Criteria for defining inadequate response to growth hormone in Noonan syndrome, Turner syndrome, and SHOX deficiency

Poster #
Sun-504

Ron Rosenfeld[1], Andrew Dauber[2], Klaus Mohnike[3], Lawrence Silverman[4], Andrea Low[5], Ming Sheng[6], Ian Sabir[6], Neil Lyon[6], Alice Huntsman Labed[6]

[1]Department of Pediatrics, Oregon Health & Science University, Portland, OR, USA; [2]Children's National Hospital, Washington, DC, USA; [3]Otto-von-Guericke-Universität, Magdeburg, Germany; [4]Division of Pediatric Endocrinology, Goryeb Children's Hospital, Atlantic Health System, Morristown, NJ, USA; [5]BioMarin Pharmaceutical Inc., Novato, CA, USA; [6]BioMarin (UK) Ltd, London, UK

Background

Inadequate growth and human growth hormone therapy

- Human growth hormone (hGH) therapy is standard in many countries for short stature conditions, including Noonan syndrome, Turner syndrome, and short stature homeobox-containing gene deficiency disorders (SHOX-Ds)[1]
- Unfortunately, many children eventually become nonresponsive to hGH therapy[2,3]
- Despite its importance, the criteria for inadequate growth on or after hGH use are poorly defined, and the lack of a definition impacts decisions to initiate and, more importantly, continue treatment

Introduction to CANOPY NS, TS, SHOX-D-2 study

- Turner syndrome, Noonan syndrome, and SHOX-Ds are associated with excess extracellular signal-regulated kinase (ERK) signaling and characterized by short stature[4–6]
- Vosoritide, an analogue of the master growth regulator C-type natriuretic peptide, is an approved targeted therapy for achondroplasia that stimulates endochondral bone growth by reducing ERK activity (**Figure 1**)[7]

Figure 1. ERK1/2 in normal chondrocyte proliferation and differentiation

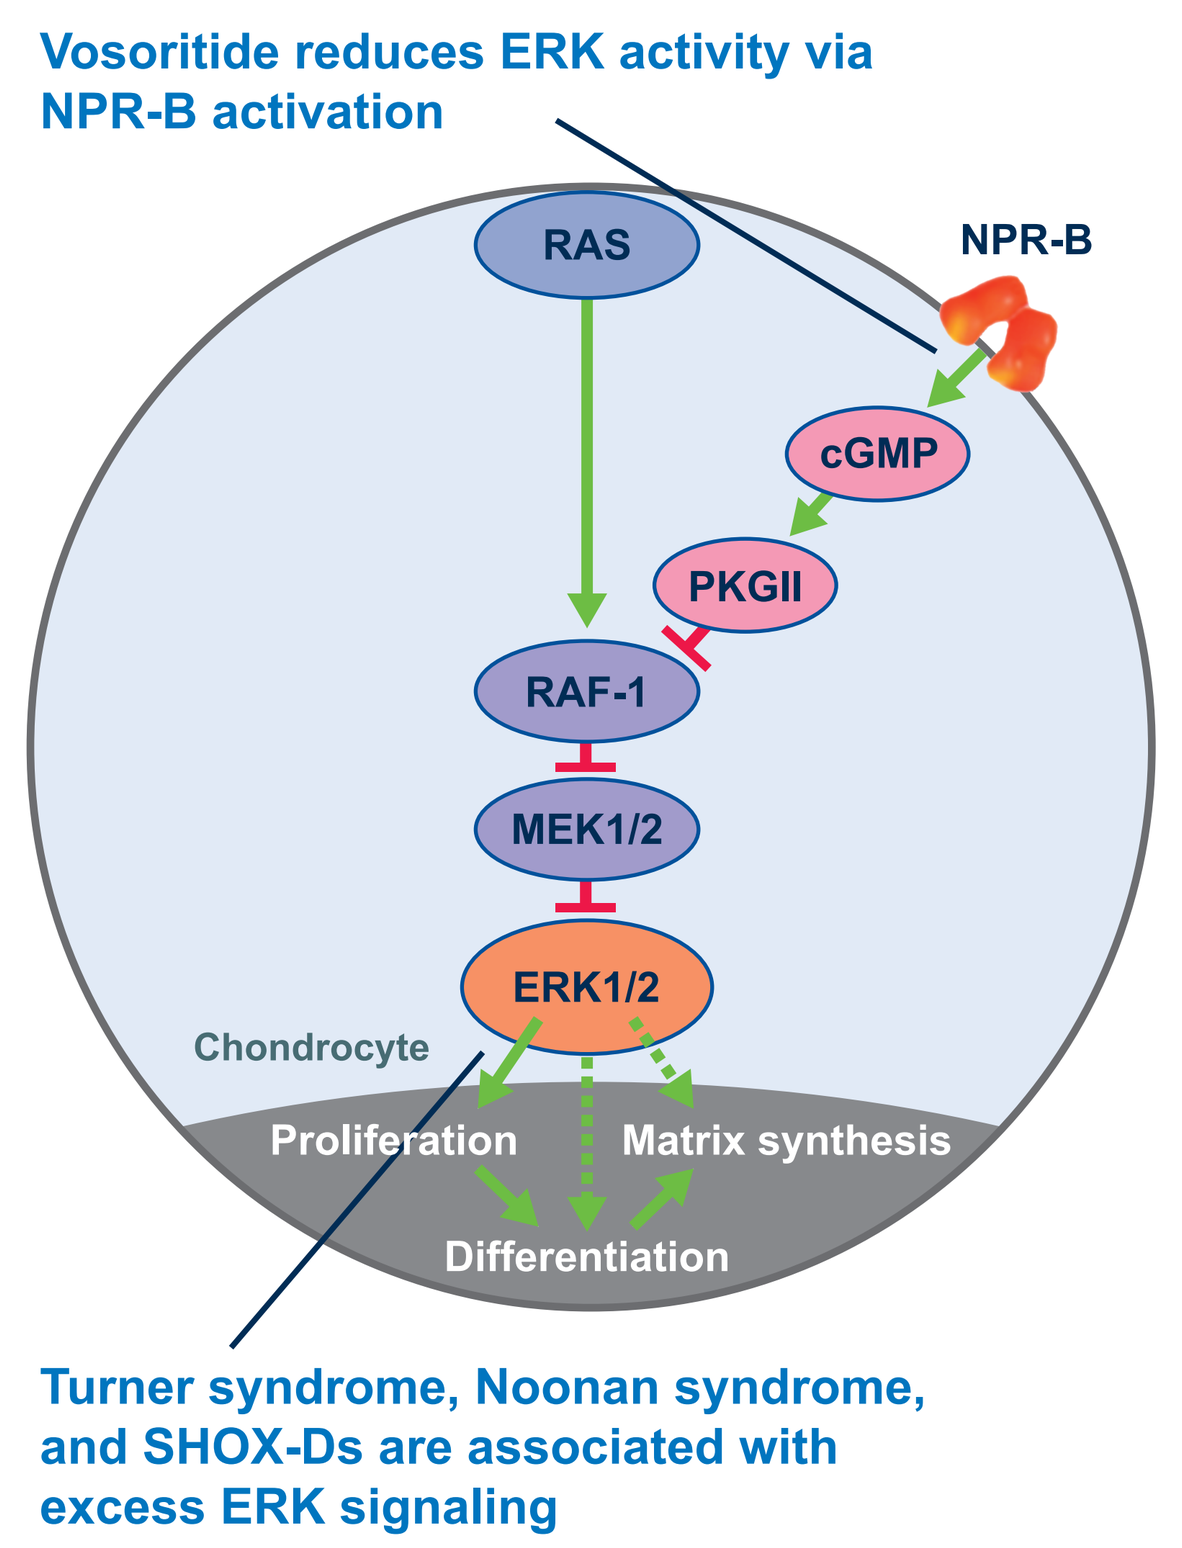
Vosoritide reduces ERK activity via NPR-B activation
RAS
NPR-B
cGMP
PKGII
RAF-1
MEK1/2
ERK1/2
Chondrocyte
Proliferation
Matrix synthesis
Differentiation
Turner syndrome, Noonan syndrome, and SHOX-Ds are associated with excess ERK signaling

cGMP, cyclic guanosine monophosphate; ERK, extracellular signal-regulated kinase; NPR-B, atrial natriuretic factor receptor B; PKGII, type II cGMP-dependent protein kinase.

- CANOPY NS, TS, SHOX-D-2 (111-211; NCT06668805) is a phase 2 randomized, multicenter basket study designed to determine the therapeutic dose of vosoritide in children with genetically confirmed Noonan syndrome, Turner syndrome, or SHOX-D with inadequate growth on or after hGH use for a phase 3 confirmatory study, CANOPY NS, TS, SHOX-D-3 (111-306)
- CANOPY NS, TS, SHOX-D-2 represents the first use case of our established definition of inadequate growth on or after hGH use

Objective

- To establish a standard definition for inadequate growth on or after hGH use and to demonstrate its use in a planned study

Methods

Identification of reference data sources for AGV → Determination of AGV over time in average-stature children → Define inadequate growth on or after hGH use based on comparison of AGV data → Planned use case of inadequate growth on or after hGH use definition

AGV, annualized growth velocity; hGH, human growth hormone.

Results

Determination of inadequate growth on or after hGH use

- US Centers for Disease Control and Prevention (CDC),[8] World Health Organization (WHO),[9] and German height chart data (Neuhauser)[10] were used to determine annualized growth velocity (AGV) by deriving the difference in median height assessments among children 12 months apart in age. Pre-derived mean (standard deviation [SD]) for AGV by age and sex (with 95% confidence intervals [CIs]) was available from a Swiss reference (Prader et al)[11]
- To assess the robustness of the approach, we charted the AGV in average-stature male (**Figure 2A**) and female (**Figure 2B**) children aged 3 to 20 years using all 4 data sources
- In general, AGV followed a similar trend in all 4 data sources and was especially consistent in males

Figure 2. AGV in average-stature children

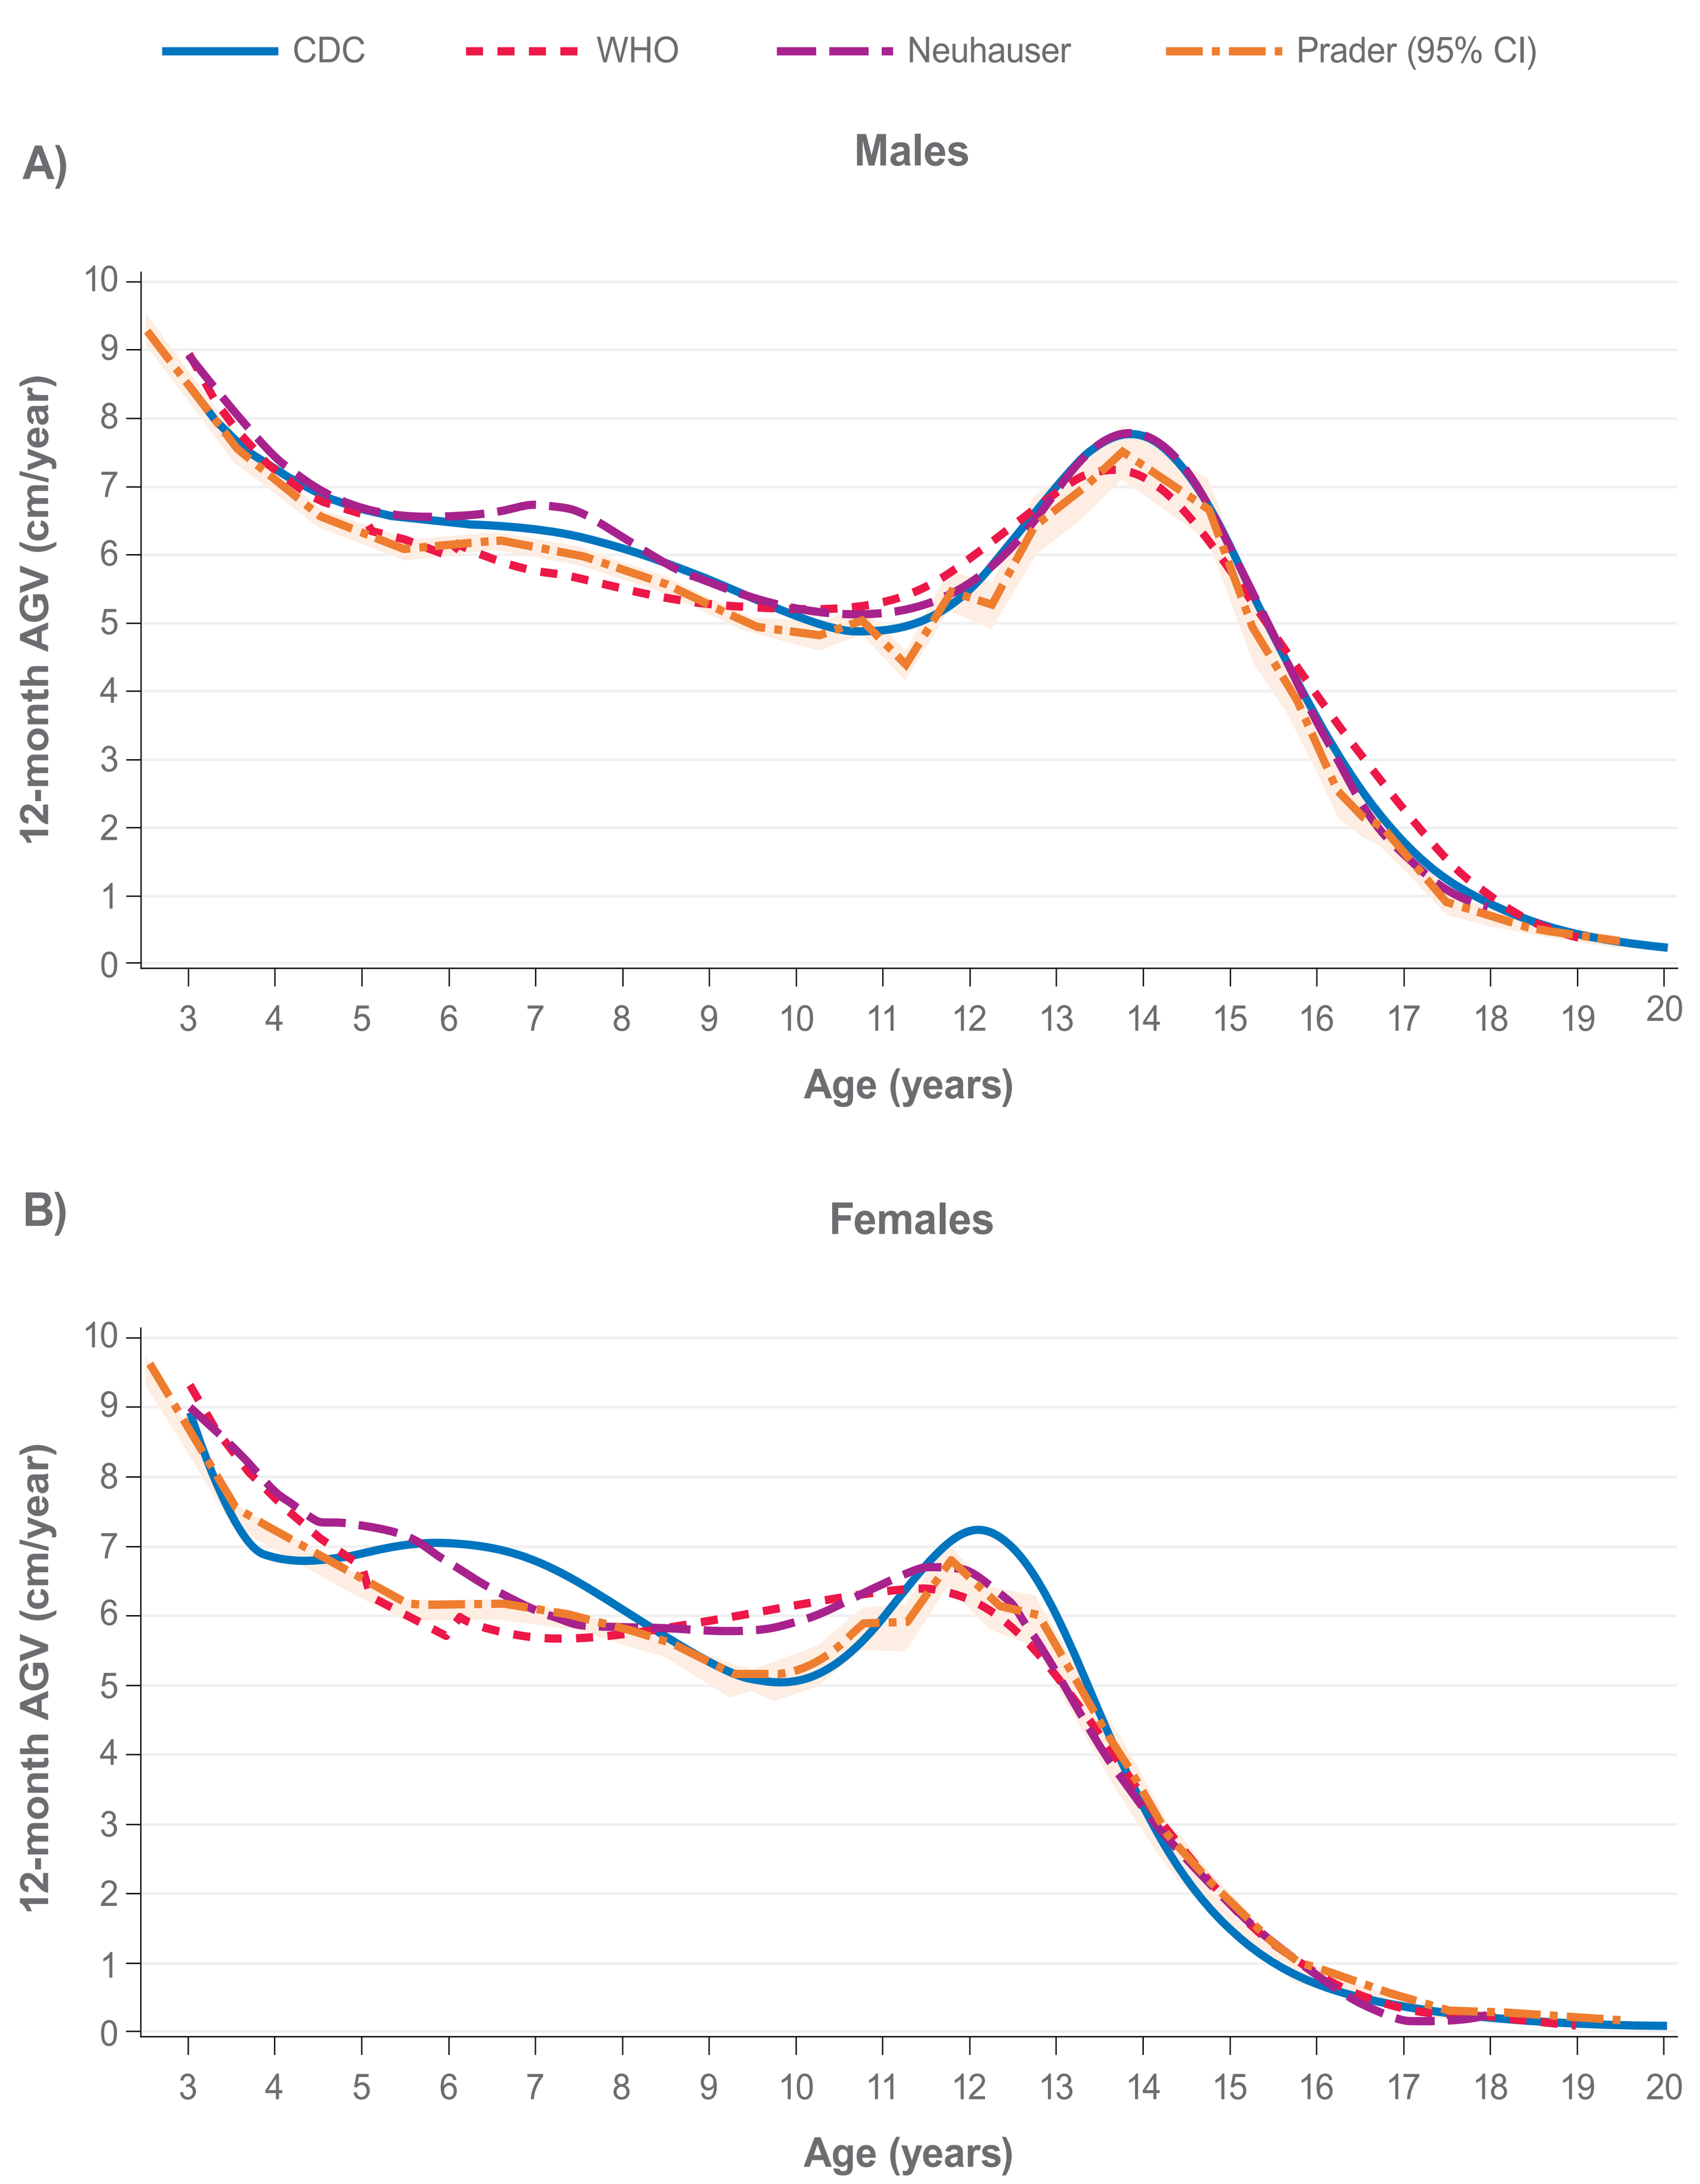
CDC
WHO
Neuhauser
Prader (95% CI)
A)
Males
B)
Females
12-month AGV (cm/year)
Age (years)

AGV from CDC,[8] WHO,[9] and Neuhauser[10] data are derived by subtracting the median height from 12 months prior to the median heights at the plotted time points. AGV from Prader et al[11] are reported directly from the data; 95% CIs are derived using the t-distribution.
AGV, annualized growth velocity; CDC, US Centers for Disease Control and Prevention; CI, confidence interval; WHO, World Health Organization.

- Using this data, we established a standard definition of inadequate growth on or after hGH use. The definition ensures that children with inadequate growth are no longer displaying catch-up growth to their average-stature peers

INADEQUATE GROWTH ON OR AFTER hGH USE: An AGV below that of the age- and sex-matched average-stature AGV determined from the CDC growth charts

Application in CANOPY NS, TS, SHOX-D-2 study (111-211; NCT06668805)*

- The newly established definition was used for the first time in the ongoing phase 2 CANOPY NS, TS, SHOX-D-2 study, designed to determine the therapeutic dose of vosoritide in children with Noonan syndrome, Turner syndrome, or SHOX-D with a history of hGH use or current use

Participants

- 54 children with genetically confirmed Turner syndrome, Noonan syndrome, or SHOX-D with a history of hGH use or current use
- Aged ≥3 to <11 years (females) or <12 years (males) and prepubertal
- Height Z-score ≤−1.28 SDs from the CDC average-stature references
- Previously or currently receiving treatment with hGH

Participants must currently demonstrate inadequate growth on or after hGH use, defined as AGV below that of average-stature children based on CDC data (**Figure 3**)

Figure 3. AGV in females and males of average stature based on CDC data[a]

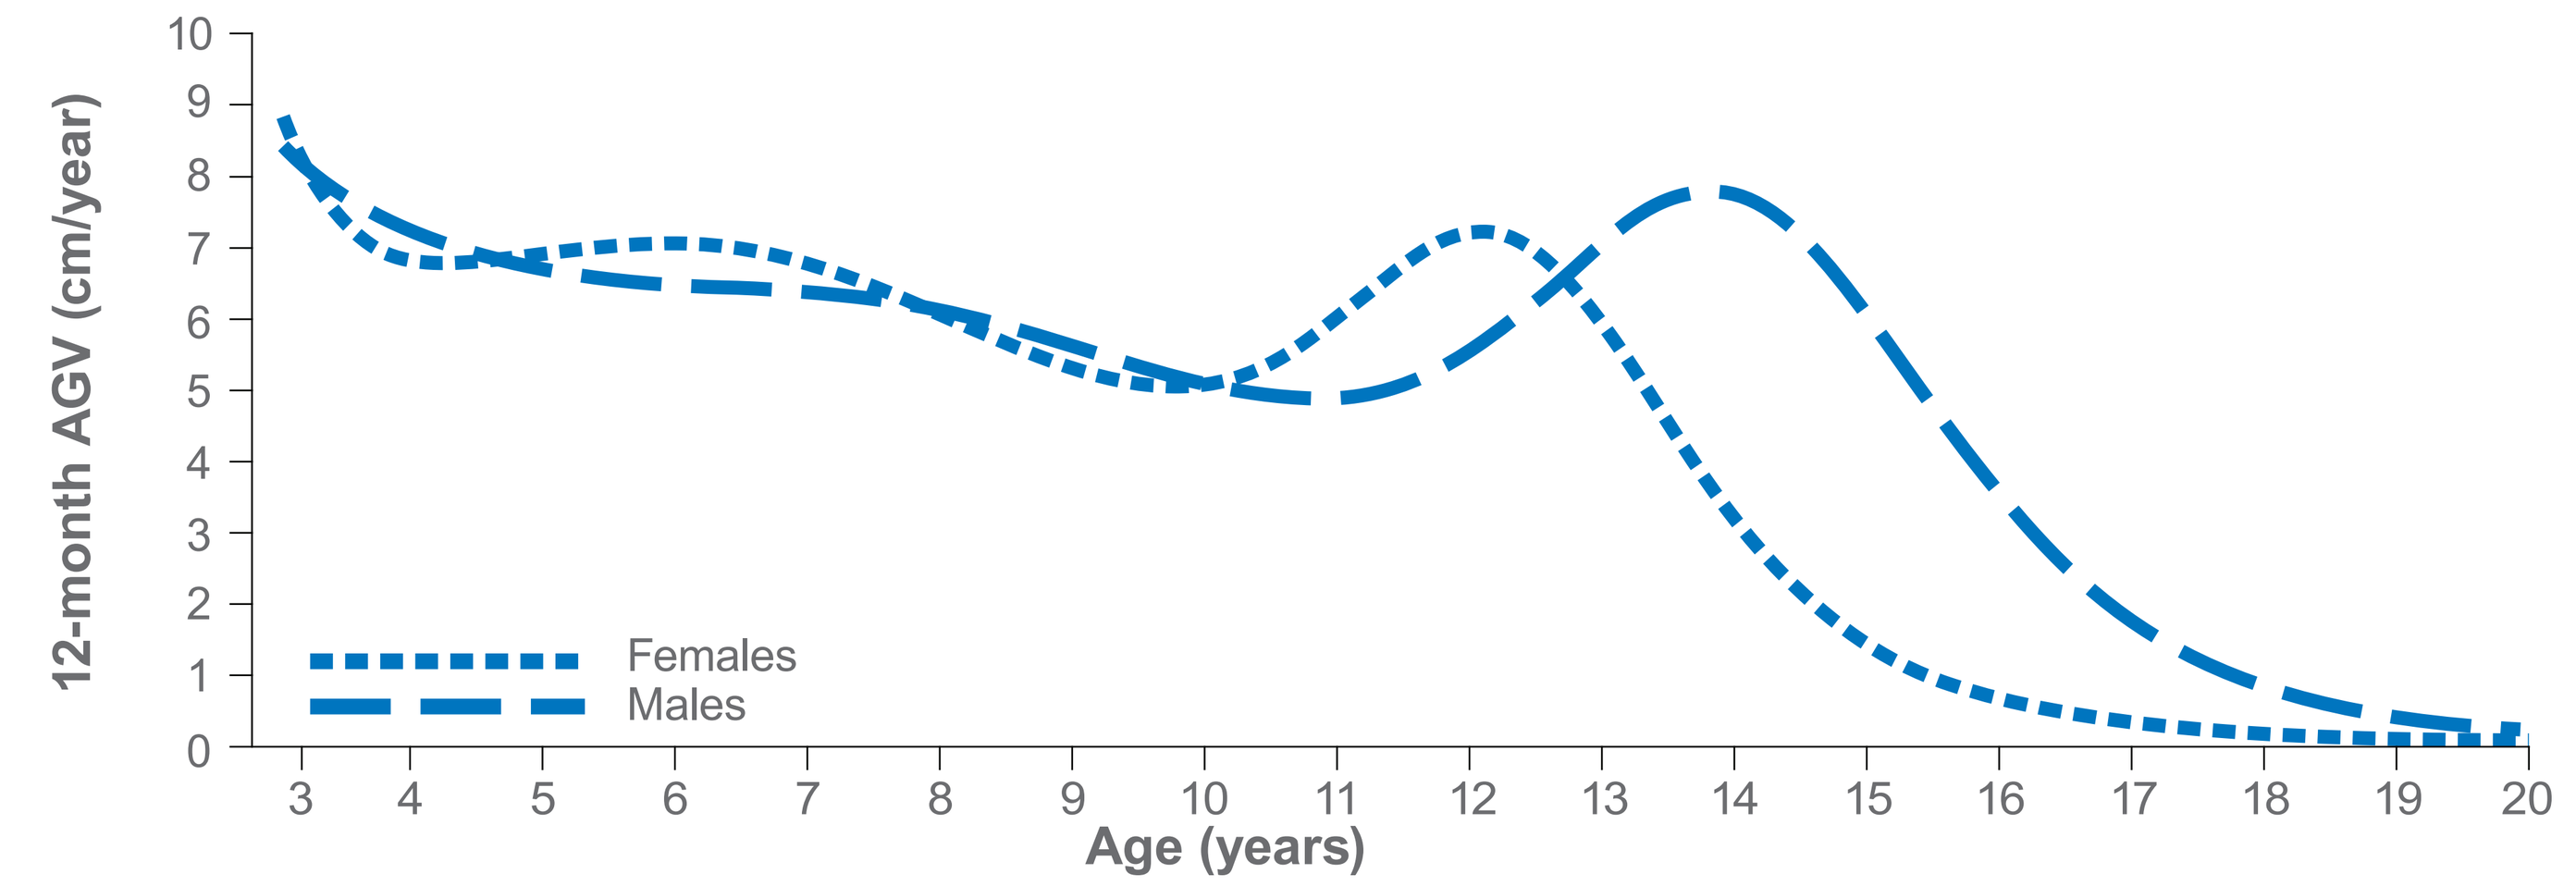
12-month AGV (cm/year)
Age (years)
Females
Males

[a]CDC height chart data were used to determine AGV by deriving the difference in median height assessments 12 months apart in age.
AGV, annualized growth velocity; CDC, US Centers for Disease Control and Prevention.

Study design

Dose-finding phase	Long-term phase
6 months	*Until near-final adult height[a]*
After a 6-month baseline growth assessment phase while on standard care, participants will be randomized to	*The therapeutic dose of vosoritide will be determined after the dose-finding phase for vosoritide participants*
7.5 µg/kg/day vosoritide; 15.0 µg/kg/day vosoritide; 22.5 µg/kg/day vosoritide	→ Therapeutic dose of vosoritide

[a]Near-final adult height is defined as decreased growth velocity (AGV <1.5 cm/year) assessed over a period of at least 6 months and growth plate fusion.
AGV, annualized growth velocity; hGH, human growth hormone; SHOX-D, short stature homeobox-containing gene deficiency disorder.

*Protocol amendments ongoing.

Study sites

- CANOPY NS, TS, SHOX-D-2 is recruiting at sites in the US, Canada, Turkey, Italy, Germany, Spain, France, and Australia

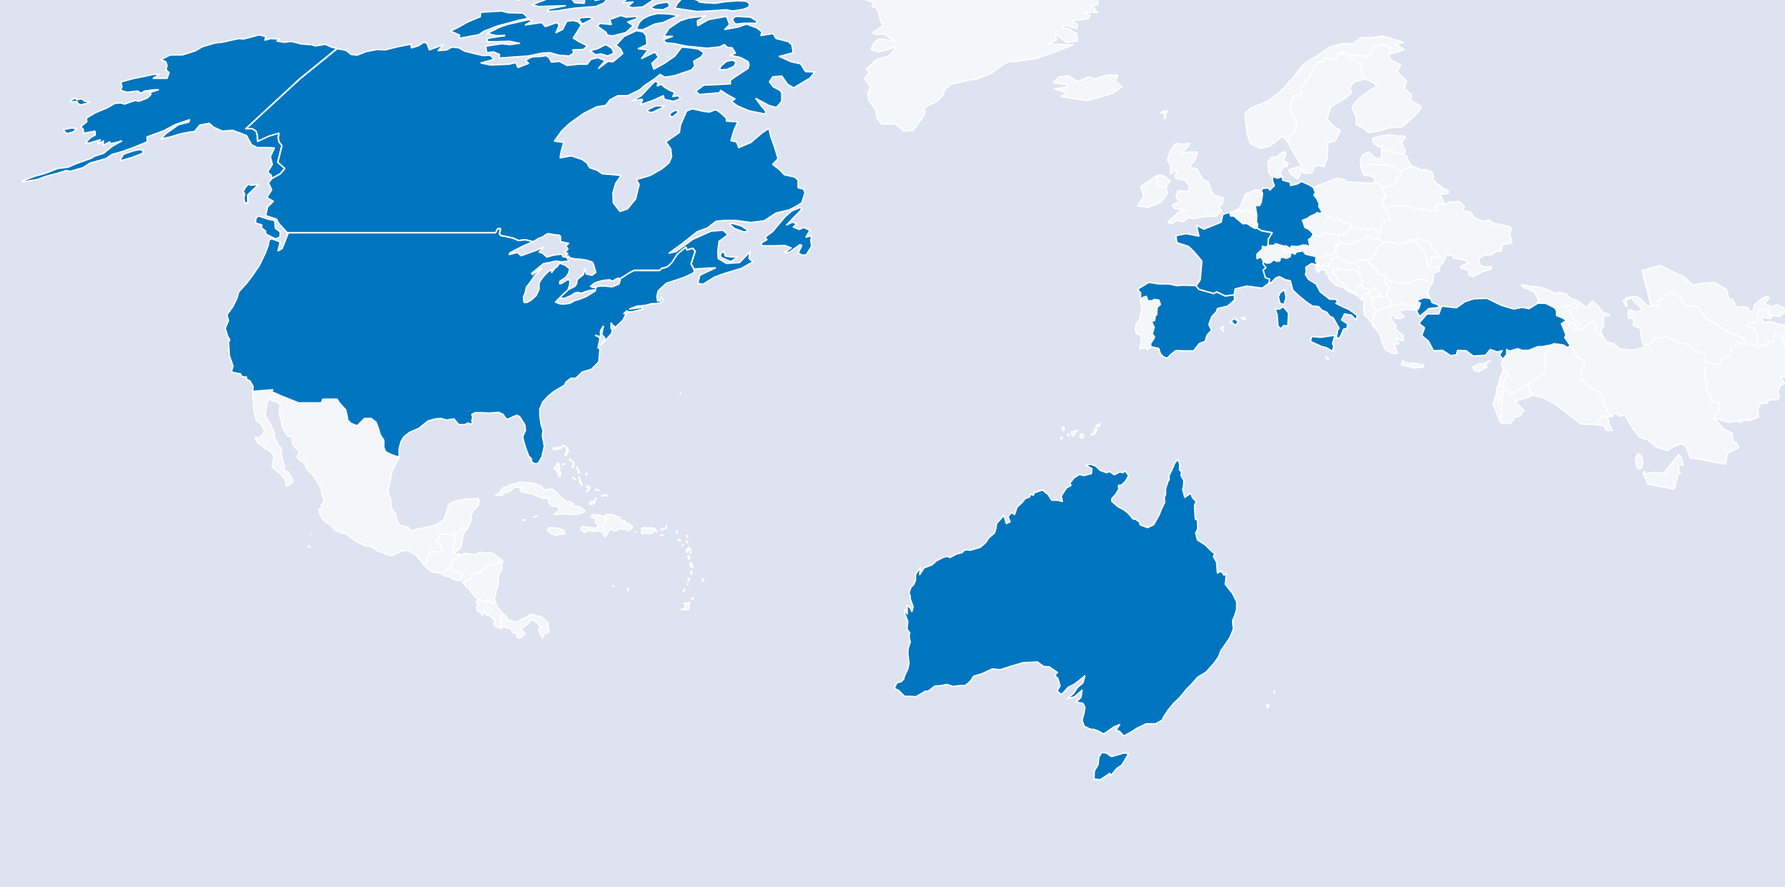

Endpoints

Endpoints	Group	Time point
Change from baseline (on or after hGH use) in AGV	3 vosoritide doses	6 months[a]
Change from baseline (on or after hGH use) in height, height Z-score,[b] and AGV	Therapeutic vosoritide dose	Closure of growth plates
Safety	*Adverse events of special interest are fracture, slipped capital femoral epiphysis, avascular necrosis or osteonecrosis, and symptomatic hypotension*	

[a]An interim analysis will occur when ≥50% of participants in each group have 6 months of follow-up. [b]Based on CDC growth charts.
AGV, annualized growth velocity; CDC, US Centers for Disease Control and Prevention; hGH, human growth hormone.

CANOPY NS,TS, SHOX-D-2
PHASE 2 STUDY

Conclusions

- Inadequate growth while on or after hGH therapy, irrespective of timing, can be defined in terms of expected growth in age- and sex-matched reference populations
- These criteria will enable optimized treatment decisions and clinical trial design for children with short stature receiving hGH therapy
- Here we also present the study design of the phase 2 study CANOPY NS, TS, SHOX-D-2 (111-211), a use-case example of the newly established definition of inadequate growth on or after hGH use

References

1. Deodati A, et al. *J Clin Res Pediatr Endocrinol*. 2017;9(suppl 2):23–32. **2.** Ross J, et al. *J Endocr Soc*. 2023;7(5):bvad026. **3.** Wu X, et al. *Mol Genet Genomic Med*. 2023;11(11):e2266. **4.** Roberts AE, et al. *Lancet*. 2013;381:333–42. **5.** Marchini A, et al. *Endo Rev*. 2016;37:417–48. **6.** Gravholt CH, et al. *Nat Rev Endocrinol*. 2019;15:601–14. **7.** European Medicines Agency. Voxzogo product information. Accessed February 28, 2025. https://www.ema.europa.eu/en/documents/product-information/voxzogo-epar-product-information_en.pdf. **8.** Kuczmarski RJ, et al. 2000 CDC growth charts for the United States: methods and development. Vital Health Stat 2002;11(246). Accessed April 25, 2025. https://www.cdc.gov/growthcharts/2000growthchart-us.pdf. **9.** World Health Organization. WHO child growth standards. November 11, 2006. Accessed April 25, 2025. https://www.who.int/publications/i/item/924154693X. **10.** Federal Health Reporting. Reference percentiles for anthropometric measures and blood pressure based on the German health interview and examination survey for children and adolescents 2003-2006 (KiGGS). Robert Koch Institute. 2016. Accessed April 25, 2025. https://edoc.rki.de/bitstream/handle/176904/3271/23s3ntHQtKbus.pdf?sequence=1. **11.** Prader A, et al. *Helv Paediatr Acta Suppl*. 1988;52(43):1–125.

Acknowledgments

Medical writing support was provided by Sanna Abbasi, PhD, of Red Nucleus, and funded by BioMarin Pharmaceutical Inc. Project management support was provided by Justin Potuzak, PhD, of BioMarin Pharmaceutical Inc.

Disclosures

RR and **AD** are consultants for BioMarin Pharmaceutical Inc. **AD** has received grant funding from BioMarin Pharmaceutical Inc. and is currently a primary investigator for a trial of vosoritide in children with genetic causes of short stature (NCT04219007). **KM** has received consulting payments from BioMarin Pharmaceutical Inc., Novo Nordisk, and QED Therapeutics; has participated as an investigator for BioMarin Pharmaceutical Inc.; and has received speaker fees and travel support from BioMarin Pharmaceutical Inc. and Novo Nordisk. **LS** is a consultant for BioMarin Pharmaceutical Inc., OPKO Health, and Pfizer and an investigator for BioMarin Pharmaceutical Inc. and Novo Nordisk. **AL**, **MS**, **IS**, **NL**, and **AHL** are employees and shareholders of BioMarin (UK) Ltd.

To view a copy of this poster, scan this QR code. Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission from the authors.

For more information on CANOPY NS, TS, SHOX-D-2, scan this QR code.

Presented at American Society for Bone and Mineral Research (ASBMR) annual meeting: September 5–8, 2025; Seattle, WA, USA

©2025 BioMarin Pharmaceutical Inc. All rights reserved.