Validation of the Phenylketonuria-Symptom Severity and Impact Scale (PKU-SSIS) in adolescents with PKU

Oko-Osi, H[1], Howerter A[2], Alfayoumi I[2], Bhargava T[2], Delaney K[1], Cook K[2], Dashiell-Aje E[1], Andrae D[1]

[1]BioMarin Pharmaceutical Inc., Novato, CA USA; [2]Clinical Outcomes Solutions, Tucson, AZ USA

PCR142

Background

- Phenylketonuria (PKU) is a rare inherited metabolic disorder resulting in elevated blood and brain phenylalanine (Phe)
- Phe regulation is vital in adolescents with PKU for maintaining cognitive, psychological, and emotional symptoms that reduce health-related quality of life
- Despite treatment options, many adolescents continue to experience persistent neuropsychological symptoms and daily impacts on their quality of life
- The Phenylketonuria Symptom Severity and Impacts Scale (PKU-SSIS; **Figure 2**) was developed as a disease-specific patient-reported outcome (PRO) measure to assess PKU-related symptoms and their impact on daily life (Quinn et al., 2022)

Objective

To evaluate the psychometric properties of the PKU-SSIS Adolescent Version in adolescents with PKU

Methods

Sample

Adolescents with PKU aged 12–17 years and their parent or guardian were recruited from the United States and Canada through patient networks, advocacy groups, and Rare Patient Voice

Study Design

A prospective, non-interventional study was conducted using online surveys administered at two timepoints. Adolescent participants completed the PKU-SSIS Adolescent version and additional PRO measures, while caregivers completed global impression measures (**Figure 1**)

Participants completed two surveys:

- Survey 1: within 48 hours of consent and enrollment
- Survey 2: approximately 10 days after Survey 1

Invitations were sent at the 10-day mark and participants had up to 1 week to complete each survey

Figure 1. Study design and assessment schedule

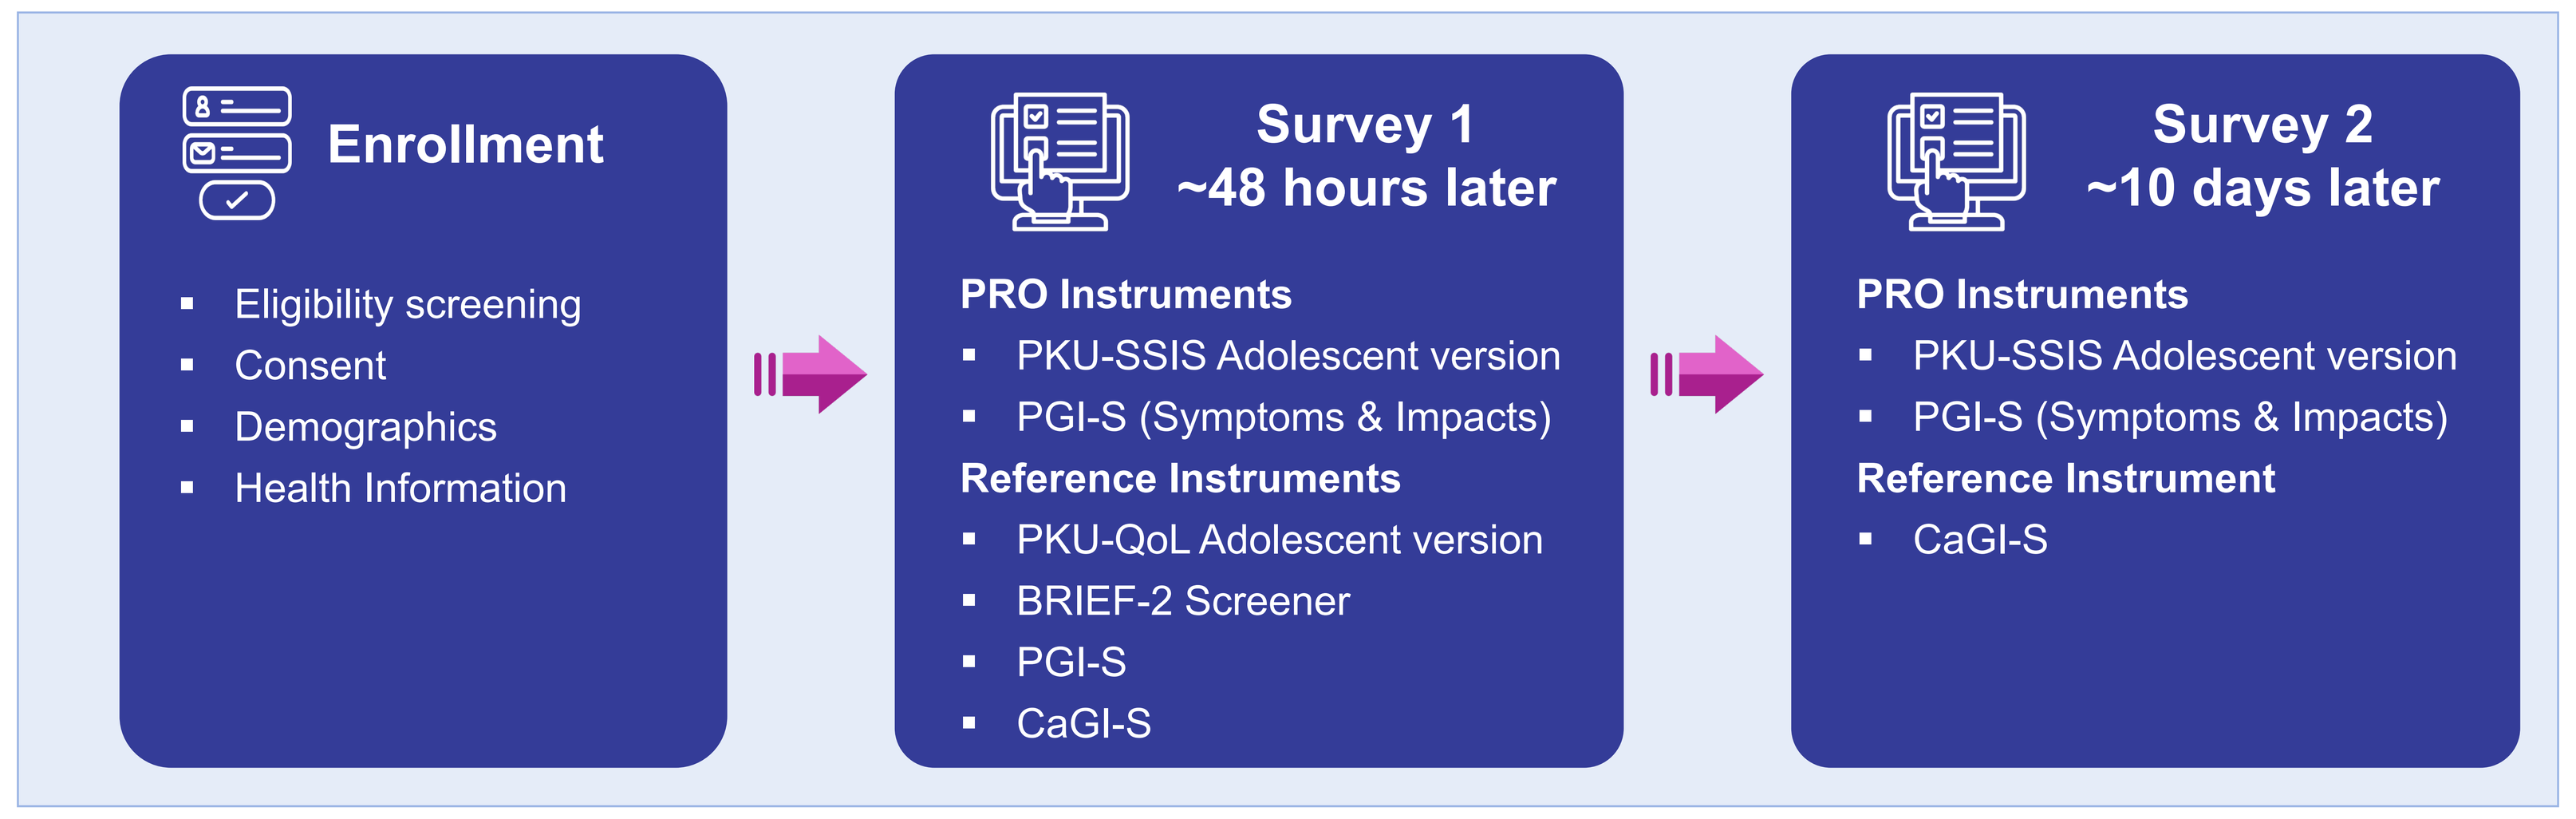
Enrollment
Eligibility screening
Consent
Demographics
Health Information
Survey 1 ~48 hours later
PRO Instruments
PKU-SSIS Adolescent version
PGI-S (Symptoms & Impacts)
Reference Instruments
PKU-QoL Adolescent version
BRIEF-2 Screener
PGI-S
CaGI-S
Survey 2 ~10 days later
PRO Instruments
PKU-SSIS Adolescent version
PGI-S (Symptoms & Impacts)
Reference Instrument
CaGI-S

Abbreviations: BRIEF-2 Screener = Behaviour Rating Inventory of Executive Function 2nd Edition, CGI-S = Caregiver Global Impression of Severity - Symptoms and Impact, PGI-S = Patient Global Impression of Severity - Symptoms and Impacts, PKU-QoL = Phenylketonuria Quality of Life Adolescent Version, PKU-SSIS = Phenylketonuria Symptom Severity and Impacts Scale Adolescent version

PKU-SSIS Adolescent Version

Concepts developed by Quinn et al. (2022) created a 22-item patient-reported instrument designed to evaluate neuropsychological symptoms and daily life impacts associated with PKU (**Figure 2**)

Higher scores indicate greater symptom severity and impact

Response options included:

- Never, Rarely, Occasionally, Frequently, Very Frequently
- Very Dissatisfied, Dissatisfied, Neutral, Satisfied, Very Satisfied

Figure 2. PKU-SSIS Adolescent version concepts

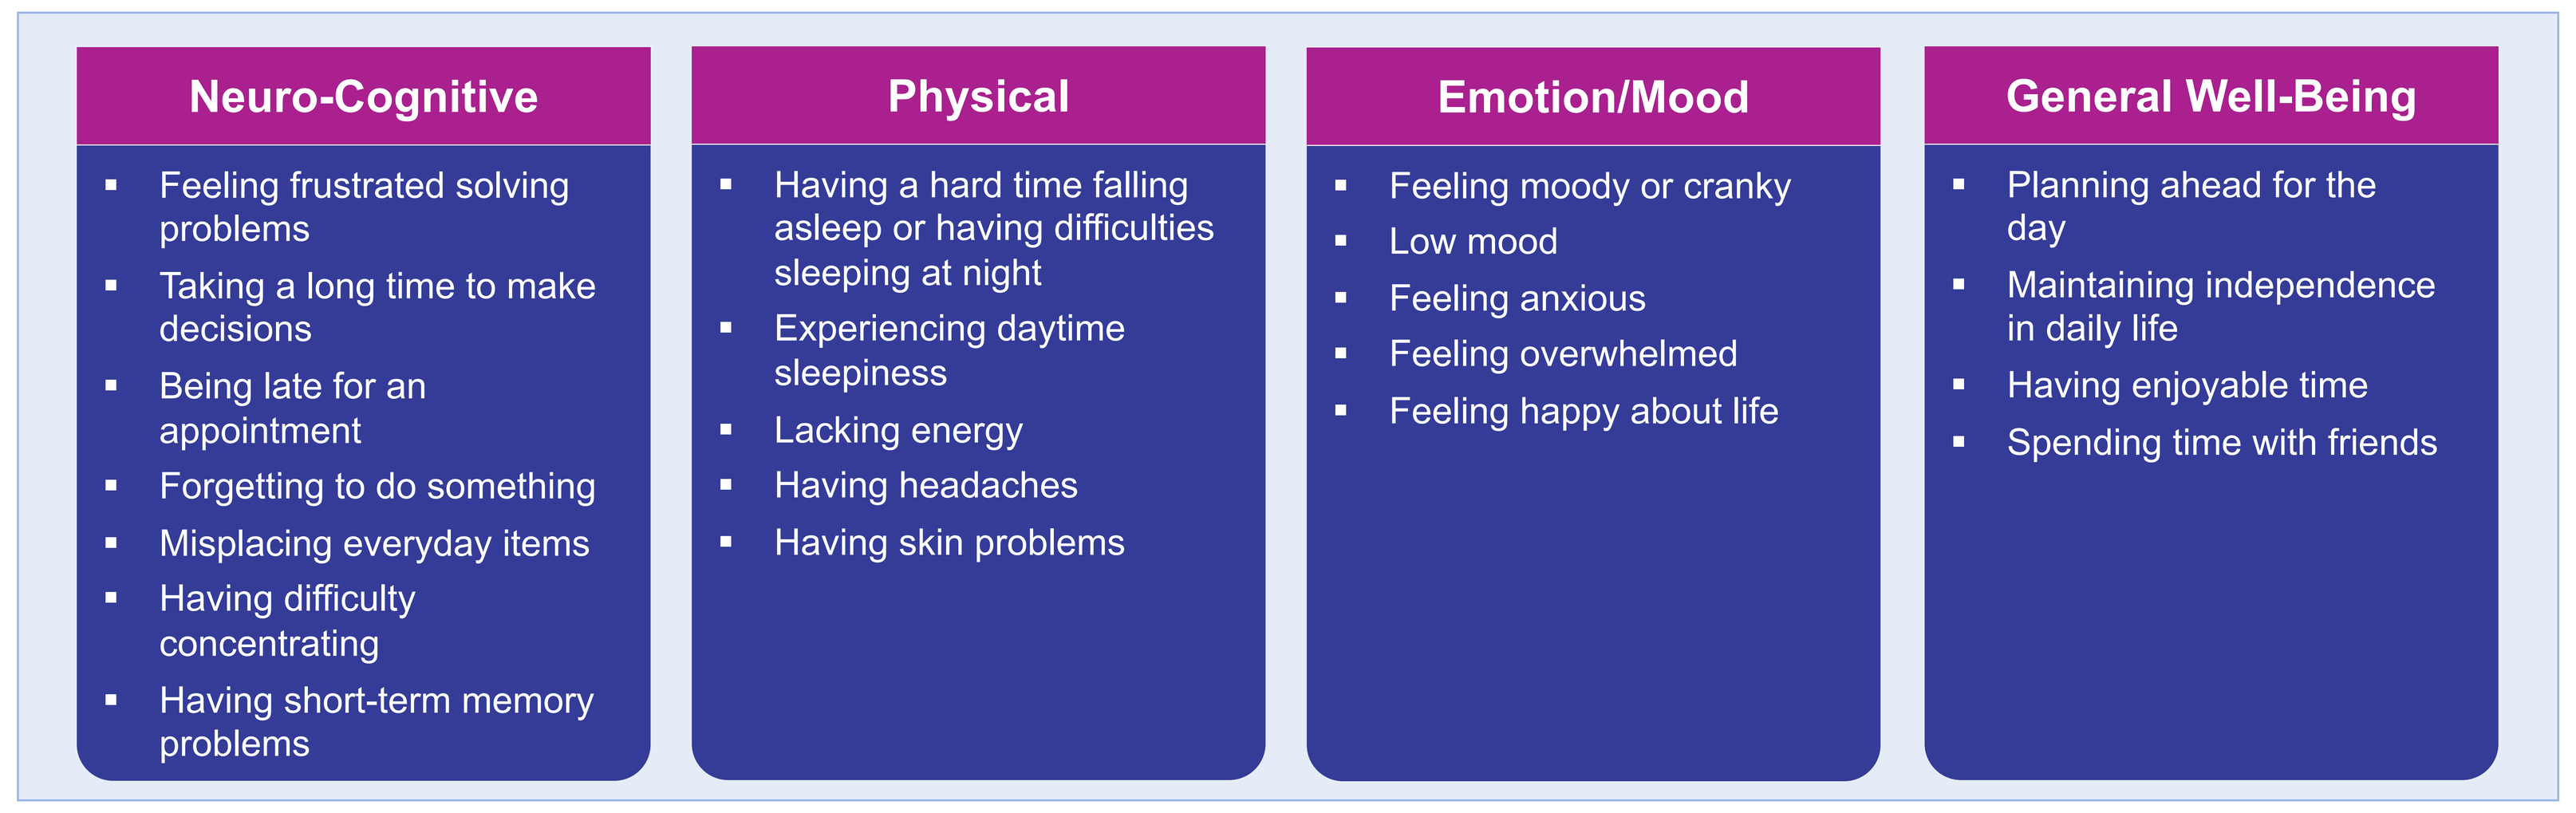
Neuro-Cognitive
Feeling frustrated solving problems
Taking a long time to make decisions
Being late for an appointment
Forgetting to do something
Misplacing everyday items
Having difficulty concentrating
Having short-term memory problems
Physical
Having a hard time falling asleep or having difficulties sleeping at night
Experiencing daytime sleepiness
Lacking energy
Having headaches
Having skin problems
Emotion/Mood
Feeling moody or cranky
Low mood
Feeling anxious
Feeling overwhelmed
Feeling happy about life
General Well-Being
Planning ahead for the day
Maintaining independence in daily life
Having enjoyable time
Spending time with friends

Results

Population

The study included **57 adolescents with PKU** and **51 caregivers**

- Adolescents had a **mean age of 14.6 years** (SD = 1.65) and were evenly distributed by sex
- Most participants identified as White (88%) and reported mild-to-moderate disease severity
- Caregivers were predominantly female (92.2%) with a **mean age of 45 years** (SD = 5.72)
- **Mean Phe levels were 6.7 mg/dL** (SD = 4.29), and the majority reported routine Phe testing at least monthly

All adolescents reported at least one strategy for PKU management including:

- **Phe-restricted diet (n = 35, 61%)**
- **Low-protein medical food (n = 25, 44%)**
- **Sapropterin treatment (n = 24, 42%)**

Dimensionality and Scoring Approach

Scoring of the PKU-SSIS Adult Version was informed by a series of analyses to assess dimensionality and to derive a scoring algorithm (**Figure 3**).

Figure 3. Analytic flow for evaluation of PKU-SSIS structure

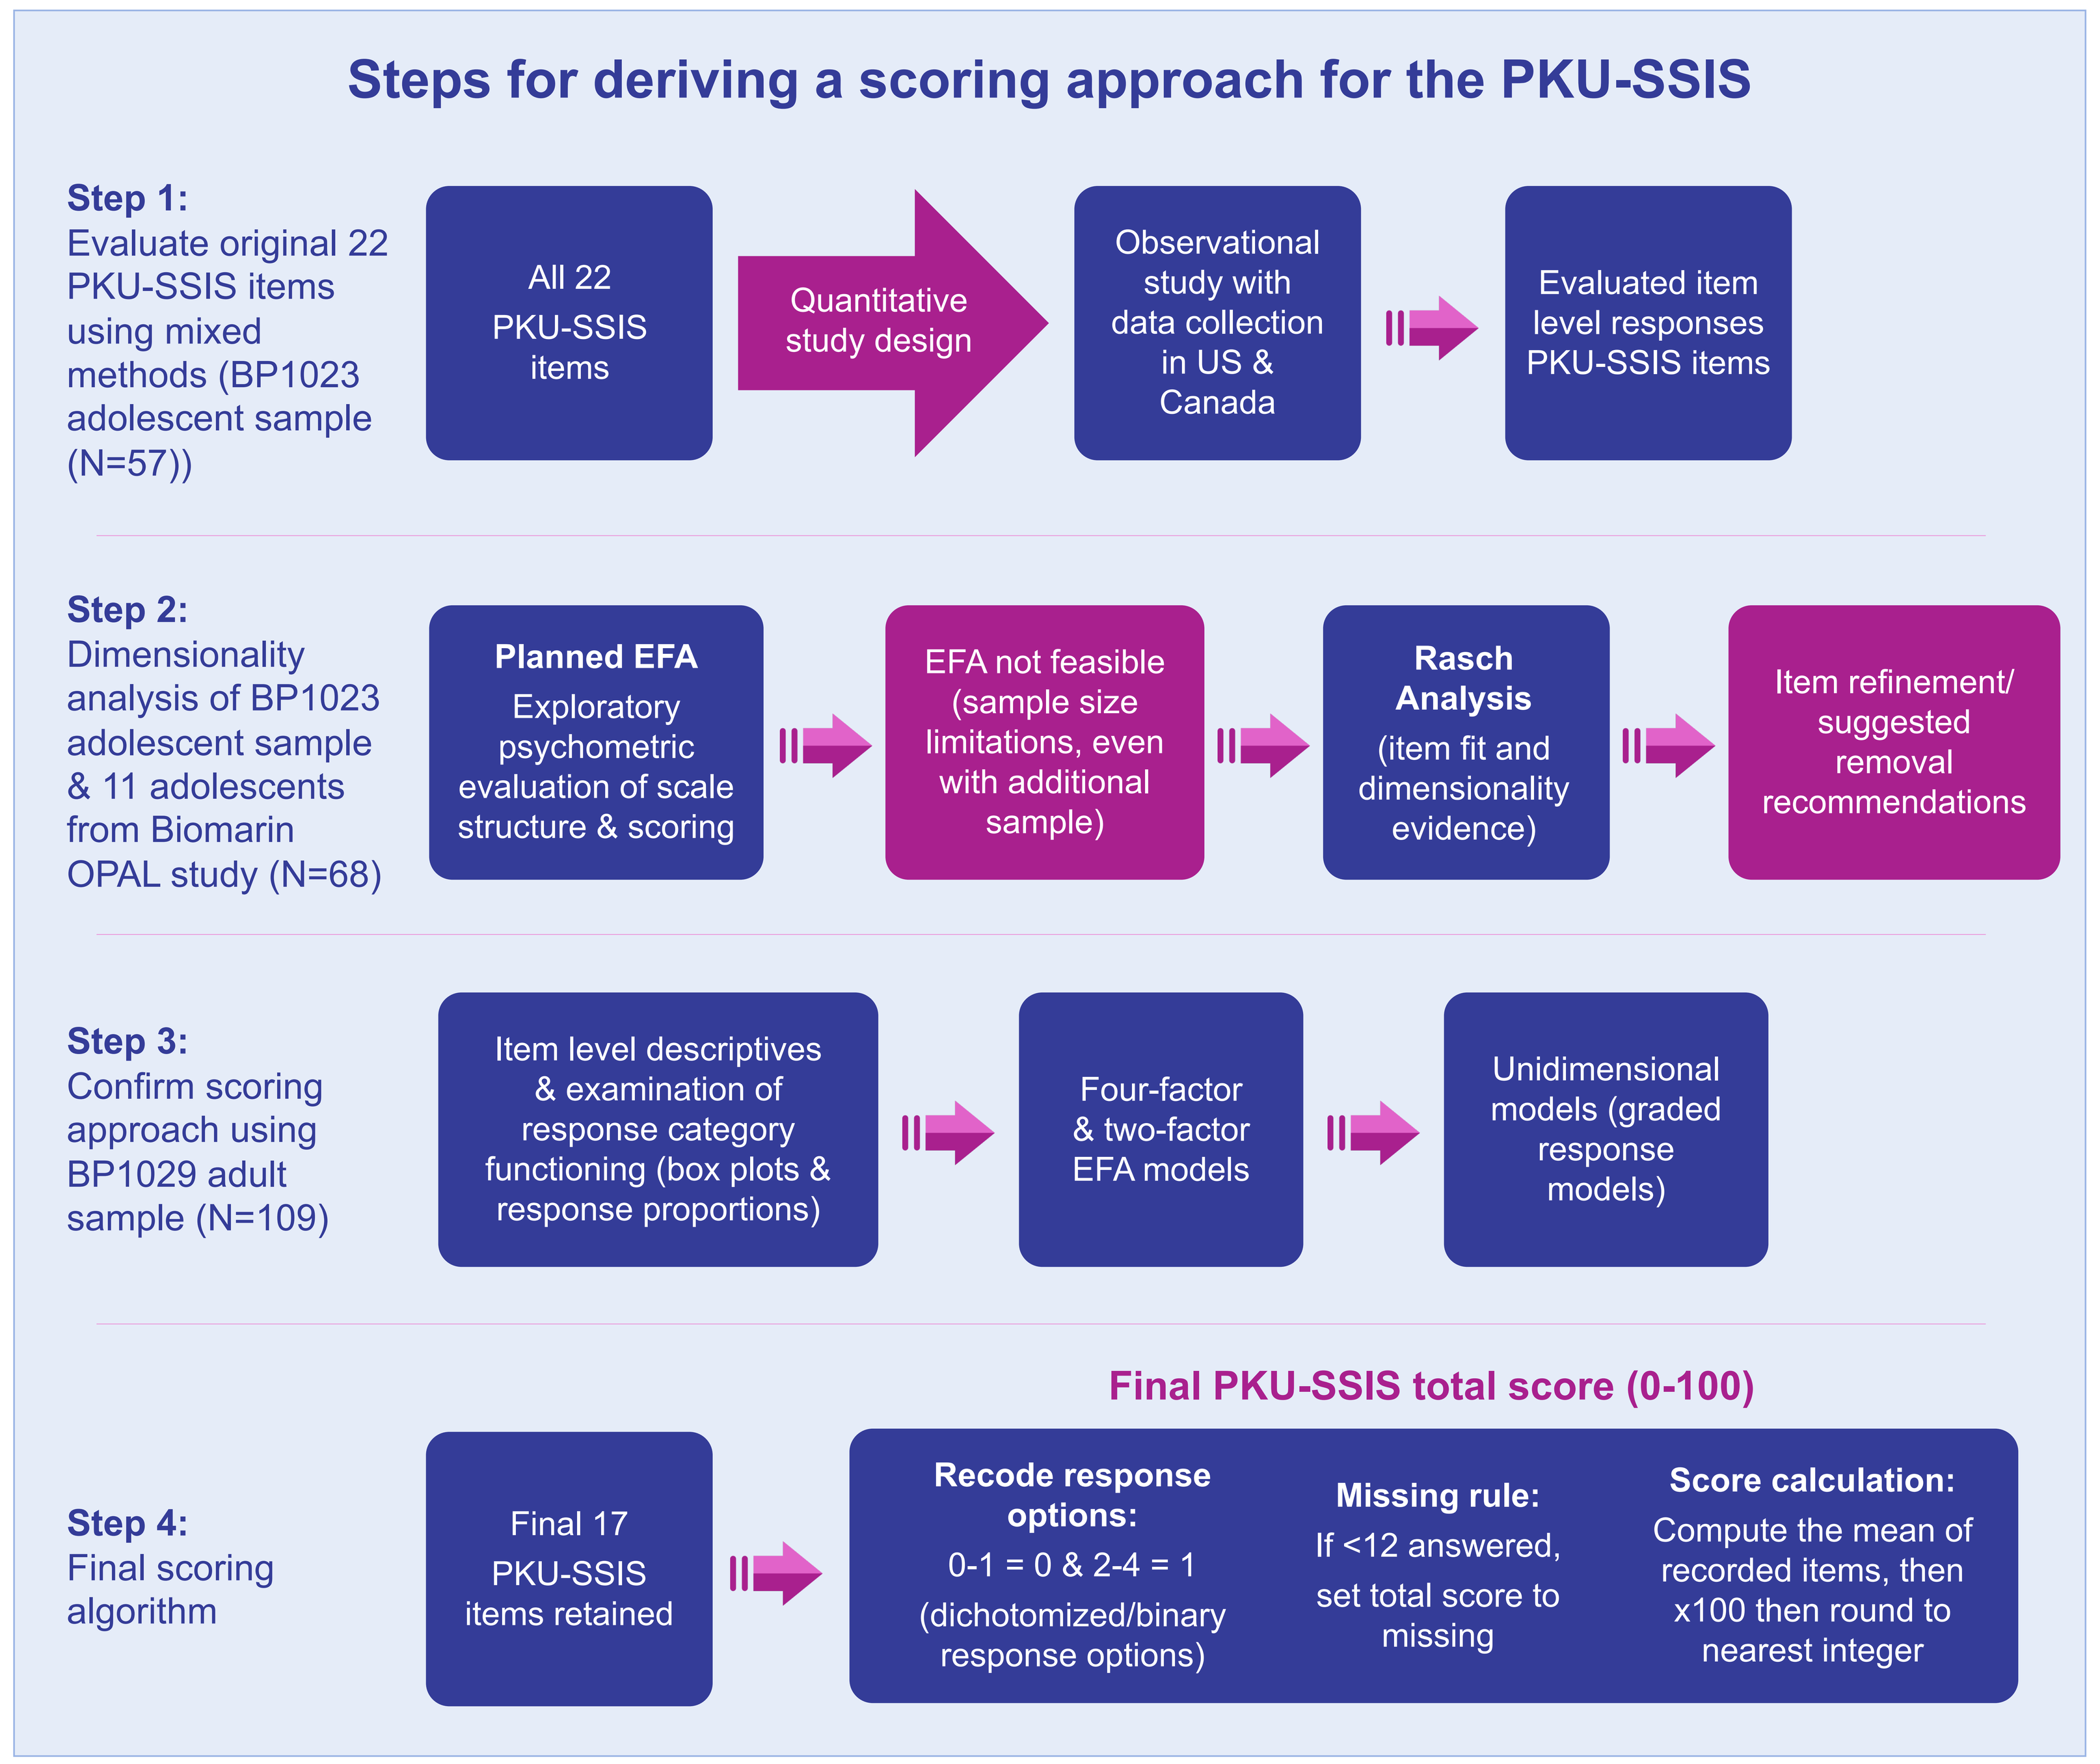
Steps for deriving a scoring approach for the PKU-SSIS
Step 1: Evaluate original 22 PKU-SSIS items using mixed methods (BP1023 adolescent sample (N=57))
All 22 PKU-SSIS items
Quantitative study design
Observational study with data collection in US & Canada
Evaluated item level responses PKU-SSIS items
Step 2: Dimensionality analysis of BP1023 adolescent sample & 11 adolescents from Biomarin OPAL study (N=68)
Planned EFA Exploratory psychometric evaluation of scale structure & scoring
EFA not feasible (sample size limitations, even with additional sample)
Rasch Analysis (item fit and dimensionality evidence)
Item refinement/ suggested removal recommendations
Step 3: Confirm scoring approach using BP1029 adult sample (N=109)
Item level descriptives & examination of response category functioning (box plots & response proportions)
Four-factor & two-factor EFA models
Unidimensional models (graded response models)
Final PKU-SSIS total score (0-100)
Step 4: Final scoring algorithm
Final 17 PKU-SSIS items retained
Recode response options: 0-1 = 0 & 2-4 = 1 (dichotomized/binary response options)
Missing rule: If <12 answered, set total score to missing
Score calculation: Compute the mean of recorded items, then x100 then round to nearest integer

EFA = Exploratory Factor Analysis; Final 17 items in PKU-SSIS for scoring were 1 – 15, 18, 19

Step 1:

- Item level examination of PKU-SSIS items, item distribution in adolescent sample

Step 2:

- Sample was expanded with additional 11 adolescents from the BioMarin OPAL study. EFA and Rasch analysis provided suggestions for item removal, however, not definitive due to sample size

Step 3:

- Examined adult study sample (n=109) with a series of models to best fit the data
- A final graded response model was examined using a binary scoring approach which was the best fit overall to balance model fit with model parsimony

Step 4:

- The final scoring approach focused on the PKU-SSIS total score, with higher scores indicating greater symptom severity and impact

Scale-Level Analysis

Psychometric properties of the PKU-SSIS total score included construct validity, item-total correlations, internal consistency reliability, and test-retest reliability. (**Table 2** and **Table 3**)

Table 2. Construct Validity of PKU-SSIS Total Score

Concept	Measure	r	Interpretation
Cognitive	BRIEF-2 Total	0.709	Strong
	Lack of concentration[1]	0.647	Moderate
	Slow thinking[1]	0.664	Moderate
Emotional	Moodiness / sadness / anxiety[1]	0.617–0.718	Moderate–Strong
	Irritability / aggressiveness[1]	0.473–0.593	Moderate
Physical	Tiredness / headache / stomach ache[1]	0.473–0.664	Moderate
Disease Impact	PKU overall impact[1]	0.558	Moderate
	PKU diet impact[1]	0.441	Moderate
	Dietary adherence[1]	0.194	Weak
Global (PRO)	PGI-S Severity / Impacts	0.611, 0.659	Moderate
Global (Observer)	CaGI-S Severity	0.460	Moderate
	CaGI-S Impacts	0.239	Weak

[1]PKU-QoL Adult domain scores
[2]The magnitude of correlations was interpreted in line with FDA recommendations: less than 0.3 = weak, between 0.3 and 0.7 = moderate, between 0.7 and 0.9 = strong, and above 0.9 = very strong

Table 3. Psychometric Analyses and Criteria for Evaluation of the PKU-SSIS Adolescent version

Type of analysis	Description	Results
Item-total correlations	Item correlations with the PKU-SSIS total score. Acceptable correlations r ≥ 0.40	Items 8, 14, 19 had low correlations with the total score (r = 0.31, 0.35, 0.28); all other items above threshold (r = 0.53 – 85)
Item Discrimination	Evaluates how well each item differentiates between adolescents with higher versus lower symptom burden	Item 14: Felt happy about life (0.242) fell below the recommended discrimination threshold and was only endorsed by 19% of the sample
Internal consistency reliability	KR-20 is a special case of alpha for dichotomous data	KR-20 = 0.84
Test-retest reliability	Assessed using ICC among stable participants across two survey timepoints using change in the PGI-S ratings	ICC = 0.89 (PGI-S Symptoms) ICC = 0.90 (PGI-S Impacts)
Concurrent validity	Evaluated by comparing PKU-SSIS total scores across caregiver-reported severity groups (CaGI-S)	PKU-SSIS total scores were higher in adolescents rated by caregivers as moderate/severe/very severe vs mild on CaGI-S Severity (p = 0.0034; SES = 0.897). Mean PKU-SSIS scores demonstrate monotonicity across the severity ratings.

Conclusions

PKU-SSIS scale structure confirmed 17 items generate a total score from 0 to 100, with higher scores representing greater symptom severity and impact of PKU

Psychometric properties were strong

- Reliability: **good internal consistency** and **excellent test-retest reliability**
- Concurrent validity: Score **monotonically increases** based on severity ratings
- Convergent validity: **Moderate to strong correlations with similar constructs** like cognitive functioning and emotional symptoms
- Divergent validity: **Smaller correlations to unrelated constructs** like diet adherence

The PKU-SSIS Adolescent version is a reliable and valid measure of symptom burden and impacts in adolescents with PKU

Further research in larger and more diverse samples is warranted to confirm dimensional structure and support broader application

To view a copy of this poster, scan this QR code.

Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission from the authors.

Reference: Quinn J, Georgiadis A, Lewis HB, Jurecki E. Measuring burden of illness in phenylketonuria (PKU): development of the PKU symptom severity and impacts scale as a robust patient-reported outcome. *Advances in Therapy*. 2022;39(2):971-991.

Disclosures: DA, ED, KD are employees and shareholders of BioMarin Pharmaceutical Inc. and HO was employed by BioMarin at the time of the analysis. AH, IA, TB, KC are employees of Clinical Outcomes Solutions who was contracted by BioMarin Pharmaceutical Inc. to conduct the study.

Presented at ISPOR – The Professional Society for Health Economics and Outcomes Research US : May 17 – 20 2026, Philadelphia, PA.

©2026 BioMarin Pharmaceutical Inc. All rights reserved.