Design of a randomized, double-blind, placebo-controlled phase 2 study to evaluate the safety and efficacy of vosoritide in infants and children with hypochondroplasia aged <3 years

Poster 12

Andrew Dauber,[1,2] Moira Cheung,[3] Julie Hoover-Fong,[4] Noriyuki Namba,[5] Oliver Semler,[6] Roberto Bassi,[7] Sajda Ghani,[8] Dane Osmond,[7] Ravi Savarirayan[9]

[1]Children's National Hospital, Washington DC, USA; [2]The George Washington, University School of Medicine and Health Sciences, Washington, DC, USA; [3]NIHR Great Ormond Street Hospital Biomedical Research Centre, Great Ormond St, London, UK; [4]Johns Hopkins University, Baltimore, MD, USA; [5]Faculty of Medicine, Tottori University, Yonago, Tottori, Japan; [6]Faculty of Medicine, Center for Rare Diseases, University Hospital Cologne, University of Cologne, Cologne, Germany; [7]BioMarin Pharmaceutical Inc., Novato, CA, USA; [8]BioMarin (UK) Ltd, London, UK; [9]Murdoch Children's Research Institute, Royal Children's Hospital, and University of Melbourne, Parkville, Victoria, Australia

Introduction

- Hypochondroplasia is a rare genetic skeletal dysplasia caused by constitutive activation of the fibroblast growth factor receptor 3 (FGFR3) protein, a negative regulator of endochondral bone growth[1]
- While there are no approved targeted treatments for hypochondroplasia to date, vosoritide, a C-type natriuretic peptide analog, increased growth velocity in children aged ≥3 years with hypochondroplasia in a phase 1/2 trial (NCT04219007)[1]
- Vosoritide is approved globally for children with achondroplasia, another FGFR3-mediated skeletal dysplasia, beginning at birth and continuing until epiphyseal closure[1,2]
- In the phase 2 CANOPY ACH-2I trial (111-206; NCT03583697), vosoritide improved growth in children with achondroplasia aged <5 years, including many <3 years, showing the potential of early treatment to maximize growth and clinical benefit for children with hypochondroplasia[2-4]
- Altogether, prior clinical trial results support the assessment of vosoritide in young children with hypochondroplasia

Objective

- CANOPY HCH-2I (111-212, NCT07126262) is a randomized, double-blind, placebo-controlled, phase 2 clinical trial that aims to evaluate the safety and efficacy of vosoritide in infants and children aged <3 years with hypochondroplasia

Participants

A planned total of 60 participants aged <3 years with genetically diagnosed hypochondroplasia will be recruited

- Hypochondroplasia-associated *FGFR3* pathogenic variant confirmed via whole genome sequencing

To be eligible, participants must have a height Z-score below average-stature references of the same sex and age, as calculated using the Centers for Disease Control and Prevention growth charts (2000)

- Participants aged 0 to <1 year: Z-score ≤ −1.0 standard deviation score (SDS)
- Participants aged ≥1 to <3 years: Z-score ≤ −2.0 SDS

Participant weight on the first visit before treatment must be ≥3 kg

Study sites

- CANOPY HCH-2I is recruiting from sites in the USA, Australia, France, Germany, Italy, Japan, and the UK

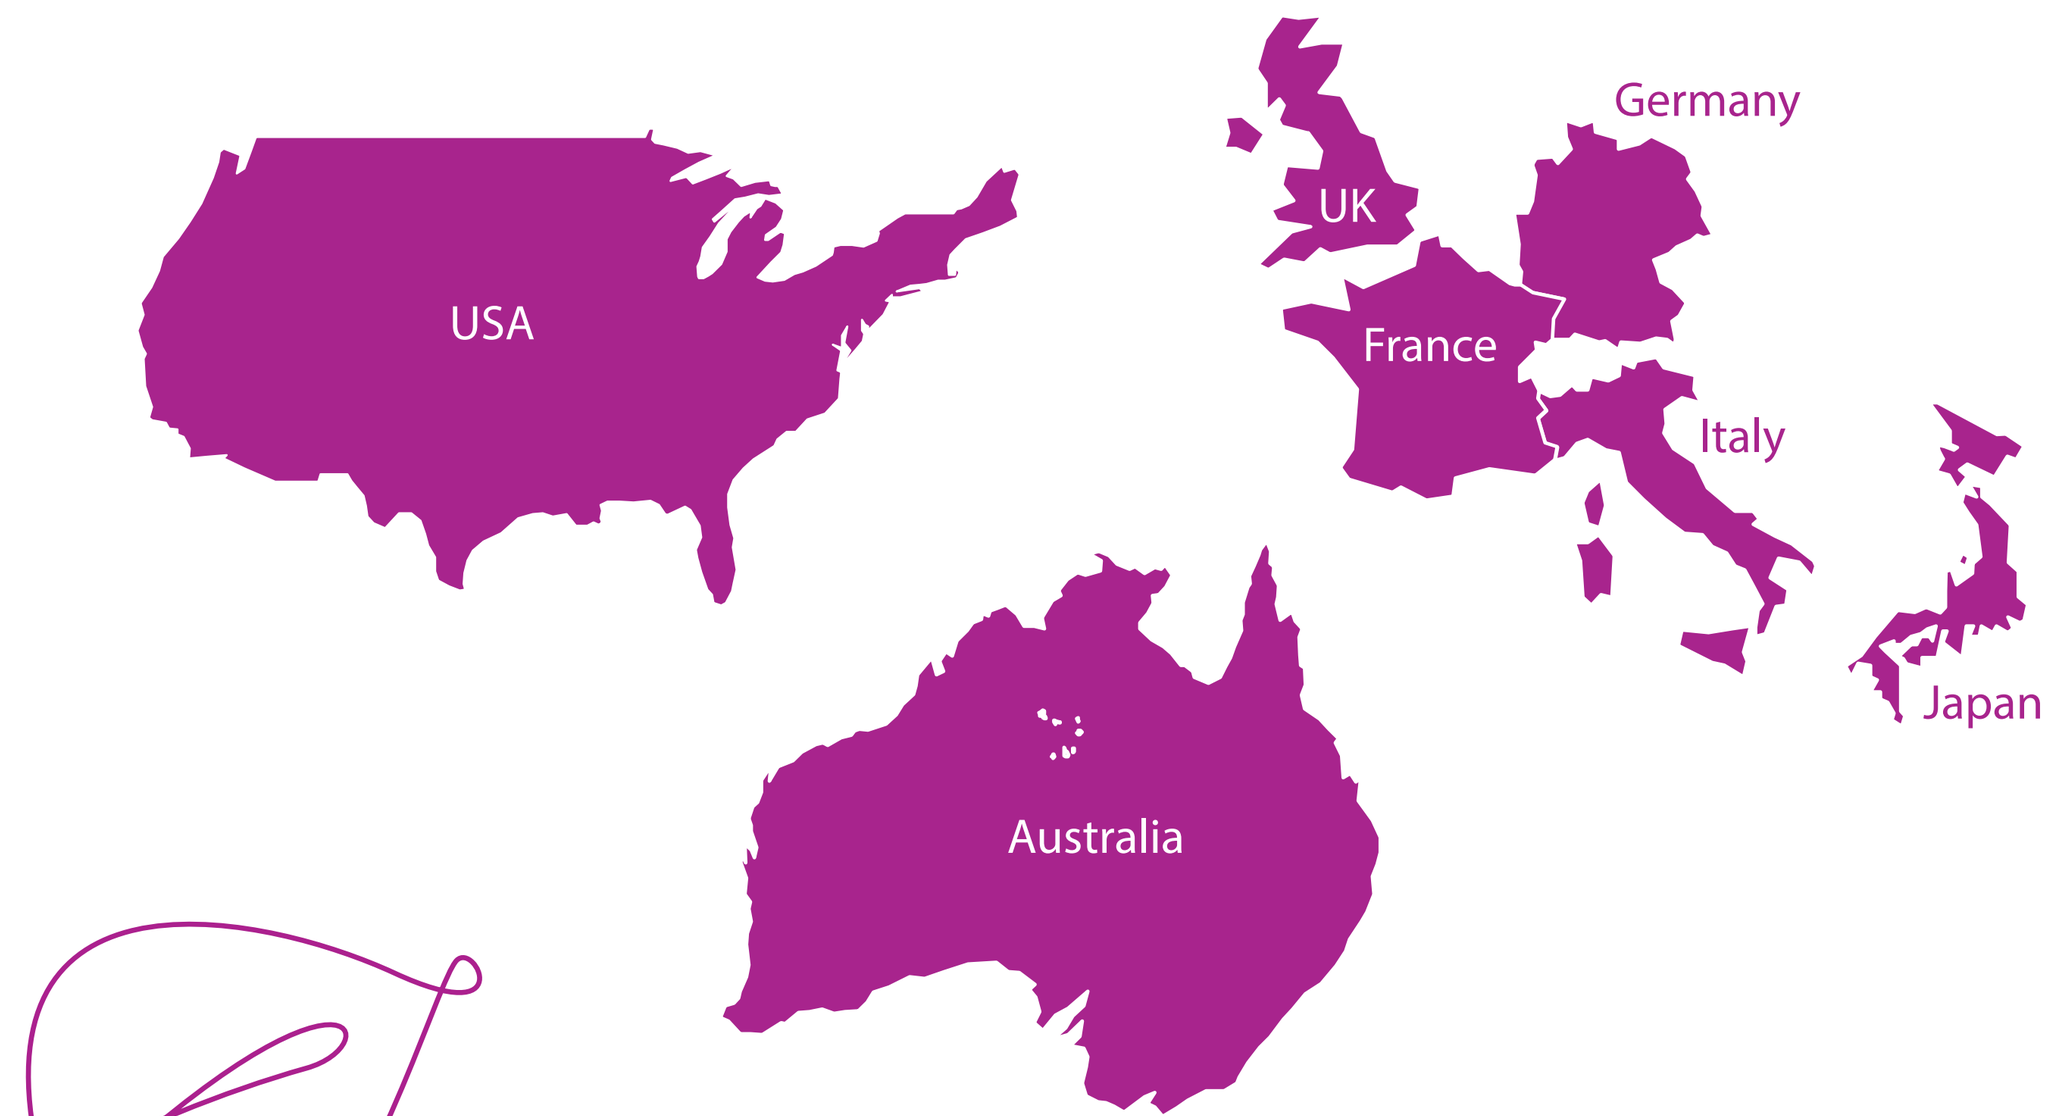
USA
UK
Germany
France
Italy
Japan
Australia

Key dates

First site activated
July 30, 2025

First participant enrolled
August 4, 2025

First participant randomized
August 19, 2025

Enrollment ongoing

Study design

- Participants will be enrolled and randomized 1:1 to receive daily subcutaneous injections of vosoritide or placebo over 52 weeks (**Figure 1**) and assessed for several primary and secondary endpoints (**Table 1**)
 - Vosoritide dosing will follow a weight-band regimen
 - Randomization will be stratified across 3 age groups:
 - 0 to <6 months (n ≥4)
 - ≥6 to <24 months (n ≥6)
 - ≥24 to <36 months (n ≥6)

Table 1. Primary and secondary endpoints

	Endpoints assessed for vosoritide vs placebo	Time
Primary	Safety, as measured by • TEAEs • Serious AEs • Standard clinical laboratory values • Vital signs	Throughout
	Height Z-score	Change from baseline to week 52
Secondary	Height	Change from baseline to week 52
	Cumulative AGV	At week 52
	6-month interval AGV	At weeks 26 and 52
	Upper-to-lower body segment ratio	Change from baseline to week 52
	Arm span	Change from baseline to week 52
	Bone quality as measured by DXA: • Total body (less head) BMD Z-score • Lumbar spine BMD Z-score • Total body (less head) BMC • Lumbar spine BMC	Change from baseline to week 52
	PK parameters	At prespecified time points
	PD biomarkers	At prespecified time points
	Incidence of otitis media	Throughout
	Seizure frequency	Throughout

AE, adverse event; AGV, annualized growth velocity; BMC, bone mineral content; BMD, bone mineral density; DXA, dual-energy X-ray absorptiometry; PD, pharmacodynamic; PK, pharmacokinetic; TEAE, treatment-emergent AE.

- At study end, participants have the option to enroll in an extension study (CANOPY HCH-EXT [111-308, NCT07073014]) assessing long-term safety and efficacy, which is currently underway with participants who enrolled from CANOPY HCH-3 (111-303)

Figure 1. Study design

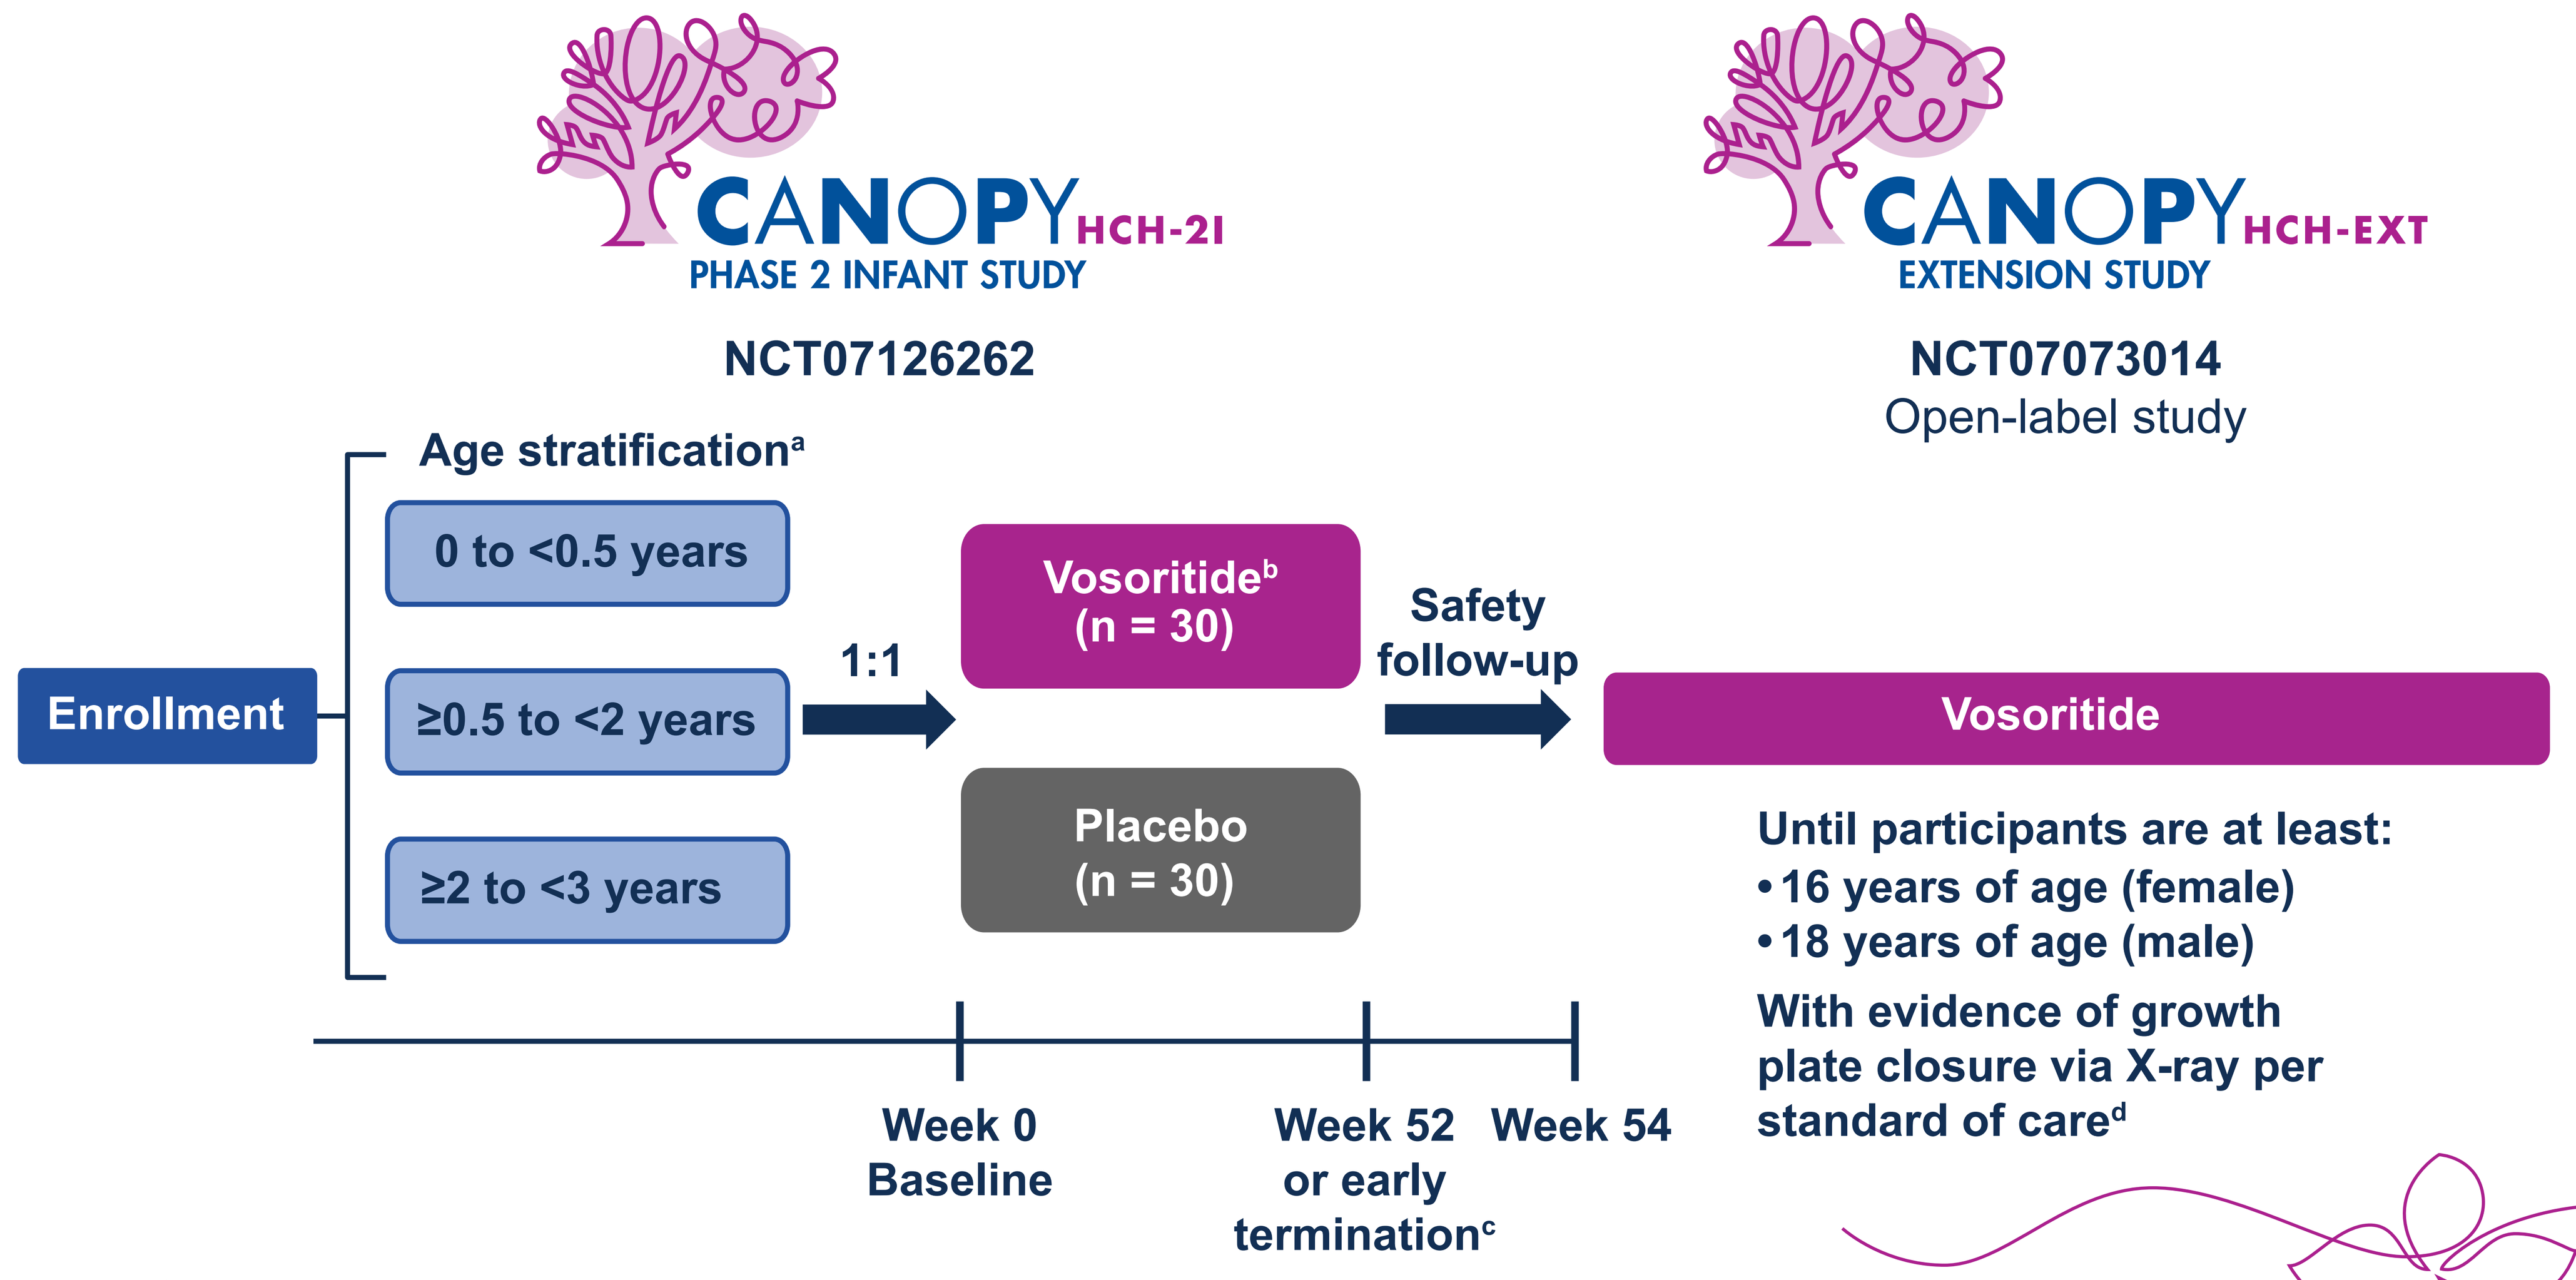
CANOPY HCH-2I
PHASE 2 INFANT STUDY
NCT07126262
CANOPY HCH-EXT
EXTENSION STUDY
NCT07073014
Open-label study
Age stratification[a]
0 to <0.5 years
≥0.5 to <2 years
≥2 to <3 years
Enrollment
1:1
Vosoritide[b] (n = 30)
Placebo (n = 30)
Safety follow-up
Vosoritide
Until participants are at least:
• 16 years of age (female)
• 18 years of age (male)
With evidence of growth plate closure via X-ray per standard of care[d]
Week 0 Baseline
Week 52 or early termination[c]
Week 54

Radiographs of lower extremities and the spine will be taken at baseline and week 52.
[a]A minimum of 4 participants are planned to be enrolled in the 0 to <0.5 years age group and at least 6 participants per group in the ≥0.5 to <2 years and ≥2 to <3 years age groups.
[b]The vosoritide dose will be based on the participant's weight and will follow the weight-band dosing regimen approved for achondroplasia. Vosoritide will be administered as a once-daily subcutaneous injection.
[c]Following the 52-week treatment period, participants will undergo a 2-week safety follow-up visit, except for participants entering the CANOPY HCH-EXT study within 2 weeks of their last dose.
[d]Definition of near-final adult height.

Conclusions

- The CANOPY HCH-2I results will complement data from children with hypochondroplasia aged ≥3 years receiving vosoritide in ongoing phase 1/2 and phase 3 CANOPY HCH-3 (111-303, NCT06455059) trials
- Initiating vosoritide treatment in infancy or early childhood may provide clinical benefit and better outcomes for children with hypochondroplasia

References

1. Dauber A, et al. *EClinicalMedicine*. 2024;71:102591. **2.** Savarirayan R, et al. *Lancet Child Adolesc Health*. 2024;8(4):301–10. **3.** Jones HL, et al. *Cureus*. 2025;17(7):e87983. **4.** Cheung MS, et al. *Am J Med Genet A*. 2024;194(2):243–52.

Acknowledgments

We thank study participants, their families, site staff, and investigators. Funding for this study was provided by BioMarin Pharmaceutical Inc. Medical writing support was provided by Sanna Abbasi, PhD, of Red Nucleus, and funded by BioMarin Pharmaceutical Inc. Project management support was provided by Justin Potuzak, PhD, of BioMarin Pharmaceutical Inc.

Disclosures

AD has received consulting fees and grants from BioMarin Pharmaceutical Inc., BridgeBio, Novo Nordisk, and Pfizer. **MC** is an investigator on clinical trials for BioMarin Pharmaceutical Inc., Kyowa Kirin, and Tyra Biosciences and has received honoraria for speaking and participating on advisory boards from BioMarin Pharmaceutical Inc., BridgeBio, Kyowa Kirin, and Tyra Biosciences. **JH-F** has received grants or contracts for clinical trials from BioMarin Pharmaceutical Inc., BridgeBio, and Pfizer; has received consulting fees from BioMarin Pharmaceutical Inc., BridgeBio, Innoskel, and Novo Nordisk; has received honoraria from BioMarin Pharmaceutical Inc. and Medscape; and has participated on advisory boards with BioMarin Pharmaceutical Inc., BridgeBio, and MCDS-Therapy Clinical Trial (unpaid). **NN** has received honoraria for lectures, presentations, speakers bureaus, manuscript writing, and/or educational events from Kyowa Kirin. **OS** has no conflicts of interest to declare. **RB** and **DO** are employees and shareholders of BioMarin Pharmaceutical Inc. **SG** is an employee of BioMarin (UK) Ltd. **RS** has received consulting fees and grants from BioMarin Pharmaceutical Inc. and was an investigator in clinical trials for achondroplasia for Ascendis Pharma A/S, BioMarin Pharmaceutical Inc., and QED Therapeutics.

To view a copy of this poster, scan this QR code.

Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission from the authors.

Presented at the Pediatric Endocrine Society (PES) annual meeting; April 30–May 3, 2026; San Francisco, CA, USA

©2026 BioMarin Pharmaceutical Inc. All rights reserved.