Patient and caregiver insights on the real-world burden of the rare genetic disorders, ENPP1 deficiency and ABCC6 deficiency

Liz Molloy[1], Christine O'Brien[1], Jelena Garafalo[2], Katie Swanner[2], Catherine Nester[2], Renuka Ramakrishnan[3], Amish Chinoy[4], M. Zulf Mughal[5]

[1]GACI Global, Argyle, TX; [2]Inozyme Pharma, Boston, MA; [3]Alder Hey Children's NHS Foundation Trust, Liverpool, UK; [4]Royal Manchester Children's Hospital, Manchester, UK; [5]Al Jalila Children's Speciality Hospital, Dubai, UAE

Poster #
Sat-352

Introduction

- Ectonucleotide pyrophosphatase/phosphodiesterase 1 (ENPP1) and ATP-binding cassette subfamily C member 6 (ABCC6) are proteins involved in the pyrophosphate (PPi)-adenosine pathway[1]
 - PPi is necessary for the regulation of bone mineralization[2] and prevention of soft tissue calcification[3]
 - Adenosine maintains blood flow by preventing stenosis of blood vessels[4]
- ENPP1 and ABCC6 deficiencies are rare genetic conditions that cause lifelong, multisystem symptoms related to impaired bone health and blood vessel function[5]
- Generalized arterial calcification of infancy (GACI), the infant-onset phenotype of ENPP1 (GACI type 1) or ABCC6 (GACI type 2) deficiency (**Figure 1**), is characterized by widespread vascular calcification, cardiovascular morbidity, and high infant mortality[1,6,7]

Figure 1. Phenotypes of ABCC6 and ENPP1 deficiencies by age

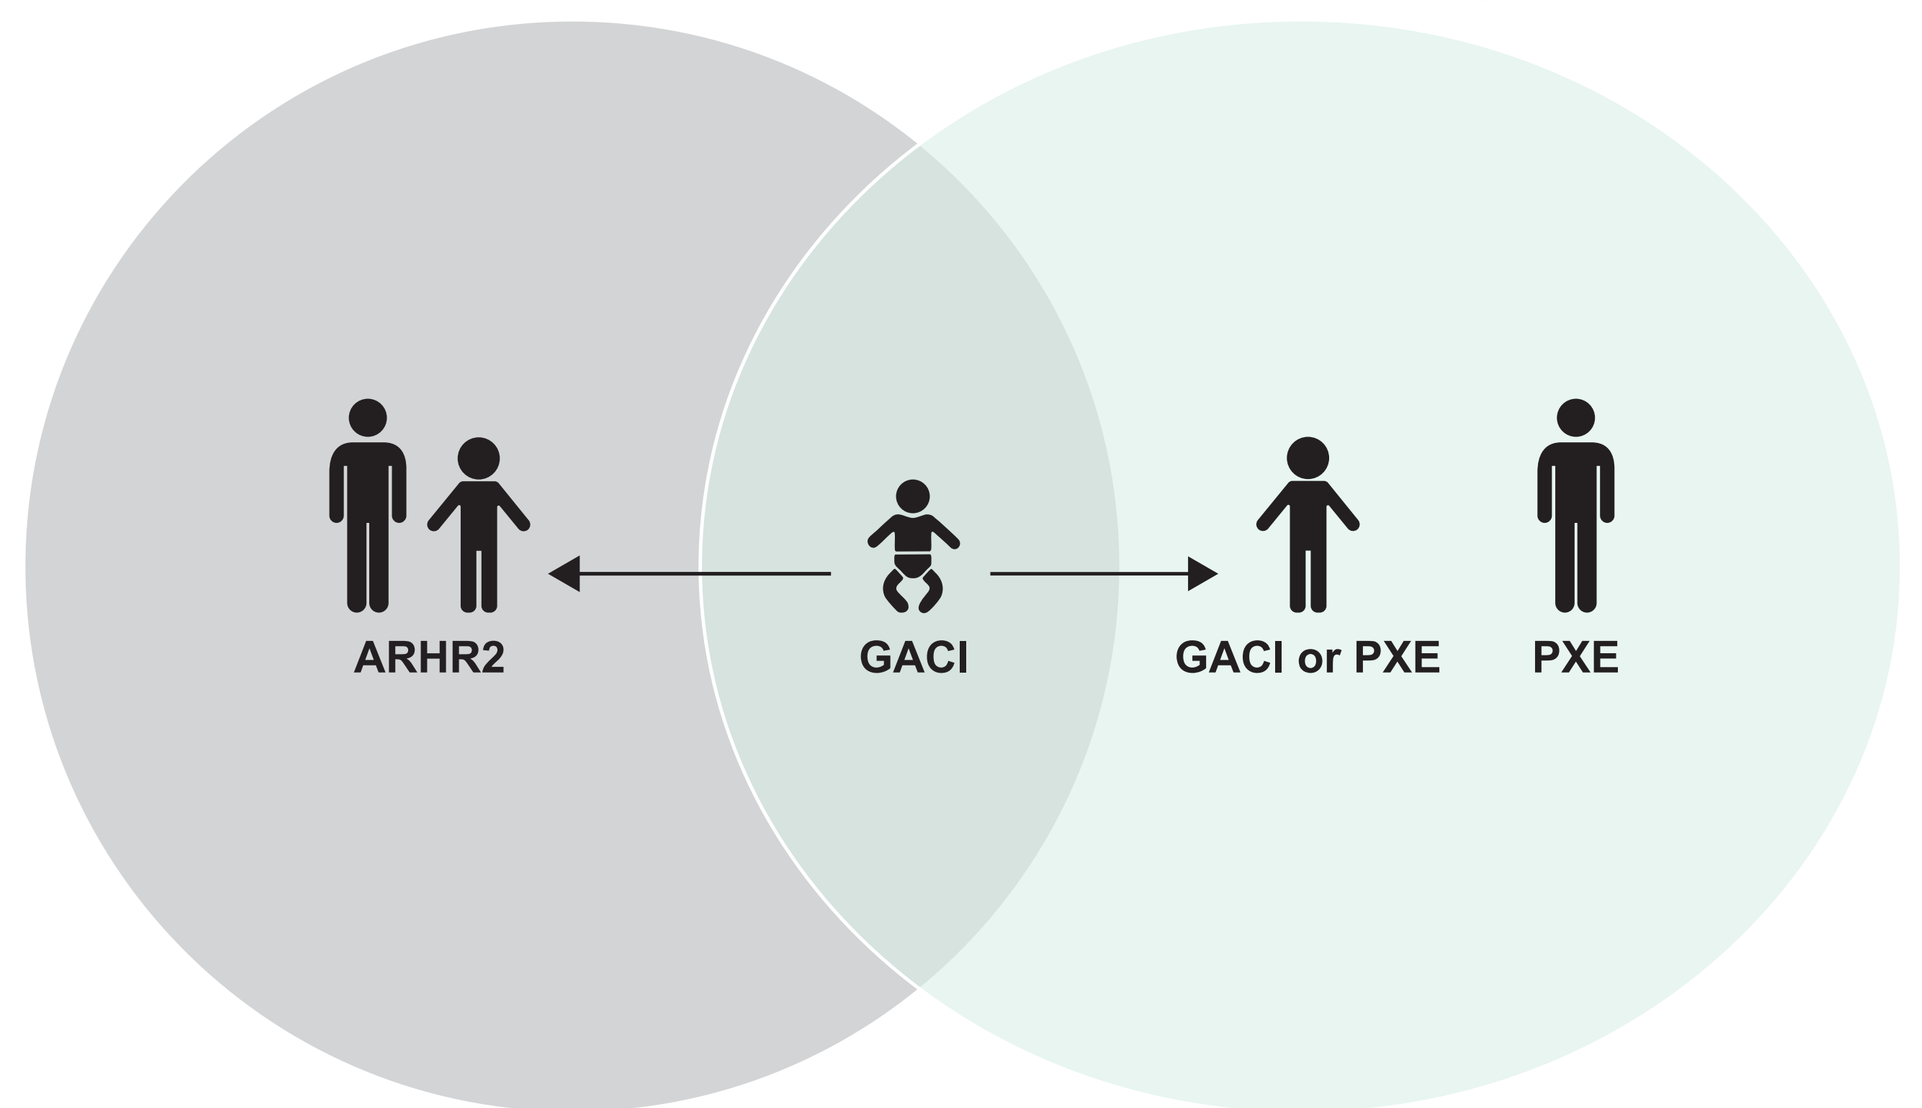
ENPP1 deficiency
ABCC6 deficiency
ARHR2
GACI
GACI or PXE
PXE

ABCC6, ATP-binding cassette subfamily C member 6; ARHR2, autosomal recessive hypophosphatemic rickets type 2; ENPP1, ectonucleotide pyrophosphatase/phosphodiesterase 1; GACI, generalized arterial calcification of infancy; PXE, pseudoxanthoma elasticum.

Objective

- Capture the real-world burden of ENPP1 and ABCC6 deficiencies on the daily lives of patients affected by the condition and their families and understand the therapeutic unmet needs

Study design

- A protocol-guided anonymous digital survey was deployed through Qualtrics® to the GACI Global community, a patient advocacy organization
- Eligible participants were patients affected by GACI/ARHR2
 - For patients aged 13 years and older, both the patient and their caregiver were invited to participate in the survey
 - For patients under the age of 13 years, only their caregiver could complete the survey
- The survey focused on 4 main topics: patient background, patient diagnosis and disease management background, patient and caregiver disease burden, and unmet needs and future therapy expectations

Results

Respondent demographics

- Between November 2024 and March 2025, 18 patients and 33 caregivers completed the survey (**Table 1**)

Table 1. Respondent demographics

		Percent of respondents (N = 51)
Respondent type	Caregiver only	63%
	Patient only	25%
	Patient under caregiver supervision	12%
Genetic diagnosis*	GACI type 1 (ENPP1 deficiency)	16%
	GACI type 2 (ABCC6 deficiency)	29%
	GACI type 1 and ARHR2 (ENPP1 deficiency)	47%
	ARHR2 (ENPP1 deficiency)	8%
Patient age, y*	Prenatal to <6 months	4%
	1 to <2	2%
	2 to <7	25%
	7 to <13	27%
	13 to <18	16%
	18 to <25	6%
	25+	20%
Patient sex*	Female	71%
	Male	29%
Age at diagnosis*	Prenatal	24%
	Birth to <1 y	35%
	1 to <13 y	33%
	13 to <18 y	0%
	>18 y	8%

*Includes caregivers' responses.
ABCC6, ATP-binding cassette subfamily C member 6; ARHR2, autosomal recessive hypophosphatemic rickets type 2; ENPP1, pyrophosphatase/phosphodiesterase family member 1; GACI, generalized arterial calcification of infancy.

HCP first consulted

- Patients with ENPP1 and ABCC6 deficiency first consulted a variety of different healthcare professionals (HCPs) with varying time to diagnosis after professional consultation (**Figure 2**)

Figure 2. Type of HCPs consulted before disease diagnosis and time to diagnosis

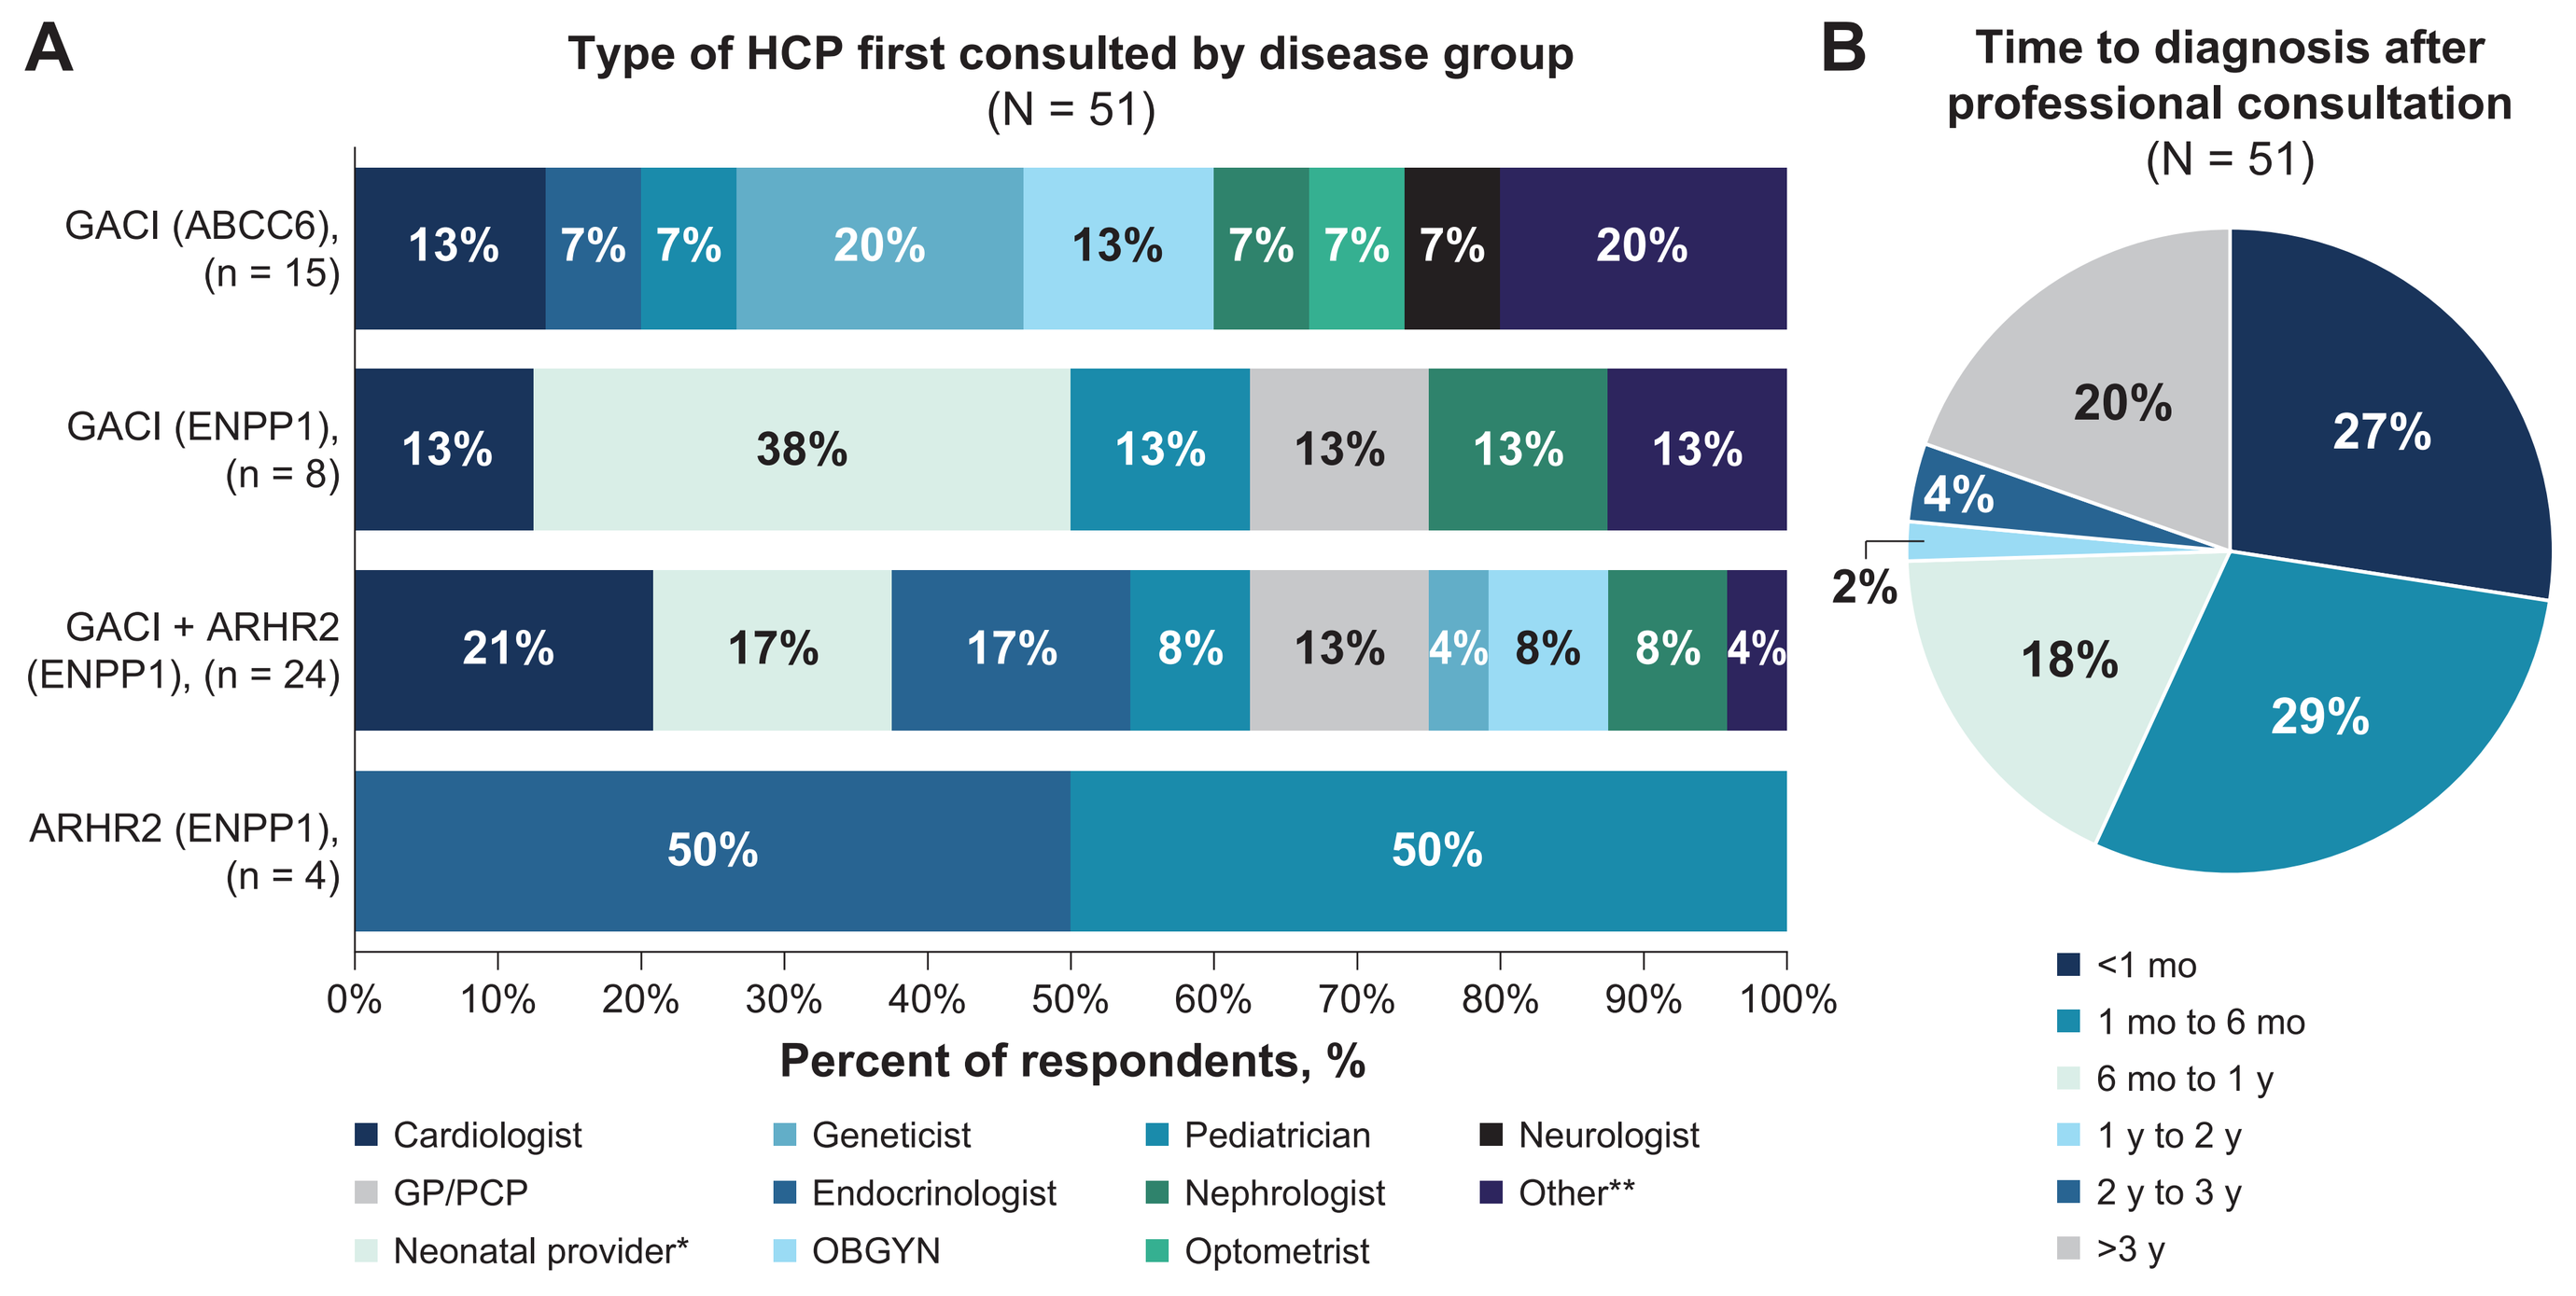
A
Type of HCP first consulted by disease group
(N = 51)
GACI (ABCC6), (n = 15)
13% 7% 7% 20% 13% 7% 7% 7% 20%
GACI (ENPP1), (n = 8)
13% 38% 13% 13% 13% 13%
GACI + ARHR2 (ENPP1), (n = 24)
21% 17% 17% 8% 13% 4% 8% 8% 4%
ARHR2 (ENPP1), (n = 4)
50% 50%
0% 10% 20% 30% 40% 50% 60% 70% 80% 90% 100%
Percent of respondents, %
Cardiologist
Geneticist
Pediatrician
Neurologist
GP/PCP
Endocrinologist
Nephrologist
Other**
Neonatal provider*
OBGYN
Optometrist
B
Time to diagnosis after professional consultation
(N = 51)
27%
29%
18%
2%
4%
20%
<1 mo
1 mo to 6 mo
6 mo to 1 y
1 y to 2 y
2 y to 3 y
>3 y

*Neonatal providers include professionals such as neonatologists, neonatal intensivists or neonatal nurse practitioner.
**Other HCPs: perinatologist, intensivist, emergency department HCP.
ABCC6, ATP-binding cassette subfamily C member 6; ARHR2, autosomal recessive hypophosphatemic rickets type 2; ENPP1, ectonucleotide pyrophosphatase/phosphodiesterase 1; GACI, generalized arterial calcification of infancy; GP, general practitioner; HCP, healthcare professional; OBGYN, obstetrics and gynecology; PCP, primary care provider.

HCP involvement in disease management

- Current disease management for patients aged 2 to <18 years of age primarily involved endocrinologists (85%) and pediatricians (86%)
- Patients with GACI + ARHR2 or ARHR2 were primarily overseen by endocrinologists
- Patients with GACI type 1 or type 2 were more often managed by cardiologists or pediatricians

ENPP1 and ABCC6 deficiency symptom burden

- ENPP1 and ABCC6 deficiencies have both unique and overlapping symptoms (**Figure 3**)

Figure 3. Top symptoms currently experienced by patients with ENPP1 or ABCC6 deficiency

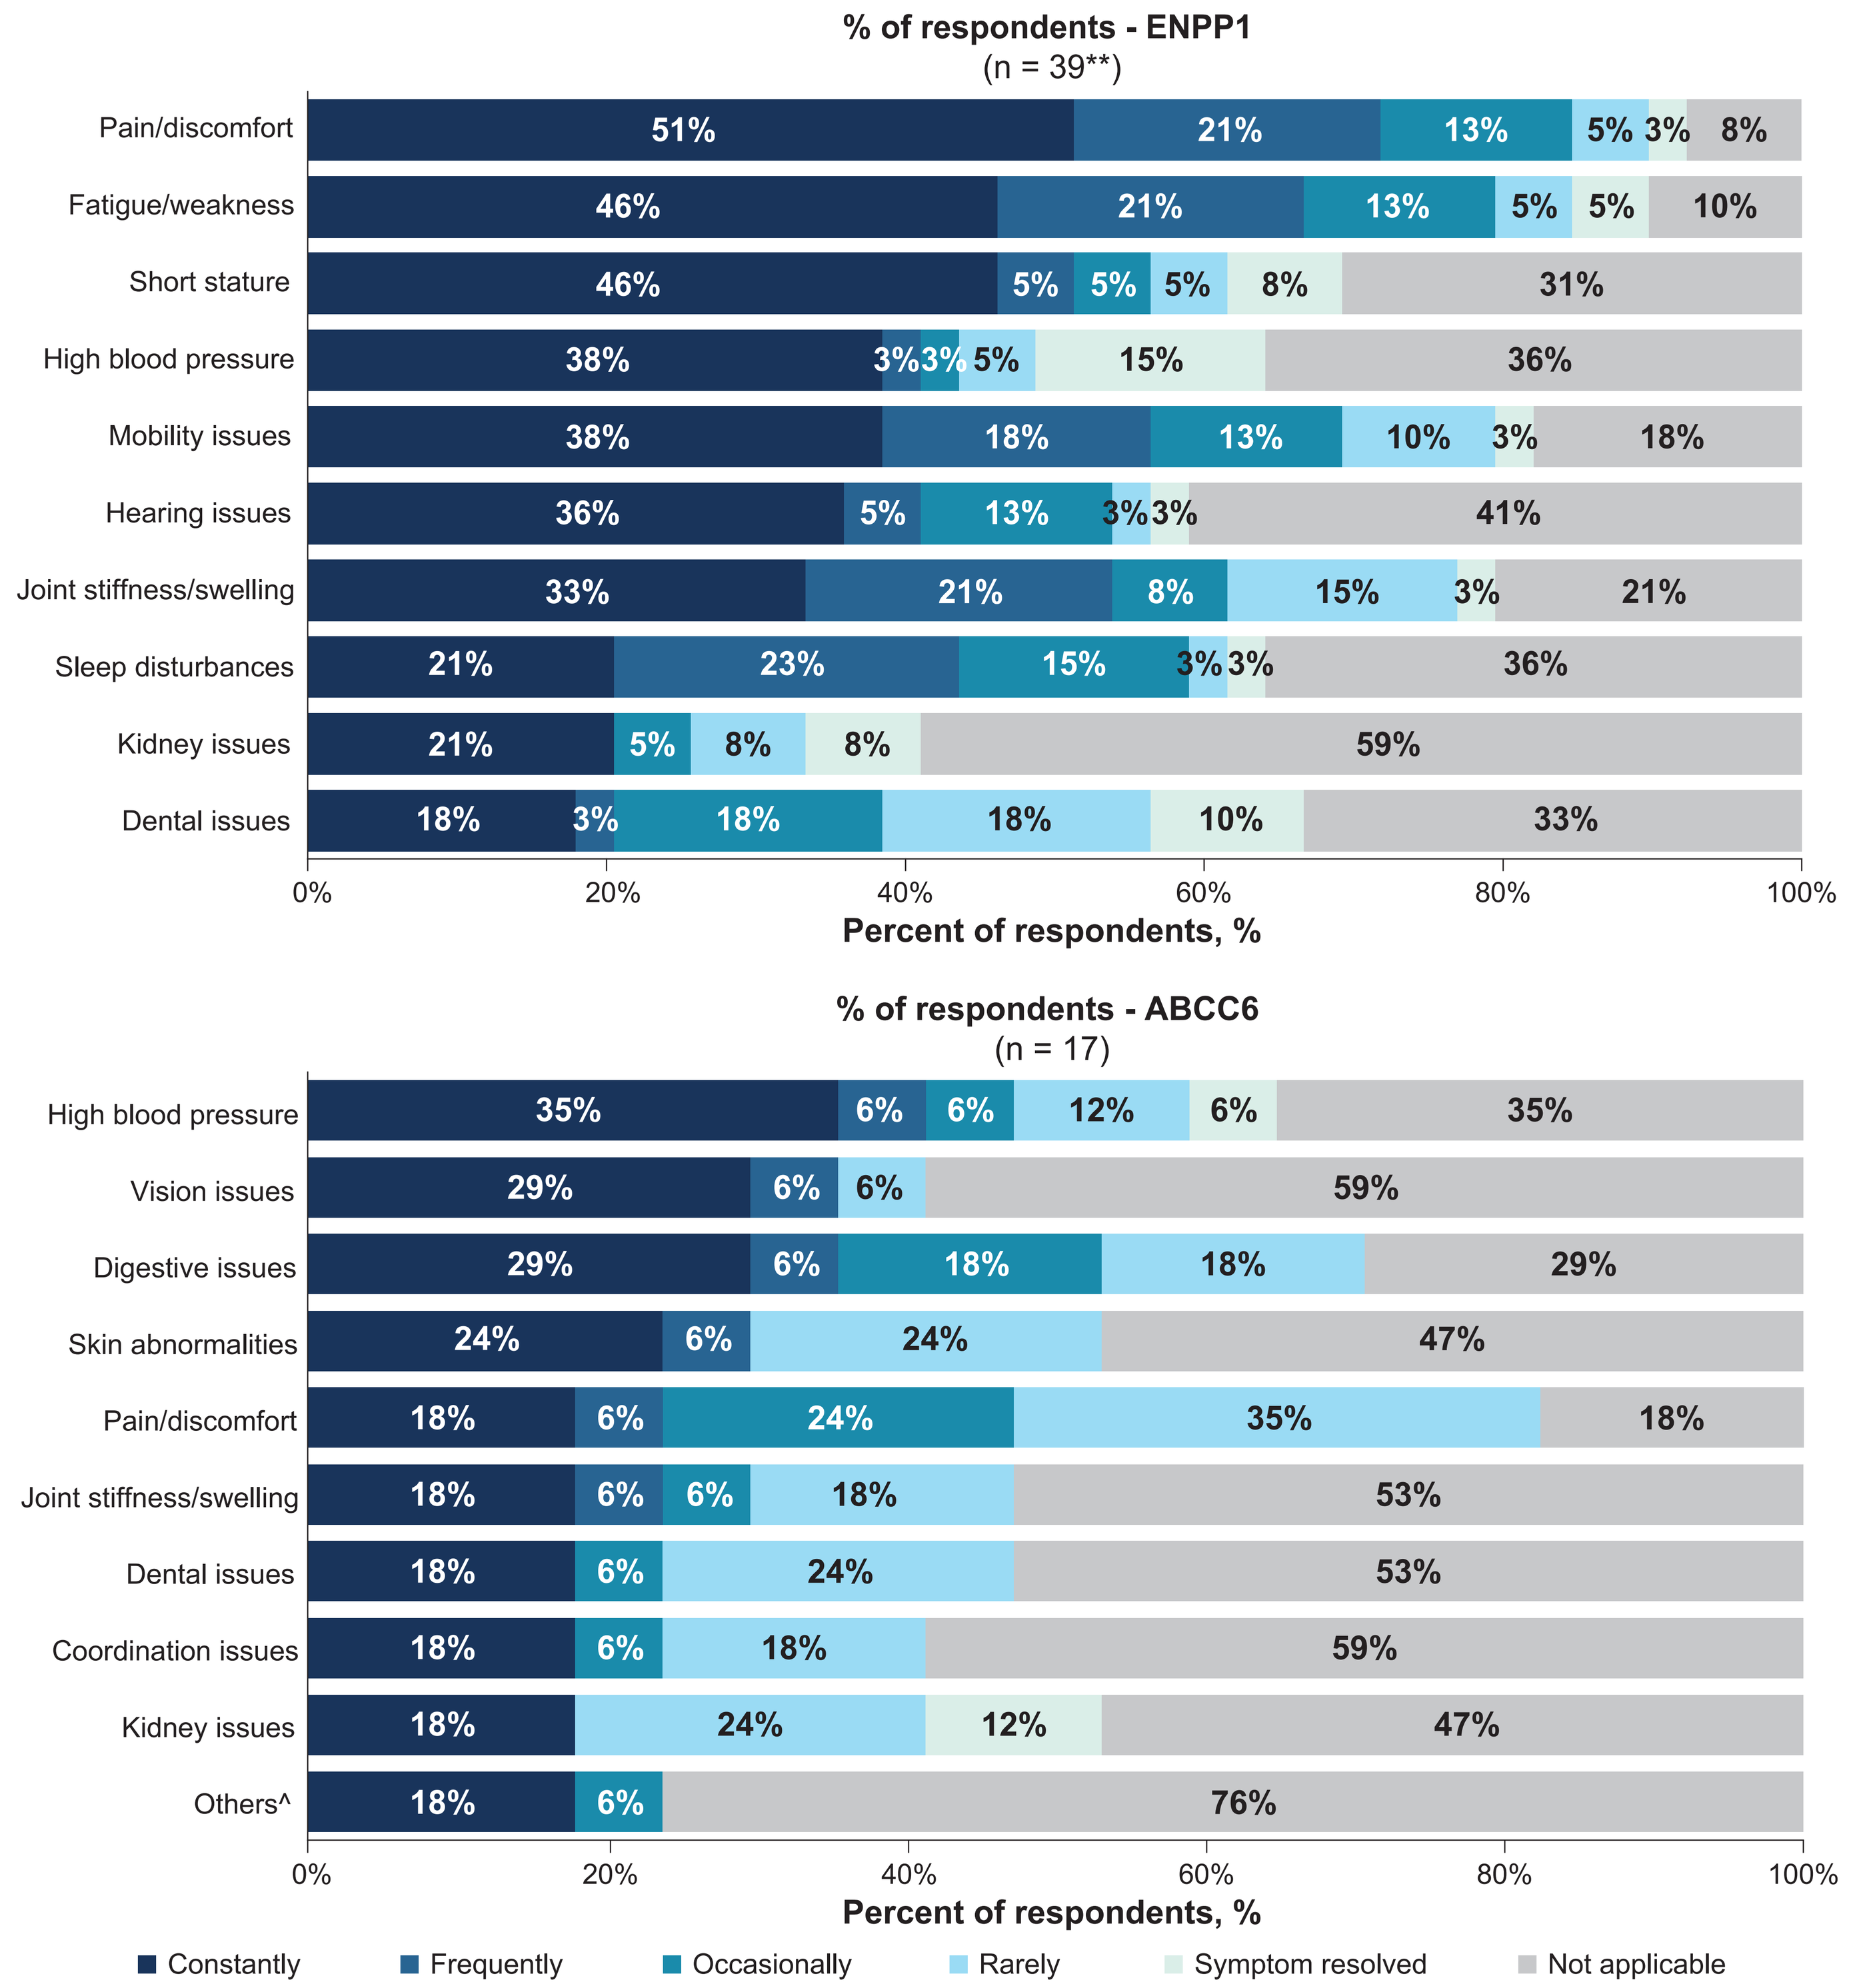
Top 10 symptoms by frequency (all disease groups) – patients and caregivers*
% of respondents - ENPP1
(n = 39**)
Pain/discomfort 51% 21% 13% 5% 3% 8%
Fatigue/weakness 46% 21% 13% 5% 5% 10%
Short stature 46% 5% 5% 5% 8% 31%
High blood pressure 38% 3% 3% 5% 15% 36%
Mobility issues 38% 18% 13% 10% 3% 18%
Hearing issues 36% 5% 13% 3% 3% 41%
Joint stiffness/swelling 33% 21% 8% 15% 3% 21%
Sleep disturbances 21% 23% 15% 3% 3% 36%
Kidney issues 21% 5% 8% 8% 59%
Dental issues 18% 3% 18% 18% 10% 33%
0% 20% 40% 60% 80% 100%
Percent of respondents, %
% of respondents - ABCC6
(n = 17)
High blood pressure 35% 6% 6% 12% 6% 35%
Vision issues 29% 6% 6% 59%
Digestive issues 29% 6% 18% 18% 29%
Skin abnormalities 24% 6% 24% 47%
Pain/discomfort 18% 6% 24% 35% 18%
Joint stiffness/swelling 18% 6% 6% 18% 53%
Dental issues 18% 6% 24% 53%
Coordination issues 18% 6% 18% 59%
Kidney issues 18% 24% 12% 47%
Others^ 18% 6% 76%
0% 20% 40% 60% 80% 100%
Percent of respondents, %
Constantly
Frequently
Occasionally
Rarely
Symptom resolved
Not applicable

*Includes both patient and caregiver responses from respondents who answered the survey together (n = 6).
**N = 1 did not respond to this question.
^Others includes heart murmur, speech issues, and anxiety.
ABCC6, ATP-binding cassette subfamily C member 6; ENPP1, ectonucleotide pyrophosphatase/phosphodiesterase 1.

Disease management

- Both ENPP1 and ABCC6 deficiencies require substantial pharmacological and nonpharmacological medical interventions and devices, pointing toward significant disease burden (**Figure 4**)

Figure 4. Pharmacological and nonpharmacological disease management

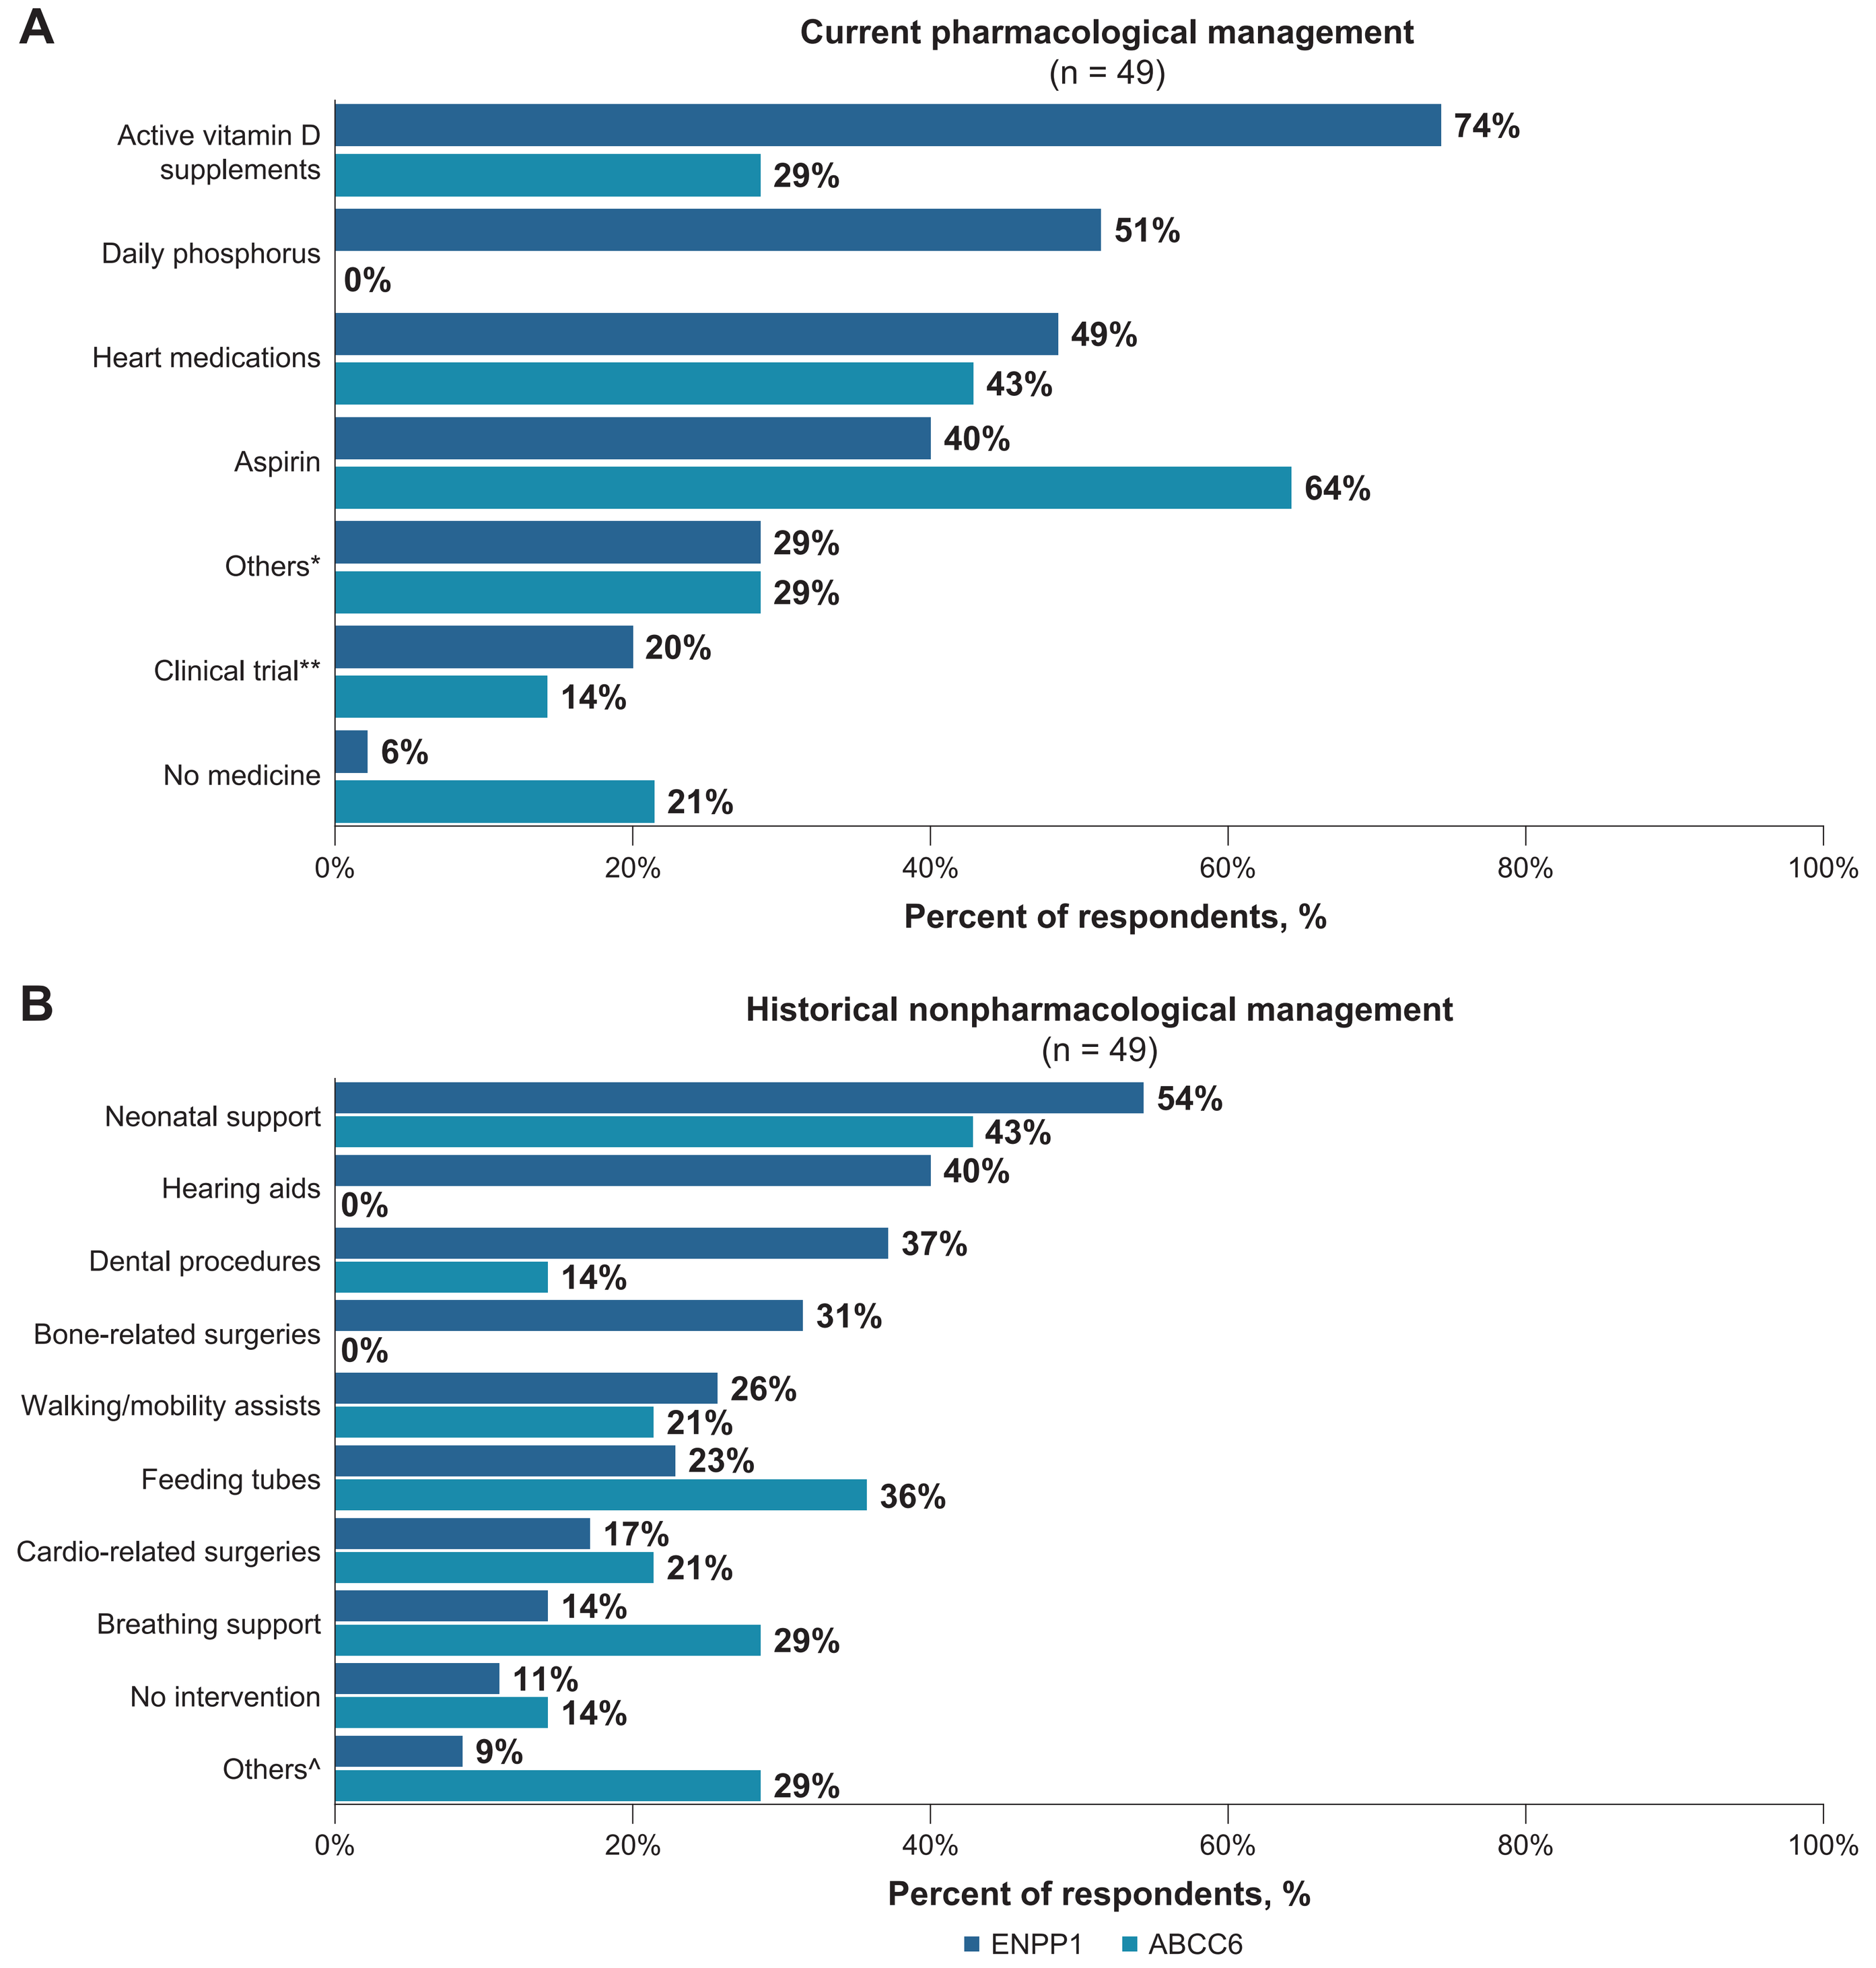
A
Current pharmacological management
(n = 49)
Active vitamin D supplements 74% 29%
Daily phosphorus 51% 0%
Heart medications 49% 43%
Aspirin 40% 64%
Others* 29% 29%
Clinical trial** 20% 14%
No medicine 6% 21%
0% 20% 40% 60% 80% 100%
Percent of respondents, %
B
Historical nonpharmacological management
(n = 49)
Neonatal support 54% 43%
Hearing aids 40% 0%
Dental procedures 37% 14%
Bone-related surgeries 31% 0%
Walking/mobility assists 26% 21%
Feeding tubes 23% 36%
Cardio-related surgeries 17% 21%
Breathing support 14% 29%
No intervention 11% 14%
Others^ 9% 29%
0% 20% 40% 60% 80% 100%
Percent of respondents, %
ENPP1
ABCC6

*Others includes vitamin supplements (B12, folic acid, etc), pain-relief medications, and hormone replacement therapy.
**Includes both Inozyme and other clinical trials (unspecified).
^Others includes interventions including mediport, bowel resection, central catheter, etc.
ABCC6, ATP-binding cassette subfamily C member 6; ENPP1, ectonucleotide pyrophosphatase/phosphodiesterase 1.

Current treatment satisfaction

- Respondents emphasized the lack of disease-modifying treatment options and reported tolerability concerns with available options for symptom management
 - The majority of very satisfied or satisfied respondents were on a combination of medications including heart medications, daily phosphorus, and vitamin D
 - All respondents reported the most important aspect for future therapies as slowing or stopping disease progression, followed by symptom management and affordability of the drug

Conclusions

- Improved outreach and education are needed for pediatricians and other HCPs managing care for patients with ENPP1 or ABCC6 deficiencies
 - Patients have difficulty finding HCPs educated on their condition, which results in delays in diagnosis
 - Multisystem complications due to ENPP1 and ABCC6 deficiencies cause patients to receive care from multiple specialists
- Patients experience significant symptom and treatment burden due to ENPP1 and ABCC6 deficiencies
- Novel therapeutics that slow disease progression and help manage symptoms in patients with ENPP1 and ABCC6 deficiencies are urgently needed

References

1. Ralph D, et al. *Am J Pathol.* 2022;192:762–7
2. Orriss IR, et al. *Curr Opin Pharmacol.* 2016;28:57–68
3. Hamczyk MR, et al. *Aging.* 2018;10(12):3652–3653
4. Guieu R, et al. *J Clin Med.* 2020;9:366
5. Ferreira CR, et al. *Ann Rev Pathol Mech Dis.* 2024;19:507–40
6. Ferreira CR, et al. *JBMR.* 2021;36:2193–202
7. Ferreira CR, et al. *JBMR Plus.* 2025;9(5):ziaf019

Acknowledgments

Medical writing support was provided by Morgan Engelhart Wynn, PhD, of Red Nucleus, and funded by BioMarin Pharmaceutical Inc. Project management support was provided by Abigail Hunt, PhD, of BioMarin Pharmaceutical Inc.

Disclosures

As of July 1, 2025, Inozyme Pharma is a wholly owned subsidiary of BioMarin Pharmaceutical Inc.

LM and **COB** have received consulting fees from Inozyme Pharma.
JG, **KS**, and **CN** are employees and stockholders of Inozyme Pharma.
RR and **AC** have nothing to disclose. **MZM** has received consulting fees from Inozyme Pharma.

Presented at American Society for Bone and Mineral Research (ASBMR) annual meeting: September 5–8, 2025; Seattle, WA, USA

©2025 BioMarin Pharmaceutical Inc. All rights reserved.