ENDURE: A phase IV, open-label trial to assess the responsiveness of patients with phenylketonuria in Denmark to treatment with sapropterin over 28 days

Ahring K[1], Enggaard K[1], Rusz M[1], Strøm P[1], Jørgensen JV[2]

[1]The Kennedy Centre, PKU Department, Glostrup, Denmark; [2]Oslo University Hospital, Department of Newborn Screening, Oslo, Norway

Introduction

- Phenylketonuria (PKU) is an inborn error of metabolism resulting in a deficiency in the activity of phenylalanine hydroxylase (PAH), this causes phenylalanine (Phe) accumulation in the blood and brain. High Phe levels are associated with neuropsychological symptoms
- Both US and European guidelines recommend life-long control of blood Phe levels via a Phe-restricted diet (supplemented with medical foods) and/or pharmacotherapy; diagnosis is generally via newborn screening programs and treatment should be initiated as soon as possible after diagnosis[1,2]
- In tetrahydrobiopterin (BH_4)-responsive patients of all ages with PKU, dietary therapy can be complemented with sapropterin dihydrochloride (sapropterin; a synthetic version of BH_4; Kuvan®, BioMarin Pharmaceutical Inc., Novato, CA)
- In Europe, BH_4-responsiveness is generally assessed via a 24 to 48-hour BH_4 loading test; patients who respond after 48 hours may not be detected. However, there is evidence to suggest that longer duration response testing may be necessary to ensure that responsive patients are not missed[3]
- The ENDURE study assessed the dynamics of early and late (after 48 hours) response to sapropterin at several time points over 28±1 days in patients with PKU in Denmark and Norway (Danish cohort results are shown)

Methods

- In this phase IV, prospective, open-label, uncontrolled, single-arm, non-randomized, non-comparative study (NCT01082328), patients aged ≥4 years received sapropterin, 20 mg/kg/day for 28±1 days
- The study took place in two centers, one in Norway and one in Denmark, between May 2010 and May 2012
 - The trial protocol and all amendments were reviewed by the relevant Independent Ethics Committees and the study was performed in accordance with Good Clinical Practice, the guiding principles of the Declaration of Helsinki, and laws and regulations for clinical research in Norway and Denmark
- Patients aged ≥4 years with PKU and with documented genotype for both PAH gene mutations were eligible to participate if they had ≥2 historical blood Phe levels >400 µmol/L, had no previous treatment with sapropterin or other BH_4 formulations, were adherent to their usual diet and were willing to adhere to the given diet for the 4-week study period
 - In Denmark, patients with known or suspected responsiveness to BH_4 were included; patients with two mutations associated with no or little residual PAH enzyme activity were excluded
 - Other key exclusion criteria included documented BH_4 deficiency, pregnancy (current or planned), or breastfeeding
- All patients attended up to eight study visits (screening; baseline; interim study visits on Days 3±1, 7±1, 14±1, 21±1, 28±1; and a follow-up visit on Day 42±3)
 - Blood Phe and tyrosine levels were analyzed from fasting (≥4 h) blood samples collected at each visit
 - At the baseline visit (Day 0), blood Phe and tyrosine levels were assessed prior to sapropterin (baseline measurements) and at 8, 16, and 24 hours post-dose (patients remained at the study center up to and including the 24-h assessment)
 - Recording of adverse events (AEs) started once written informed consent was obtained at the screening visit
- The primary efficacy endpoint was the proportion of responders (defined as those achieving a ≥30% reduction from baseline in blood Phe at any point during the 28±1 days post initiation of sapropterin administration)
- Secondary endpoints included the proportion of non-responders, partial responders (10–<30% reduction), early responders (≤7±1 days), and late responders (≥30% after 7±1 days)
- Safety endpoints included the incidence and severity of AEs, abnormal laboratory values, serious AEs (SAEs) and differences in safety endpoints by age group

Results

Patient disposition, demographics and baseline characteristics

- All 19 patients screened in the Danish cohort (mean [range] age 10.6 [4.3–27.2] years) patients were included in the analyses **(Table 1)**

Table 1. Patient demographics and baseline characteristics (safety population)

	Age 4 to 16 years (n = 17)	Age >16 years (n = 2)	Total (N = 19)
Females	8 (43%)	1 (5%)	9 (47%)
Males	9 (47%)	1 (5%)	10 (53%)
Age, years			
Mean (SD)	10.3 (3.3)	21.9 (7.9)	10.6 (5.1)
Median (range)	11 (4–14)	21.9 (16–27)	(4–27)
Screening history			
Newborn screening completed	17 (89%)	2 (11%)	19 (100%)
Confirmatory testing completed	17 (89%)	1 (11%)	19 (100%)
Highest observed neonatal Phe concentration, µmol/L			
Mean (SD)	874 (492)	776.5 (235)	864 (467)
Median (range)	735 (340–2062)	776.5 (753–800)	742 (340–2062)

- Based on blood Phe measurements from the first 4 weeks of life, 5 patients were classified as having classic PKU, 10 as having mild PKU, and 4 with mild hyperphenylalaninemia (HPA)
- PAH genotype data were available for all patients enrolled in the study
 - The most frequently occurring PAH gene mutations were Y414C (11/19 patients), R408W (4/19 patients), and IVS-12 (6/19 patients; **Table 2**)

Table 2. Mutations occurring in 1 or more patients (n = 19)

Patients	Mutation 1	Mutation 2	Type	Phenotype
1	R408W	Y414C	mild	classic/mild
2	IVS12+1G>A	I65T	moderate	classic/moderate
3	IVS12+1G>A	T63P-H64N	mild	classic/mild
4	IVS12+1G>A	L48S	mild	classic/mild
5	G46S	Y414C	mild	mild/mild
6	G46S	Y414C	mild	mild/mild
7	F39L	Y414C	mild	moderate/mild
8	P281L	Y414C	mild	classic/mild
9	E221D222fsdelAG	Y414C	mild	classic/mild
10	L348V	Y414C	mild	moderate/mild
11	R408W	Y414C	mild	classic/mild
12	IVS1nt5g->t	A104D	mild	classic/mild
13	IVS12+1G>A	Y414C	mild	classic/mild
14	R408W	Y414C	mild	classic/mild
15	R408W	R241H	mild	classic/mild
16	R408W	E390G	MHP	classic/MHP
17	W120X	Y414C	mild	classic/mild
18	Y386C	R158Q	moderate	moderate/classic
19	IVS12+1G>A	Y414C	mild	classic/mild

Response to sapropterin treatment

- For the primary endpoint, 17 of the 19 patients (89%) responded to sapropterin over the course of 28±1 days and 2 were partial responders: the majority (16/17) were early responders and 1 patient was a late responder

Table 2. Mutations occurring in 1 or more patients (n = 19)

		Response status, n/N (%)*		
Age group	**Phenotype**	**Early response**	**Late response**	**Partial response**
4 to 16 years (n = 18)	Mild PKU Moderate PKU Mild HPA	14/18 (78%) 2/18 (11%) 1/18 (6%)	1/18 (6%) 0/18 (0%) 0/18 (0%)	0/18 (0%) 0/18 (0%) 0/18 (0%)
>16 years (n = 1)	Mild PKU	0/1 (0%)	0/1 (0%)	1/1 (100%)
N=19	All	17/19 (89%)	1/19 (5%)	1/19 (5%)

*Percentages may not total 100% due to rounding.

- Greater relative reductions in blood phenylalanine concentration over the treatment period were observed in patients with mild PKU and mild HPA than in patients with classic PKU **(Figure 1)**

Figure 1. Relative change from baseline in blood Phe, stratified by PKU phenotype (full analysis population)

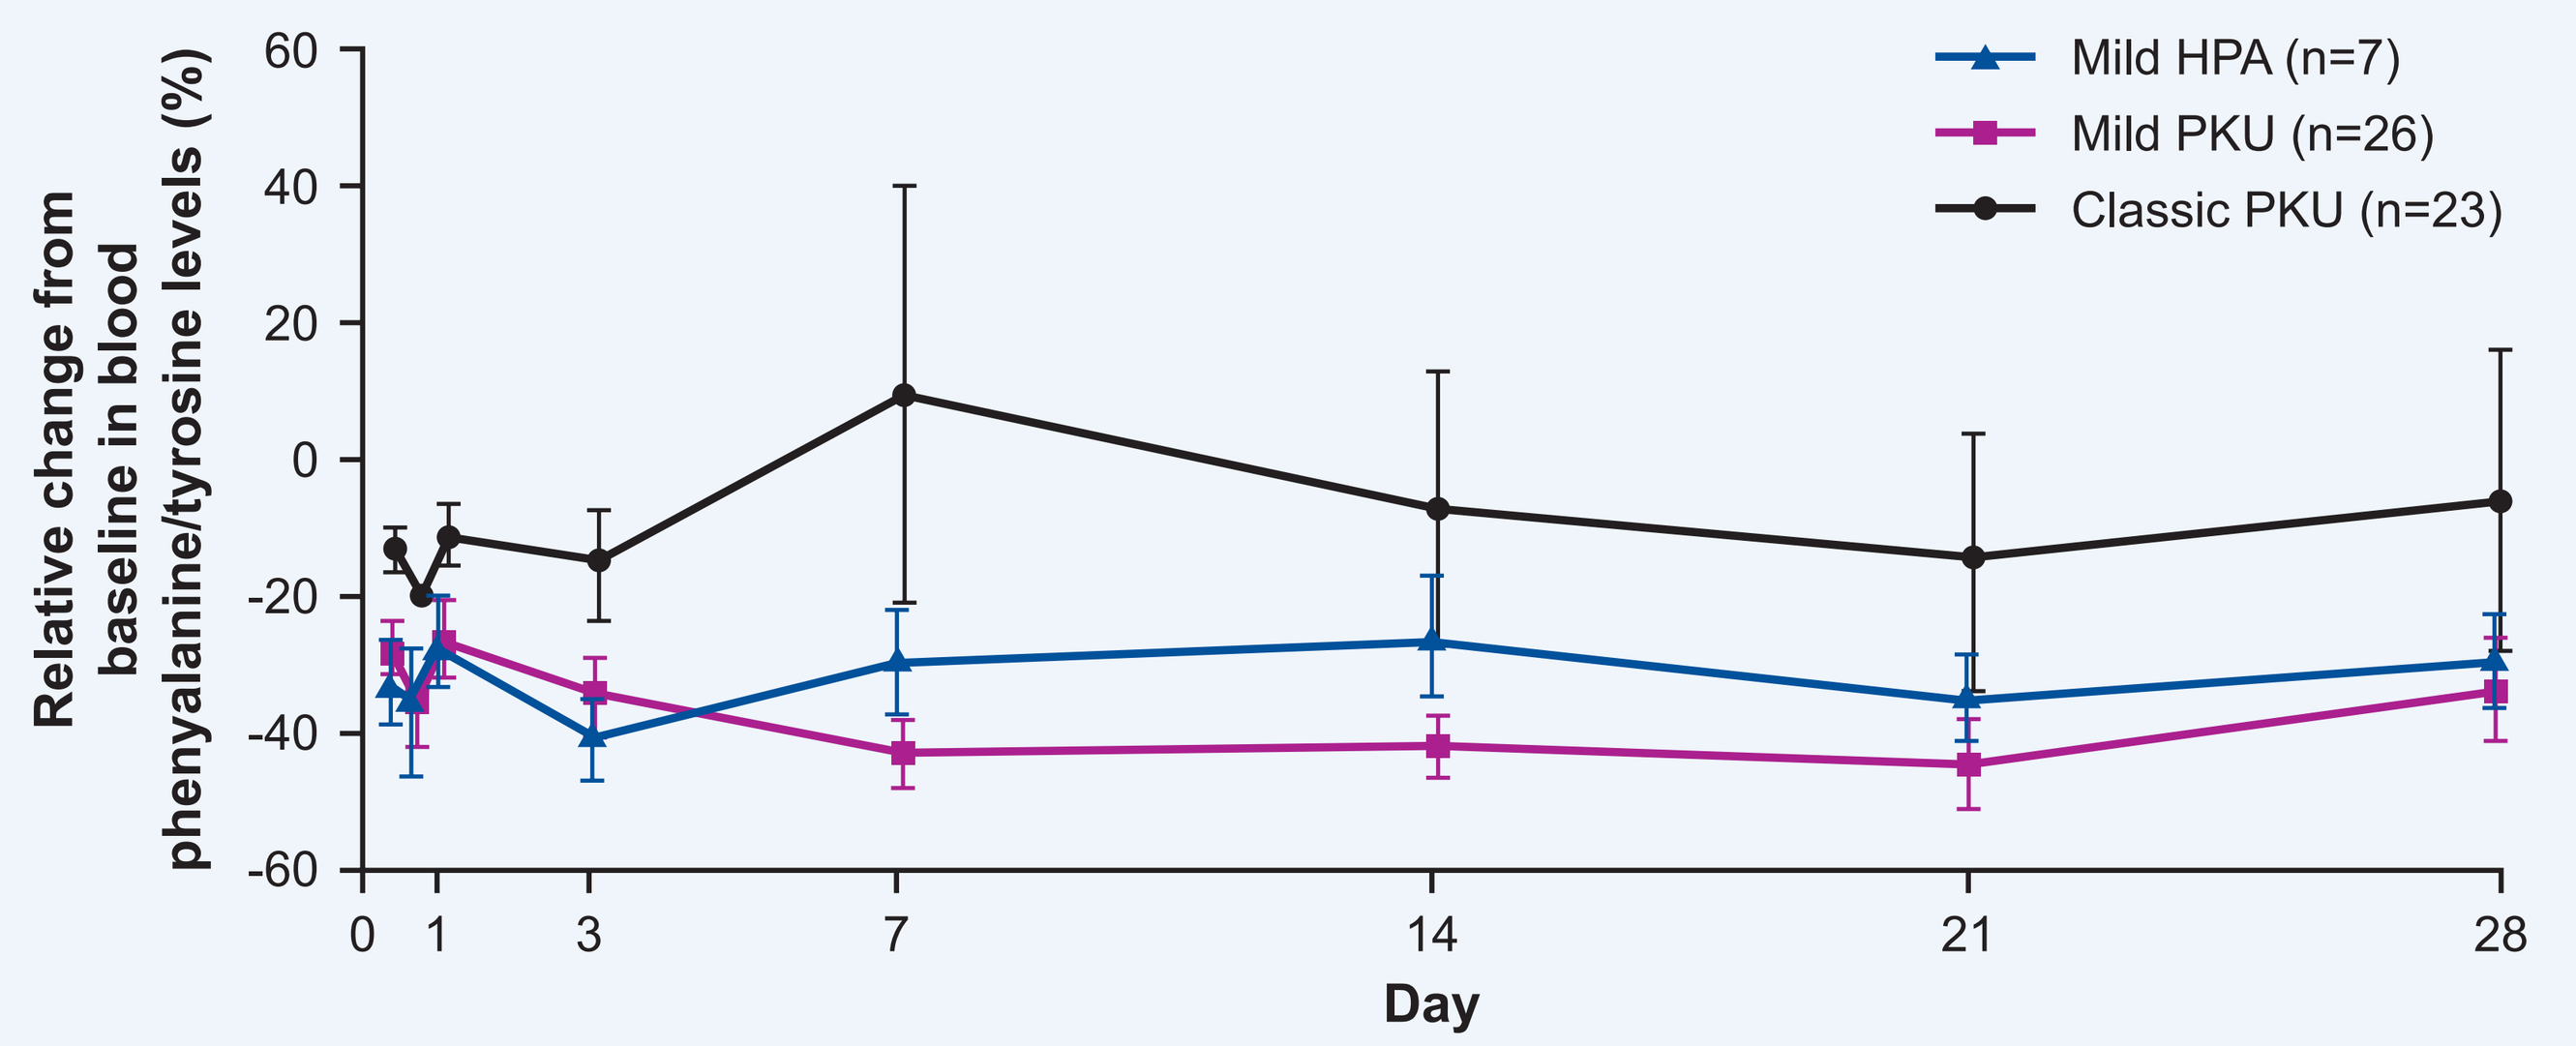
Mild HPA (n=7)
Mild PKU (n=26)
Classic PKU (n=23)
Relative change from baseline in blood phenyalanine/tyrosine levels (%)
60
40
20
0
-20
-40
-60
0
1
3
7
14
21
28
Day

Data for three patients with unknown PKU phenotype are not shown. Data are presented as mean (standard error)

- Treatment-related adverse events (all mild/moderate) were reported for 15 (79%) patients; no deaths or SAEs occurred during the study
- Guidance for the evaluation of responsiveness to sapropterin from an international group of healthcare professionals treating PKU patients was published in 2019 and recommended a 24-hour loading test for neonates, while test duration of ≥48 h or a 4-week trial was recommended for older infants, children, adolescents, and adults[3]

Conclusions

- Although most BH_4-responsive patients with PKU responded to sapropterin within 7 days, 6% of responsive patients responded after 7 days, highlighting the need for longer test periods
- The relevance of partial response requires further study

References

1. Van Wegberg AMJ et al. *Orphanet J Rare Dis.* 2017;12(1):162.
2. Vockley J et al. *Genet Med.* 2014;16(2):188-200.
3. Muntau AC et al. *Mol Genet Metab.* 2019;127(1):1-11.

Funding Disclosure

The study was supported by Merck, Darmstadt, Germany, and BioMarin Pharmaceutical Inc., Novato, CA, USA. Medical writing support was provided by Dr. Paul Lane (BioMarin). Poster design was provided by Terry Manspeaker (Show Your Science, LLC) and was funded by BioMarin.

Presented at the GMDI Virtual Conference: April 28–May 1, 2021

©2021 BioMarin Pharmaceutical Inc. All rights reserved.