Real-world long-term effectiveness and safety outcomes of vosoritide in adolescents with achondroplasia in Japan

Poster 58

Toshimi Michigami,[1] Shirou Matsumoto,[2] Taichi Kitaoka,[3] Jeanne Pimenta,[4] Veronika Horvathova,[4] Hirofumi Tokuoka,[5] Soraya Azmi,[6] Hiroshi Kitoh[7]

[1]Osaka Women's and Children's Hospital, Osaka, Japan; [2]Kumamoto University Hospital, Kumamoto, Japan; [3]ISEIKAI International General Hospital, Osaka, Japan; [4]BioMarin (UK) Ltd, London, UK; [5]BioMarin Pharmaceutical Japan KK, Tokyo, Japan; [6]BioMarin Pharmaceutical Inc., Novato, CA, USA; [7]Aichi Children's Health and Medical Center, Obu, Japan

Introduction

- Achondroplasia is a rare skeletal dysplasia caused by pathogenic fibroblast growth factor receptor 3 gene (*FGFR3*) variants that impair endochondral bone growth
- Vosoritide is a C-type natriuretic peptide analog approved in multiple countries for achondroplasia treatment in children with open epiphyses[1,2]
- In June 2022, Japan became the first country to approve vosoritide from birth until epiphyseal closure[3,4]
- Drug use survey 111-604 is evaluating long-term real-world safety and effectiveness of vosoritide treatment in Japan

Objective

- Using data from 111-604, we report interim long-term vosoritide safety and effectiveness in Japanese adolescents (aged ≥11 years at initiation)

Methods

Study design

- 111-604 collects prospective data beginning August 2022 from the medical records of children and adolescents with achondroplasia prescribed vosoritide in Japan
- This analysis included adolescents (age ≥11 years at initiation)

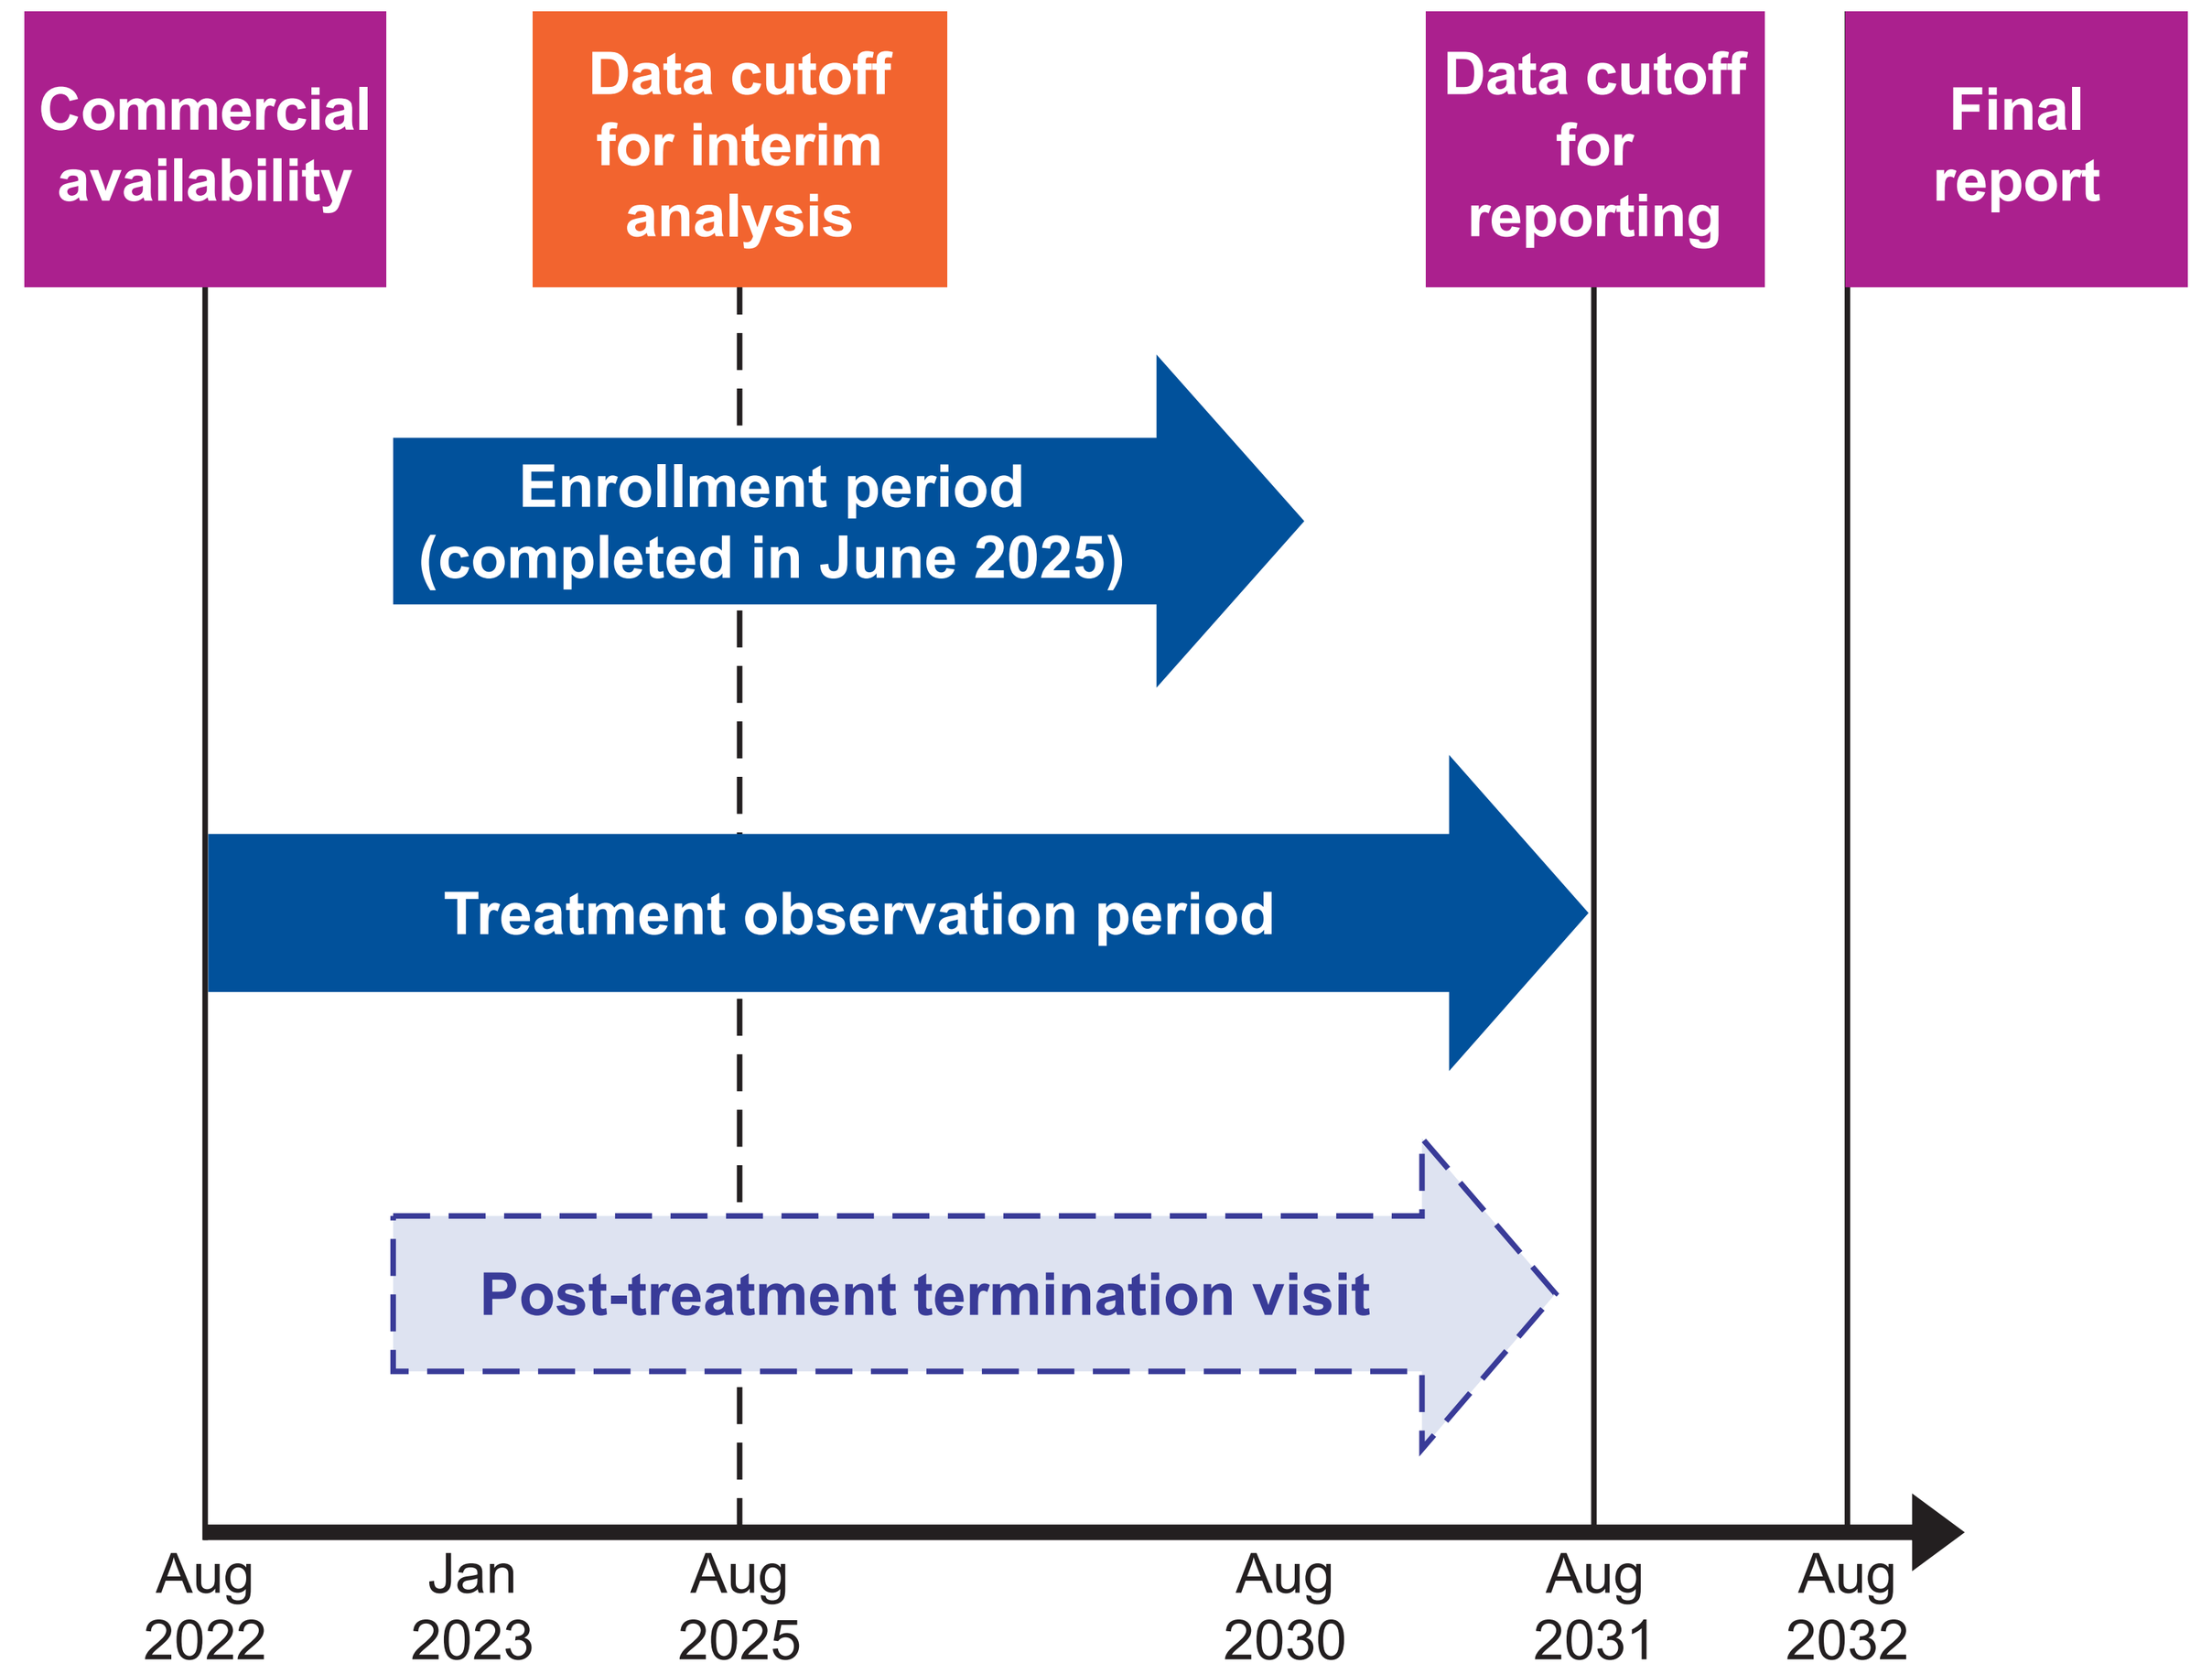
Commercial availability
Data cutoff for interim analysis
Data cutoff for reporting
Final report
Enrollment period (completed in June 2025)
Treatment observation period
Post-treatment termination visit
Aug 2022
Jan 2023
Aug 2025
Aug 2030
Aug 2031
Aug 2032

Endpoints

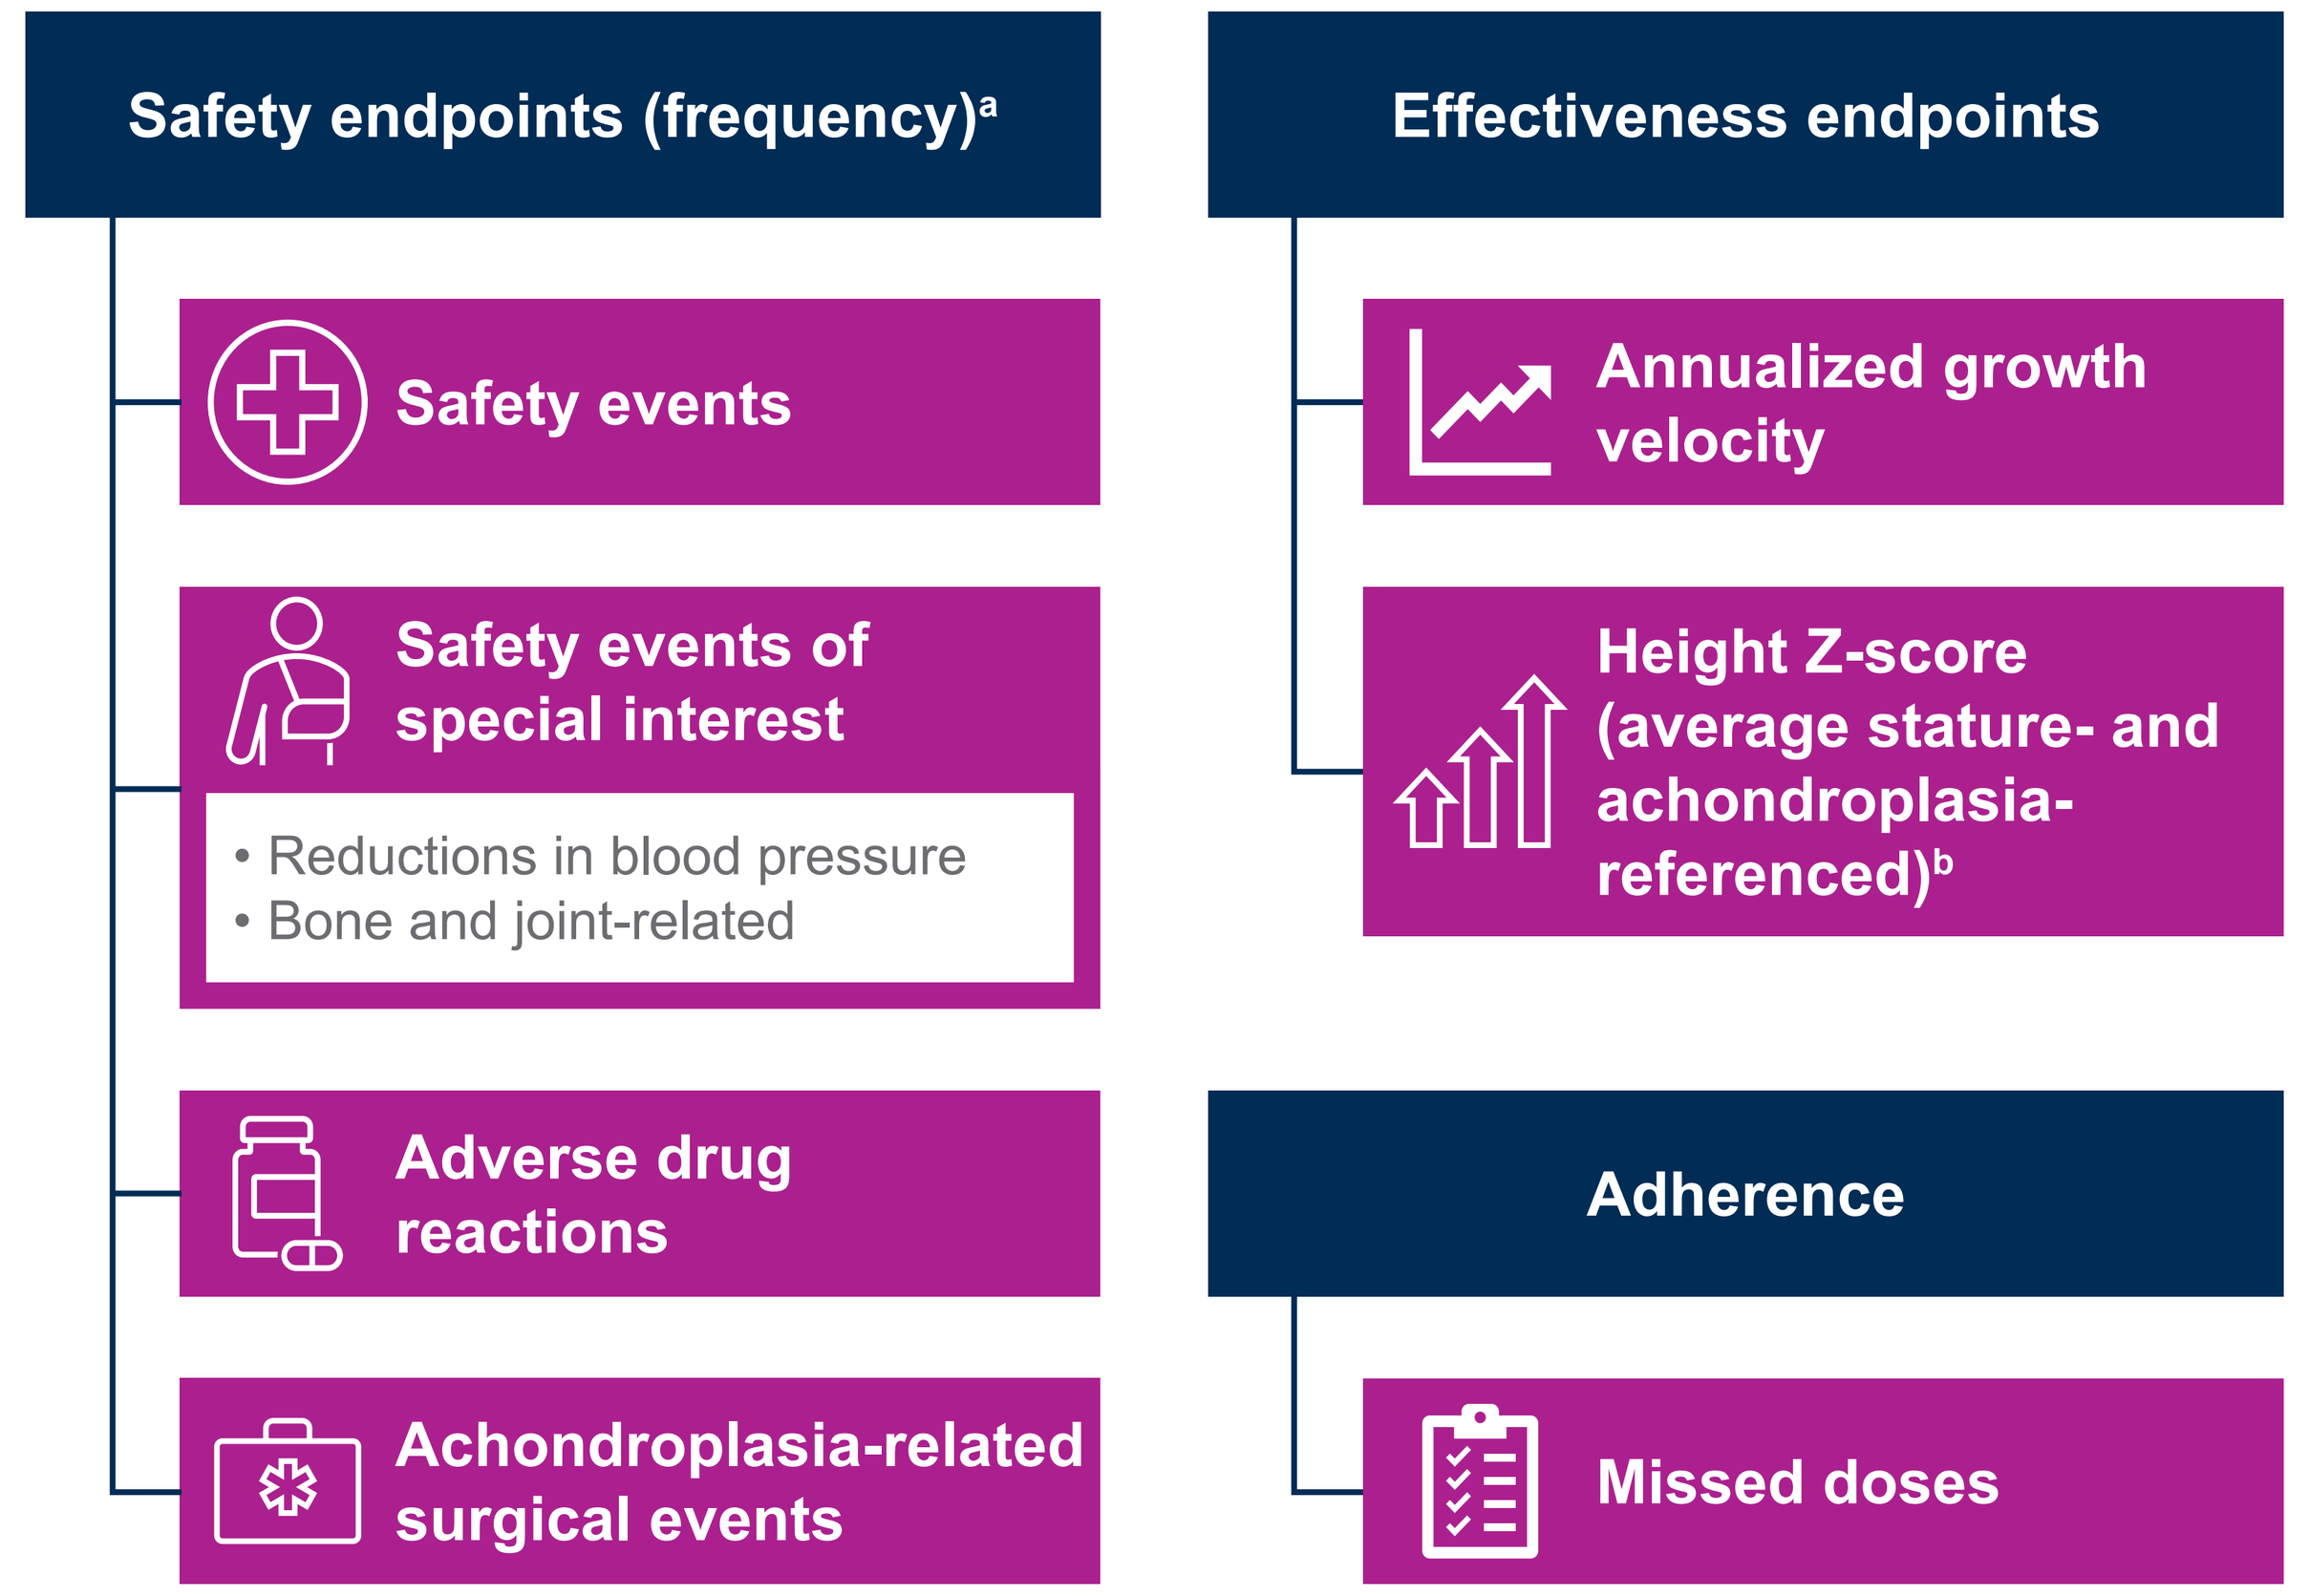
Safety endpoints (frequency)[a]
Safety events
Safety events of special interest
• Reductions in blood pressure
• Bone and joint-related
Adverse drug reactions
Achondroplasia-related surgical events
Effectiveness endpoints
Annualized growth velocity
Height Z-score (average stature- and achondroplasia-referenced)[b]
Adherence
Missed doses

[a]Coded using MedDRA/J version 28.0. [b]Height Z-scores were referenced to age- and sex-matched children of average stature (CDC)[5] or untreated children with achondroplasia (CLARITY).[6]
CDC, US Centers for Disease Control and Prevention; MedDRA/J, Japanese translation of Medical Dictionary for Regulatory Activities.

Results

Participants

- At the data cutoff (August 25, 2025), 632 participants were enrolled, 322 met quality control criteria for inclusion and consented to publication, and 50 were aged ≥11 years at treatment initiation (**Figure 1, Table 1**)

Figure 1. Participant disposition

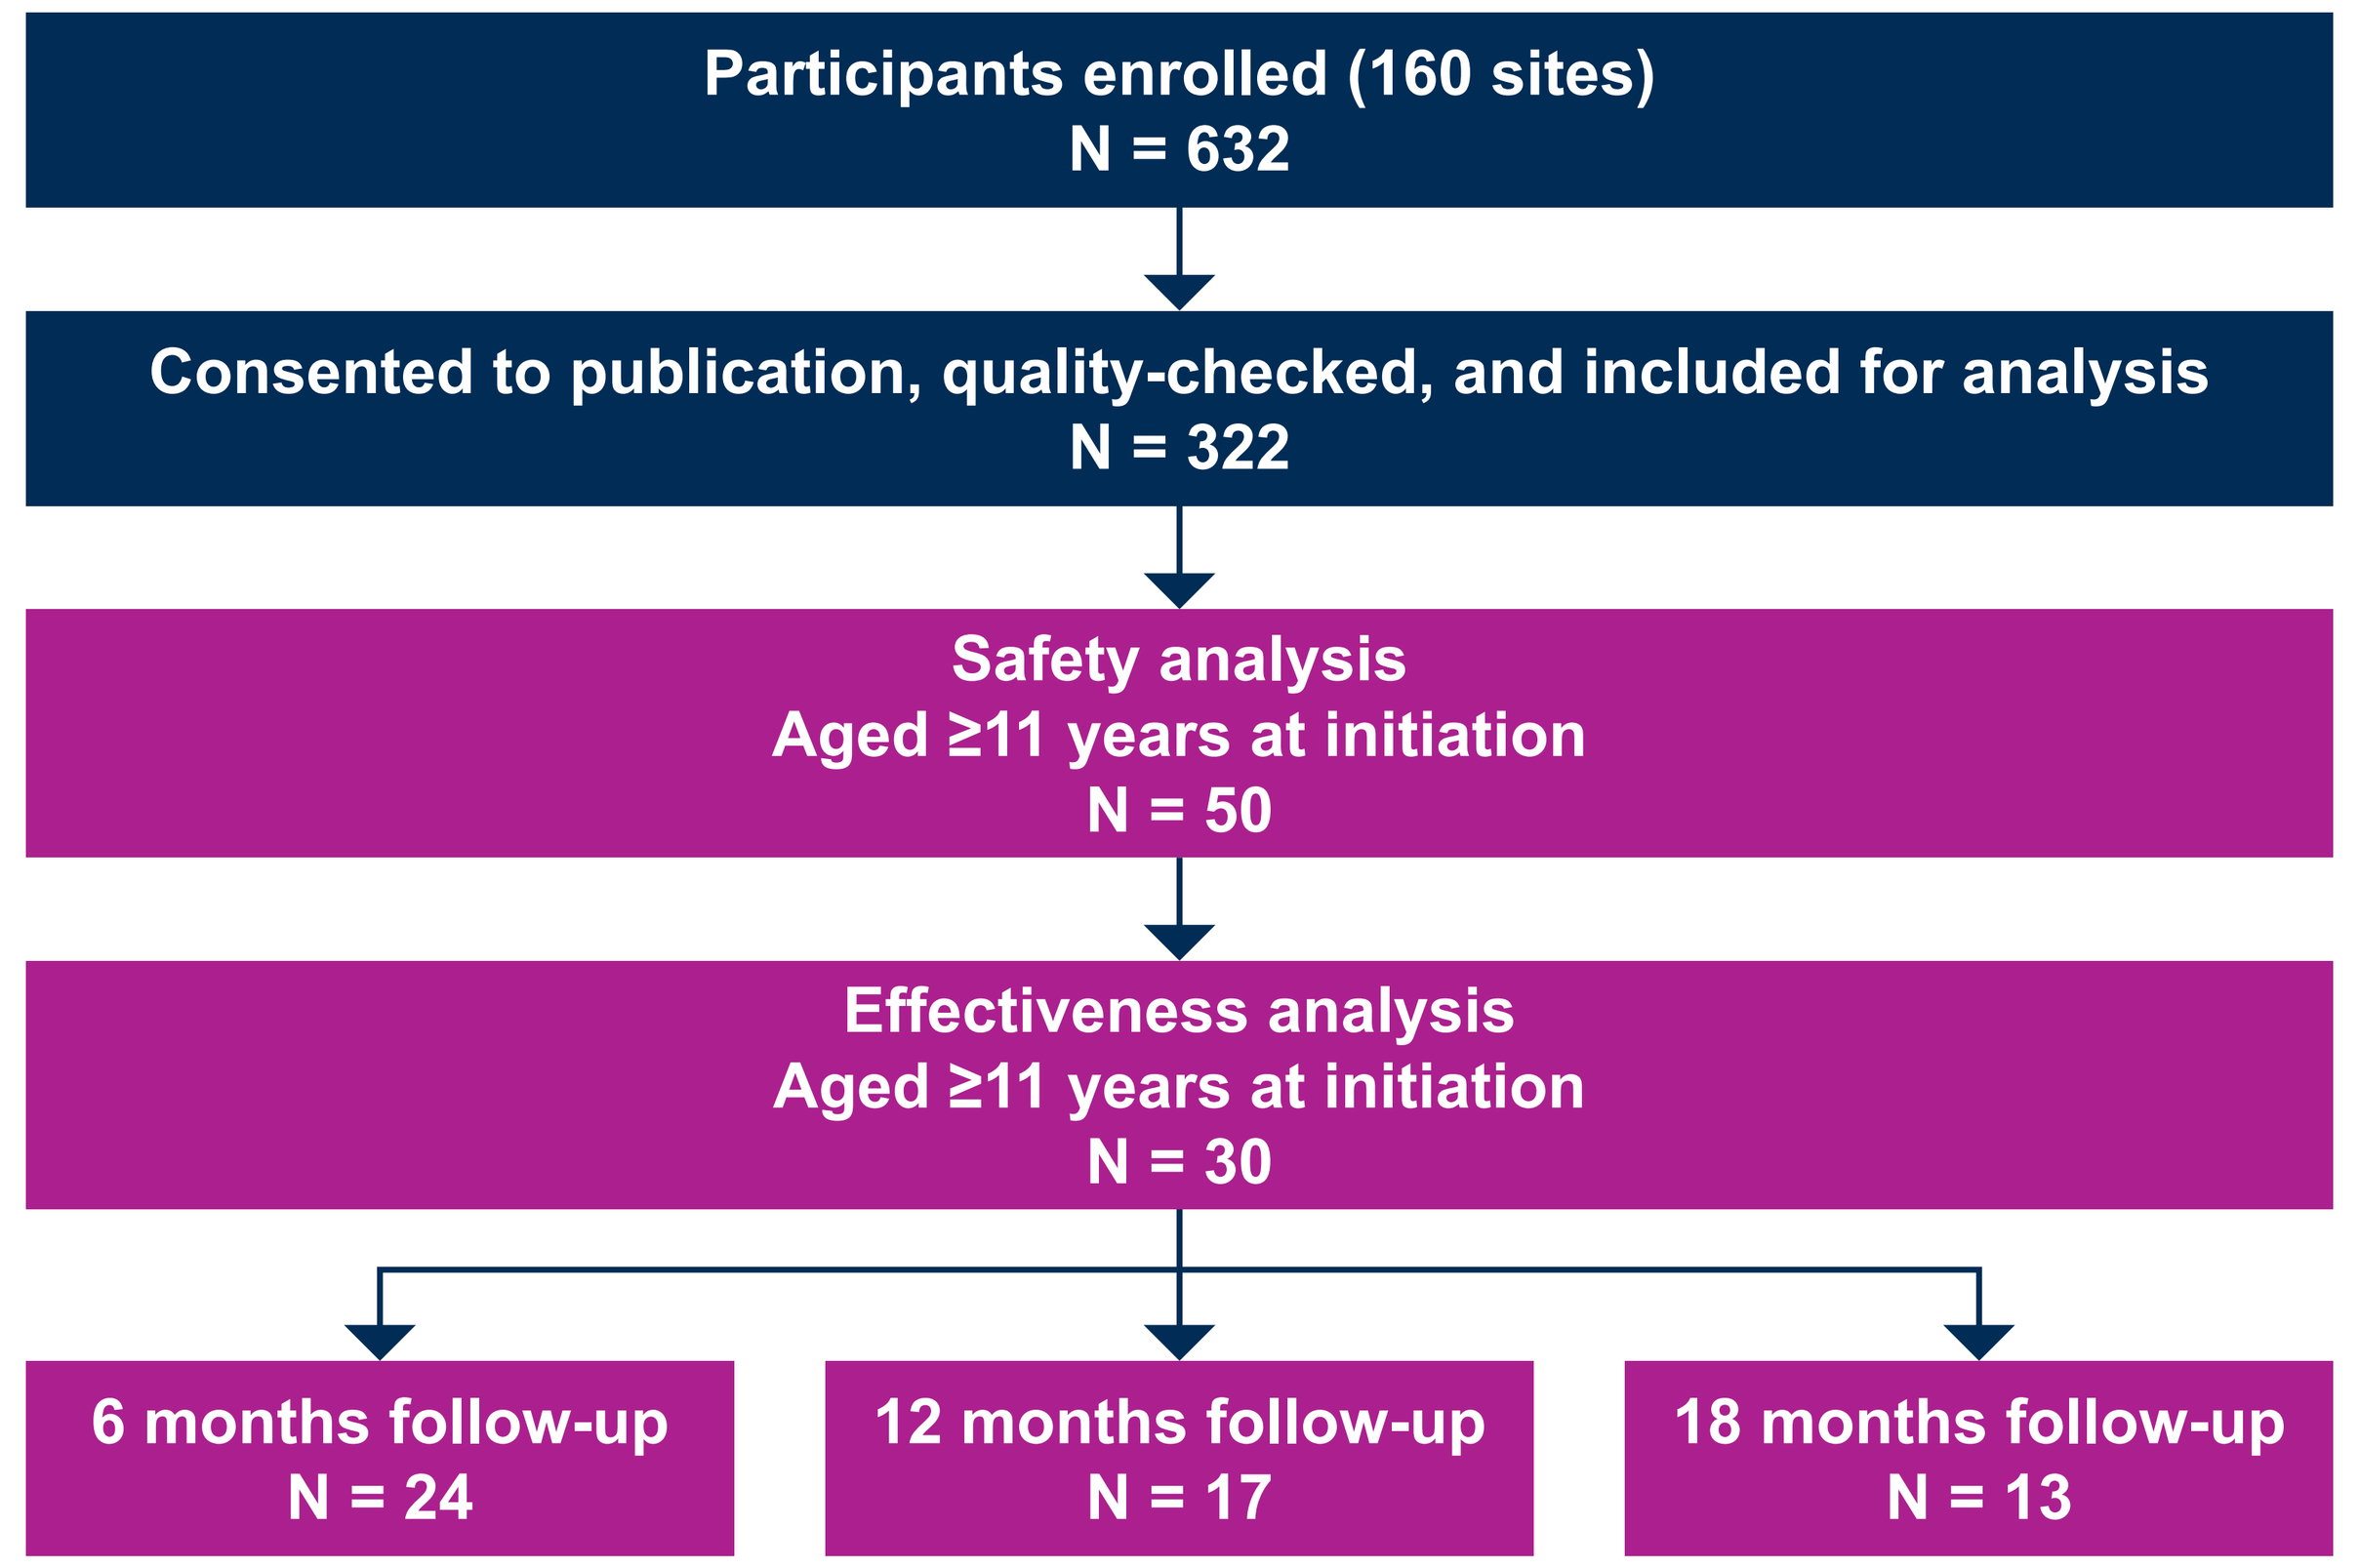
Participants enrolled (160 sites)
N = 632
Consented to publication, quality-checked, and included for analysis
N = 322
Safety analysis
Aged ≥11 years at initiation
N = 50
Effectiveness analysis
Aged ≥11 years at initiation
N = 30
6 months follow-up
N = 24
12 months follow-up
N = 17
18 months follow-up
N = 13

Table 1. Demographics, baseline characteristics, and treatment adherence

	Participants aged ≥11 years (n = 50)
Sex, n (%)	
Male	28 (56.0)
Female	22 (44.0)
Age at treatment initiation, years, mean ± SD	12.6 ± 1.2
Min, max	11.1, 16.6
Prior growth hormone use, n (%)	18 (36.0)
Growth parameters, mean ± SD	
Standing height, cm	118.33 ± 8.65
Height Z-score (average stature-referenced[5])	−4.63 ± 1.10
Height Z-score (achondroplasia-referenced[6])	1.00 ± 1.15
Follow-up duration, months	
Mean ± SD	24.3 ± 9.3
Median (range)	27.2 (0.5–34.7)
Treatment discontinuation, n (%)	9 (18.0)
Physician's judgment	5 (10.0)
Occurrence of safety events	1 (2.0)
Other	3 (6.0)
Treatment interruption (missed >7 consecutive days of daily treatment), n (%)	2 (4.0)
Region, n (%)	
Hokkaido	1 (2.0)
Tohoku	3 (6.0)
Kanto	17 (34.0)
Chubu	6 (12.0)
Kinki	15 (30.0)
Chugoku	2 (4.0)
Shikoku	1 (2.0)
Kyushu and Okinawa	5 (10.0)

Max, maximum; min, minimum; SD, standard deviation.

Safety

- Ten (20.0%) participants reported adverse drug reactions and 10 (20.0%) participants experienced safety events; none were serious (**Figure 2**)
- Two (4.0%) participants had events associated with blood pressure reduction, and 1 (2.0%) participant had a bone/joint-related safety event
- Seven (14.0%) participants had ≥1 achondroplasia-related surgical event (limb lengthening, 6 [12.0%]; surgery for postoperative limb-lengthening complications, 1 [2.0%]; other, 1 [2.0%])

Figure 2. Frequency of adverse drug reactions

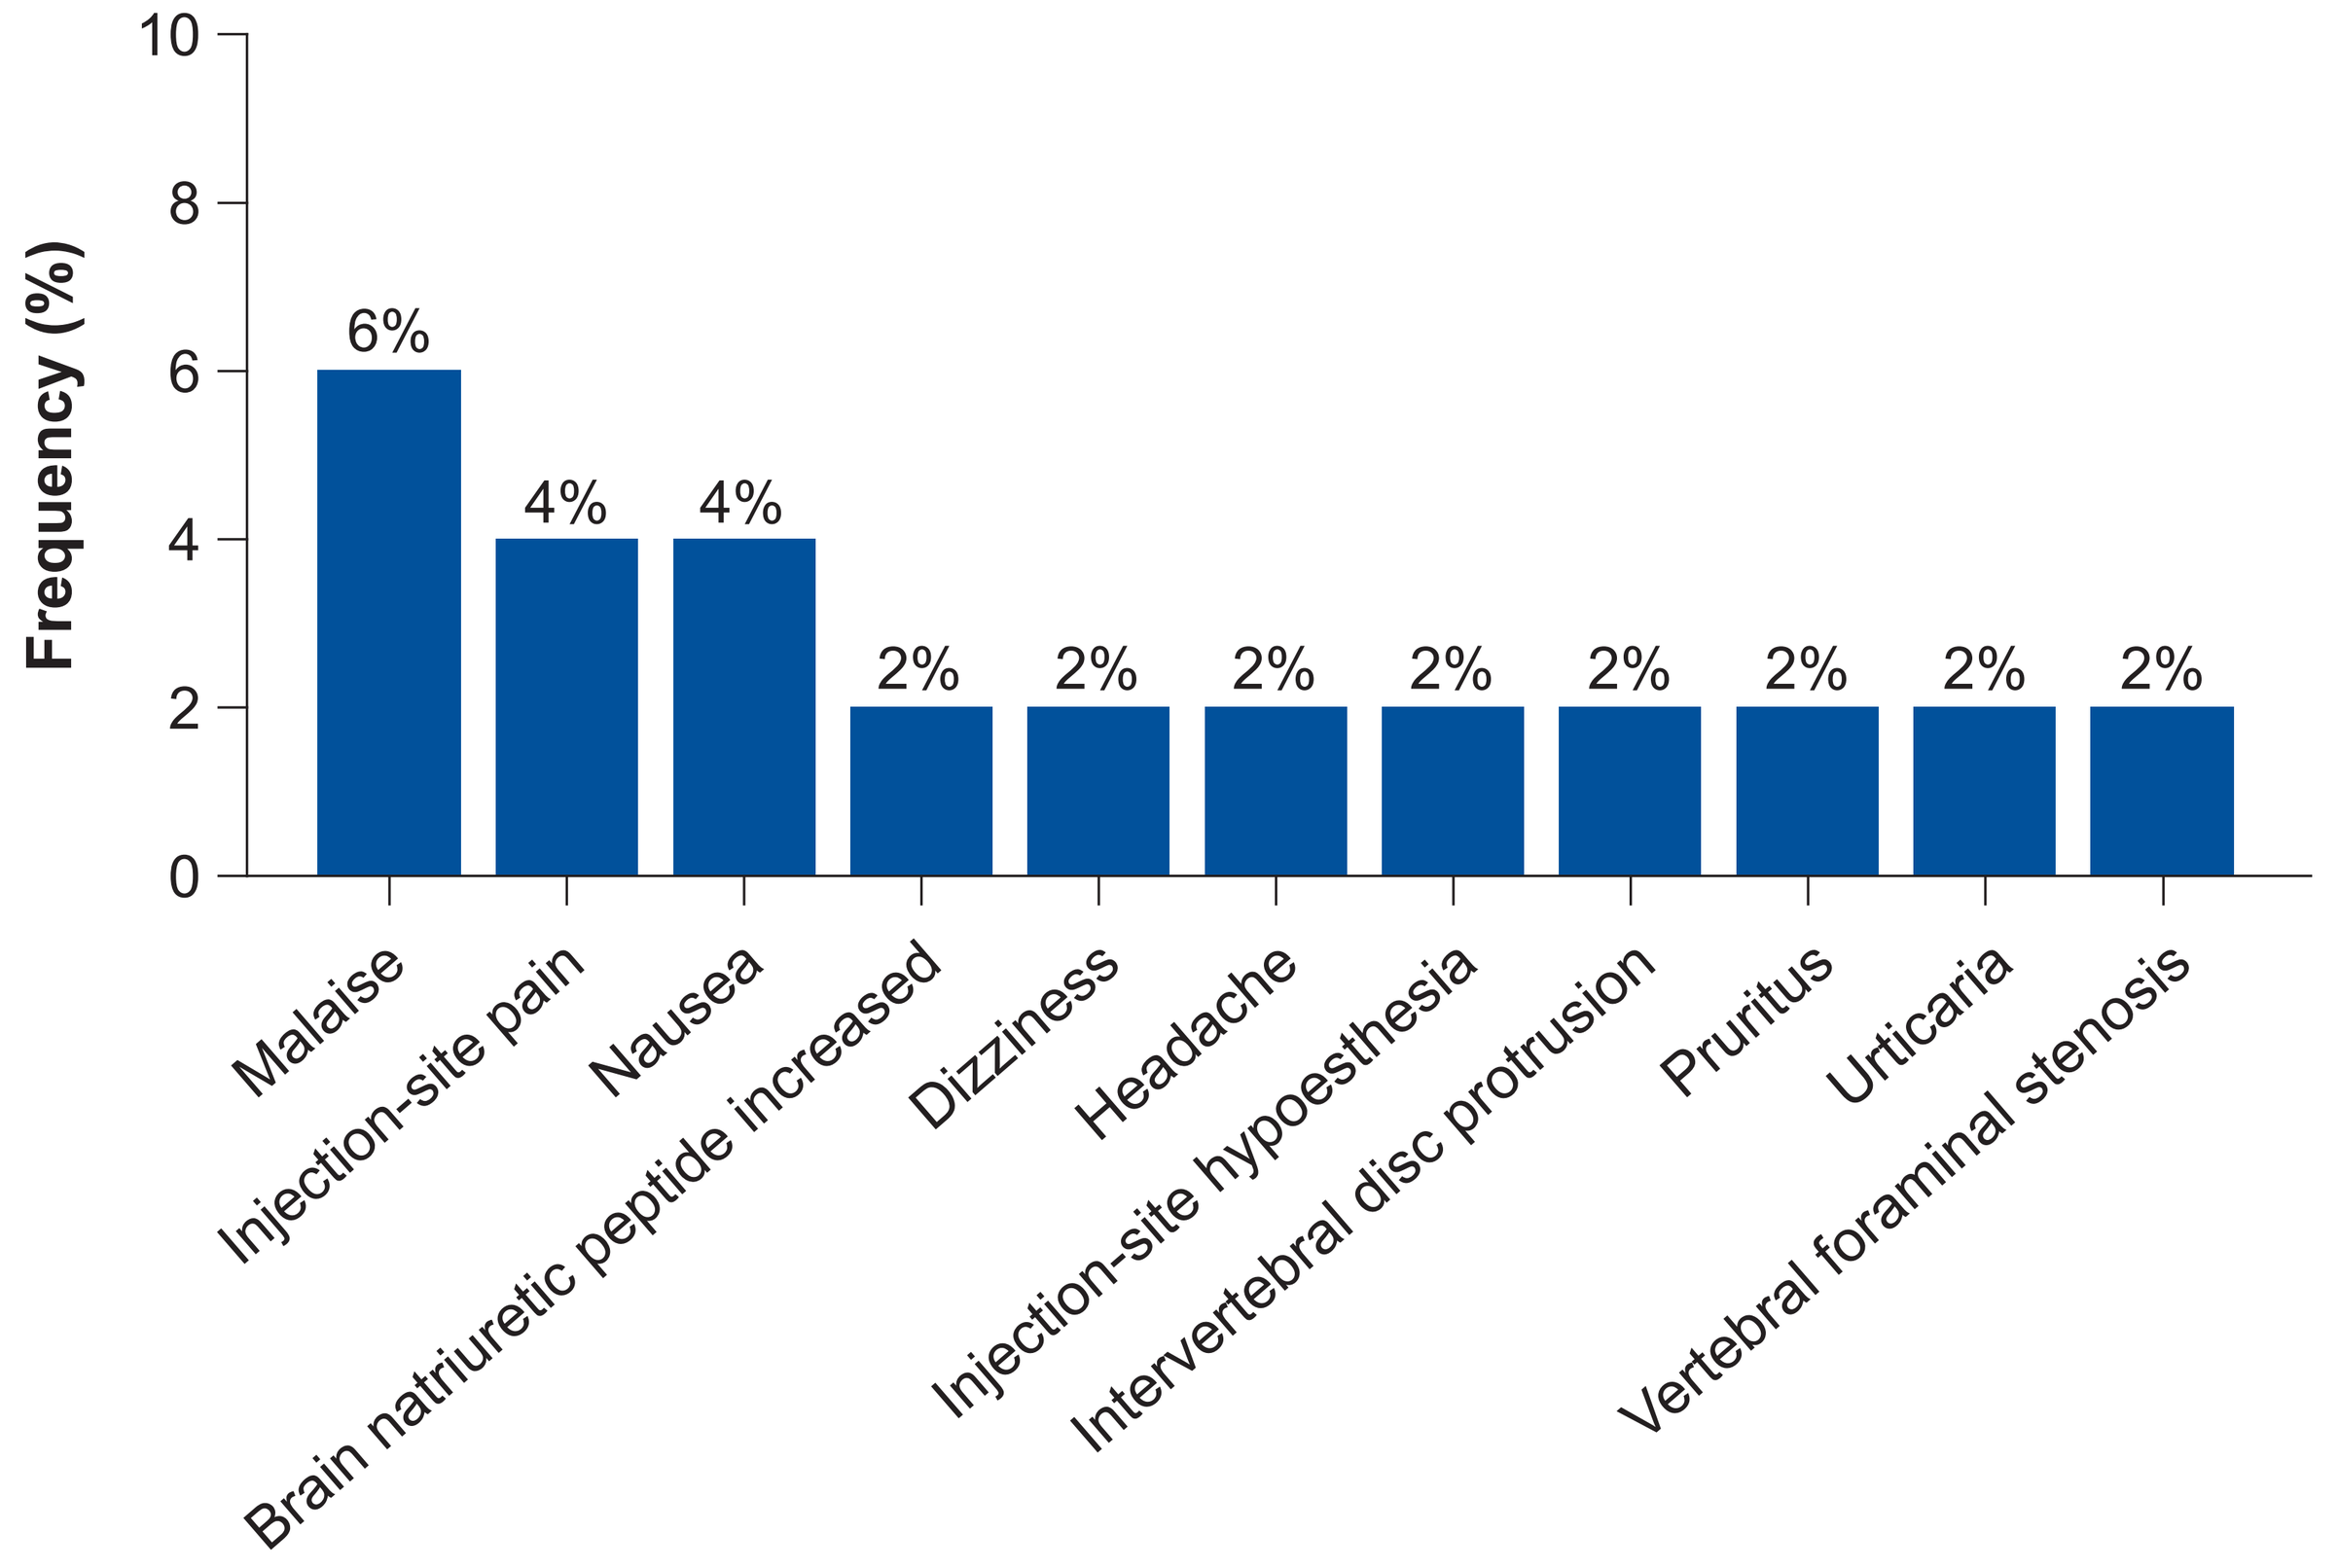
Frequency (%)
10
8
6
4
2
0
6%
4%
4%
2%
2%
2%
2%
2%
2%
2%
2%
Malaise
Injection-site pain
Nausea
Brain natriuretic peptide increased
Dizziness
Headache
Injection-site hypoesthesia
Intervertebral disc protrusion
Pruritus
Urticaria
Vertebral foraminal stenosis

Percentages are calculated based on the total number of participants aged ≥11 years at treatment initiation (n = 50).

Effectiveness

- Change from baseline height Z-scores at 6, 12, and 18 months of follow-up were consistent with clinical trial data (**Figure 3**)
- Consistent cumulative annualized growth velocities were observed at 6, 12, and 18 months (**Figure 4**)

Figure 3. Mean change from baseline height Z-scores at 6, 12, and 18 months

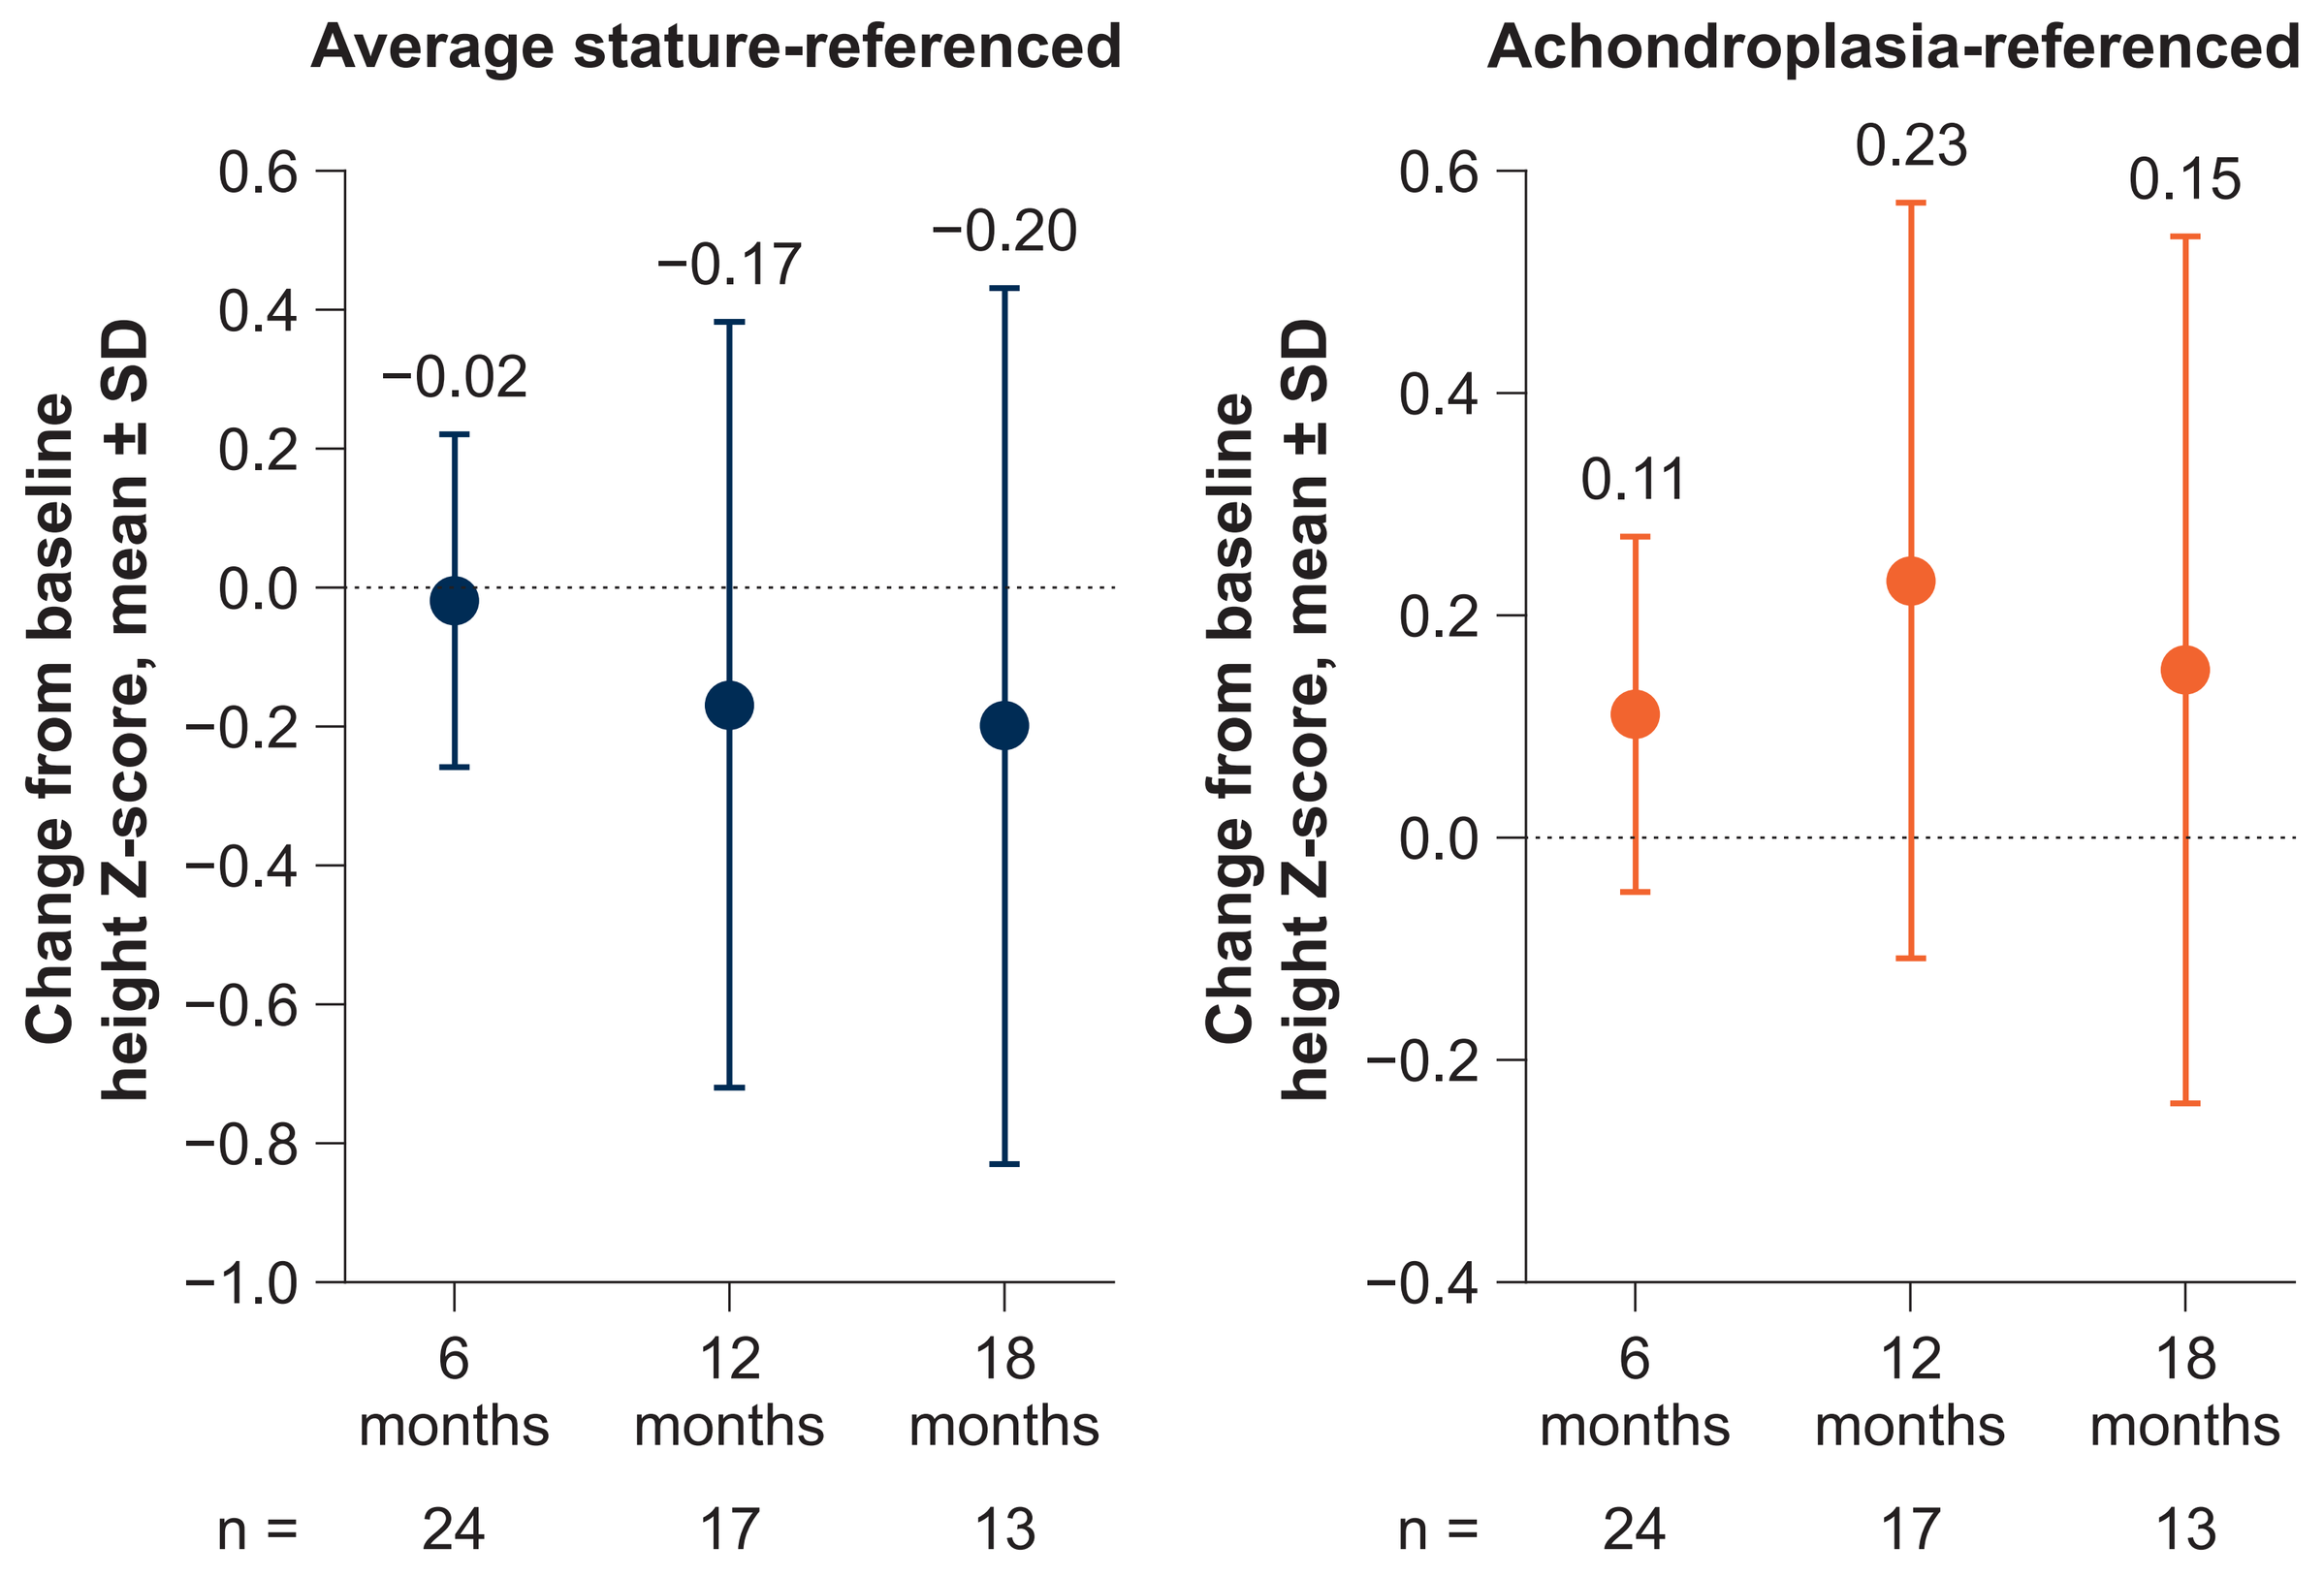
Average stature-referenced
Change from baseline height Z-score, mean ± SD
0.6
0.4
0.2
0.0
−0.2
−0.4
−0.6
−0.8
−1.0
−0.02
−0.17
−0.20
6 months
12 months
18 months
n = 24 17 13
Achondroplasia-referenced
Change from baseline height Z-score, mean ± SD
0.6
0.4
0.2
0.0
−0.2
−0.4
0.11
0.23
0.15
6 months
12 months
18 months
n = 24 17 13

SD, standard deviation.

Figure 4. Mean cumulative annualized growth velocity at 6, 12, and 18 months

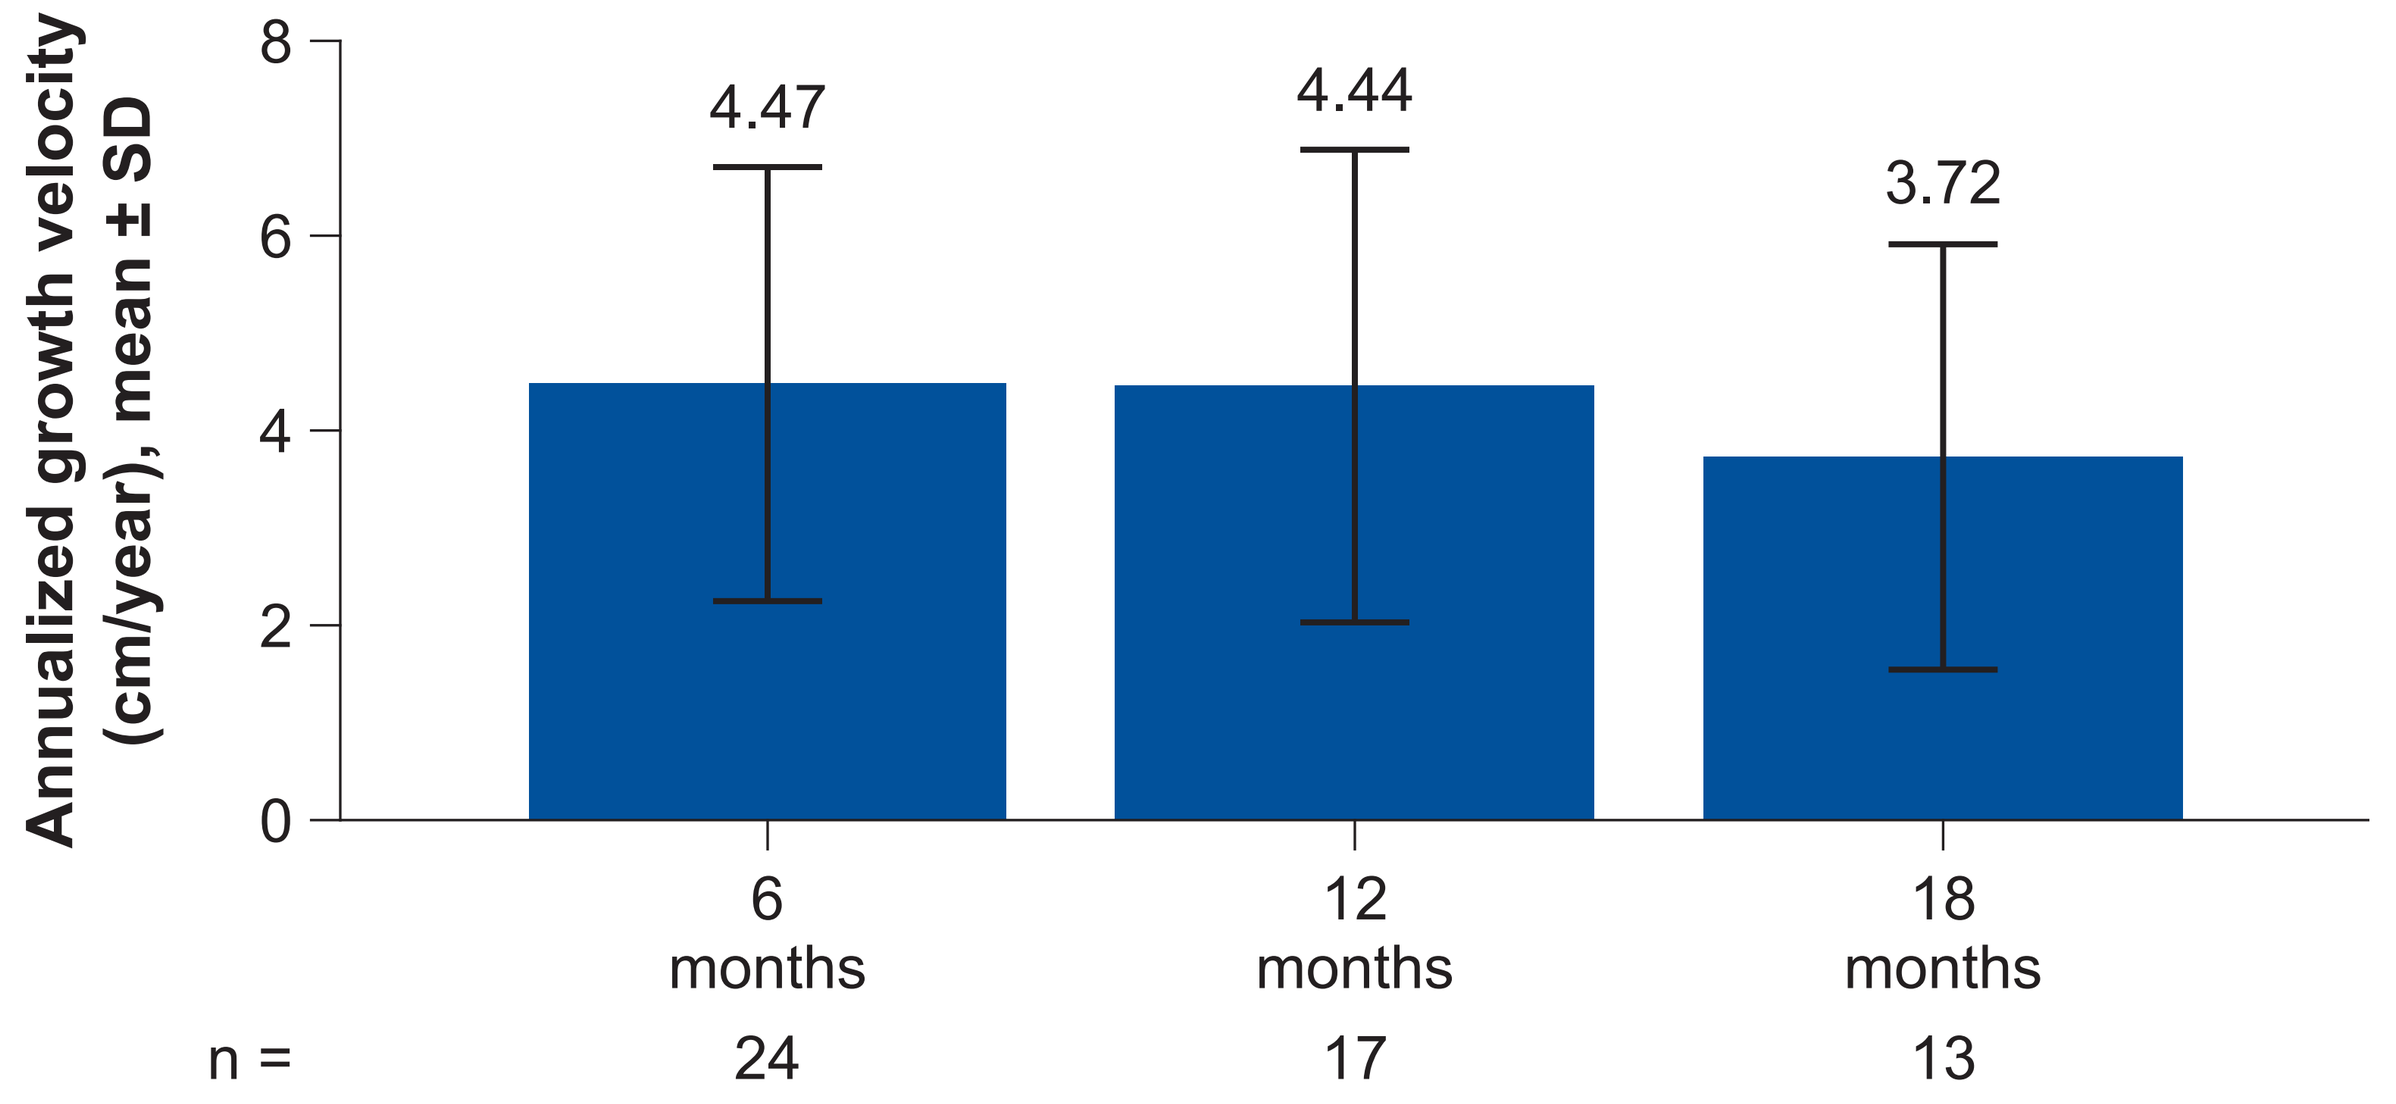
Annualized growth velocity (cm/year), mean ± SD
8
6
4
2
0
4.47
4.44
3.72
6 months
12 months
18 months
n = 24 17 13

Annualized growth velocity calculated from baseline to each time point (6, 12, and 18 months).
SD, standard deviation.

Conclusions

- In Japanese adolescents initiating vosoritide aged ≥11 years, sustained gains in linear growth were observed, consistent with clinical trial findings
- Vosoritide was well tolerated, with no serious safety events and low rates of clinically meaningful treatment interruption and discontinuation through up to 34.7 months of follow-up in real-world clinical practice
- These interim data support the effectiveness and safety of initiating vosoritide during adolescence in routine care settings

References

1. Holmes G, et al. *PLoS One.* 2018;13(7):e0201492. **2.** Savarirayan R, et al. *Nat Rev Endocrinol.* 2025;21(5):314–24. **3.** BioMarin announces the Ministry of Health, Labor and Welfare (MHLW) in Japan granted approval for VOXZOGO® (vosoritide) for injection for the treatment of children with achondroplasia, whose growth plates are not closed. BioMarin Pharmaceutical Inc. June 21, 2023. Accessed February 26, 2026. https://www.biomarin.com/news/press-releases/biomarin-announces-the-ministry-of-health-labor-and-welfare-mhlw-in-japan-granted-approval-for-voxzogo-vosoritide-for-injection-for-the-treatment-of-children-with-achondroplasia-whose-grow/. **4.** Michigami T, et al. *Genet Med Open.* 2025;3(suppl 2):102173. **5.** US Centers for Disease Control and Prevention. CDC growth charts. National Center for Health Statistics. Accessed January 29, 2026. https://www.cdc.gov/growthcharts/clinical_charts.htm. **6.** Hoover-Fong JE, et al. *Orphanet J Rare Dis.* 2021;16(1):522.

Acknowledgments

We thank all trial participants, their families, study-site staff, and investigators. Medical writing support was provided by Johnson Ying, PhD, of Red Nucleus, and funded by BioMarin Pharmaceutical Inc. Project management support was provided by Pooja Dhupkar, PhD, of BioMarin Pharmaceutical Inc.

Disclosures

TM has received lecture fees from Alexion Pharma Japan, BioMarin Pharmaceutical Japan, and Kyowa Kirin. **SM** and **HK** report no conflict of interest. **TK** received speaking and lecture fees and travel reimbursement from BioMarin Pharmaceutical Inc. **JP** and **VH** are employees and stockholders of BioMarin Pharmaceutical Inc. **SA** is an employee of BioMarin Pharmaceutical Inc. **HT** is an employee and stockholder of BioMarin Pharmaceutical Japan KK.

To view a copy of this poster, scan this QR code.

Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission from the authors.

Presented at the Pediatric Endocrine Society (PES) annual meeting: April 30–May 3, 2026; San Francisco, CA, USA

©2026 BioMarin Pharmaceutical Inc. All rights reserved.