SUN-212

BMN 333 Achieves High Sustained Released Vosoritide Exposure with Favorable Safety: Phase 1 Results that Support Phase 2/3 Trials in Achondroplasia

Nina Mitchell[1], Yulan Qi[2], Lynn Han[2], Daniel Wendt[2], Heather Wenzel[2], Russell Soon[2], Huiyu Zhou[2]

[1]BioMarin (UK) Ltd, London, UK; [2]BioMarin Pharmaceutical Inc., Novato, CA, USA.

Introduction

- Achondroplasia is a rare skeletal dysplasia caused by excess fibroblast growth factor 3 (FGFR3) signaling, causing diminished endochondral bone ossification that hinders early growth and often results in multisystem complications[1,2]
- Currently approved therapies include vosoritide, a recombinant C-type natriuretic peptide (CNP) analog that counteracts excess FGFR3 signaling to promote endochondral bone growth[2,3]
- Clinical and real-world data have demonstrated that vosoritide is well tolerated and provides sustained improvements in growth and proportionality with once-daily administration[4-6]
 - Vosoritide is currently the only approved therapy for achondroplasia with long-term clinical and real-world data
- There is potentially further opportunity to improve efficacy through greater vosoritide exposure while maintaining a robust safety profile
- BMN 333 is an investigational drug in early clinical development that is fully biodegradable and designed to time-dependently release vosoritide, its active moiety, to extend exposure (**Figure 1**)

Figure 1. BMN 333 structure and preclinical results

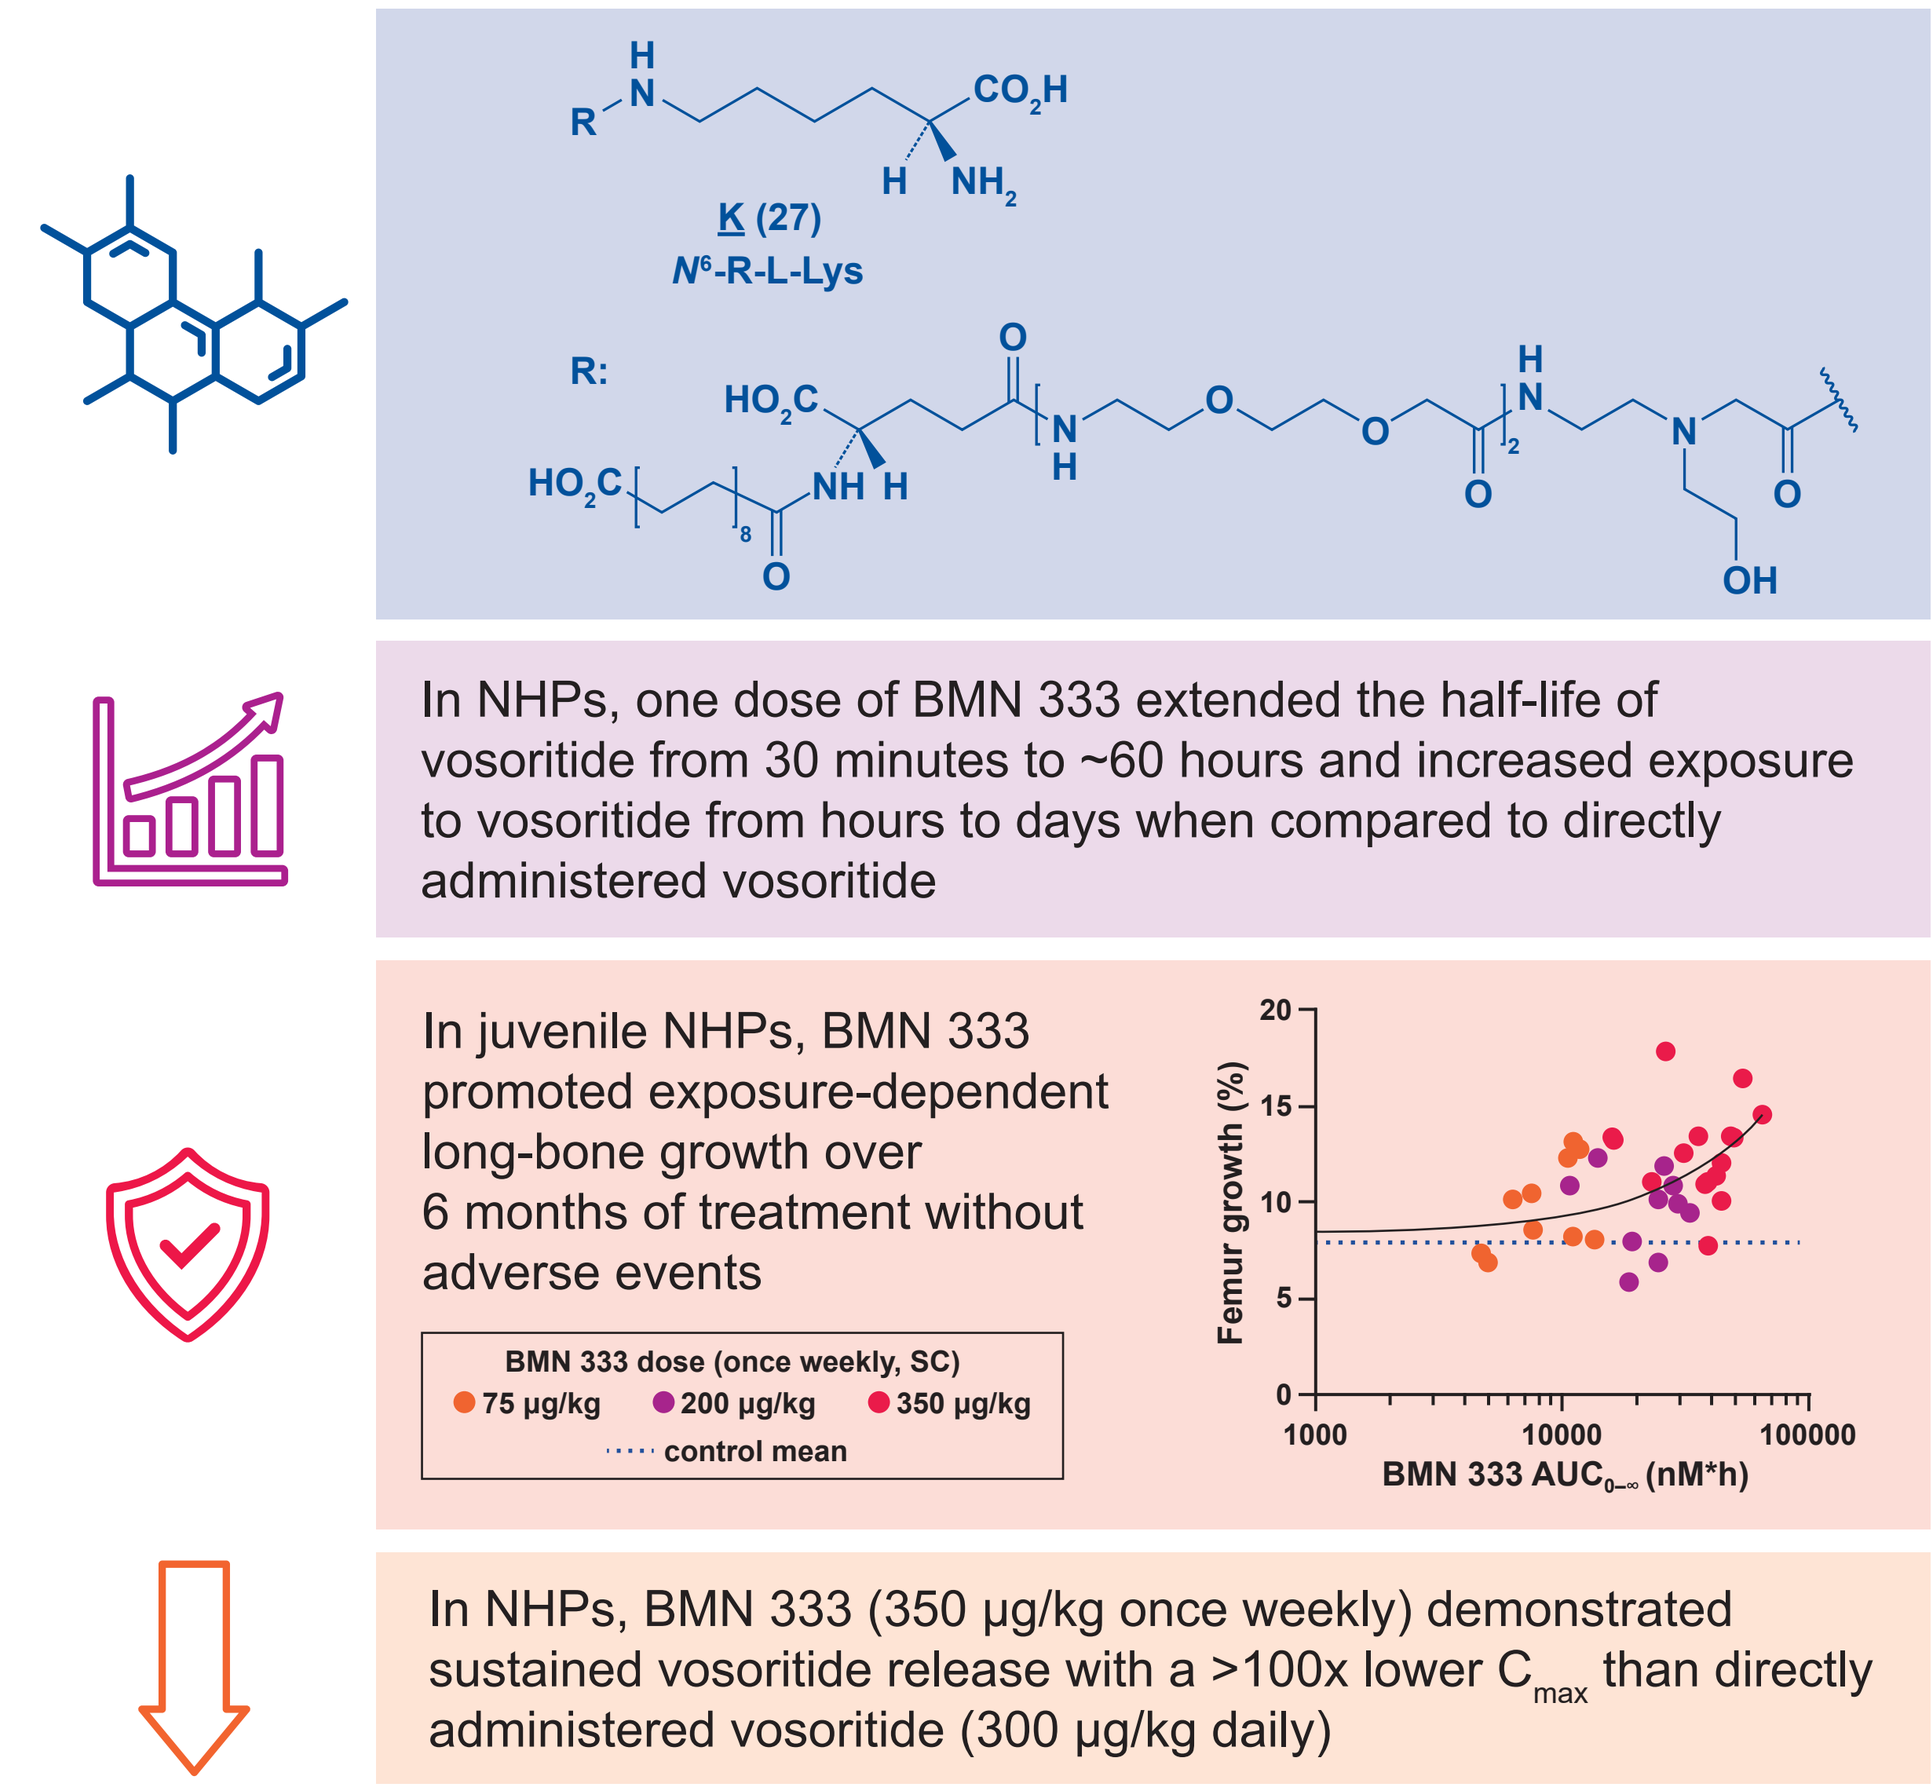
K (27)
N^6-R-L-Lys
R:
In NHPs, one dose of BMN 333 extended the half-life of vosoritide from 30 minutes to ~60 hours and increased exposure to vosoritide from hours to days when compared to directly administered vosoritide
In juvenile NHPs, BMN 333 promoted exposure-dependent long-bone growth over 6 months of treatment without adverse events
BMN 333 dose (once weekly, SC)
75 µg/kg 200 µg/kg 350 µg/kg
control mean
Femur growth (%)
BMN 333 $AUC_{0-\infty}$ (nM*h)
In NHPs, BMN 333 (350 µg/kg once weekly) demonstrated sustained vosoritide release with a >100x lower C_{max} than directly administered vosoritide (300 µg/kg daily)

$AUC_{0-\infty}$, area under the plasma concentration-time curve from time 0 to infinity; C_{max}, maximum observed plasma concentration; NHP, non-human primate; SC, subcutaneous.

- Here, we present safety, pharmacokinetic (PK), and pharmacodynamic (PD) results from the first-in-human phase 1 study of BMN 333 (study 333-101)

Objective

- To evaluate the ability of BMN 333 to safely extend exposure to vosoritide in healthy volunteers

Study Design

- This phase 1, randomized, double-blind, placebo-controlled, single ascending dose study enrolled healthy adults to receive a single subcutaneous dose of 15, 50, 100, 200, 350, or 500 µg/kg BMN 333 or placebo (7:3 ratio)
- Vital signs, adverse events (AEs), PK parameters, and PD parameters (via cyclic guanosine monophosphate [cGMP]) were collected
 - PK and safety follow-up data were collected at baseline, immediately following injection, and on days 2, 4, 6, 8, 15, 22, and 29 post-injection
 - PD follow-up data were collected at baseline, 12 hours after injection, and on days 2, 3, 4, 5, and 8 post-injection

Results

Participants

- A total of 60 participants enrolled and received BMN 333 (n = 42) or placebo (n = 18; **Table 1**)

Table 1. Participant demographics and baseline characteristics

	Overall BMN 333 (n = 42)	Pooled placebo (n = 18)
Age (years), mean ± SD	41.6 ± 11.5	37.8 ± 10.6
Sex, male	21 (50.0)	11 (61.1)
Race		
Black or African American	20 (47.6)	8 (44.4)
White	20 (47.6)	7 (38.9)
Asian	1 (2.4)	1 (5.6)
Unknown	1 (2.4)	0
Multiple	0	2 (11.1)
Weight (kg), mean ± SD	77.60 ± 11.56	80.38 ± 11.94

Data shown as n (%) unless otherwise specified.
SD, standard deviation.

Safety

- BMN 333 was well tolerated, with no treatment-related serious AEs, dose-limiting toxicities, changes to laboratory evaluations, or electrocardiogram parameters attributed to BMN 333
- The incidence of AEs was similar between BMN 333 (15/42 participants; 35.7%) and placebo (6/18 participants; 33.3%), and all were mild or moderate in severity with the exception of 1 grade 3 AE in both the placebo and 500-µg/kg dose group; neither event was considered related to study drug (**Figure 2**)

Figure 2. Incidence of adverse events following administration of BMN 333 or placebo

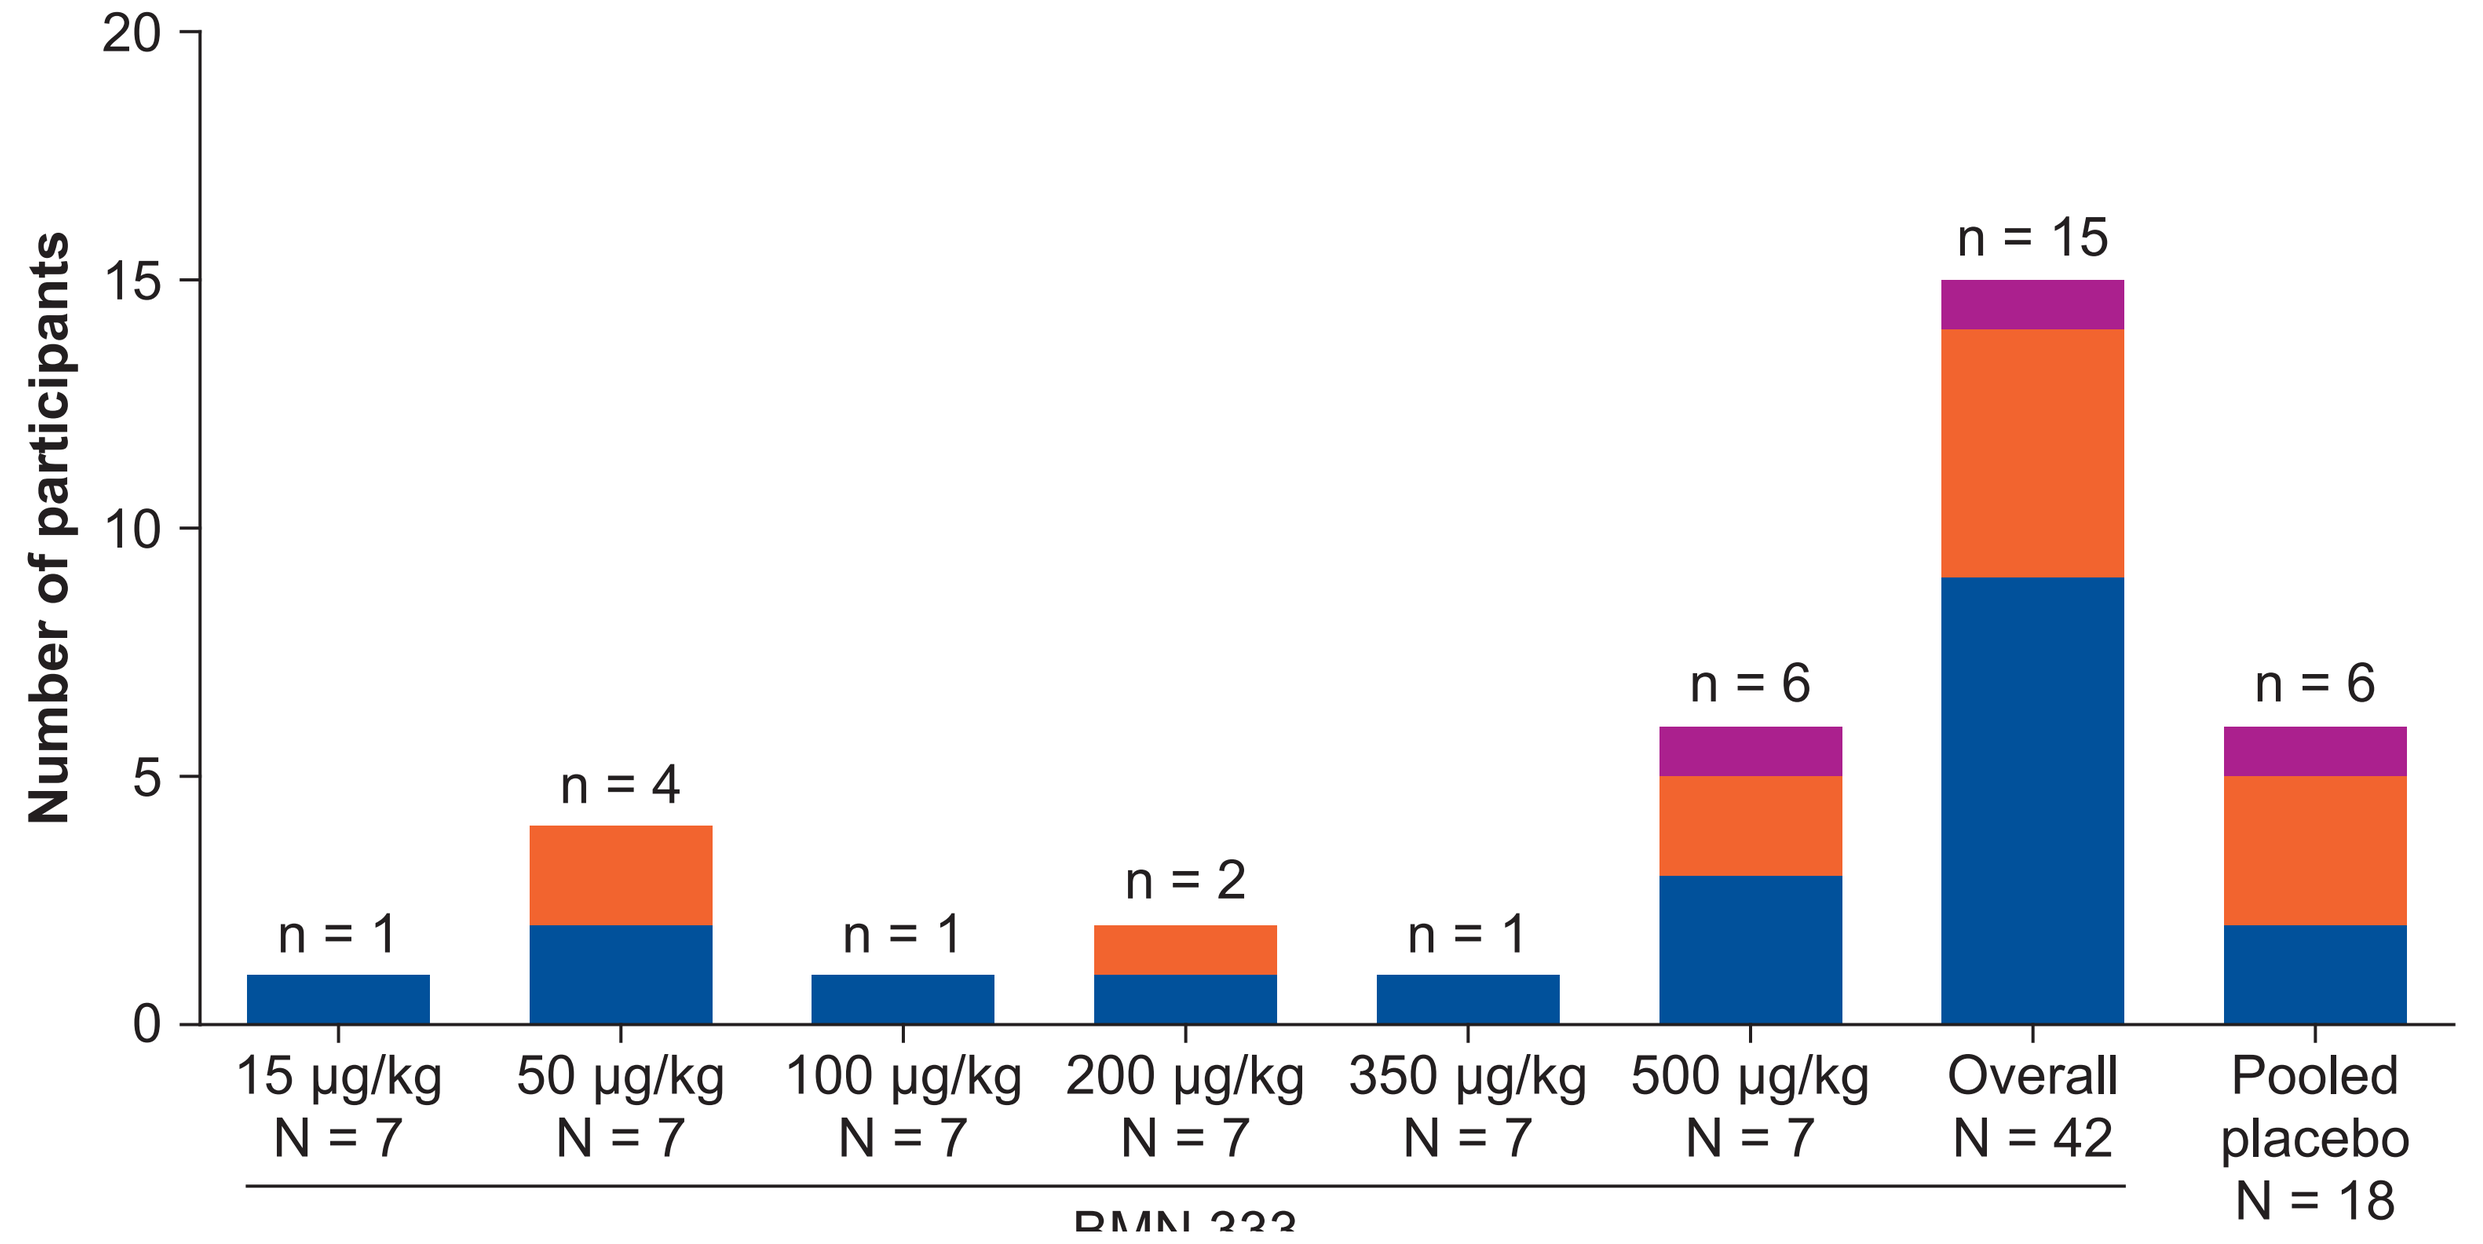
Number of participants
n = 1
n = 4
n = 1
n = 2
n = 1
n = 6
n = 15
n = 6
15 µg/kg N = 7
50 µg/kg N = 7
100 µg/kg N = 7
200 µg/kg N = 7
350 µg/kg N = 7
500 µg/kg N = 7
Overall N = 42
Pooled placebo N = 18
BMN 333

Grade 1, mild; grade 2, moderate; grade 3, severe.

- BMN 333 was not associated with hemodynamic effects, with heart rate and blood pressure being comparable to placebo and generally consistent over time (**Figure 3**) and across doses (data not shown)
 - BMN 333 did not have a postural effect, as lying heart rate and blood pressure were not impacted (data not shown)

Figure 3. Effect of BMN 333 and placebo on A) standing heart rate and B) diastolic and systolic blood pressure

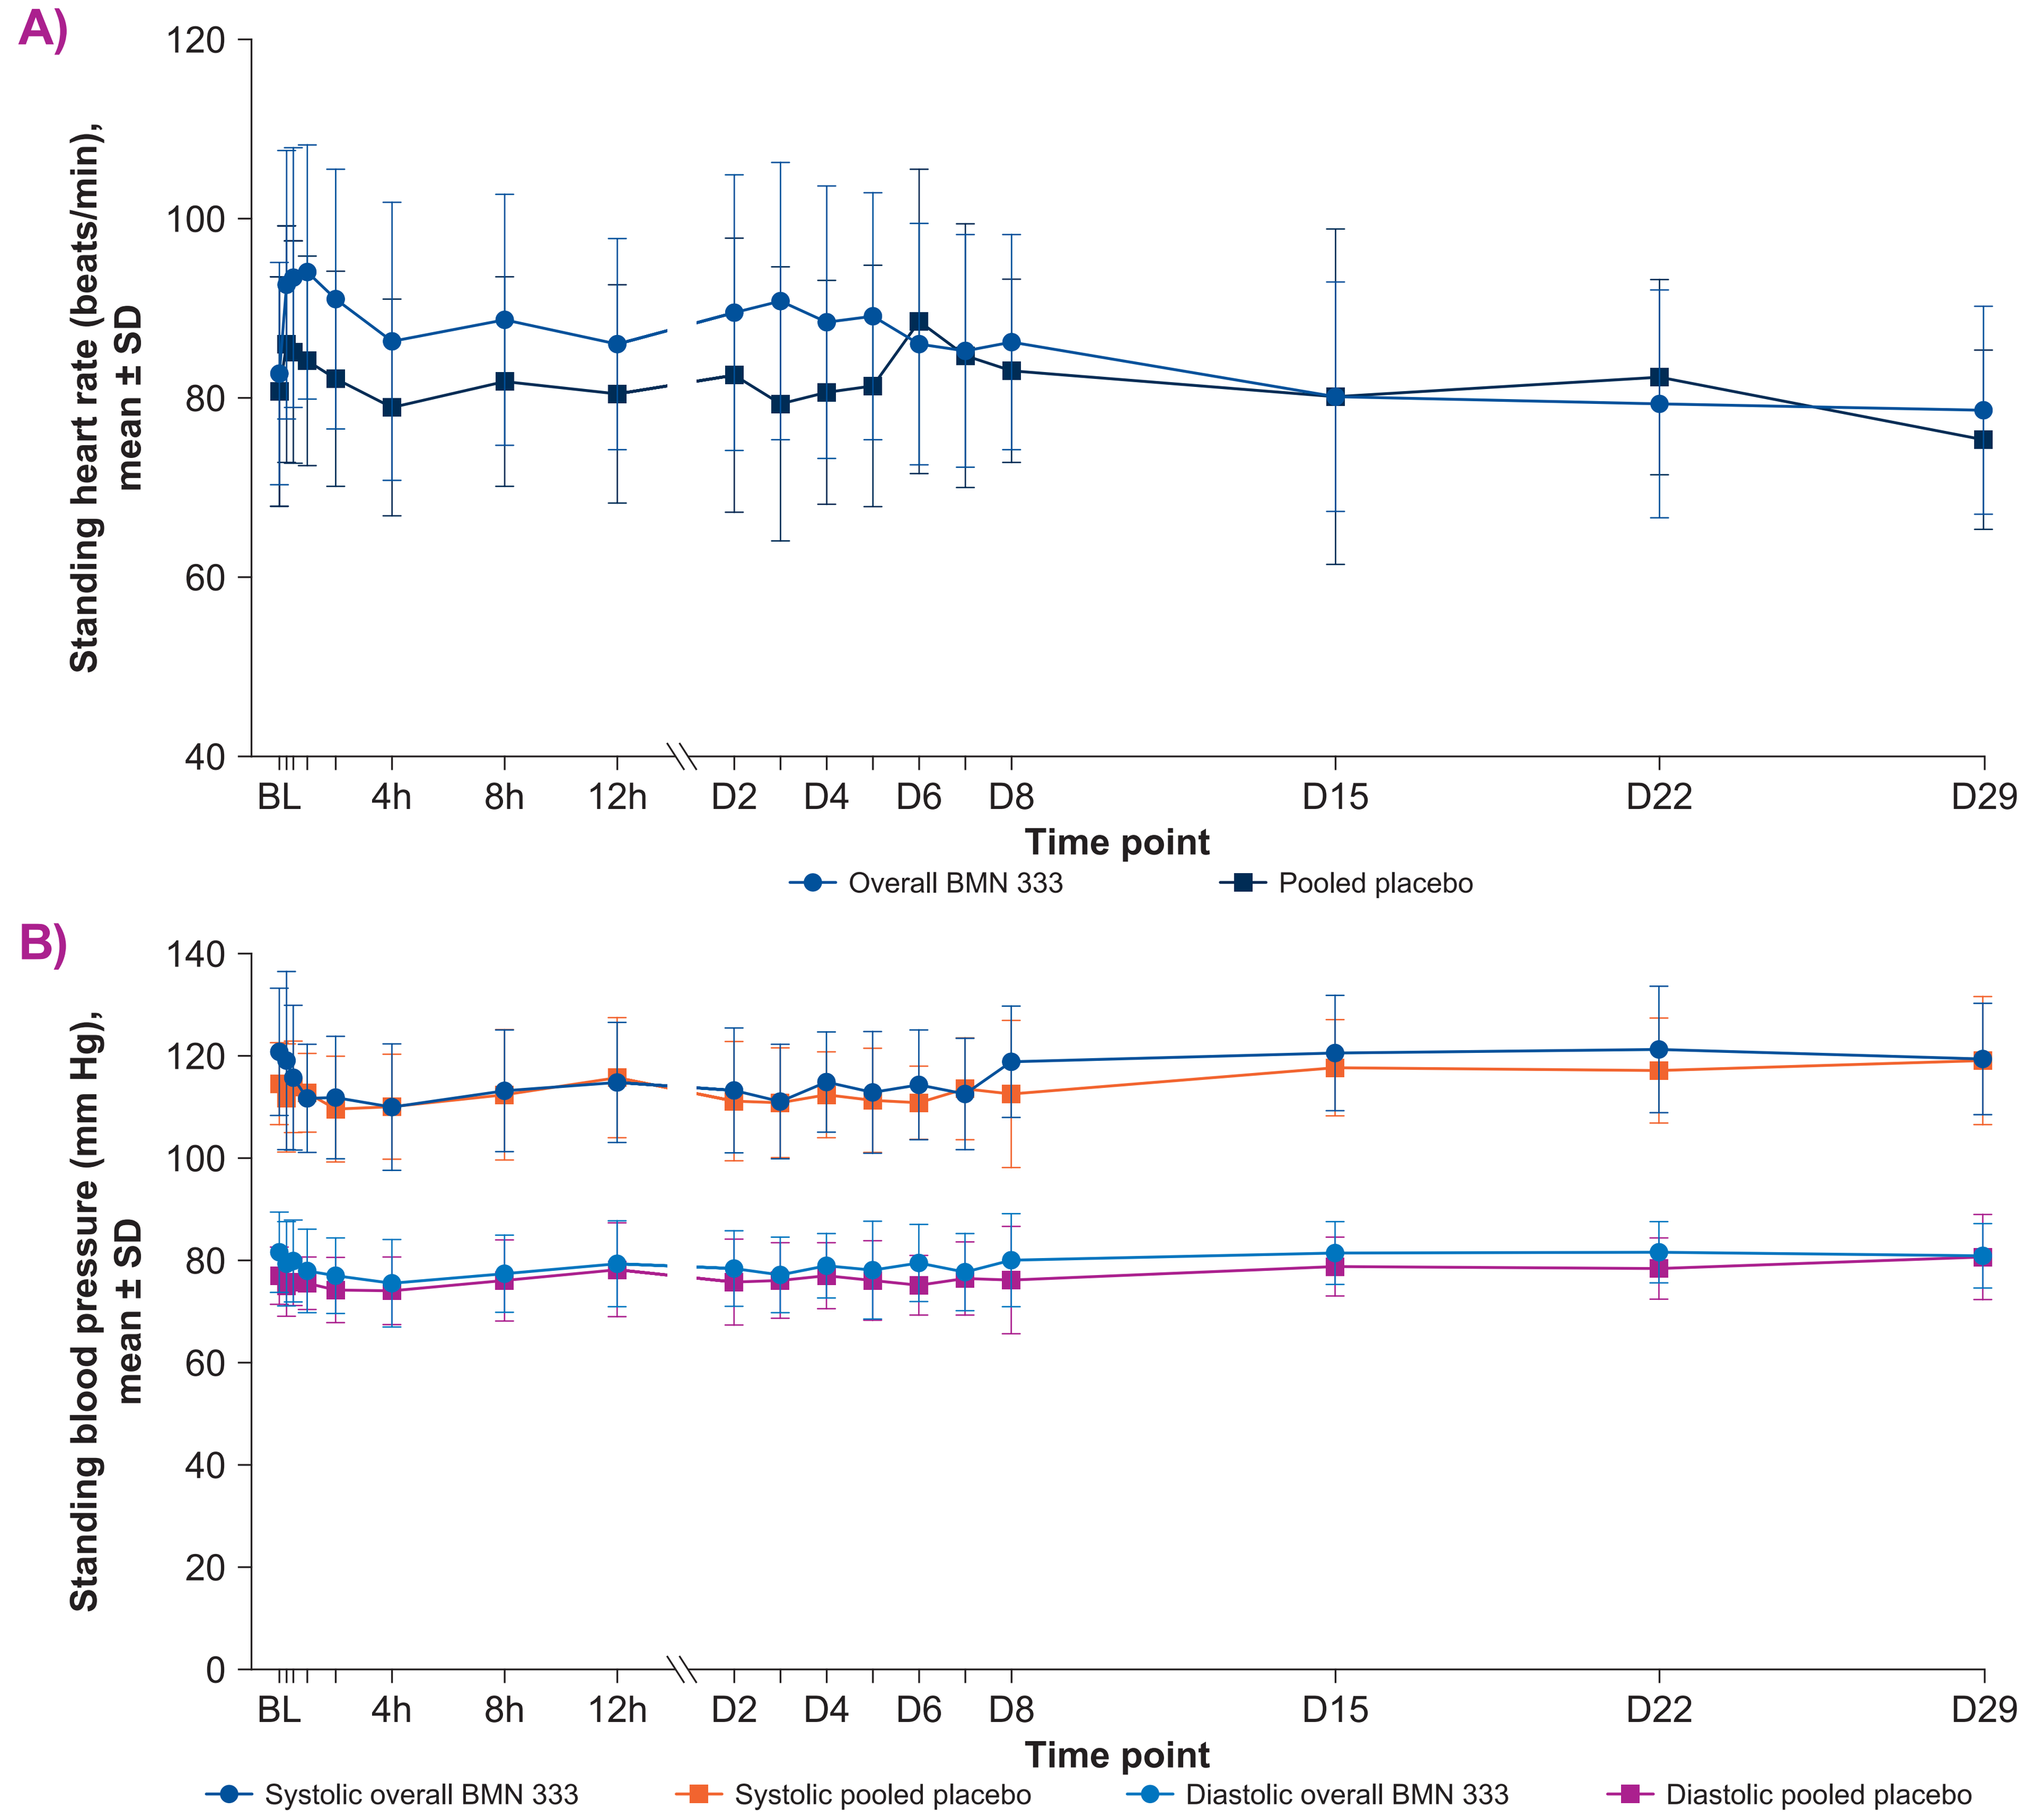
A)
Standing heart rate (beats/min), mean ± SD
BL 4h 8h 12h D2 D4 D6 D8 D15 D22 D29
Time point
Overall BMN 333
Pooled placebo
B)
Standing blood pressure (mm Hg), mean ± SD
Time point
Systolic overall BMN 333
Systolic pooled placebo
Diastolic overall BMN 333
Diastolic pooled placebo

BL, baseline; D, day; SD, standard deviation.

PK parameters

- BMN 333 exhibited linear, dose-proportional PK, with sustained and continuous exposure to released vosoritide that was up to >13 times higher than another long-acting CNP agent[7] (**Table 2**; **Figure 4**)

Table 2. Pharmacokinetic parameters for released vosoritide

	BMN 333 dose					
	15 µg/kg (n = 7)	50 µg/kg (n = 7)	100 µg/kg (n = 7)	200 µg/kg (n = 7)	350 µg/kg (n = 7)	500 µg/kg (n = 7)
C_{max}, pM	10.7 ± 5.97	23.6 ± 8.12	52.3 ± 20.6	180 ± 112	348 ± 352	512 ± 143
$t_{1/2}$, h	ND	ND	108 ± 13.9[a]	95.9 ± 15.1	99.5 ± 21.4	123 ± 9.95
AUC_{0-t}, pM*h	831 ± 781	2530 ± 958	6960 ± 2570	15,200 ± 6360	24,800 ± 17,100	44,800 ± 15,000
$AUC_{0-\infty}$, pM*h	ND	ND	9910 ± 1790[a]	16,900 ± 7140	26,500 ± 17,600	46,200 ± 14,900

Data shown as mean ± SD.
[a]n = 4.
AUC_{0-t}, area under the plasma concentration-time curve from 0 to the time of last measurable concentration; $AUC_{0-\infty}$, area under the plasma concentration-time curve from time 0 to infinity; C_{max}, maximum observed plasma concentration; ND, not determined; SD, standard deviation; $t_{1/2}$, elimination half-life.

Figure 4. Dose-proportional increases in systemic released vosoritide. A) Plasma concentration of released vosoritide, B) correlation between released vosoritide and BMN 333 dose, and C) AUC_{0-t} compared with another long-acting agent

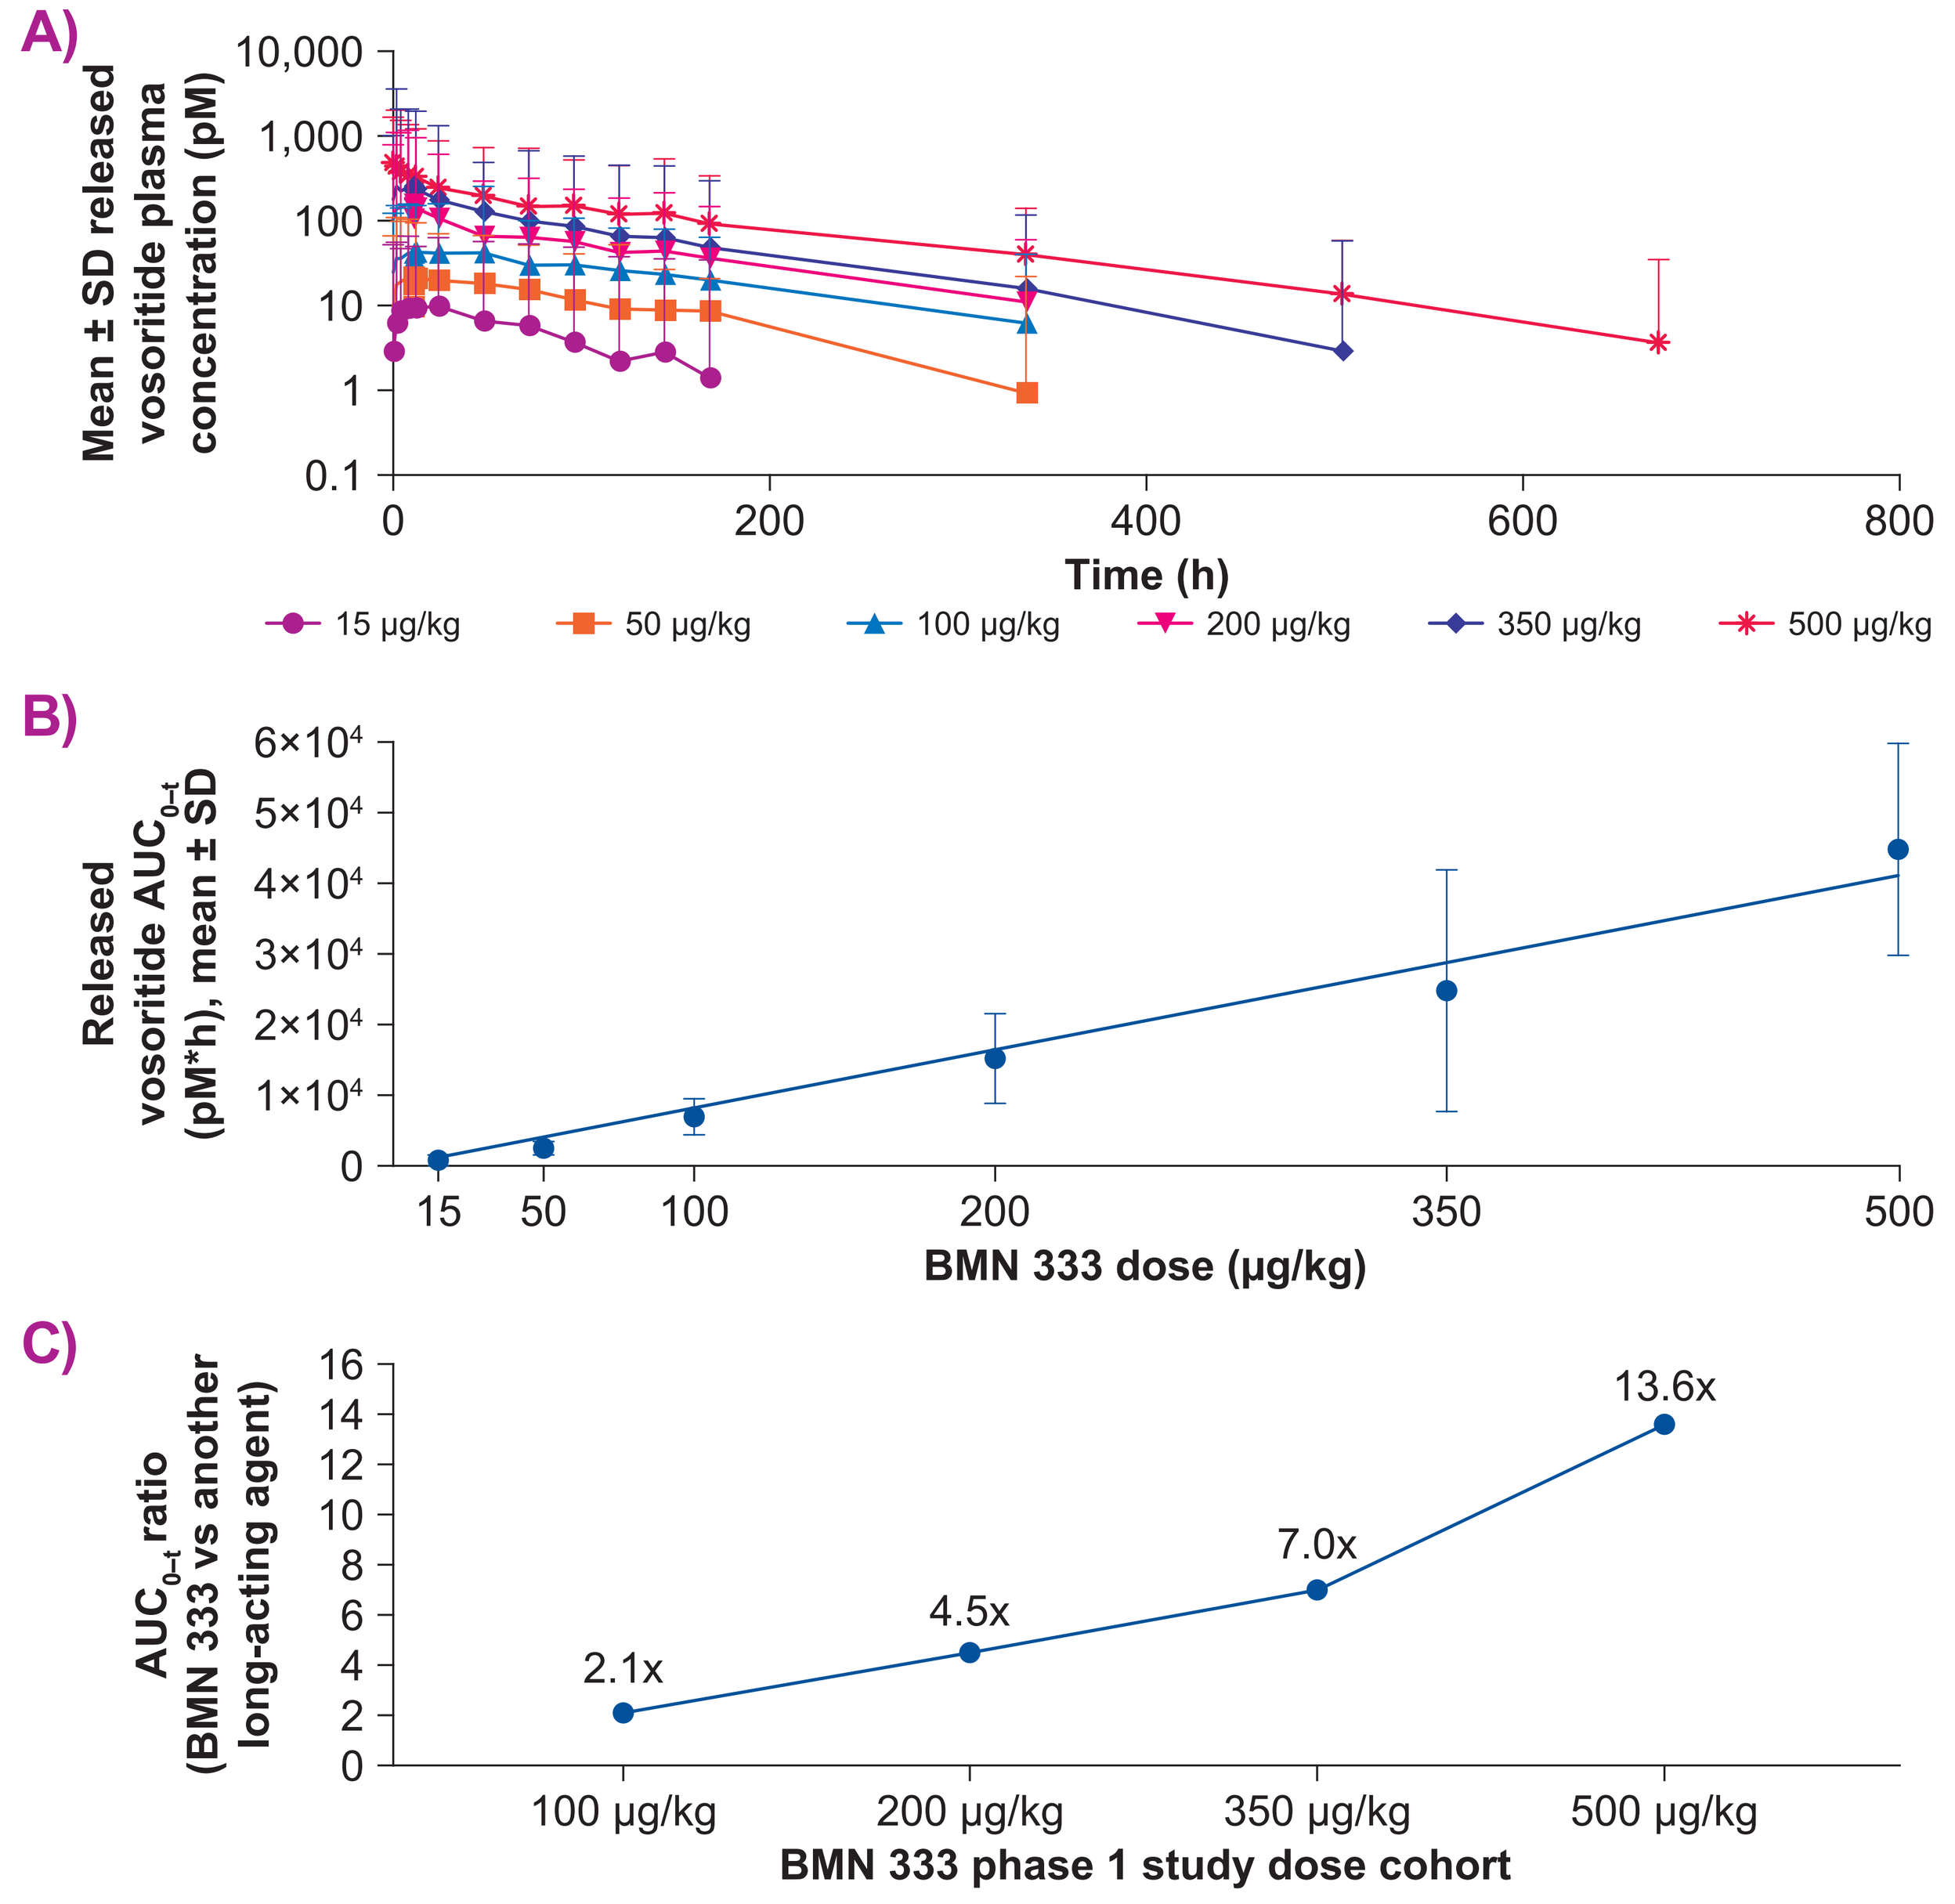
A)
Mean ± SD released vosoritide plasma concentration (pM)
Time (h)
15 µg/kg
50 µg/kg
100 µg/kg
200 µg/kg
350 µg/kg
500 µg/kg
B)
Released vosoritide AUC_{0-t} (pM*h), mean ± SD
BMN 333 dose (µg/kg)
C)
AUC_{0-t} ratio (BMN 333 vs another long-acting agent)
2.1x
4.5x
7.0x
13.6x
100 µg/kg
200 µg/kg
350 µg/kg
500 µg/kg
BMN 333 phase 1 study dose cohort

[a]Reference to geometric mean of AUC_{0-t} in Breinholt et al, *Br J Clin Pharmacol.* 2022;88(12):5203–15.
AUC_{0-t}, area under the plasma concentration-time curve from 0 to the time of last measurable concentration; SD, standard deviation.

- When compared with directly administered vosoritide, BMN 333 had a >10-fold lower C_{max}, extended the t_{max} from minutes to hours, and provided more sustained plasma vosoritide levels in healthy adults (**Figure 5**)

Figure 5. Systemic vosoritide levels in healthy adults following BMN 333 or direct administration

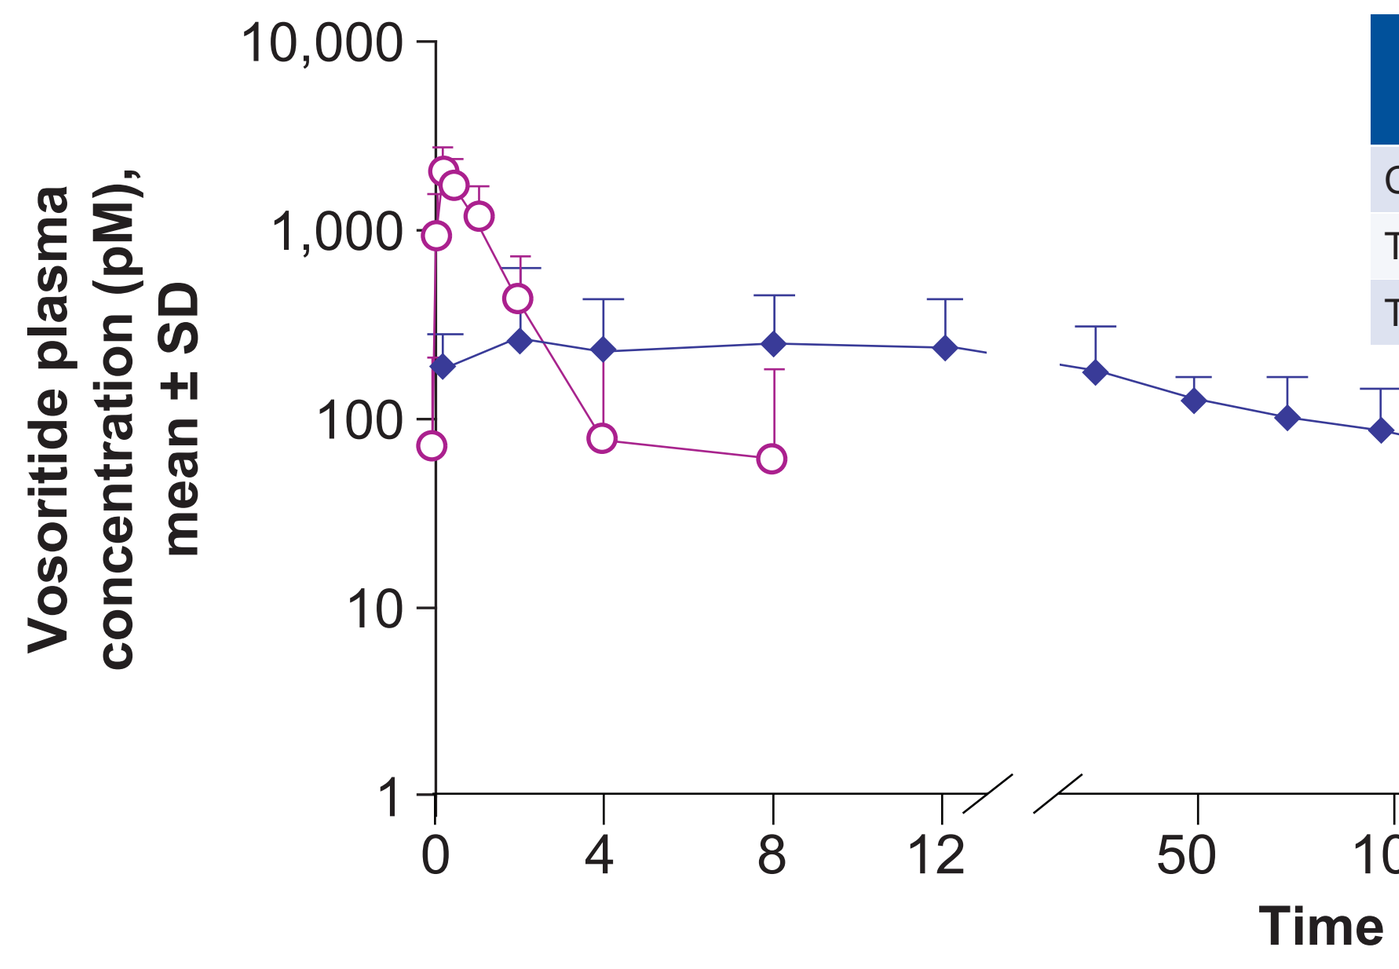
Vosoritide plasma concentration (pM), mean ± SD
Time (hours)

	Direct vosoritide (15 µg/kg)	Released vosoritide from BMN 333 (350 µg/kg)
C_{max}, (pM), mean (SD)	2152 (702)	348 (352)
T_{max}, (h), mean	0.4	8
$T_{1/2}$ (h), mean	1.2	99.5

Data for BMN 333 (study 333-101) and directly administered vosoritide (study 111-101)[8] are from 2 separate studies and are shown for descriptive purposes.
C_{max}, maximum observed plasma concentration; SD, standard deviation; t_{max}, time to reach maximum observed plasma concentration; $t_{1/2}$, elimination half-life.

PD parameters

- BMN 333 target engagement, as measured by cGMP, increased in a dose-dependent manner and remained elevated through day 7 at higher doses (**Figure 6**)

Figure 6. Plasma cGMP levels over time

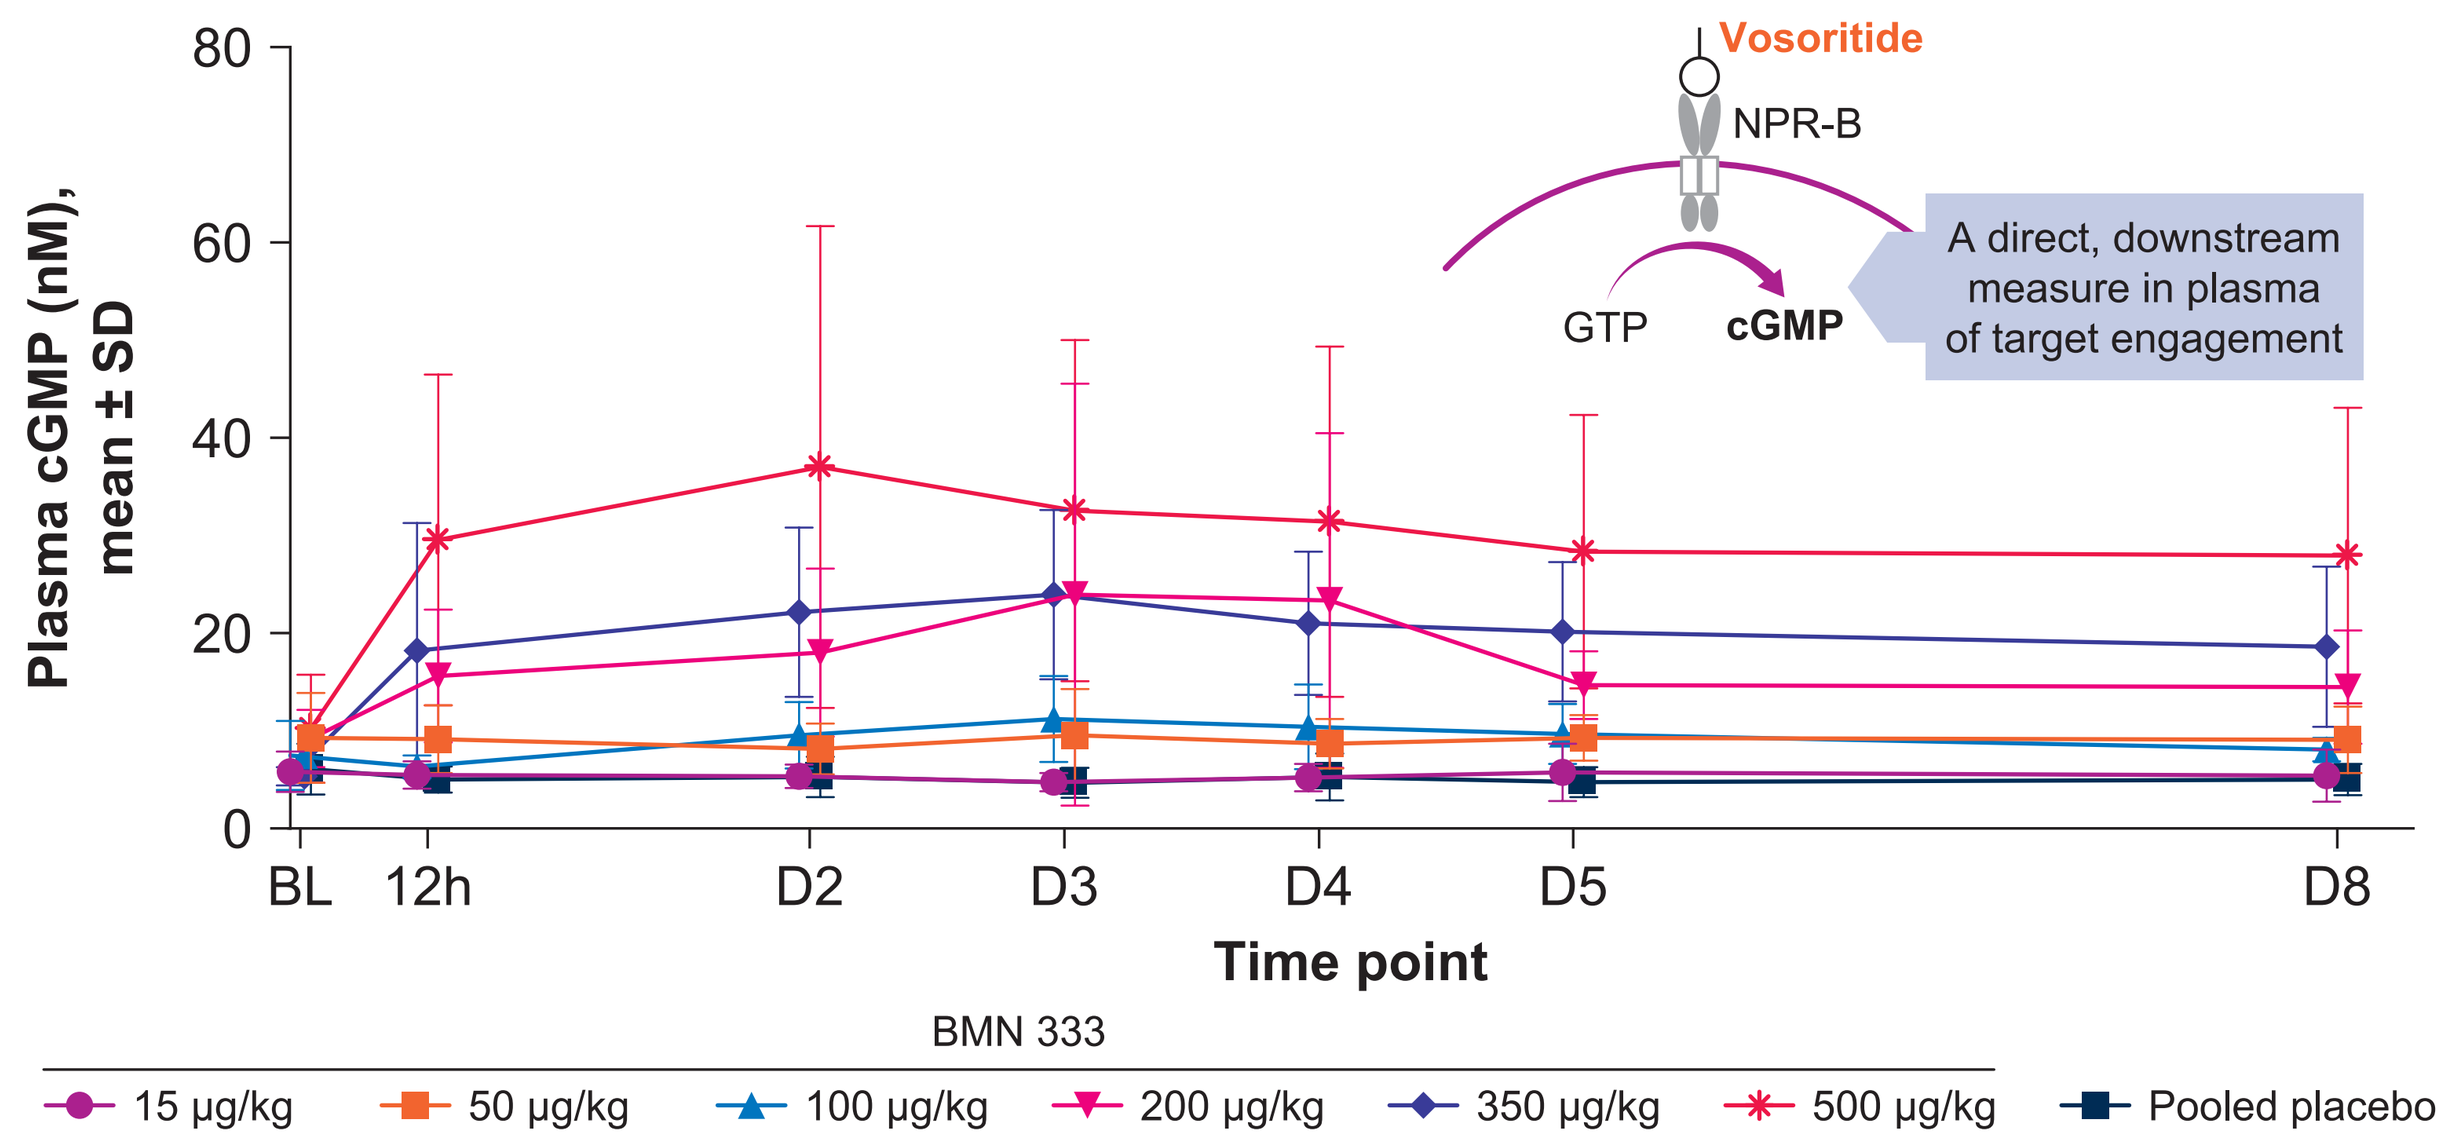
Vosoritide
NPR-B
GTP
cGMP
A direct, downstream measure in plasma of target engagement
Plasma cGMP (nM), mean ± SD
BL 12h D2 D3 D4 D5 D8
Time point
BMN 333
15 µg/kg
50 µg/kg
100 µg/kg
200 µg/kg
350 µg/kg
500 µg/kg
Pooled placebo

BL, baseline; cGMP, cyclic guanosine monophosphate; D, day; GTP, guanosine triphosphate; NPR-B, natriuretic peptide receptor B; SD, standard deviation.

Conclusions

- BMN 333 was well tolerated in this single ascending dose study in healthy adults, and the lower C_{max} relative to directly administered vosoritide may be relevant to the observed absence of consistent vital sign changes
- BMN 333 provided extended systemic vosoritide exposure compared with directly administered vosoritide
 - The maximum examined dose of BMN 333 (500 µg/kg) increased exposure to free CNP by >13x compared with another long-acting CNP agent[7]
- Sustained PK was associated with prolonged cGMP elevation, supporting continued natriuretic peptide receptor B pathway activation
- BMN 333 is a long-acting drug with an extended half-life that improves the dosing frequency and provides sustained exposure with a lower C_{max} that may improve growth efficacy
- These data support further clinical evaluation in children with achondroplasia, via phase 2 and 3 trials, to determine whether sustained vosoritide exposure with BMN 333 translates into improved growth outcomes while maintaining an acceptable safety profile

References

1. Savarirayan R, et al. *Lancet Child Adolesc Health*. 2024;8(4):301–10. **2.** Voxzogo (vosoritide). Prescribing information. BioMarin Pharmaceutical Inc; 2021. **3.** Savarirayan R, et al. *Nat Rev Endocrinol*. 2025;21:314–24. **4.** Savarirayan R, et al. *NEJM*. 2019;381:25–35. **5.** Savarirayan R, et al. *Lancet*. 2020;396:684–92. **6.** Savarirayan R, et al. *Med*. 2025;6(5):100566. **7.** Breinholt VM, et al. *Br J Clin Pharmacol*. 2022;88(12):5203–15. **8.** Center for Drug Evaluation and Research. Vosoritide (VOXZOGO) integrated review. Application number: 214938Orig1s000. Accessed March 25, 2026. https://www.accessdata.fda.gov/drugsatfda_docs/nda/2021/214938Orig1s000IntegratedR.pdf

Acknowledgements

We would like to thank all study participants, their families, and study site personnel.

Medical writing support was provided by Morgan Engelhart Wynn, PhD, of Red Nucleus, and funded by BioMarin Pharmaceutical Inc. Project management support was provided by Justin Potuzak, PhD, of BioMarin Pharmaceutical Inc.

Disclosures

NM, **YQ**, **LH**, **DW**, **HW**, **RS**, and **HZ** are employees and shareholders of BioMarin Pharmaceutical Inc.

To view a copy of this poster, scan this QR code.
Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission from the authors.

Presented at the Endocrine Society (ENDO) annual meeting; June 13–16, 2026; Chicago, IL, USA

©2026 BioMarin Pharmaceutical Inc. All rights reserved.