Effect of long-term vosoritide treatment on growth in children with achondroplasia in open-label, multicenter clinical trials

Poster 32

Ravi Savarirayan,[1] Carlos A. Bacino,[2] Julie E. Hoover-Fong,[3] Christine Rivat,[4] Richard Rowell,[5] Swati Mukherjee,[4] Alice Huntsman Labed,[4] Lynda E. Polgreen[6]

[1]Murdoch Children's Research Institute, Royal Children's Hospital, and University of Melbourne, Parkville, Victoria, Australia; [2]Baylor College of Medicine, Houston, TX, USA; [3]Johns Hopkins University, Baltimore, MD, USA; [4]BioMarin (UK) Ltd, London, UK; [5]BioMarin Pharmaceutical Inc., Novato, CA, USA; [6]Lundquist Institute for Biomedical Innovation at Harbor-UCLA Medical Center, Torrance, CA, USA

Introduction

- Achondroplasia is a skeletal dysplasia caused by diminished endochondral bone ossification that hinders early growth and often results in multisystem complications[1,2]
- Vosoritide, a first-in-class recombinant C-type natriuretic peptide analog, targets impaired endochondral bone growth and is approved in 47 countries to treat achondroplasia until closure of the epiphyses[2,3]
 - In pivotal phase 2 and 3 trials, vosoritide demonstrated statistically significant improvements in growth vs placebo in children of all ages with a favorable safety profile[4,5,6]

Objective

- To demonstrate the impact of long-term vosoritide treatment on growth vs natural history

Methods

Confirmatory data package: CANOPY ACH-EXT (111-208, 111-205, 111-302) study design

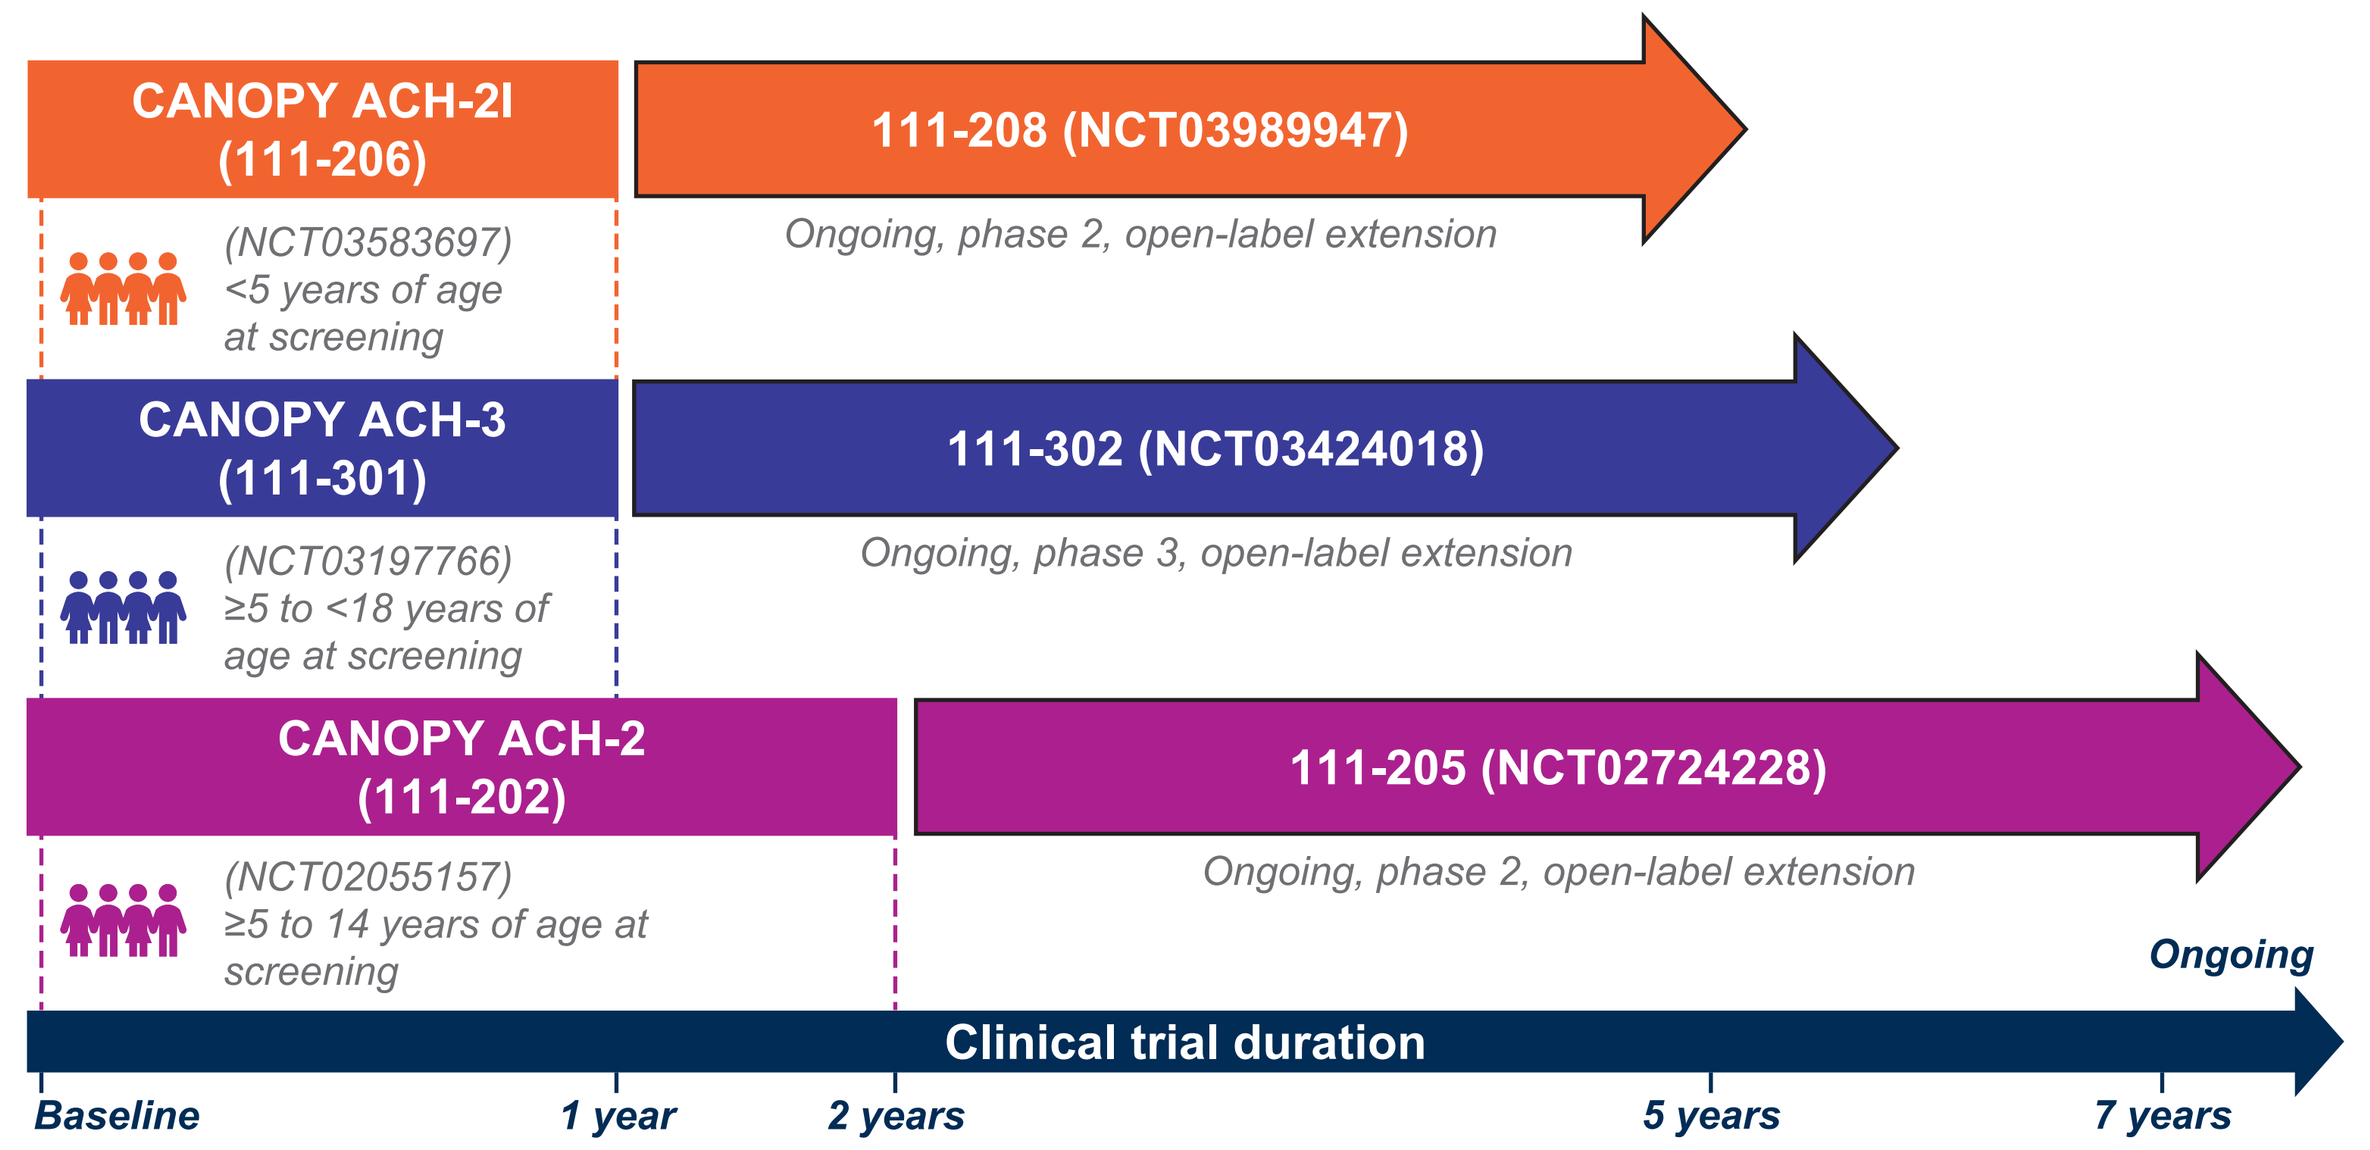
CANOPY ACH-2I (111-206)
111-208 (NCT03989947)
(NCT03583697) <5 years of age at screening
Ongoing, phase 2, open-label extension
CANOPY ACH-3 (111-301)
111-302 (NCT03424018)
(NCT03197766) ≥5 to <18 years of age at screening
Ongoing, phase 3, open-label extension
CANOPY ACH-2 (111-202)
111-205 (NCT02724228)
(NCT02055157) ≥5 to 14 years of age at screening
Ongoing, phase 2, open-label extension
Ongoing
Clinical trial duration
Baseline
1 year
2 years
5 years
7 years

All analyses for CANOPY ACH-EXT studies included data from the first dose of vosoritide in the CANOPY ACH core studies.

CANOPY ACH-EXT study endpoints

Study	Endpoints	Statistical analysis	Statistical comparator datasets
111-208, 111-205, 111-302	Height Z-score (achondroplasia-referenced)[7]	Descriptive summary statistics	
	Height	Baseline-adjusted cross-sectional analysis	Hoover-Fong et al 2021 (CLARITY)[7]
	Height Z-score (average stature-referenced)[8]	Baseline-adjusted cross-sectional analysis	Hoover-Fong et al 2021 (CLARITY)[7]
	Arm span Z-score (achondroplasia-referenced)[9]	Descriptive summary statistics	
	Arm span-to-height ratio	Descriptive summary statistics	

Data are included in analyses after the start of vosoritide treatment. Cross sectional analyses adjusted for baseline differences by subtracting the matched difference at baseline from the matched difference between the treated and untreated arms at the follow up time point. Height Z-scores were referenced to age- and sex-matched children of average stature (CDC)[8] or untreated children with achondroplasia (CLARITY).[7] Arm span Z-scores were referenced to age- and sex-matched untreated children with achondroplasia.[9]
CDC, US Centers for Disease Control and Prevention; HRQOL, health-related quality of life; QoLISSY, Quality of Life in Short Stature Youth questionnaire.

Results

Participants and treatment adherence

- As of August 2025, 222 children enrolled in CANOPY ACH-EXT trials (**Table 1**)

Table 1. Participant baseline characteristics and study disposition

Parameter	111-208	111-205	111-302
Enrolled, n	73	30	119
Age on day 1, years, mean ± SD	2.6 ± 1.7	8.2 ± 1.6	9.2 ± 2.6
Male sex, n (%)	37 (50.7)	13 (43.3)	63 (52.9)
Standing height, cm, mean ± SD	74.07 ± 10.60	100.16 ± 8.03	103.57 ± 11.91
Height Z-score (achondroplasia-referenced), mean ± SD	0.29 ± 0.78	0.39 ± 0.91	0.28 ± 0.99
Height Z-score (average stature-referenced), mean ± SD	−4.18 ± 1.11	−5.12 ± 1.00	−5.12 ± 1.09
Baseline arm span Z-score (achondroplasia-referenced), mean ± SD	−0.36 ± 1.19[a]	−0.12 ± 1.07	−0.32 ± 1.25
Treatment duration, years, mean ± SD	5.0 ± 1.5	7.4 ± 2.2	5.2 ± 1.4
Median (min, max)	5.4 (1.0, 7.2)	7.2 (6.1, 9.2)	5.5 (1.7, 7.4)

[a]n = 40.
Max, maximum; min, minimum; SD, standard deviation.

- In studies 111-205 and 111-302, 13/30 (43.3%) and 22/119 (18.5%) participants discontinued from treatment after reaching near-final adult height with evidence of growth plate closure and <1.5 cm/year annualized growth velocity
 - One participant discontinued from treatment due to adverse events in each of the 3 studies
- Across all 3 studies, treatment adherence remained high over time, with children aged ≥3 months to <5 years (111-208) and ≥5 to <18 years (111-205 and 111-302) at treatment initiation remaining compliant to the study protocol, with a mean (standard deviation [SD]) percent adherence of 96.3% (5.1), 97.7% (4.2), and 97.1% (4.9), respectively

Efficacy

Linear growth

- Height Z-score (achondroplasia-referenced) continued to increase over time for participants in all 3 studies (**Figure 1**)

Figure 1. Height Z-score (achondroplasia-referenced) improved over time across all ages independent of age at treatment initiation

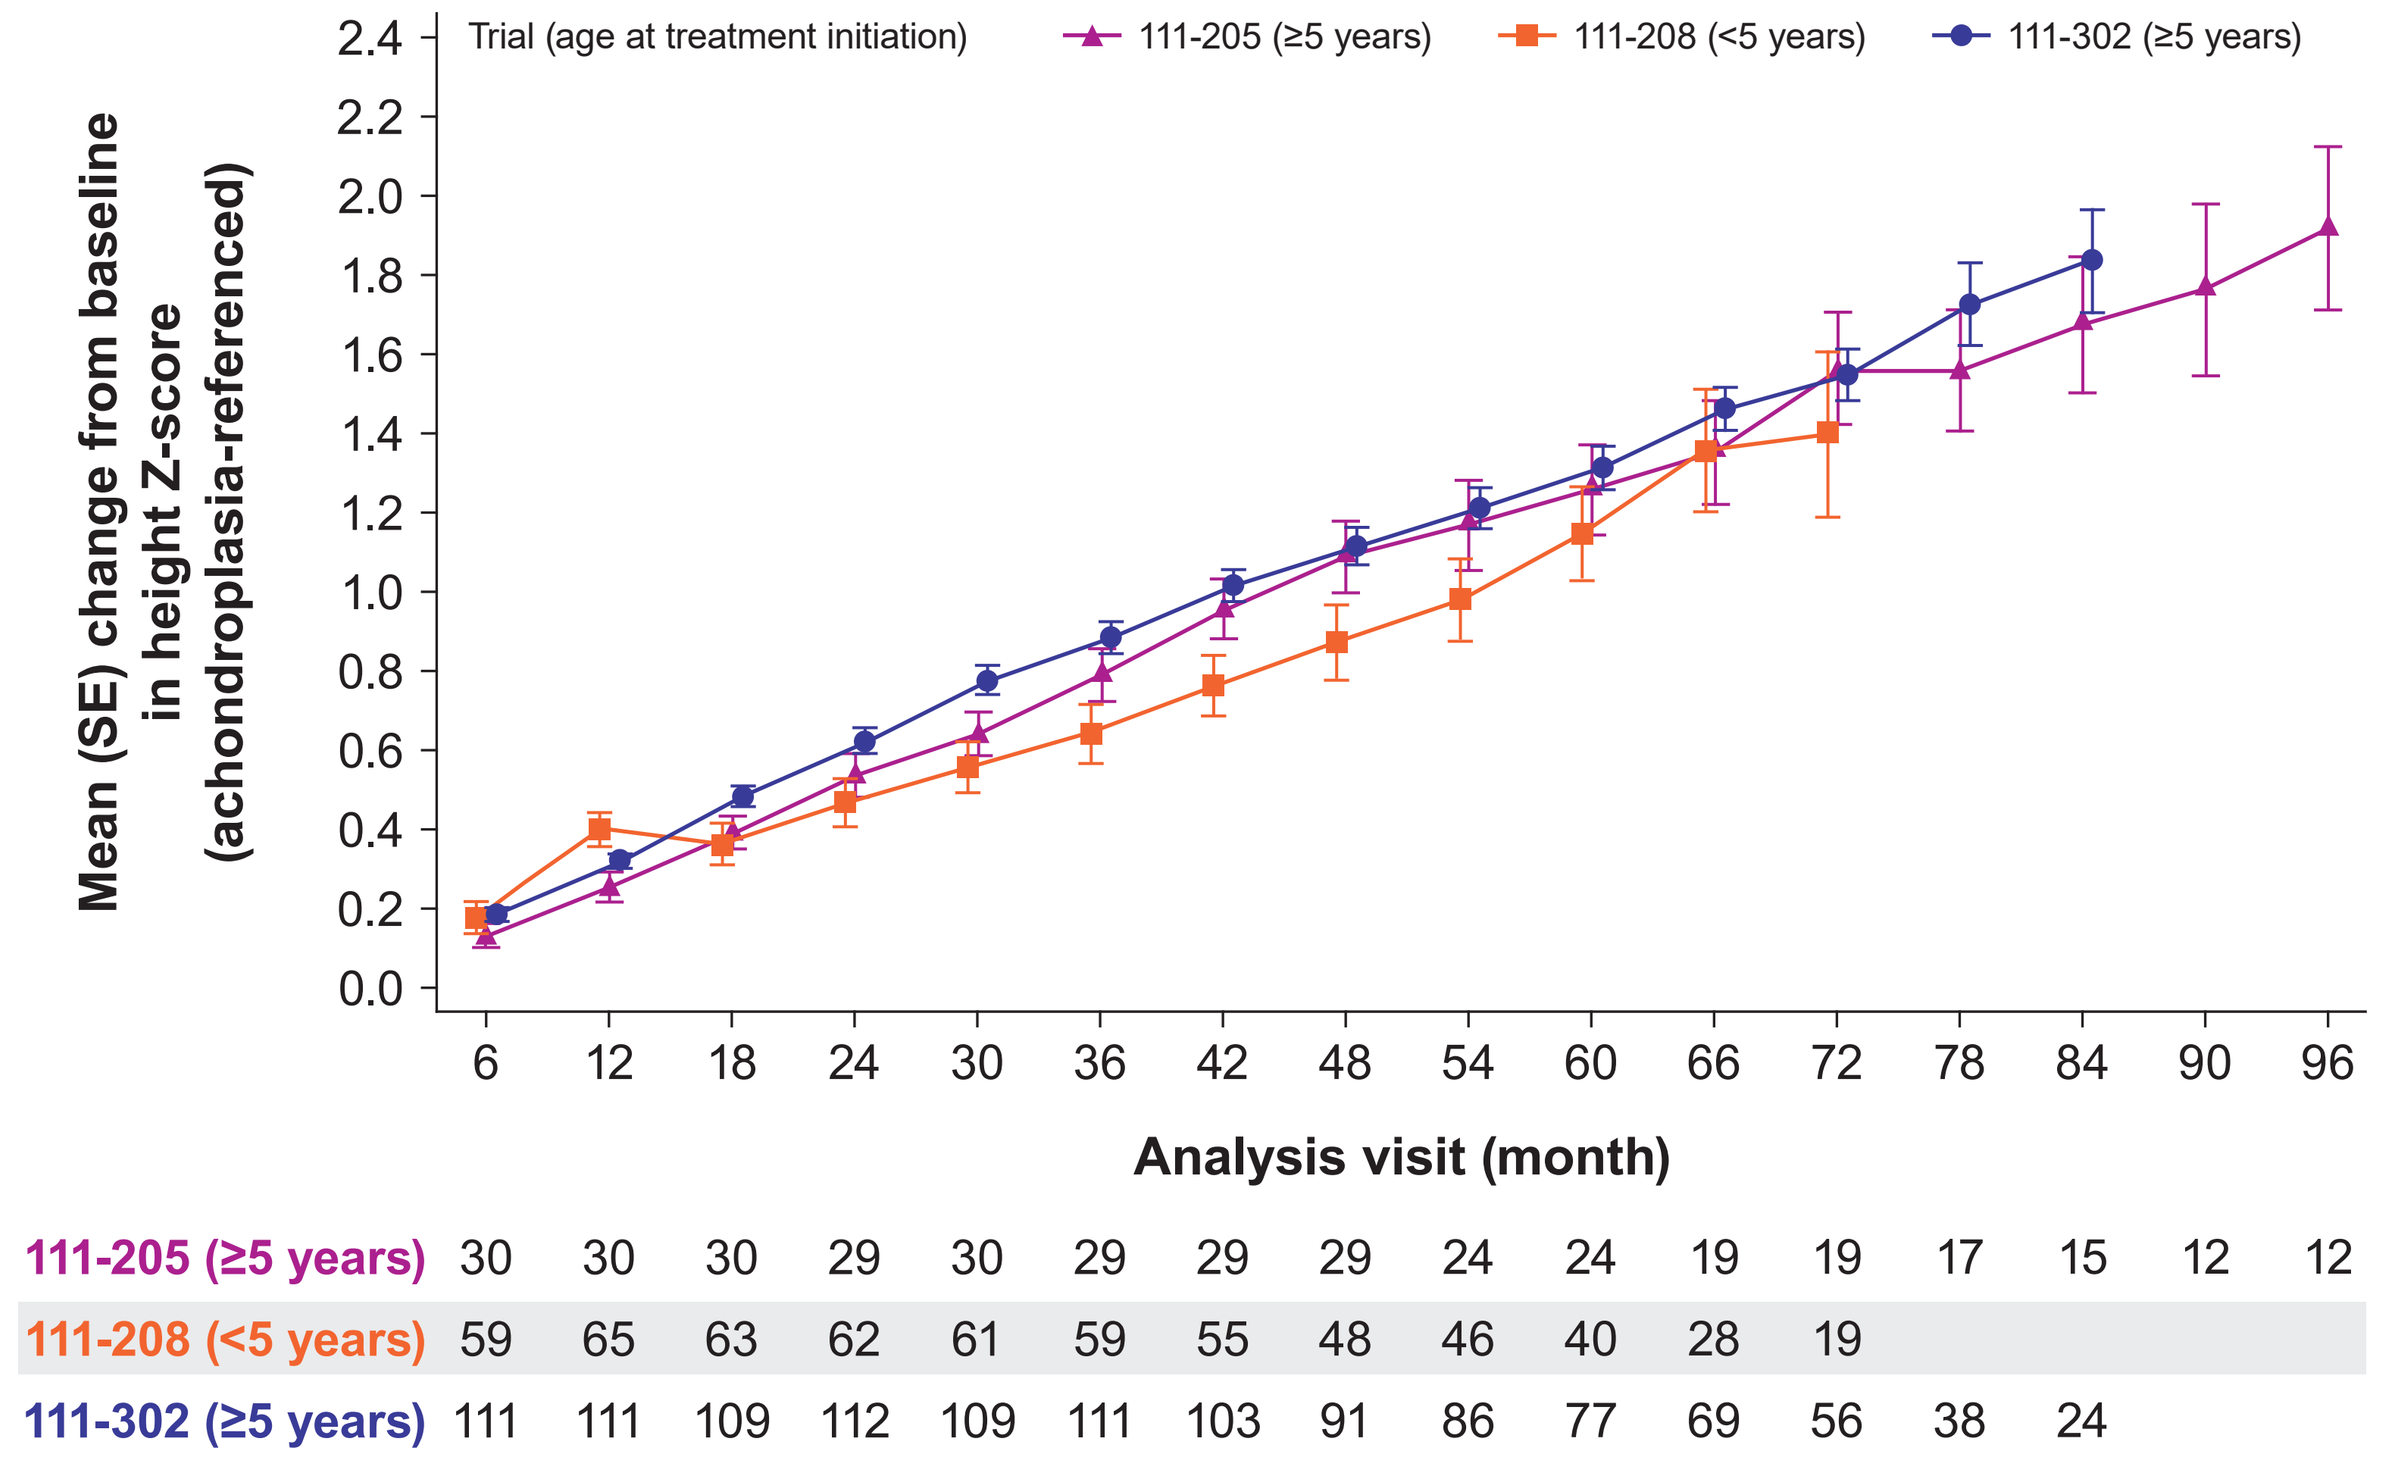
Trial (age at treatment initiation)
111-205 (≥5 years)
111-208 (<5 years)
111-302 (≥5 years)
Mean (SE) change from baseline in height Z-score (achondroplasia-referenced)
2.4 2.2 2.0 1.8 1.6 1.4 1.2 1.0 0.8 0.6 0.4 0.2 0.0
6 12 18 24 30 36 42 48 54 60 66 72 78 84 90 96
Analysis visit (month)
111-205 (≥5 years) 30 30 30 29 30 29 29 29 24 24 19 19 17 15 12 12
111-208 (<5 years) 59 65 63 62 61 59 55 48 46 40 28 19
111-302 (≥5 years) 111 111 109 112 109 111 103 91 86 77 69 56 38 24

Height Z-scores were referenced to age- and sex-matched untreated children with achondroplasia (CLARITY).[7]
SE, standard error.

- Gains in height (**Figure 2A**) and height Z-score (**Figure 2B**; average stature-referenced) continued to increase over time compared with untreated children with achondroplasia independent of study and age at treatment initiation

Figure 2. Height (achondroplasia natural history referenced) and height Z-score (average stature-referenced) improved in all ages over time

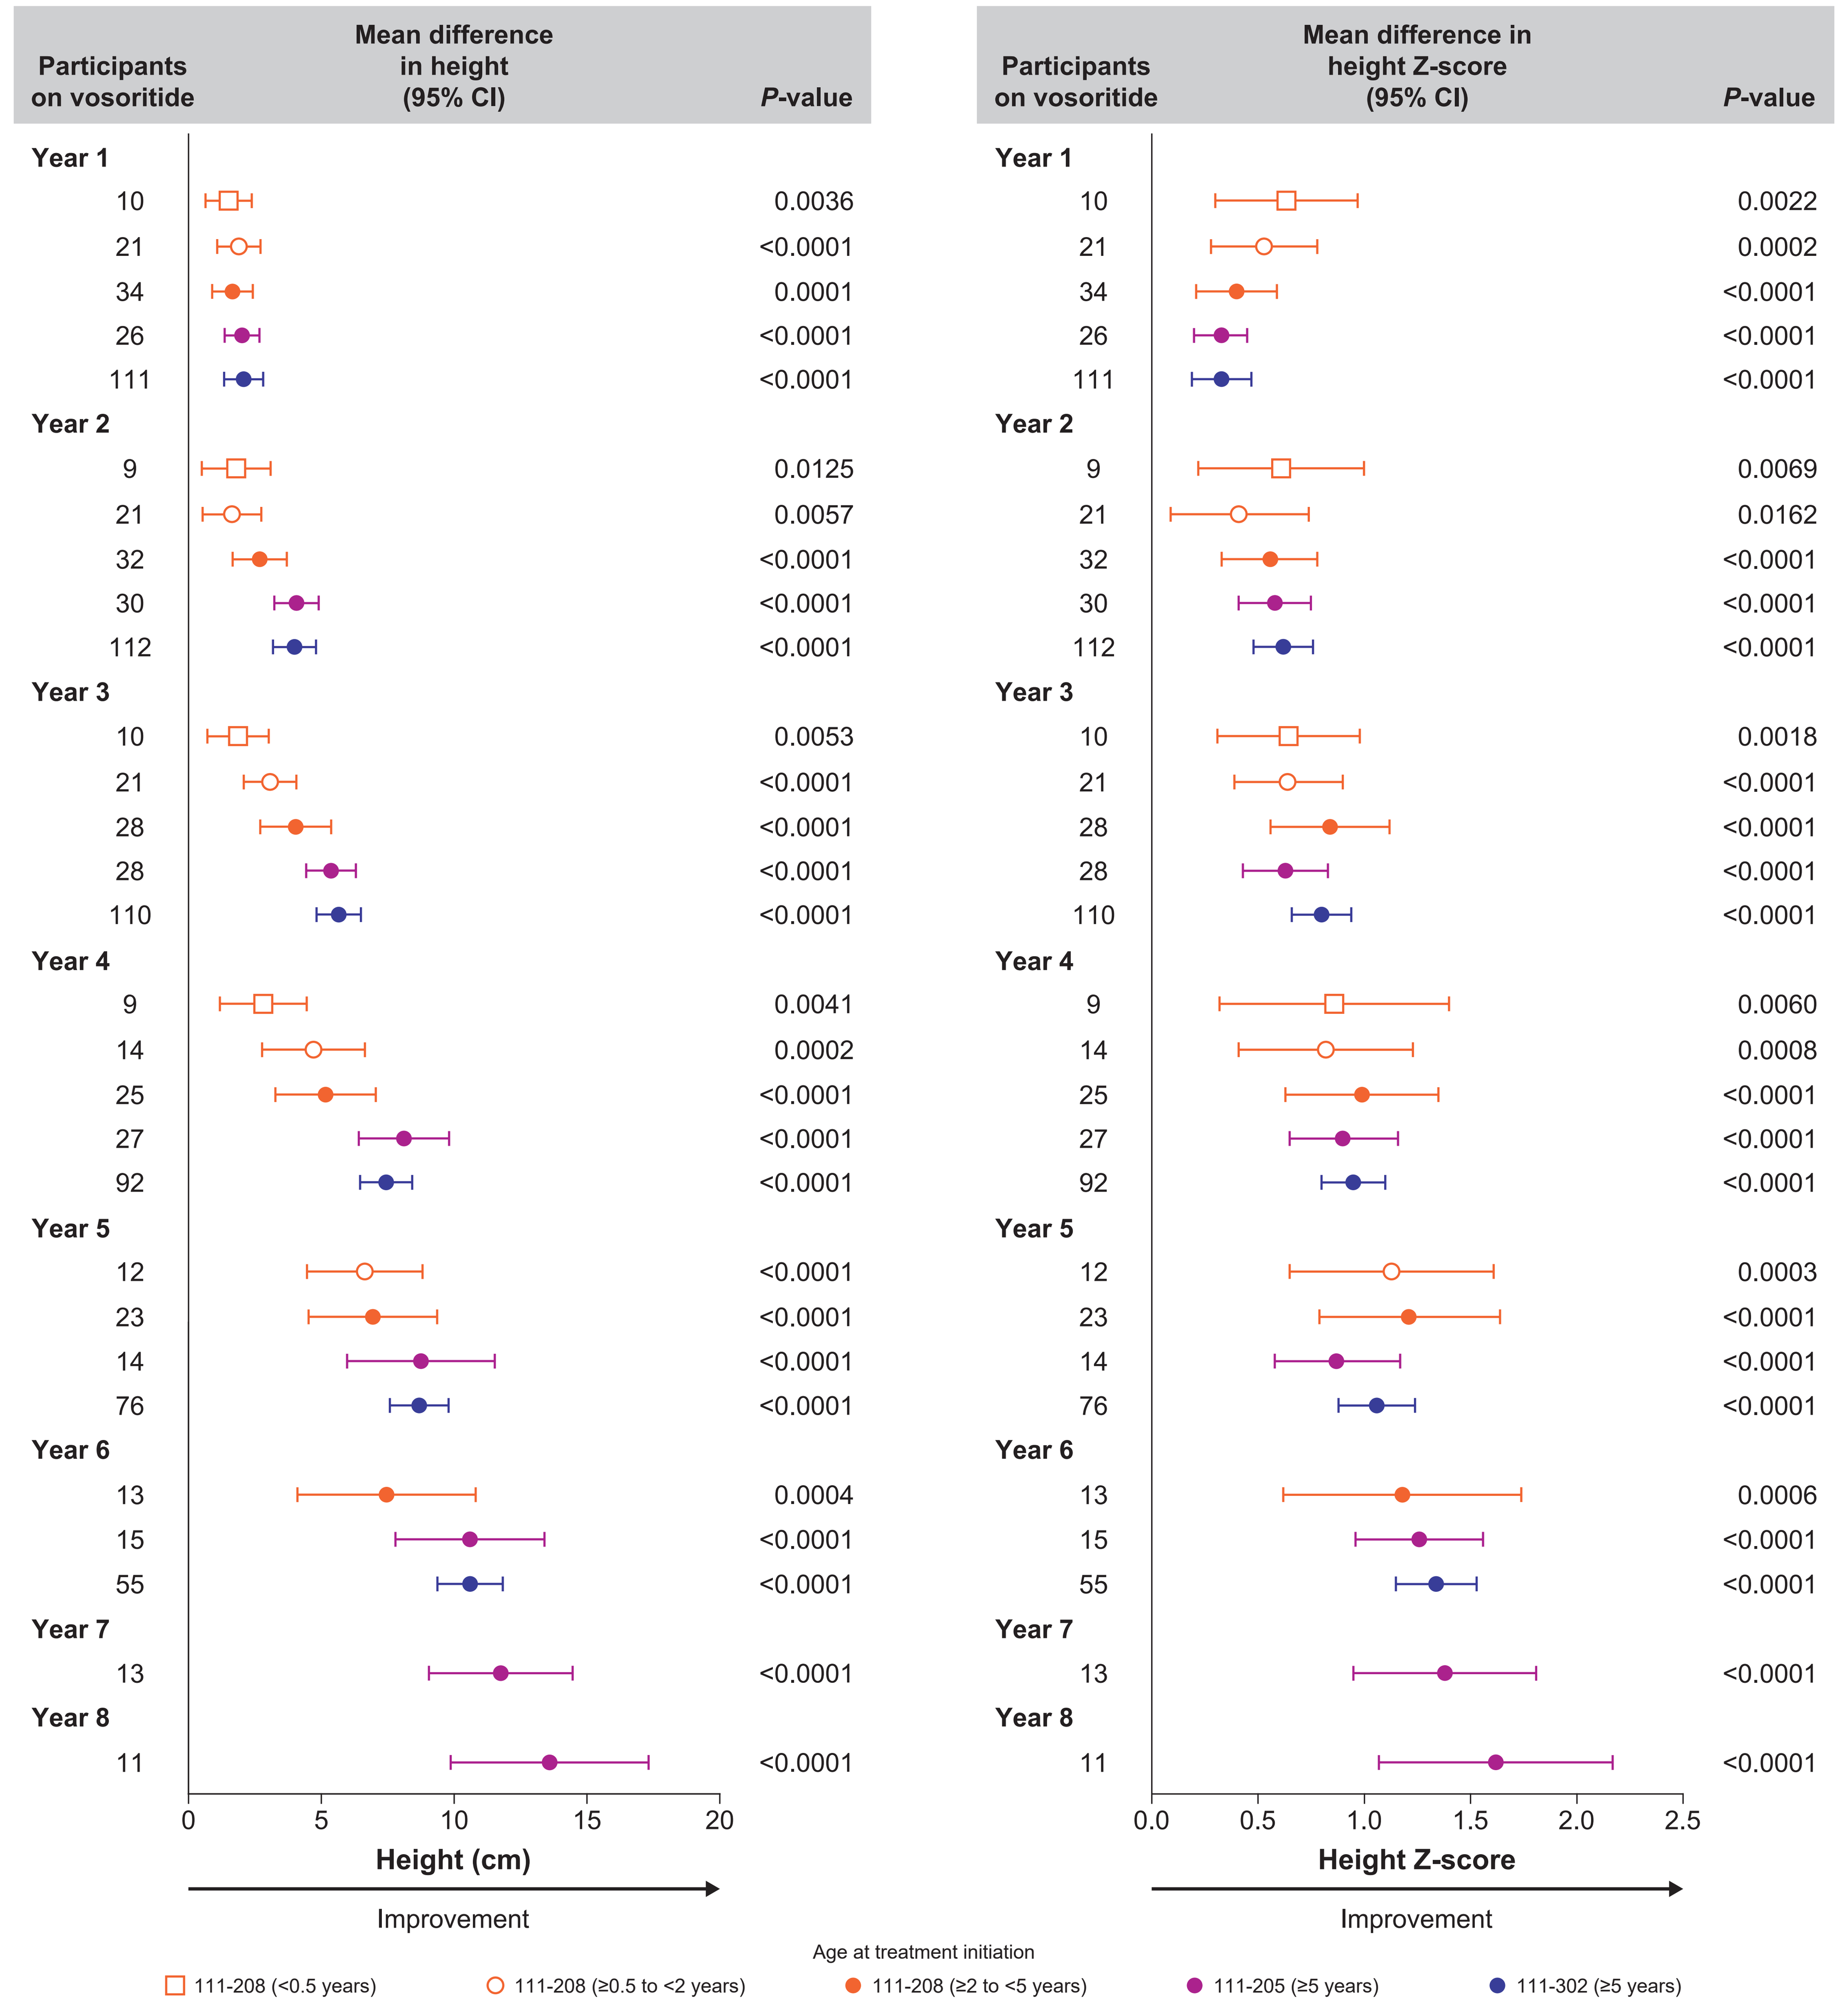
A) Height
Participants on vosoritide
Mean difference in height (95% CI)
P-value
Year 1
10 0.0036
21 <0.0001
34 0.0001
26 <0.0001
111 <0.0001
Year 2
9 0.0125
21 0.0057
32 <0.0001
30 <0.0001
112 <0.0001
Year 3
10 0.0053
21 <0.0001
28 <0.0001
28 <0.0001
110 <0.0001
Year 4
9 0.0041
14 0.0002
25 <0.0001
27 <0.0001
92 <0.0001
Year 5
12 <0.0001
23 <0.0001
14 <0.0001
76 <0.0001
Year 6
13 0.0004
15 <0.0001
55 <0.0001
Year 7
13 <0.0001
Year 8
11 <0.0001
0 5 10 15 20
Height (cm)
Improvement
B) Height Z-score (average stature-referenced)
Participants on vosoritide
Mean difference in height Z-score (95% CI)
P-value
Year 1
10 0.0022
21 0.0002
34 <0.0001
26 <0.0001
111 <0.0001
Year 2
9 0.0069
21 0.0162
32 <0.0001
30 <0.0001
112 <0.0001
Year 3
10 0.0018
21 <0.0001
28 <0.0001
28 <0.0001
110 <0.0001
Year 4
9 0.0060
14 0.0008
25 <0.0001
27 <0.0001
92 <0.0001
Year 5
12 0.0003
23 <0.0001
14 <0.0001
76 <0.0001
Year 6
13 0.0006
15 <0.0001
55 <0.0001
Year 7
13 <0.0001
Year 8
11 <0.0001
0.0 0.5 1.0 1.5 2.0 2.5
Height Z-score
Improvement
Age at treatment initiation
111-208 (<0.5 years)
111-208 (≥0.5 to <2 years)
111-208 (≥2 to <5 years)
111-205 (≥5 years)
111-302 (≥5 years)

Baseline-adjusted cross-sectional analyses were performed using achondroplasia natural history (CLARITY)[7] as a comparator. Results from 111-205 were rebaselined, with day 1 being the day of the first dose of 15 or 30 µg/kg vosoritide in study 111-202.
CI, confidence interval.

Arm span

- Over time, arm span Z-scores (achondroplasia-referenced) continued to increase for participants in all 3 studies (**Figure 3**), while arm span-to-height ratio remained stable across all years of treatment (data not shown)

Figure 3. Arm span Z-score (achondroplasia-referenced) improved over time across all ages independent of age at treatment initiation

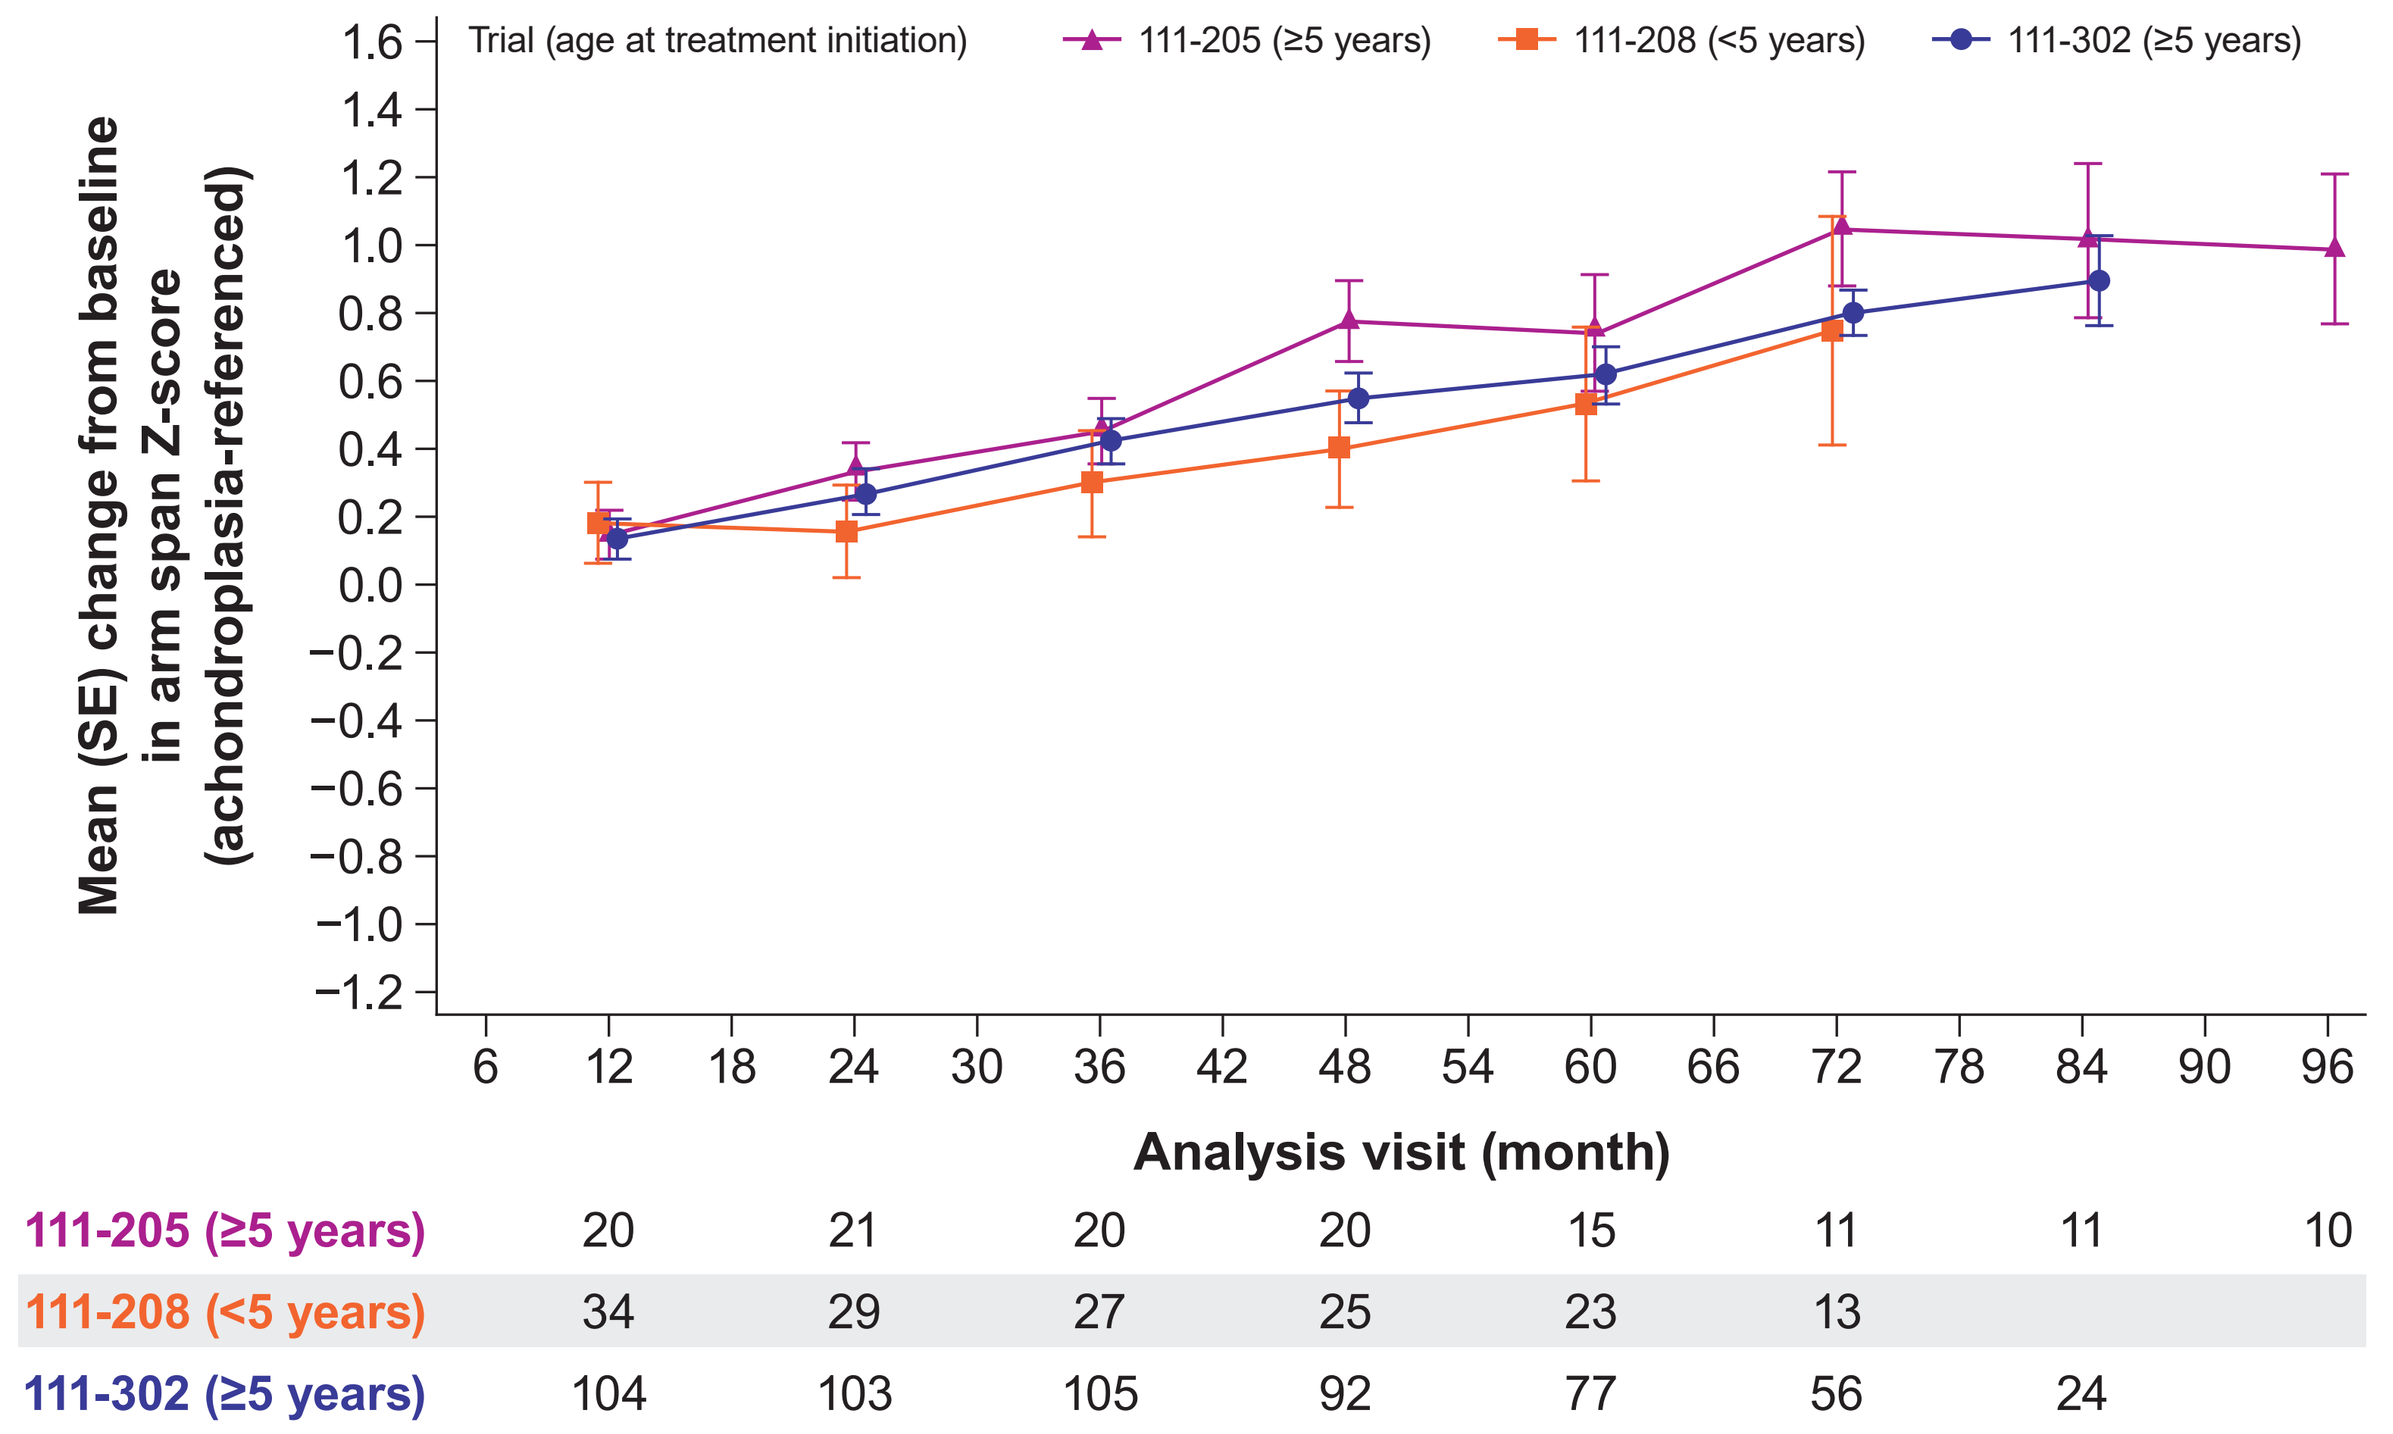
Trial (age at treatment initiation)
111-205 (≥5 years)
111-208 (<5 years)
111-302 (≥5 years)
Mean (SE) change from baseline in arm span Z-score (achondroplasia-referenced)
1.6 1.4 1.2 1.0 0.8 0.6 0.4 0.2 0.0 −0.2 −0.4 −0.6 −0.8 −1.0 −1.2
6 12 18 24 30 36 42 48 54 60 66 72 78 84 90 96
Analysis visit (month)
111-205 (≥5 years) 20 21 20 20 15 11 11 10
111-208 (<5 years) 34 29 27 25 23 13
111-302 (≥5 years) 104 103 105 92 77 56 24

Arm span Z-scores were referenced to age- and sex-matched untreated children with achondroplasia.[9]
SE, standard error.

Safety

- The long-term safety profile of vosoritide across 10,638 person-years of exposure in clinical trials and the commercial setting remained manageable and consistent with the previous reports (**Table 2**)
- No slipped capital femoral epiphysis events were observed across all 3 studies

Table 2. Safety summary

	111-208 N = 73 369.94 PY		111-205 N = 30 241.46 PY		111-302 N = 119 686.58 PY	
	Incidence %	Event rate/PY	Incidence %	Event rate/PY	Incidence %	Event rate/PY
AEs	98.6	6.982	100	5.39	99.2	3.57
Adverse drug reactions[a]						
ISRs[b]	20.5	0.387	56.7	0.18	10.9	0.05
Hypotension[c]	2.7	0.008	46.7	0.14	14.3	0.03
Fatigue	2.7	0.005	13.3	0.03	7.6	0.015
Dizziness	6.8	0.032	23.3	0.05	12.6	0.033
Syncope	1.4	0.003	3.3	0.01	1.7	0.003
Pre-syncope	1.4	0.003	13.3	0.02	6.7	0.013
Nausea	6.8	0.032	20.0	0.05	12.6	0.033
Hypertrichosis[d]	4.1	0.008	0	0	1.7	0.004
Femur fracture	2.7	0.005	6.7	0.01	4.2	0.007
Discontinuation due to AEs	1.4	0.005	3.3	0.004	0.8	0.001
SAEs	28.8	0.087	26.7	0.05	25.2	0.07
Related SAEs	1.4	0.003	0	0	1.7	0.01
Grade ≥3 AEs[e]	24.7	0.070	30.0	0.05	21.8	0.06

[a]Identified for vosoritide. Additionally, increased alkaline phosphatase was identified as an adverse drug reaction based on laboratory findings and occurred at a frequency of >10% in the pivotal phase 3 study CANOPY ACH-3 (111-301). [b]ISRs include Grade ≥2 events or events lasting for >24 hours (excluding bruising). [c]Includes both symptomatic and asymptomatic events. [d]Includes preferred terms of hypertrichosis and hair growth abnormal. [e]Severity based on CTCAE grade.
AE, adverse event; CTCAE, Common Terminology Criteria for Adverse Events; ISR, injection site reaction; PY, person-years; SAE, serious AE.

Conclusions

- Vosoritide provides durable and consistent improvements in growth for children with achondroplasia, with height and arm span Z-scores consistently increasing over time, compared to untreated children across all ages
- Vosoritide offers a growth benefit across age groups regardless of age at treatment initiation
- Vosoritide maintains a well-established safety profile with up to 8 years of follow-up and high treatment adherence

References

1. Savarirayan R, et al. *Lancet Child Adolesc Health*. 2024;8(4):301–10. **2.** Voxzogo. FDA packet insert. BioMarin Pharmaceutical Inc. 2021. **3.** Savarirayan R, et al. *Nat Rev Endocrinol*. 2025;21:314–24. **4.** Savarirayan R, et al. *Lancet Child Adolesc Health*. 2024;8(1):40–50. **5.** Savarirayan R, et al. *N Engl J Med*. 2019;381(1):25–35. **6.** Savarirayan R, et al. *Lancet*. 2020;396(10252):684–92. **7.** Hoover-Fong J, et al. *Genet Med*. 2021;23:1498–505. **8.** US Centers for Disease Control and Prevention. CDC growth charts. National Center for Health Statistics. Accessed January 29, 2026. https://www.cdc.gov/growthcharts/clinical_charts.htm. **9.** Merker A, et al. *Am J Med Genet*. 2018;176A:1819–29.

Acknowledgments

We thank all trial participants, their families, study-site staff, and investigators. Medical writing support was provided by Morgan Engelhart Wynn, PhD, of Red Nucleus, and funded by BioMarin Pharmaceutical Inc. Project management support was provided by Justin Potuzak, PhD, of BioMarin Pharmaceutical Inc.

Disclosures

RS has received consulting fees and travel support from Ascendis, BioMarin Pharmaceutical Inc., and QED Therapeutics and has participated as a clinical trial investigator for Ascendis, BioMarin Pharmaceutical Inc., QED Therapeutics, and Sanofi. **CAB** has received consulting fees from BioMarin Pharmaceutical Inc. and has participated as a clinical trial investigator for Ascendis, BioMarin Pharmaceutical Inc., and Roche. **JEH-F** has participated in advisory boards and served as a consultant for Ascendis, AstraZeneca, BioMarin Pharmaceutical Inc., Children's Mercy Research Institute, Medscape, Pfizer, QED Therapeutics, and Therachon and has received research support from AstraZeneca, BioMarin Pharmaceutical Inc., and QED Therapeutics. **CR**, **SM**, and **AHL** are employees and stockholders of BioMarin (UK) Ltd. **RR** is an employee and stockholder of BioMarin Pharmaceutical Inc. **LEP** has received honoraria from BioMarin Pharmaceutical Inc., participated on an advisory board for RegenXBio, and consulted for BioMarin Pharmaceutical Inc. and Denali.

To view a copy of this poster, scan this QR code.

Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission from the authors.

Presented at the Pediatric Endocrine Society (PES) annual meeting; April 30–May 3, 2026; San Francisco, CA, USA

©2026 BioMarin Pharmaceutical Inc. All rights reserved.