184/PF

Phase 3 PRISM studies: Efficacy and safety of pegvaliase in patients aged 16 and 17 years with phenylketonuria

Vockley J[1], Stuy MZ[2], Northrup H[3], Zori RT[4], Lounsbury D[5], Li M[5], Olbertz J[5], Weng HH[5], Thomas JA[6]

[1]University of Pittsburgh and Children's Hospital of Pittsburgh, Pittsburgh, PA, USA; [2]Indiana University, Indianapolis, IN, USA; [3]University of Texas Health Science Center at Houston, Houston, TX, USA; [4]University of Florida, Gainesville, FL, USA; [5]BioMarin Pharmaceutical Inc., Novato, CA, USA; [6]University of Colorado School of Medicine and Children's Hospital Colorado, Aurora, CO, USA

Background

- Phenylketonuria (PKU) is caused by deficiency of the phenylalanine hydroxylase enzyme resulting in phenylalanine (Phe) accumulation[1]
- Pegvaliase, PEGylated recombinant *Anabaena variabilis* phenylalanine ammonia lyase (PAL), converts Phe to trans-cinnamic acid and ammonia[2–4]
- Pegvaliase (Palynziq®) was approved for PKU patients with uncontrolled blood Phe concentrations >600 µmol/L on existing management in the US in May 2018 for adults (≥18 years) at doses of up to 40 mg once daily[5] and by the European Commission in May 2019 for patients ≥16 years at doses of up to 60 mg once daily[6]
- Adolescent/young adult PKU patients aged 16 to 17 years with blood Phe levels ≤360 µmol/L have been shown to perform better at neuropsychological and neurocognitive tasks[7,8] and elicit improvements in behavior and school performance following reduction in blood Phe below this threshold[9]

Methods

- Study designs for PRISM-1 (165-301, NCT01819727) and PRISM-2 (165-302, NCT01889862) are shown in **Figure 1**
- Herein we report on the safety, efficacy, and immunogenicity of pegvaliase for the 11 subjects who were aged 16 or 17 years at the time of consent
- Final study results are reported from the last study visit on 5 February 2019

Figure 1. Study design of PRISM-1 and PRISM-2

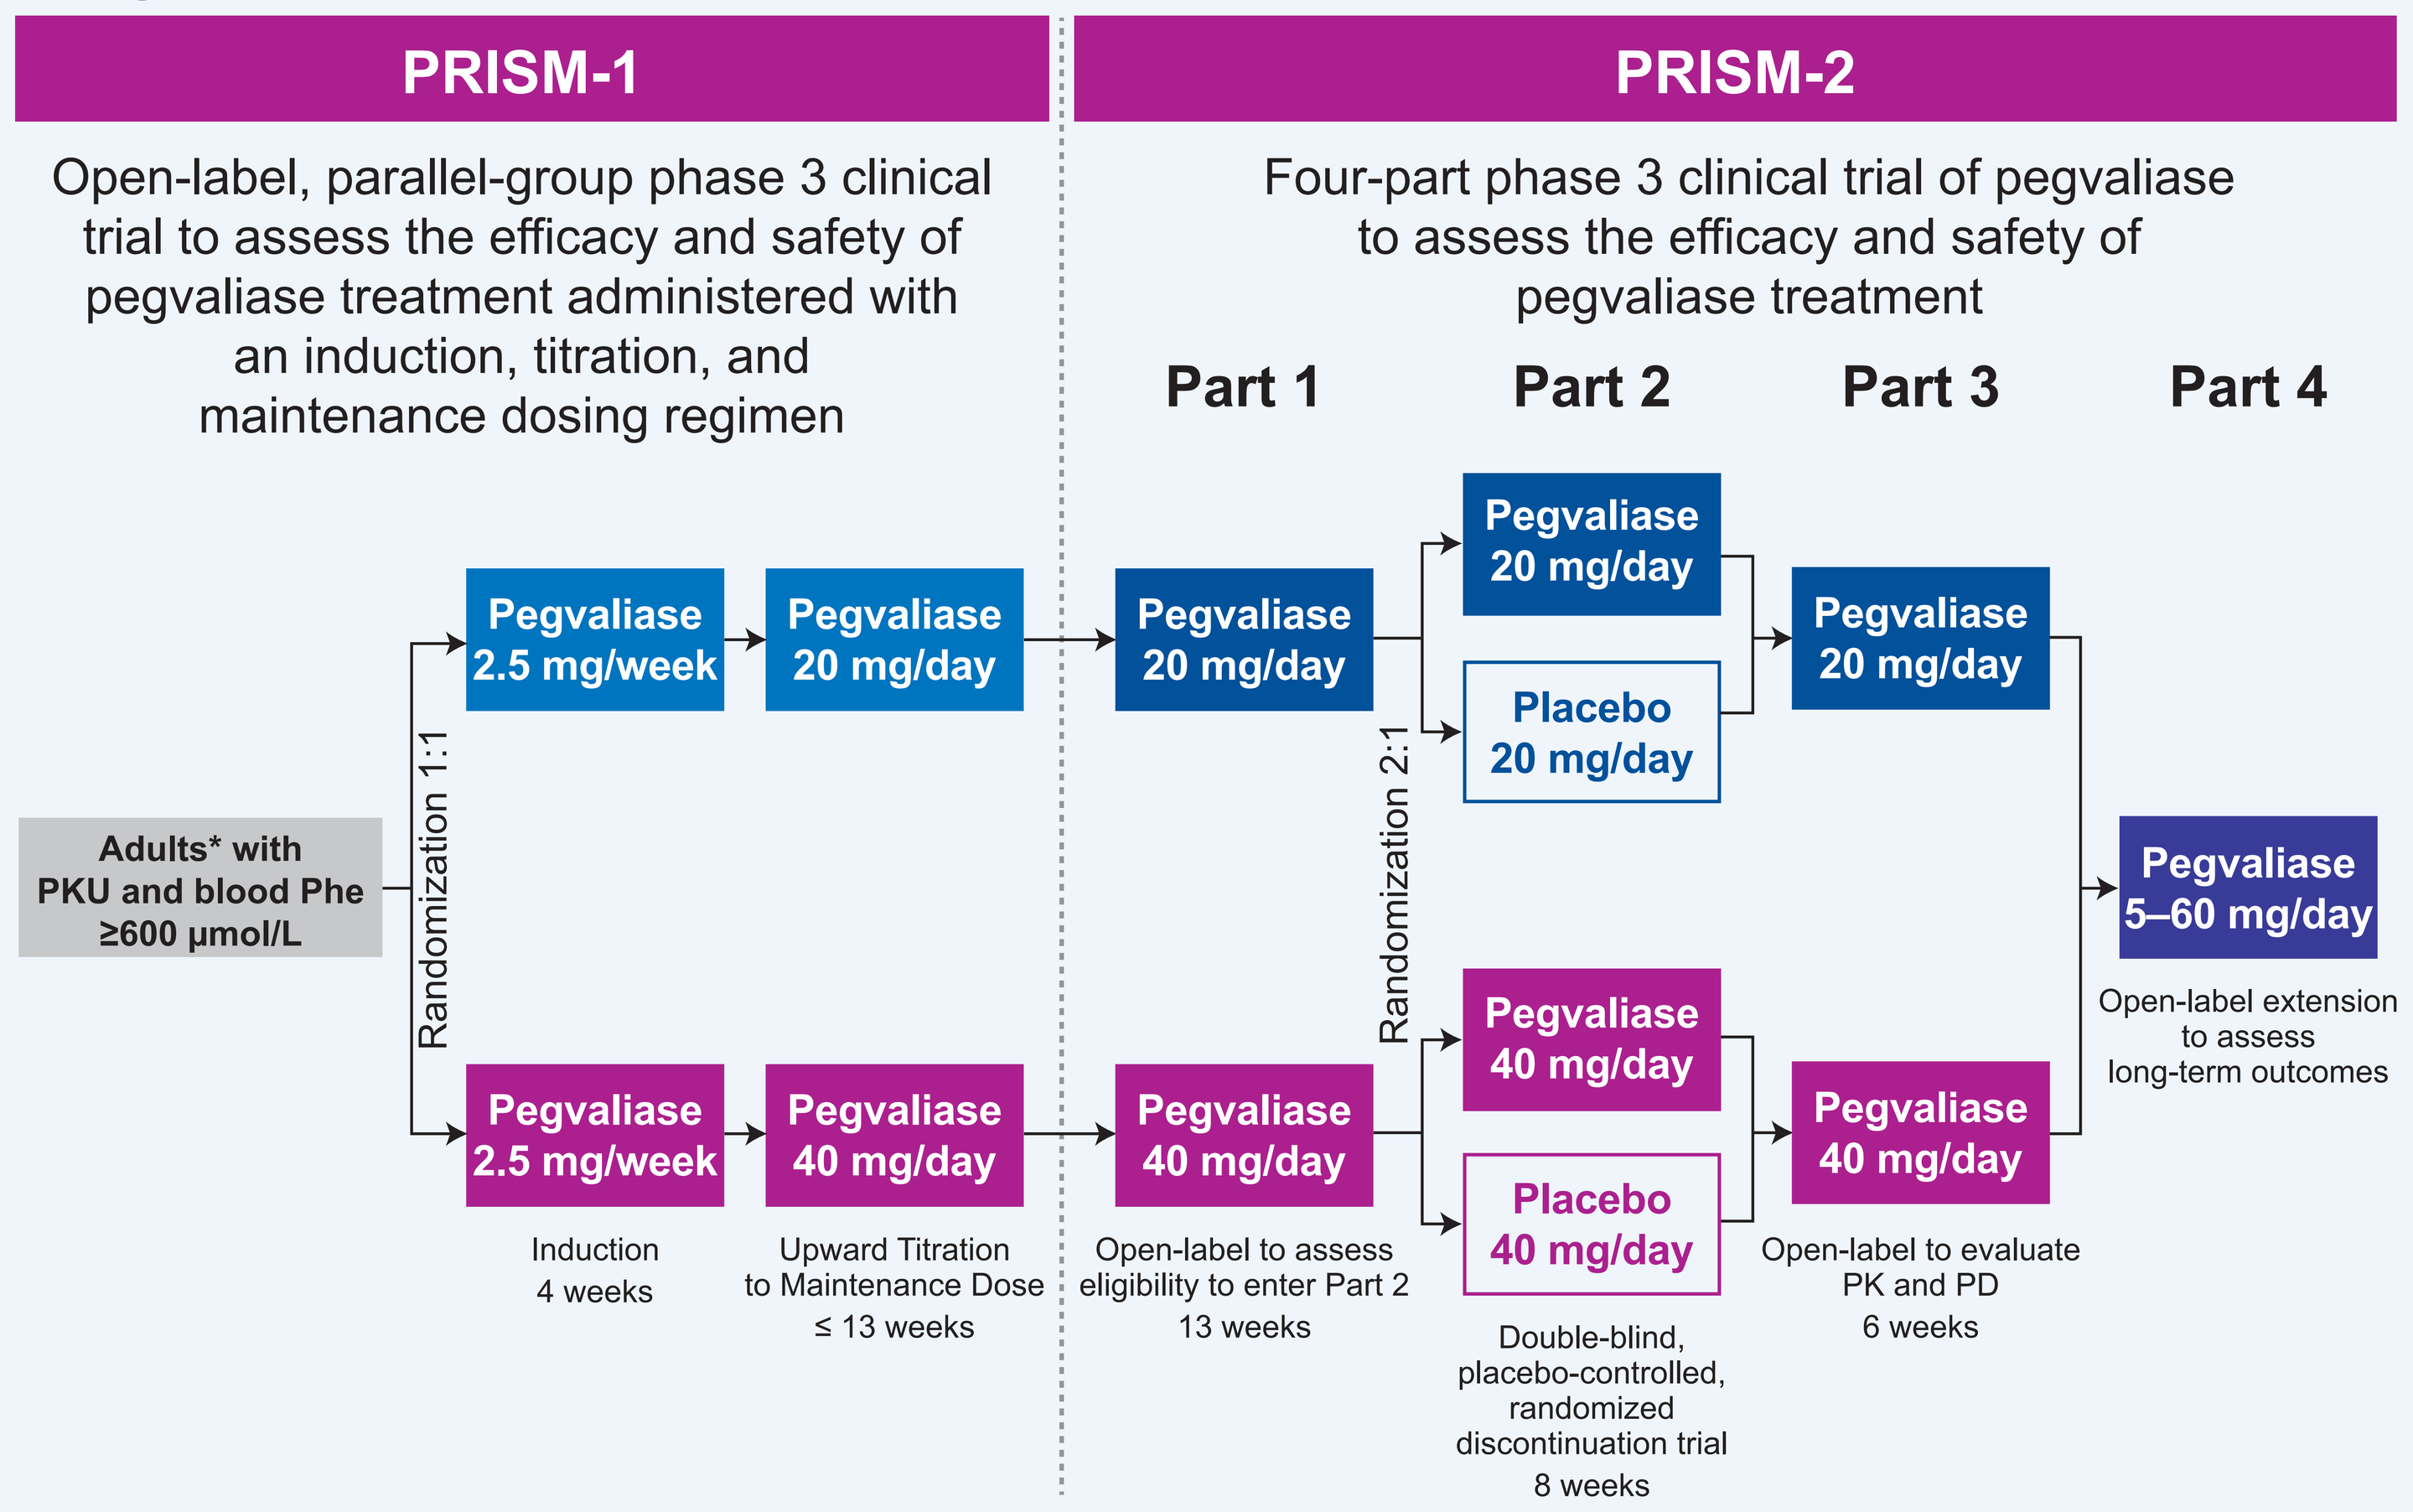
PRISM-1
Open-label, parallel-group phase 3 clinical trial to assess the efficacy and safety of pegvaliase treatment administered with an induction, titration, and maintenance dosing regimen
PRISM-2
Four-part phase 3 clinical trial of pegvaliase to assess the efficacy and safety of pegvaliase treatment
Part 1
Part 2
Part 3
Part 4
Adults* with PKU and blood Phe ≥600 µmol/L
Randomization 1:1
Pegvaliase 2.5 mg/week
Pegvaliase 20 mg/day
Pegvaliase 2.5 mg/week
Pegvaliase 40 mg/day
Induction 4 weeks
Upward Titration to Maintenance Dose ≤ 13 weeks
Pegvaliase 20 mg/day
Pegvaliase 40 mg/day
Open-label to assess eligibility to enter Part 2 13 weeks
Randomization 2:1
Pegvaliase 20 mg/day
Placebo 20 mg/day
Pegvaliase 40 mg/day
Placebo 40 mg/day
Double-blind, placebo-controlled, randomized discontinuation trial 8 weeks
Pegvaliase 20 mg/day
Pegvaliase 40 mg/day
Open-label to evaluate PK and PD 6 weeks
Pegvaliase 5–60 mg/day
Open-label extension to assess long-term outcomes

*Young adult/adolescents (subjects 16 and 17 years of age) were originally allowed under the PRISM-1 protocol, but inclusion criteria were revised in 2014 to exclude subjects <18 years old from participating to mitigate potential safety risks; previously enrolled young adult subjects were allowed to continue participating in the study.

Results

Subject exposure and disposition

- Exposure to pegvaliase and disposition for the 11 adolescent/young adult subjects in PRISM-2 were similar to the adult (≥18 years) population
 - The mean (SD) duration of exposure for the adolescent/young adult subjects was 885.5 (645.06) days, with a mean (SD) daily dose of 36.9 (12.70) mg/day. Most adolescent/young adult subjects were administered a mean dose of ≥40 mg/day to <60 mg/day (45.5% [n = 5]) or ≥20 mg/day to <40 mg/day (36.4% [n = 4]), with 18.2% (n = 2) administered a mean dose <20 mg/day and none were administered a mean dose ≥60 mg/day
 - 4 (36%) of the 11 adolescent/young adult subjects discontinued from study drug: 2 (18%) due to an AE, 1 (9%) withdrawn from study drug per investigator decision (due to subject non-compliance), and 1 (9%) lost to follow-up

Baseline demographics and characteristics

Table 1. Baseline demographics and characteristics by age for subjects entering PRISM-1

	Adolescent/Young Adult	Adults
Number of subjects	11	250
Age at consent Mean (SD), years	16.6 (0.5)	29.7 (8.5)
Sex Female, n (%)	7 (63.6)	123 (49.2)
Body mass index Mean (SD), kg/m²	24.7 (4.4)	28.6 (6.8)
Weight Mean (SD), kg	64.7 (8.8)	81.2 (20.8)
Blood Phe Mean (SD), µmol/L Median, µmol/L	1038.1 (280.5) 968.0	1241.3 (388.5) 1236.5
Protein from intact food Mean (SD), g/day	21.1 (15.3)	39.3 (27.9)

Efficacy

- Like adults, adolescent/young adults receiving long-term dosing of pegvaliase in PRISM-2 Part 4 were able to achieve clinically relevant blood Phe thresholds (<600 µmol/L [European guideline target for patients aged >12 years], <360 µmol/L ACMG target for all patients, <120 µmol/L [upper limit of normal]) **(Figure 2)** and had a substantial and sustained reduction in mean blood Phe over time, with a mean (SD) of 595.8 (539.07) µmol/L by Week 49 and 500.0 (625.01) µmol/L by Week 169, a reduction from baseline Phe values of 36.8% and 47.1%, respectively

Figure 2. Percentage of subjects who met blood Phe threshold (A) ≤600 µmol/L, (B) ≤360 µmol/L, and (C) ≤120 µmol/L over time by age group

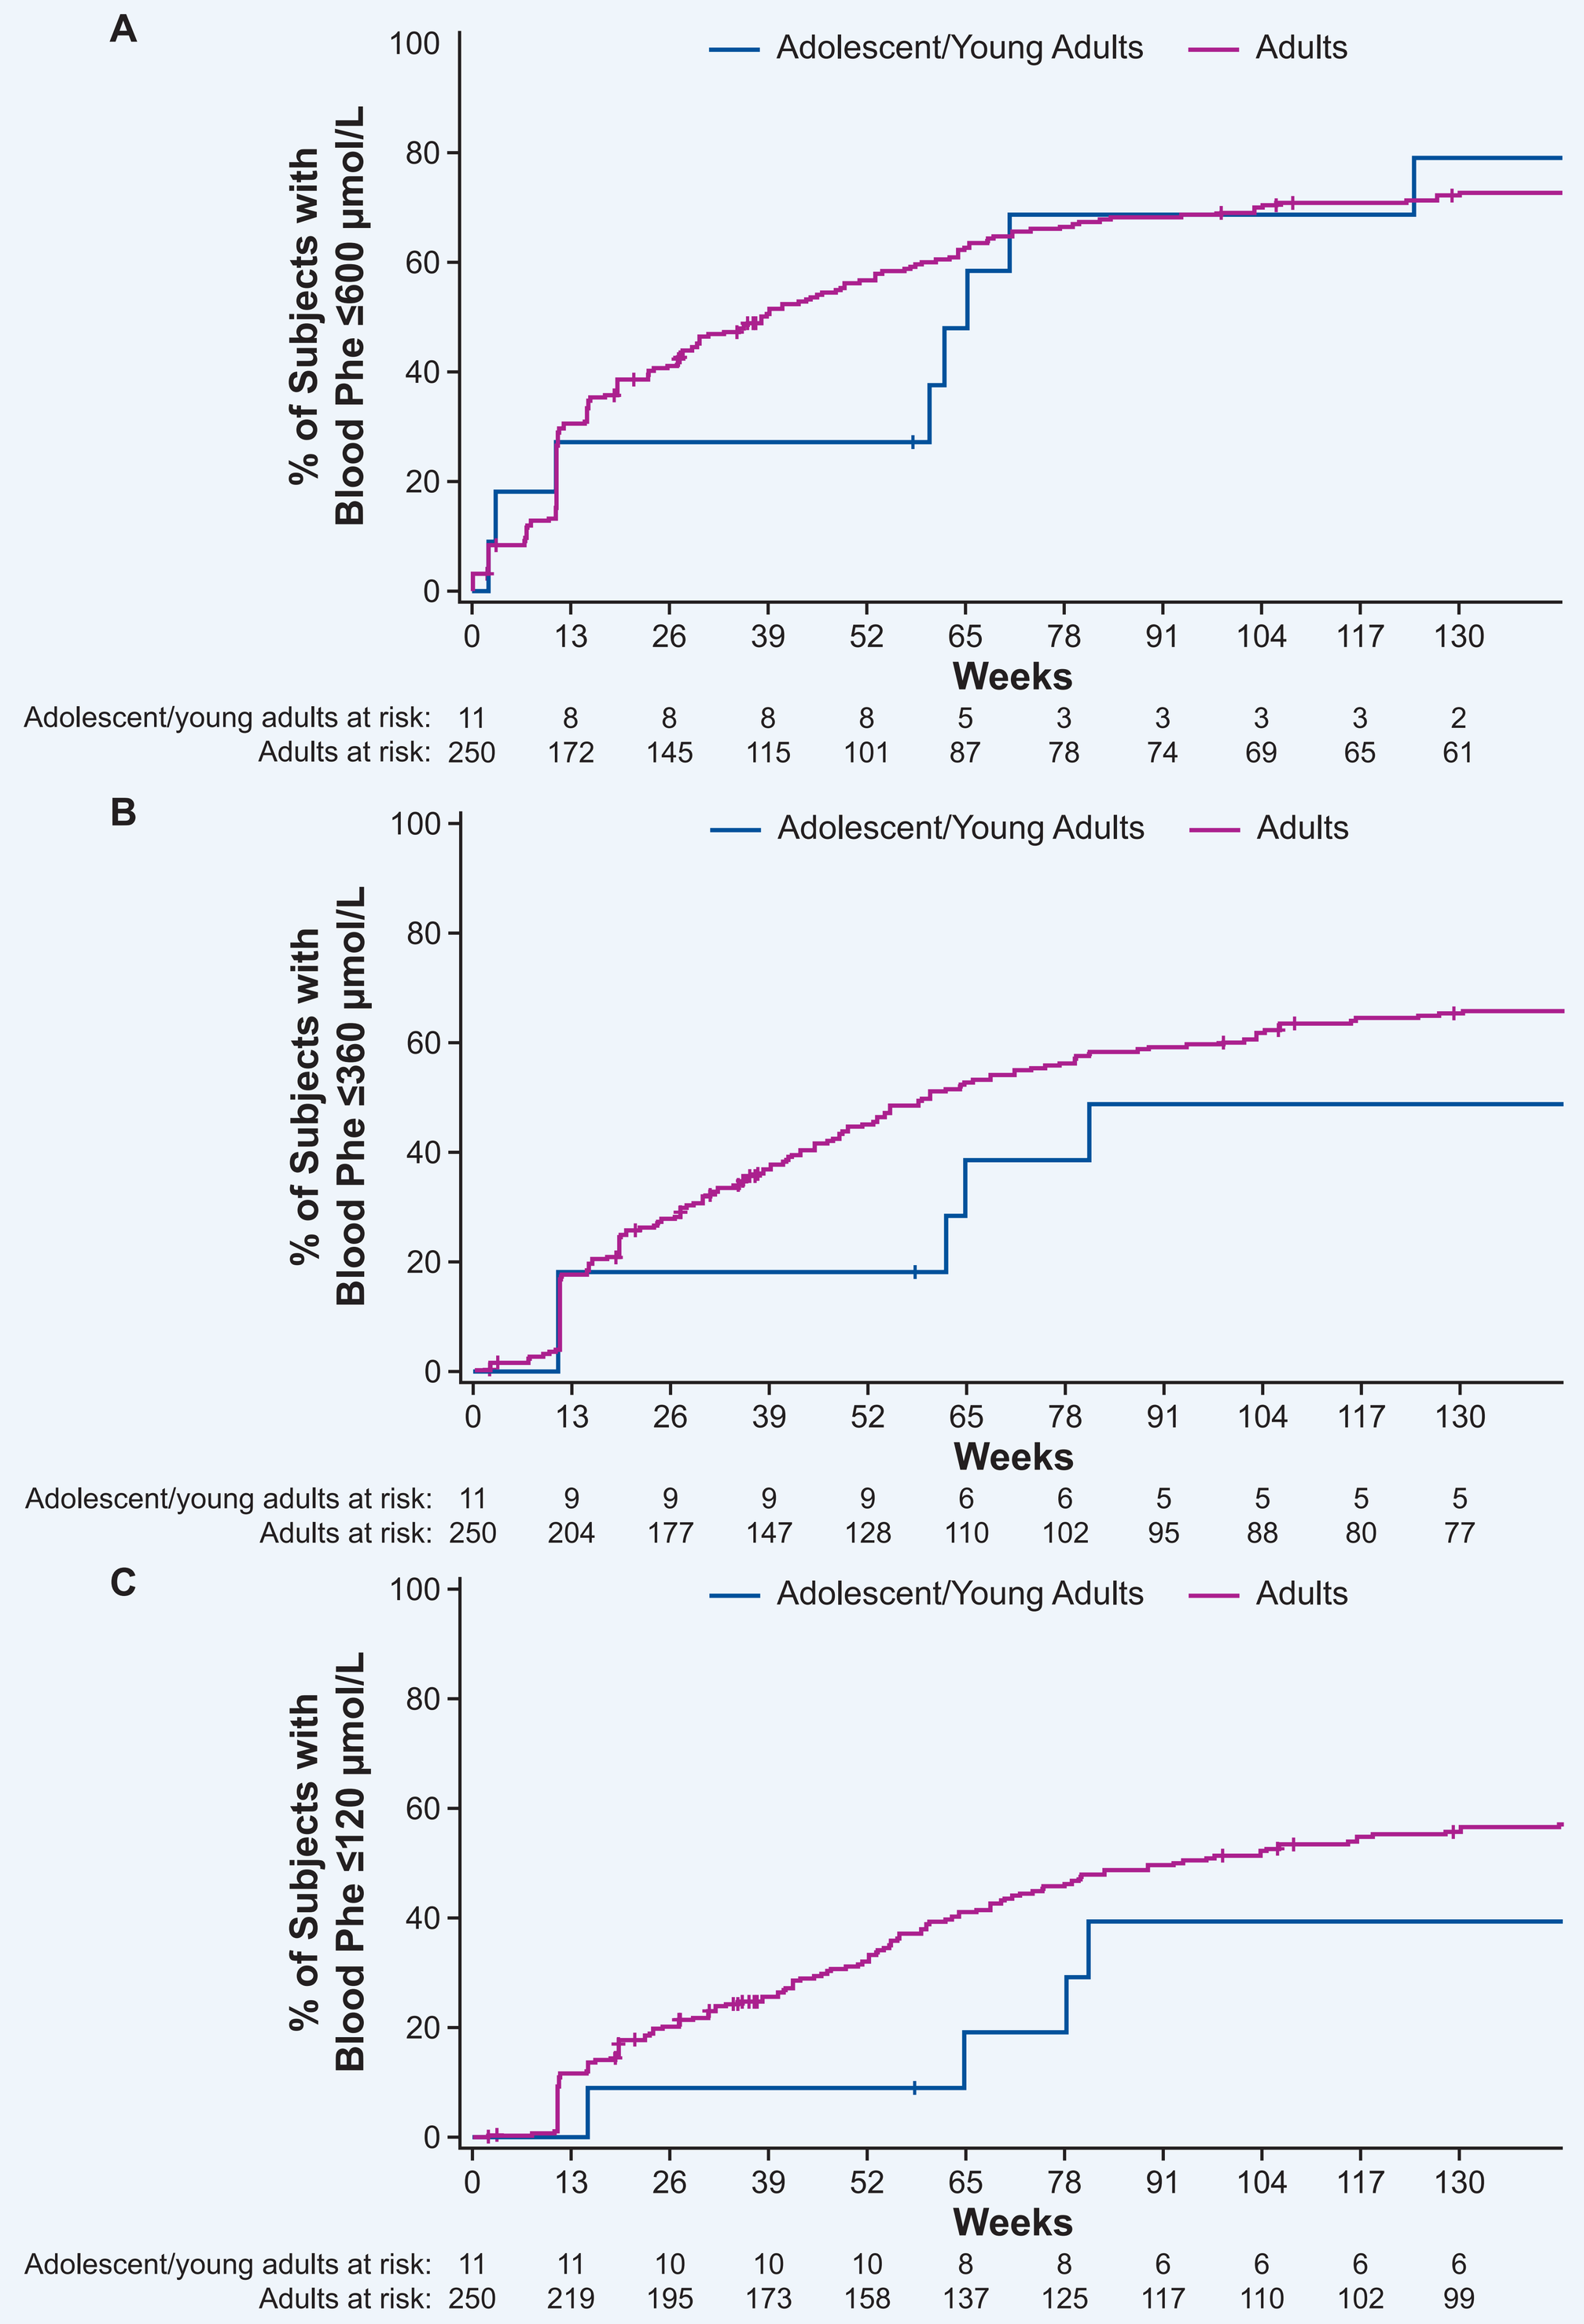
A
Adolescent/Young Adults
Adults
% of Subjects with Blood Phe ≤600 µmol/L
Weeks
0 13 26 39 52 65 78 91 104 117 130
Adolescent/young adults at risk: 11 8 8 8 8 5 3 3 3 3 2
Adults at risk: 250 172 145 115 101 87 78 74 69 65 61
B
Adolescent/Young Adults
Adults
% of Subjects with Blood Phe ≤360 µmol/L
Weeks
0 13 26 39 52 65 78 91 104 117 130
Adolescent/young adults at risk: 11 9 9 9 9 6 6 5 5 5 5
Adults at risk: 250 204 177 147 128 110 102 95 88 80 77
C
Adolescent/Young Adults
Adults
% of Subjects with Blood Phe ≤120 µmol/L
Weeks
0 13 26 39 52 65 78 91 104 117 130
Adolescent/young adults at risk: 11 11 10 10 10 8 8 6 6 6 6
Adults at risk: 250 219 195 173 158 137 125 117 110 102 99

Subjects at risk on x-axis indicates the number of subjects who have not been censored or reached specific efficacy thresholds at each time point. The time of censor for subjects who terminated early from the study or from study drug due to an adverse event, death, physician decision, or withdrawal by subject was the maximum study day of blood Phe assessment plus 1 day. For subjects who terminated early from the study or study drug due to other reasons, the time was censored to the last blood Phe assessment.

Safety/Immunogenicity

- AEs occurred at a similar rate in both age groups **(Table 2)**
 - None of the acute systemic hypersensitivity reactions were associated with drug-specific immunoglobulin E and all events resolved without sequelae
 - Neither of the 2 adolescent/young adult subjects who experienced an SAE discontinued from study drug or from the study due to the event
- Immunogenicity and PK/PD profile were consistent between the two age groups
 - PAL IgG antibodies found in 100% of subjects and neutralizing antibodies (NAbs) found in most subjects with mean detectable levels remaining stable or decreasing over time in both age groups

Table 2. Summary of adverse event rates by age group for subjects enrolled in PRISM-2

	Adolescent/Young Adults (n=11)	Adults (n=204)
Adverse event (AE)	**11 (100.0%)**	**200 (98.0%)**
Any AE assessed by the investigator as drug related	10 (90.9%)	192 (94.1%)
Any AE causing permanent study drug discontinuation	3 (27.3%)	9 (4.4%)
Serious adverse event (SAE)	**2 (18.2%)**	**38 (18.6%)**
SAE assessed by the investigator as drug related	1 (9.1%)	18 (8.8%)
SAE causing permanent study drug discontinuation	0	5 (2.5%)
SAE assessed by the investigator as drug related causing study discontinuation	0	3 (1.5%)
SAE causing study discontinuation	0	3 (1.5%)
Any hypersensitivity AE	**10 (90.9%)**	**175 (85.8%)**
Acute systemic hypersensitivity reaction	1 (9.1%)	7 (3.4%)
Severe acute systemic hypersensitivity reaction	0	0
Generalized skin reaction (≥14 Days)	0	90 (44.1%)
Injection site skin reaction (≥14 Days)	6 (54.5%)	84 (41.2%)
Arthralgia	7 (63.6%)	140 (68.6%)
Injection site reaction	10 (90.9%)	154 (75.5%)
Death	**0**	**0**

Overall summary of incidence of adverse events by age group (adolescent/young adults vs. adults) from subjects who entered PRISM-2 (N=215).

Conclusions

- Adolescent/young adult subjects aged 16 to 17 years achieved substantial and sustained blood Phe reductions with pegvaliase dosages up to 60 mg/day with a manageable safety profile for most subjects with long term treatment
- The efficacy, safety, and immunogenicity results in adolescent/young adults are consistent with those found in adults, demonstrating a positive benefit:risk profile and supporting inclusion of adolescent/young adult PKU patients aged 16 to 17 years for treatment with pegvaliase
- As adherence to dietary management begins to deteriorate during adolescence, pharmacotherapy should be considered to achieve optimum blood Phe control in this patient population

References

1. Vockley J et al. *Genet Med.* 2014;16:188–200. **2.** Bell SM et al. *PLoS One.* 2017;12:e0173269.
3. Longo N et al. *Lancet.* 2014;384:37–44. **4.** Sarkissian CN et al. *Proc Natl Acad Sci USA.* 1999;96:2339–44.
5. Palynziq [package insert]. Novato, CA: BioMarin Pharmaceutical Inc.; 2018. **6.** Palynziq (pegvaliase) [EU Product Information]. Shanbally, Ireland: BioMarin International Ltd.; 2019.
7. Huijbregts SC et al. *J Inherit Metab Dis.* 2002;25(6):419–30. **8.** Jahja R et al. *J Pediatr.* 2014;164(4):895–9.
9. Koch R et al. *J Inherit Metab Dis.* 2002;25(5):333–46.

Acknowledgements and Disclosures

The authors would like to thank the study investigators, study coordinators, study site support staff, and patients that participated in the study. This meeting was sponsored by BioMarin Pharmaceutical Inc. Medical writing support was provided by Kaleigh Bulloch Whitehall (BioMarin).

HN has received research grants and personal fees from BioMarin. RTZ has received consulting fees, honoraria, and travel support from BioMarin. MZS and JAT have participated in clinical studies and received consulting fees and research support from BioMarin. DL, ML, JO, and HHW are employees of BioMarin. JV declares no conflicts of interest.

Presented at the 2020 ACMG Annual Clinical Genetics Meeting

©2020 BioMarin Pharmaceutical Inc. All rights reserved.