CR28

Plain language summary of 4-year efficacy and safety data in the GENEr8-1 trial of valoctocogene roxaparvovec, a gene therapy for severe hemophilia A

Andrew D. Leavitt[1], Johnny Mahlangu[2], Emily Symington[3], Doris V. Quon[4], Adam Giermasz[5], Nigel S. Key[6], Steven W. Pipe[7], Francisco P. Careta[8], Patrick James Lynch[9], Andres Ruiz[10], Hua Yu[11], Christine Rivat[11], Margareth C. Ozelo[12]

[1]Adult Hemophilia Treatment Center, University of California San Francisco, San Francisco, CA, USA; [2]Hemophilia Comprehensive Care Center, Charlotte Maxeke Johannesburg Academic Hospital, and Department of Molecular Medicine and Haematology, Faculty of Health Sciences, University of the Witwatersrand and NHLS, Johannesburg, South Africa; [3]Cambridge University Hospitals NHS Foundation Trust, Cambridge, UK; [4]Orthopaedic Hemophilia Treatment Center, Los Angeles, CA, USA; [5]Hemophilia Treatment Center, University of California Davis, Sacramento, CA, USA; [6]UNC Blood Research Center, University of North Carolina, Chapel Hill, NC, USA; [7]Departments of Pediatrics and Pathology, University of Michigan, Ann Arbor, MI, USA; [8]Universidade Federal do Espírito Santo, Alegre, ES, Brazil; [9]Believe Ltd, Hawthorne, CA, USA; [10]National Bleeding Disorders Foundation, Oakland Park, FL, USA; [11]BioMarin Pharmaceutical Inc., Novato, CA, USA; [12]Hemocentro UNICAMP, Department of Internal Medicine, School of Medical Sciences, University of Campinas, Campinas, SP, Brazil

What is hemophilia A?

Hemophilia A is an inherited disorder that causes people to bleed easily because they have lower-than-normal levels of ***FVIII*** (factor VIII; "factor eight"), a blood-clotting protein

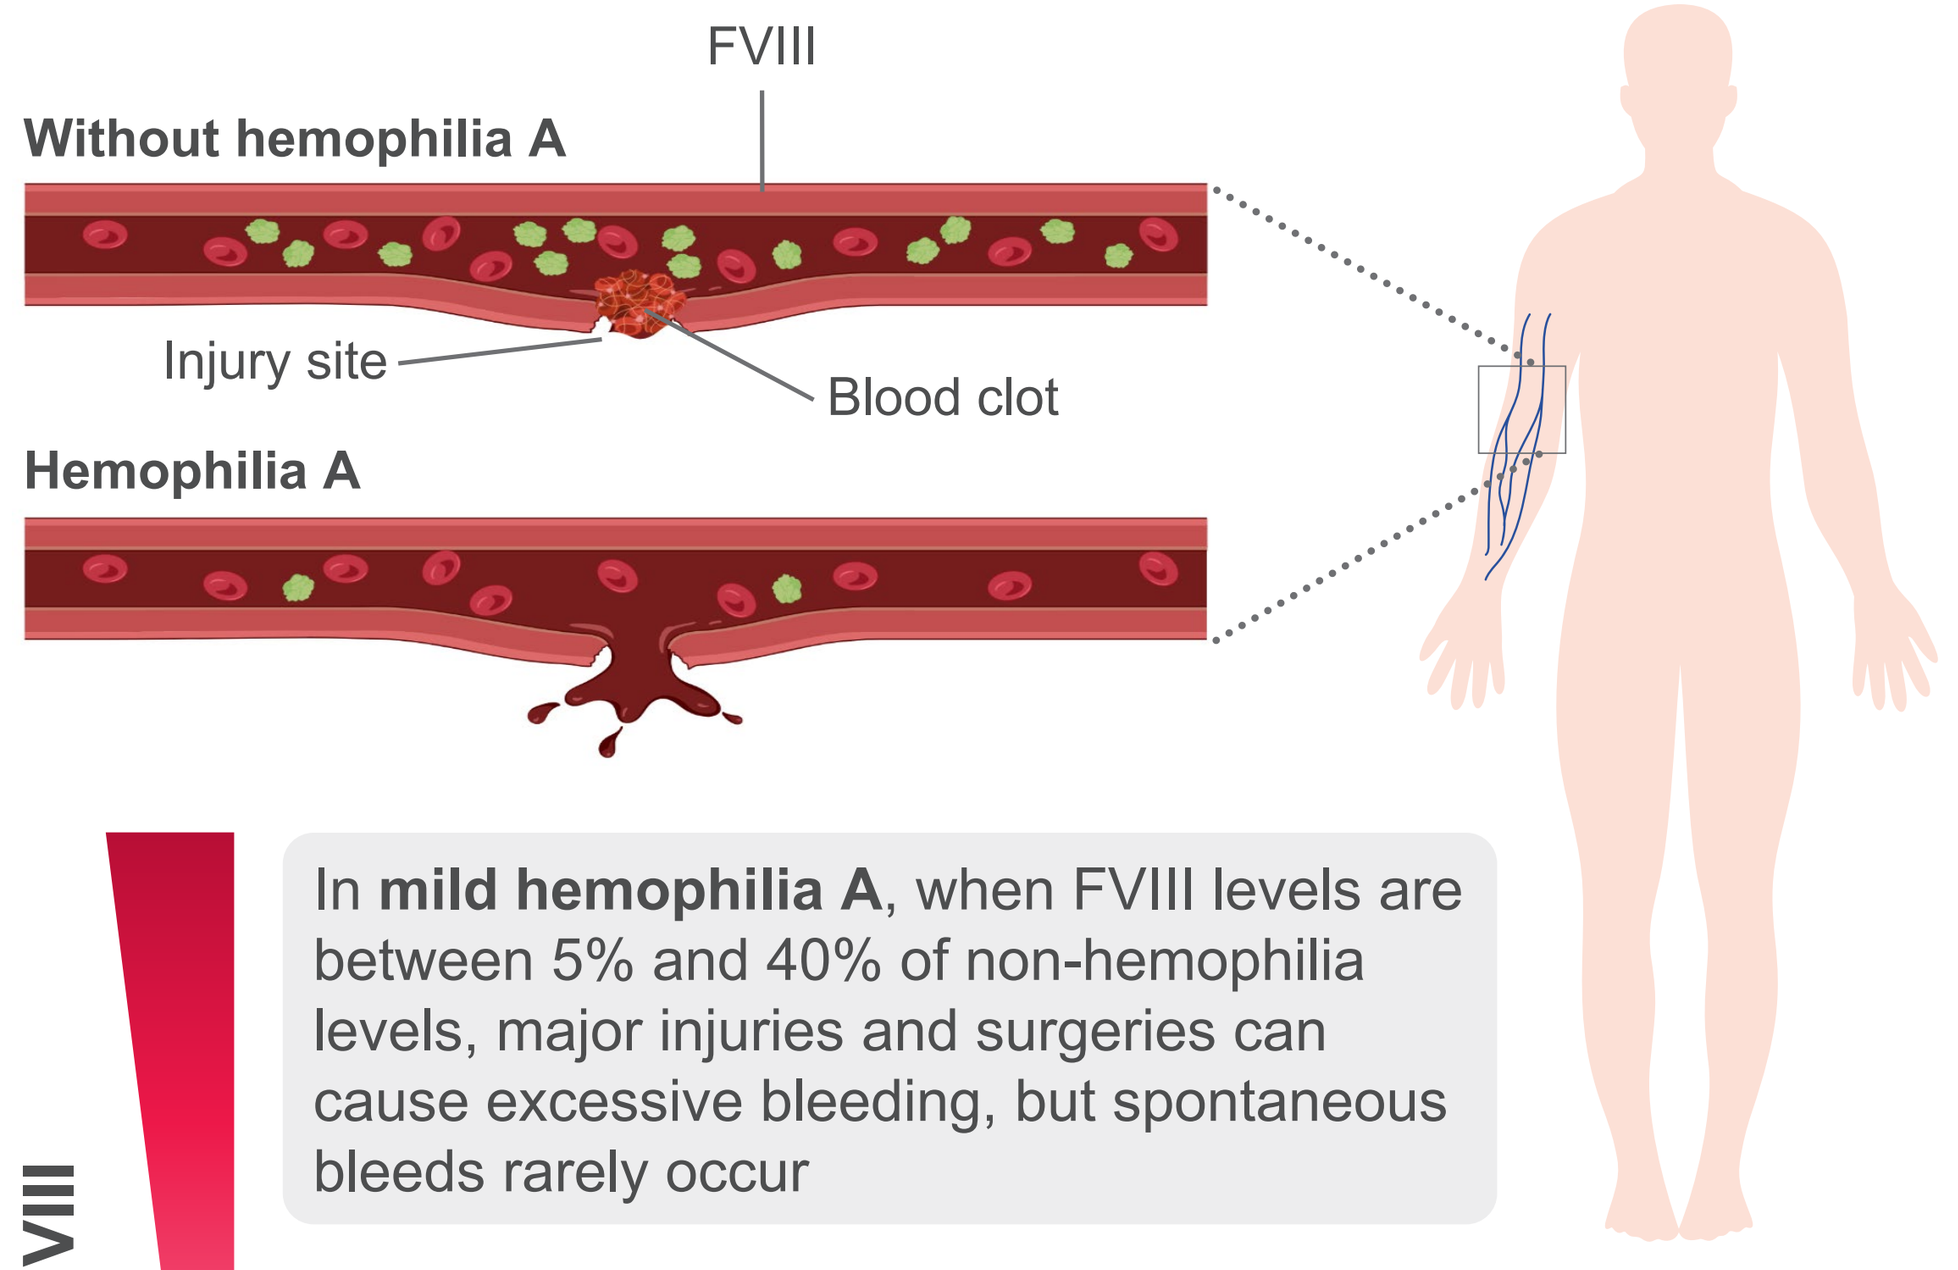
FVIII
Without hemophilia A
Injury site
Blood clot
Hemophilia A

Factor VIII

In **mild hemophilia A**, when FVIII levels are between 5% and 40% of non-hemophilia levels, major injuries and surgeries can cause excessive bleeding, but spontaneous bleeds rarely occur

In **moderate hemophilia A**, when FVIII levels are between 1% and 5%, excess bleeding occurs with minor injuries, and spontaneous bleeds occur occasionally

In **severe hemophilia A**, when FVIII levels are less than 1%, bleeding occurs even with no apparent injury, often into joints and muscles

What is valoctocogene roxaparvovec?

Valoctocogene roxaparvovec is the first approved gene therapy for hemophilia A. Gene therapy is meant to address the lack of genetic instructions for making FVIII

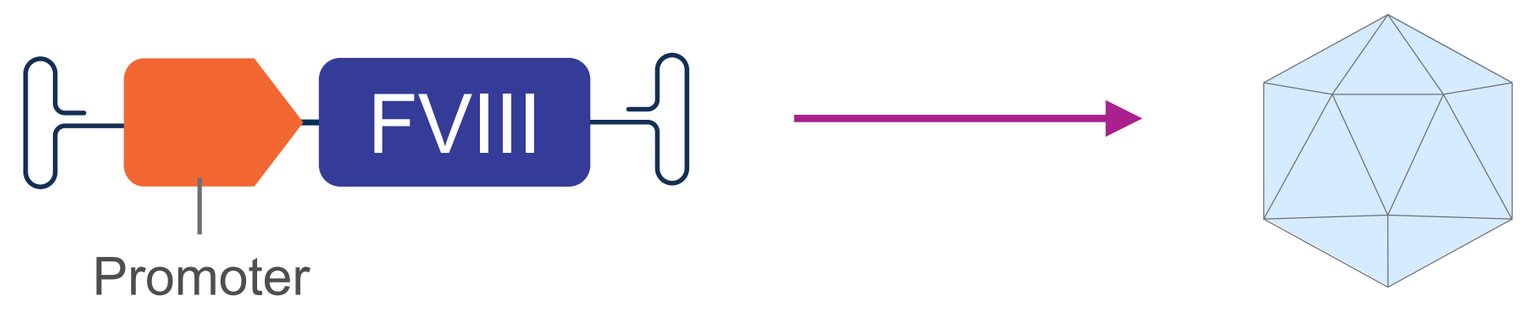
Vector genome
Vector
FVIII
Promoter

The *vector genome* contains instructions for the body to make FVIII

The *promoter* specifies that only liver cells should read the instructions

The *vector* is the empty "shell" of the *adeno-associated virus*

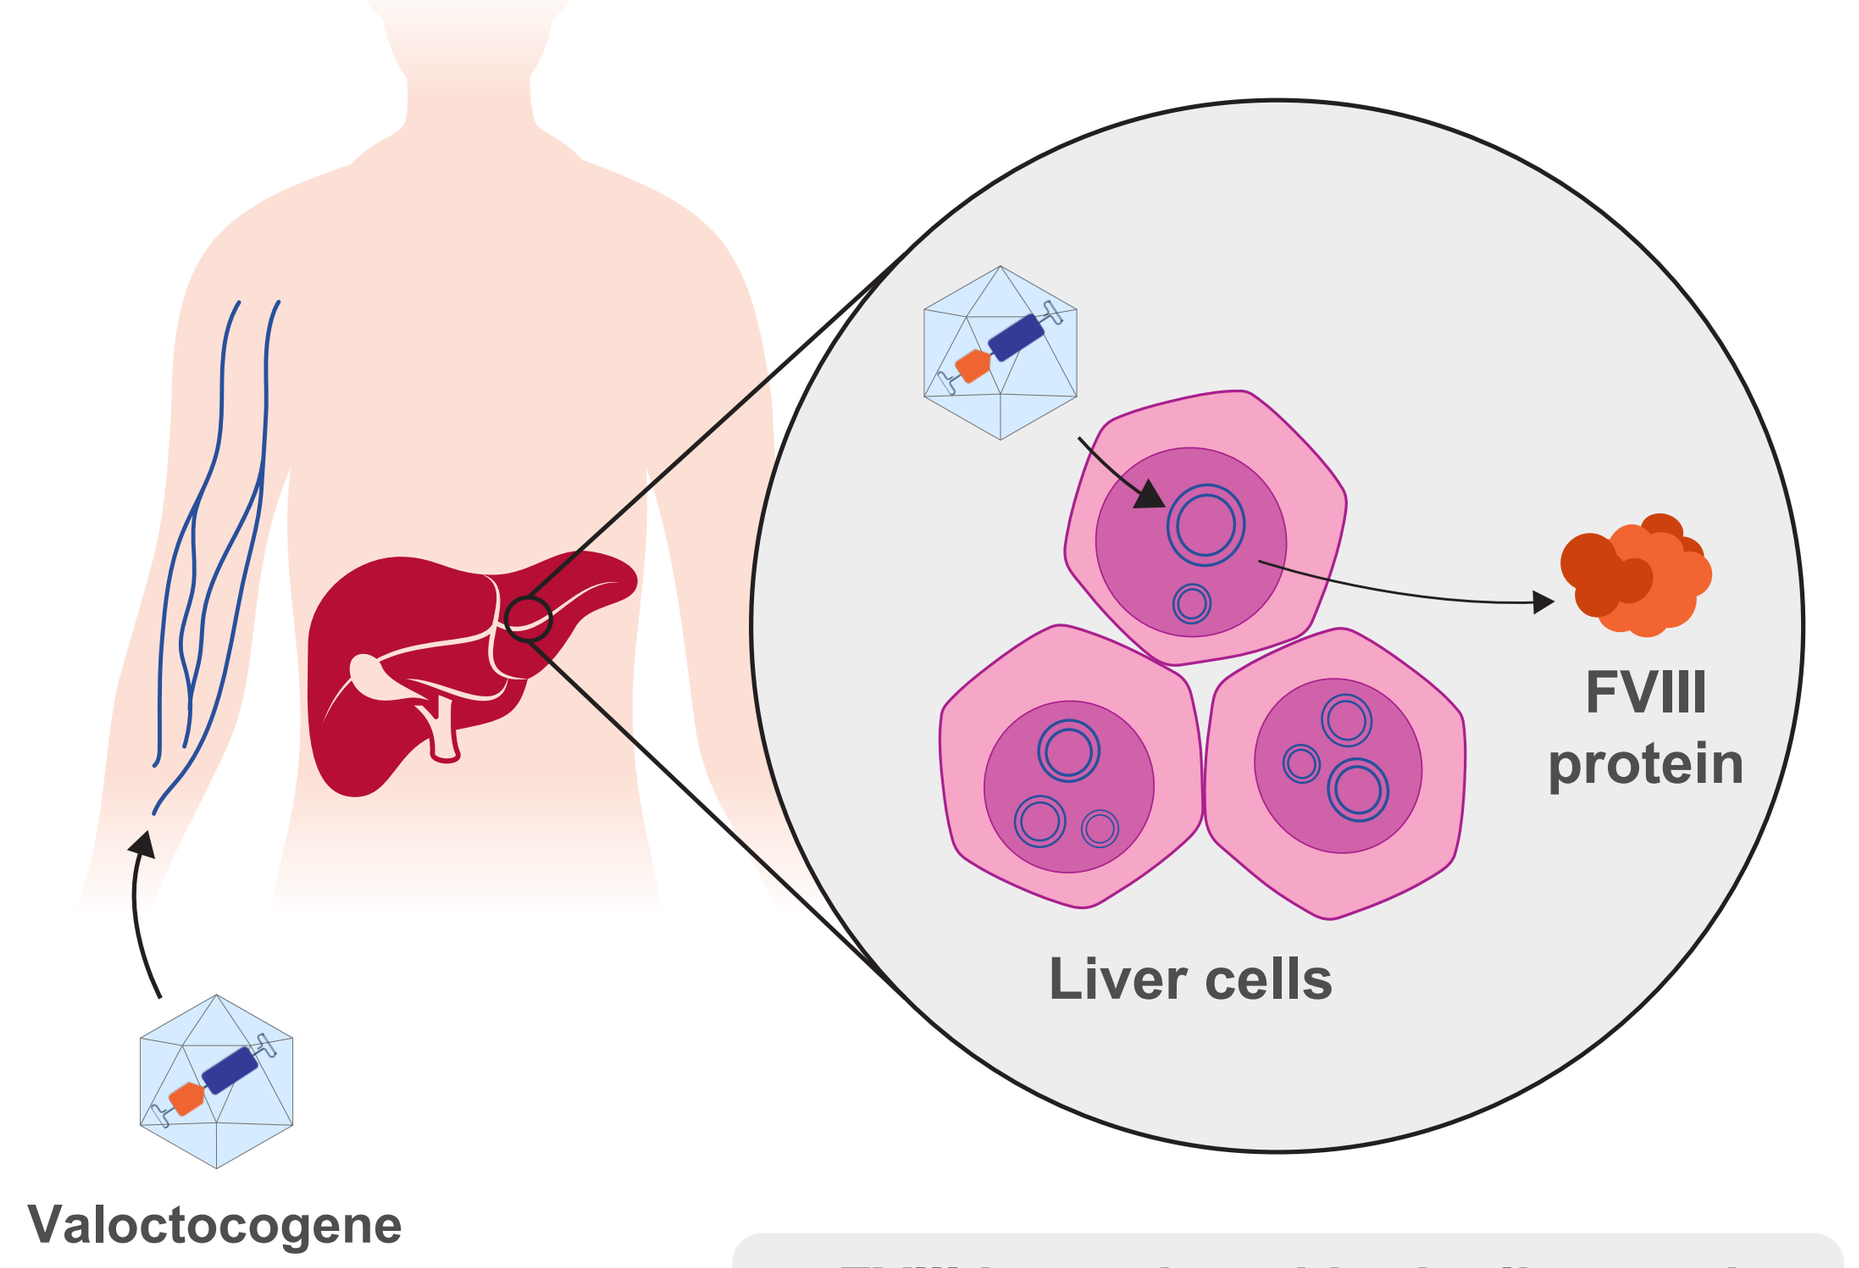
Valoctocogene roxaparvovec
Liver cells
FVIII protein

FVIII is produced in the liver and circulates throughout the body

What is the GENEr8-1 study?

GENEr8-1 is a *phase 3 clinical trial* that is testing the *efficacy* and safety of a single dose of valoctocogene roxaparvovec

What were the goals of the study?

To see how well valoctocogene roxaparvovec prevents bleeding
- How many bleeds participants experienced per year that needed to be treated with FVIII
- How many total bleeds participants experienced per year
- How much FVIII the participants made

To determine how safe valoctocogene roxaparvovec is
- How many *adverse events* occurred
- How many *ALT* elevations occurred, a type of adverse event that suggests liver inflammation

Who could participate in the study?

All participants were
- Males with severe hemophilia A
- Age 18 years or older
- Previously treated with exogenous FVIII *prophylaxis* for at least 1 year before joining

Patients could not join the study if they had
- Liver or kidney problems
- Antibodies to the vector
- FVIII inhibitors

Who participated in the study?

112 participants enrolled following an observational study of FVIII infusions in hemophilia A

22 participants enrolled directly into GENEr8-1

All **134** participants received a single dose of **valoctocogene roxaparvovec**

Years 1–3 after gene therapy

Year 4 after gene therapy

129 participants are still in the study at least 4 years after the gene therapy was given

What were the efficacy results?

Treated bleeding

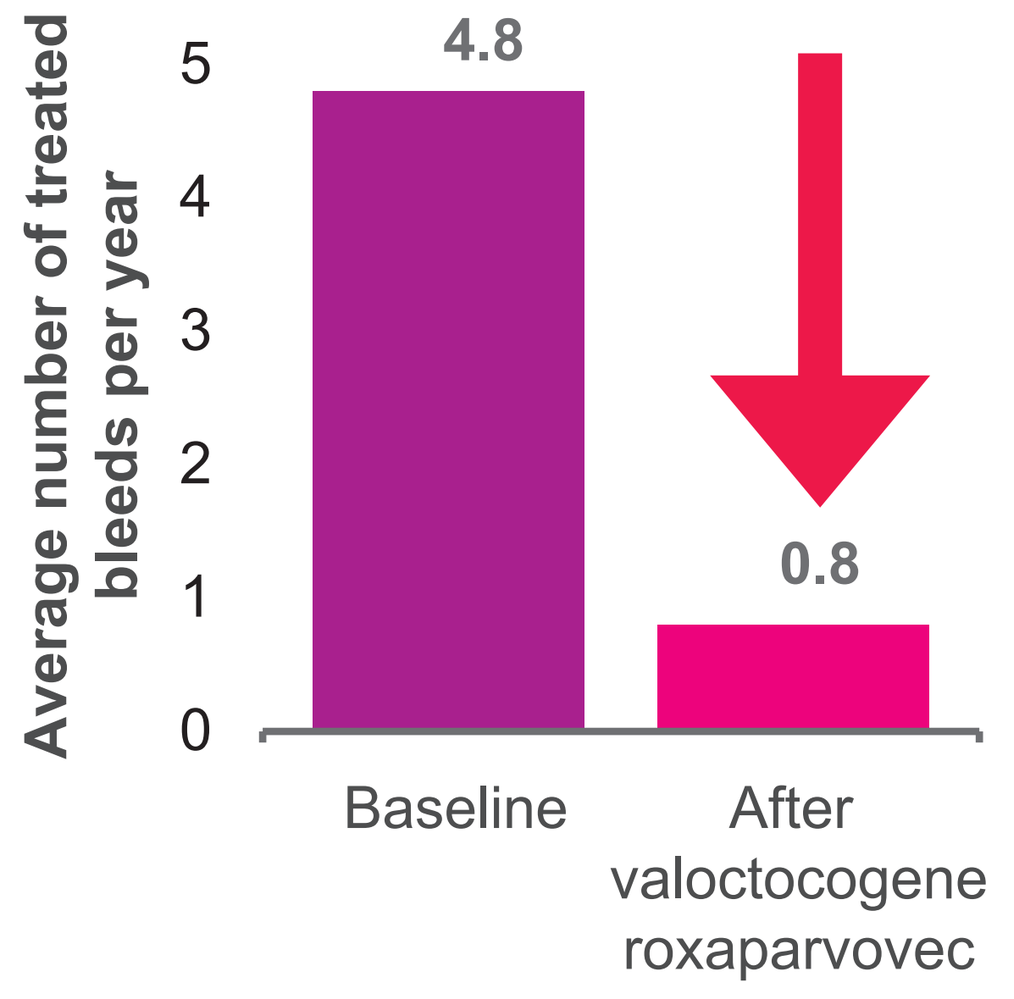
Average number of treated bleeds per year
5
4
3
2
1
0
4.8
0.8
Baseline
After valoctocogene roxaparvovec

The 112 participants who joined from the observational study had **83%** fewer bleeds per year **that needed treatment** after receiving valoctocogene roxaparvovec compared with before

Participants with no treated bleeds

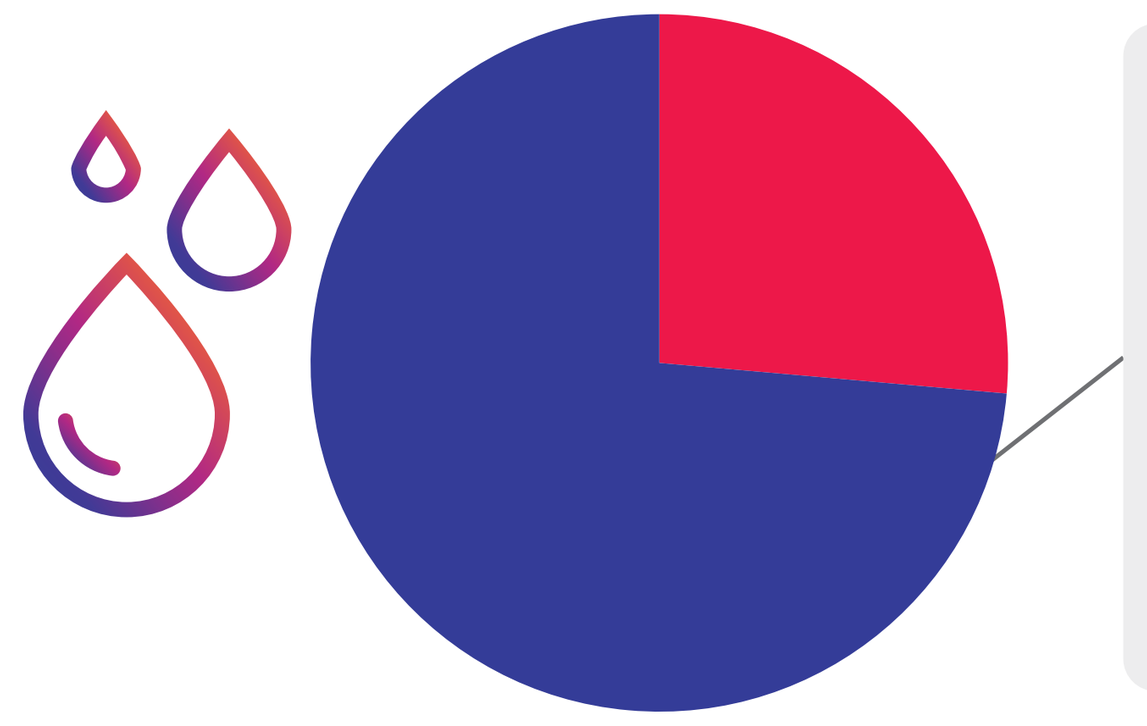

74% of participants who enrolled from the observational study had **no treated bleeds** during year 4

Overall bleeding

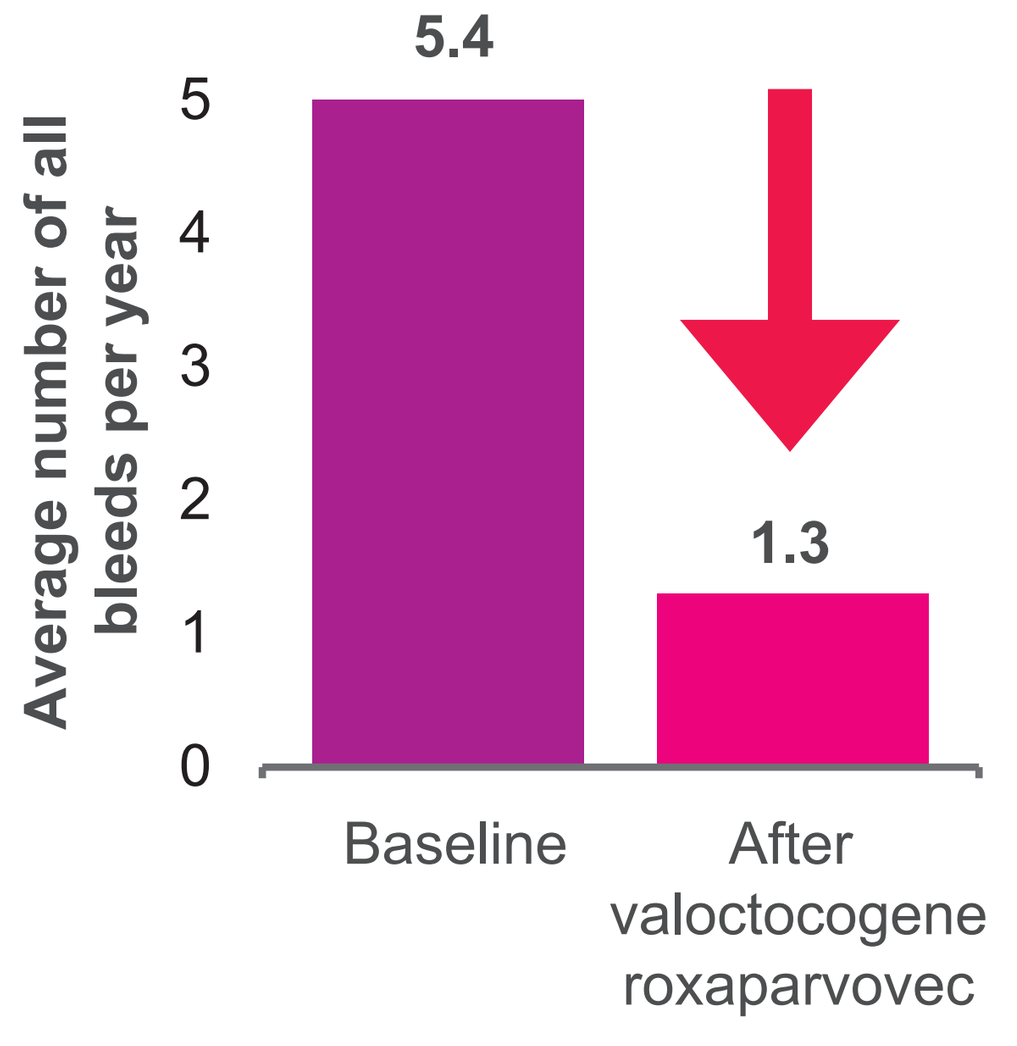
Average number of all bleeds per year
5
4
3
2
1
0
5.4
1.3
Baseline
After valoctocogene roxaparvovec

The 112 participants who joined from the observational study had **77%** fewer bleeds per year **regardless of treatment** after receiving valoctocogene roxaparvovec compared with before

Timing of return to prophylaxis

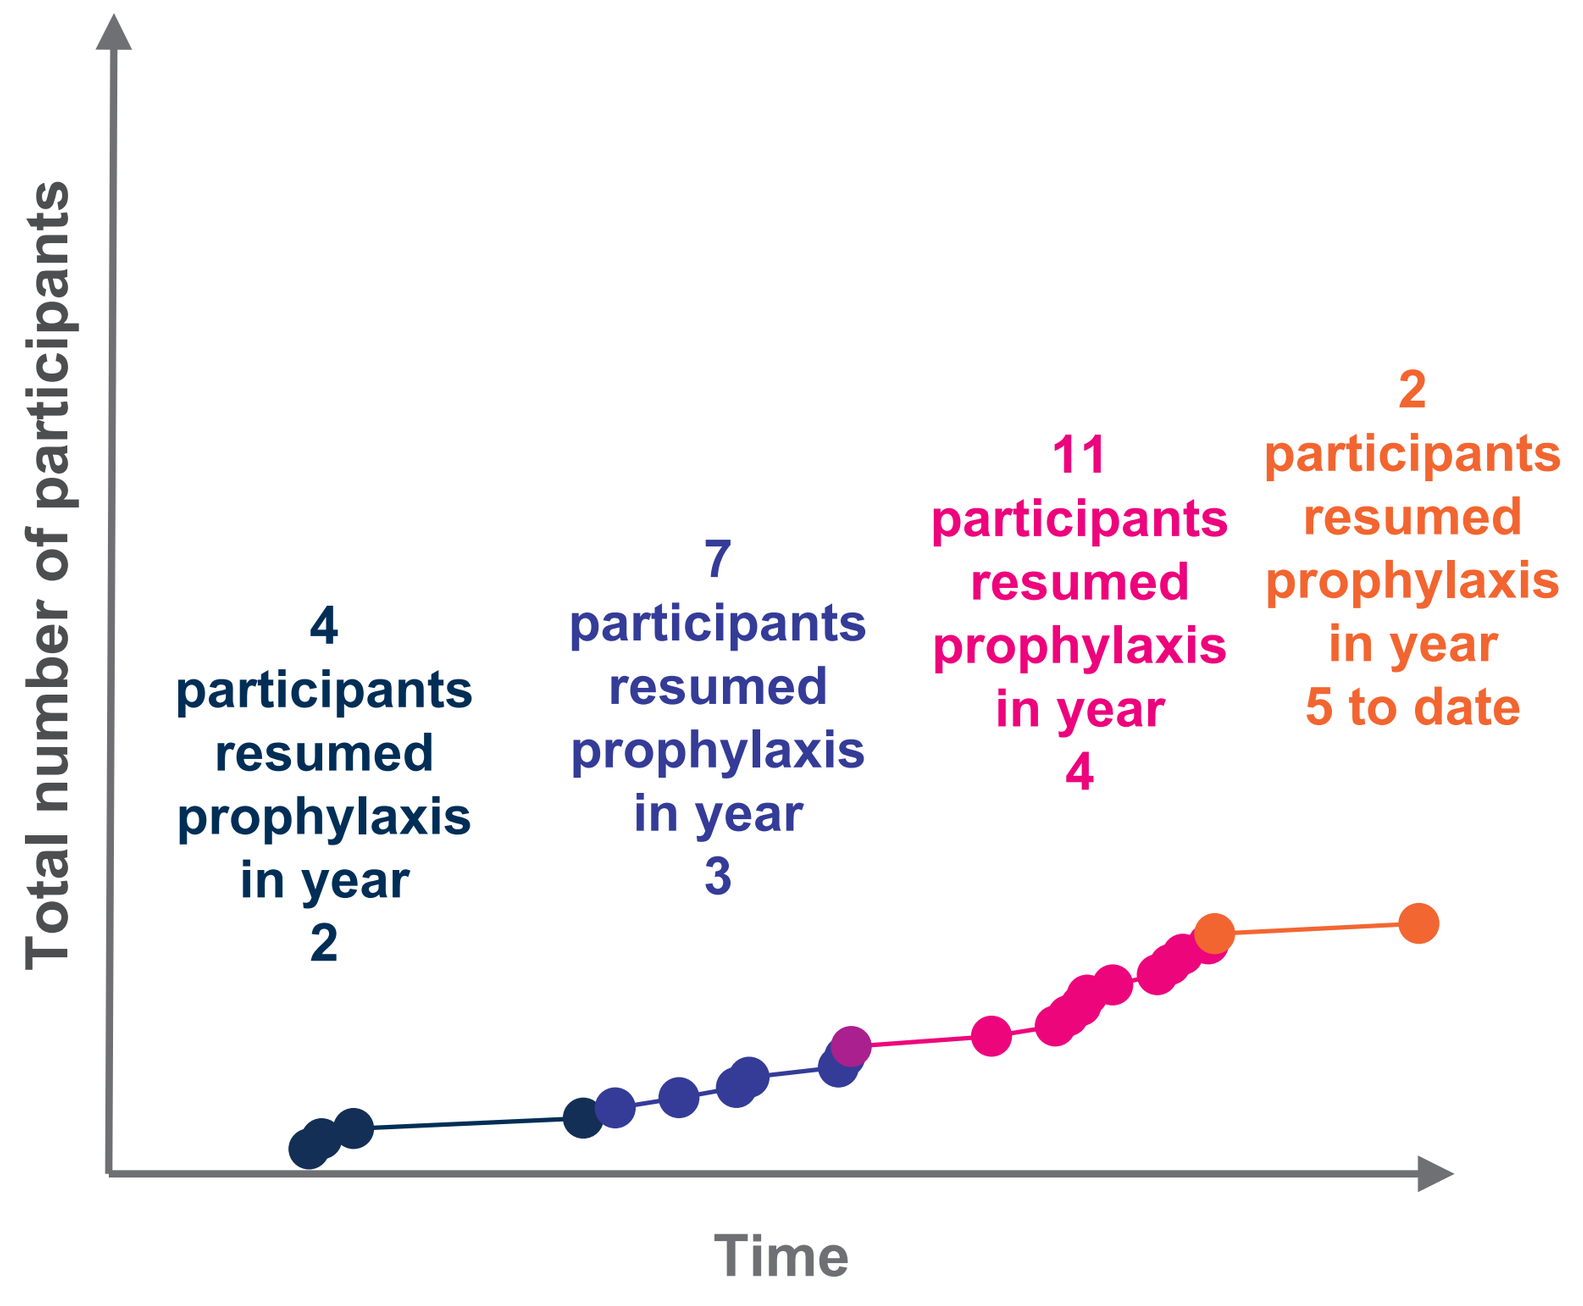
Total number of participants
Time
4 participants resumed prophylaxis in year 2
7 participants resumed prophylaxis in year 3
11 participants resumed prophylaxis in year 4
2 participants resumed prophylaxis in year 5 to date

110 participants remain off prophylaxis. Of these, the evaluable participants had **92%** fewer treated bleeds per year after receiving valoctocogene roxaparvovec compared with before

24 participants have returned to receiving regular prophylaxis with either FVIII concentrate or *emicizumab*. Of these, the evaluable participants had **21%** fewer treated bleeds per year from the time they received valoctocogene roxaparvovec to when they resumed prophylaxis compared with before

The decision to resume prophylaxis is part of a shared decision-making process between patients and their physicians and includes many individual factors, such as FVIII activity, bleeds, and physical activity level

FVIII levels over time

Average one-stage FVIII levels were **27%** at the end of year 4

For 17 participants who received valoctocogene roxaparvovec at least 5 years ago, average FVIII levels were **26%**

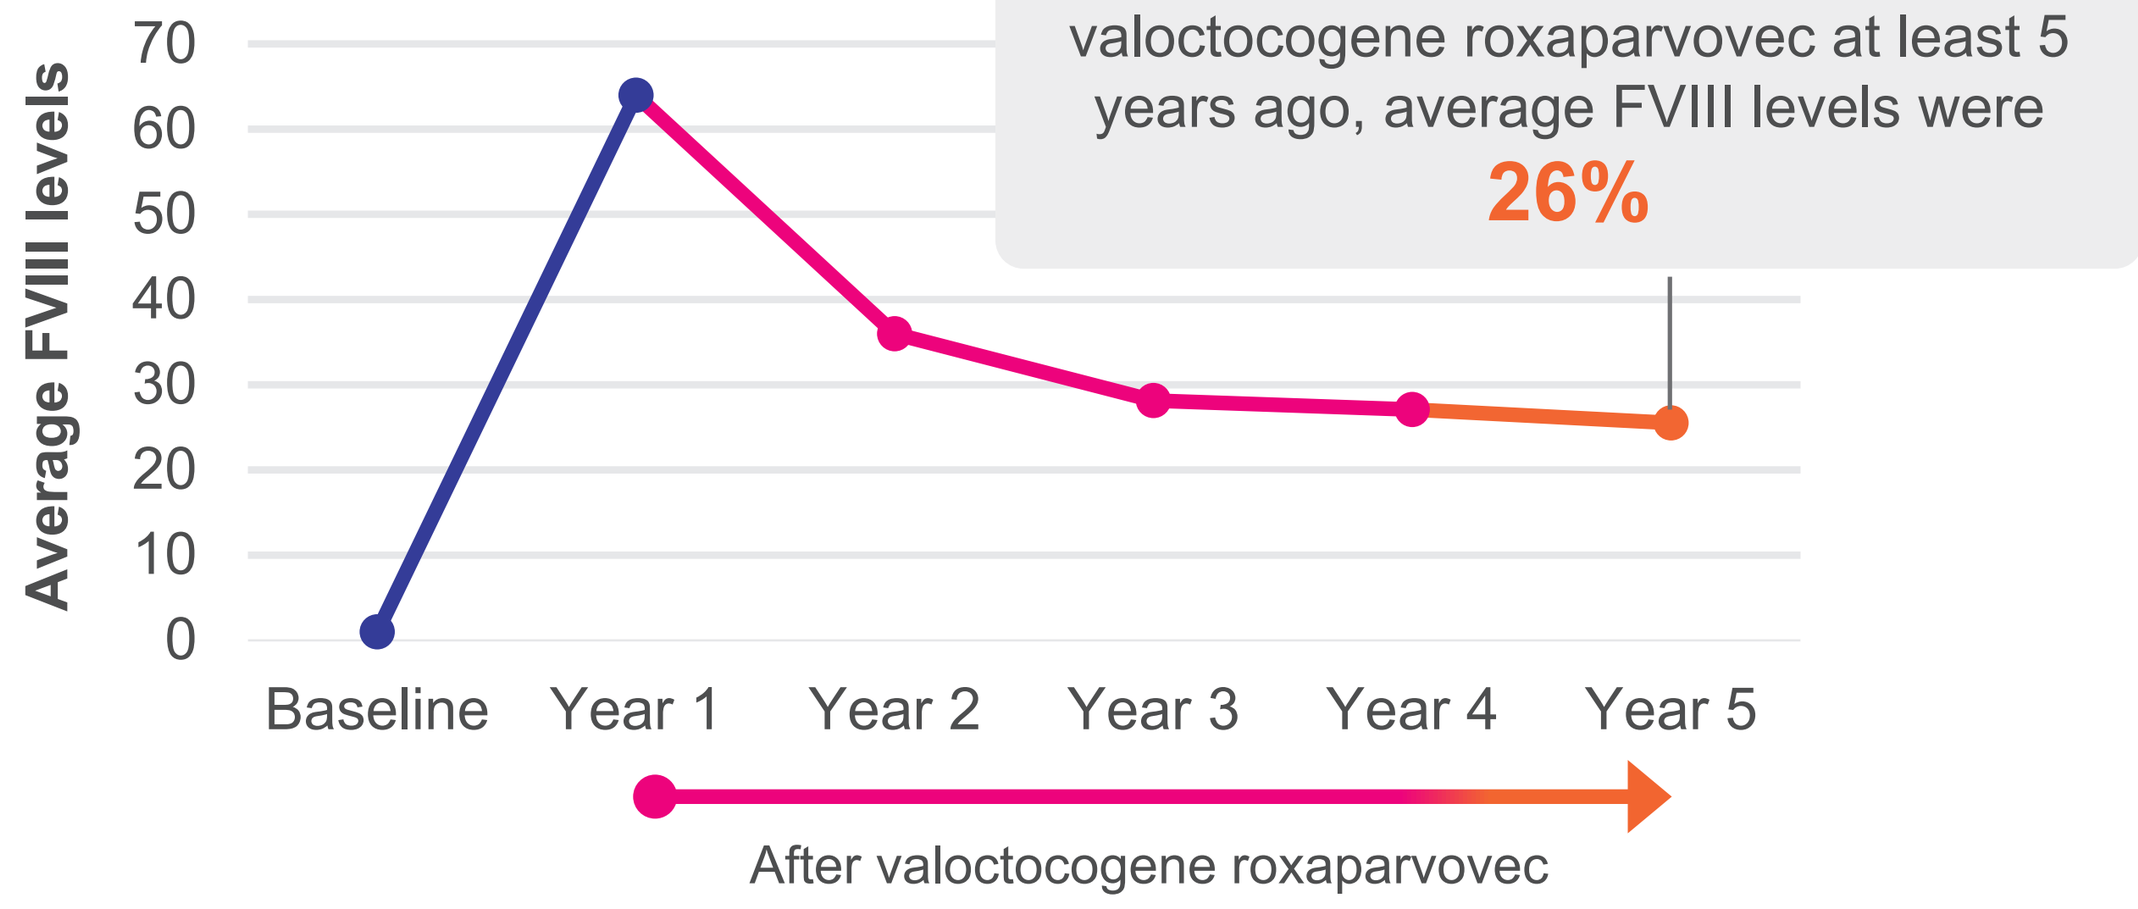
Average FVIII levels
70
60
50
40
30
20
10
0
Baseline
Year 1
Year 2
Year 3
Year 4
Year 5
After valoctocogene roxaparvovec

The FVIII levels reported here are based on the **one-stage assay**.
If your center uses the chromogenic assay, please note that those values tend to be ~1.5 times lower

FVIII levels at the end of year 4

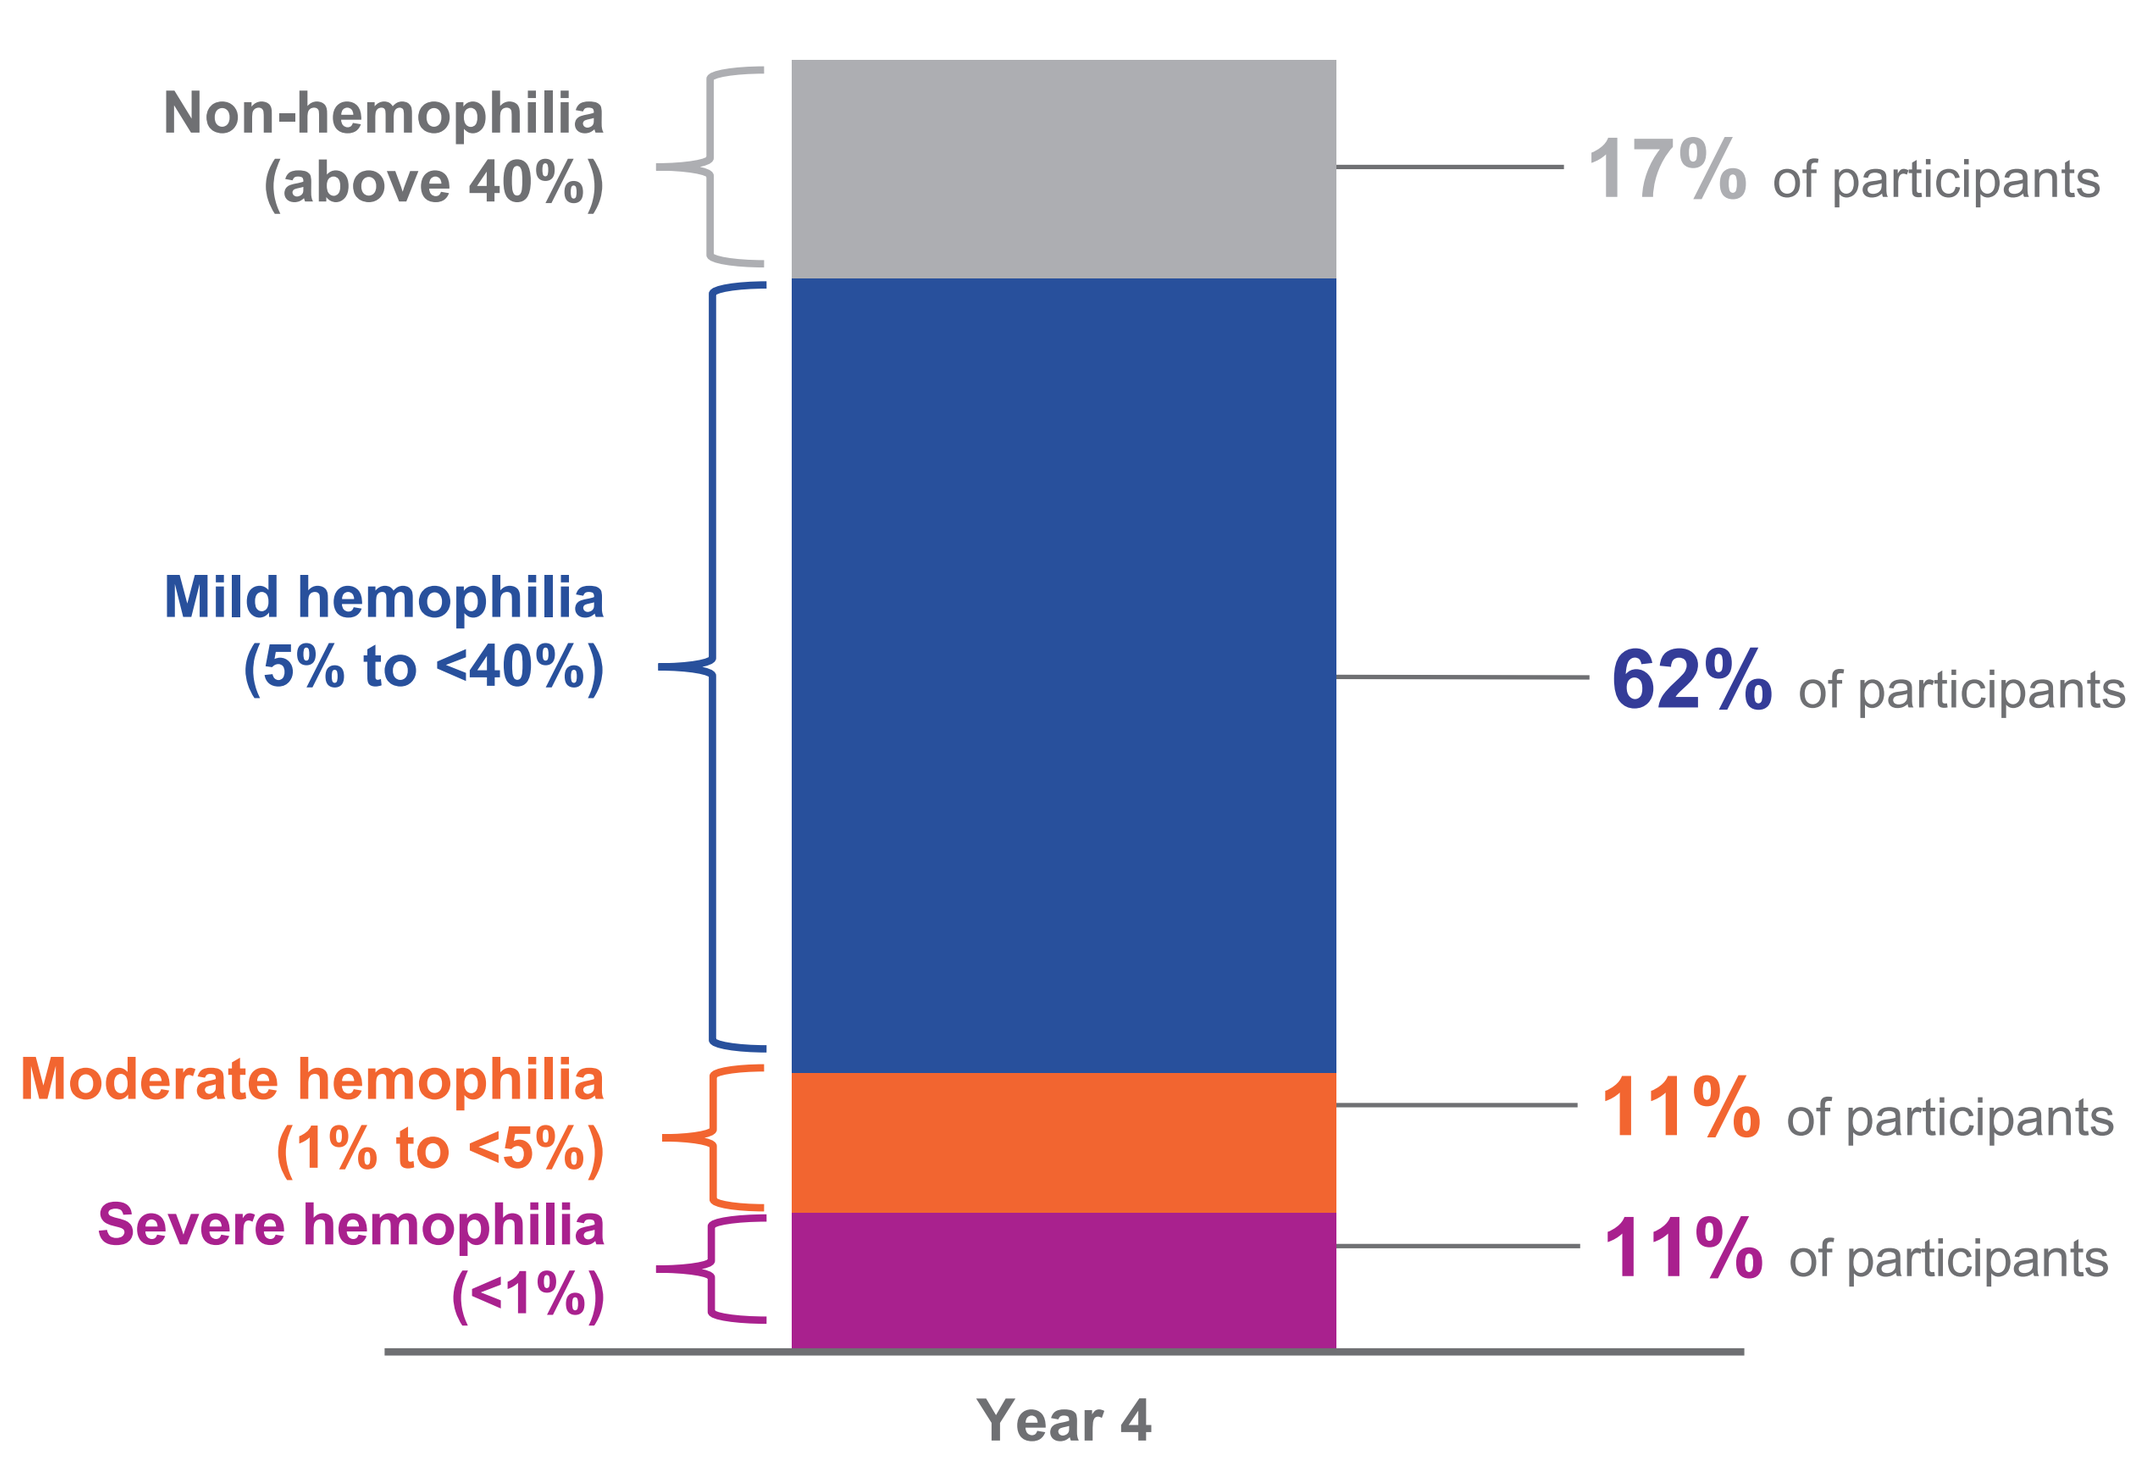
FVIII levels:
At the end of year 4:
Non-hemophilia (above 40%)
17% of participants
Mild hemophilia (5% to <40%)
62% of participants
Moderate hemophilia (1% to <5%)
11% of participants
Severe hemophilia (<1%)
11% of participants
Year 4

What were the safety results?

ALT elevation can be a sign of liver inflammation. Since valoctocogene roxaparvovec targets the FVIII instructions to the liver, ALT elevations were tracked closely in GENEr8-1

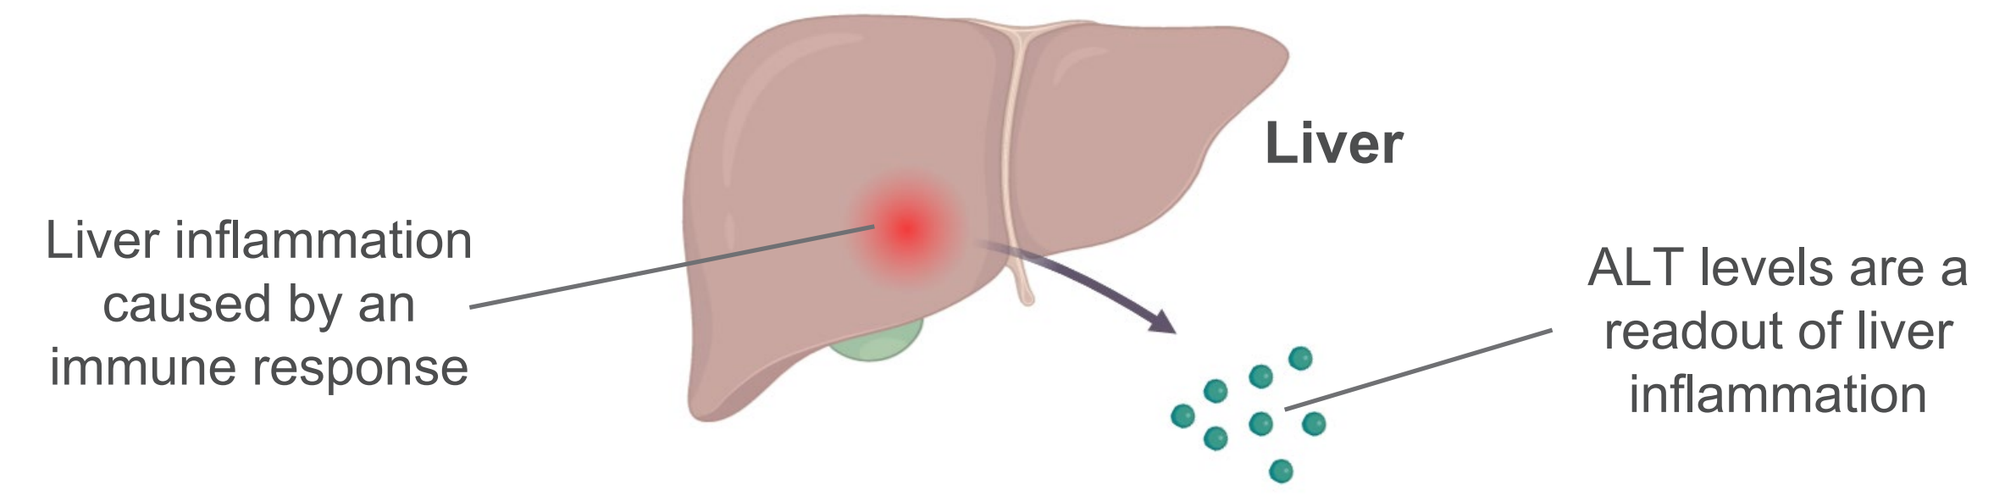
Liver
Liver inflammation caused by an immune response
ALT levels are a readout of liver inflammation

ALT elevations

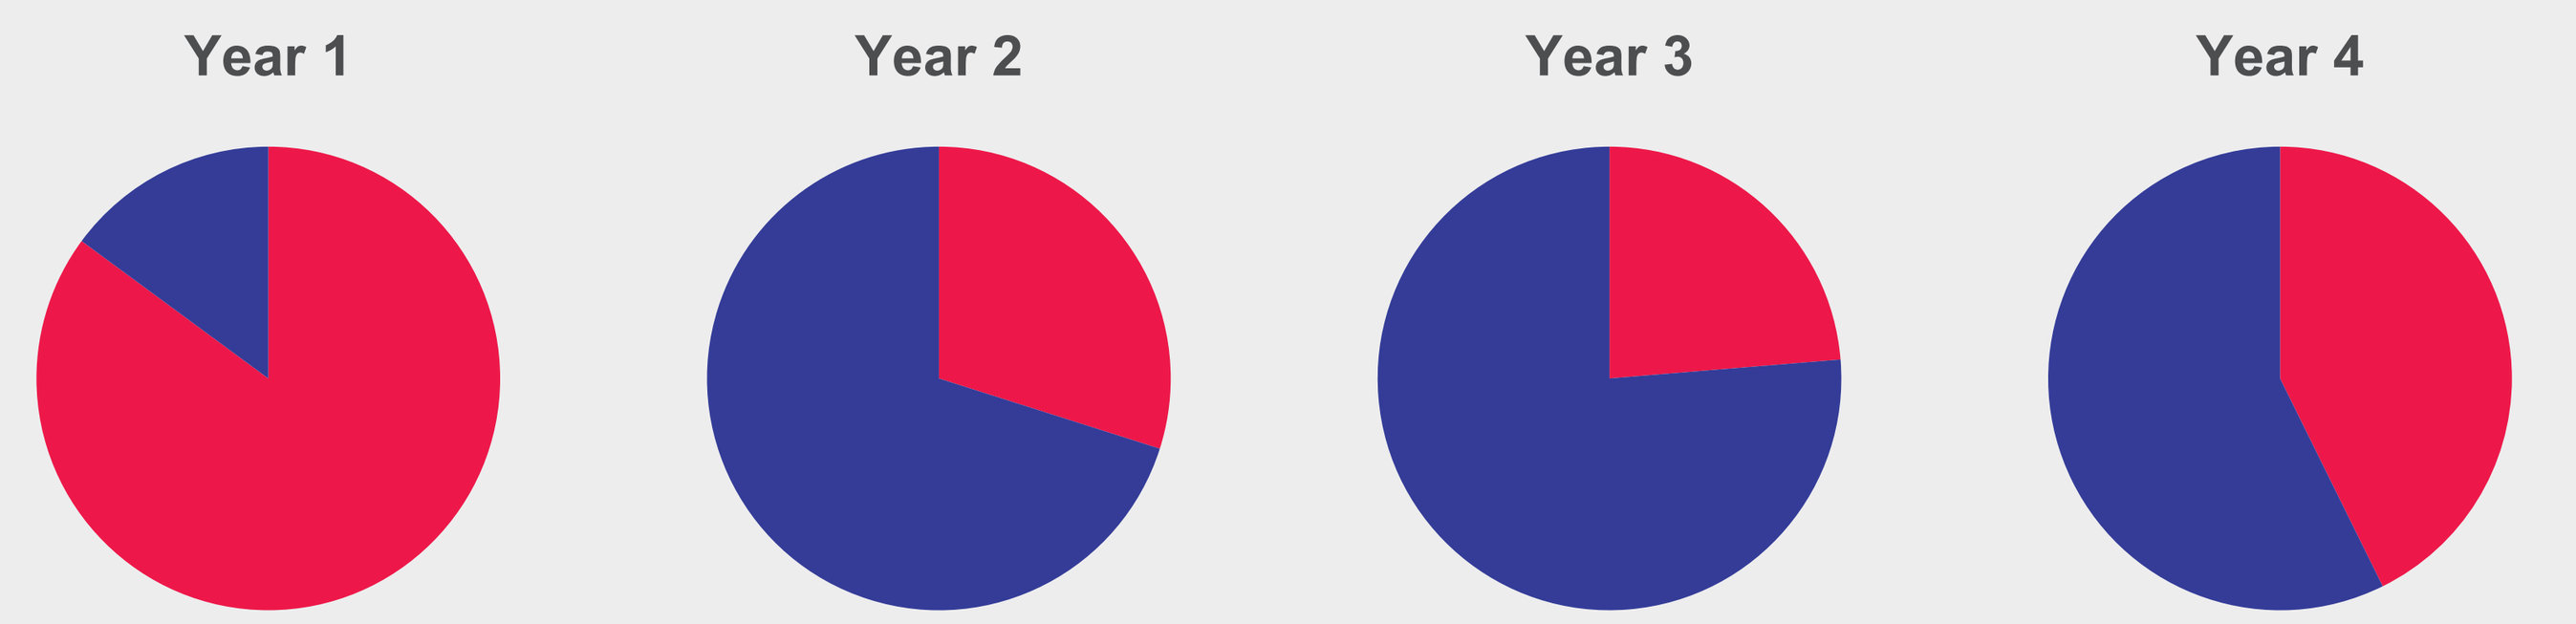
Year 1
Year 2
Year 3
Year 4

Year 1	Year 2	Year 3	Year 4
85% of participants experienced an ALT elevation	**30%** of participants experienced an ALT elevation	**24%** of participants experienced an ALT elevation	**43%** of participants experienced an ALT elevation

- During year 4, **ALT elevations occurred in 43% of participants; most were mild, and none were treatment-related**. As an extra precaution, a low threshold for identifying ALT elevation was chosen
- Overall, no participants developed inhibitors, treatment-related malignancies, or dangerous blood clots from too-high FVIII
- Other common adverse events were **headache** and **joint pain,** which occurred in fewer participants during year 4 compared with earlier years. **Nausea** also occurred in consistently fewer participants in year 4 compared with year 1

What do the results mean?

- A single infusion of valoctocogene roxaparvovec continued to provide **increased protection from bleeding** compared with regular FVIII prophylaxis after the fourth year
- **No new safety concerns** were found in the fourth year of the trial
- Overall, the **risk-benefit balance was unchanged** after the fourth year of the trial

Glossary

Adeno-associated virus: a virus that does not cause disease in humans. For valoctocogene roxaparvovec, the virus has been engineered to remove its ability to replicate and repurposed to carry genetic instructions for FVIII

Adverse event: health-related events that occur after a participant receives a treatment. Not all adverse events are related to the study treatment and may be incidental

ALT (alanine aminotransferase): an enzyme (a type of protein) that is commonly monitored by doctors as a sign of liver inflammation

Efficacy: in clinical trials, efficacy is the ability of a treatment to produce the desired effect

Emicizumab: an antibody that is designed to mimic the ability of FVIII to clot blood; emicizumab is an approved therapy for hemophilia A that is used for prophylaxis against bleeding

FVIII (factor VIII): a critical protein for blood clotting, FVIII is produced by reading the instructions in the *F8* gene

Phase 3 clinical trial: after finding the most effective safe dose in phase 1 and 2 trials, phase 3 trials use a larger group of people to determine whether the benefits of the treatment outweigh the risks and whether the treatment works better than a currently approved treatment

Promoter: a part of the vector genome that directs certain types of cells to read the instructions. In the case of valoctocogene roxaparvovec, the promoter is designed to allow only liver cells to produce FVIII based on the instructions

Prophylaxis: a treatment aimed at preventing an event from occurring

Vector: a means to deliver a vector genome to the desired cells within the body. In the case of valoctocogene roxaparvovec, the vector is the capsid ("shell") of the AAV

Vector genome: the genetic material that the gene therapy delivers to the body to produce the desired benefit. Vector genomes are the units in which a gene therapy dose is measured

Funding

GENEr8-1 is funded by BioMarin Pharmaceutical Inc.

Acknowledgements

We thank all trial participants, investigators, and site staff. Medical writing support was provided by Amin Ghane, PhD, of AlphaBioCom, a Red Nucleus company, and funded by BioMarin. Project management support was provided by Gillian Clague, CMPP, of BioMarin.

Financial disclosures

ADL has received research funding from BioMarin and Pfizer and has participated in advisory boards for BioMarin, CSL, Pfizer, Sanofi, and Sobi. **JM** has received research funding from BioMarin, Catalyst, Roche, Novo Nordisk, Pfizer, Sandoz, Sanofi, and Spark Therapeutics. **ES** has received travel grants from CSL Behring and Novo Nordisk. **DVQ** has served on advisory boards and speaker bureaus or as a consultant for Bayer, BioMarin, Genentech, Novo Nordisk, Octapharma, Sanofi, Takeda, and uniQure. **AG** has served on advisory boards for BioMarin, Genentech, Hema Biologics, Pfizer, Sanofi Genzyme, and uniQure; served on speaker bureaus for BioMarin and Sanofi Genzyme; and received research funding from Freeline, Genentech, Pfizer, Sangamo, Spark Therapeutics, and uniQure. **NSK** has received consulting fees from Centessa, Novo Nordisk, and Pfizer. **SWP** has served as a consultant for ApcinteX, Bayer, BioMarin, CSL Behring, Equilibra Bioscience, GeneVentiv, HEMA Biologics, LFB, Novo Nordisk, Pfizer, Regeneron, Roche, Sanofi, Siemens, Spark, Takeda, and uniQure. **PJL** is an employee of Believe Ltd. **HY** and **CR** are employees and stockholders of BioMarin. **MCO** has participated in advisory boards for Bayer, BioMarin, Pfizer, Sanofi, and Takeda and received honoraria from BioMarin, Biotest, Novo Nordisk, Pfizer, Roche, Takeda, and Sanofi. **FPC** and **AR** have no conflicts to declare.

Presented at the 76th Bleeding Disorders Conference 2024: September 12–14, 2024, Atlanta, GA, USA

©2024 BioMarin Pharmaceutical Inc. All rights reserved.