Effect of long-term vosoritide treatment in pediatric participants with achondroplasia on bone mineral density and bone mineral content measured with dual X-ray absorptiometry

Poster 16

Jeanette White,[1] Eric Roberts[1]

[1]BioMarin Pharmaceutical Inc., Novato, CA, USA

Introduction

- Achondroplasia is a skeletal dysplasia caused by impaired endochondral bone growth and associated with decreased measured bone mineral density (BMD)[1–3]
- Vosoritide, a targeted recombinant C-type natriuretic peptide analog approved for the treatment of children with achondroplasia, provides sustained improvements in linear growth and associated health outcomes[1]
 - Clinical trial data showed that long-term vosoritide treatment in children with achondroplasia was associated with increases in bone size and BMD as measured with peripheral quantitative computed tomography[4]
- In the ongoing phase 3, open-label extension study CANOPY ACH-EXT (111-302, NCT03424018), bone growth parameters were measured in children who received vosoritide long-term for up to 7.8 years, beginning at ≥5 years of age[1]

Objective

- To assess long-term changes in bone mineral content (BMC) and BMD in children who received vosoritide long-term as assessed by dual X-ray absorptiometry (DXA) adjusted for age and sex but not height in this chondrodysplastic population

Methods

Study design

- CANOPY ACH-EXT (111-302) is a phase 3, open-label, long-term extension of the completed randomized, placebo-controlled trial CANOPY ACH-3 (111-301, NCT03197766) that aims to evaluate the safety and efficacy of vosoritide in children with achondroplasia who initiated treatment >5 years of age
- Results for participants with up to 6 years of treatment are presented here. The data cutoff was February 25, 2025

DXA scans

- Scans were acquired every year using the Lunar Prodigy (GE HealthCare) or Discovery Horizon (Hologic) system and were performed for whole body less head (WBLH) and lumbar spine
- Participants with scans available for both baseline and yearly time points from the CANOPY ACH-EXT trial were included, except those who switched scanners during the study

BMC and BMD Z-score calculations

- Changes in BMC (g) and in BMD Z-score were recorded longitudinally
- BMD Z-scores were derived using age- and sex-specific reference data for average-stature children
- While the practice of adjustment of BMD and BMC for height Z-score is common,[5] available coefficients are based solely on children without skeletal dysplasias and were not intended for generalization to this population. Therefore, Z-scores are only adjusted for age and sex

Height Z-score

- Age- and sex-adjusted height Z-scores were calculated using both an average-stature reference (Centers for Disease Control and Prevention) and an achondroplasia natural history reference (CLARITY)[6]

Results

Participants

- CANOPY ACH-EXT included 119 participants with a mean ± standard deviation treatment duration of 5.2 ± 1.4 years
- Of the 119 participants (**Table 1**), 105 used the same scanner throughout the study, and their DXA data are presented here

Table 1. Baseline demographics and characteristics

Characteristic	CANOPY ACH-EXT participants N = 119
Age at day 1, years, mean ± SD	9.18 ± 2.60
Sex, n (%)	
Male	63 (52.9)
Female	56 (47.1)
Race, n (%)	
White	85 (71.4)
Asian	21 (17.6)
Other	15 (12.6)
Japanese	6 (5.0)
Multiple	8 (6.7)
Black or African American	5 (4.2)
Ethnicity, n (%)	
Not Hispanic or Latino	111 (93.3)
Hispanic or Latino	8 (6.7)
Tanner stage, n (%)	
I	89 (74.8)
>I	29 (24.4)
Not done	1 (0.8)
Weight, kg, mean ± SD	25.19 ± 9.32
Weight Z-score, mean ± SD[a]	−1.54 ± 1.35
BMI, kg/m², mean ± SD	22.82 (4.84)
BMI Z-score, mean ± SD[a]	1.75 (0.60)
AGV, cm/year, mean ± SD[b]	4.01 (1.43)
Height Z-score (average-stature reference), mean ± SD[a]	−5.12 (1.09)
Height Z-score (achondroplasia reference), mean ± SD[c]	0.28 (0.99)
Standing height, cm, mean ± SD	103.57 (11.91)
Sitting height, cm, mean ± SD	68.51 (6.57)
Upper-to-lower body segment ratio, mean ± SD	1.98 (0.19)

Day 1 is the day of the first dose of vosoritide in either CANOPY (111-301) or CANOPY ACH-EXT (111-302).
[a]Derived using age- and sex-specific reference data (means and SDs) for average-stature children per the US Centers for Disease Control and Prevention.
[b]AGV at baseline is defined as [(height at baseline − last height measurement at least 6 months prior to day 1)/(date of baseline assessment − date of last height assessment at least 6 months prior to day 1)] x 365.25.
[c]Derived using achondroplasia age- and sex-specific reference data (means and SDs) from CLARITY.[6]
AGV, annualized growth velocity; BMI, body mass index; SD, standard deviation.

BMC

- WBLH BMC values increased over time for children with achondroplasia receiving vosoritide (**Figure 1**)
- The change from baseline in WBLH BMC consistently trended upward over time (**Figure 2A**)
- A similar upward trend over time was also observed for the change from baseline in lumbar spine BMC (**Figure 2B**)
- Trends from both DXA scanners were similar

Figure 1. Absolute BMC over time for WBLH

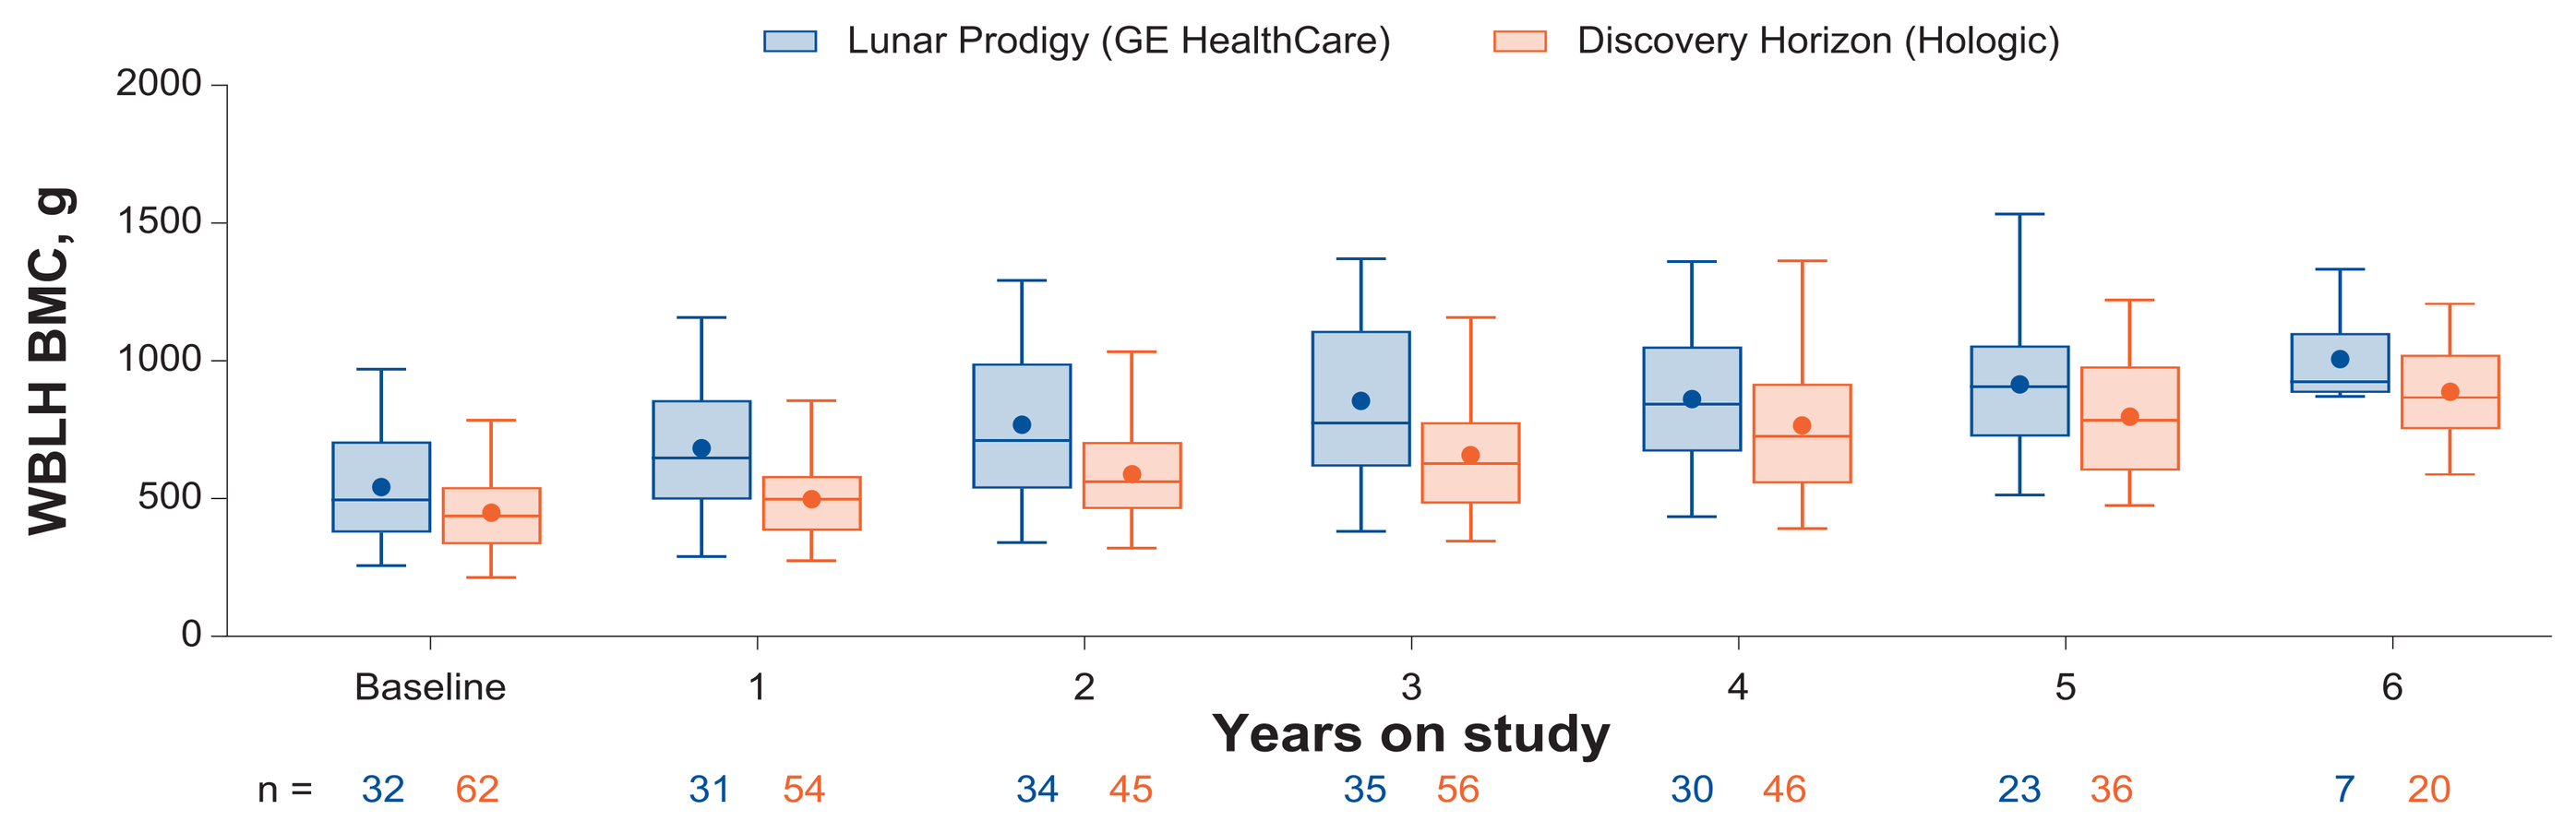
Lunar Prodigy (GE HealthCare)
Discovery Horizon (Hologic)
WBLH BMC, g
2000
1500
1000
500
0
Baseline
1
2
3
4
5
6
Years on study
n = 32 62
31 54
34 45
35 56
30 46
23 36
7 20

Data from participants who switched DXA machines are not included.
BMC, bone mineral content; DXA, dual X-ray absorptiometry; WBLH, whole body less head.

Figure 2. Change from baseline in BMC over time

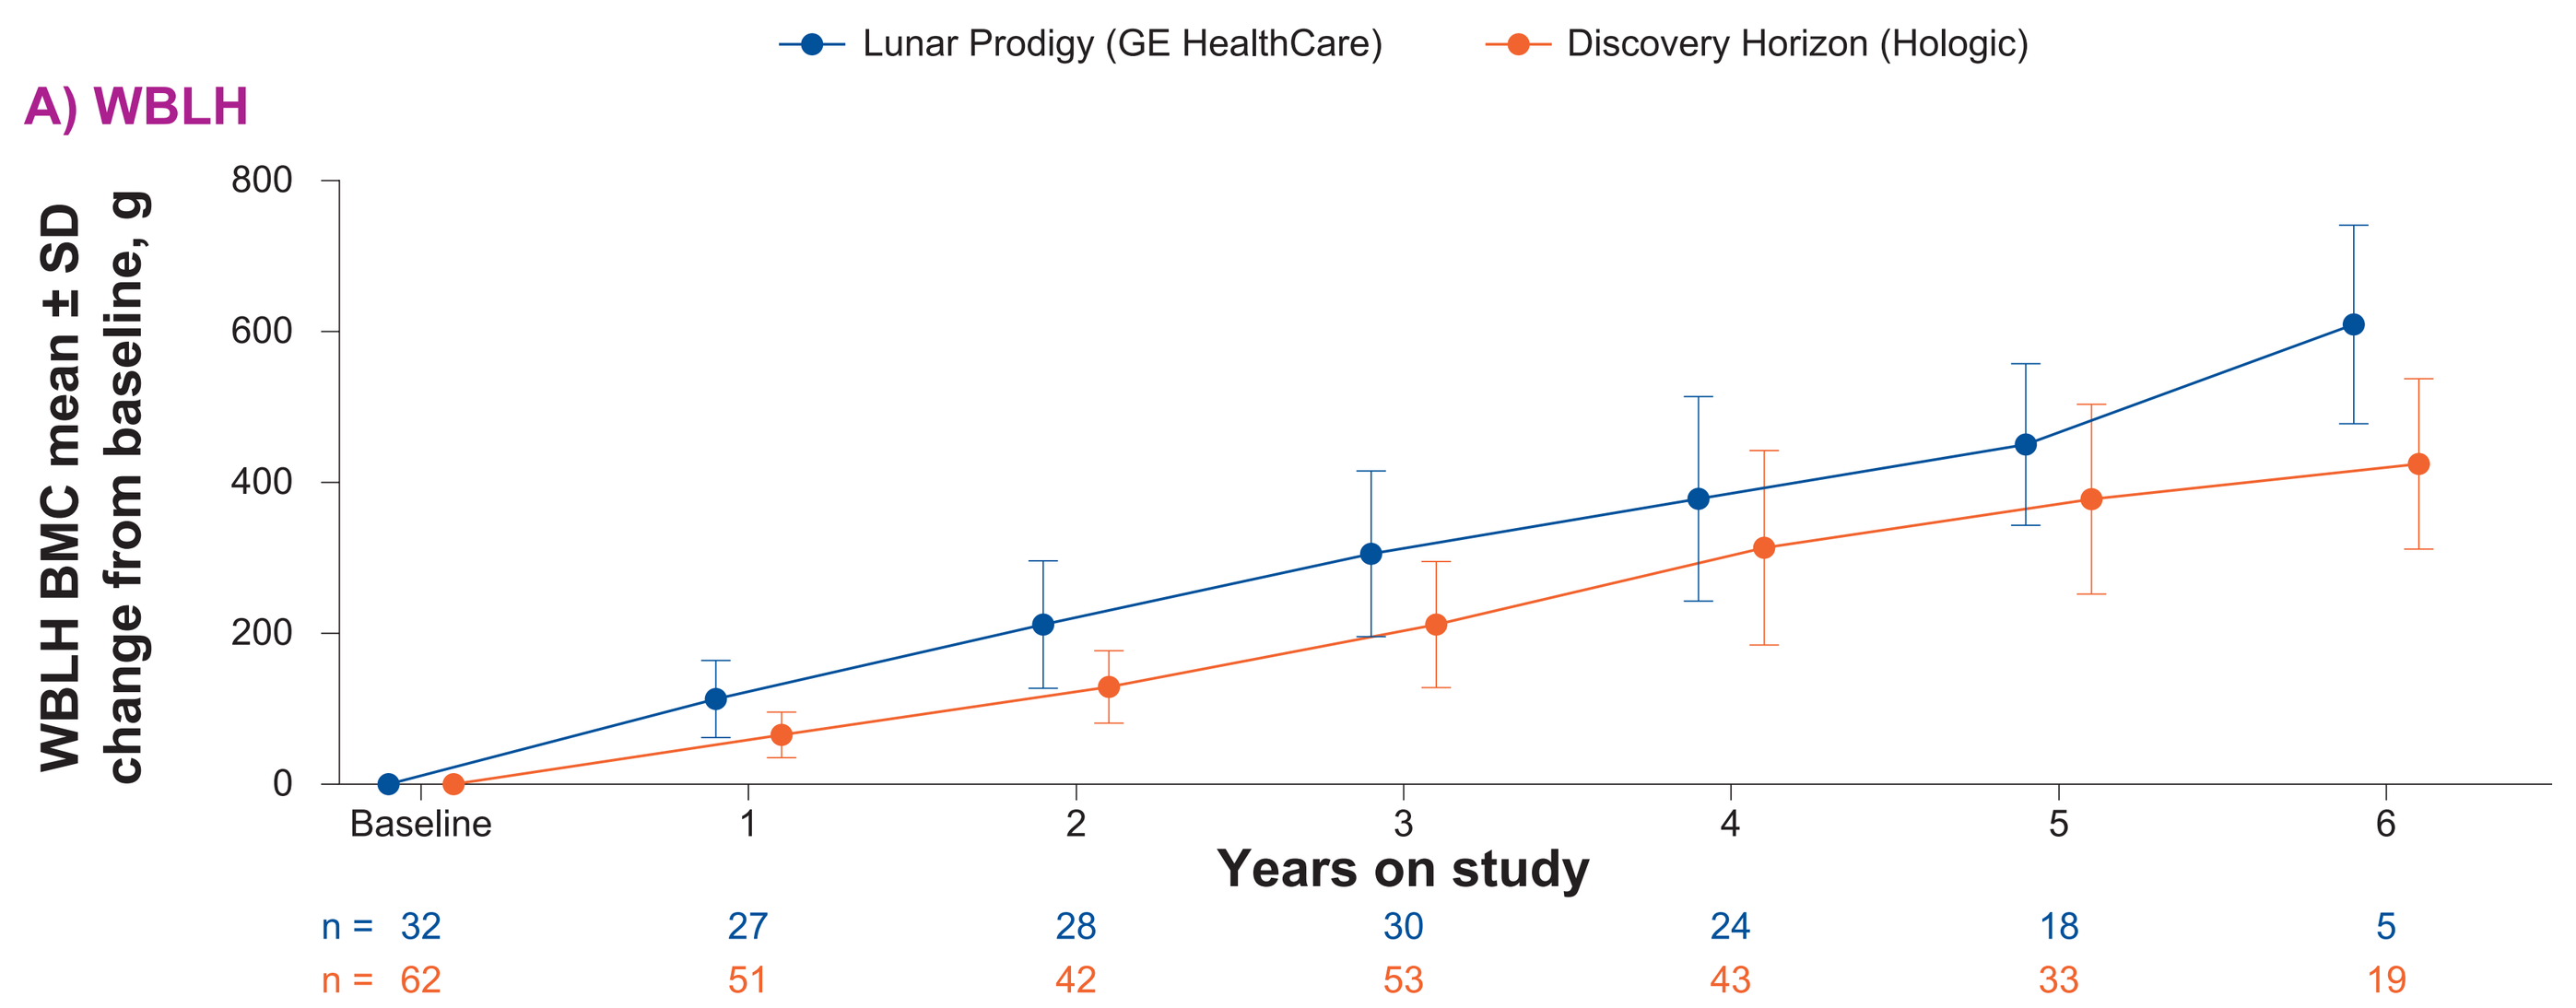
Lunar Prodigy (GE HealthCare)
Discovery Horizon (Hologic)
A) WBLH
WBLH BMC mean ± SD change from baseline, g
800
600
400
200
0
Baseline
1
2
3
4
5
6
Years on study
n = 32 27 28 30 24 18 5
n = 62 51 42 53 43 33 19

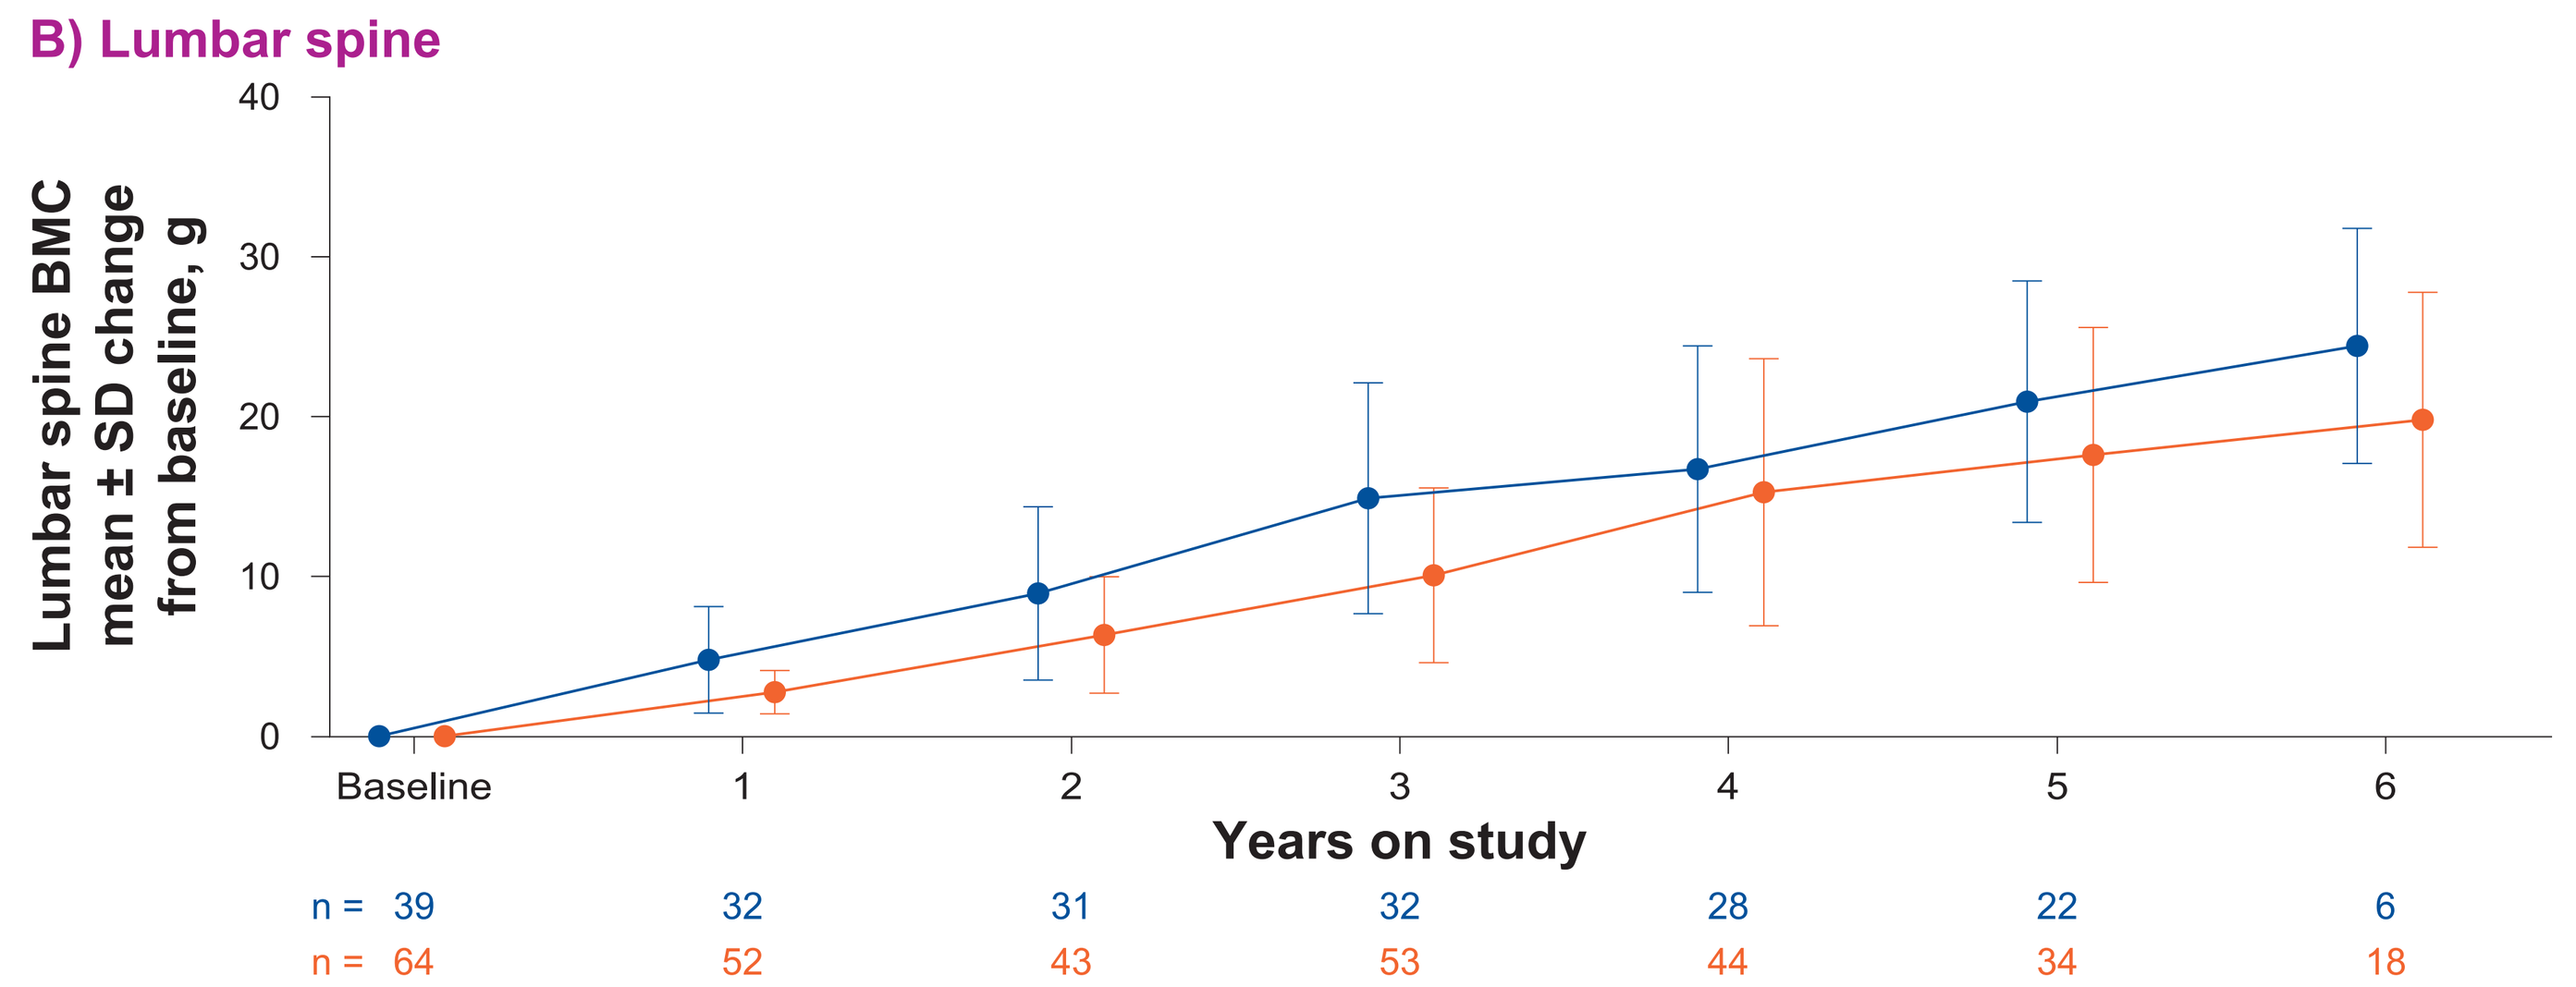
B) Lumbar spine
Lumbar spine BMC mean ± SD change from baseline, g
40
30
20
10
0
Baseline
1
2
3
4
5
6
Years on study
n = 39 32 31 32 28 22 6
n = 64 52 43 53 44 34 18

Data from participants who switched DXA machines are not included.
BMC, bone mineral content; DXA, dual X-ray absorptiometry; SD, standard deviation; WBLH, whole body less head.

BMD Z-score

- WBLH BMD Z-score remained relatively stable over time in children with achondroplasia receiving vosoritide. The readings from the Discovery Horizon (Hologic) system were consistently lower than those of Lunar Prodigy (GE HealthCare) system (**Figure 3**)
- The changes from baseline in BMD Z-scores at each year were consistent over time for WBLH (**Figure 4A**) and lumbar spine (**Figure 4B**)

Figure 3. Absolute BMD Z-score over time for WBLH

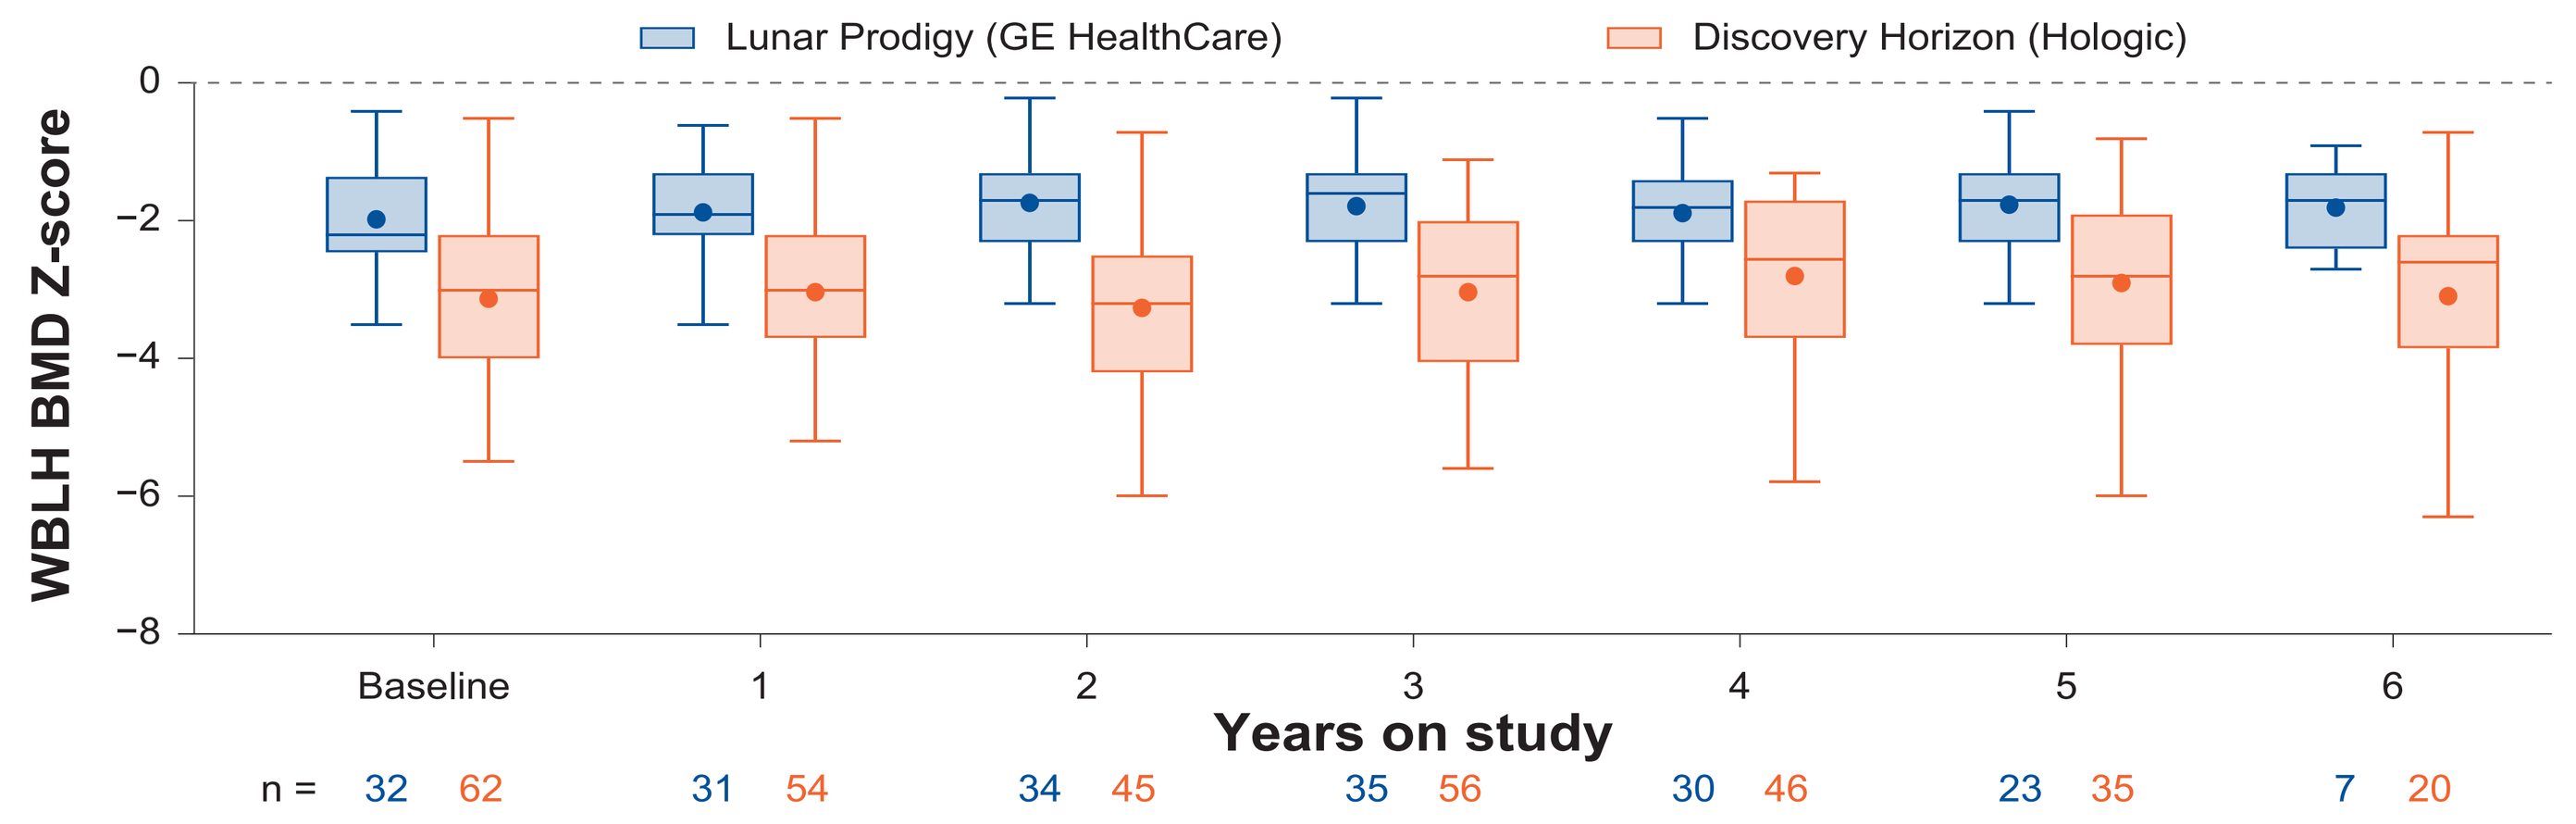
Lunar Prodigy (GE HealthCare)
Discovery Horizon (Hologic)
WBLH BMD Z-score
0
−2
−4
−6
−8
Baseline
1
2
3
4
5
6
Years on study
n = 32 62
31 54
34 45
35 56
30 46
23 35
7 20

Data from participants who switched DXA machines are not included.
BMC, bone mineral content; DXA, dual X-ray absorptiometry; WBLH, whole body less head.

Figure 4. Change from baseline in BMD Z-score over time

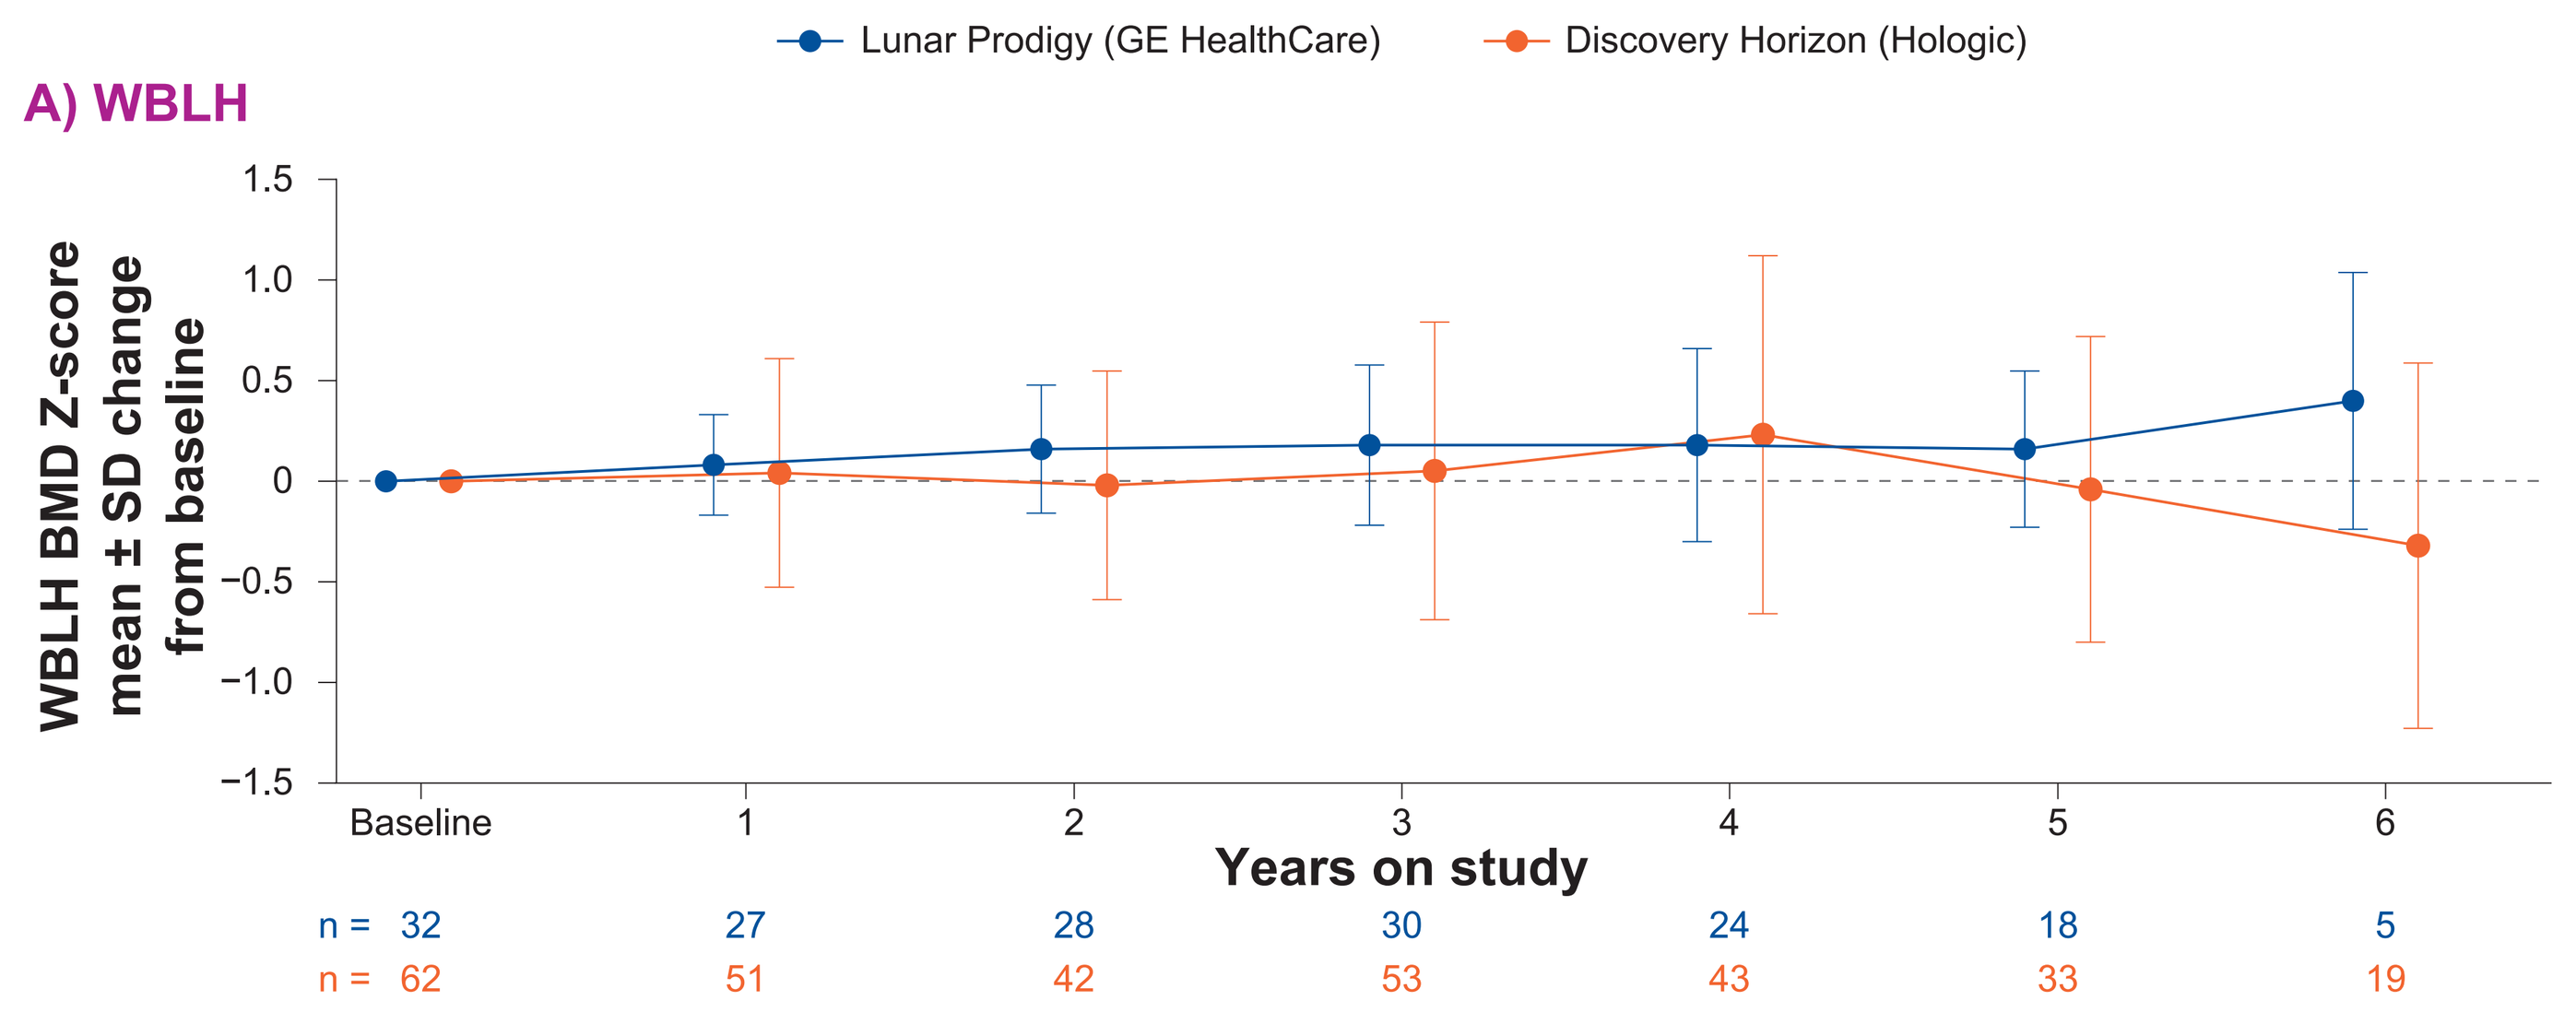
Lunar Prodigy (GE HealthCare)
Discovery Horizon (Hologic)
A) WBLH
WBLH BMD Z-score mean ± SD change from baseline
1.5
1.0
0.5
0
−0.5
−1.0
−1.5
Baseline
1
2
3
4
5
6
Years on study
n = 32 27 28 30 24 18 5
n = 62 51 42 53 43 33 19

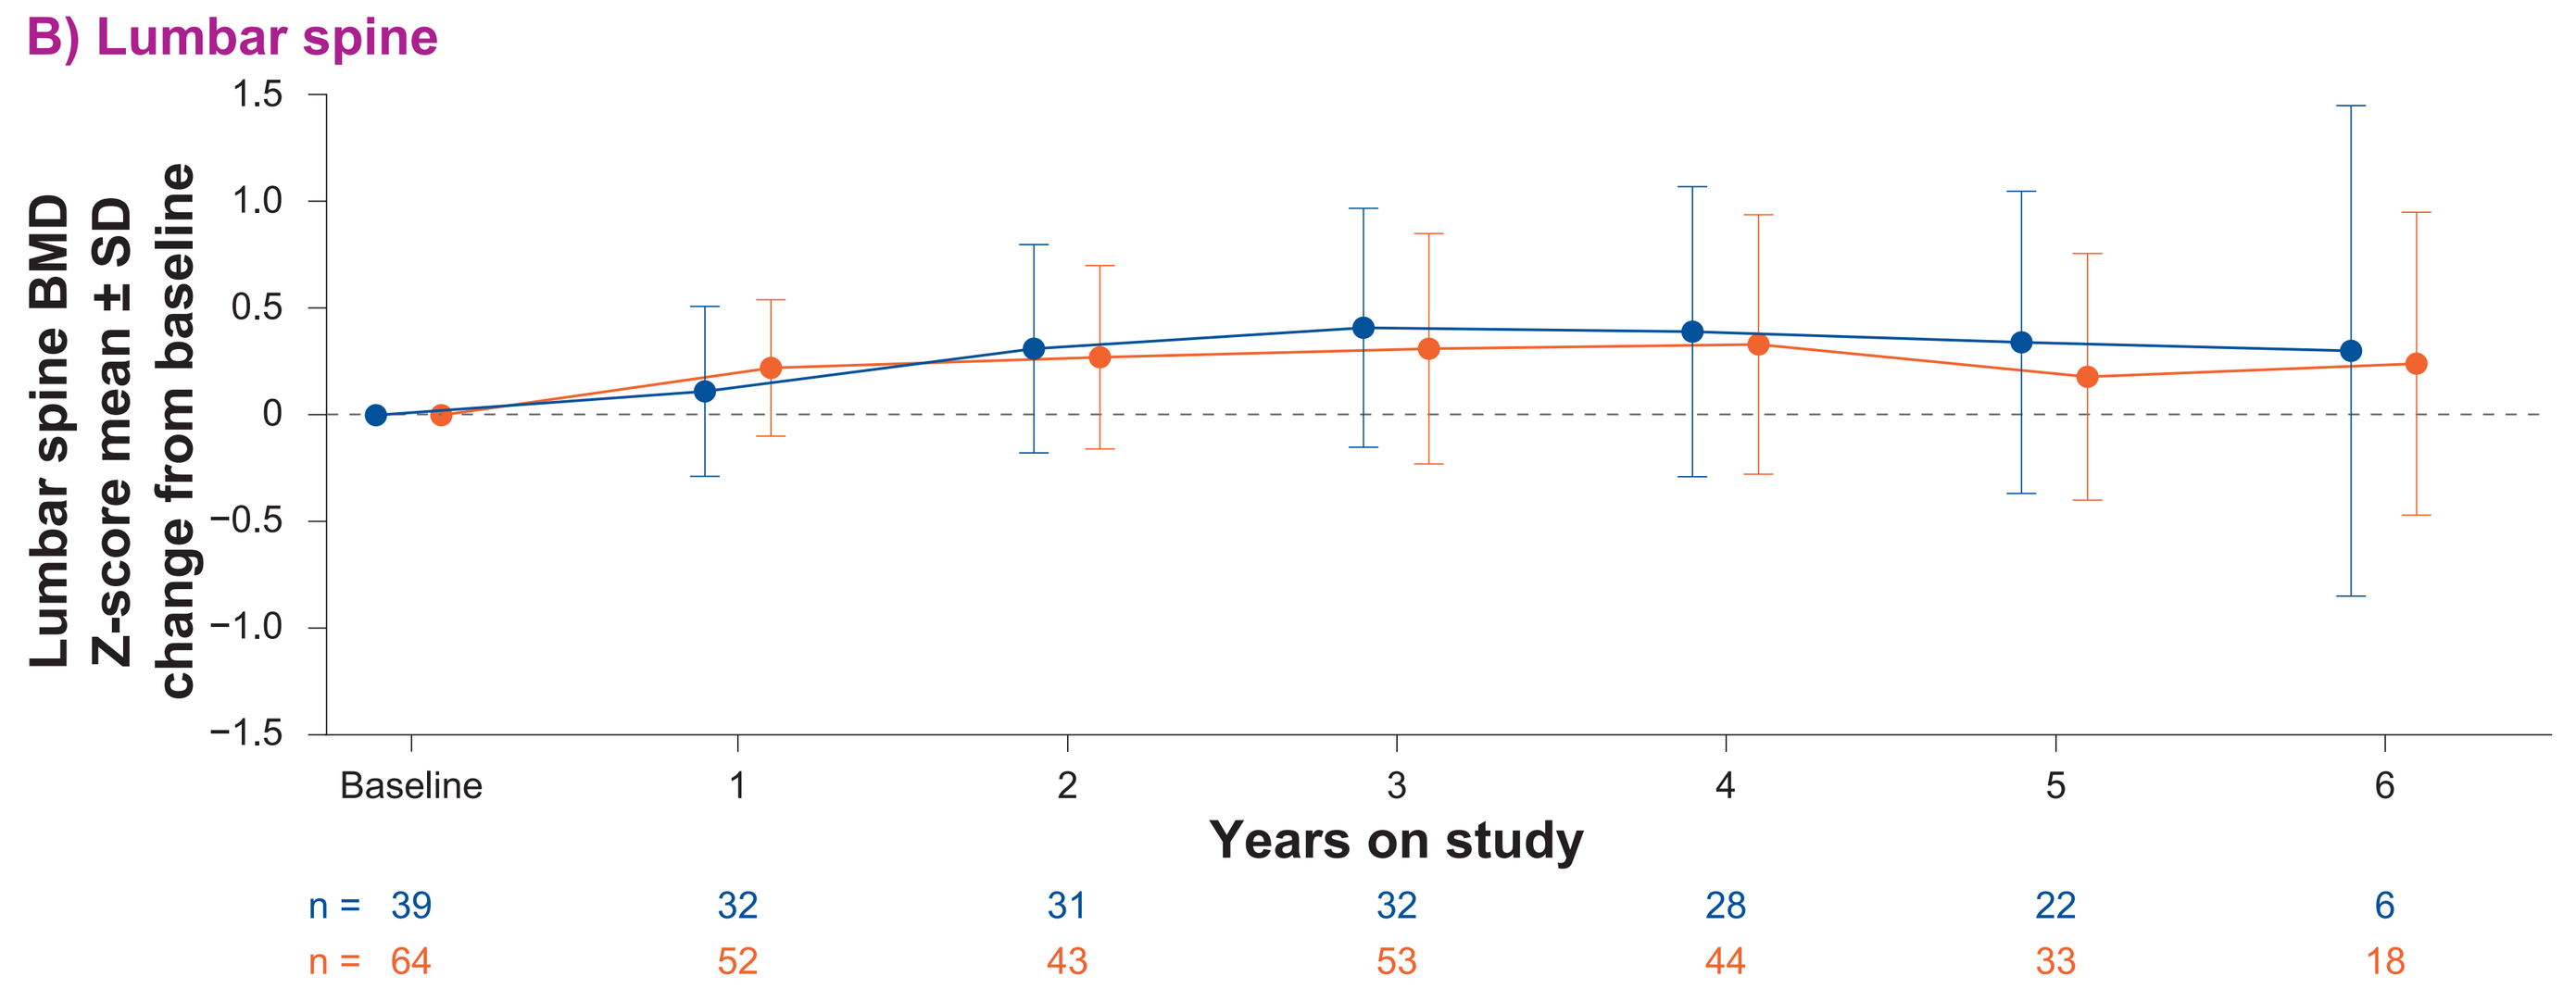
B) Lumbar spine
Lumbar spine BMD Z-score mean ± SD change from baseline
1.5
1.0
0.5
0
−0.5
−1.0
−1.5
Baseline
1
2
3
4
5
6
Years on study
n = 39 32 31 32 28 22 6
n = 64 52 43 53 44 33 18

Z-scores were derived using age- and sex-specific reference data for average-stature children per scanner-specific reference data. Data from participants who switched DXA machines are not included.
BMD, bone mineral density; DXA, dual X-ray absorptiometry; SD, standard deviation; WBLH, whole body less head.

Height Z-score

- At the same time, height Z-scores (both average stature- and achondroplasia-referenced) increased from baseline over time for CANOPY ACH-EXT participants (**Figure 5**)

Figure 5. Change from baseline in height Z-score for CANOPY ACH-EXT participants

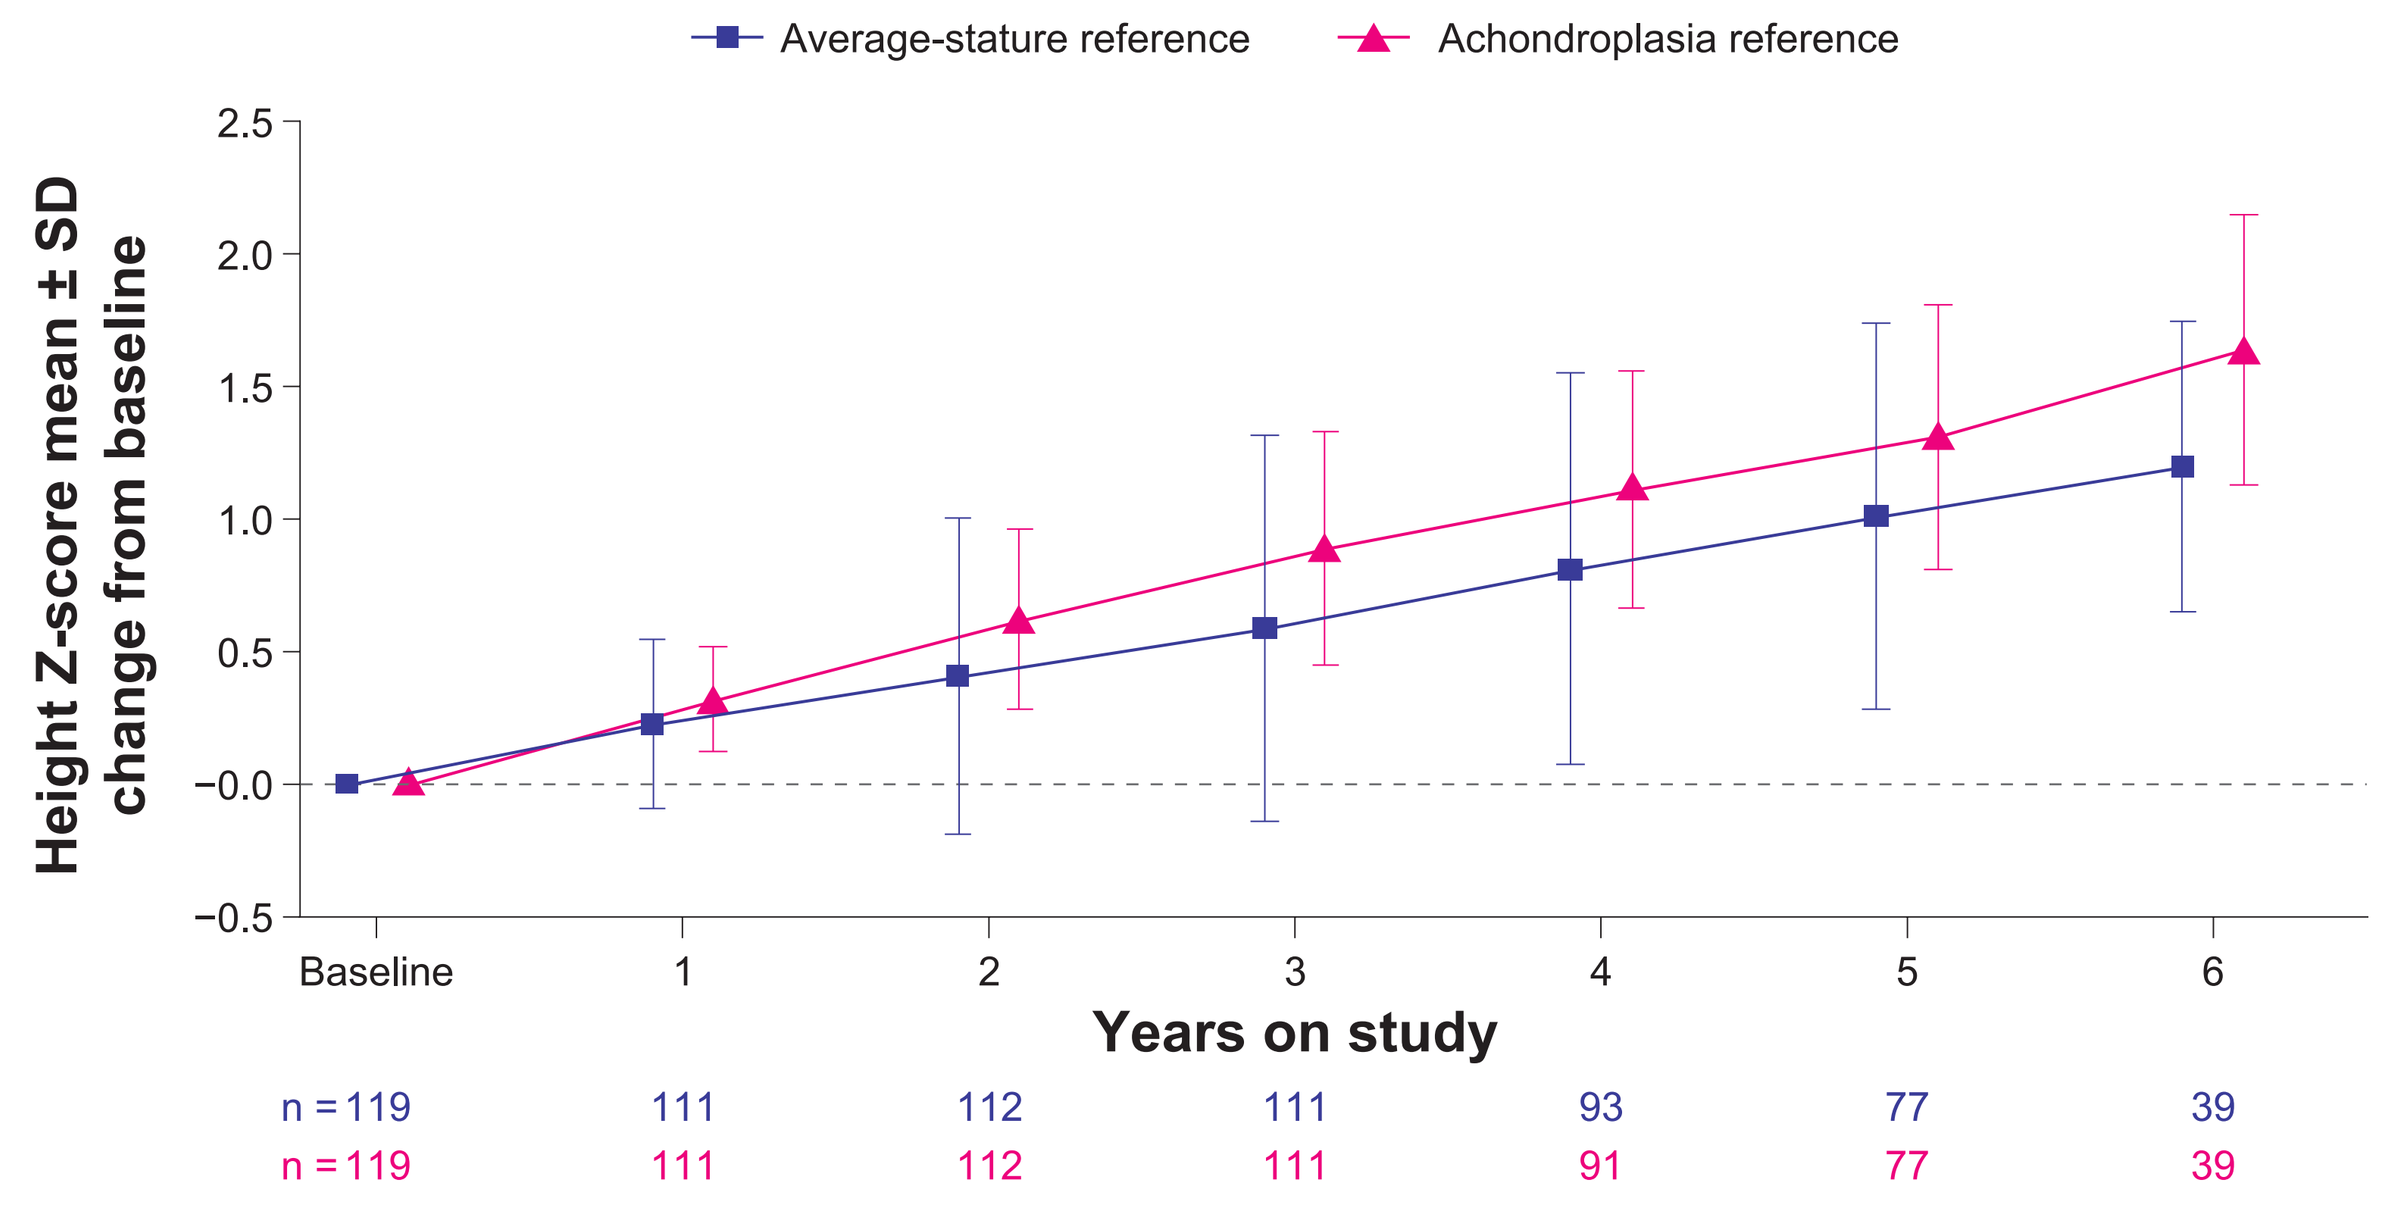
Average-stature reference
Achondroplasia reference
Height Z-score mean ± SD change from baseline
2.5
2.0
1.5
1.0
0.5
−0.0
−0.5
Baseline
1
2
3
4
5
6
Years on study
n = 119 111 112 111 93 77 39
n = 119 111 112 111 91 77 39

Height Z-scores (average-stature reference) were derived using age- and sex-specific reference data (means and SDs) for average-stature children per the CDC. Height Z-scores (achondroplasia reference) were derived using achondroplasia age- and sex-specific reference data (means and SDs) from CLARITY.[6]
CDC, US Centers for Disease Control and Prevention; SD, standard deviation.

Conclusions

- As measured with DXA, BMC values in children with achondroplasia receiving vosoritide increased over time for both WBLH and lumbar spine scans, while BMD Z-scores remained relatively stable. At the same time, height Z-scores increased in vosoritide-treated children with achondroplasia over time compared to baseline
- These longitudinal data from the CANOPY ACH-EXT trial suggest that bone health is maintained with long-term vosoritide treatment up to 6 years in children with achondroplasia with the improvement of growth

References

1. Savarirayan R, et al. *Med.* 2024;6(5):100566. **2.** Bellus GA, et al. *Am J Hum Genet.* 1995;56:368–73. **3.** Matsushita M, et al. *Pediatr Int.* 2016;58:705–8. **4.** Engelke K, et al. Oral presentation at the Pre-meeting Symposium for the ASBMR 2025 Annual Meeting; September 5-8. PM-089. **5.** Zemel BS, et al. *J Clin Endocrinol Metab.* 2025;110(11):3034–48. **6.** Hoover-Fong JE, et al. *Genet Med.* 2021;23(8):1498–1505.

Acknowledgments

We thank all trial participants and their families. Funding for this study was provided by BioMarin Pharmaceutical Inc. Medical writing support was provided by Tony Sallese, PhD, of Red Nucleus, and funded by BioMarin Pharmaceutical Inc. Project management support was provided by Pooja Dhupkar, PhD, of BioMarin Pharmaceutical Inc.

Disclosures

JW and **ER** are employees and shareholders of BioMarin Pharmaceutical Inc.

To view a copy of this poster, scan this QR code.

Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission from the authors.

Presented at the Pediatric Endocrine Society (PES) annual meeting; April 30–May 3, 2026; San Francisco, CA, USA

©2026 BioMarin Pharmaceutical Inc. All rights reserved.