Real-World Impact of Enzyme Replacement Therapy on Endurance in Patients with MPS IVA

Pascal Reisewitz[1], David Hinds[1], Sara Došenović[1], Yonghao Ma[1], Ashok Jha[1], Abigail Hunt[1], Barbara Burton[2], Nathalie Guffon[3]

[1] BioMarin Pharmaceutical Inc., Novato, CA, USA; [2] Ann & Robert H. Lurie Children's Hospital of Chicago, Chicago, IL, USA; [3] Reference Centre of Inherited Metabolic Disease, HCL Hospital, Lyon, France

Introduction

- Initiation of long-term enzyme replacement therapy (ERT) is recommended for all patients with mucopolysaccharidosis (MPS) IVA as soon as possible after a confirmed diagnosis [1]
- Longitudinal comparisons of the real-world impact of ERT in the MPS IVA population have been challenging to describe due to limited natural history data
- We conducted a cross-sectional analysis comparing 6-minute walk test distance (6MWT; a measure of endurance) in ERT-treated and untreated patients at different ages using data from the Morquio A Registry Study (MARS) and the Morquio A Clinical Assessment Program (MorCAP) natural history study
 - MARS is an ongoing multinational, observational study of patients with MPS IVA: patients with a confirmed diagnosis of MPS IVA are eligible to participate and data are collected as part of routine care; the study includes both ERT-treated and untreated patients [2]
 - The MorCAP natural history study was a multinational, observational study of patients with MPS IVA: patients were assessed at study entry and over time thereafter [3,4]

Methods

- Patients with a 6MWT measurement (in either data set) at specific ages of interest were identified: parallel analyses were conducted for patients with a 6MWT measurement at age 10 (±1 year) and those with a 6MWT measurement at age 15 (±1 year)
- Within each age group, patients were stratified on the basis of ERT exposure prior to the 6MWT measurement of interest: ERT-treated patients were those who had received ERT treatment for at least 180 days prior to the 6MWT measurement of interest; untreated patients were those with no prior ERT exposure
- Characteristics of ERT-treated and untreated patients were assessed and regression analyses were performed to assess associations between patient characteristics (including ERT exposure) and 6MWT distance
 - Variables found to be associated with 6MWT distance in univariate analyses ($P<0.1$) were subsequently included in multivariate regression analyses

Figure 1: Analytic approach

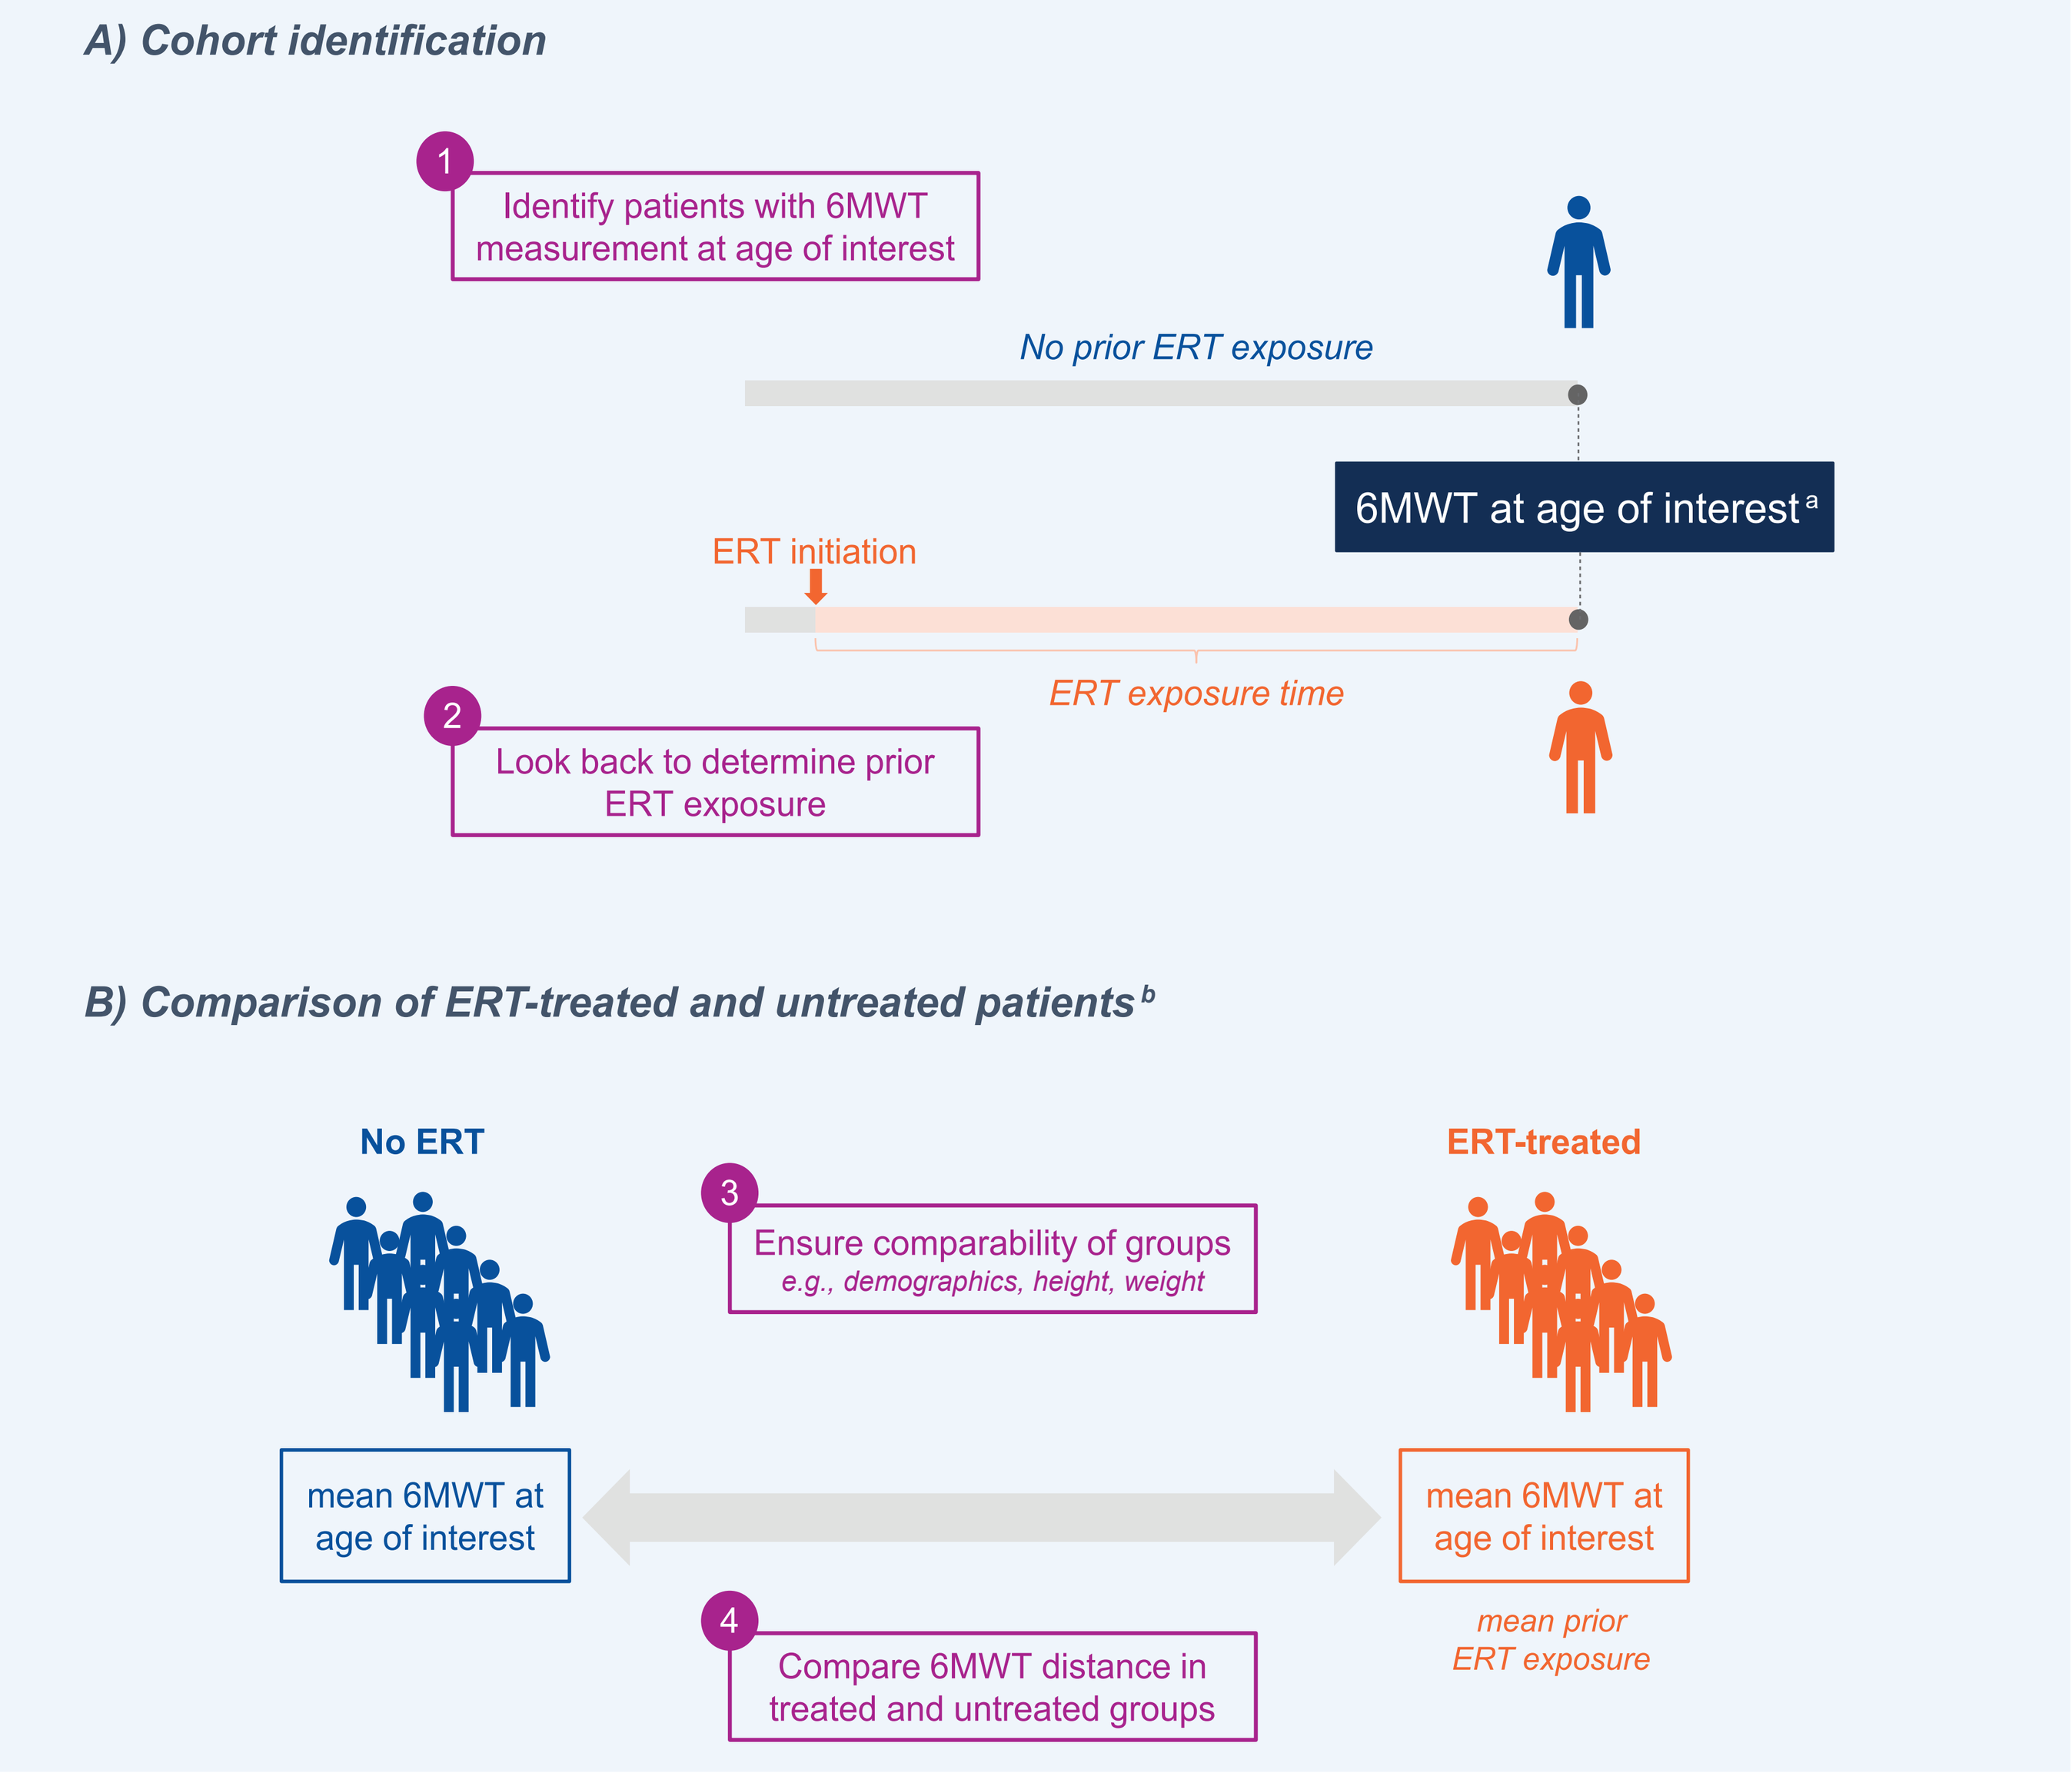
A) Cohort identification
1
Identify patients with 6MWT measurement at age of interest
No prior ERT exposure
6MWT at age of interest[a]
ERT initiation
ERT exposure time
2
Look back to determine prior ERT exposure
B) Comparison of ERT-treated and untreated patients[b]
No ERT
ERT-treated
3
Ensure comparability of groups
e.g., demographics, height, weight
mean 6MWT at age of interest
mean 6MWT at age of interest
4
Compare 6MWT distance in treated and untreated groups
mean prior ERT exposure

[a] Assessment closest to the age of interest was utilized for analysis
[b] Groups were mutually exclusive: individual subjects identified in both datasets were included in the no ERT exposure group only

Results

6MWT at age 10 years

- Among subjects with 6MWT assessment at age 10, mean (range) ERT exposure time for ERT-treated patients was 4.5 (0.5-9.5) years
- Sex, age at assessment, mean standing height, and weight were similar for ERT-treated and untreated groups (**Table 1**)
- At 10 years of age, mean (SD) 6MWT distance was 308.0 (153.8) m for ERT-treated and 242.6 (116.8) m for untreated patients; median (IQR) 6MWT distance was 330.5 (190.0, 436.0) m for ERT-treated and 258.3 (173.6, 332.0) m for untreated patients (**Figure 2**)

Table 1: Characteristics of patients with 6MWT measurement at age 10 years

	No ERT N=69	ERT N=58
Age at 6MWT assessment, y		
mean (SD)	9.6 (0.5)	9.7 (0.5)
Sex, n (%)		
Female	32 (46.4)	20 (34.5)
Male	37 (53.6)	38 (65.5)
Race, n (%)		
Asian	8 (11.6)	15 (25.9)
Black	1 (1.4)	2 (3.4)
White	51 (73.9)	29 (50.0)
Other	7 (10.1)	5 (8.6)
Unknown	2 (2.9)	7 (12.1)
Height, cm[a]	*n=61*	*n=52*
mean (SD)	105.7 (12.8)	107.6 (11.8)
median (range)	100.9 (84.1, 146.5)	104.3 (87.2, 137.5)
Weight, kg[a]	*n=66*	*n=55*
mean (SD)	22.3 (6.5)	21.9 (6.0)
median (range)	21.0 (14.4, 47.3)	20.0 (14.1, 42.8)
FEV_1, L[b]	*n=35*	*n=33*
mean (SD)	0.95 (0.45)	1.04 (0.48)
FVC, L[b]	*n=35*	*n=33*
mean (SD)	1.11 (0.58)	1.18 (0.52)

[a]within ±365 days of 6MWT assessment of interest
[b]within ±180 days of 6MWT assessment of interest
Additional variables assessed included region. walking aid use during 365 days prior to 6MWT assessment, and incidence of surgeries within 180 days prior to 30 days after 6MWT assessment
FEV_1, forced expiratory volume in 1 second; FVC, forced vital capacity; SD, standard deviation

Figure 2: 6MWT distance at age 10 years

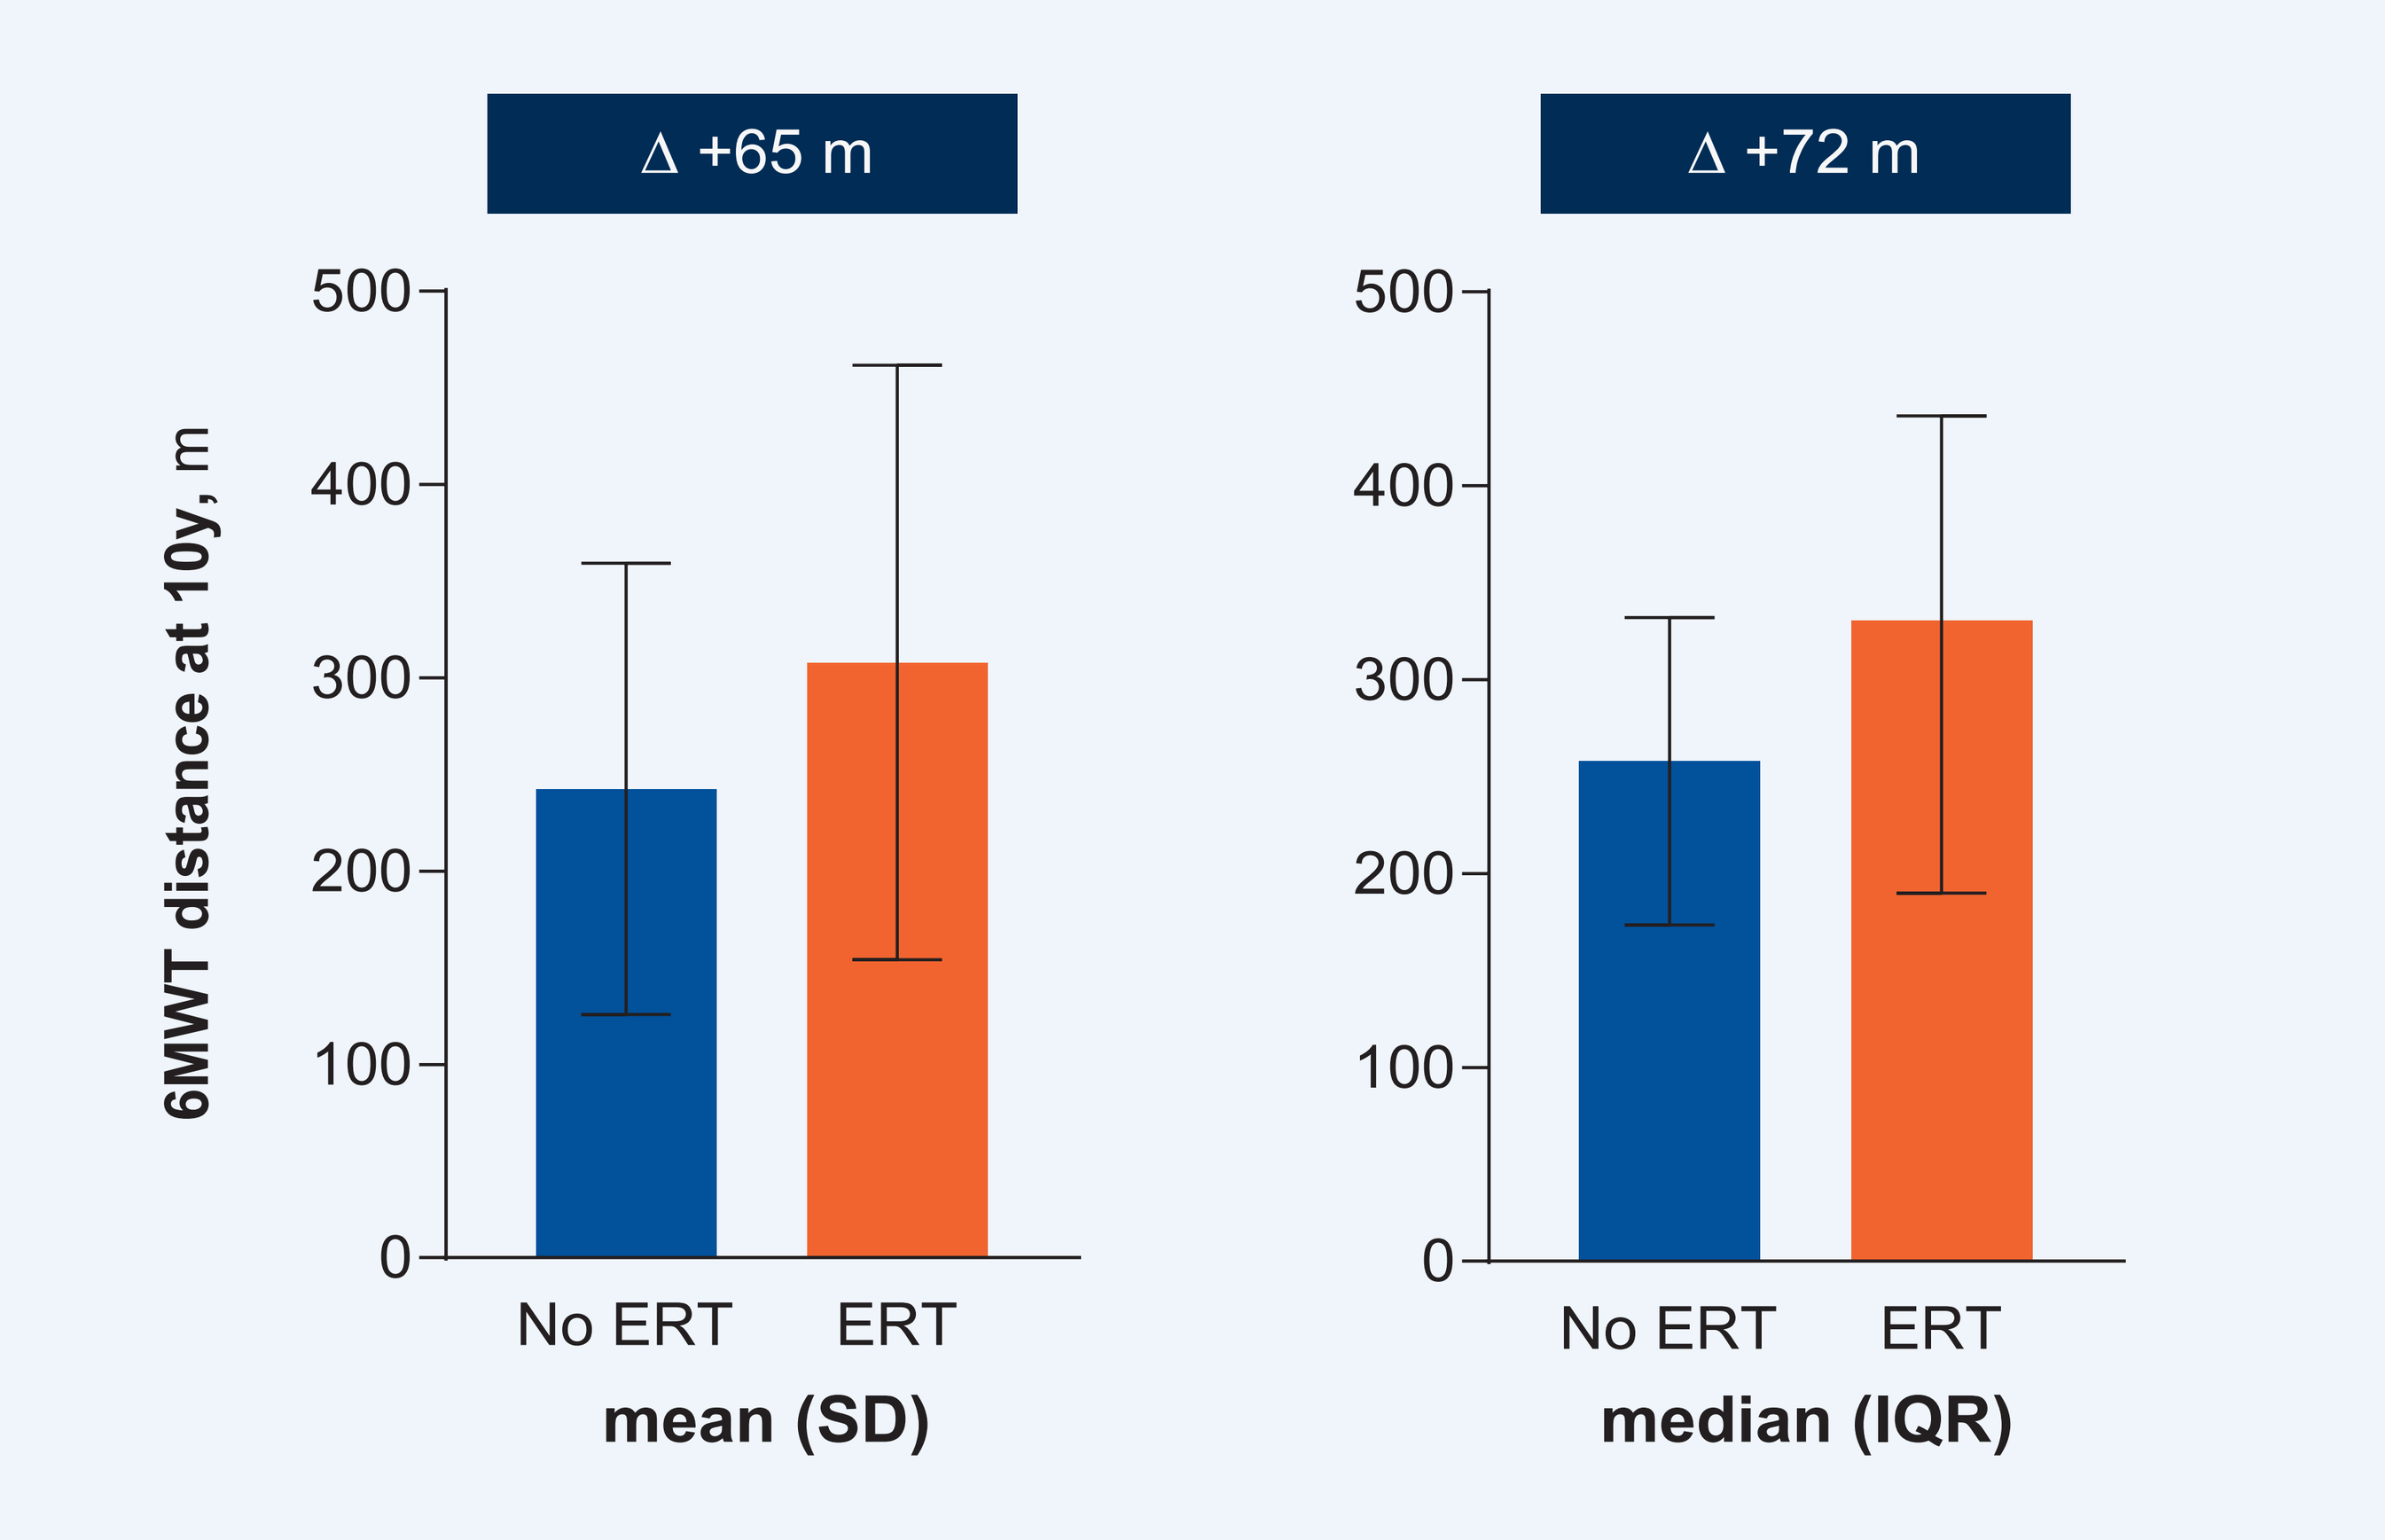
Δ +65 m
Δ +72 m
6MWT distance at 10y, m
500
400
300
200
100
0
No ERT
ERT
mean (SD)
median (IQR)

IQR, interquartile range; SD, standard deviation

6MWT at age 15 years

- Among subjects with 6MWT assessment at age 15, mean (range) ERT exposure time for ERT-treated patients was 5.3 (0.5,10.4) years
- Sex, age at assessment, mean standing height, and weight were similar for ERT-treated and untreated groups (**Table 2**)
- At 15 years of age, mean (SD) 6MWT distance was 244.5 (200.8) m for ERT-treated and 167.2 (168.7) m for untreated patients; median (IQR) 6MWT distance was 246.0 (61.0, 341.0) m for ERT-treated and 105.0 (0, 305.5) m for untreated patients (**Figure 3**)

Table 2: Characteristics of patients with 6MWT measurement at age 15 years

	No ERT N=65	ERT N=37
Age at 6MWT assessment, y		
mean (SD)	14.6 (0.5)	14.6 (0.5)
Sex, n (%)		
Female	33 (50.8)	18 (48.6)
Male	32 (49.2)	19 (51.4)
Race, n (%)		
Asian	7 (10.8)	10 (27.0)
Black	3 (4.6)	1 (2.7)
White	41 (63.1)	17 (45.9)
Other	12 (18.5)	6 (16.2)
Unknown	2 (3.1)	3 (8.1)
Height, cm[a]	*n=48*	*n=31*
mean (SD)	117.5 (21.2)	116.0 (23.6)
median (range)	113.3 (89.2, 166.8)	103.5 (87.0, 165.0)
Weight, kg[a]	*n=63*	*n=35*
mean (SD)	30.4 (12.2)	33.4 (14.6)
median (range)	28.0 (13.3, 67.6)	26.9 (17.3, 69.4)
FEV_1, L[b]	*n=32*	*n=23*
mean (SD)	1.26 (0.92)	1.36 (1.12)
FVC, L[b]	*n=32*	*n=23*
mean (SD)	1.48 (1.09)	1.60 (1.24)

[a]within ±365 days of 6MWT assessment of interest
[b]within ±180 days of 6MWT assessment of interest
Additional variables assessed included region. walking aid use during 365 days prior to 6MWT assessment, and incidence of surgeries within 180 days prior to 30 days after 6MWT assessment
FEV_1, forced expiratory volume in 1 second; FVC, forced vital capacity; SD, standard deviation

Figure 3: 6MWT distance at age 15 years

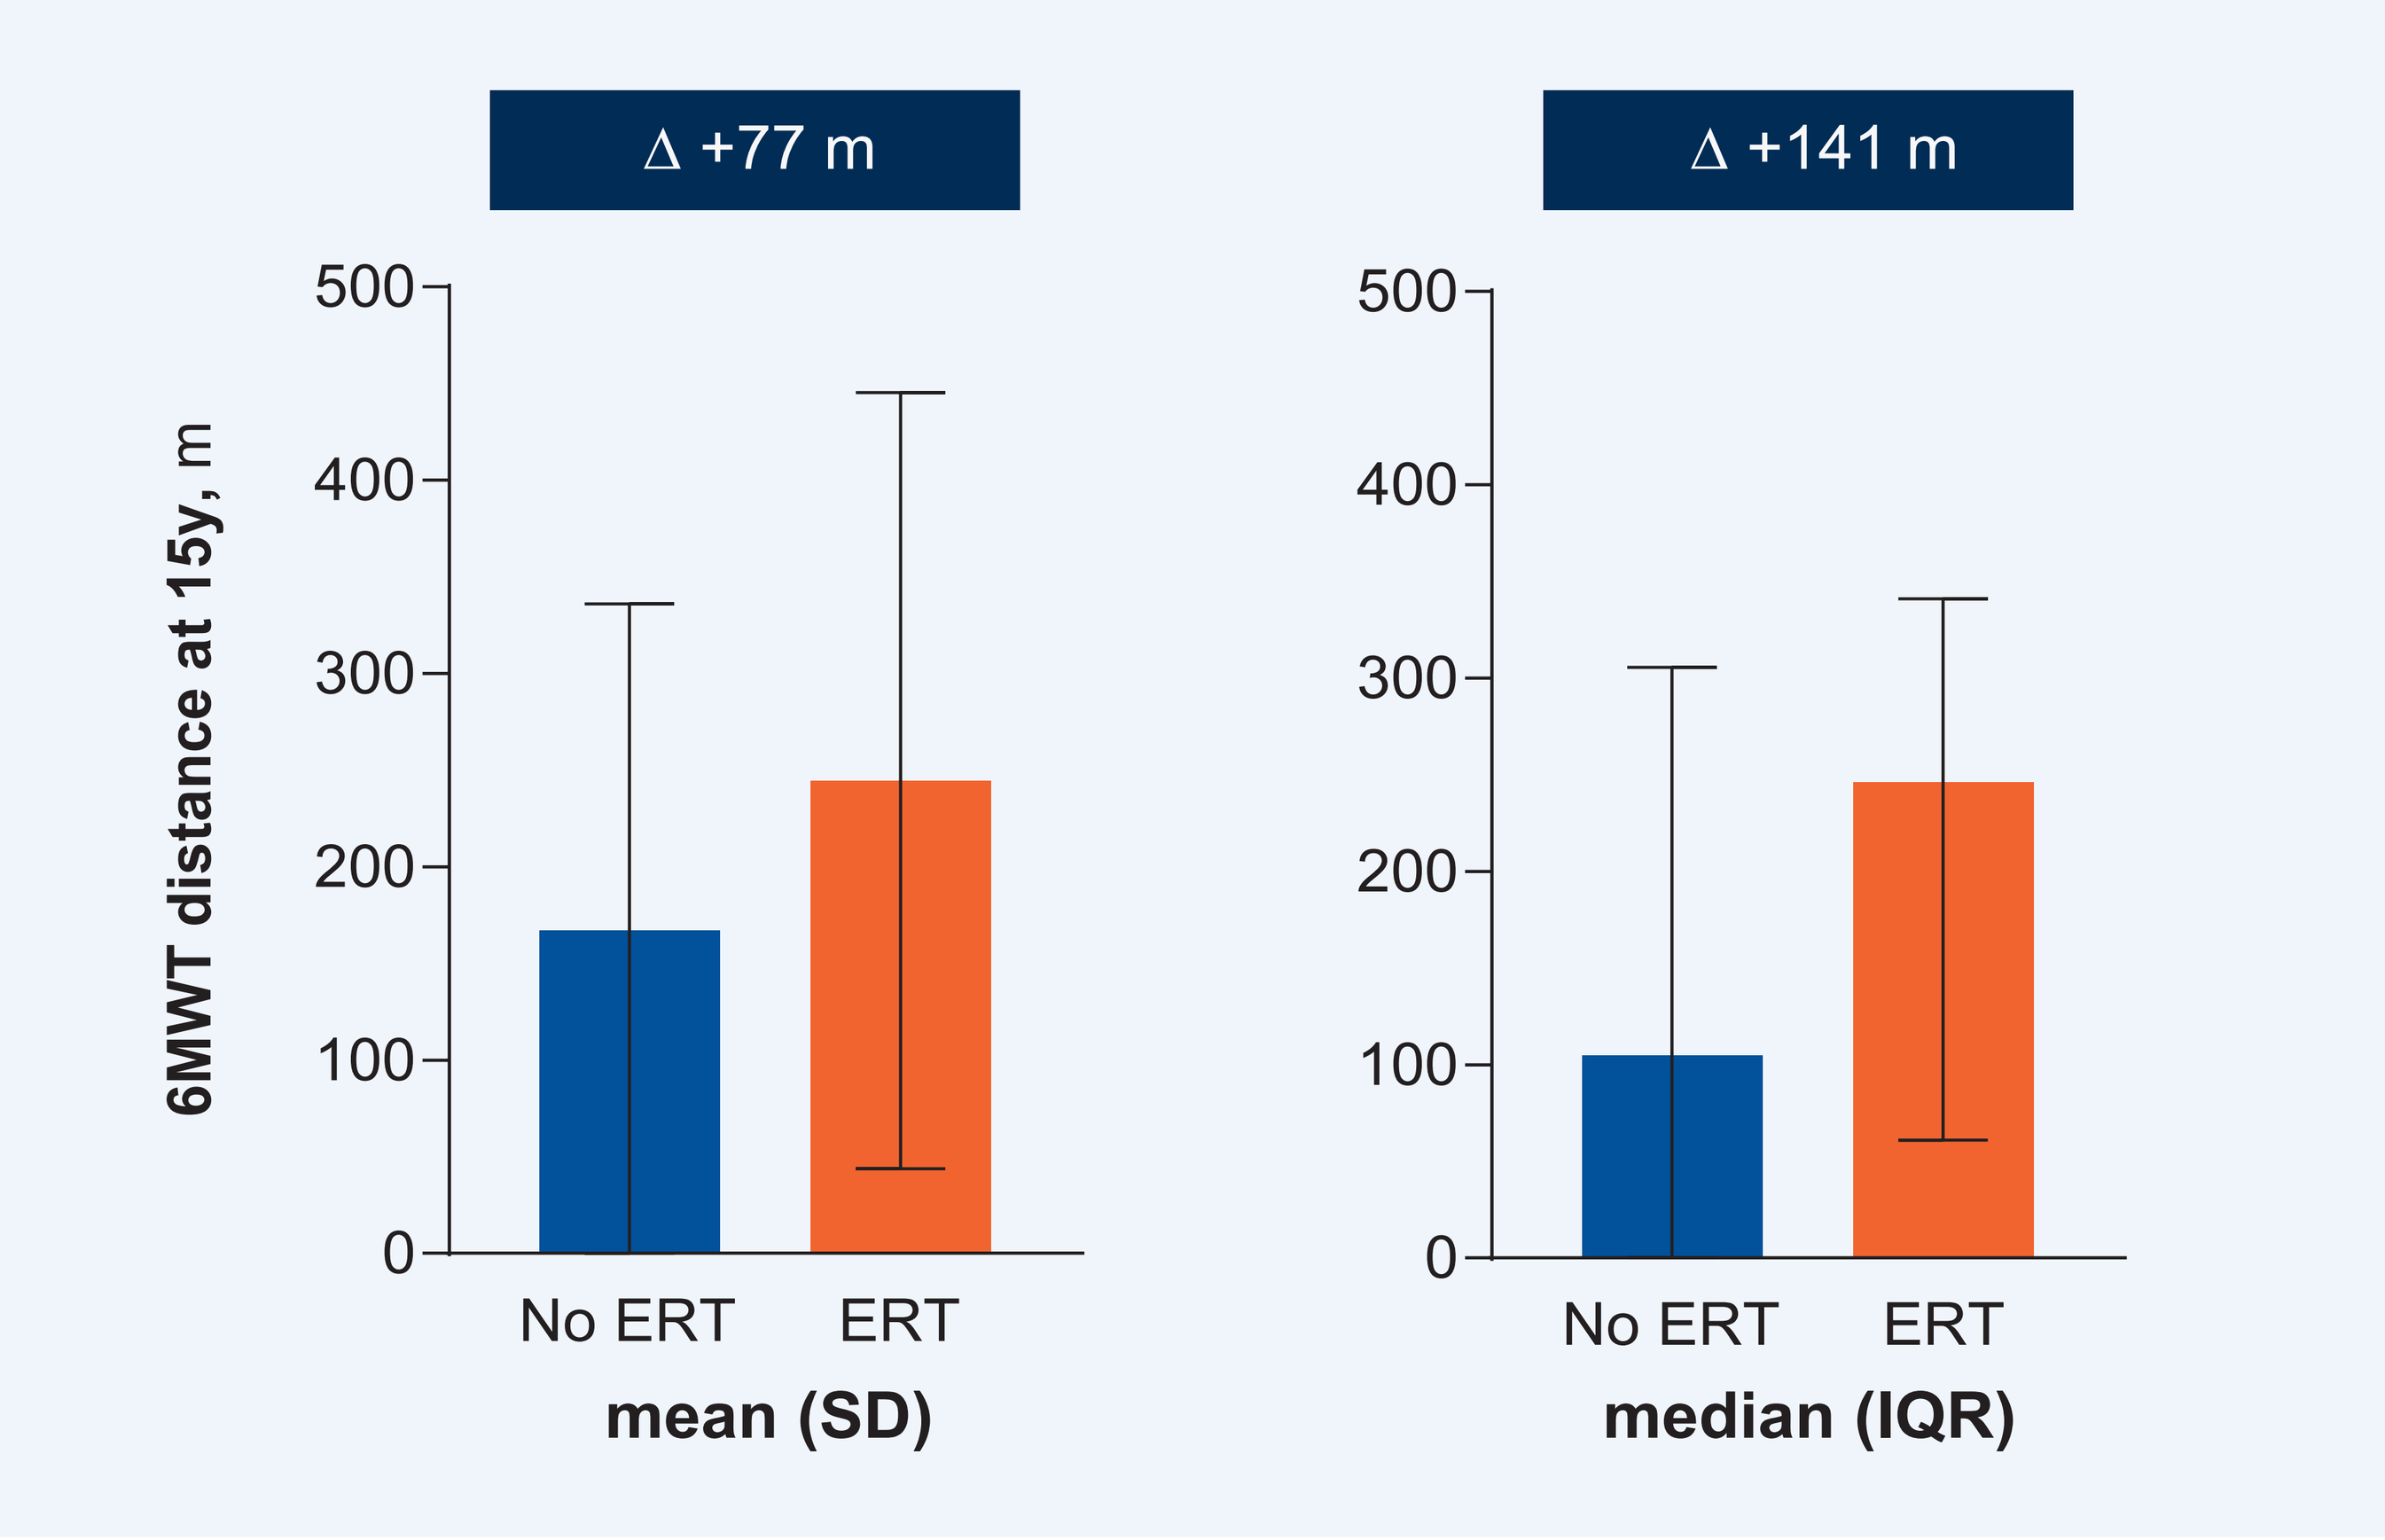
Δ +77 m
Δ +141 m
6MWT distance at 15y, m
500
400
300
200
100
0
No ERT
ERT
mean (SD)
median (IQR)

IQR, interquartile range; SD, standard deviation

Regression Analyses

- In univariate linear regression analyses, ERT exposure, standing height, and weight (age 15 only) were found to be significantly associated ($P<0.1$) with 6MWT distance and were subsequently included in multivariate analyses
- ERT exposure (age 10 only, $P=0.02$) and standing height (both age groups: age 10, $P=0.0008$; age 15, $P=0.003$) remained statistically significantly associated with 6MWT distance in multivariate analyses (**Table 3**)

Table 3: Associations with 6MWT distance, multivariate linear regression

	Age 10		Age 15	
	Estimate (95% CI)	*P*-value	Estimate (95% CI)	*P*-value
ERT treatment	55.6 (8.6, 102.7)	0.02	50.7 (-16.2, 117.6)	0.14
Standing height, per cm	3.3 (1.4, 5.2)	0.0008	4.4 (1.5, 7.2)	0.003
Weight, per kg			1.5 (-3.1, 6.2)	0.51

Variables determined to be significantly associated with 6MWT distance in univariate analyses ($p<0.1$) were included in multivariate analyses
Base case variables assessed in univariate analyses included: ERT treatment, sex, race, region, standing height, weight
CI, confidence interval

- Quantile regression analyses yielded similar results:
 - In univariate quantile regression analyses, ERT exposure, standing height, and weight (age 15 only) were associated ($P < 0.1$) with 6MWT distance
 - Subsequent multivariate analyses also followed a similar pattern (age 10: ERT exposure [$P=0.05$], standing height [$P=0.007$]; age 15: ERT exposure [$P=0.06$], standing height [$P=0.0005$], weight [$P=0.83$])

Conclusions

- Patients treated with ERT demonstrated greater endurance (as determined by 6MWT distance) than age-matched untreated patients at both 10 years and 15 years of age
- This analytic approach enabled assessment of the associations between ERT exposure and endurance, despite limited availability of longitudinal natural history data
- These results demonstrate a consistent impact of ERT treatment on endurance in a real-world MPS IVA population
- Further analyses to explore associations between ERT exposure and endurance in additional age groups are planned

References

1. Akyol MU, Alden TD, Amartino H, et al. *Orphanet J Rare Dis* 2019;14:137.
2. Mitchell JJ, Burton BK, Bober MB, et al. *Mol Genet Metab* 2022;137:164-72.
3. Harmatz P, Mengel KE, Giugliani R, et al. *Mol Genet Metab* 2013;109:54-61.
4. Harmatz PR, Mengel KE, Giugliani R, et al. *Mol Genet Metab* 2015;114:186-94.

Acknowledgements

The MARS and MorCAP studies and the development of this poster were supported by BioMarin Pharmaceutical, Inc. BB and NG received payments from BioMarin for attending advisory boards related to the MARS study. Medical writing support for the development of this poster was provided by Ismar Healthcare, Lier, Belgium.

Presented at the 19th Annual WORLD Symposium: February 22–26, 2023; Orlando, FL, USA

©2023 BioMarin Pharmaceutical Inc. All rights reserved.