PO124

Seven-year follow-up of valoctocogene roxaparvovec gene therapy for hemophilia A

Symington E[1], Rangarajan S[2], Lester W[3], Madan B[4], Pierce GF[5], Raheja P[6], Millar C[7], Osmond D[8], Li M[8], Robinson TM[8]

[1]Cambridge University Hospitals NHS Foundation Trust, Cambridge, UK; [2]Faculty of Medicine, University of Southampton, Southampton, UK; [3]University Hospitals Birmingham NHS Foundation Trust, Birmingham, UK; [4]Guy's and St Thomas' NHS Foundation Trust, London, UK; [5]Independent Consultant, La Jolla, CA, USA; [6]Haemophilia Centre Royal London Hospital, Barts Health NHS Trust, London, UK; [7]Imperial College Healthcare NHS Trust and Centre for Haematology, Department of Immunology and Inflammation, Imperial College London, London, UK; [8]BioMarin Pharmaceutical Inc., Novato, CA, USA

Introduction

- Valoctocogene roxaparvovec (AAV5-hFVIII-SQ) is an adeno-associated virus serotype 5 (AAV5)-mediated gene therapy that transfers a factor VIII (FVIII) coding sequence to increase endogenous FVIII expression in individuals with severe hemophilia A (FVIII activity level, ≤1 international unit [IU]/dL)[1-5]
- Here, we present the updated safety and efficacy results of valoctocogene roxaparvovec up to 7 years after dosing in a phase 1/2 clinical study (NCT02576795)

Figure 1. Valoctocogene roxaparvovec gene transfer of FVIII coding sequence

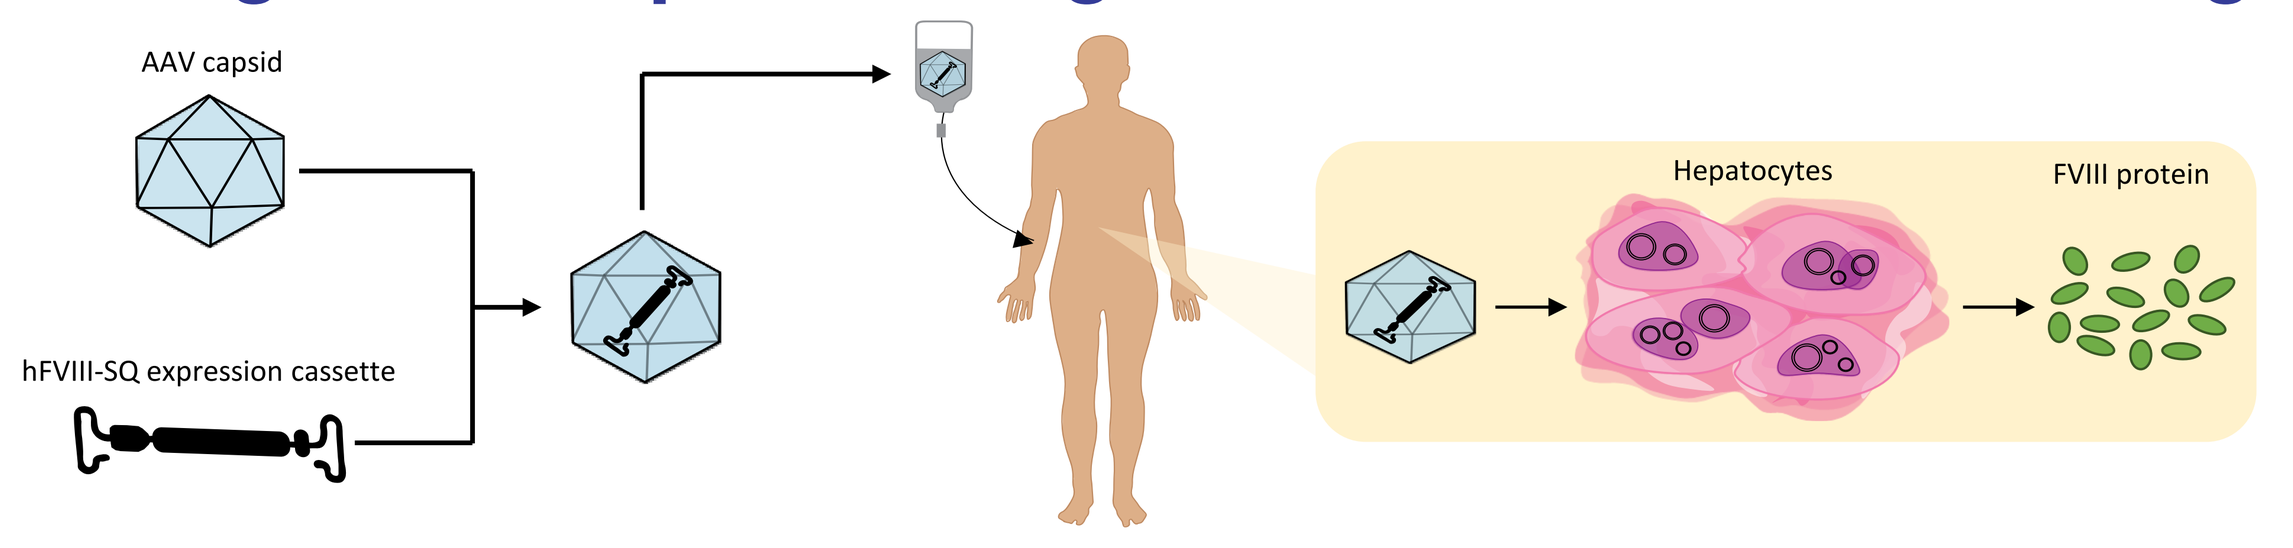
AAV capsid
hFVIII-SQ expression cassette
Hepatocytes
FVIII protein

Methods

Assessments

- Safety was evaluated with laboratory assessments and adverse events (AEs)
- Annualized treated bleeding rates (ABRs) and FVIII infusion rates were calculated as described previously[1,2]
- FVIII activity was assessed via chromogenic substrate assay (CSA) and the one-stage assay (OSA)[1-4]
- Liver ultrasounds were performed at the time of screening, at year-end visits starting at year 5, and at the discretion of the physician[4]

Statistics

- Data were summarized with descriptive statistics; missing data were not imputed
- Yearly rate of change in FVIII activity was determined using a linear regression model (FVIII activity = intercept + [slope × week], with random intercept and slope)

Results

- As previously reported, males aged ≥18 years with severe hemophilia A (FVIII ≤1 IU/dL) who were previously receiving exogenous FVIII received valoctocogene roxaparvovec treatment and were followed for 7 (6x10^{13} vg/kg; n = 7) and 6 (4x10^{13} vg/kg; n = 6) years[1-4]

Figure 2. Dosing schema

15 participants enrolled and dosed in 4 cohorts

- **1 participant in the 6×10^{12} vg/kg dose cohort**
- **1 participant in the 2×10^{13} vg/kg dose cohort**
- **7 participants in the 6×10^{13} vg/kg dose cohort**
- **6 participants in the 4×10^{13} vg/kg dose cohort**

Prior to infusion, all but 1 participant in the 6x10^{13} dose cohort were receiving FVIII prophylaxis; the participant who was not on prophylaxis was using FVIII on-demand.
FVIII, factor VIII; vg, vector genomes.

Table 1. Baseline demographics and rates of bleeding and FVIII infusion

Baseline characteristics	6x10^{13} vg/kg cohort (n = 7)	4x10^{13} vg/kg cohort (n = 6)
Age, y		
Mean (SD)	30.4 (5.8)	31.3 (9.6)
Median	30.0	30.5
Min, max	23.0, 42.0	22.0, 45.0
Race, n (%)		
Asian	1 (14.3)	0
Black	0	1 (16.7)
White	6 (85.7)	5 (83.3)
Baseline annualised number of FVIII infusions, infusions/y		
Mean (SD)	120.1 (45.9)	142.8 (48.8)
Median	121.4	155.8
Min, max	27.4, 158.5	53.8, 184.3
Baseline ABR (treated bleeds), bleeds/y		
Mean (SD)	17.6 (14.7)	12.2 (15.4)
Median	24.0	8.0
Min, max	0, 40.0	0, 41.0

All participants were male and not Hispanic or Latino. Eligible participants had no history of FVIII inhibitors or anti-AAV5 antibodies, and exclusion criteria included significant liver dysfunction, significant liver fibrosis, and liver cirrhosis.[1-4]
AAV5, adeno-associated virus serotype 5; ABR, annualized bleeding rate; FVIII, factor VIII; max, maximum; min, minimum; SD, standard deviation.

Safety

- No alanine aminotransferase (ALT) elevations reported in the last year
- In the last year, 1 participant in each cohort reported grade 1 treatment-related AEs:
 - Hepatomegaly: one 6x10^{13} cohort participant
 - Splenomegaly and hepatic steatosis: one 4x10^{13} cohort participant
- No treatment-related serious AEs (SAEs) occurred after year 1
- One non-treatment-related SAE occurred in the past year:
 - Grade 4 internal carotid artery (ICA) bleed: one 6x10^{13} cohort participant

Table 2. Safety over 7 years

	6x10^{13} vg/kg cohort (n = 7)							4x10^{13} vg/kg cohort (n = 6)					
	Y1	Y2	Y3	Y4	Y5	Y6	Y7	Y1	Y2	Y3	Y4	Y5	Y6
Any AE	7 (100)	6 (85.7)	7 (100)	7 (100)	7 (100)	5 (71.4)	5 (71.4)	6 (100)	5 (83.3)	5 (83.3)	4 (66.7)	6 (100)	4 (66.7)
Any SAE	0	1 (14.3)	1 (14.3)	1 (14.3)	0	1 (14.3)	1 (14.3)†	1 (16.7)	0	1 (16.7)	1 (16.7)	1 (16.7)	0
Any treatment-related AE	6 (85.7)	1 (14.3)	1 (14.3)	2 (28.6)	1 (14.3)	0	1 (14.3)¥	6 (100)	0	0	0	1 (16.7)	1 (16.7)£
Any treatment-related SAE	0	0	0	0	0	0	0	1 (16.7)*	0	0	0	0	0
AEs of special interest													
ALT elevation§	6 (85.7)	0	0	1 (14.3)	1 (14.3)	0	0	4 (66.7)	0	1 (16.7)	0	0	0
AEs of liver dysfunction#	6 (85.7)	1 (14.3)	0	1 (14.3)	1 (14.3)	0	0	5 (83.3)	0	1 (16.7)	0	0	0
Infusion-related reactions	3 (42.9)	0	0	0	0	0	0	4 (66.7)	0	0	0	0	0

Data are presented as n (%).
*Pyrexia on study day 2. †Grade 4 SAE of spontaneous ICA bleeding during Y7. ¥Grade 1 hepatomegaly during Y7. £Grade 1 splenomegaly, in addition to a worsening of hepatic steatosis during Y6.
§Defined as ALT ≥1.5x ULN or ALT ≥1.5x baseline. #Identified with a MedDRA search strategy using the high-level term "liver function analyses."
AE, adverse event; ALT, alanine aminotransferase; ICA, internal carotid artery; MedDRA, Medical Dictionary for Regulatory Activities; SAE, serious AE; ULN, upper limit of normal; Y, year.

Efficacy

Figure 3. Sustained reduction in annualized treated bleeding rate

6x10^{13} vg/kg cohort (n = 7*) | 4x10^{13} vg/kg cohort (n = 6)

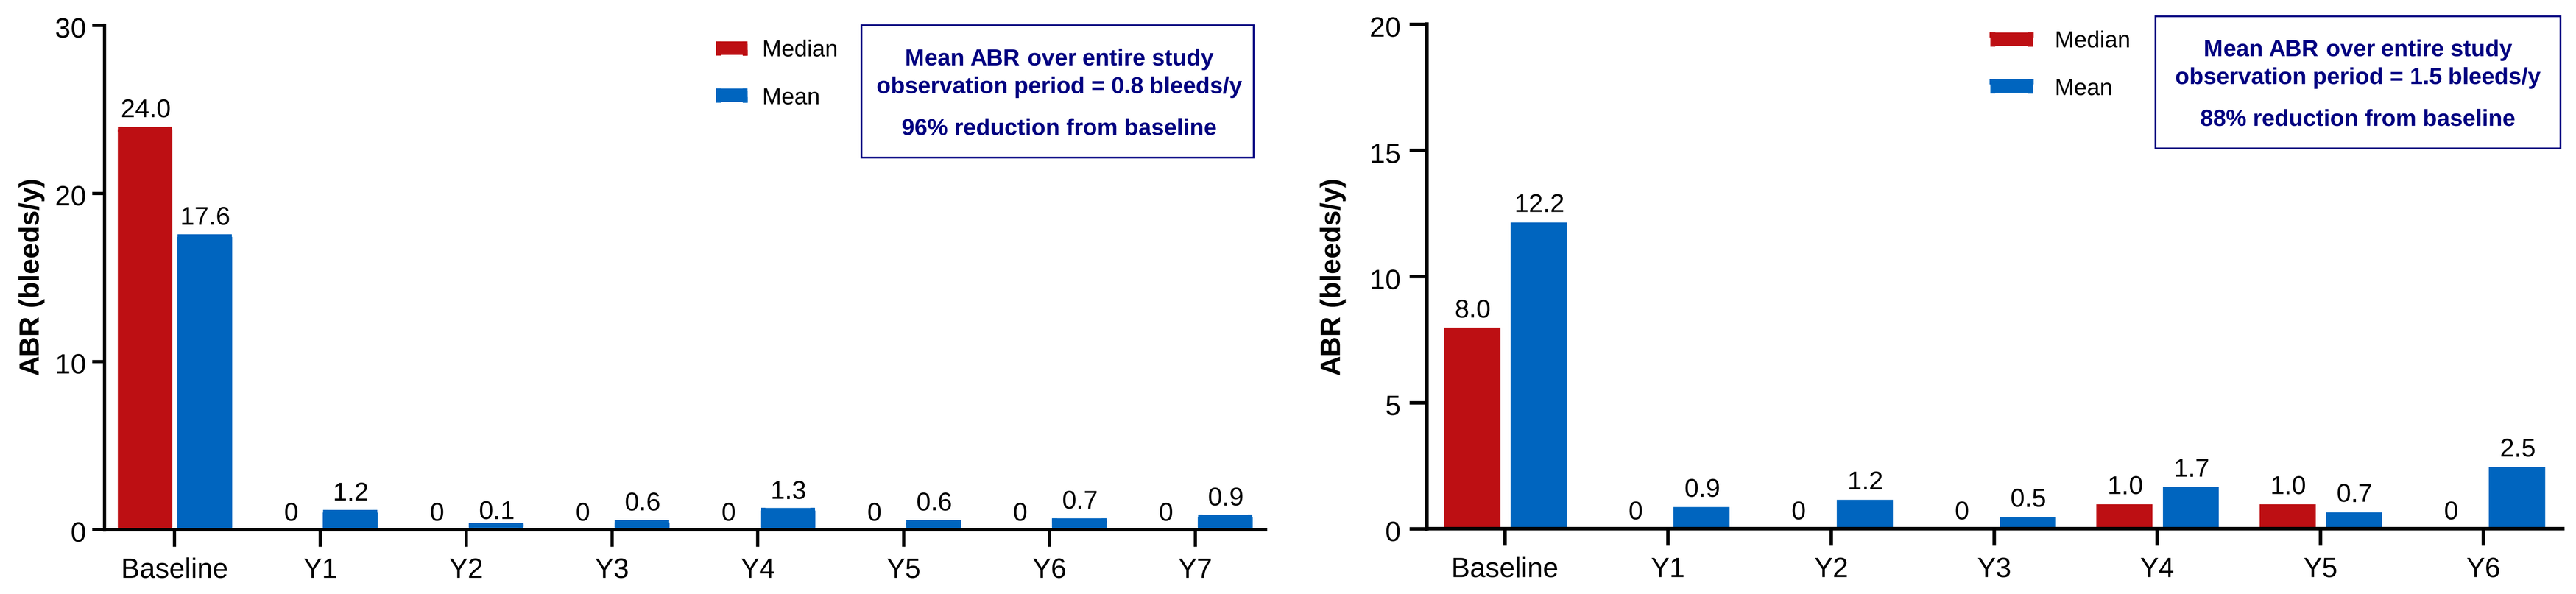
ABR (bleeds/y)
Median
Mean
Mean ABR over entire study observation period = 0.8 bleeds/y
96% reduction from baseline
Baseline 24.0 17.6; Y1 0 1.2; Y2 0 0.1; Y3 0 0.6; Y4 0 1.3; Y5 0 0.6; Y6 0 0.7; Y7 0 0.9
Mean ABR over entire study observation period = 1.5 bleeds/y
88% reduction from baseline
Baseline 8.0 12.2; Y1 0 0.9; Y2 0 1.2; Y3 0 0.5; Y4 1.0 1.7; Y5 1.0 0.7; Y6 0 2.5

n (%) participants bleed free (n = 7)							
Baseline	Y1	Y2	Y3	Y4	Y5	Y6	Y7
1 (14)	5 (71)	6 (86)	6 (86)	5 (71)	6 (86)	4 (57)	4 (57)

n (%) participants bleed free (n = 6)						
Baseline	Y1	Y2	Y3	Y4	Y5	Y6
1 (17)	5 (83)	4 (67)	4 (67)	3 (50)	2 (33)	5 (83)

*Six of the 7 participants in the 6x10^{13} vg/kg cohort were receiving regular FVIII prophylaxis at baseline (1 participant was receiving on-demand FVIII prophylaxis and was excluded). Baseline (n = 6) mean and median ABR were 16.3 bleeds/y and 16.5 bleeds/y, and the mean ABR over the entire study was 0.8 bleeds/y, representing a 95% decrease from baseline.
ABR, annualized bleeding rate; FVIII, factor VIII; Y, year.

- Two participants from the 6x10^{13} vg/kg cohort returned to prophylaxis (FVIII and emicizumab) during year 7 and continued through the cutoff
 - Five 6x10^{13} vg/kg cohort participants chose to remain off FVIII prophylaxis
- All 5 remaining participants in the 4x10^{13} vg/kg cohort have chosen to remain off prophylaxis

Figure 4. Sustained reduction in annualized FVIII infusion rate

6x10^{13} vg/kg cohort (n = 7*) | 4x10^{13} vg/kg cohort (n = 6)

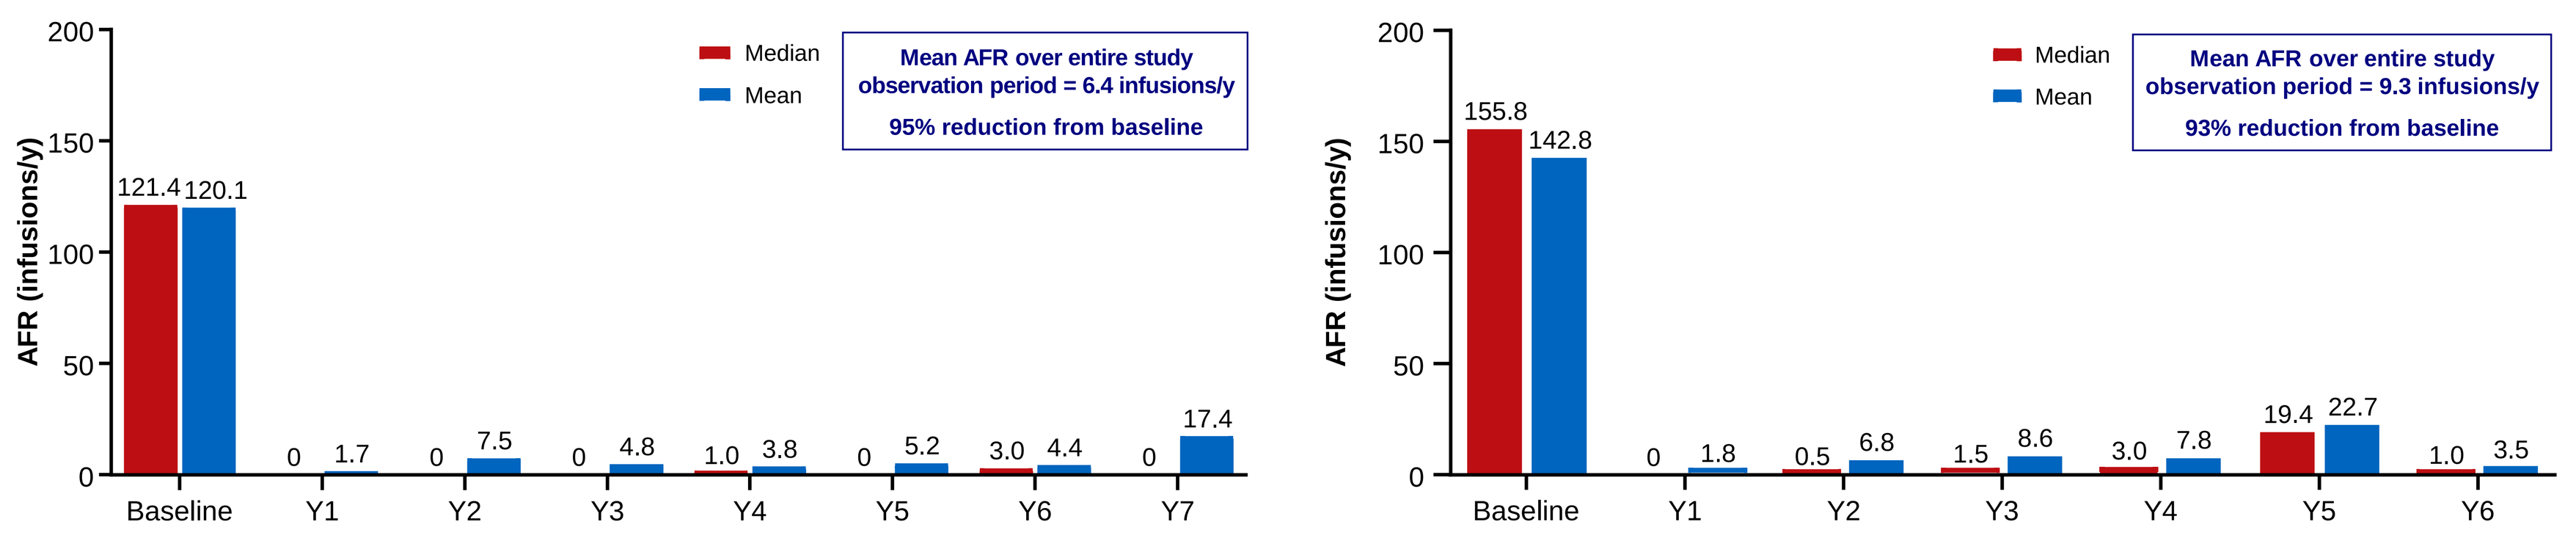
AFR (infusions/y)
Median
Mean
Mean AFR over entire study observation period = 6.4 infusions/y
95% reduction from baseline
Baseline 121.4 120.1; Y1 0 1.7; Y2 0 7.5; Y3 0 4.8; Y4 1.0 3.8; Y5 0 5.2; Y6 3.0 4.4; Y7 0 17.4
Mean AFR over entire study observation period = 9.3 infusions/y
93% reduction from baseline
Baseline 155.8 142.8; Y1 0 1.8; Y2 0.5 6.8; Y3 1.5 8.6; Y4 3.0 7.8; Y5 19.4 22.7; Y6 1.0 3.5

Infusion rate by reason after week 4 (n = 7)				
no./y	Treatment for bleed	Usual prophylaxis	Surgery/ procedures	One-time prophylaxis
Mean	1.5	1.8	2.1	1.0
Median	0.6	0	1.0	0

Infusion rate by reason after week 4 (n = 6)				
no./y	Treatment for bleed	Usual prophylaxis	Surgery/ procedures	One-time prophylaxis
Mean	3.5	0.7	2.5	2.5
Median	2.1	0	0.7	1.0

*Six of the 7 participants were receiving regular FVIII prophylaxis at baseline (1 participant was receiving on-demand FVIII prophylaxis and was excluded). Baseline (n = 6) mean and median AFR were 135.6 infusions/y and 136.6 infusions/y, respectively, and the mean AFR over the entire study period was 7.2 infusions/y, representing a 95% reduction from baseline.
AFR, annualized FVIII infusion rate; FVIII, factor VIII; no., number; Y, year.

Figure 5. FVIII activity rate of change over time

6x10^{13} vg/kg cohort (n = 7) | 4x10^{13} vg/kg cohort (n = 6)

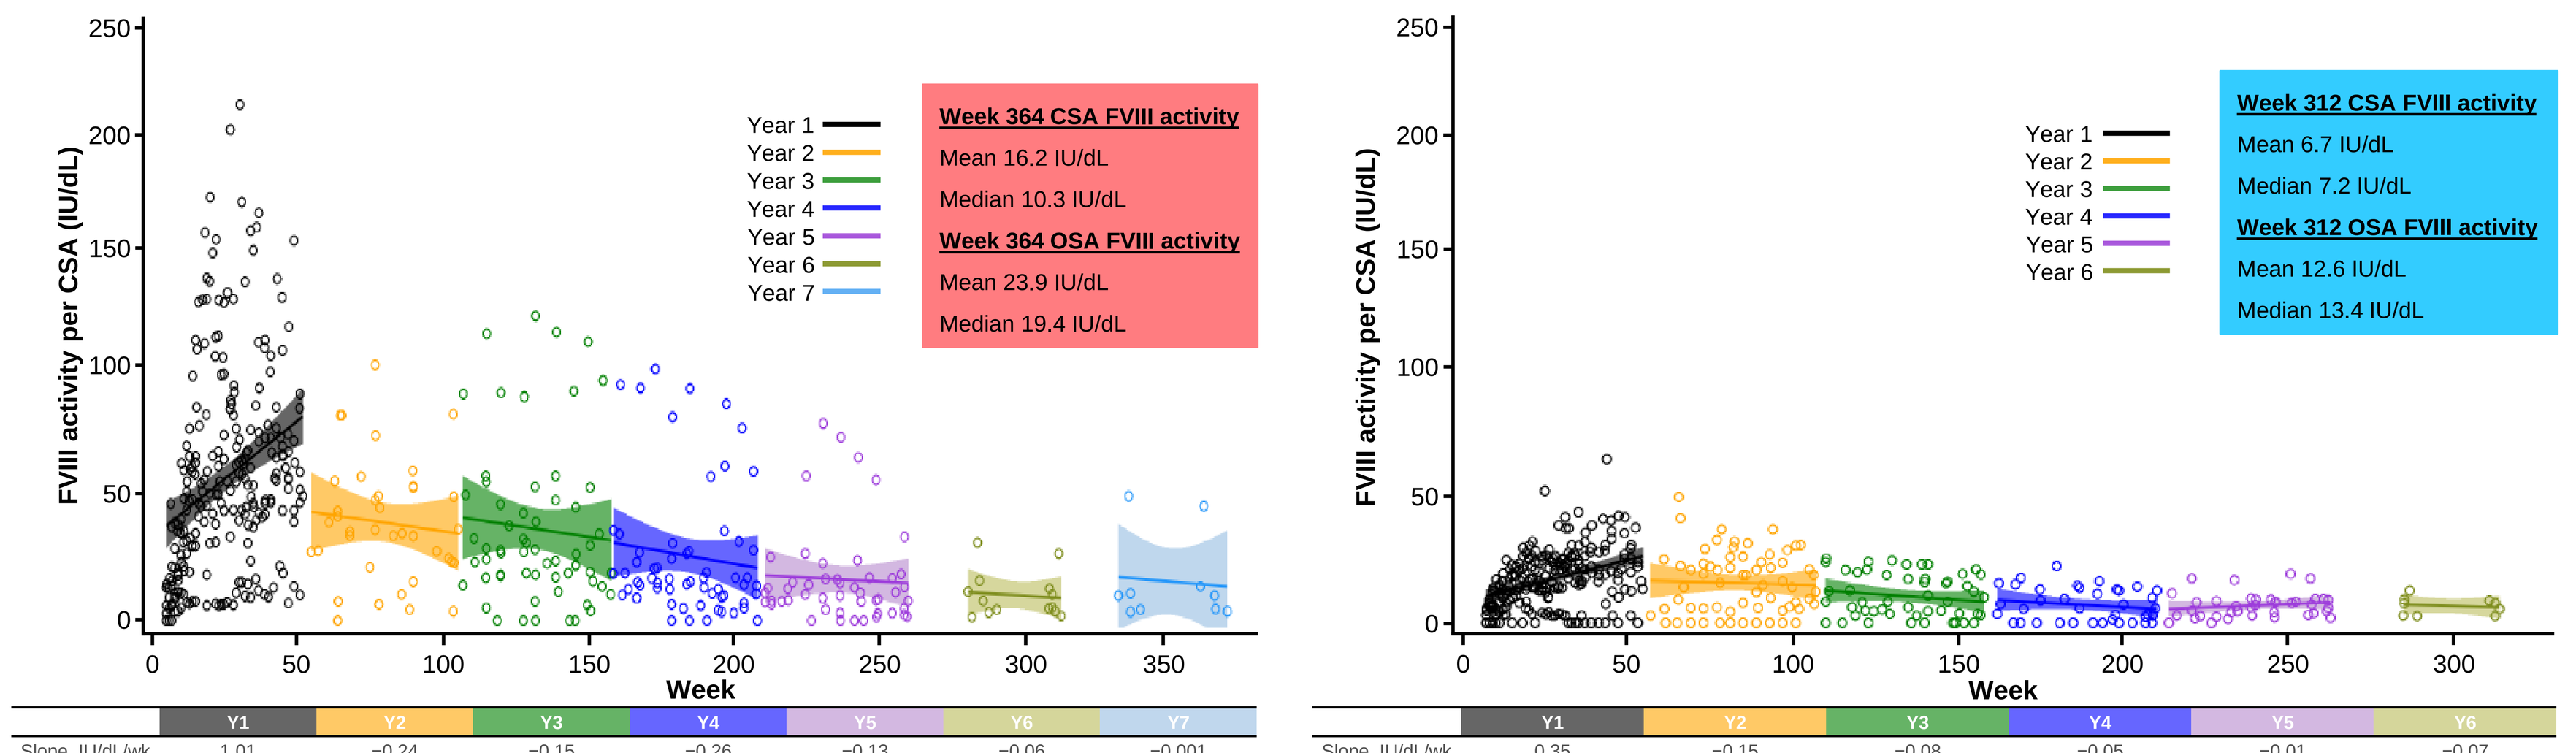
FVIII activity per CSA (IU/dL)
Week
Year 1
Year 2
Year 3
Year 4
Year 5
Year 6
Year 7
Week 364 CSA FVIII activity
Mean 16.2 IU/dL
Median 10.3 IU/dL
Week 364 OSA FVIII activity
Mean 23.9 IU/dL
Median 19.4 IU/dL
Week 312 CSA FVIII activity
Mean 6.7 IU/dL
Median 7.2 IU/dL
Week 312 OSA FVIII activity
Mean 12.6 IU/dL
Median 13.4 IU/dL

	Y1	Y2	Y3	Y4	Y5	Y6	Y7
Slope, IU/dL/wk (95% CI)	1.01 (−0.04, 2.06)	−0.24 (−0.68, 0.21)	−0.15 (−0.47, 0.16)	−0.26 (−0.49, −0.03)	−0.13 (−0.31, 0.04)	−0.06 (−0.20, 0.08)	−0.001 (−0.18, 0.17)
Annual absolute % change (sample size)	NA	−43% (N = 7)	−10% (N = 7)	−26% (N = 6)	−50% (N = 7)	−19% (N = 7)	65% (N = 5)

	Y1	Y2	Y3	Y4	Y5	Y6
Slope, IU/dL/wk (95% CI)	0.35 (−0.01, 0.71)	−0.15 (−0.37, 0.07)	−0.08 (−0.17, 0.02)	−0.05 (−0.13, 0.02)	−0.01 (−0.08, 0.07)	−0.07 (−0.20, 0.06)
Annual absolute % change (sample size)	NA	−30% (N = 5)	−33% (N = 6)	−39% (N = 6)	27% (N = 5)	−12% (N = 4)

Values from participants who returned to prophylaxis were excluded after they returned to prophylaxis to reflect the true treatment effect by removing the impact from resuming prophylaxis. Missing data were not imputed. Slope (95% CI) is for FVIII activity per CSA.
CI, confidence interval; CSA, chromogenic substrate assay; FVIII, factor VIII; IU, international unit; NA, not applicable; OSA, one-stage assay; Y, year.

Figure 6. Individual participant FVIII activity per CSA

6x10^{13} vg/kg cohort (n = 7) | 4x10^{13} vg/kg cohort (n = 6)

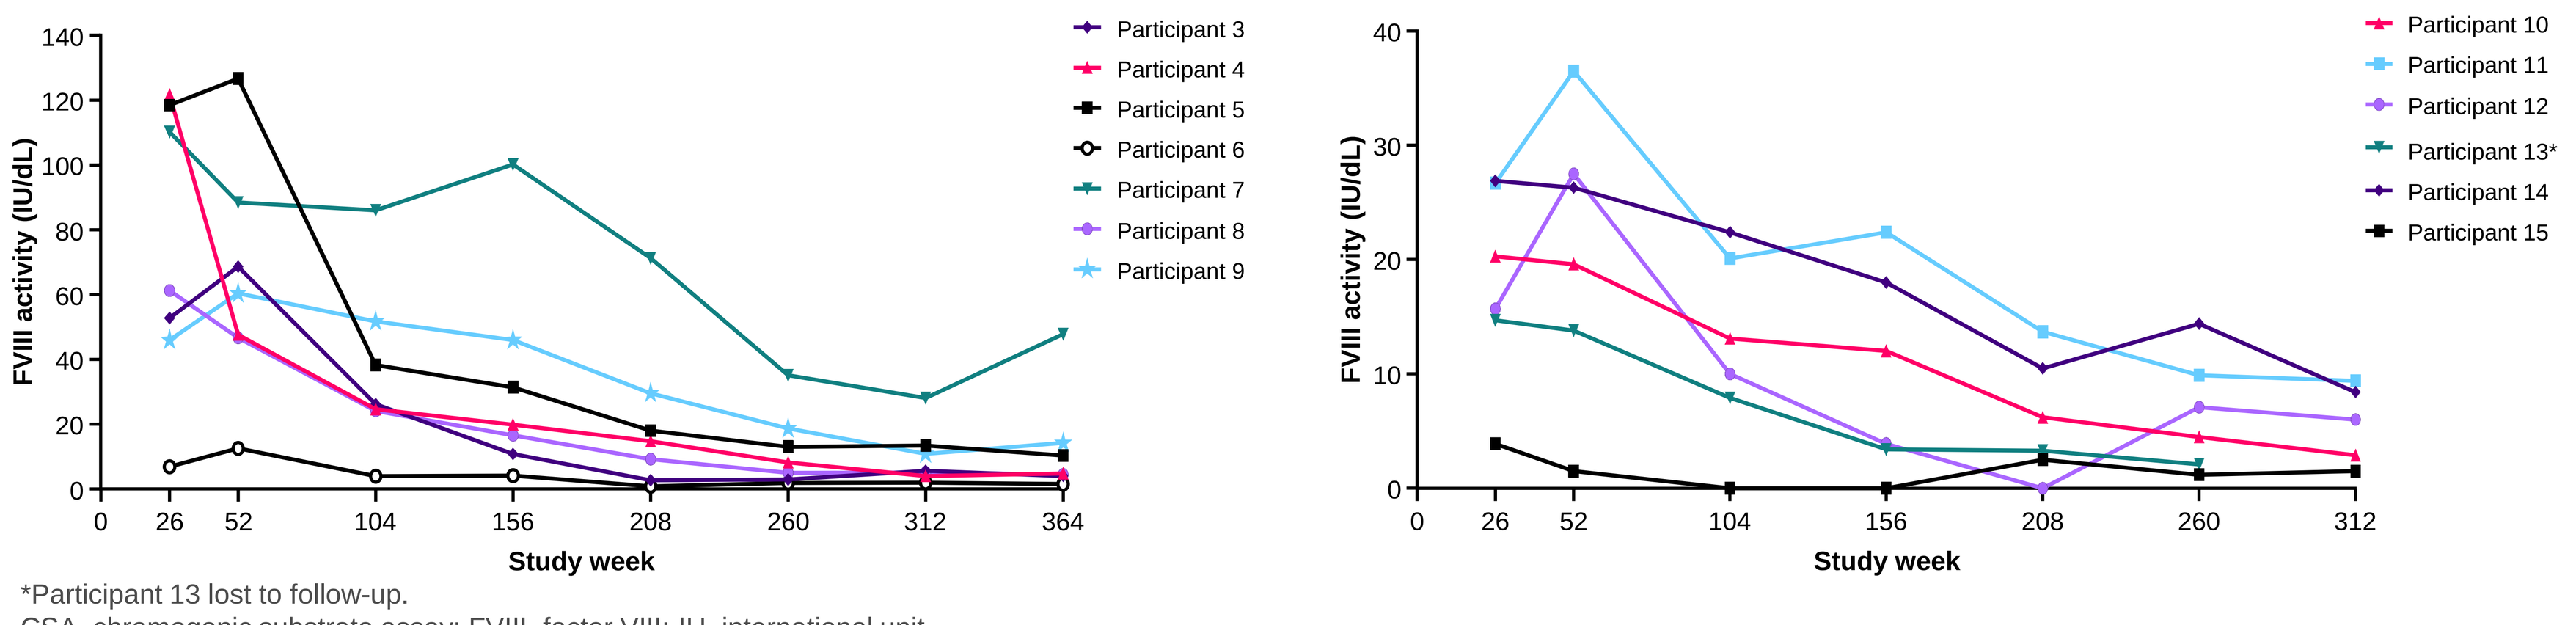
FVIII activity (IU/dL)
Study week
Participant 3
Participant 4
Participant 5
Participant 6
Participant 7
Participant 8
Participant 9
Participant 10
Participant 11
Participant 12
Participant 13*
Participant 14
Participant 15

*Participant 13 lost to follow-up.
CSA, chromogenic substrate assay; FVIII, factor VIII; IU, international unit.

Figure 7. Individual FVIII activity, ABR, and FVIII infusion rates

6x10^{13} vg/kg cohort (n = 7) | 4x10^{13} vg/kg cohort (n = 6)

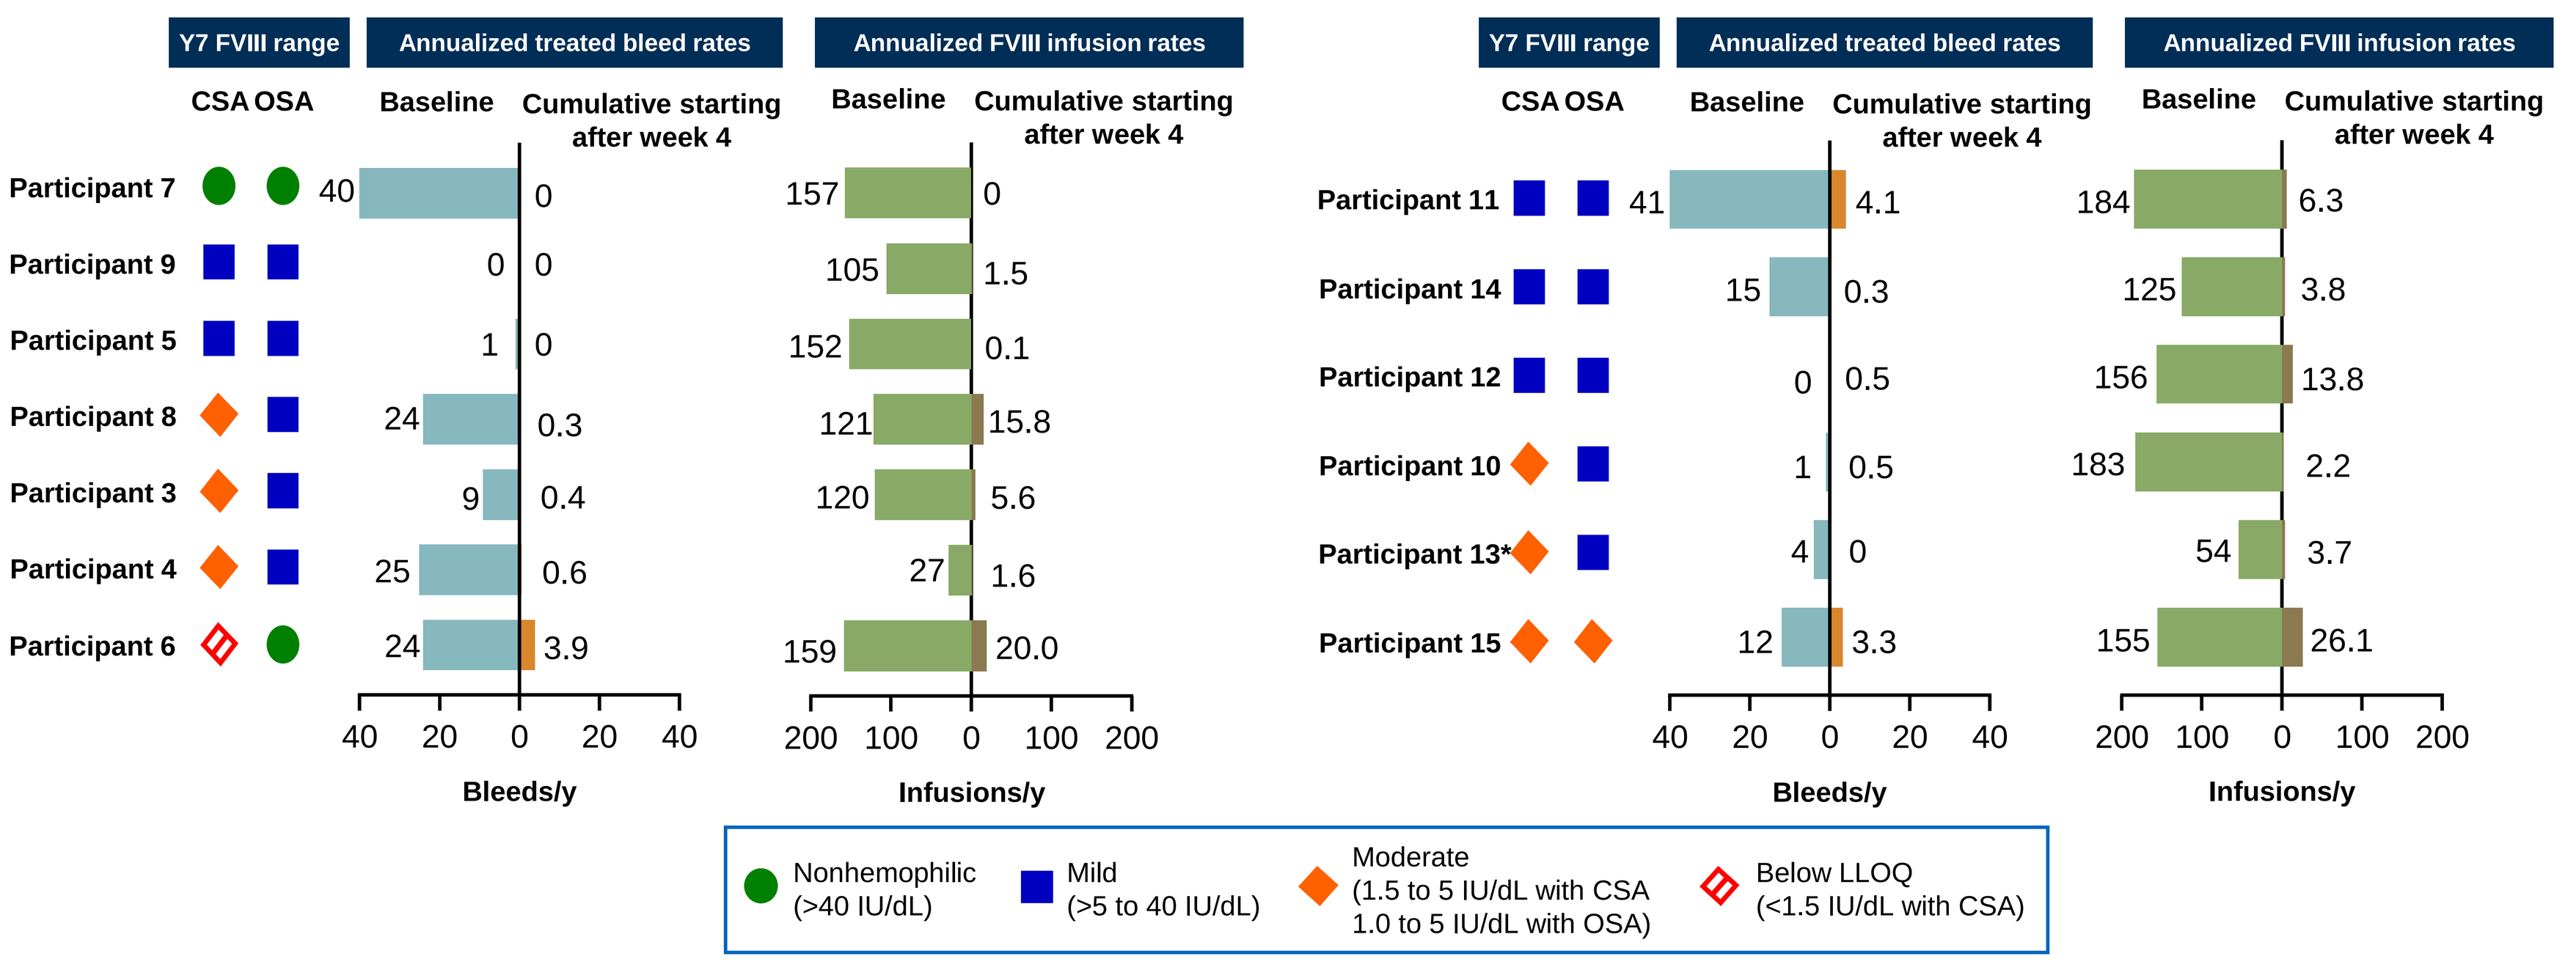
Y7 FVIII range
Annualized treated bleed rates
Annualized FVIII infusion rates
CSA OSA
Baseline
Cumulative starting after week 4
Participant 7
Participant 9
Participant 5
Participant 8
Participant 3
Participant 4
Participant 6
Participant 11
Participant 14
Participant 12
Participant 10
Participant 13*
Participant 15
Bleeds/y
Infusions/y
Nonhemophilic (>40 IU/dL)
Mild (>5 to 40 IU/dL)
Moderate (1.5 to 5 IU/dL with CSA 1.0 to 5 IU/dL with OSA)
Below LLOQ (<1.5 IU/dL with CSA)

Participants 6 and 8 resumed FVIII prophylaxis during Y7.
FVIII activity is for week 364 for the 6x10^{13} vg/kg cohort and week 312 for the 4x10^{13} vg/kg cohort (week 286 for participant 13).
*Participant 13 lost to follow-up after week 286.
ABR, annualized bleeding rate; CSA, chromogenic substrate assay; FVIII, factor VIII; LLOQ, lower limit of quantitation; OSA, one-stage assay; Y, year.

Conclusions

- Over the course of 7 and 6 years, safety outcomes following respective doses of 6x10^{13} and 4x10^{13} vg/kg of valoctocogene roxaparvovec remained consistent with previous reports
- No long-term sequalae were observed as a result of corticosteroid treatment, and no participants experienced thrombotic events
- No participants developed FVIII inhibitors
- Two participants returned to prophylaxis to treat bleeding events; the remaining participants chose not to resume FVIII prophylaxis
- Despite the slow decline in FVIII levels, valoctocogene roxaparvovec continues to support hemostasis for the majority of the trial population

Acknowledgements

We thank all the trial participants, their families, and study-site personnel. Medical writing support was provided by Taryn Bosquez-Berger, PhD, of AlphaBioCom, a Red Nucleus company, and funded by BioMarin Pharmaceutical Inc. Project management support was provided by Jonathan Morton, PhD, of BioMarin Pharmaceutical Inc. Funding for this trial was provided by BioMarin Pharmaceutical Inc.

Conflict of interest statement

ES received grants from BioMarin Pharmaceutical Inc. and travel support from CSL Behring and Novo Nordisk. **SR** received grants from Roche and Sangamo, travel support from Reliance Life Sciences and Shire/Takeda, and consulting payments from Pfizer, Reliance Life Sciences, Sanofi, and Shire/Takeda. **WL** received grants from BioMarin Pharmaceutical Inc.; personal fees from Bayer, LFB Biopharmaceuticals, Novo Nordisk, Sobi, and Takeda; and travel support from CSL Behring and Takeda. **GFP** received consulting payments from BioMarin Pharmaceutical Inc., Decibel Therapeutics, Frontera, Generation Bio, Regeneron Pharmaceuticals, Spark Therapeutics, and Third Rock Ventures and is a board member of Be Bio, the Medical and Scientific Advisory Council of the US National Hemophilia Foundation, Metagenomi, Pfizer, Spark Therapeutics, Voyager Therapeutics, and the World Federation of Hemophilia. **PR** has received grant/travel support from CSL Behring, Sobi, and Takeda and advisory honoraria from Idogen, Sigilon, Sobi, and Pfizer. **TMR**, **DO**, and **ML** are employees and shareholders of BioMarin Pharmaceutical Inc. **ML** received grants from BioMarin Pharmaceutical Inc.; personal fees from Bayer, LEO Pharma, LFB Biopharmaceuticals, Pfizer, Roche, Shire, and Sobi; and travel support from Bayer, LFB Biopharmaceuticals, and Sobi. **CM** has received research support from Baxter/Takeda, CSL Behring, and Grifols and honoraria or consultation fees from CSL Behring, LFB Biopharmaceuticals, Octapharma, and Takeda. She has participated in advisory boards for CSL Behring and Takeda. **BM** has no conflicts to disclose.

References

1) Rangarajan S, et al. *N Engl J Med*. 2017;377(26):2519-30. **2)** Pasi KJ, et al. *N Engl J Med*. 2020;382:29-40. **3)** Pasi KJ, et al. *Haemophilia*. 2021;1:1-10. **4)** Symington E, et al. *Haemophilia*. 2023: In Review. **5)** Ozelo MC, et al. *N Engl J Med*. 2022;386:1013-25.

Presented at the European Association for Haemophilia and Allied Disorders (EAHAD) Annual Meeting: February 6–9, 2024; Frankfurt, Germany

©2024 BioMarin Pharmaceutical Inc. All rights reserved.

MMRC-BMN27-00803