P032

Systematic literature review of the prevalence and severity of somatic comorbidities in adults with phenylketonuria

Whitehall KB[1], Rose S[1], Clague GE[1], Ahring KK[2], Bilder DA[3], Harding CO[4], Hermida Á[5], Inwood A[6], Longo N[7], Maillot F[8], Muntau AC[9], Pessoa ASL[10], Rocha JC[11], Rohr F[12], Sivri S[13], Said J[14], Oshinbolu S[15], Sibbring GC[14]

[1]BioMarin Pharmaceutical Inc., Novato, CA, USA; [2]Center for PKU, Copenhagen University Hospital, Copenhagen, Denmark; [3]Huntsman Mental Health Institute, University of Utah, Salt Lake City, UT, USA; [4]Oregon Health & Science University, Portland, OR, USA; [5]University of Santiago de Compostela, Santiago de Compostela, Spain; [6]Queensland Lifespan Metabolic Medicine Service, Queensland Children's Hospital, South Brisbane, Queensland, Australia; [7]University of Utah School of Medicine, Salt Lake City, UT, USA; [8]CHRU and the University of Tours, Tours, France; [9]University Children's Hospital, University Medical Center Hamburg-Eppendorf, Hamburg, Germany; [10]Ceará State University and Hospital Infantil Albert Sabin, Fortaleza, CE, Brazil; [11]NOVA Medical School, Faculdade de Ciências Médicas (NMS|FCM), Universidade NOVA de Lisboa, Lisboa, Portugal, and Reference Centre of Inherited Metabolic Diseases, Centro Hospitalar Universitário de Lisboa Central, Lisboa, Portugal, and Nutrition & Metabolism, CINTESIS, NOVA Medical School, Faculdade de Ciências Médicas (NMS, FCM), Universidade NOVA de Lisboa, Lisboa, Portugal; [12]Met Ed Consultants, Boulder, CO, USA; [13]Hacettepe University, Ankara, Turkey; [14]Prime Global, Knutsford, UK; [15]Prime Global, London, UK

Introduction

- Phenylketonuria (PKU) is an inborn error of phenylalanine (Phe) metabolism that, if untreated, causes Phe accumulation in the blood and brain leading to neurophysiological alterations and poor developmental outcomes[1]
- Lifelong management of PKU centers on medical nutrition therapy with a Phe-restricted diet to achieve (and maintain) appropriate blood Phe levels[2,3]; sapropterin dihydrochloride is indicated for responsive individuals[4,5] and pegvaliase is an option for adults with uncontrolled blood Phe levels despite intervention[6,7]
- Many adults are unable to achieve blood Phe levels within guideline-recommended ranges, either due to waning adherence to Phe-restricted diet or an inadequate response to treatment, including due to disease severity[8,9]; uncontrolled blood Phe levels lead to adverse neurocognitive and neuropsychiatric outcomes[10-12]
- Comorbidities across organ systems have been reported in adults with PKU, with claims-based studies finding a higher prevalence of somatic comorbidities compared with a general population[13,14]
- A systematic literature review (SLR) was conducted to evaluate the prevalence and severity of non-neurocognitive and non-neuropsychiatric comorbidities in adults with PKU by intervention, disease severity, and adherence, to provide insight into specific treatment situations

Methods

- The SLR is registered with the Research Registry (reviewregistry1476)
- Eligibility criteria were established using the Population, Intervention, Comparator, Outcome, Study design (PICOS) framework (**Figure 1**)

Figure 1. Inclusion criteria established using the PICOS framework

P	**Inclusion:** Adult patients (aged ≥16 years, or as defined by the study) with confirmed PKU or described as having PKU[a] **Exclusion:** Children aged <16 years
I	**Inclusion:** • Sapropterin dihydrochloride or pegvaliase • Protein substitutes, in liquid, powdered, semi-solid, or solid forms • Low protein foods
C	**Inclusion:** • No therapeutic intervention (not receiving medical nutritional therapy/Phe-restricted diet, and/or sapropterin dihydrochloride or pegvaliase, including healthy controls or a reference population) • Protein substitutes and/or low protein foods
O	**Inclusion:** The prevalence or severity of different somatic comorbidities in patients with PKU on medical nutritional therapy/Phe-restricted diet, and/or sapropterin dihydrochloride or pegvaliase compared with: • healthy controls • the general population (including standard reference values) • patients with PKU not receiving any form of therapeutic intervention • patients with PKU who did not adhere to treatment • patients with PKU who interrupted/discontinued treatment The prevalence or severity of different somatic comorbidities in patients across the PKU disease spectrum on medical nutritional therapy/Phe-restricted diet, and/or sapropterin dihydrochloride or pegvaliase
S	**Inclusion:** Randomized controlled trials; single-arm clinical trials; cohort studies (prospective and retrospective); cross-sectional studies and surveys **Exclusion:** Systematic reviews and meta-analyses; narrative (non-systematic) review articles; animal or *in vitro* studies; letters, editorials, and commentaries; guidelines and best practice; congress abstracts; non-peer-reviewed articles
Other	**Inclusion:** Language of publication: English; date of publication: up to February 1, 2022; countries: all **Exclusion:** Language of publication: non-English

[a]Studies that included children but did not stratify outcomes by patient age were included.

- Literature was retrieved via the PubMed interface and included publications from MEDLINE from earliest coverage (1946) to February 1, 2022, using pre-defined free text and Medical Subject Headings (MeSH) search terms based on the PICOS framework
- A two-stage screening process identified records eligible for data extraction
 - Records were screened by abstract, and those considered potentially eligible were screened by full text to confirm eligibility (concordance of eligibility decisions was assessed by independent review of 10% of records selected at random)
- Data extraction was conducted by one reviewer into a pre-designed spreadsheet (Microsoft Excel®) and was checked for accuracy by an independent reviewer
- Studies were grouped by PKU populations identified and by comorbidity types
 - Vote counting was used for data synthesis, allowing direction of effect to be determined
- Vote counting followed the methods described in the Cochrane handbook[15] and was reported according to the Synthesis Without Meta-analysis (SWiM) guidelines[16]
 - A standardized binary metric was created by allocating votes to the groups compared in studies according to the direction of a higher comorbidity burden, regardless of statistical significance of differences between the groups
 - The number of votes allocated to each group was then compared to determine the direction of effect

Results

Study selection and characteristics

- In total, 1,128 unique records were screened; 459 records were identified as potentially eligible for inclusion and 73 studies spanning 10,640 patients were confirmed as eligible for inclusion (**Figure 2**)
 - Most were of observational design and most were conducted in European countries (n=53) and/or in North America (n=19)

Figure 2. PRISMA diagram showing article selection process

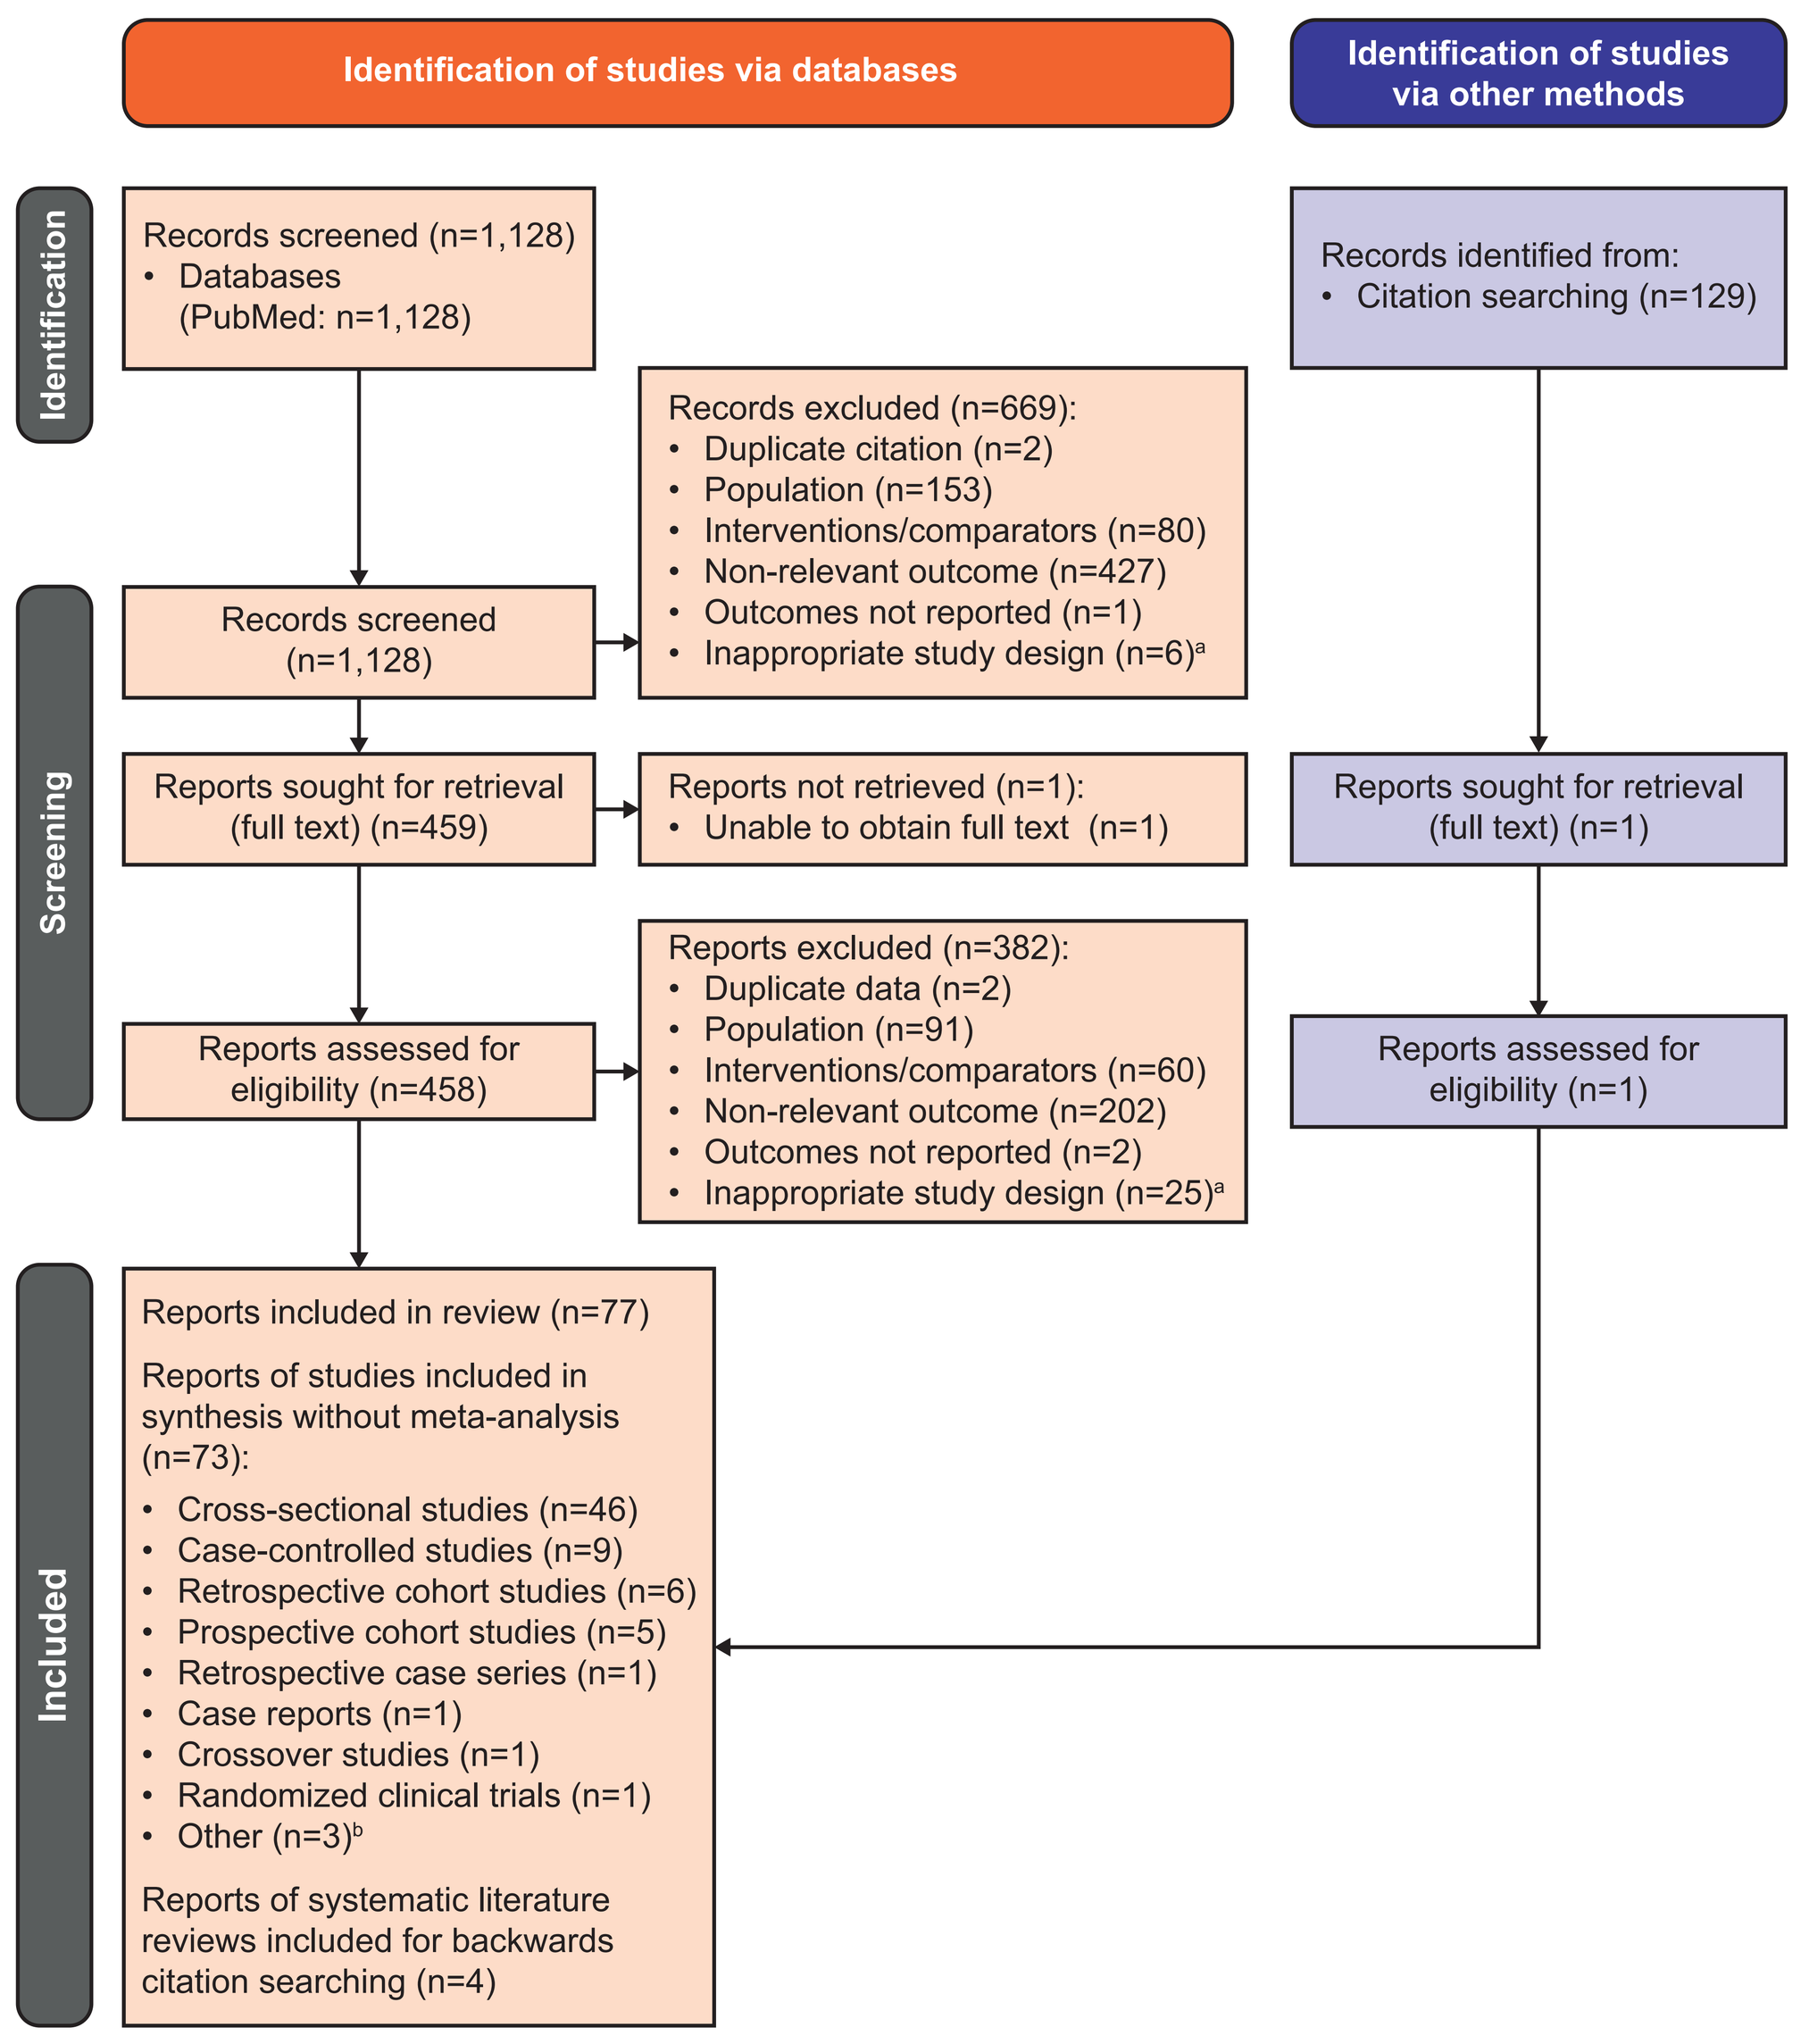
Identification of studies via databases
Identification of studies via other methods
Identification
Records screened (n=1,128)
• Databases (PubMed: n=1,128)
Records identified from:
• Citation searching (n=129)
Records excluded (n=669):
• Duplicate citation (n=2)
• Population (n=153)
• Interventions/comparators (n=80)
• Non-relevant outcome (n=427)
• Outcomes not reported (n=1)
• Inappropriate study design (n=6)a
Screening
Records screened (n=1,128)
Reports sought for retrieval (full text) (n=459)
Reports not retrieved (n=1):
• Unable to obtain full text (n=1)
Reports sought for retrieval (full text) (n=1)
Reports excluded (n=382):
• Duplicate data (n=2)
• Population (n=91)
• Interventions/comparators (n=60)
• Non-relevant outcome (n=202)
• Outcomes not reported (n=2)
• Inappropriate study design (n=25)a
Reports assessed for eligibility (n=458)
Reports assessed for eligibility (n=1)
Included
Reports included in review (n=77)
Reports of studies included in synthesis without meta-analysis (n=73):
• Cross-sectional studies (n=46)
• Case-controlled studies (n=9)
• Retrospective cohort studies (n=6)
• Prospective cohort studies (n=5)
• Retrospective case series (n=1)
• Case reports (n=1)
• Crossover studies (n=1)
• Randomized clinical trials (n=1)
• Other (n=3)b
Reports of systematic literature reviews included for backwards citation searching (n=4)

PRISMA = Preferred Reporting Items for Systematic Reviews and Meta-Analyses. Articles were excluded on a hierarchical basis, in the order that questions were asked (i.e., if the answer to the first question was no, this was given as the main reason for exclusion, but articles may have met or not met other criteria).
[a]Includes studies that did not present outcomes in a meaningful way which answered one or more of the pre-specified research questions.
[b]Includes open interventional trials, pooled analyses, and cost analyses.

- More than 15 different comorbidity types were reported across the 73 studies: white matter abnormalities and bone-related abnormalities were the most commonly reported
- Studies were grouped according to PKU populations (**Table 1**) and then according to comorbidity type

Table 1. Grouping of studies by PKU population

Study groupings		Number of studies[a]	Number of patients[a]	Vote counting conducted
Intervention population	Comparator population			
Patients with PKU on a Phe-restricted diet with or without pharmacological therapy	Healthy controls or reference values	58[13,14,17-72]	9,938	Yes[b]
Patients with PKU adherent to a Phe-restricted diet	Patients with PKU non-adherent to a Phe-restricted diet	8[8,44,65,73-77]	417	Yes[c]
Patients with PKU on a specific Phe-restricted diet	Patients with PKU on a different Phe-restricted diet	4[78-81]	96	No[d]
Patients with more severe PKU on a Phe-restricted diet[e]	Patients with HPA or less severe PKU on a Phe-restricted diet[e]	3[82-84]	123	No[d]
Patients with PKU treated with pegvaliase or sapropterin dihydrochloride[f]	Patients with PKU treated with placebo	2[85,86]	166	No[d]

HPA = hyperphenylalaninemia.
[a]Two studies[44,65] are counted twice as both compare patients with PKU on a Phe-restricted diet with or without pharmacological therapy versus healthy controls or reference values, and patients with PKU who adhered to a Phe-restricted diet versus non-adherent. [b]Vote counting was conducted on 47 studies: nine of the 58 studies did not report results for the healthy controls group and were excluded; another two studies were also excluded as it was not possible to confirm treatment with a Phe-restricted diet in the full study population. [c]Vote counting was conducted on seven studies: one study was excluded due to no clear correlation between the severity of comorbidity and dietary history of the patients. [d]Fewer than five studies resulting in insufficient data, vote counting could not be conducted. [e]Includes studies of patients with classical PKU versus patients with mild/moderate PKU and patients with PKU versus patients with HPA. [f]One study[85] included five participants who followed a Phe-restricted diet (study did not specify whether these patients were in the pegvaliase or placebo group). Another study[86] did not indicate patients were on a Phe-restricted diet.

Data synthesis

- In the vote-counting analysis shown in **Figure 3**, a higher burden of ≥1 comorbidity (or outcome measure) in patients with PKU versus healthy controls or reference values was indicated in all 47 studies, and a higher burden of ≥1 comorbidity (or outcome measure) in healthy controls or reference values versus patients with PKU was indicated in seven studies
 - Bone-related abnormalities and white matter abnormalities were the most commonly reported comorbidities with a higher burden of ≥1 comorbidity (or outcome measure) in patients with PKU on a Phe-restricted diet with or without pharmacological therapy versus healthy controls or reference values

Figure 3. Burden of comorbidities in patients with PKU on a Phe-restricted diet with or without pharmacological therapy versus healthy controls or reference values (assessed by vote counting, n=47 studies)

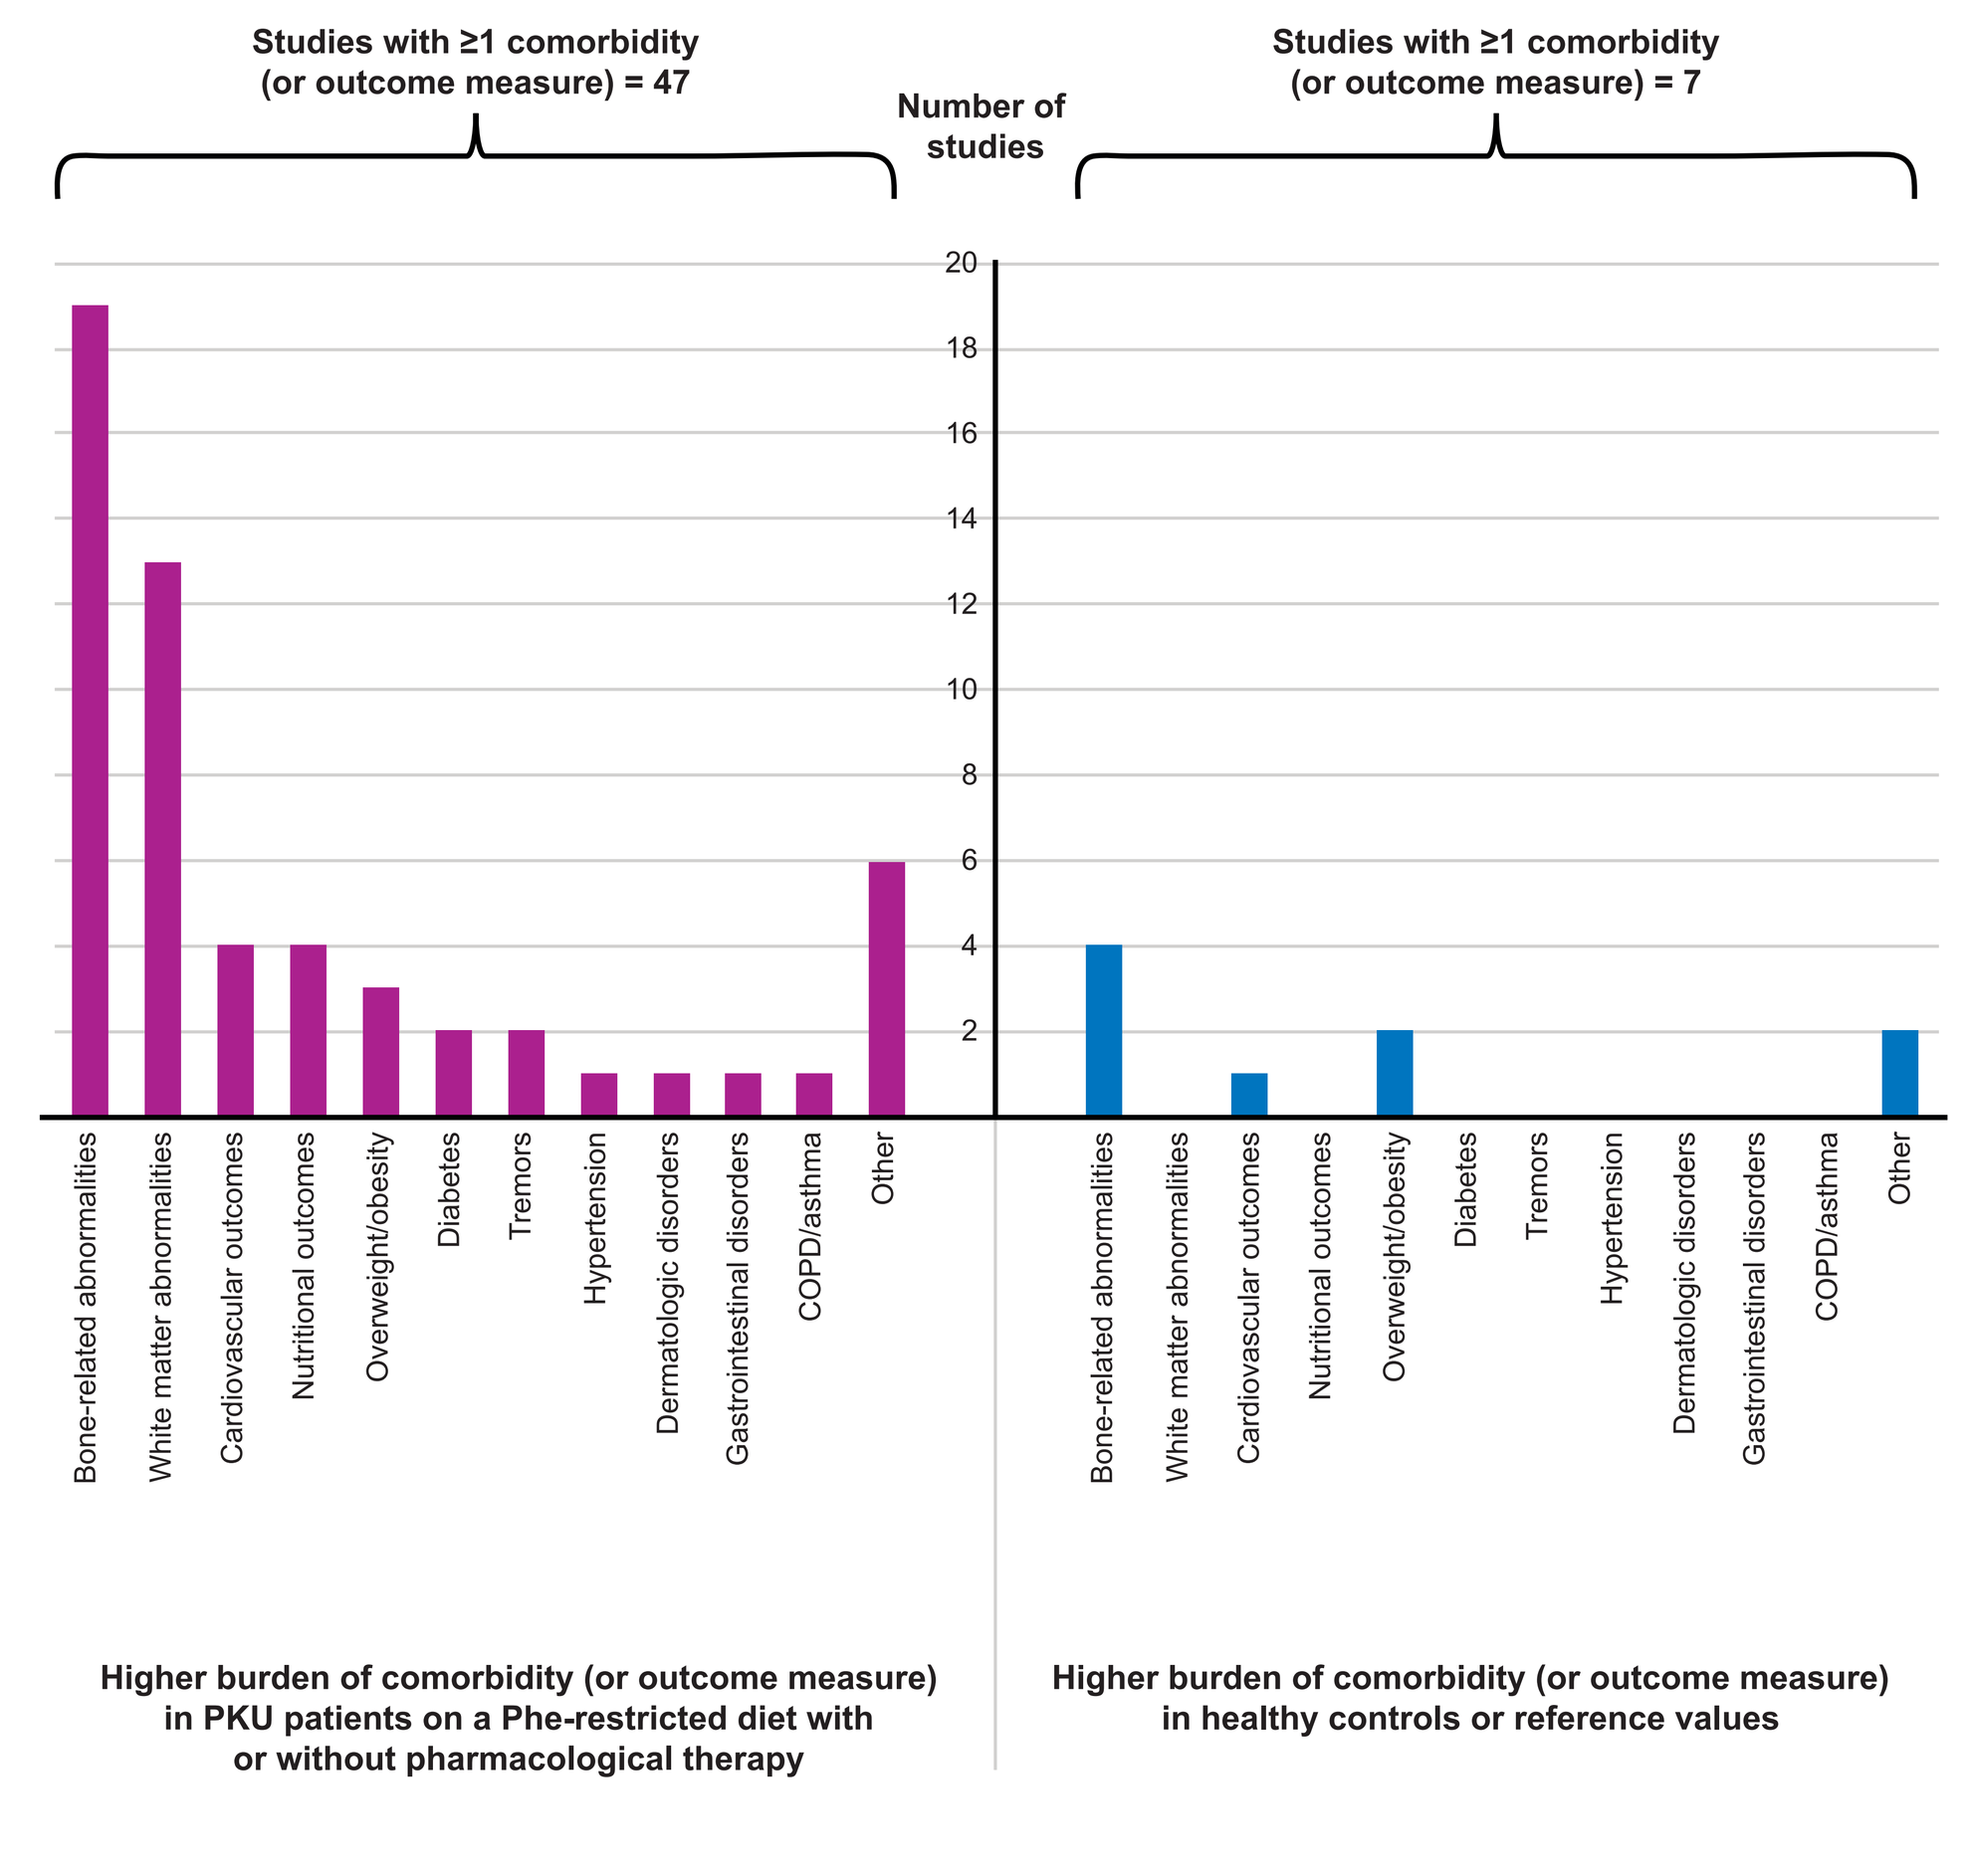
Studies with ≥1 comorbidity (or outcome measure) = 47
Number of studies
Studies with ≥1 comorbidity (or outcome measure) = 7
20
18
16
14
12
10
8
6
4
2
Bone-related abnormalities
White matter abnormalities
Cardiovascular outcomes
Nutritional outcomes
Overweight/obesity
Diabetes
Tremors
Hypertension
Dermatologic disorders
Gastrointestinal disorders
COPD/asthma
Other
Higher burden of comorbidity (or outcome measure) in PKU patients on a Phe-restricted diet with or without pharmacological therapy
Higher burden of comorbidity (or outcome measure) in healthy controls or reference values

COPD = chronic obstructive pulmonary disease.
Figure indicates the number of studies with a higher burden of ≥1 comorbidity or outcome measure (some studies reported more than one comorbidity/outcome measure or had a differing direction of effect between comorbidities/outcome measure). Vote counting was conducted regardless of statistical significance. In most studies, all patients were on a Phe-restricted diet, except: one study with a mixture of patients on and not on a Phe-restricted diet[23]; one study in which some patients received sapropterin, some patients were on a Phe-restricted diet, and, for some patients, it was not clear whether they were on a Phe-restricted diet or not[14]; and one study in which some patients were treated with sapropterin in addition to dietary treatment[35].
Bone-related abnormalities: Higher burden in PKU (n=19)[14,17,22,23,26,28,29,33,40,42-45,50,56,57,63,70,71]; higher burden in controls (n=4)[17,23,29,45]. **White matter abnormalities:** Higher burden in PKU (n=13)[30,31,34,39,41,49,52-55,64,66,69]. **Cardiovascular outcomes:** Higher burden in PKU (n=4)[14,18,35,36]; higher burden in controls (n=1)[36]. **Nutritional outcomes:** Higher burden in PKU (n=4)[37,38,67,68]. **Overweight/obesity:** Higher burden in PKU (n=3)[14,58,60]; higher burden in controls (n=2)[58,60]. **Diabetes:** Higher burden in PKU (n=2)[27]. **Tremors:** Higher burden in PKU (n=2)[47,53]. **Hypertension:** Higher burden in PKU (n=1)[14]. **Dermatologic disorders:** Higher burden in PKU (n=1)[14]. **Gastrointestinal disorders:** Higher burden in PKU (n=1)[14]. **COPD/asthma:** Higher burden in PKU (n=1)[14]. **Other:** Higher burden in PKU (n=6)[14,20,23-25,65]; higher burden in controls (n=2)[23,60].

- A range of outcome measures for bone-related abnormalities and white matter abnormalities was reported across studies (**Figure 4**)
 - The most commonly reported (in ≥4 studies) outcome measures for bone-related abnormalities were Z-scores, markers for bone resorption and bone formation, and prevalence of osteopenia/osteoporosis
 - The most commonly reported (in ≥4 studies) outcome measures for white matter abnormalities were magnetic resonance imaging grading, apparent diffusion coefficient, and fractional anisotropy
- In the vote-counting analysis comparing the burden of comorbidities in patients with PKU adhering to a Phe-restricted diet versus those non-adherent, four studies indicated a higher burden of ≥1 comorbidity (or outcome measure) in patients who adhered versus those non-adherent, and six studies indicated a higher burden of ≥1 comorbidity (or outcome measure) in patients who did not adhere versus those adherent
- A range of comorbidities was reported in the other PKU population comparisons:
 - White matter abnormalities were reported in a study of patients with classical PKU versus mild/moderate disease (including a mixture of patients off diet and patients on a Phe-restricted diet); no significant difference was found in terms of the severity of white matter involvement between the disease severity groups (p=0.36)[82]
 - Bone-related abnormalities were reported in a study of patients who were on a Phe-restricted diet with classical PKU versus mild/moderate disease; the prevalence of osteopenia and osteoporosis was reported to be similar between the disease severity groups (no statistical comparison reported)[83]
 - Nutritional outcomes were measured in patients with PKU and patients with HPA; differences in the concentrations of serum pre-albumin, zinc, and iron between groups were not statistically significant, but there was a statistically significant lower concentration of selenium in adult patients with PKU compared with adult patients with HPA (p=0.006)[84]
 - Headache was reported as an adverse event in studies of pegvaliase or sapropterin dihydrochloride[85,86]
 - One study of patients with PKU reported more headaches in the placebo group than in the pegvaliase group (no statistical comparison reported)[85]
 - Another study reported headaches in patients with PKU or HPA receiving sapropterin dihydrochloride; headaches (in 1 patient), which were considered to be probably related to sapropterin and headaches (in 8 patients) and migraines (in 4 patients), which were considered to be possibly related to sapropterin[86]
 - Three studies evaluating different Phe-restricted diets reported on prevalence of overweight/obesity[78-80]. Two studies reported no significant differences between diets (p=1.000[79], p=0.367[80]); one study reported body mass index remained unchanged between diets[78]

Figure 4. Measures used to report A) bone-related abnormalities and B) white matter abnormalities in patients with PKU on a Phe-restricted diet with or without pharmacological therapy versus healthy controls or reference values

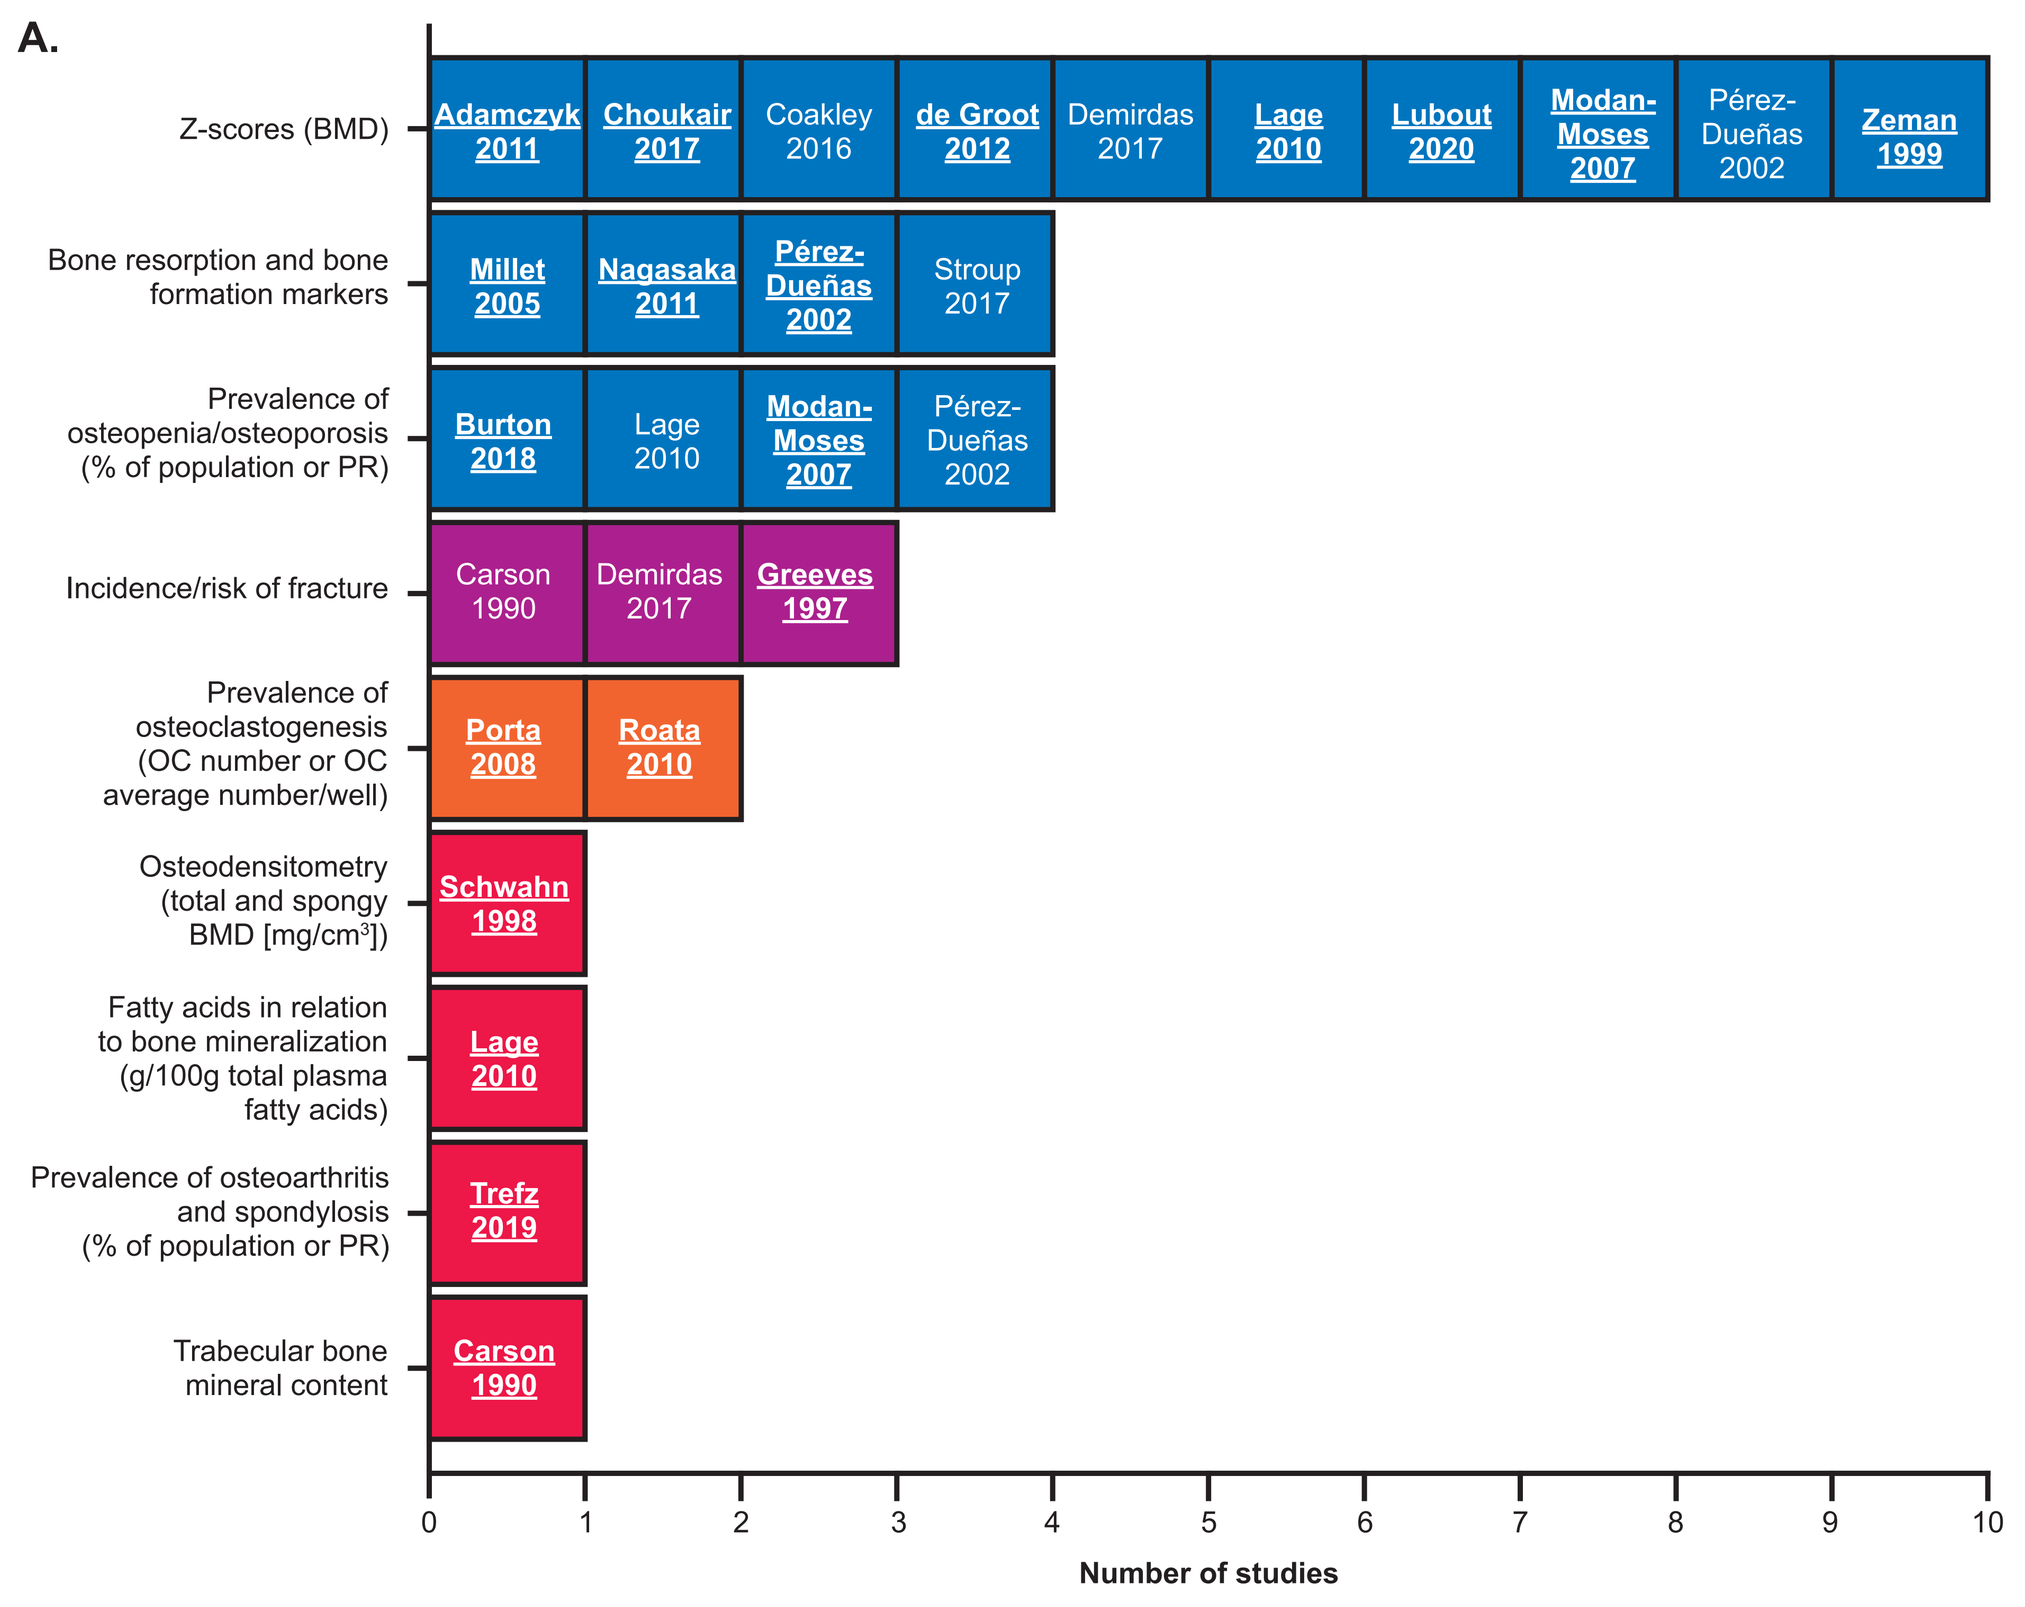
A.
Z-scores (BMD)
Adamczyk 2011
Choukair 2017
Coakley 2016
de Groot 2012
Demirdas 2017
Lage 2010
Lubout 2020
Modan-Moses 2007
Pérez-Dueñas 2002
Zeman 1999
Bone resorption and bone formation markers
Millet 2005
Nagasaka 2011
Pérez-Dueñas 2002
Stroup 2017
Prevalence of osteopenia/osteoporosis (% of population or PR)
Burton 2018
Lage 2010
Modan-Moses 2007
Pérez-Dueñas 2002
Incidence/risk of fracture
Carson 1990
Demirdas 2017
Greeves 1997
Prevalence of osteoclastogenesis (OC number or OC average number/well)
Porta 2008
Roata 2010
Osteodensitometry (total and spongy BMD [mg/cm3])
Schwahn 1998
Fatty acids in relation to bone mineralization (g/100g total plasma fatty acids)
Lage 2010
Prevalence of osteoarthritis and spondylosis (% of population or PR)
Trefz 2019
Trabecular bone mineral content
Carson 1990
0 1 2 3 4 5 6 7 8 9 10
Number of studies

BAP = bone alkaline phosphatase; BMD = bone mineral density; ICTP = pyridinoline cross-linked telopeptide domain of type I collagen; OC = osteoclastogenesis; PR = prevalence ratio.
Underlined studies showed a statistically significant difference between groups. All 20 studies indicated a higher prevalence and/or severity in the PKU group compared with healthy controls or reference values; with 17 reporting a statistically significant difference[13,14,17,22,23,28,33,40,42-45,50,56,57,63,70], one that did not find a statistically significant difference[26], and two that did not test for statistical significance between PKU group and controls[29,71]. Further details on this figure can be found in the forthcoming manuscript.

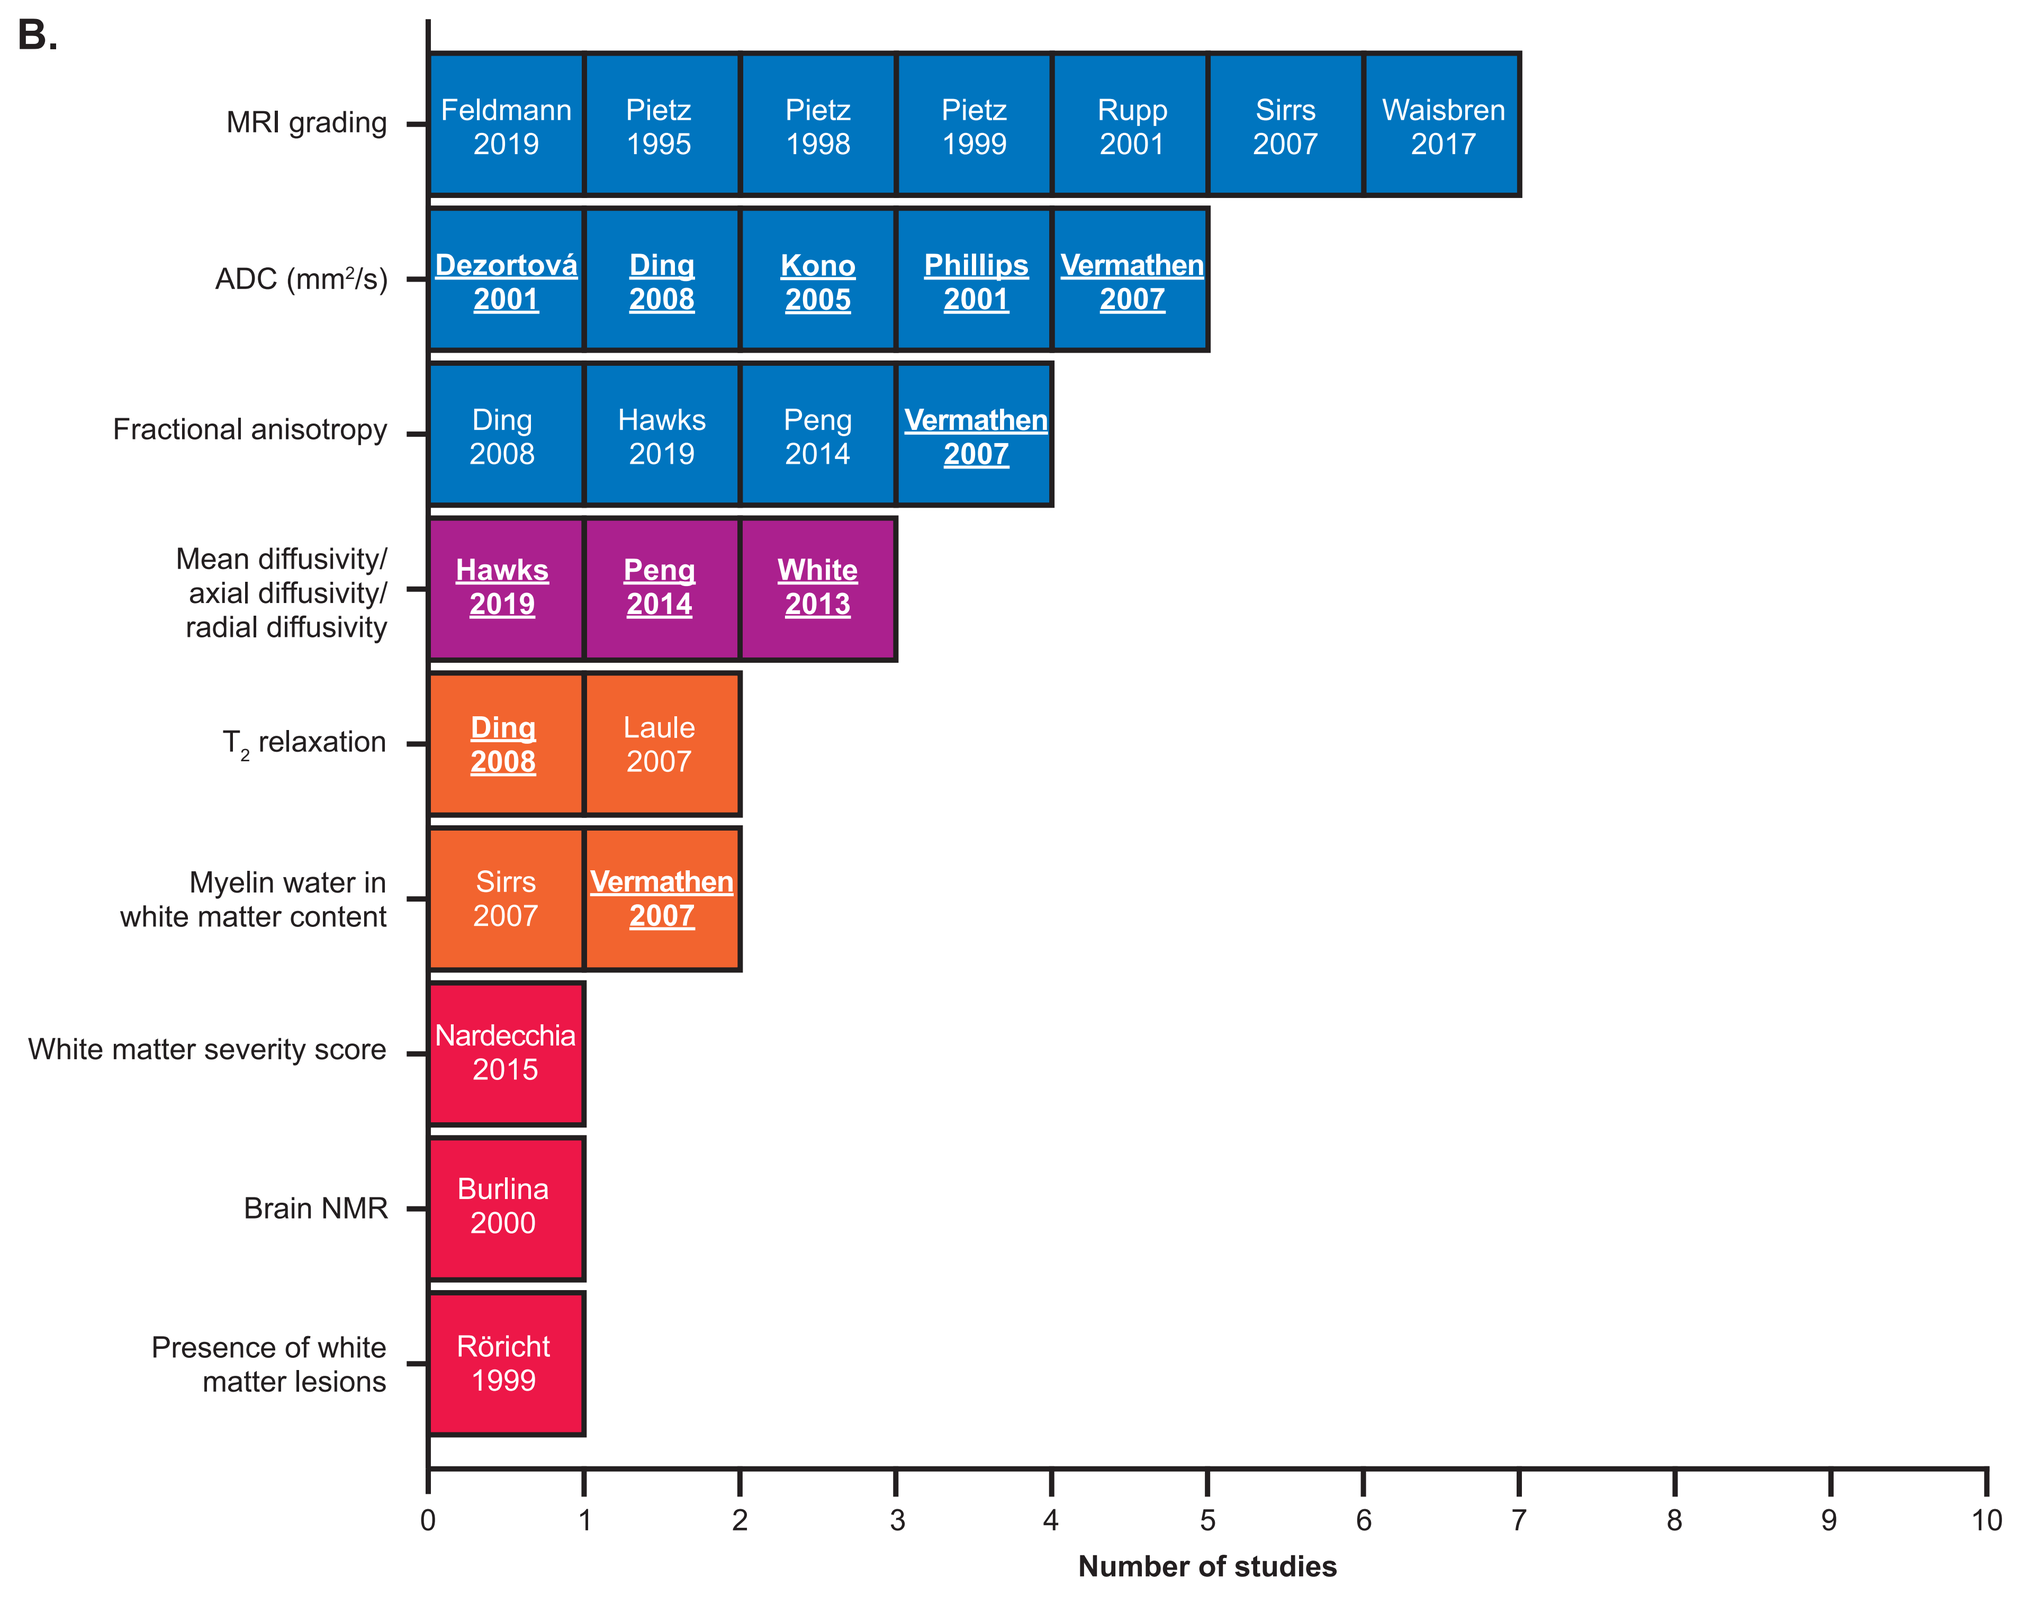
B.
MRI grading
Feldmann 2019
Pietz 1995
Pietz 1998
Pietz 1999
Rupp 2001
Sirrs 2007
Waisbren 2017
ADC (mm2/s)
Dezortová 2001
Ding 2008
Kono 2005
Phillips 2001
Vermathen 2007
Fractional anisotropy
Ding 2008
Hawks 2019
Peng 2014
Vermathen 2007
Mean diffusivity/ axial diffusivity/ radial diffusivity
Hawks 2019
Peng 2014
White 2013
T2 relaxation
Ding 2008
Laule 2007
Myelin water in white matter content
Sirrs 2007
Vermathen 2007
White matter severity score
Nardecchia 2015
Brain NMR
Burlina 2000
Presence of white matter lesions
Röricht 1999
0 1 2 3 4 5 6 7 8 9 10
Number of studies

ADC = Apparent diffusion coefficient; IW = intermediate weighted; NMR = nuclear magnetic resonance.
Out of 19 studies, seven studies did not report outcome values for healthy controls despite a clear indication of a control group, and therefore the results are not comparative[21,32,54,61,62,72]. From the 12 studies that did report outcome values for both healthy controls/reference values and patients with PKU[30,31,34,39,41,46,49,52,53,55,66,69], all 12 indicated a higher prevalence and/or severity in the PKU group compared with healthy controls or reference values; with eight studies reporting statistical significance[30,31,34,39,49,52,66,69] and four studies that did not test for statistical significance[41,53,55,64]. Underlined studies showed a statistically significant difference between groups. Further details on this figure can be found in the forthcoming manuscript.

Conclusions

- This SLR provides evidence for an unmet need in the current treatment landscape, highlighting the higher somatic, non-neurocognitive and non-neuropsychiatric comorbidity burden on patients with PKU versus a non-PKU population. To potentially avoid the clinical and economic implications of managing comorbidities, there is a need for improved access to therapeutic interventions to maintain blood Phe levels within recommended ranges over the long term
- No conclusions could be drawn from vote counting of the diet adherent versus non-adherent populations, as numbers of studies in each population indicating a higher burden were similar
- The scarcity of data from studies using similar designs and patient populations, as well as consistency of outcome measures for many comorbidities, restricted the synthesis methods that could be used to evaluate the somatic comorbidity burden
 - Although vote counting is considered an acceptable alternative when meta-analysis is not feasible[15], not all studies could be included in the analysis due to lack of comparative data; the analysis does not account for differences in the relative sizes of the studies or methodological aspects, and provides no information on the magnitude of effect
- More robust studies reporting consistent outcome measures and evaluating the relationship between effective metabolic control and comorbidity burden are needed

References

1. Williams RA et al. *Clin Biochem Rev.* 2008;29:31-41. 2. Vockley J et al. *Genet Med.* 2014;16:188-200. 3. van Wegberg AMJ et al. *Orphanet J Rare Dis.* 2017;12:162. 4. BioMarin Pharmaceutical Inc. KUVAN Summary of Product Characteristics. 2020. Available at: https://www.ema.europa.eu/en/documents/product-information/kuvan-epar-product-information_en.pdf. Accessed January 19, 2023. 5. BioMarin Pharmaceutical Inc. KUVAN Prescribing information. 2021. Available at: http://kuvan.com/wp-content/file/KUVAN_Prescribing_Information1.pdf. Accessed January 19, 2023. 6. BioMarin Pharmaceutical Inc. PALYNZIQ Summary of Product Characteristics. 2022. Available at: https://www.ema.europa.eu/en/documents/product-information/palynziq-epar-product-information_en.pdf. Accessed January 19, 2023. 7. BioMarin Pharmaceutical Inc. PALYNZIQ Prescribing information. 2020. Available at: https://www.palynziq.com/prescribinginformation.pdf. Accessed January 19, 2023. 8. Green B et al. *Nutrients.* 2019;11. 9. Kanufre V et al. *Nutrients.* 2021;13:3118. 10. Pilotto A et al. *Neurology.* 2021;96:e399-e411. 11. Bilder DA et al. *Dev Neuropsychol.* 2016;41:245-260. 12. Romani C et al. *Neurosci Biobehav Rev.* 2022;143:104925. 13. Burton BK et al. *Mol Genet Metab.* 2018;125:228-234. 14. Trefz KF et al. *Orphanet J Rare Dis.* 2019;14:181. 15. McKenzie JE et al. Cochrane Handbook for Systematic Reviews of Interventions. Chapter 12: Synthesizing and presenting findings using other methods. 2022. Available at: https://training.cochrane.org/handbook/current/chapter-12. Accessed January 19, 2023.

16. Campbell M et al. *BMJ.* 2020;368:l6890. 17. Adamczyk P et al. *J Bone Miner Metab.* 2011;29:236-244. 18. Azabdaftari A et al. *Orphanet J Rare Dis.* 2019;14:213. 19. Bilder DA et al. *Mol Genet Metab.* 2017;121:1-8. 20. Bodner KE et al. *Mol Genet Metab.* 2012;107:302-307. 21. Burlina AB et al. *J Inherit Metab Dis.* 2000;23:313-316. 22. Carson DJ et al. *Pediatr Radiol.* 1990;20:598-599. 23. Choukair D et al. *J Inherit Metab Dis.* 2017;40:219-226. 24. Christ SE et al. *Mol Genet Metab.* 2010;99 Suppl 1:S33-40. 25. Christ SE et al. *Mol Genet Metab.* 2016;118:3-8. 26. Coakley KE et al. *J Inherit Metab Dis.* 2016;39:363-372. 27. Couce ML et al. *Orphanet J Rare Dis.* 2018;13:103. 28. de Groot MJ et al. *Mol Genet Metab.* 2012;105:566-570. 29. Demirdas S et al. *Ann Nutr Metab.* 2017;70:111-121. 30. Dezortová M et al. *Acta Radiol.* 2001;42:459-466. 31. Ding XQ et al. *J Magn Reson Imaging.* 2008;27:998-1004. 32. Feldmann R et al. *Mol Genet Metab.* 2019;126:246-249. 33. Greeves LG et al. *Acta Paediatr.* 1997;86:242-244. 34. Hawks Z et al. *Neuroimage Clin.* 2019;23:101916. 35. Hermida-Ameijeiras A et al. *Medicine (Baltimore).* 2017;96:e9322. 36. Htun P et al. *PLoS One.* 2015;10:e0135930. 37. Huemer M et al. *Mol Genet Metab.* 2008;94:46-51. 38. Hvas AM et al. *J Inherit Metab Dis.* 2006;29:47-53. 39. Kono K et al. *Radiology.* 2005;236:630636. 40. Lage S et al. *J Inherit Metab Dis.* 2010;33 Suppl 3:S363-371. 41. Laule C et al. *J Magn Reson Imaging.* 2007;26:1117-1121. 42. Lubout CMA et al. *J Inherit Metab Dis.* 2020;43:251-258. 43. Millet P et al. *Clin Biochem.* 2005;38:1127-1132. 44. Modan-Moses D et al. *J Inherit Metab Dis.* 2007;30:202-208. 45. Nagasaka H et al. *J Bone Miner Metab.* 2011;29:737-743. 46. Nardecchia F et al. *Mol Genet Metab.* 2015;115:84-90. 47. Nardecchia F et al. *Mol Genet Metab.* 2019;128:53-56. 48. Pedersen HE. *Acta Neurol Scand.* 1974;50:599-610. 49. Peng H et al. *J Inherit Metab Dis.* 2014;37:237-243. 50. Pérez-Dueñas B et al. *Acta Paediatr.* 2002;91:899-904. 51. Pérez-Dueñas B et al. *Neurology.* 2006;66:1074-1078.

52. Phillips MD et al. *AJNR Am J Neuroradiol.* 2001;22:1583-1586. 53. Pietz J et al. *Eur J Pediatr.* 1998;157:824-830. 54. Pietz J et al. *Pediatr Res.* 1995;38:657-663. 55. Pietz J et al. *J Clin Invest.* 1999;103:1169-1178. 56. Porta F et al. *J Inherit Metab Dis.* 2008;31 Suppl 2:S339-342. 57. Roato I et al. *PLoS One.* 2010;5:e14167. 58. Robertson LV et al. *J Hum Nutr Diet.* 2013;26 Suppl 1:1-6. 59. Robinson M et al. *J Pediatr.* 2000;136:545-547. 60. Rocha JC et al. *Mol Genet Metab.* 2012;107:659-663. 61. Röricht S et al. *J Neurol.* 1999;246:21-30. 62. Rupp A et al. *J Cereb Blood Flow Metab.* 2001;21:276-284. 63. Schwahn B et al. *Acta Paediatr.* 1998;87:61-63. 64. Sirrs SM et al. *Radiology.* 2007;242:236-243. 65. Sumanszki C et al. *Ann Nutr Metab.* 2019;75:16-23. 66. Vermathen P et al. *Magn Reson Med.* 2007;58:1145-1156. 67. Vugteveen I et al. *Mol Genet Metab.* 2011;102:13-17. 68. Weigel C et al. *Ann Nutr Metab.* 2008;53:91-95. 69. White DA et al. *Mol Genet Metab.* 2013;110:213-217. 70. Zeman J et al. *Acta Paediatr.* 1999;88:1348-1351. 71. Stroup BM et al. *J Nutr Metab.* 2017;2017:1909101. 72. Waisbren SE et al. *JIMD Rep.* 2017;34:77-86. 73. Guest JF et al. *J Intellect Disabil Res.* 2013;57:567-579. 74. Koch R et al. *J Inherit Metab Dis.* 2002;25:333-346. 75. Schulz B et al. *Acta Paediatr.* 1995;84:743-748. 76. Pearsen KD et al. *Radiology.* 1990;177:437-440. 77. Shaw DW et al. *AJNR Am J Neuroradiol.* 1991;12:403-406. 78. MacDonald A et al. *J Inherit Metab Dis.* 2004;27:127-135. 79. Pena MJ et al. *Orphanet J Rare Dis.* 2021;16:84. 80. Pinto A et al. *Eur J Clin Nutr.* 2017;71:1230-1234. 81. Das AM et al. *JIMD Rep.* 2014;13:149-158. 82. Leuzzi V et al. *J Inherit Metab Dis.* 2007;30:209-216. 83. Mirás A et al. *Mol Genet Metab.* 2013;108:149-154. 84. Procházková D et al. *Biol Trace Elem Res.* 2013;154:178-184. 85. Harding CO et al. *Mol Genet Metab.* 2018;124:20-26. 86. Lee P et al. *Am J Med Genet A.* 2008;146a:2851-2859.

Acknowledgements

Technical support, including screening and data extraction, was provided by Sofie Norregaard, MSc; medical writing support was provided by Alison Blackburn, PhD, and editorial support was provided by Michelle Seddon, Dip Psych, fact checking was supported by Penelope Cervelo Bouzo, MRes, and Philippine Sauzey, MSc, all of Prime Global, UK, and supported by BioMarin, according to Good Publication Practice guidelines. The Sponsor was involved in the study design, collection, analysis, and interpretation of data, as well as data checking of information provided in the manuscript. However, ultimate responsibility for opinions, conclusions, and data interpretation lies with the authors.

Disclosures

SR is an employee of BioMarin.

Funding

This work was supported by BioMarin.

Presented at the 2023 ACMG Annual Clinical Genetics Meeting: March 14–18, Salt Lake City, UT, USA

©2023 BioMarin Pharmaceutical Inc. All rights reserved.