Plasma pyrophosphate (PPi) levels correlate with severity of clinical manifestations of ENPP1 deficiency

Kevin O'Brien[1], Caitlin Sullivan[1], Jennifer Howe[1], Denis Schrier[1], Hervé Husson[1], and Yves Sabbagh[1]

[1]Inozyme Pharma, Boston, MA, USA

inozyme pharma

Poster # Sun-298

Introduction

- Ectonucleotide pyrophosphatase/phosphodiesterase 1 (ENPP1) deficiency is a rare autosomal recessive genetic condition that results in extensive medial vascular calcification, vascular stenosis, and death in approximately 50% of patients within the first year of life.[1] Those who survive experience lifelong cardiovascular and skeletal symptoms due to low levels of circulating pyrophosphate (PPi)[2]
 - The ENPP1 enzyme is essential to the PPi-adenosine pathway, which converts adenosine triphosphate (ATP) to PPi and adenosine monophosphate (AMP), which is further broken down to adenosine by the action of CD73[1,3]
 - PPi and adenosine are essential for maintaining bone and blood vessel health[4-6]
- INZ-701 (BMN 401), a Fc fusion protein containing recombinant ENPP1 to cleave ATP and restore circulating PPi and AMP levels, is currently under investigation for treatment of ENPP1 deficiency and other disorders caused by low PPi (**Figure 1**)[7]
- Treatment with BMN 401 in ENPP1-deficient mice normalized PPi levels, protected against vascular calcification, and prevented bone abnormalities[7,8]
- Though PPi levels are clearly associated with disease (ie, low levels are associated with ENPP1 deficiency and high levels with hypophosphatasia),[9] there is limited evidence linking plasma PPi directly to clinical phenotypes such as ectopic calcification

Figure 1. Mechanism of ENPP1 deficiency and mechanism of action of BMN 401

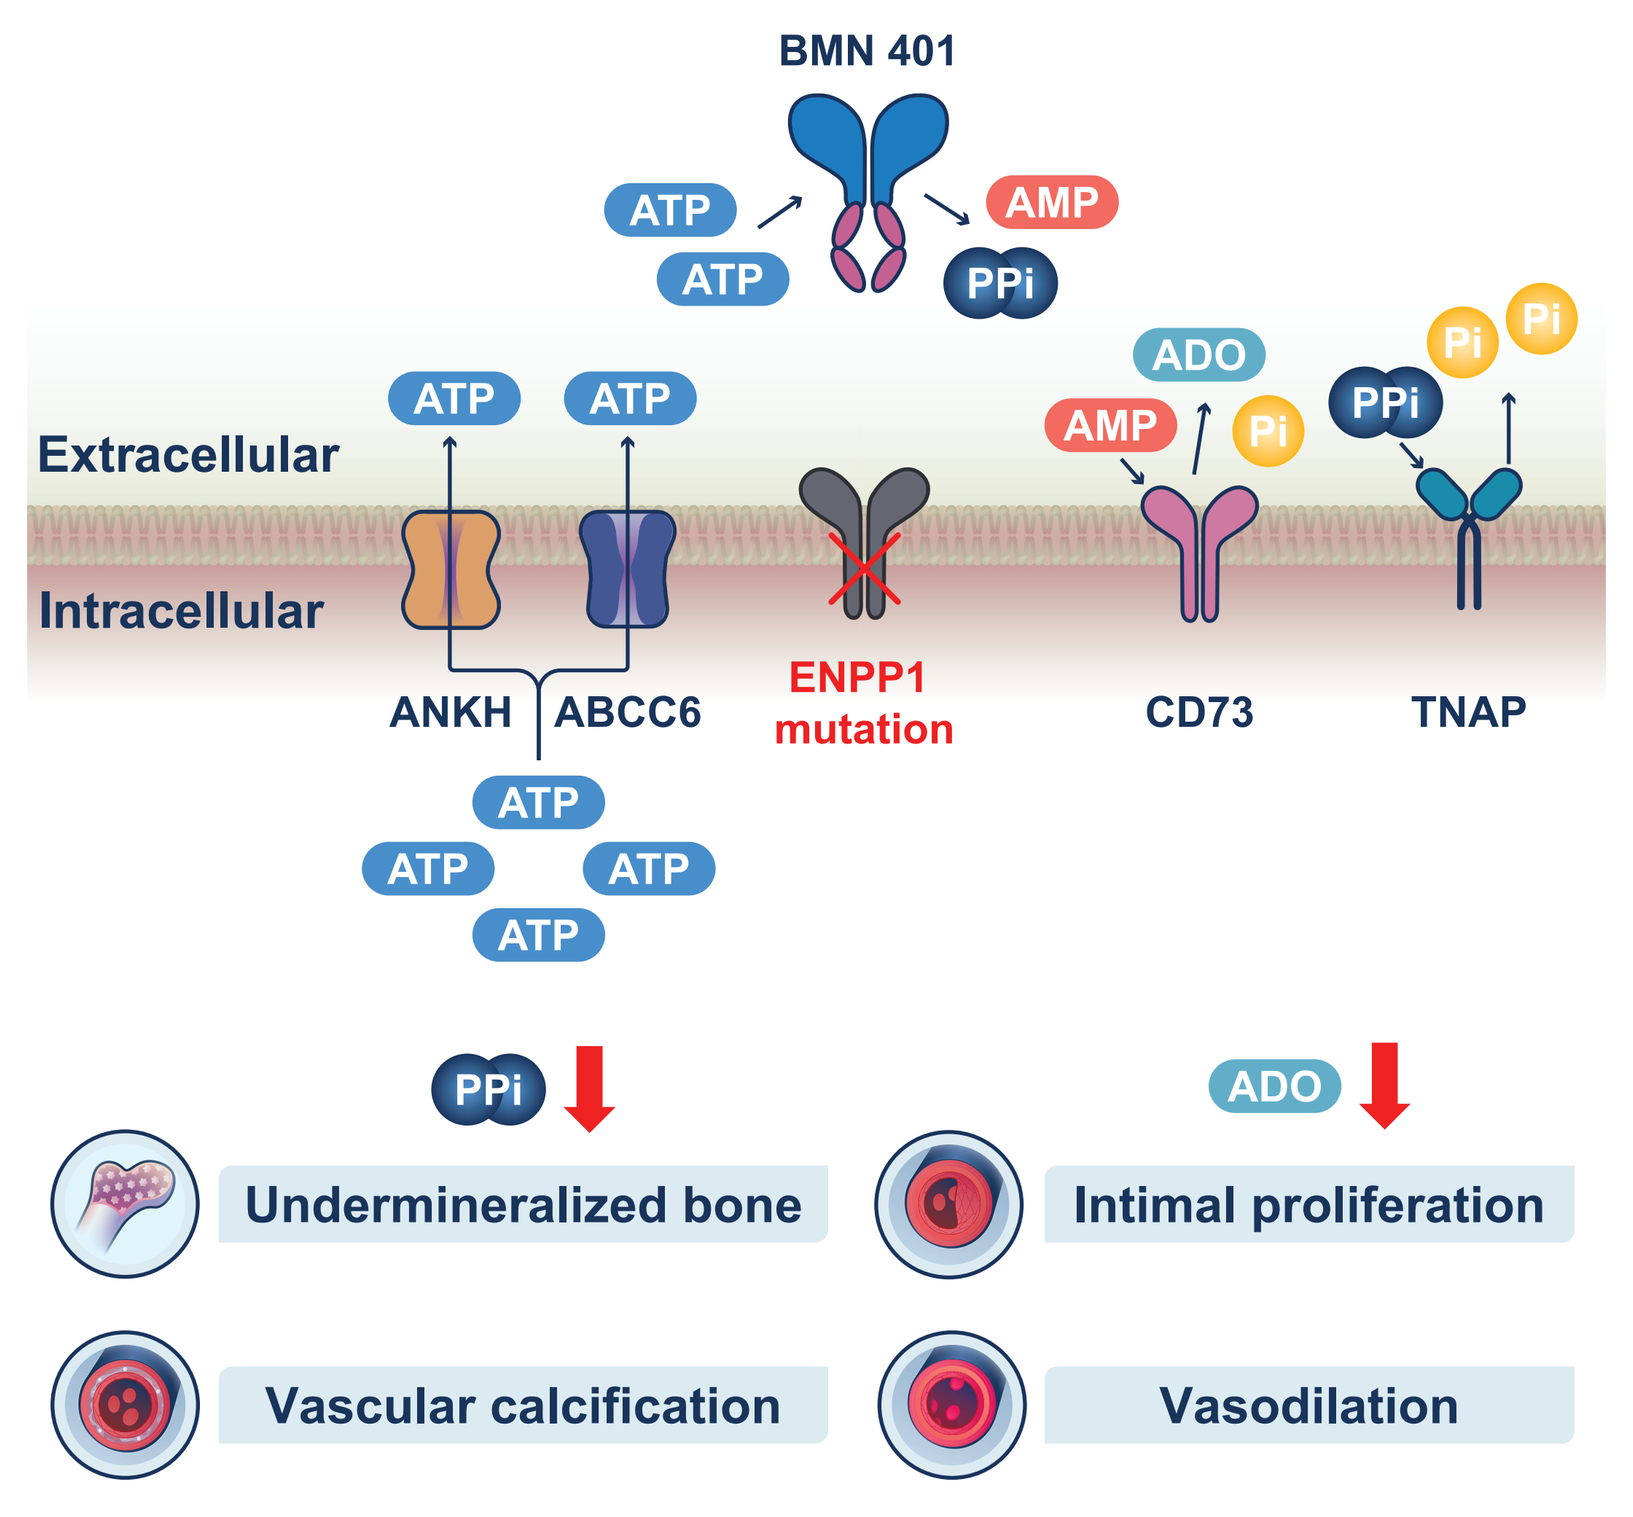
BMN 401
ATP
ATP
AMP
PPi
Extracellular
Intracellular
ANKH
ABCC6
ENPP1 mutation
CD73
TNAP
ADO
Pi
PPi
Undermineralized bone
Intimal proliferation
Vascular calcification
Vasodilation

ABCC6, ATP-binding cassette sub-family C member 6; ADO, adenosine; AMP, adenosine monophosphate; ANKH, ankylosis homolog; ATP, adenosine triphosphate; CD, cluster of differentiation; ENPP1, ectonucleotide pyrophosphatase/phosphodiesterase 1; Pi, phosphate; PPi, pyrophosphate; TNAP, alkaline phosphatase.

Objective

- To demonstrate correlation between PPi plasma levels with multiple disease endpoints using BMN 401 in murine models of ENPP1 deficiency

Study Design

- Numerous mouse models of ENPP1 deficiency exist. We used 2 models to assess the effect of BMN 401 on PPi levels and symptom manifestation
 - ENPP1$^{asj/asj}$ with acceleration diet (high phosphate/low magnesium to promote mineralization)
 - ENPP1$^{asj\text{-}2J}$ without acceleration diet
- To create a range of plasma PPi levels, 2-week-old mice were treated with varying doses of BMN 401 or vehicle every other day for 8 weeks (**Figure 2**)

Figure 2. Study design

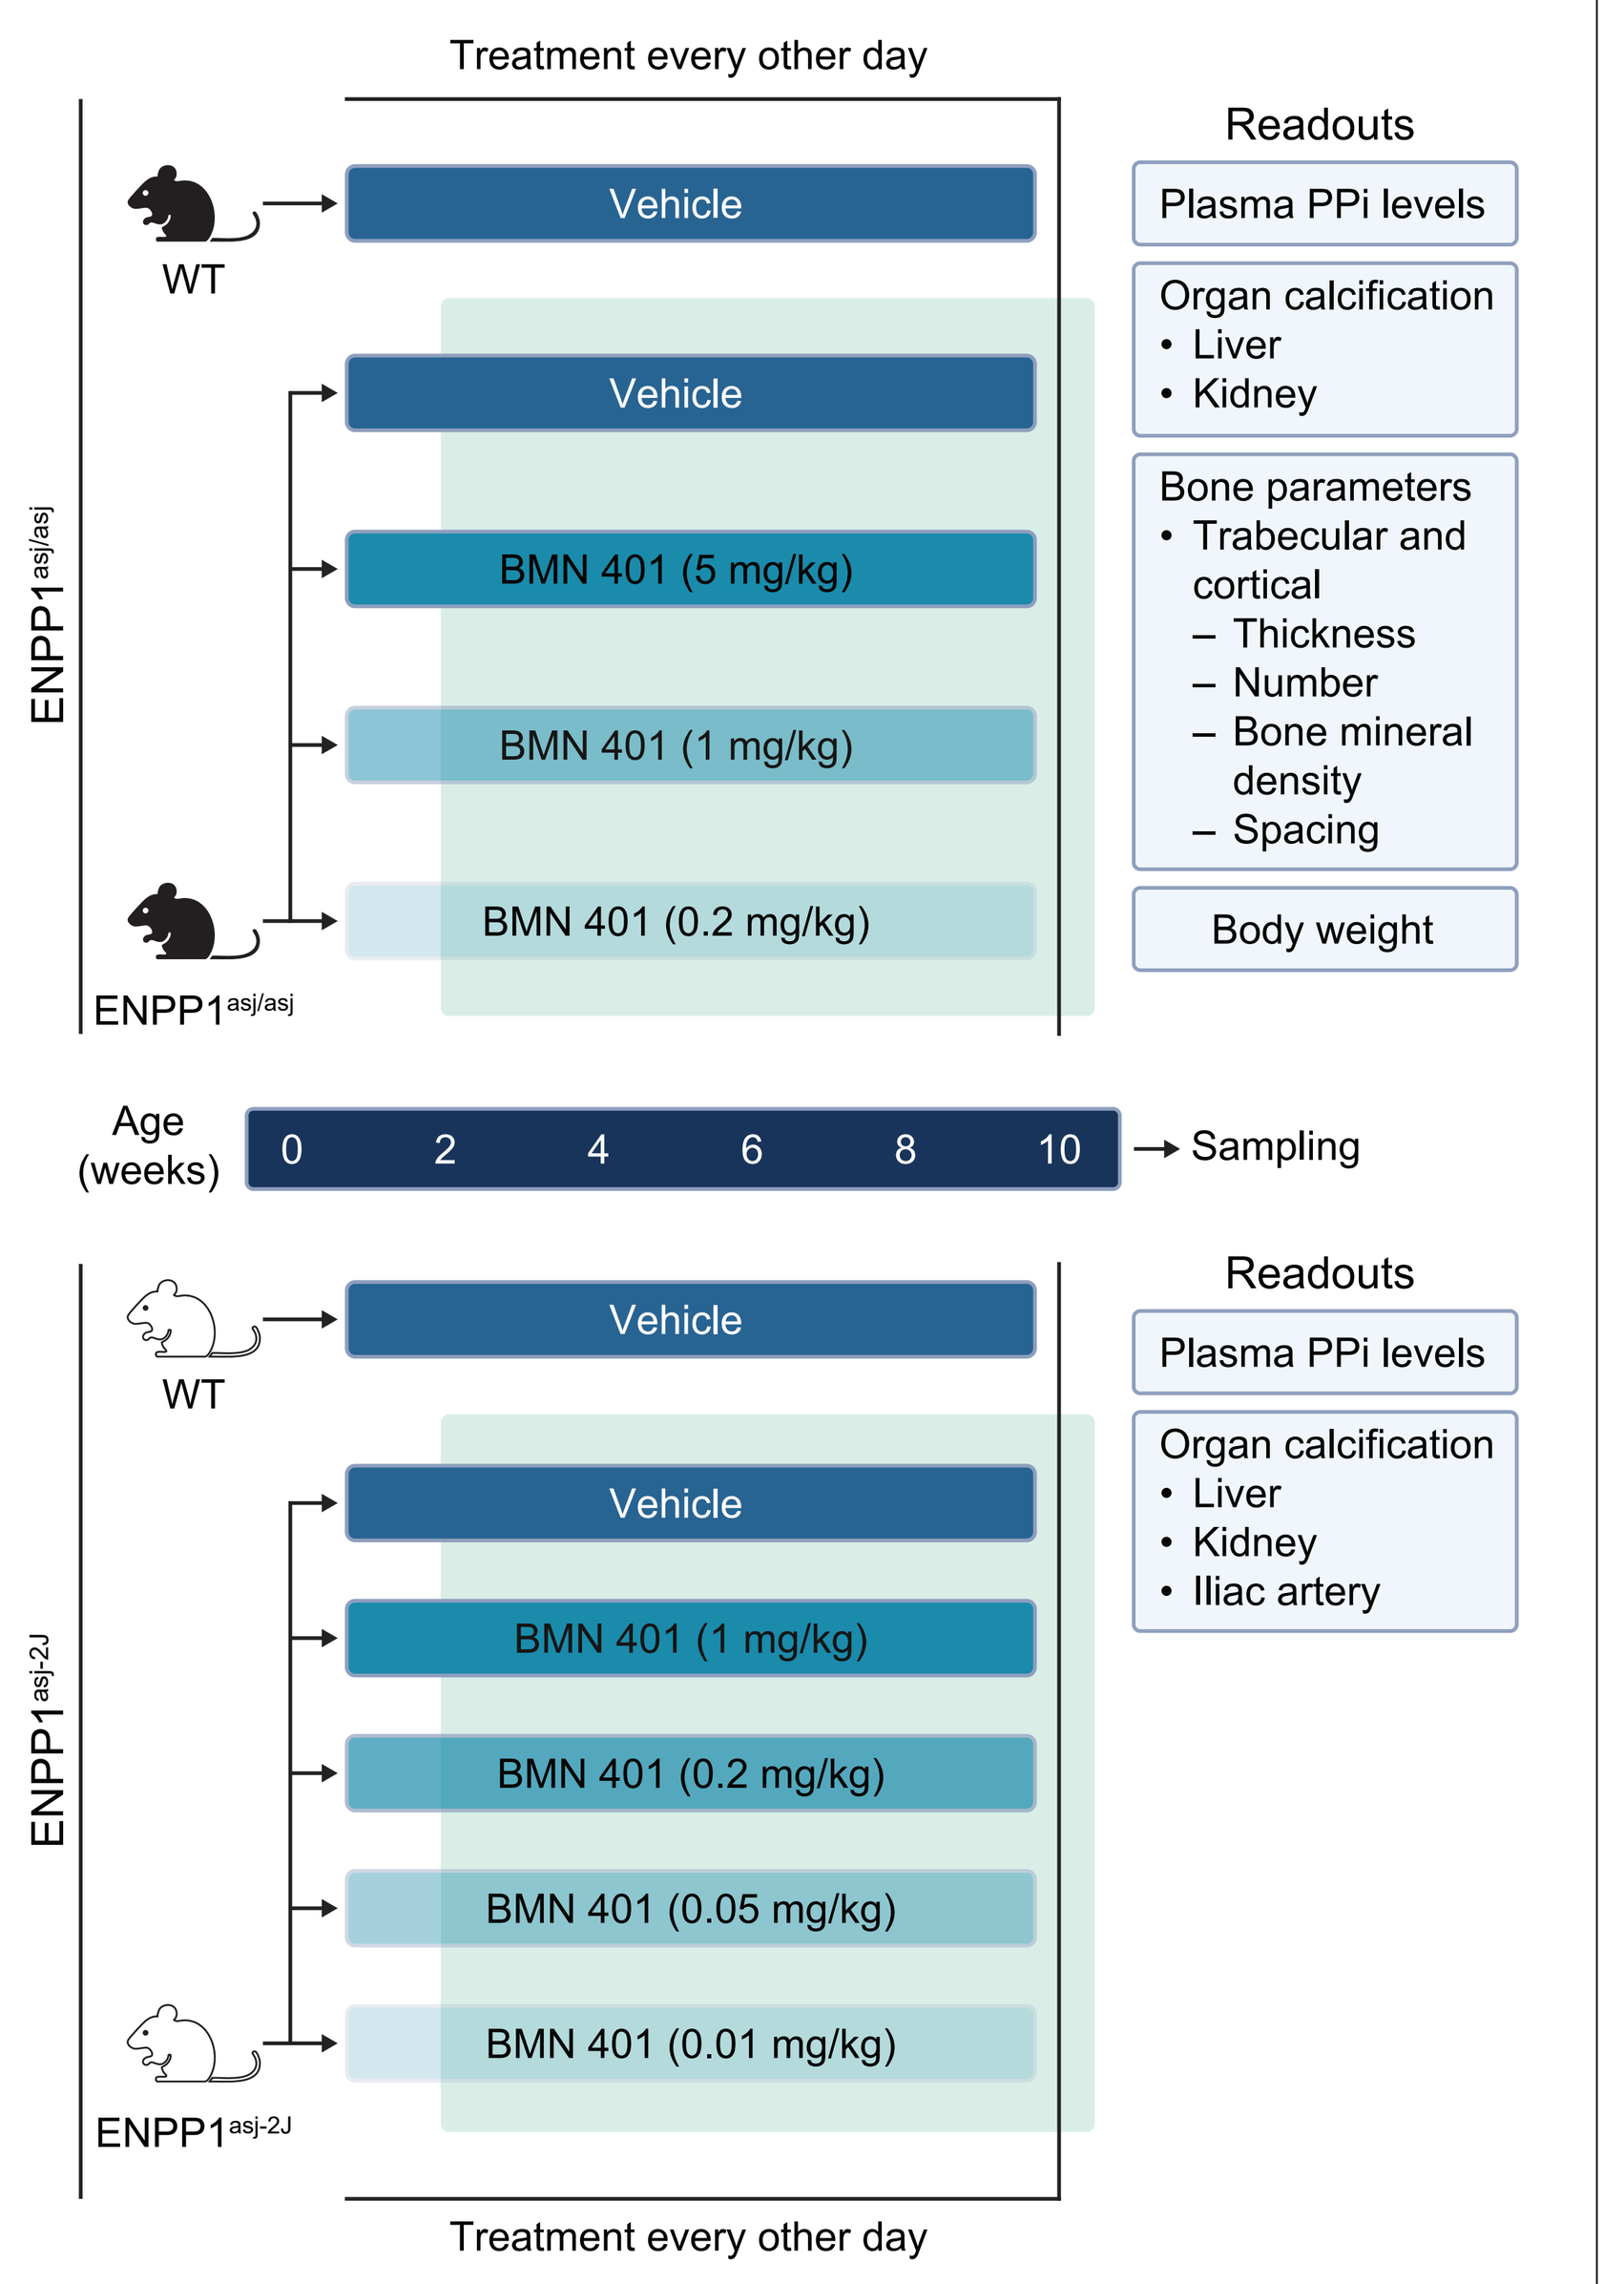
Treatment every other day
Readouts
WT
Vehicle
Vehicle
BMN 401 (5 mg/kg)
BMN 401 (1 mg/kg)
BMN 401 (0.2 mg/kg)
ENPP1asj/asj
Plasma PPi levels
Organ calcification
• Liver
• Kidney
Bone parameters
• Trabecular and cortical
– Thickness
– Number
– Bone mineral density
– Spacing
Body weight
Age (weeks)
0 2 4 6 8 10
Sampling
Readouts
WT
Vehicle
Vehicle
BMN 401 (1 mg/kg)
BMN 401 (0.2 mg/kg)
BMN 401 (0.05 mg/kg)
BMN 401 (0.01 mg/kg)
ENPP1asj-2J
Plasma PPi levels
Organ calcification
• Liver
• Kidney
• Iliac artery
Treatment every other day

asj, ages with stiffened joints; ENPP1, ectonucleotide pyrophosphatase/phosphodiesterase 1; PPi, pyrophosphate; WT, wild type.

- Associations between plasma PPi and outcomes were determined with a Pearson correlation analysis

Results

Calcification

- Plasma PPi level was significantly inversely correlated with the level of calcification in the liver and kidney in dosed ENPP1$^{asj/asj}$ mice (**Figure 3**) and in the liver, kidney, and iliac artery of ENPP1$^{asj\text{-}2J}$ mice (**Figure 4**)

Figure 3. Correlation between PPi level and organ calcification in ENPP1$^{asj/asj}$ mice treated with BMN 401

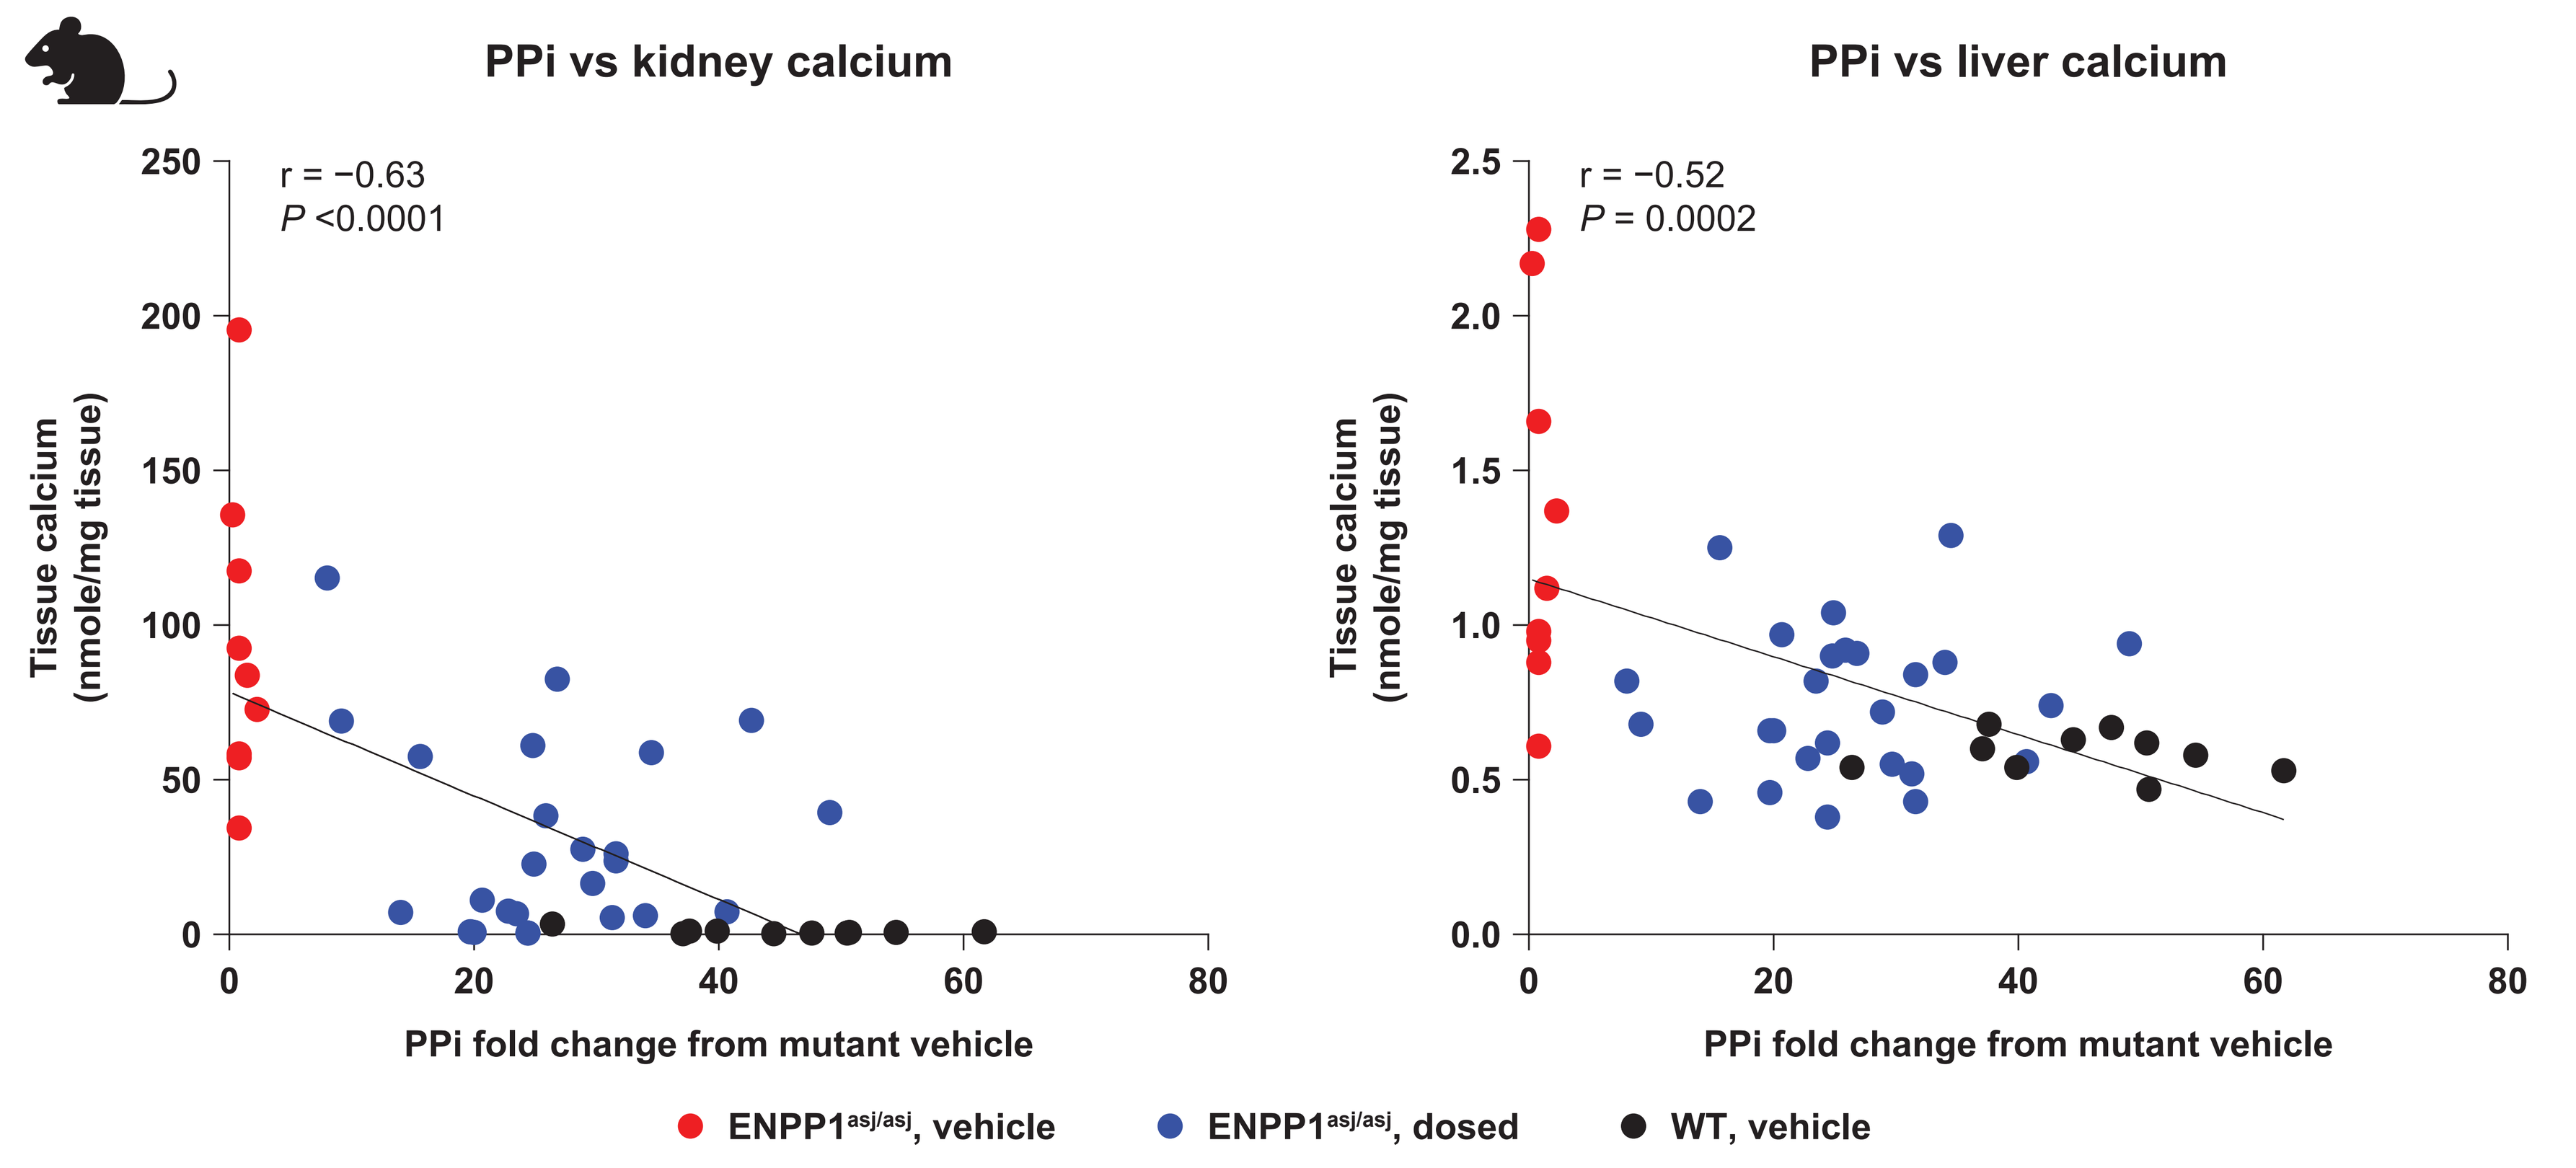
PPi vs kidney calcium
r = −0.63
P <0.0001
PPi vs liver calcium
r = −0.52
P = 0.0002
Tissue calcium (nmole/mg tissue)
PPi fold change from mutant vehicle
ENPP1asj/asj, vehicle
ENPP1asj/asj, dosed
WT, vehicle

asj, ages with stiffened joints; ENPP1, ectonucleotide pyrophosphatase/phosphodiesterase 1; PPi, pyrophosphate; WT, wild type.

Figure 4. Correlation between PPi level and organ calcification in ENPP1$^{asj\text{-}2J}$ mice treated with BMN 401

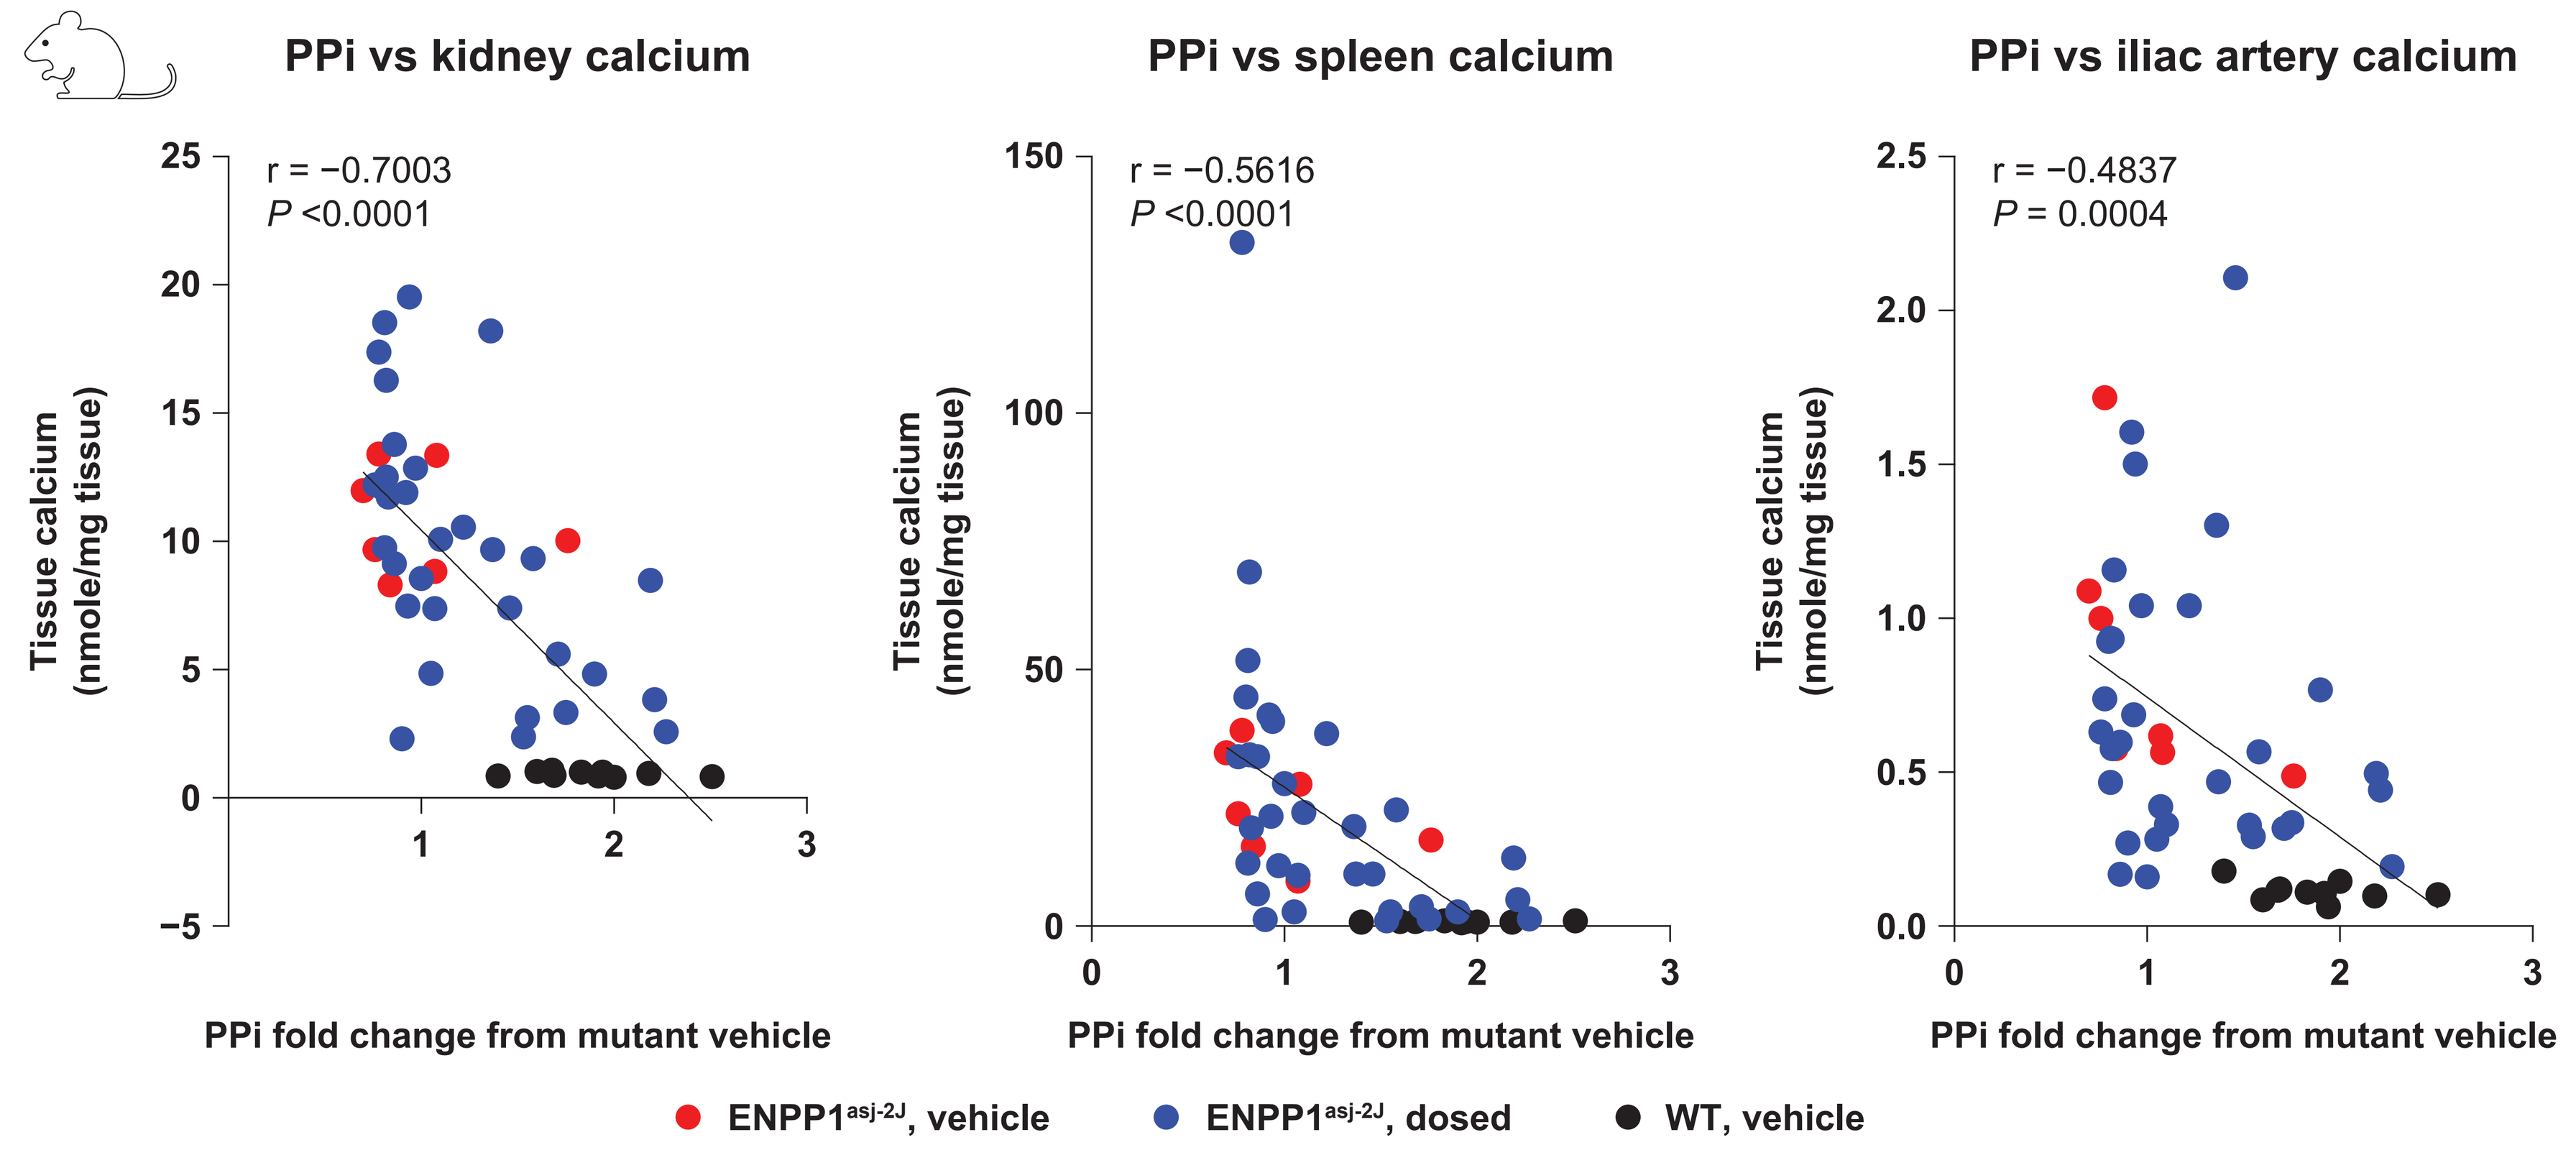
PPi vs kidney calcium
r = −0.7003
P <0.0001
PPi vs spleen calcium
r = −0.5616
P <0.0001
PPi vs iliac artery calcium
r = −0.4837
P = 0.0004
Tissue calcium (nmole/mg tissue)
PPi fold change from mutant vehicle
ENPP1asj-2J, vehicle
ENPP1asj-2J, dosed
WT, vehicle

asj, ages with stiffened joints; ENPP1, ectonucleotide pyrophosphatase/phosphodiesterase 1; PPi, pyrophosphate; WT, wild type.

Bone parameters

- Increasing plasma PPi levels were positively correlated with trabecular and cortical bone parameters (**Figure 5**)

Figure 5. Correlation of PPi level with trabecular and cortical bone parameters in ENPP1$^{asj/asj}$ mice treated with BMN 401

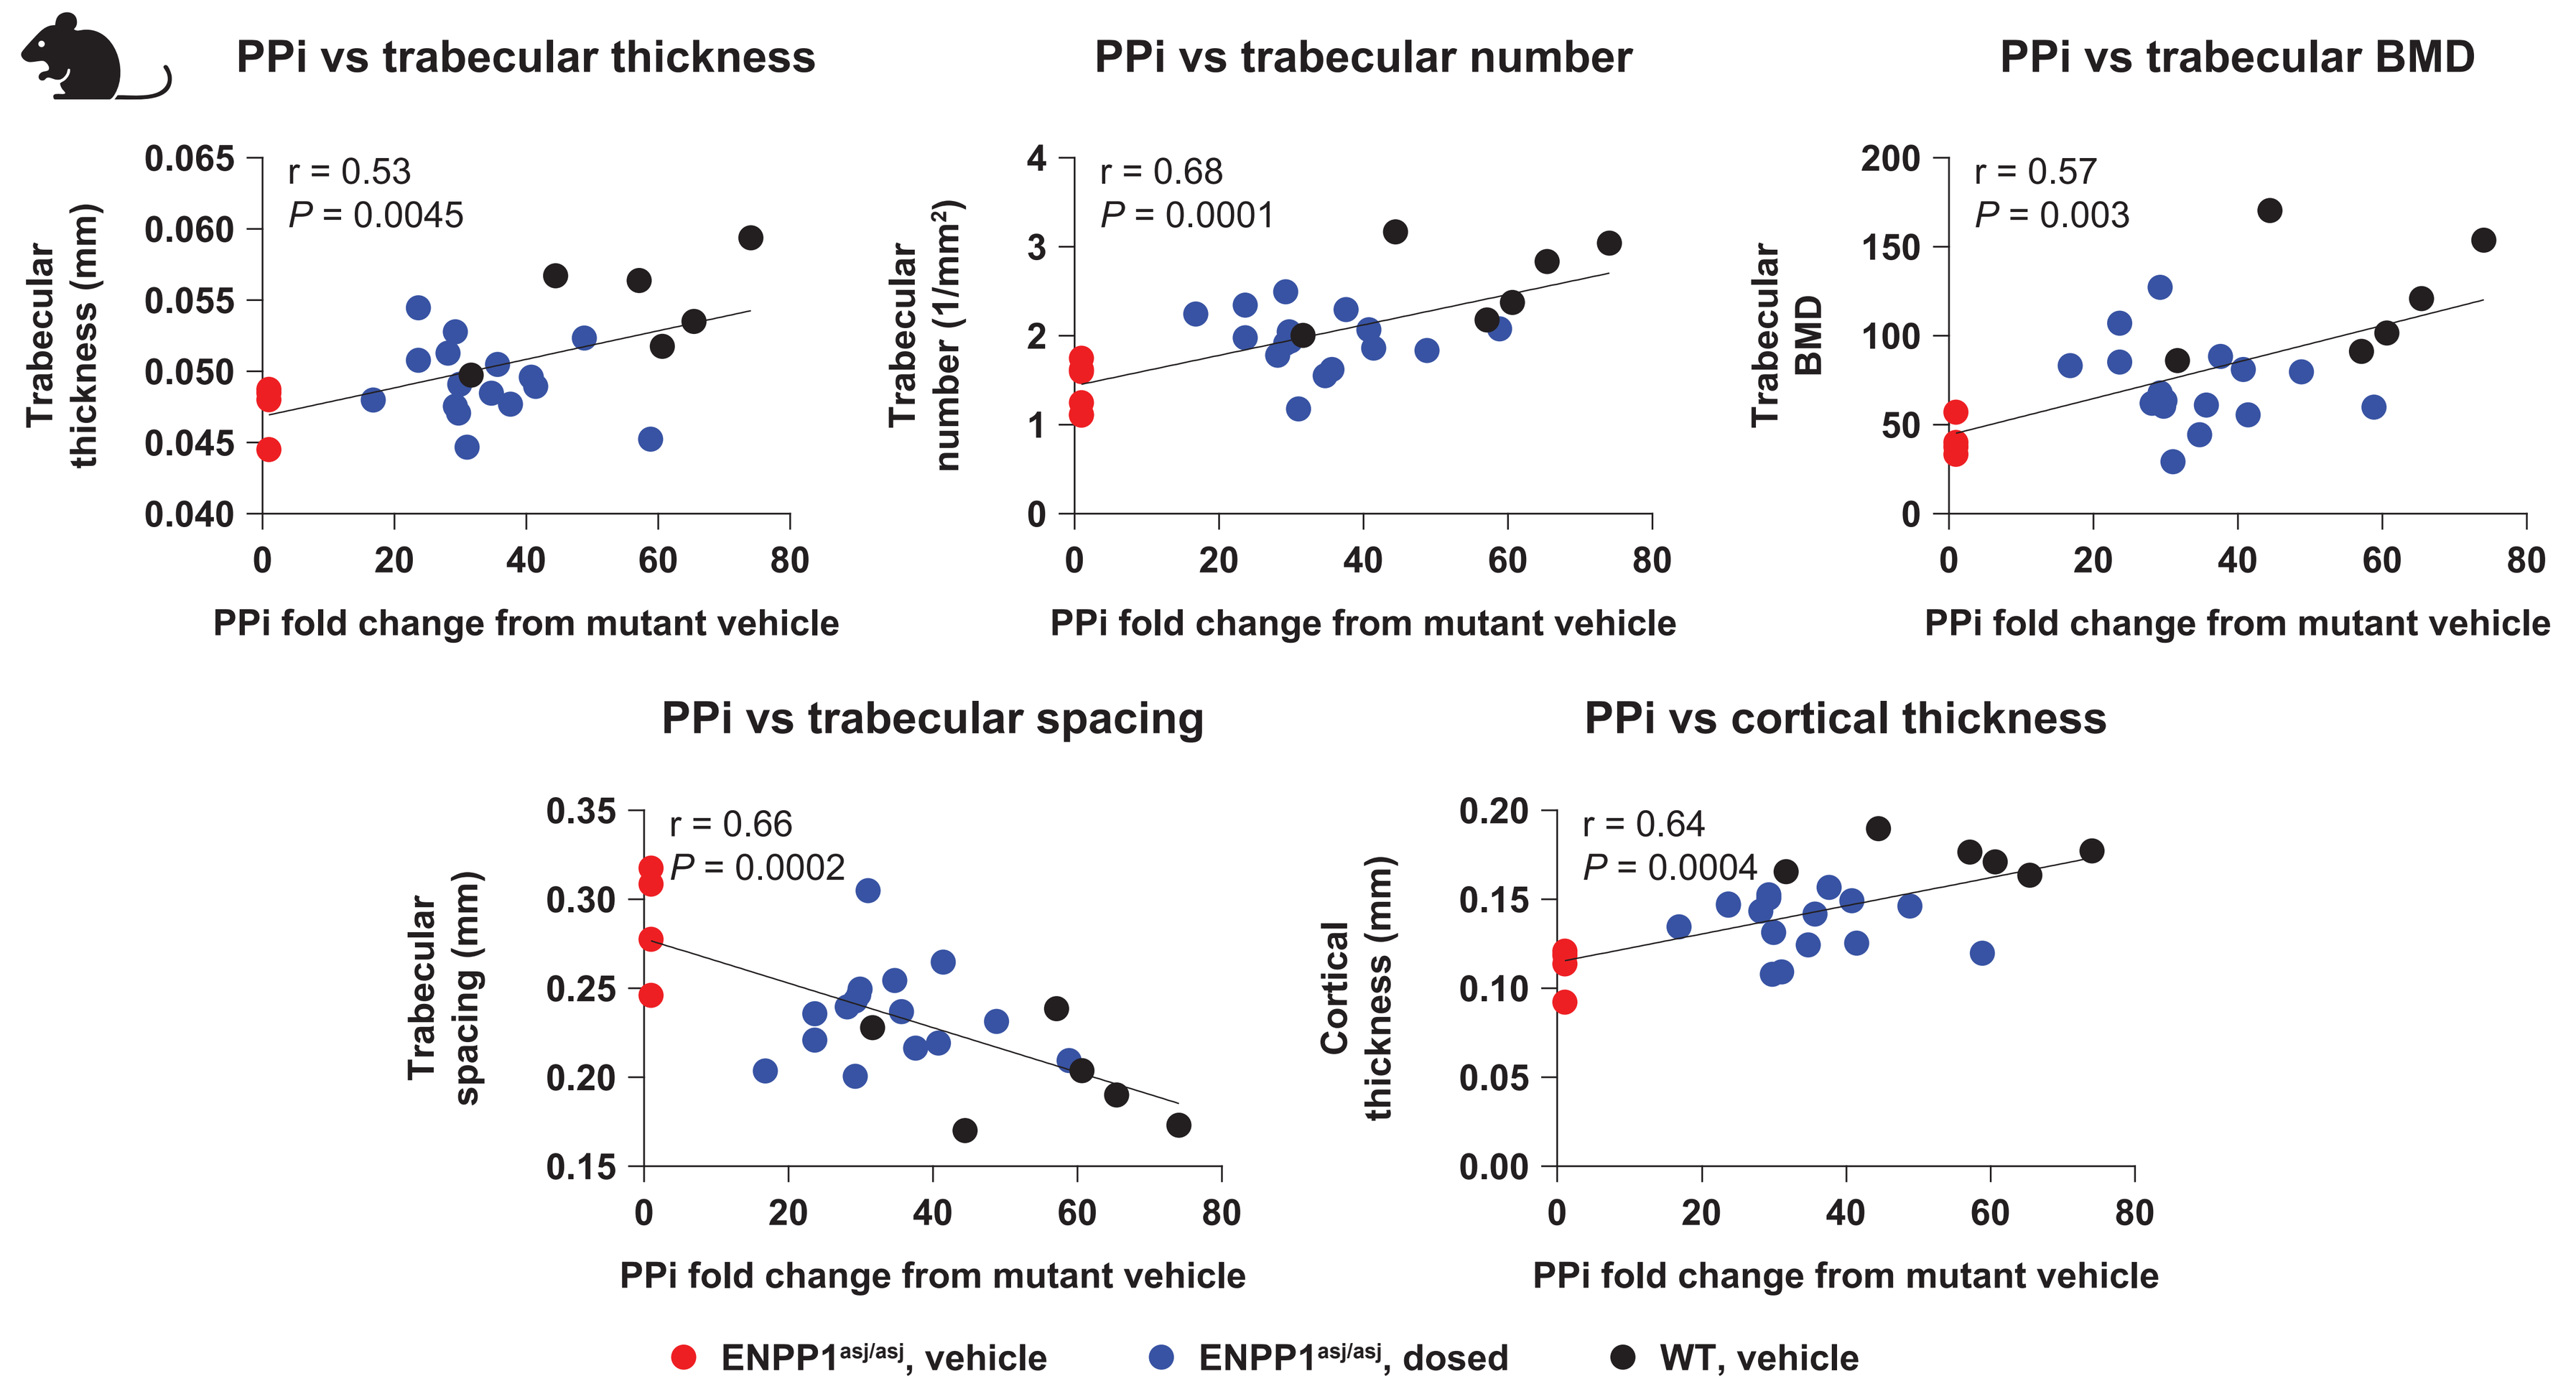
PPi vs trabecular thickness
r = 0.53
P = 0.0045
Trabecular thickness (mm)
PPi vs trabecular number
r = 0.68
P = 0.0001
Trabecular number (1/mm²)
PPi vs trabecular BMD
r = 0.57
P = 0.003
Trabecular BMD
PPi vs trabecular spacing
r = 0.66
P = 0.0002
Trabecular spacing (mm)
PPi vs cortical thickness
r = 0.64
P = 0.0004
Cortical thickness (mm)
PPi fold change from mutant vehicle
ENPP1asj/asj, vehicle
ENPP1asj/asj, dosed
WT, vehicle

asj, ages with stiffened joints; BMD, bone mineral density; ENPP1, ectonucleotide pyrophosphatase/phosphodiesterase 1; PPi, pyrophosphate; WT, wild type.

Body weight

- Plasma PPi positively correlated with body weight (**Figure 6**)

Figure 6. Correlation between PPi and body weight in ENPP1 deficiency murine models treated with BMN 401

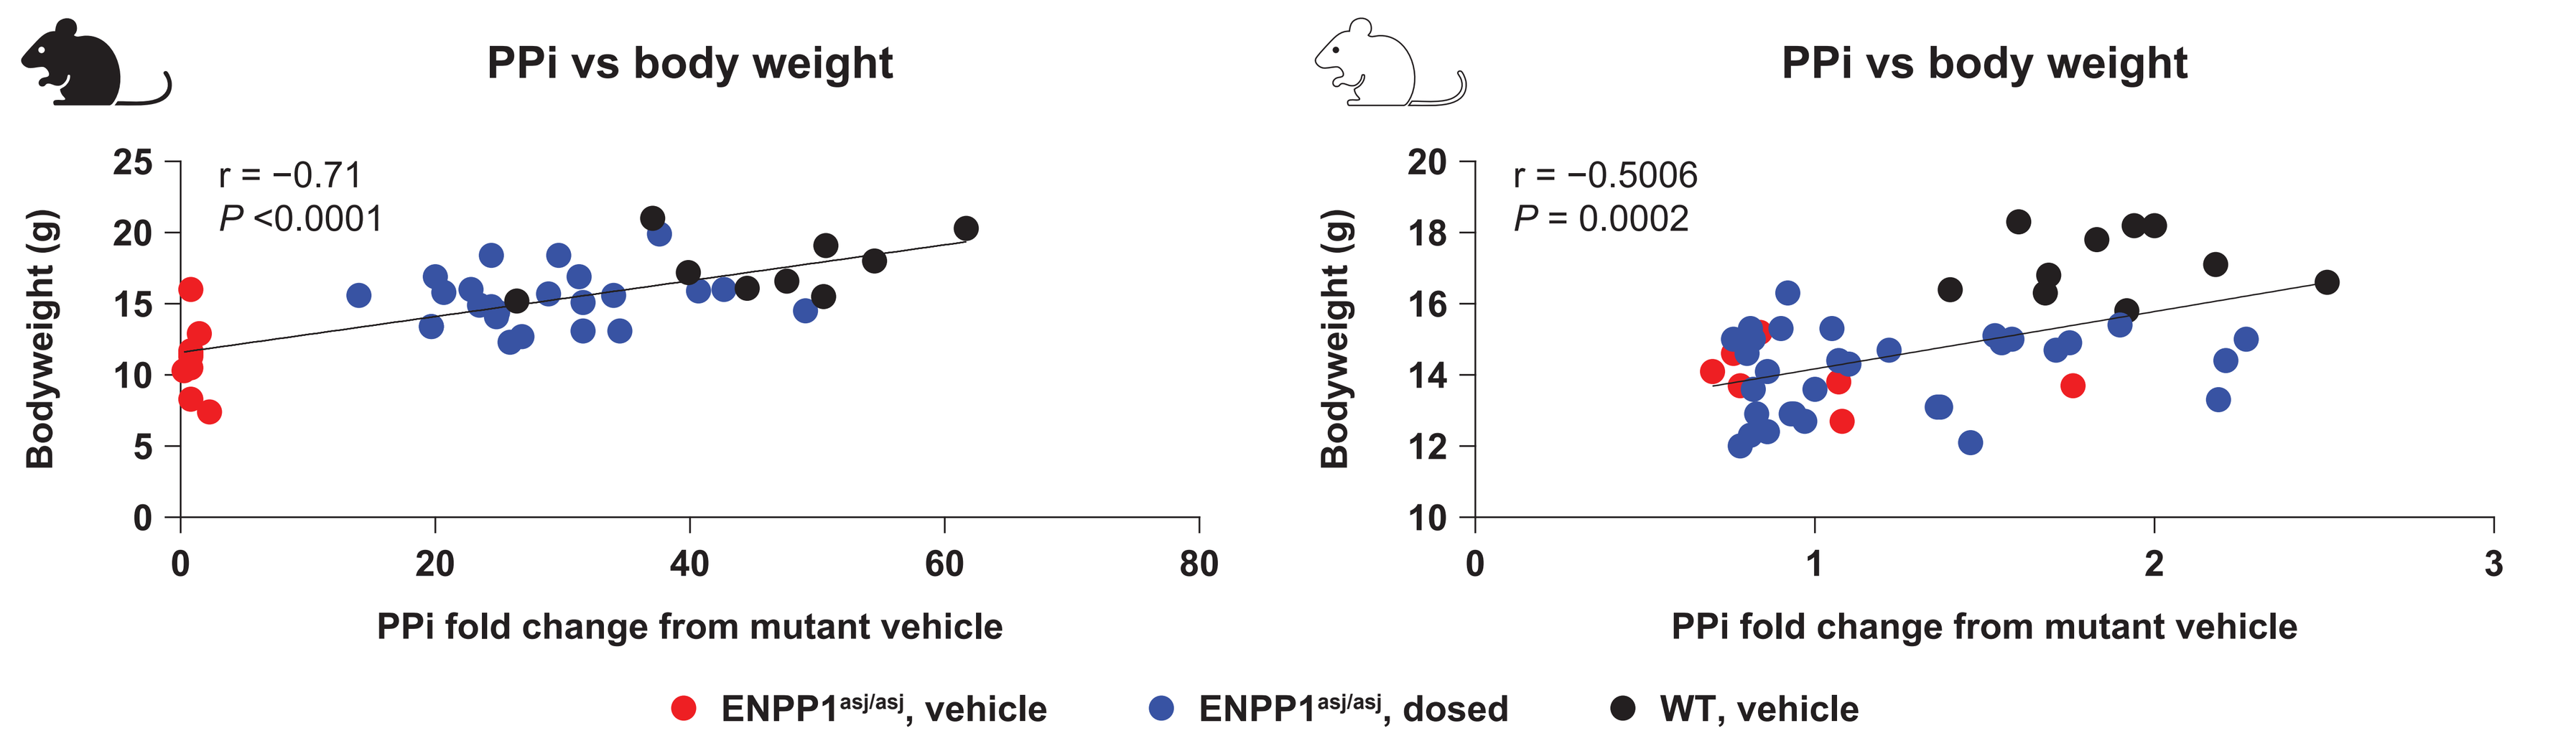
PPi vs body weight
r = −0.71
P <0.0001
PPi vs body weight
r = −0.5006
P = 0.0002
Bodyweight (g)
PPi fold change from mutant vehicle
ENPP1asj/asj, vehicle
ENPP1asj/asj, dosed
WT, vehicle

asj, ages with stiffened joints; ENPP1, ectonucleotide pyrophosphatase/phosphodiesterase 1; PPi, pyrophosphate; WT, wild type.

Conclusions

- Improvements in plasma PPi levels are reflective of a reduction in vascular calcification and restoration of bone abnormalities
- These data provide crucial evidence indicating that maintaining PPi within the normal range is important
- Plasma PPi is significantly positively correlated with improvements in bone parameters and significantly inversely correlated with vascular and tissue calcification
- PPi is a key pharmacodynamic marker of efficacy to assess enzyme replacement therapy in disorders like ENPP1 deficiency characterized by low PPi

References

1. Ferreira CR, et al. *JBMR Plus*. 2025;9(5):ziaf019
2. Ferreira CR, et al. *Ann Rev Pathol Mech Dis*. 2024;19:507–40
3. Ralph D, et al. *Am J Pathol*. 2022;192:762–70
4. Orriss IR, et al. *Curr Opin Pharmacol*. 2016;28:57–68
5. Hamczyk MR, et al. *Aging*. 2018;10(12):3652–3
6. Guieu R, et al. *J Clin Med*. 2020;9:366
7. Cheng Z, et al. *J Bone Miner Res*. 2021;36:1594–604
8. Nitschke Y, et al. *Exp Mol Med*. 2018:50(10):1–12
9. Bernhard E, et al. *J Clin Endo Metab*. 2022;107:109–18

Acknowledgments

Medical writing support was provided by Morgan Engelhart Wynn, PhD, of Red Nucleus, and funded by BioMarin Pharmaceutical Inc. Project management support was provided by Abigail Hunt, PhD, of BioMarin Pharmaceutical Inc.

Disclosures

As of July 1, 2025, Inozyme Pharma is a wholly owned subsidiary of BioMarin Pharmaceutical Inc.

KOB, **CS**, **JH**, **DS**, **HH**, and **YS** were employees and stockholders of Inozyme Pharma at the time of data generation and analysis.

Presented at American Society for Bone and Mineral Research (ASBMR) annual meeting: September 5–8, 2025; Seattle, WA, USA

©2025 BioMarin Pharmaceutical Inc. All rights reserved.