Quantitative evaluation of hypophosphatemic rickets due to ENPP1 deficiency

Leanne Ward[1], David Weber[2], Frank Rutsch[3], Alix Bensacon[4], Klaus Dieterich[5], Thomas Perouse de Montclos[6], Suma Uday[7], Deborah Wenkert[8], Nagwa Wilson[9], Khaldoun Koujok[9], Kevin Smit[10], Stefan Jackowski[11], Maya Scharke[11], Diane Piper[12], Kurt C. Gunter[12], M. Zulf Mughal[13], Thomas Thacher[14]

[1]Children's Hospital of Eastern Ontario, University of Ottawa, ON, Canada; [2]Children's Hospital of Philadelphia, PA, United States; [3]Münster University Children's Hospital, Münster, Germany; [4]Hôpital Universitaire Necker Enfants Malades, Paris, France; [5]CHU Grenoble Alpes, La Tronche, France; [6]CHU de Lyon, Lyon, France; [7]Birmingham Women's and Children's Hospital, Birmingham, United Kingdom; [8]Wenkert & Young, LLC, Thousand Oaks, CA, United States; [9]Department of Medical Imaging, Children's Hospital of Eastern Ontario, University of Ottawa, ON, Canada; [10]Department of Surgery, Children's Hospital of Eastern Ontario, University of Ottawa, ON, Canada; [11]The Ottawa Pediatric Bone Health Research Group, Children's Hospital of Eastern Ontario Research Institute, Ottawa, ON, Canada; [12]Inozyme Pharma, Boston, MA, United States; [13]Al Jalila Children's Specialty Hospital, Dubai, United Arab Emirates; [14]Mayo Clinic, Rochester, MN, United States

inozyme pharma

CHEO RESEARCH INSTITUTE INSTITUT DE RECHERCHE

Introduction

- Ectonucleotide pyrophosphatase/phosphodiesterase family member 1 (ENPP1) is a critical enzyme that generates pyrophosphate (PPi), a mineralization inhibitor, and adenosine, a regulator of vascular smooth muscle cell proliferation[1-4]
- Patients with biallelic, loss-of-function *ENPP1* variants typically present at birth with severe arterial calcification and cardiovascular complications, a phenotype described as Generalized Arterial Calcification of Infancy Type 1 (GACI type 1)[5-6]
- All patients with ENPP1 deficiency who survive beyond infancy are expected to develop autosomal recessive hypophosphatemic rickets type 2 (ARHR2), a PPi- and Fibroblast Growth Factor 23 (FGF23)-mediated hypophosphatemia causing rickets and osteomalacia
- ARHR2 leads to impaired growth and skeletal deformities, with substantial impacts on quality of life

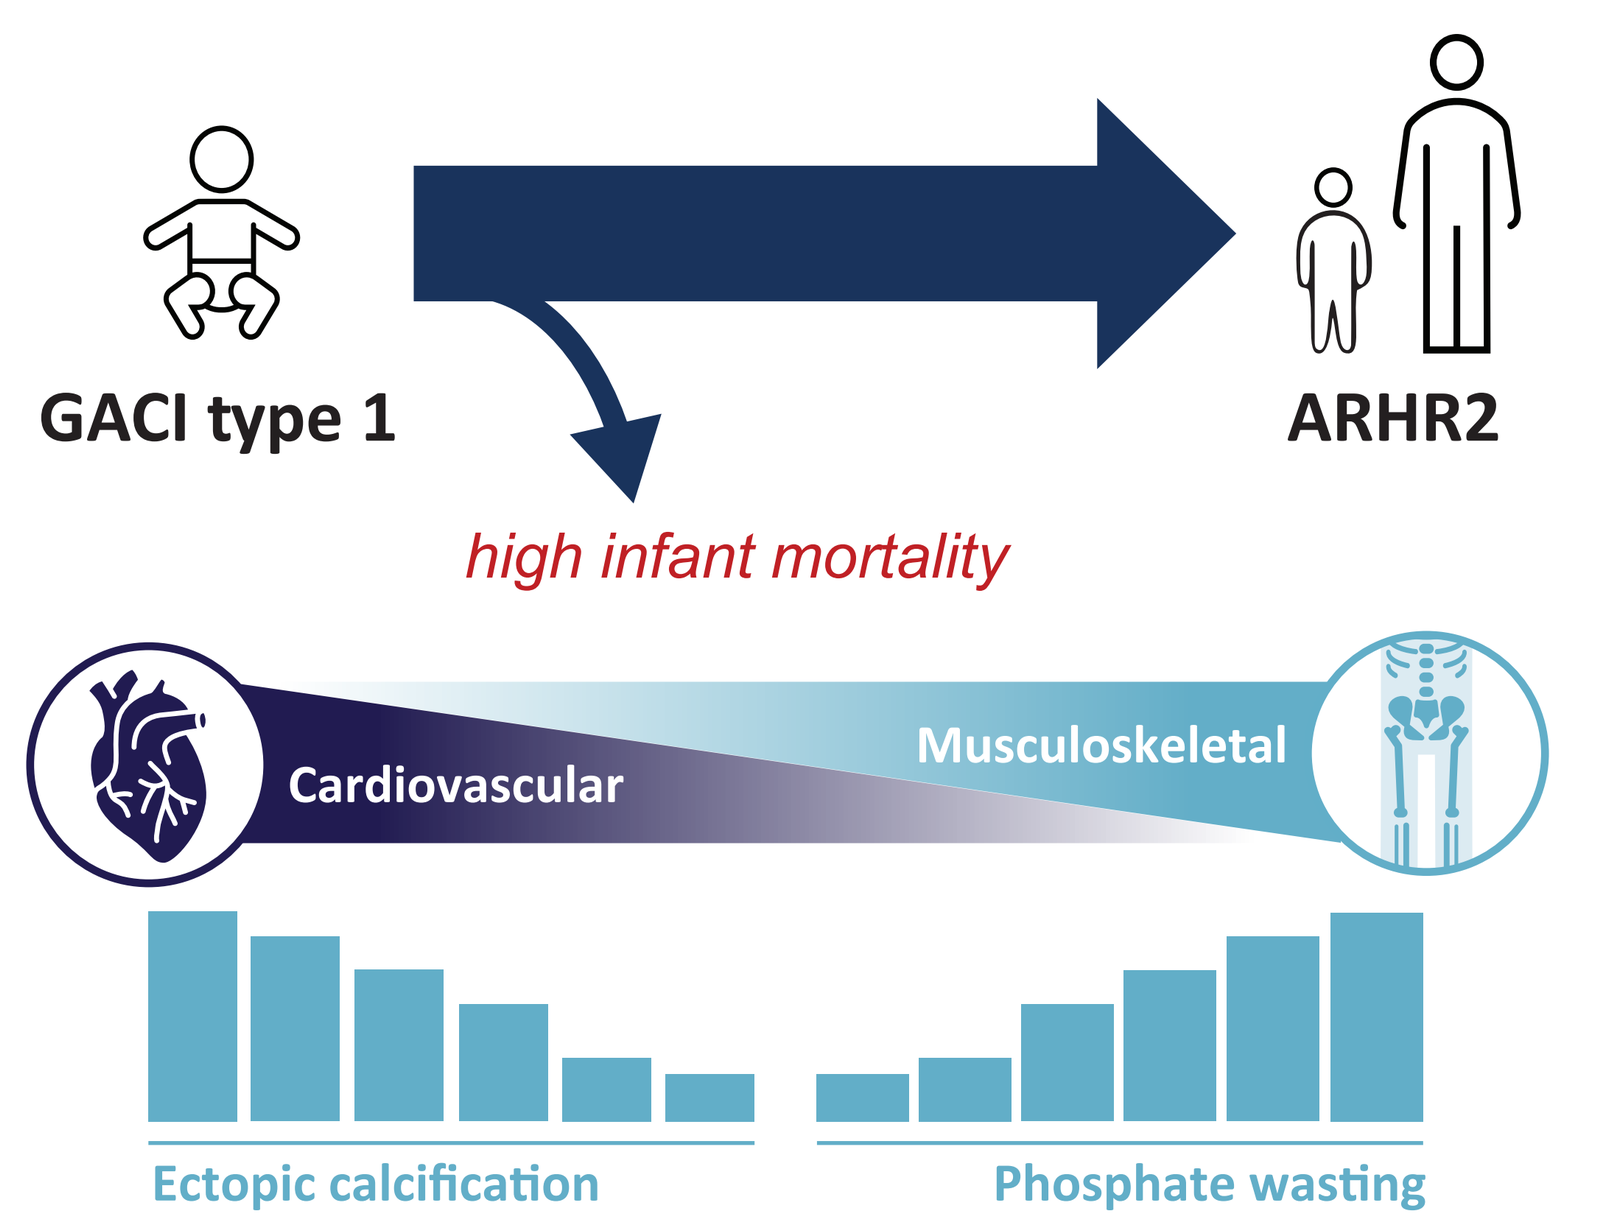

GACI type 1
ARHR2
high infant mortality
Cardiovascular
Musculoskeletal
Ectopic calcification
Phosphate wasting

Objective & methods

Objective: To describe the clinical phenotype in children and adults with ARHR2 receiving current standard of care, by characterizing their height, phosphate metabolism, treatments/interventions and (for children), their Rickets Severity Score (RSS), rickets biomarkers, bone age, and Bone Health Index (cortical thickness and mineralization of three metacarpal bones, a surrogate for bone strength)

Methods:

- Sub-group analysis (N = 14) of a multicenter longitudinal retrospective chart review, including patients with clinical and genetic diagnosis of ENPP1 deficiency
- In a post hoc analysis, bone age and Bone Health Index were calculated (in children) using BoneXpert™ software

Results

Demographics & medical history

Demographic, N = 14	
Age at enrollment	
Median (range), years	19.5 (4–33)
Sex, n (%)	
Male	8 (57.1%)
Female	6 (42.9%)
GACI diagnosis	
Participants, n (%)	7 (50.0%)
Age at GACI diagnosis (months)	
Median (range)	2.0 (0–8)
ARHR2 diagnosis	
Participants, n (%)	10 (71.4%)
Age at ARHR2 diagnosis (months)	
Median (range)	61 (4–255)
Mortality, n (%)	2 (14.3%)
Treatment for GACI age <1 year, n (%)	7 (50.0%)
Bisphosphonates or STS	5 (35.7%)
Respiratory support[a]	4 (33.3%)
Anti-hypertensives	3 (21.4%)
Heart failure medications	1 (7.1%)
Medicinal treatment for rickets, n (%)	9 (64.2%)
Active vitamin D3[b]	9 (64.2%)
Oral phosphate	8 (57.1%)
Concomitant medications[a] age >1 year, n (%)	
Cholecalciferol	8 (66.7%)
Bisphosphonates	7 (50.0%)
Anti-hypertensives	5 (23.7%)
Heart failure medications	2 (14.3%)
Orthopedic surgery, n (%)	9 (64.3%)
Epiphysiodesis	7 (50.0%)
Osteotomy	5 (35.7%)
Hearing aids, n (%)	6 (42.9%)
Physiotherapy, n (%)	4 (28.6%)

[a]Includes mechanical ventilation and supplemental oxygen.
[b]Included calcitriol and alfacalcidiol.
ARHR2, autosomal recessive hypophosphatemic rickets type 2; GACI, Generalized Arterial Calcification of Infancy; STS, sodium thiosulfate.

Bone age & bone health index Z-scores by Bone Xpert

CA at X ray acquisition (years)	Bone age by BoneXpert (years)	CA – BA (difference, years)	Bone health index (Z-score)
9.2 (6.0)	8.4 (5.6)	0.9 (1.3)*	-1.3 (1.3)**

All results expressed as mean (SD); *p<0.01; **p<0.001.
CA, chronological age; BA, bone age.

- We have carried out internal validation of BoneXpert in ENPP1 deficiency, showing high levels of agreement between BoneXpert and radiologists
- Here we show the bone age and Bone Health Index results by BoneXpert in 9 patients with ARHR2
- These results affirm a slight bone age delay (Greulich and Pyle method) and an average Bone Health Index below the mean, both as expected in a rachitic condition

Blood phosphate

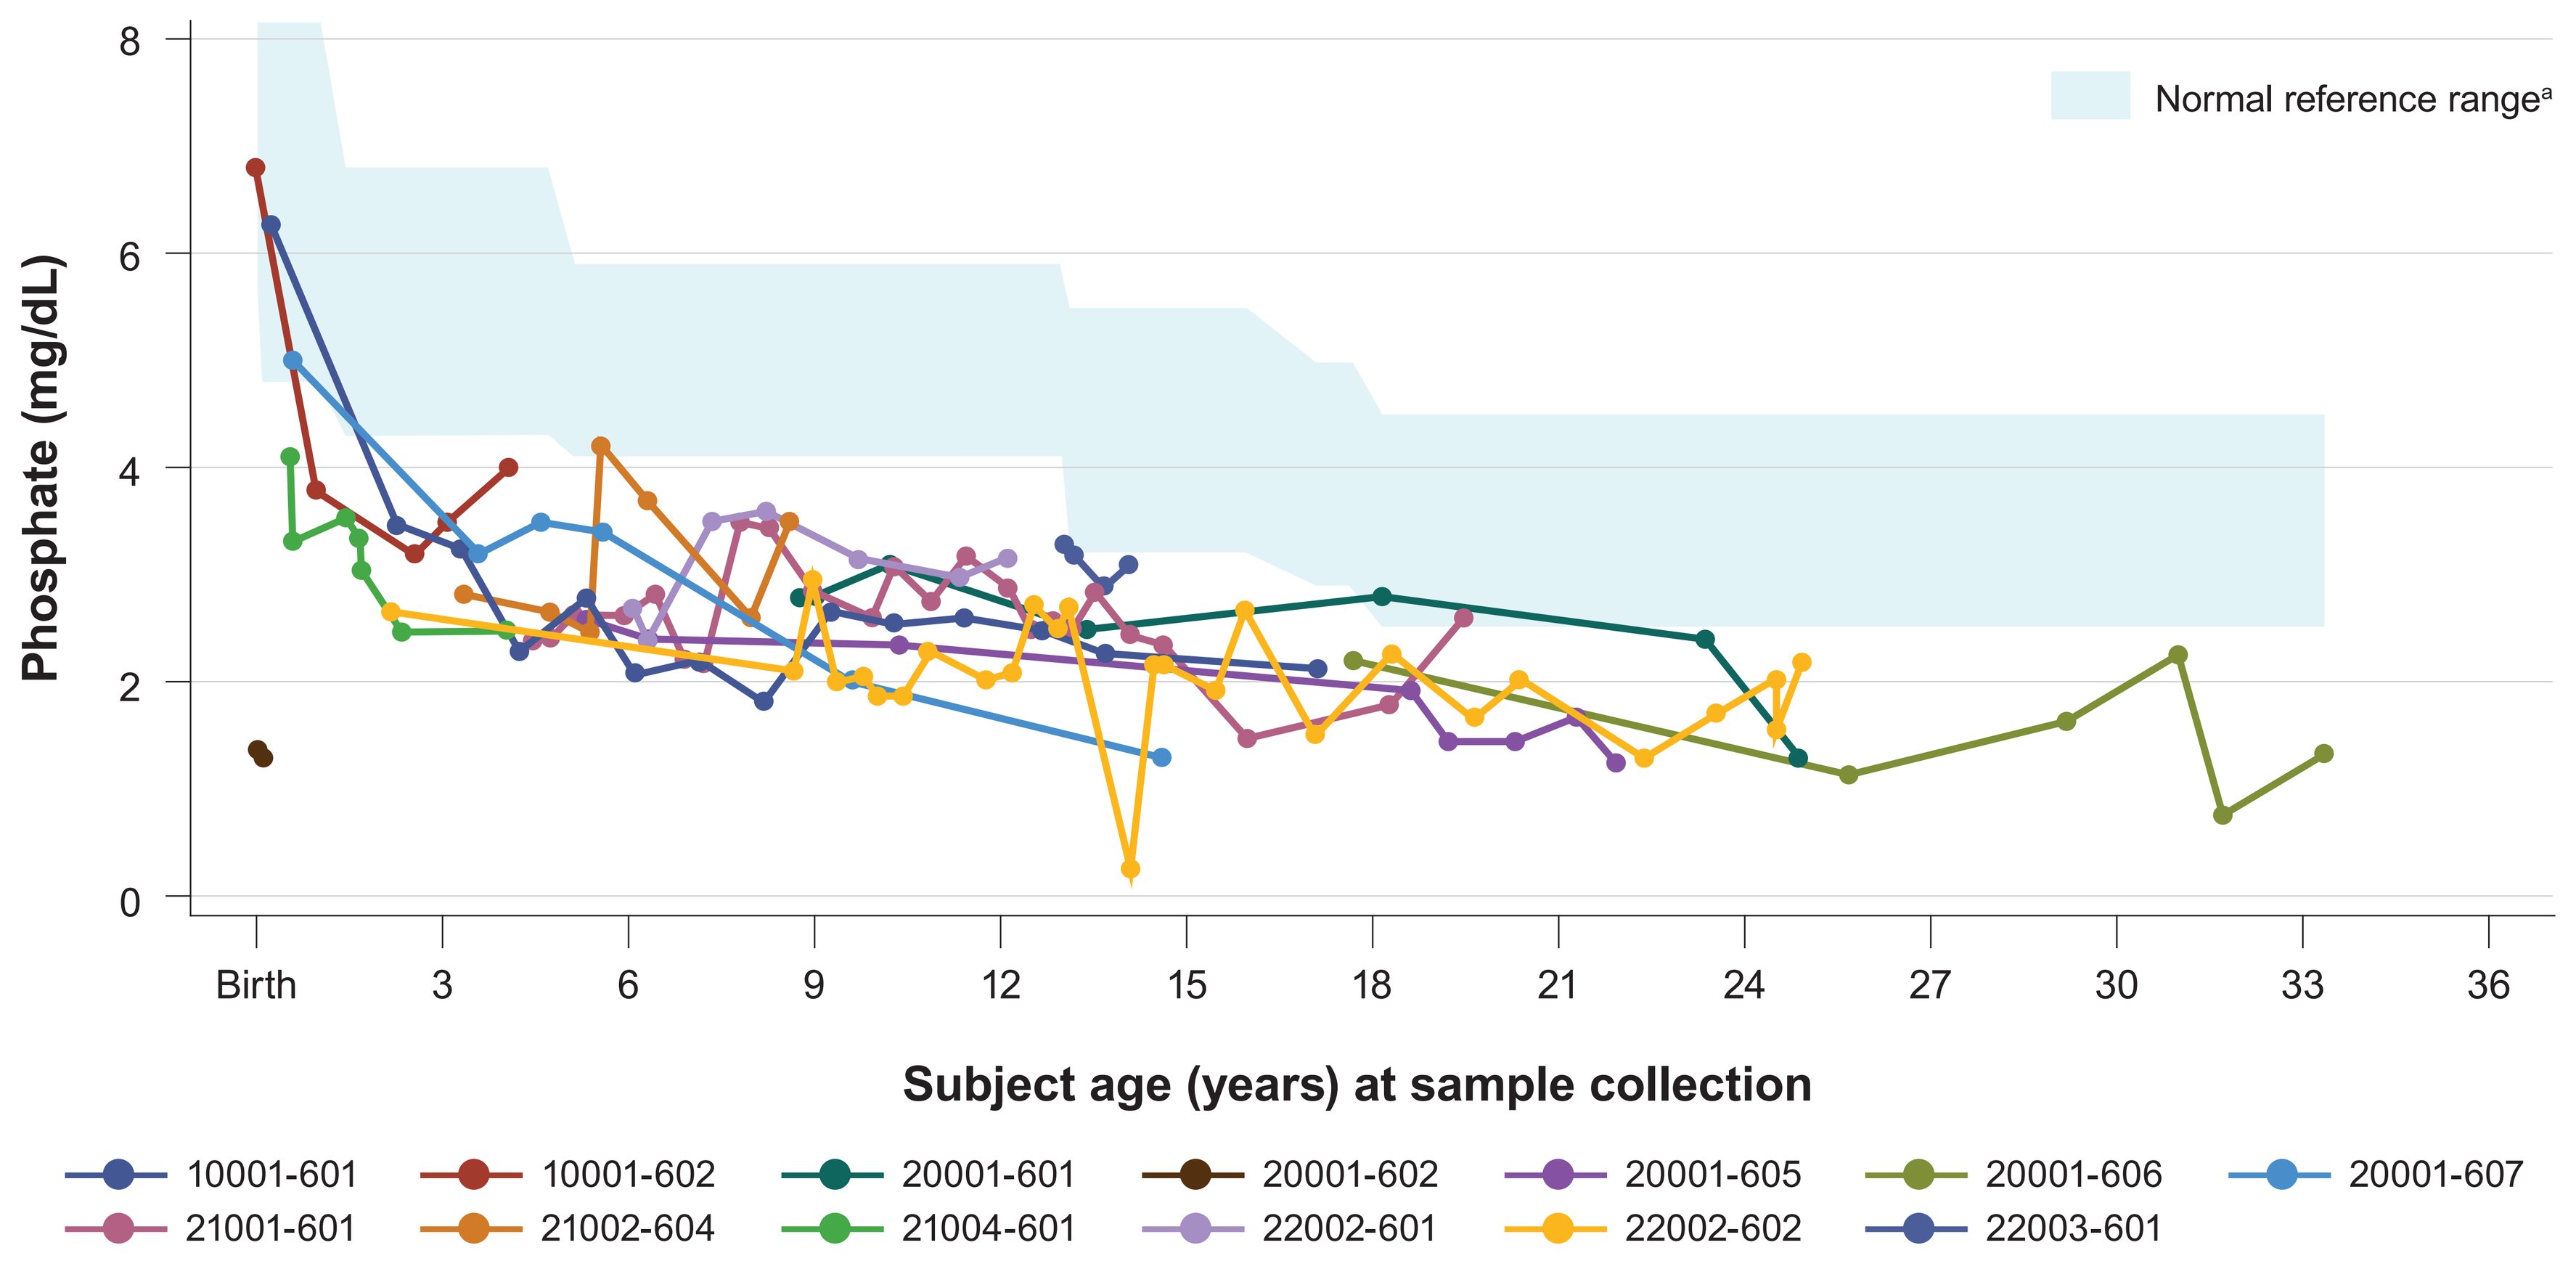

Normal reference range[a]
Phosphate (mg/dL)
Subject age (years) at sample collection
Birth 3 6 9 12 15 18 21 24 27 30 33 36
10001-601 10001-602 20001-601 20001-602 20001-605 20001-606 20001-607
21001-601 21002-604 21004-601 22002-601 22002-602 22003-601

[a]As published in Cárdenas Aguilera 2024.

- Progressive decline in serum/plasma phosphate was observed throughout the ages assessed, with levels below the lower limit of normal
- Most precipitous declines were in the first three years of life
- Even the patients receiving phosphate supplementation and active vitamin D3 (9/13) were persistently hypophosphatemic, as expected for an FGF23-mediated, renal phosphate-wasting disorder

Blood alkaline phosphatase (ALP)

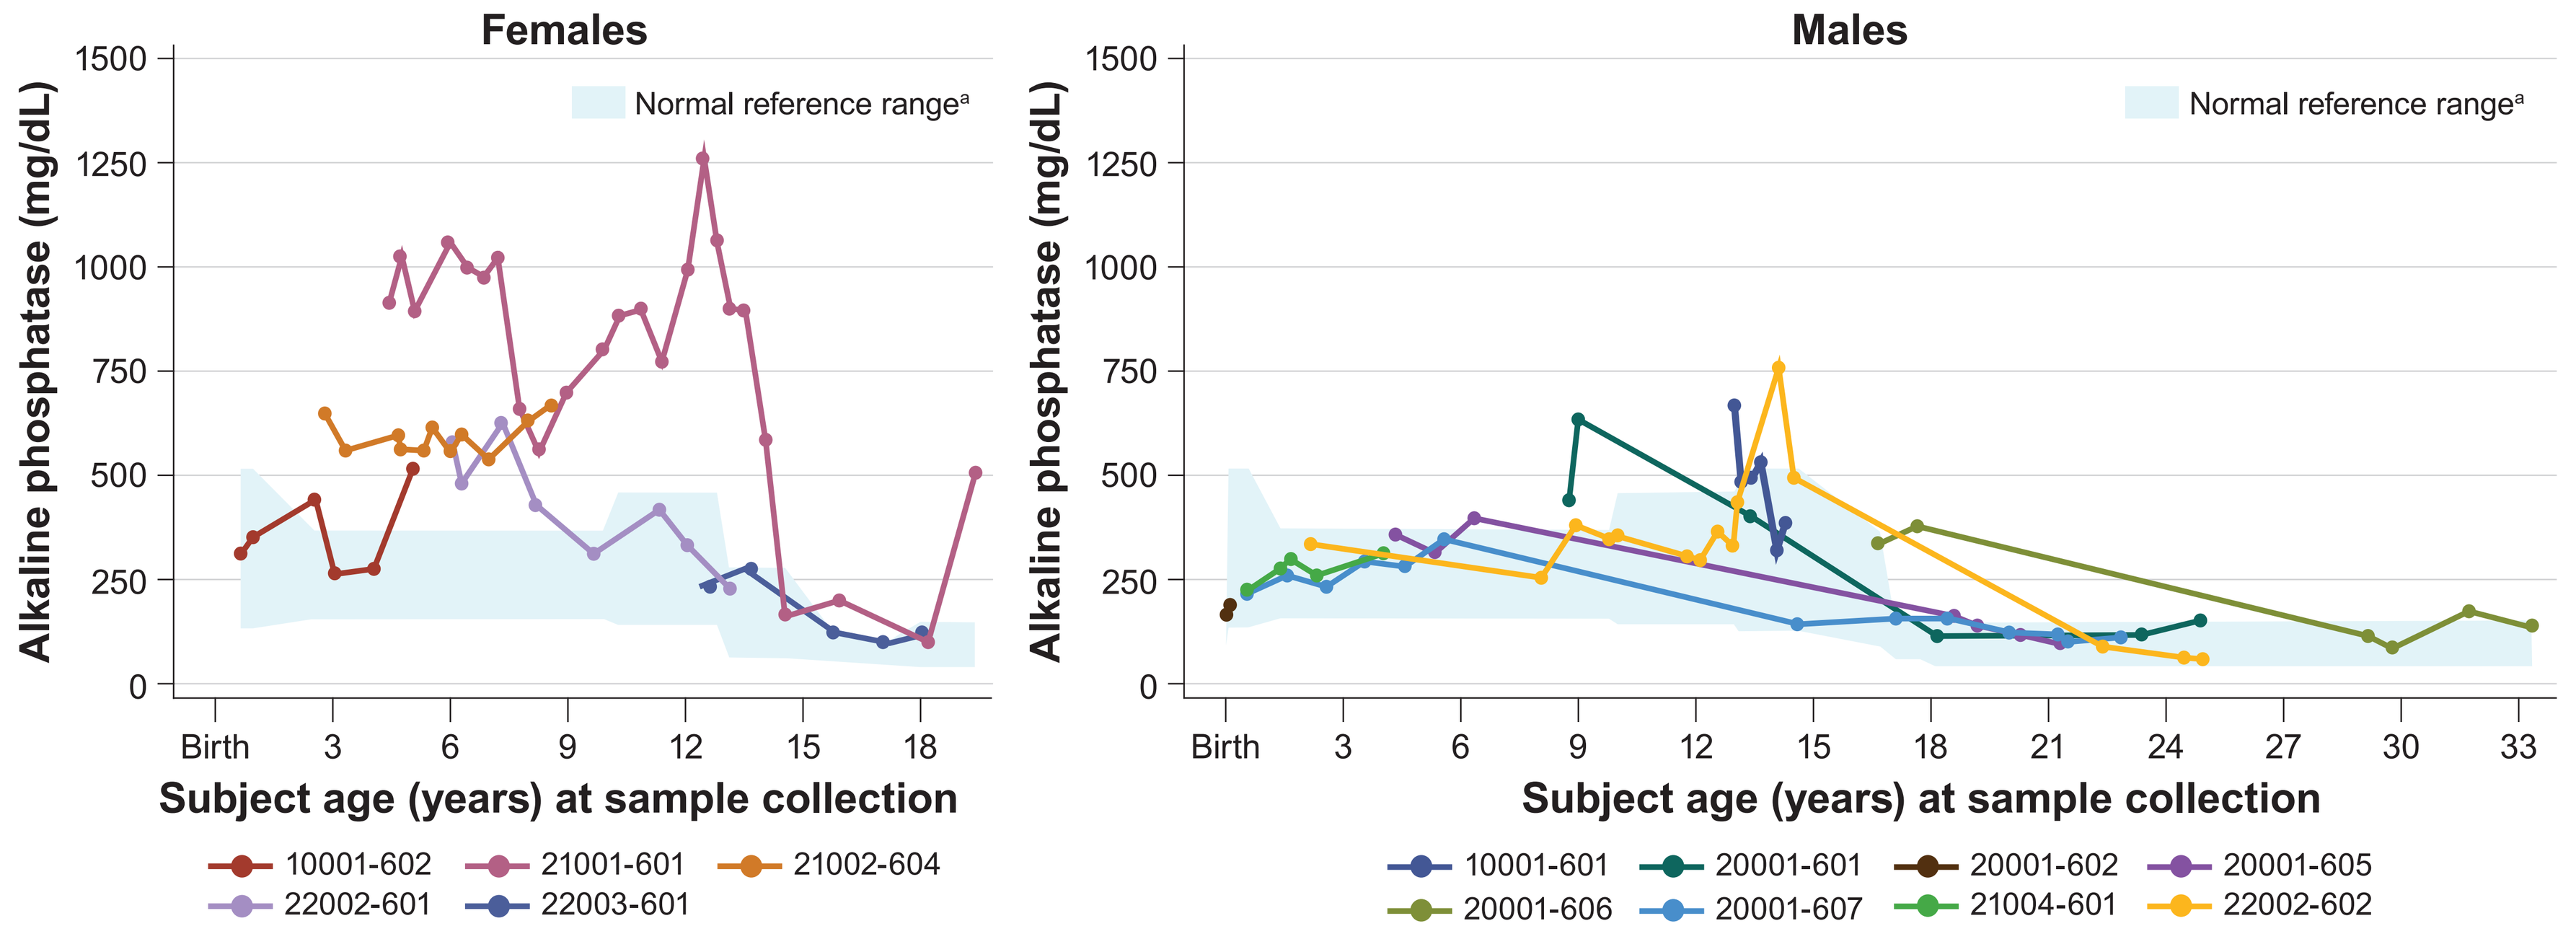

Females
Males
Normal reference range[a]
Alkaline phosphatase (mg/dL)
Subject age (years) at sample collection
10001-602 21001-601 21002-604 22002-601 22003-601
10001-601 20001-601 20001-602 20001-605 20001-606 20001-607 21004-601 22002-602

[a]As published in Cárdenas Aguilera 2024.

- Variability in inter-patient ALP, where high ALP is a marker of active rickets
- Majority of patients had elevated ALP at some point in time, with highest values observed in childhood and early adolescence

Height Z-scores

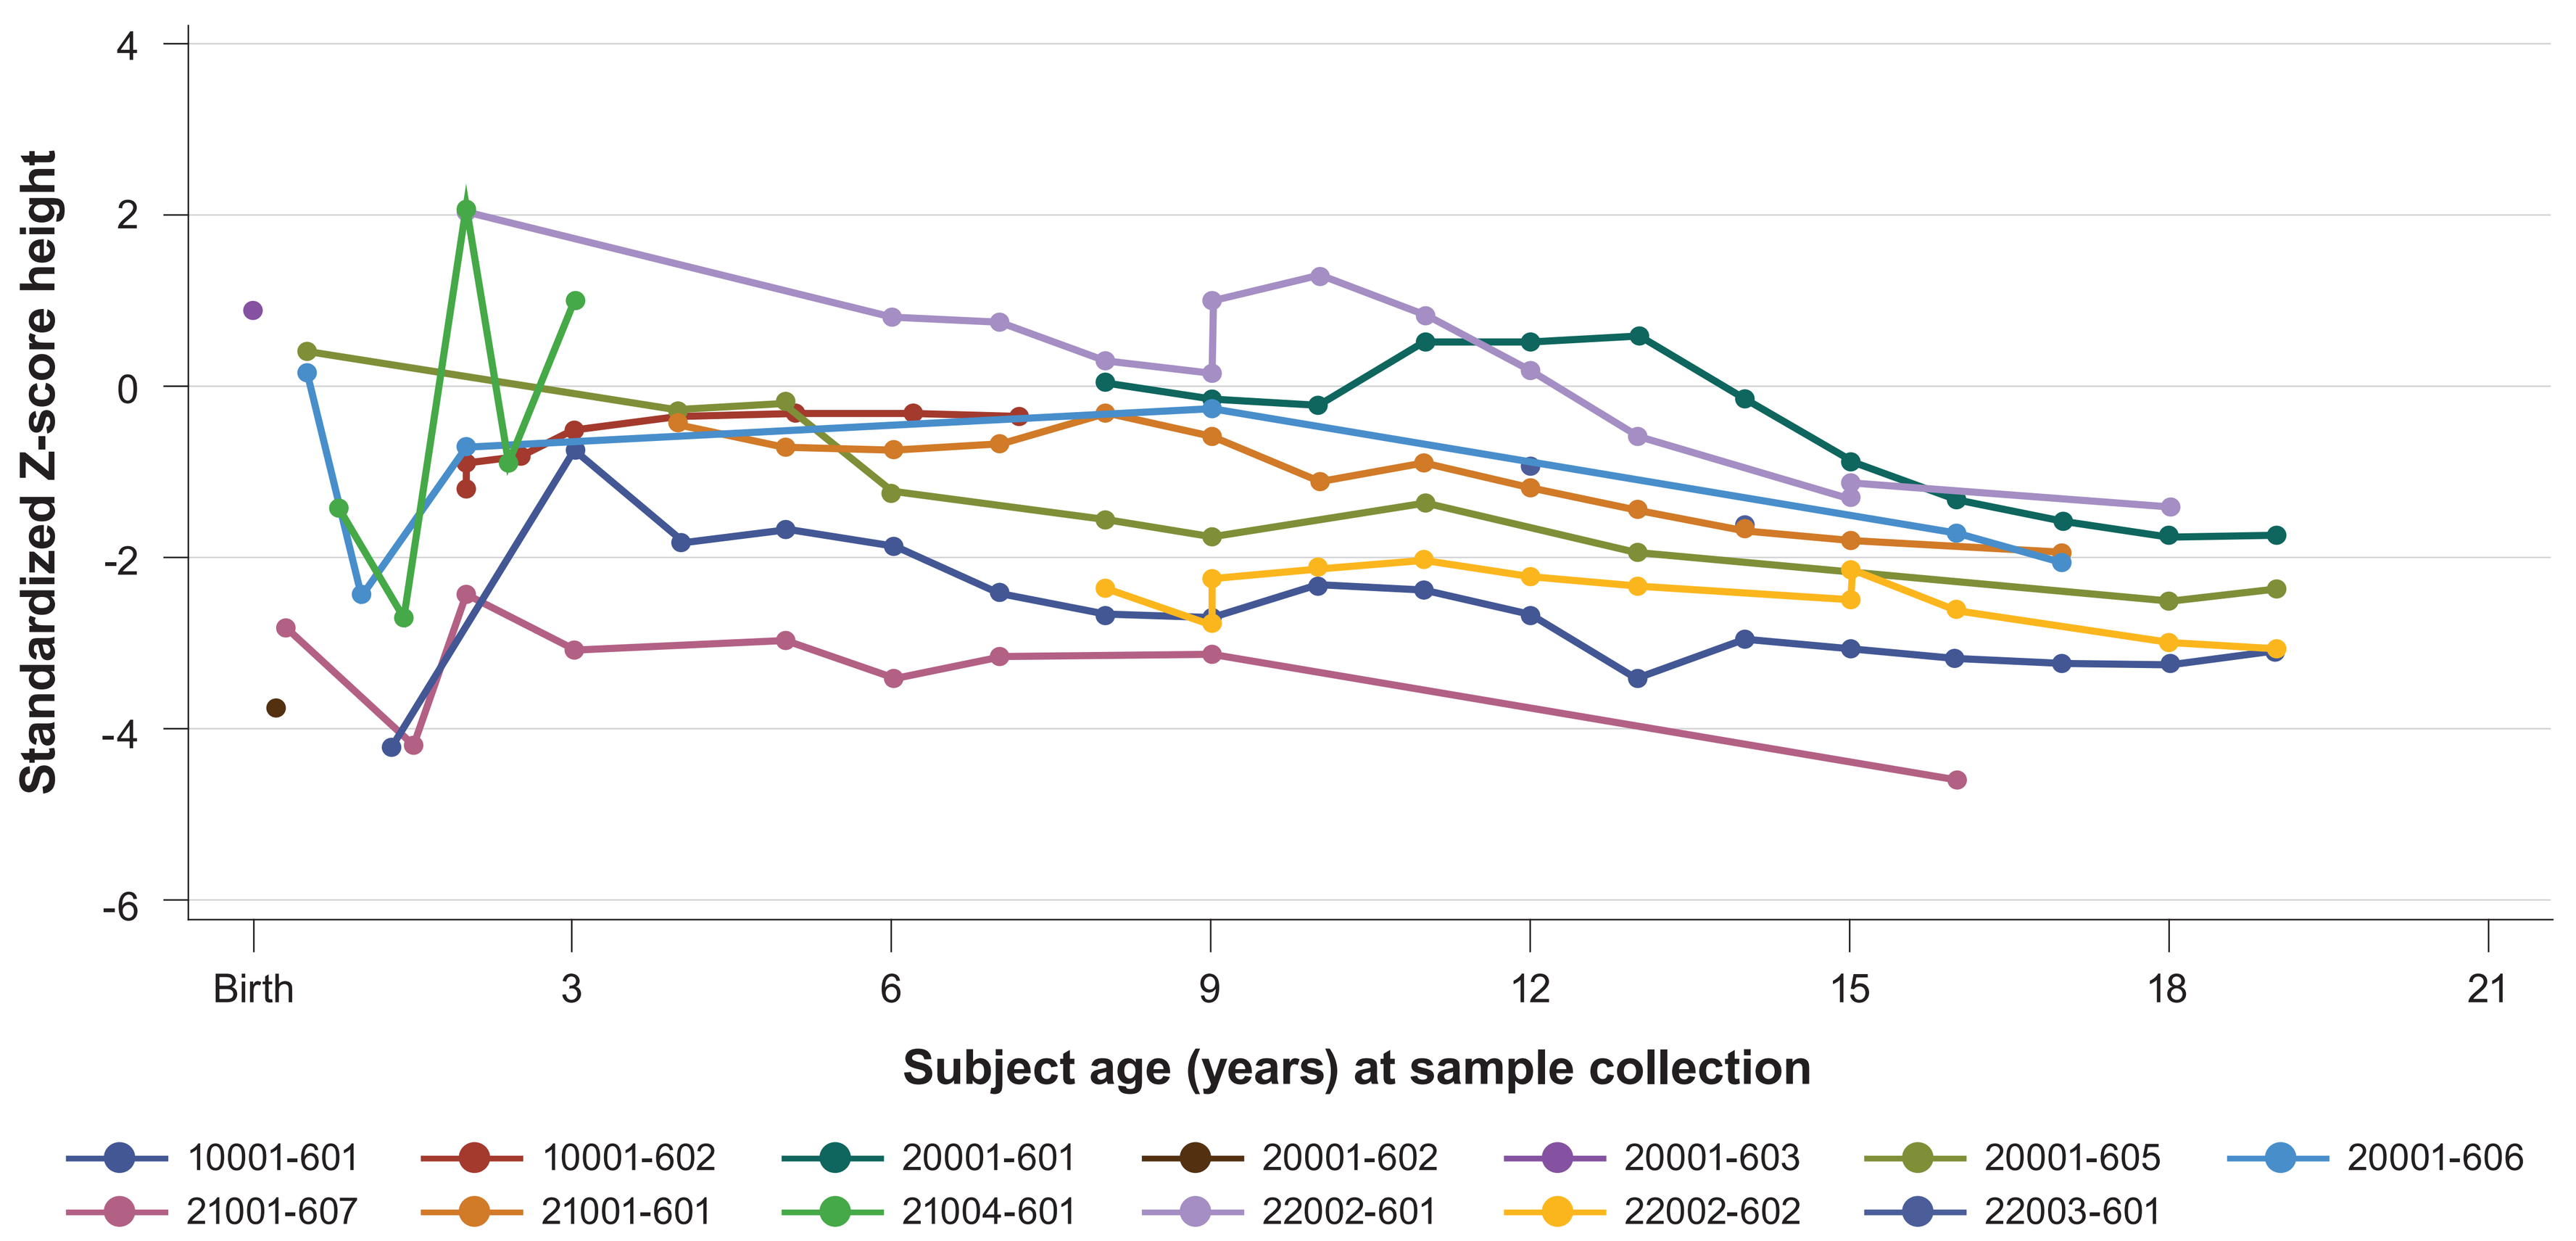

Standardized Z-score height
Subject age (years) at sample collection
Birth 3 6 9 12 15 18 21
10001-601 10001-602 20001-601 20001-602 20001-603 20001-605 20001-606
21001-607 21001-601 21004-601 22002-601 22002-602 22003-601

- Height Z-scores tended to decline over time
- At final follow up, 5/13 (38%) patients had short stature (Z-score ≤-2) including 3 patients with stature Z-scores more than 3.0 standard deviations below the mean

Rickets severity score (RSS)

Patient A: 3.5-year-old girl
Global RSS = 2
- Wrist RSS = 1
- Knee RSS = 1

Maximum global RSS in participants < 13 years old, N = 8	
Mean (SD)	1.6 (1.1)
Median	1.8
Min, Max	0.0, 3.0

Patient B: 12.9-year-old boy
Global RSS = 3
- Wrist RSS = 1.5
- Knee RSS = 1.5

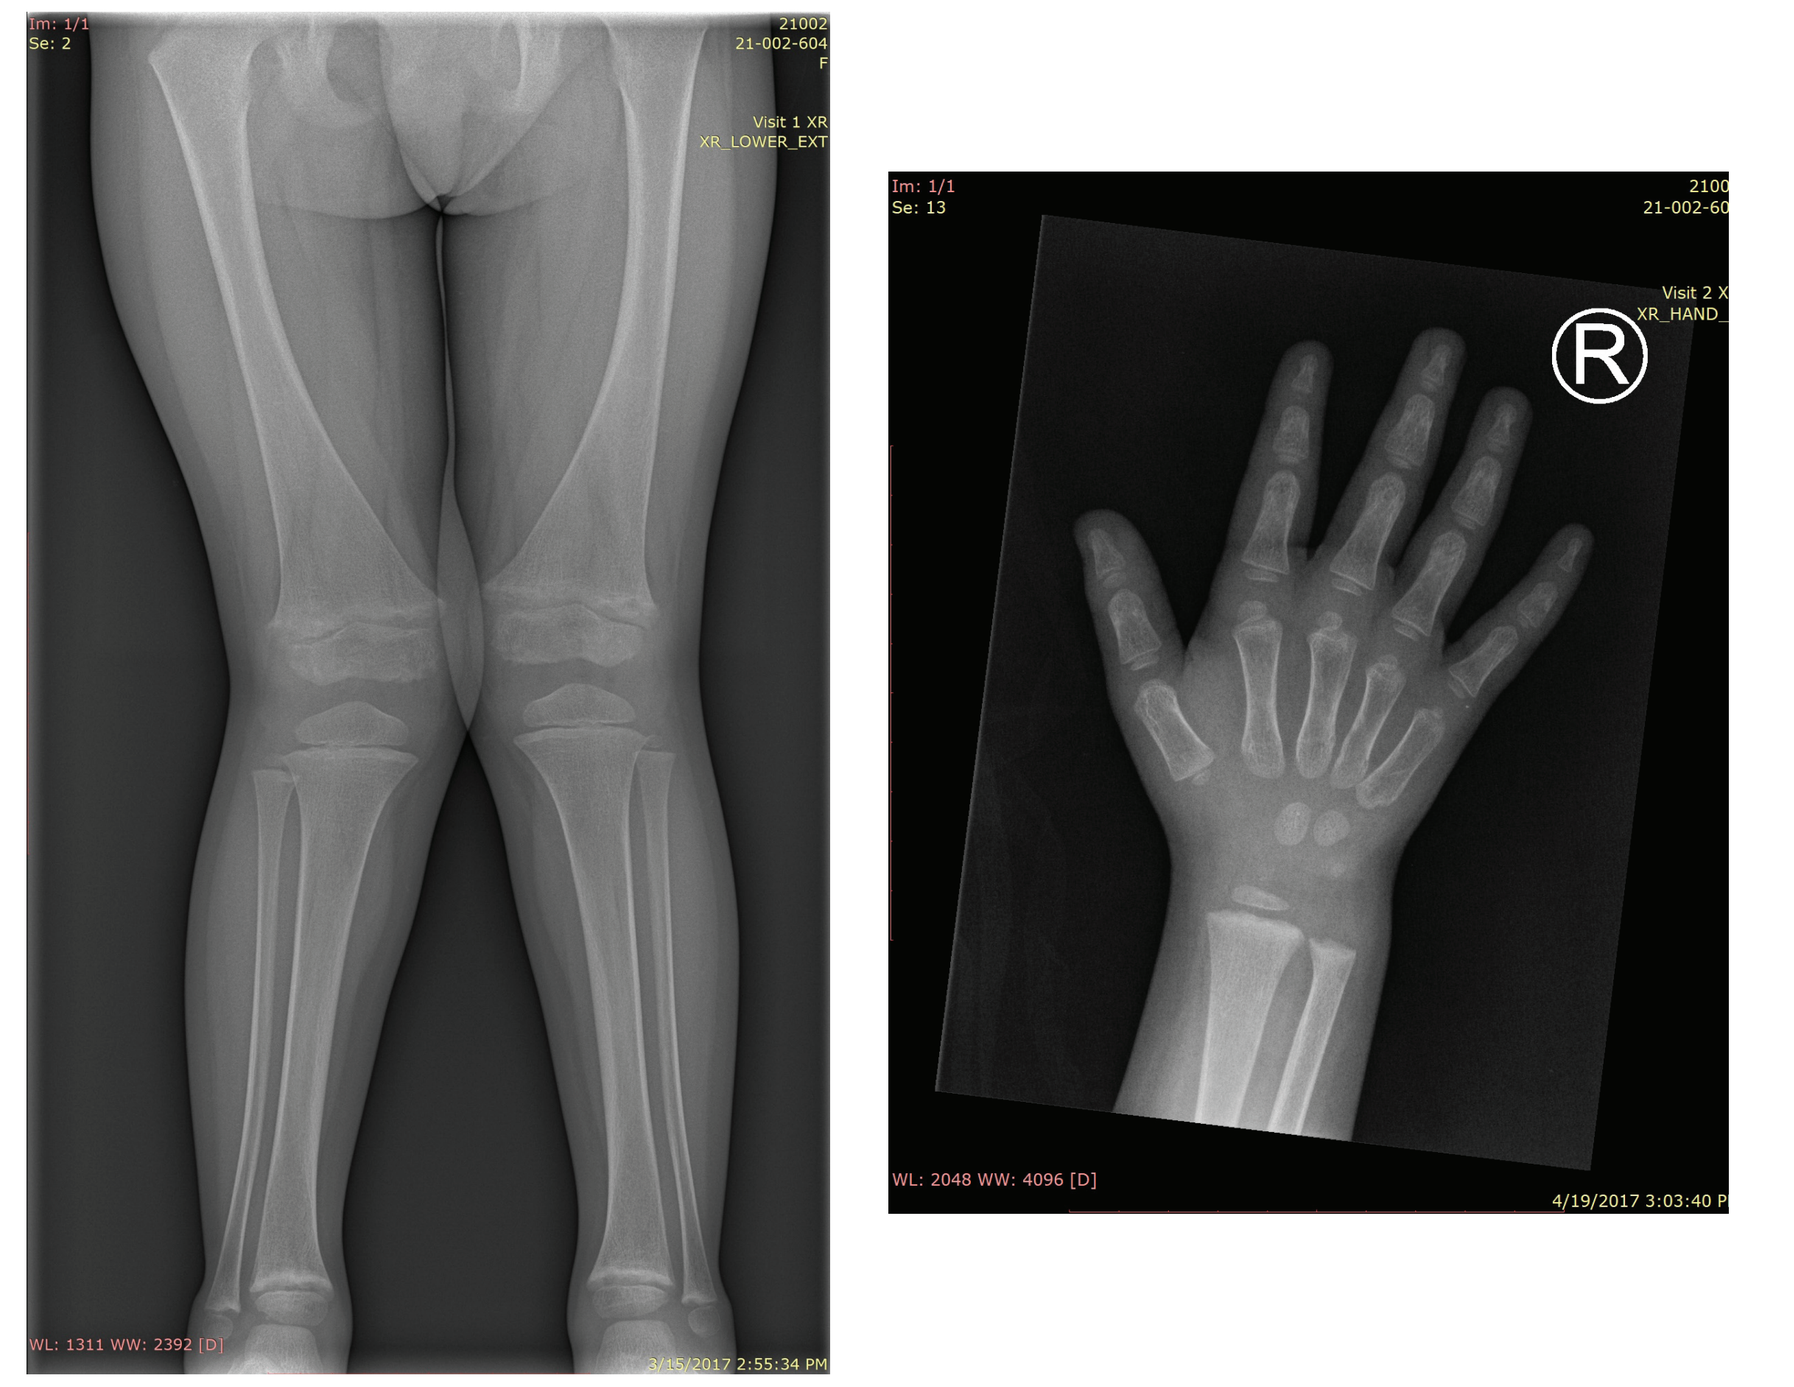
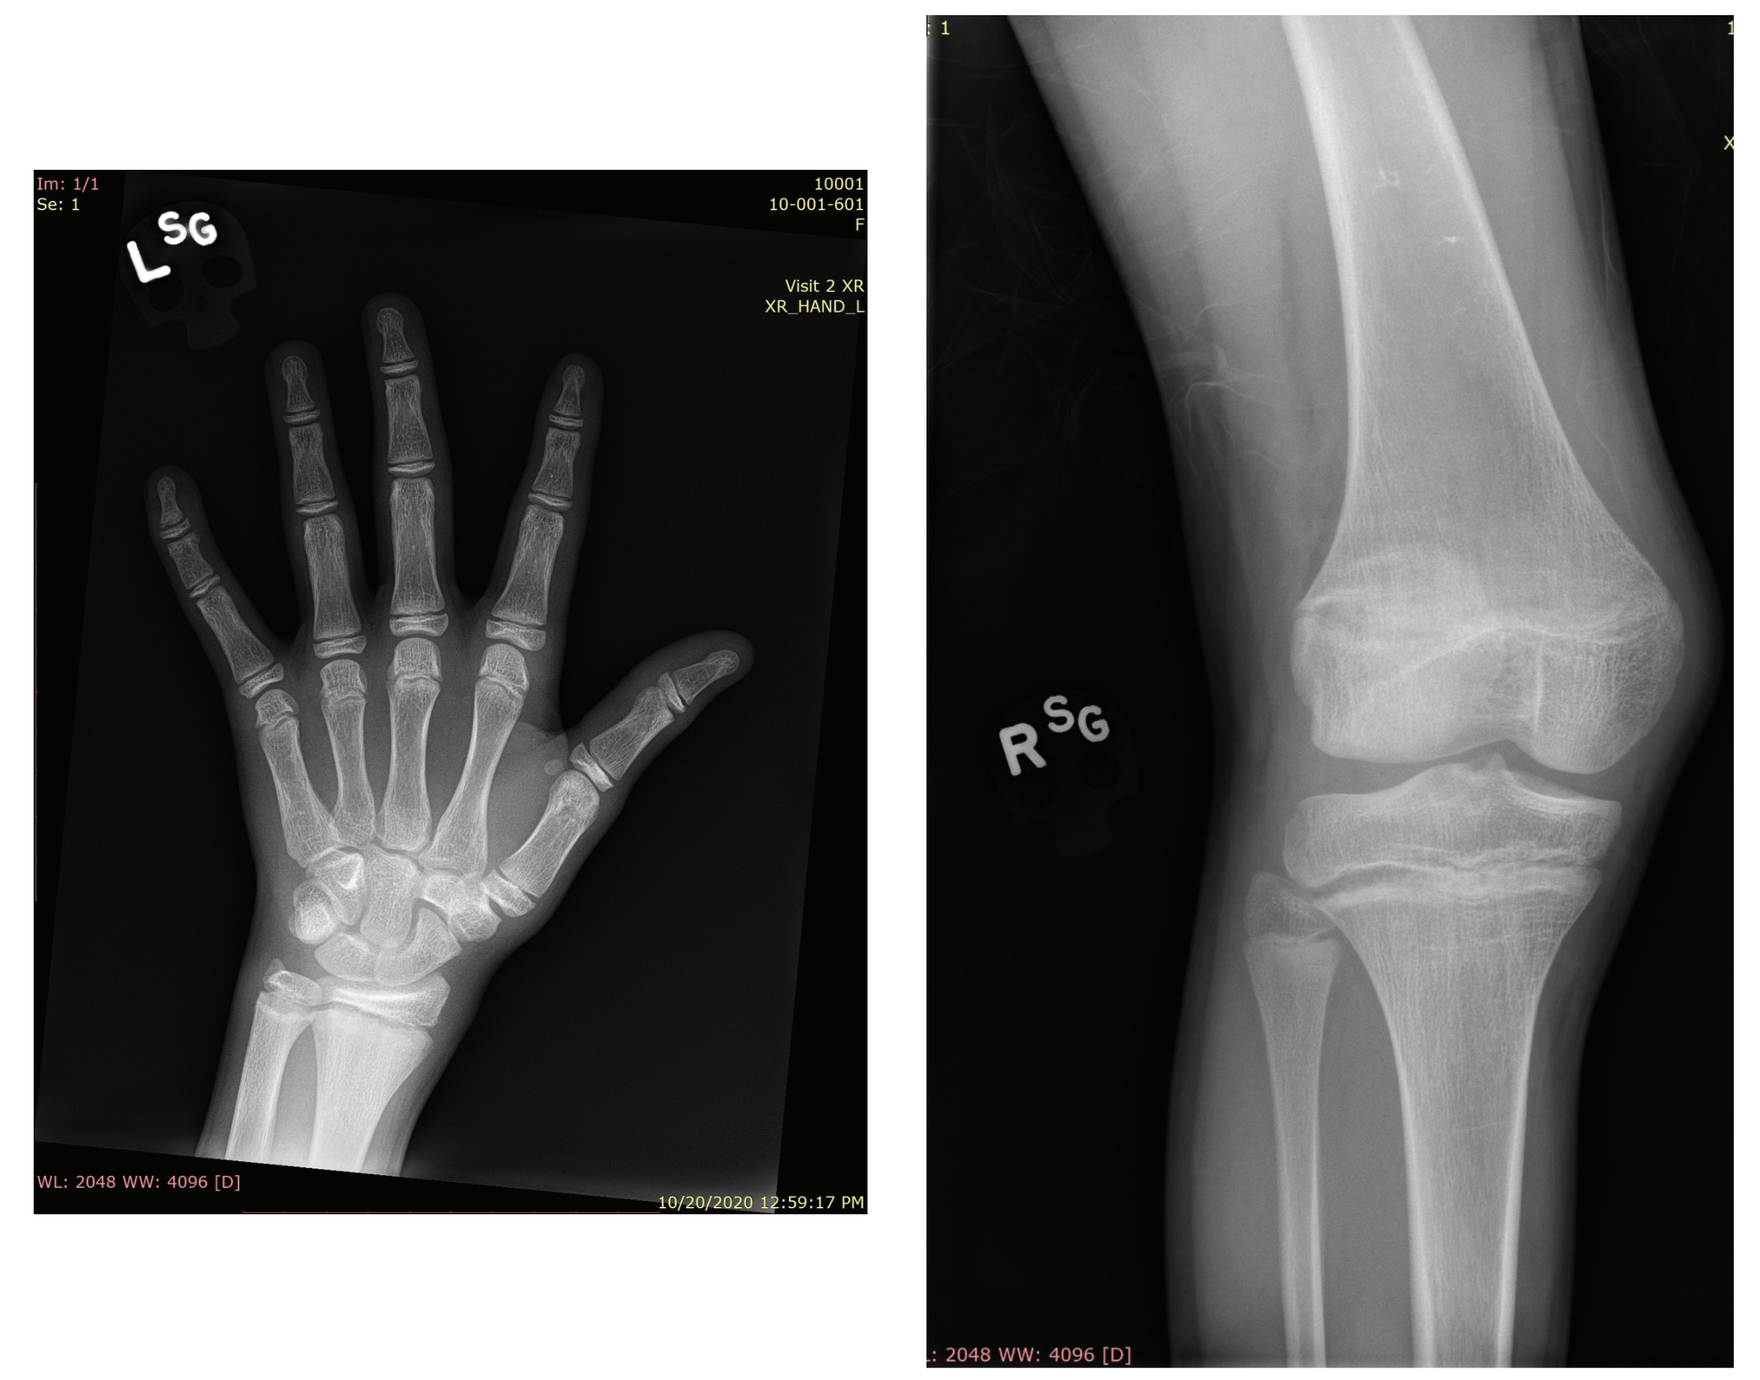

Conclusions

- Children with ENPP1 deficiency under current standard of care exhibit features consistent with FGF23-mediated hypophosphatemia, including:
 - — Persistently low plasma phosphate due to renal phosphate-wasting
 - — Variable serum ALP
 - — Rickets (median RSS 1.8), with features indistinguishable from X-linked hypophosphatemia
 - — Impaired growth and short stature
 - — Delayed bone age and Bone Health Index by BoneXpert
- Many surviving patients may require treatments/interventions for hypertension and hearing loss
- High phenotypic overlap of ARHR2 with other hereditary hypophosphatemias supports the recommendation for genetic testing to obtain accurate diagnosis

References

1. Ralph D, et al. *Am J Pathol.* 2022;192:762–770
2. Oriss IR, et al. *Curr Opin Pharmacol.* 2016;28:57-68
3. Nitschke Y, et al. *Exp Mol Med.* 2018;50(10):1–12
4. Albayrak G, et al. *Vascular.* 2015;23:124–131
5. Ferreira CR, et al. *Ann Rev Pathol Mech Dis.* 2024;19:507–540
6. Ferreira CR, et al. *JBMR Plus.* 2025;9(5):ziaf019
7. Cárdenas Aguilera JG, et al. *Rev. Colomb. Nefrol.* 2024:11(1):https://doi.org/10.22265/acnef.11.1.754

Disclosures & acknowledgments

As of July 1, 2025, Inozyme Pharma is a wholly owned subsidiary of BioMarin Pharmaceutical Inc.

This study was sponsored by Inozyme Pharma. The authors would like to thank the patients and their families who participated in this study. We acknowledge the contributions of site staff.

DP and KCG are employees and stockholders of Inozyme Pharma. D Wenkert is a former employee and stockholder of Inozyme Pharma. LW has participated in clinical trials with Inozyme (standardized central imaging), with funds to institution. D Weber and MZM are Inozyme site PI's and have received consulting fees and grant support from Inozyme. TT receives consulting fees from Leanne Ward, CHEO, in collaboration with Inozyme.

Presented at American Society for Bone and Mineral Research (ASBMR) annual meeting: September 5–8, 2025; Seattle, WA, USA

©2025 BioMarin Pharmaceutical Inc. All rights reserved.