Comprehensive panel of pharmacokinetic, pharmacodynamic, and biomarker measures drives clinical development of nivudirsen (BMN 351) for Duchenne muscular dystrophy

Kevin Larimore, Kristin Obrochta Moss, Huiyu Zhou, Katja Hebestreit, Evan Witt, Laura Wetzel, Maria Abbasi, Lexa Staskus, Annie Greenslade, Ara Yu, Kristen Jahr, Pavan Chityala, Rob Power, Peter Velázquez, Andrew Melton

BioMarin Pharmaceutical Inc., Novato, CA, USA

Introduction

- Duchenne muscular dystrophy (DMD) is a neuromuscular disorder characterized by progressive muscle weakness resulting from truncated, non-functional dystrophin protein[1,2]
- BMN 351 (nivudirsen) is an exon 51-skipping antisense oligonucleotide (ASO) designed to enable synthesis of functional, near full-length dystrophin protein[3,4]
- Study 351-201 (EUCT#2023-506737-30-00; NCT06280209) is an ongoing, open-label, phase 1/2 clinical trial of nivudirsen in participants with DMD amenable to exon 51 skipping
- Measurement of ASO drug concentration and localization in muscle tissue provides pharmacokinetic (PK) data
- Exon skipping, near full-length dystrophin protein, and gene expression biomarkers are pharmacodynamic (PD) measures of nivudirsen activity from muscle biopsies
- Systemic muscle injury biomarkers are global PD readouts from longitudinal serum and urine samples

Objectives

- The objective of this poster is to describe a multifaceted approach to understand drug exposure, mechanism of action, and impact on muscle gene expression and systemic biomarkers of muscle injury resulting from nivudirsen treatment
- Results from clinical study 351-201 are presented in additional posters by BioMarin Pharmaceutical Inc. (see QR codes)

Methods

- Muscle biopsy samples were collected from the biceps of participants in the 351-201 trial at baseline and after 13 or 25 weekly nivudirsen administrations
- The concentration of nivudirsen was measured in muscle biopsy tissue using a qualified quantitative hybridization ELISA (**Figure 1**)

Figure 1. Quantitative hybridization ELISA to measure nivudirsen in muscle biopsies

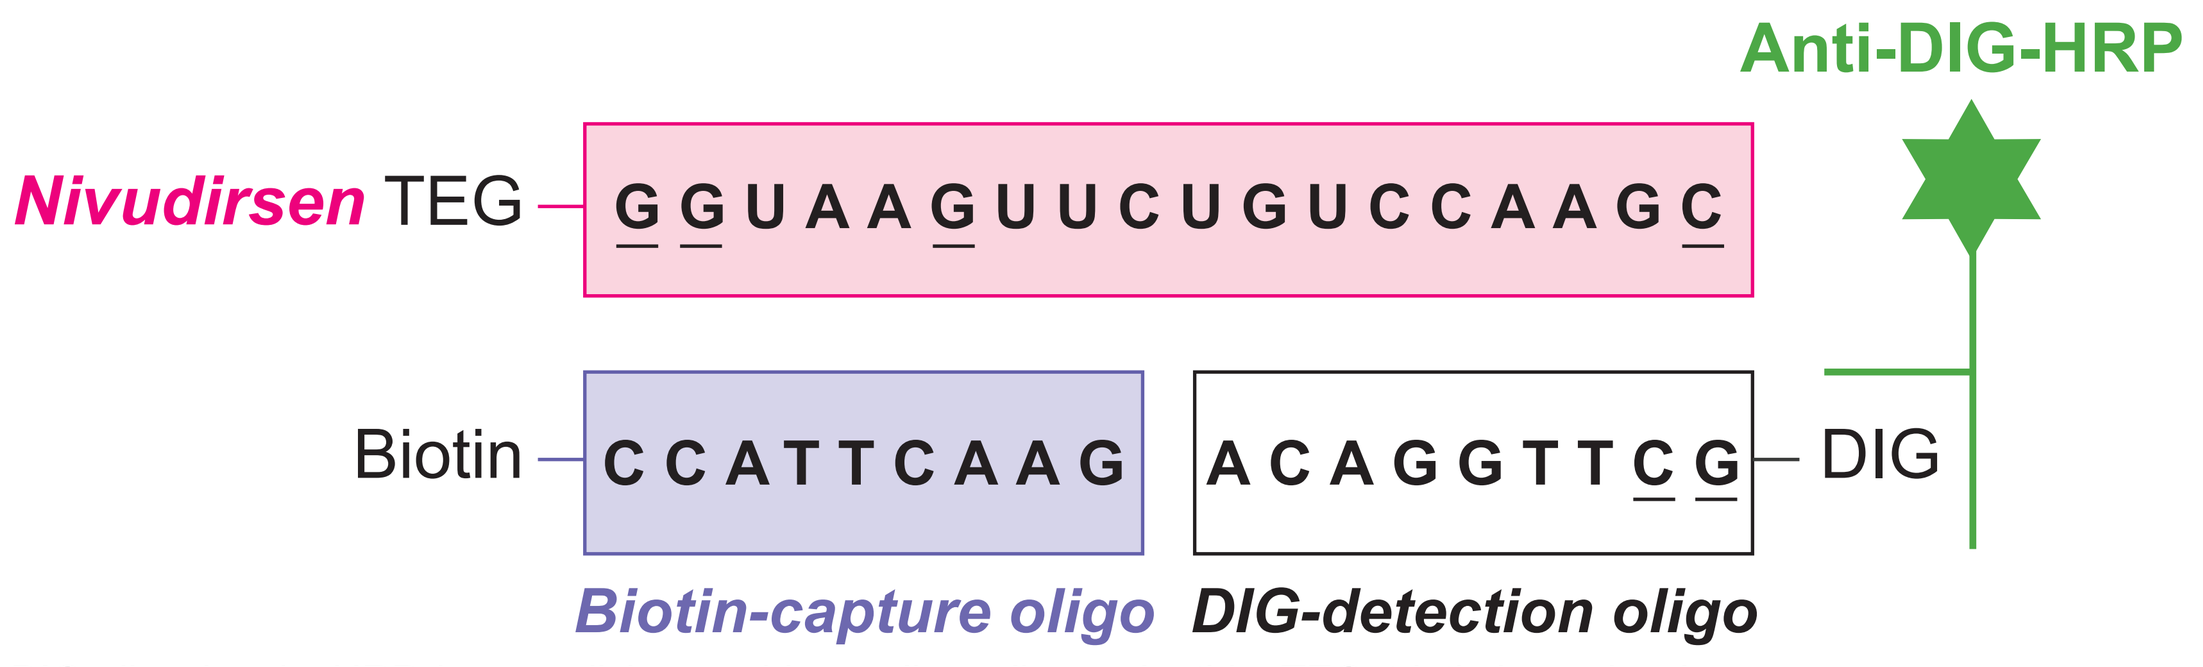
Anti-DIG-HRP
Nivudirsen TEG
GGUAAGUUCUGUCCAAGC
Biotin
CCATTCAAG
ACAGGTTCG
DIG
Biotin-capture oligo
DIG-detection oligo

DIG, digoxigenin; HRP, horseradish peroxidase; oligo, oligonucleotide; TEG, triethylene glycol.

- The distribution of nivudirsen in muscle was characterized by exploratory miRNAscope fluorescence in situ hybridization (FISH)
- *DMD* mRNA exon 51 skipping was measured in muscle using a characterized droplet digital PCR (ddPCR) method (**Figure 2**)

Figure 2. Quantitative ddPCR *DMD* exon 51 skip assay

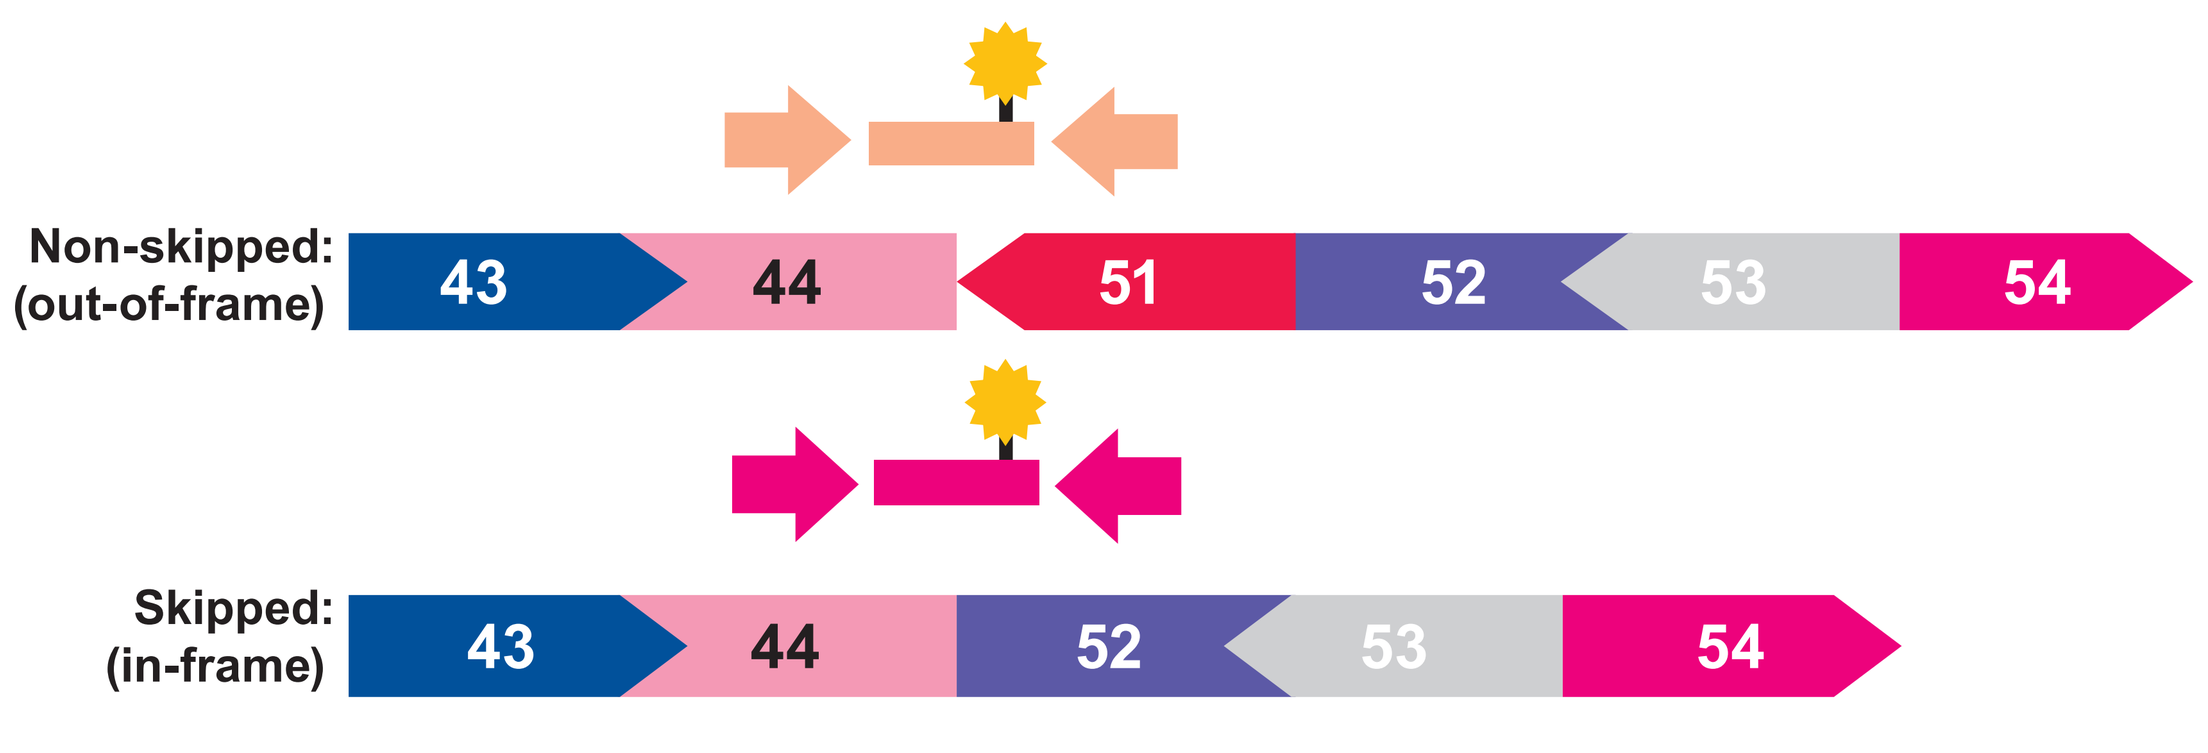
Non-skipped:
(out-of-frame)
43
44
51
52
53
54
Skipped:
(in-frame)
43
44
52
53
54

The illustration shows a representative genotype of exon 45 to 50 deletion; a skipped ddPCR transcript is only produced when exon 51 is skipped. Arrows represent primers, and bars represent fluorescently labeled probes.
ddPCR, droplet digital PCR.

- Near full-length dystrophin protein was measured in muscle by quantitative immunoaffinity liquid chromatography-tandem mass spectrometry (LC-MS; **Figure 3**)

Figure 3. Schematic of IA-UPLC-MS/MS analyte

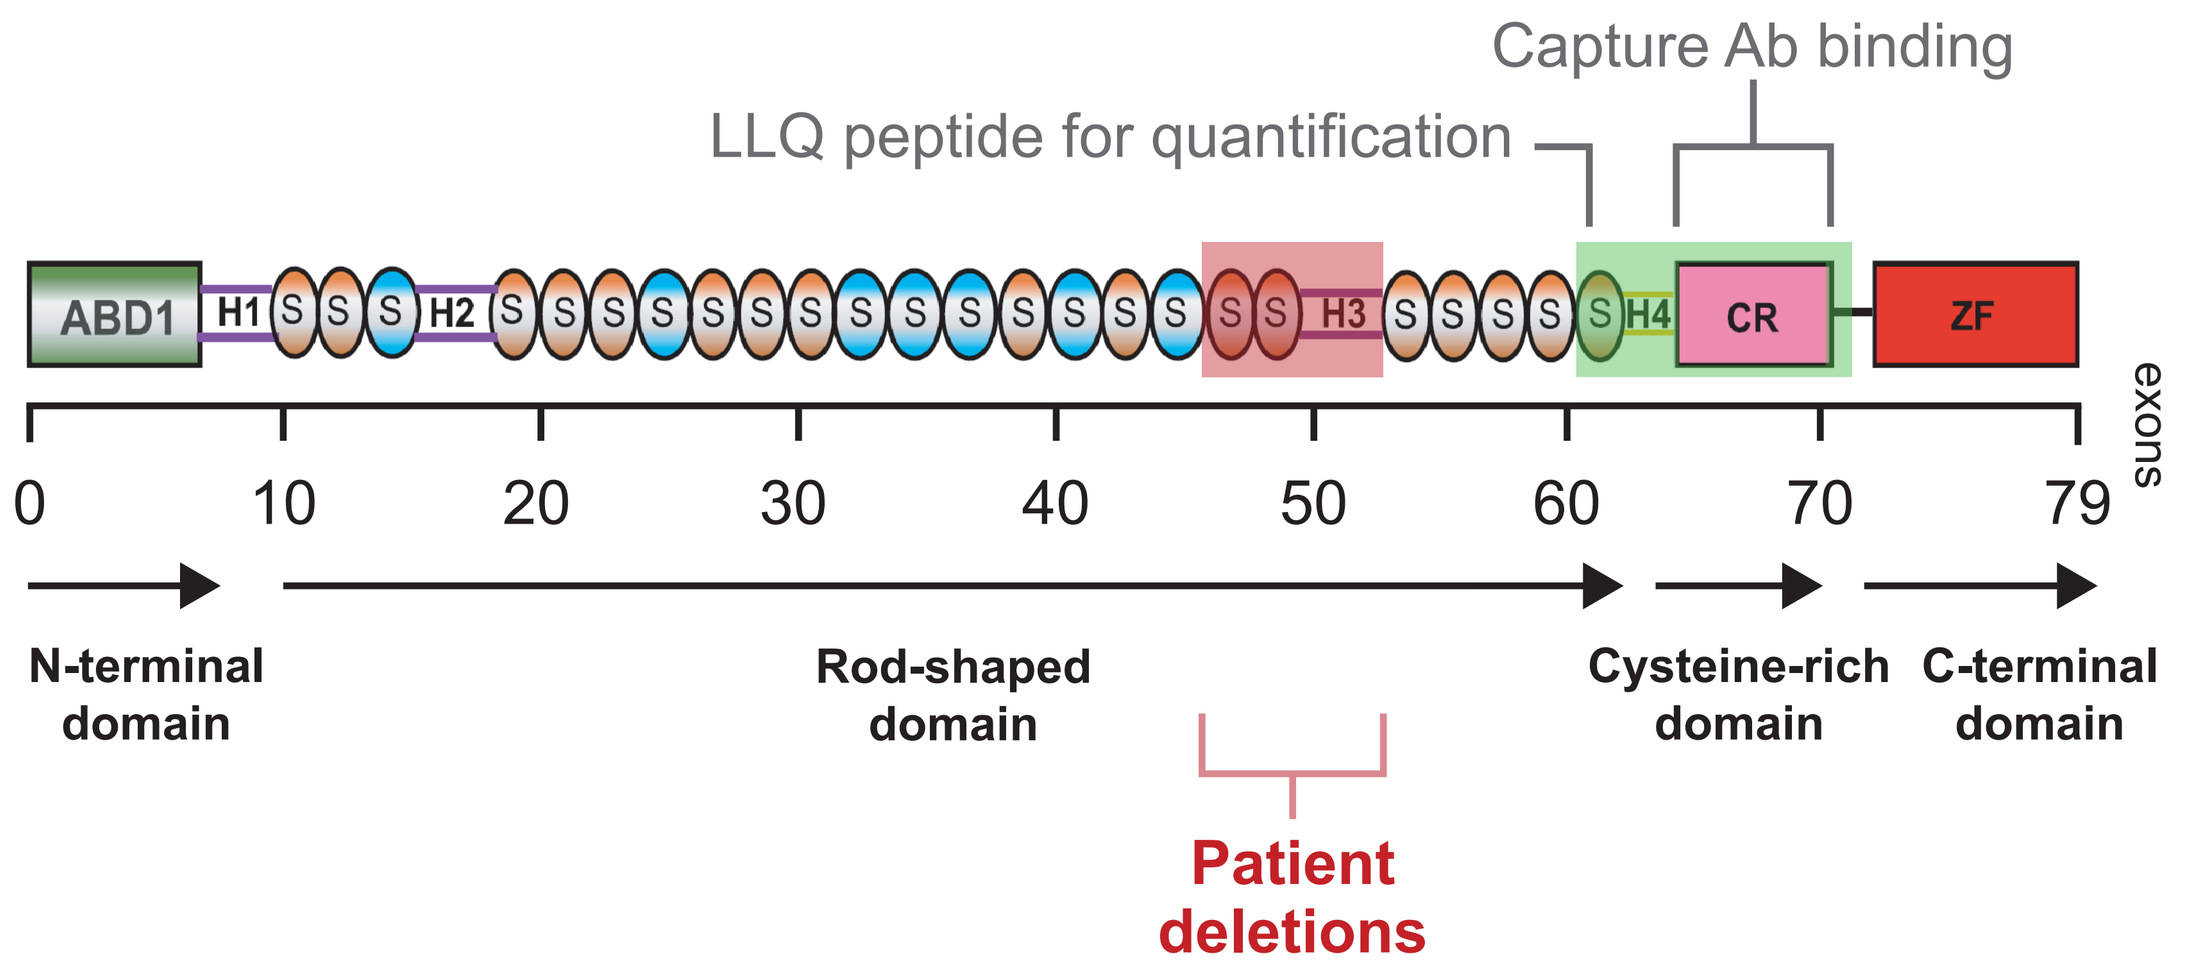
Capture Ab binding
LLQ peptide for quantification
ABD1
H1
H2
H3
H4
CR
ZF
0
10
20
30
40
50
60
70
79
exons
N-terminal domain
Rod-shaped domain
Cysteine-rich domain
C-terminal domain
Patient deletions

Ab, antibody; ABD, actin-binding domain; CR, cysteine-rich domain; H, hinge region; LLQ, a signature peptide (with the sequence of LLQVAVEDR); S, spectrin; IA-UPLC-MS/MS, immunoaffinity ultra-performance liquid chromatography coupled with tandem mass spectrometry; ZF, zinc-finger domain.
Figure 3 reproduced with minor changes from Wang Y, et al. Prenatal diagnosis of Duchenne muscular dystrophy revealed a novel mosaic mutation in dystrophin gene: a case report. *BMC Med Genet.* 2020;21(1):222. Licensed under CC BY 4.0 and cropped from original.

- Muscle RNA sequencing data were generated from wild-type or hDMDdel52/*mdx* mice treated for 13 weeks with vehicle or nivudirsen (18 mg/kg administered weekly)[3]
 - RNA sequencing was analyzed using a custom Snakemake pipeline; alignment and gene-level quantification were determined using STAR. Differential expression was assessed with DESeq2, and principal components analyses and gene set variation analyses were performed in R using base stats and the Gene Set Variation Analysis (GSVA) packages
- Myoblast- and myofiber-expressed genes were measured by quantitative ddPCR and used to calculate a myoblast-to-myofiber gene expression ratio
- Muscle function in mice was measured by MotoRater kinematic gait analysis
- Lactate dehydrogenase (LDH) and creatine kinase (CK) were measured in serum using validated enzymatic colorimetry assays
- Titin was measured in urine by ELISA and normalized to osmolality

Results

Figure 4. Measuring drug in muscle biopsies: Dose dependent increases of nivudirsen in study 351-201

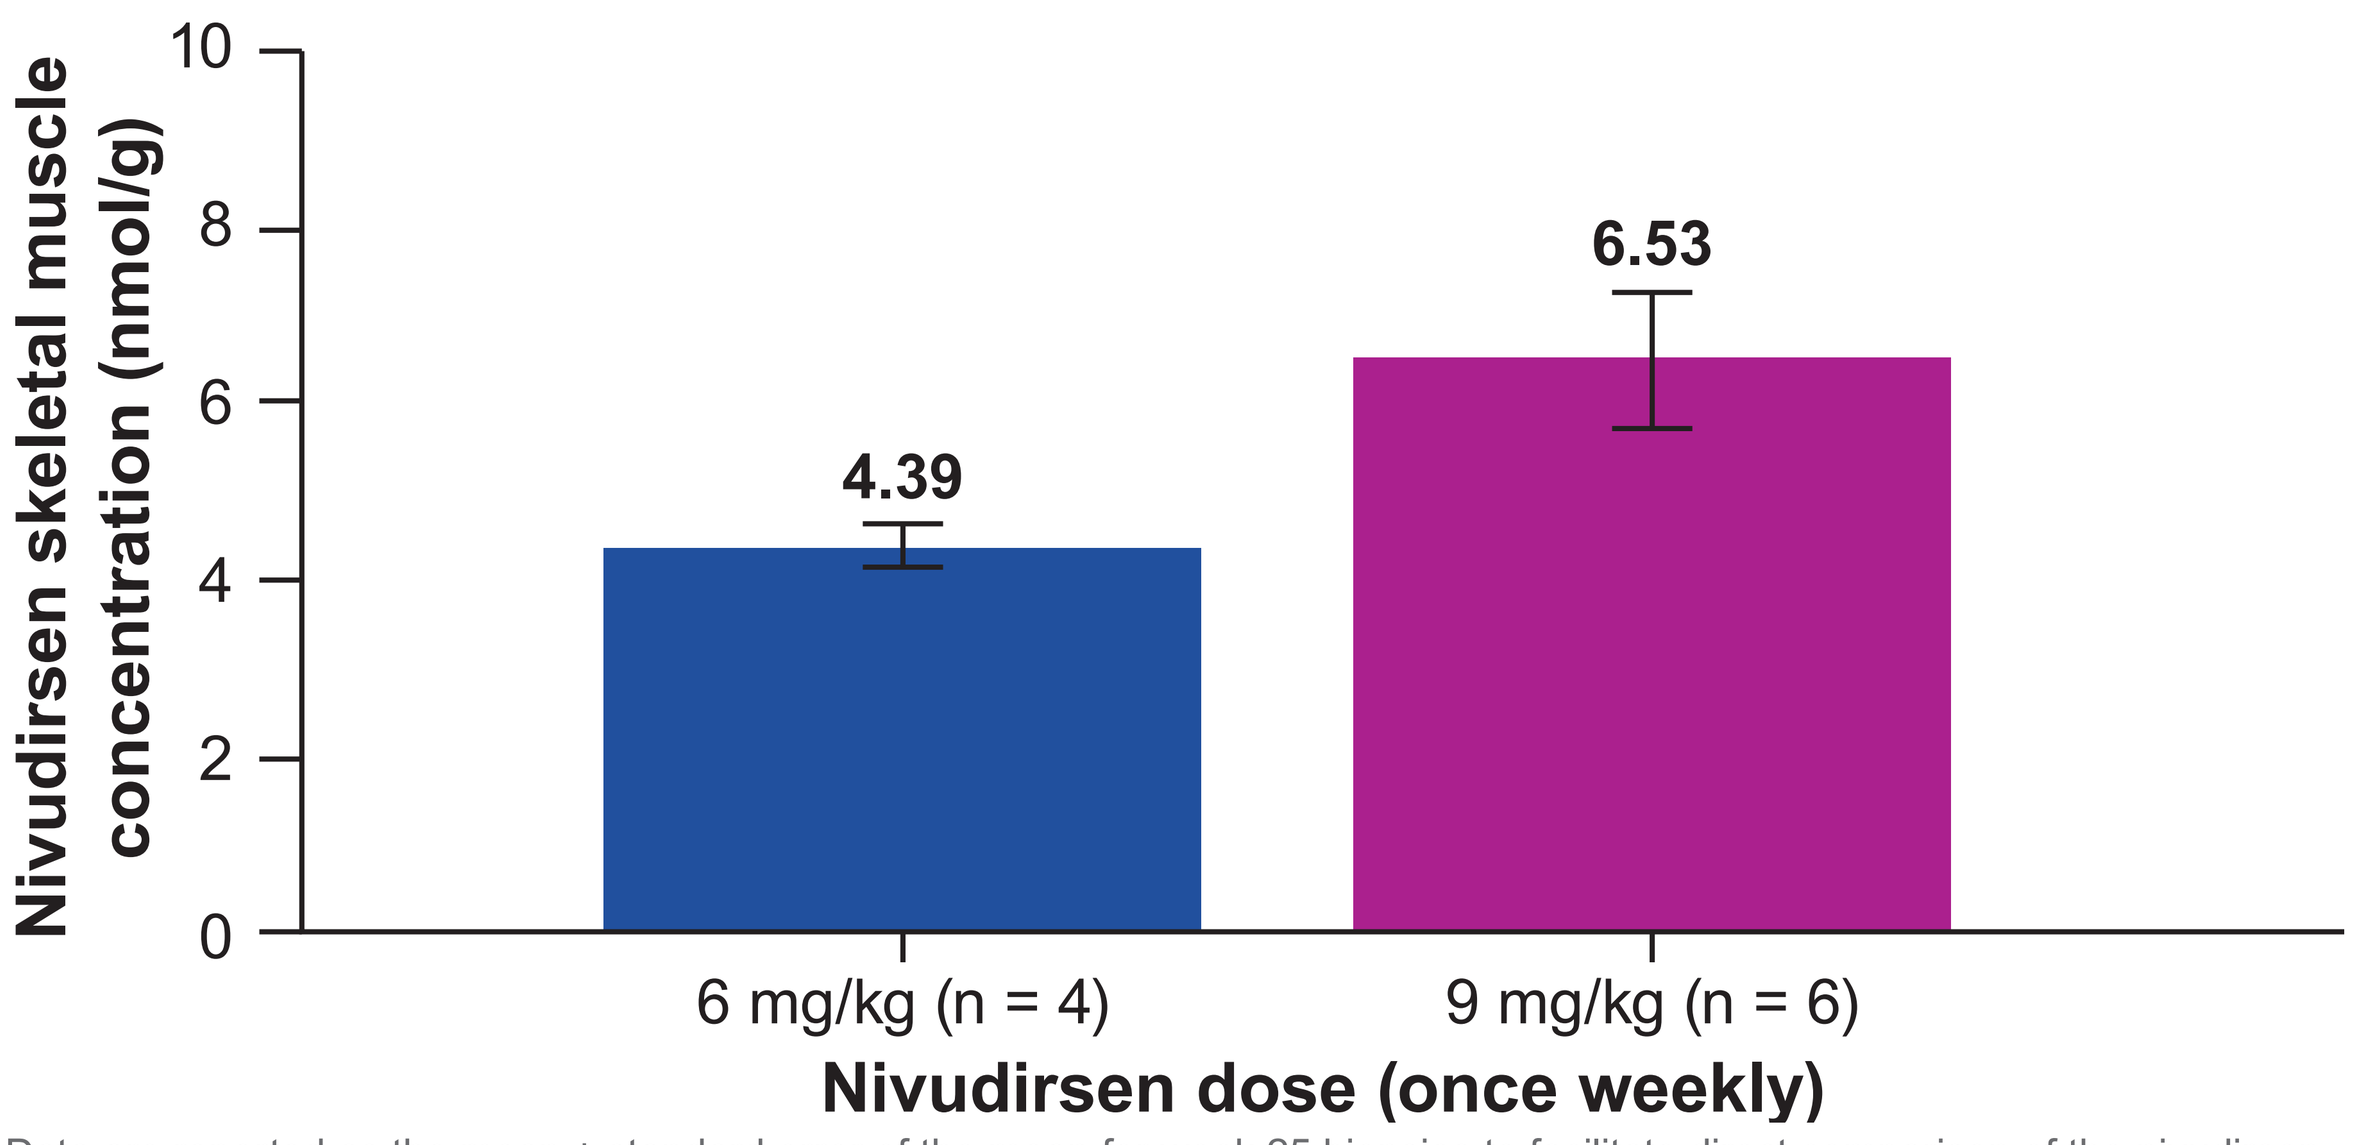
Nivudirsen skeletal muscle concentration (nmol/g)
10
8
6
4
2
0
4.39
6.53
6 mg/kg (n = 4)
9 mg/kg (n = 6)
Nivudirsen dose (once weekly)

Data are reported as the mean ± standard error of the mean for week 25 biopsies to facilitate direct comparison of the nivudirsen concentration (cohort 1A 6-mg/kg biopsies collected at week 13 were not included in the analysis).

Figure 5. Characterizing drug distribution in muscle in study 351-201: Nivudirsen reaches myofiber nuclei

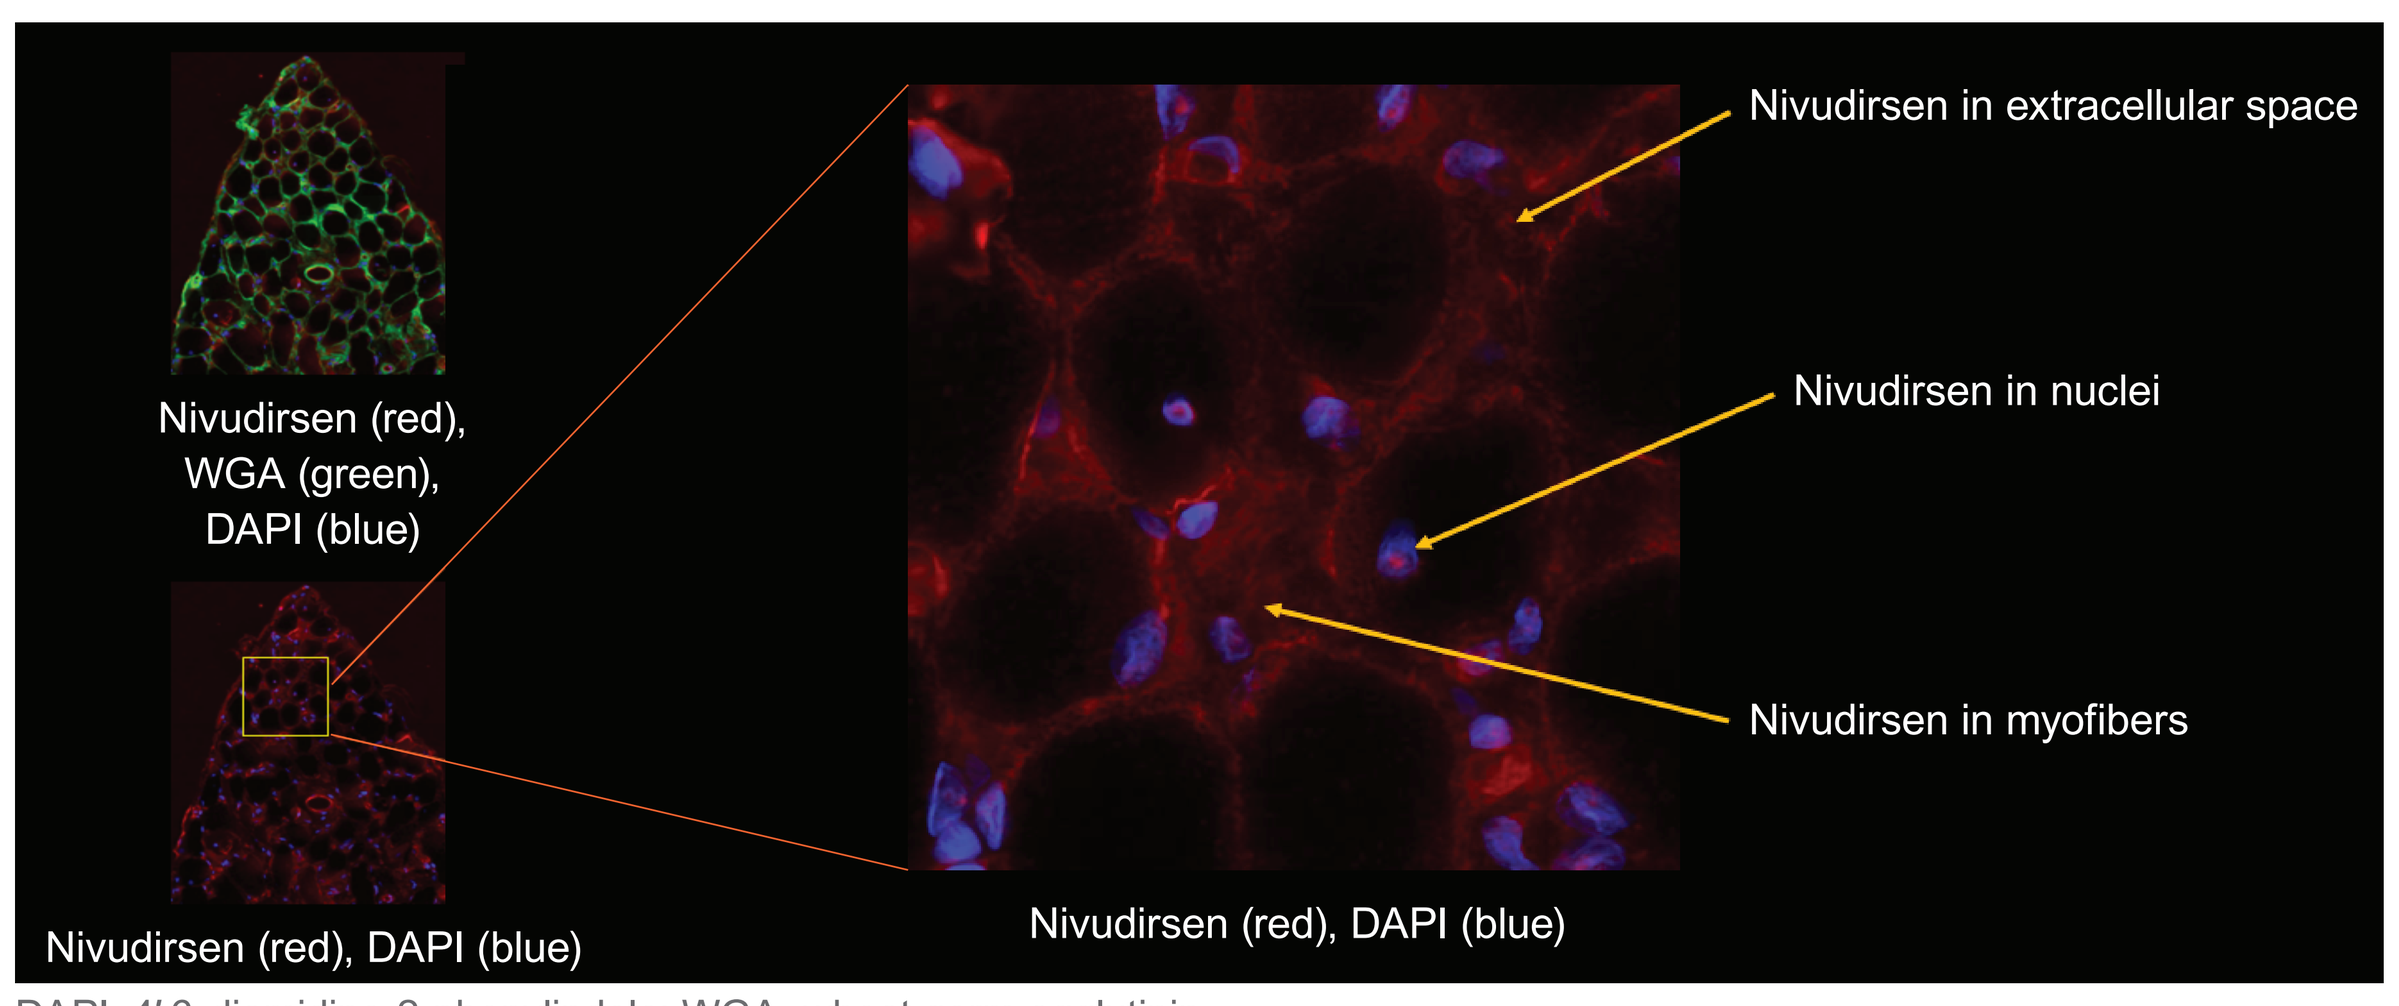
Nivudirsen (red), WGA (green), DAPI (blue)
Nivudirsen (red), DAPI (blue)
Nivudirsen in extracellular space
Nivudirsen in nuclei
Nivudirsen in myofibers
Nivudirsen (red), DAPI (blue)

DAPI, 4′,6-diamidino-2-phenylindole; WGA, wheat germ agglutinin.

Figure 6. Measuring proximal pharmacodynamics in muscle biopsies: Exon-skipped *DMD* mRNA correlated with near full-length dystrophin protein in study 351-201

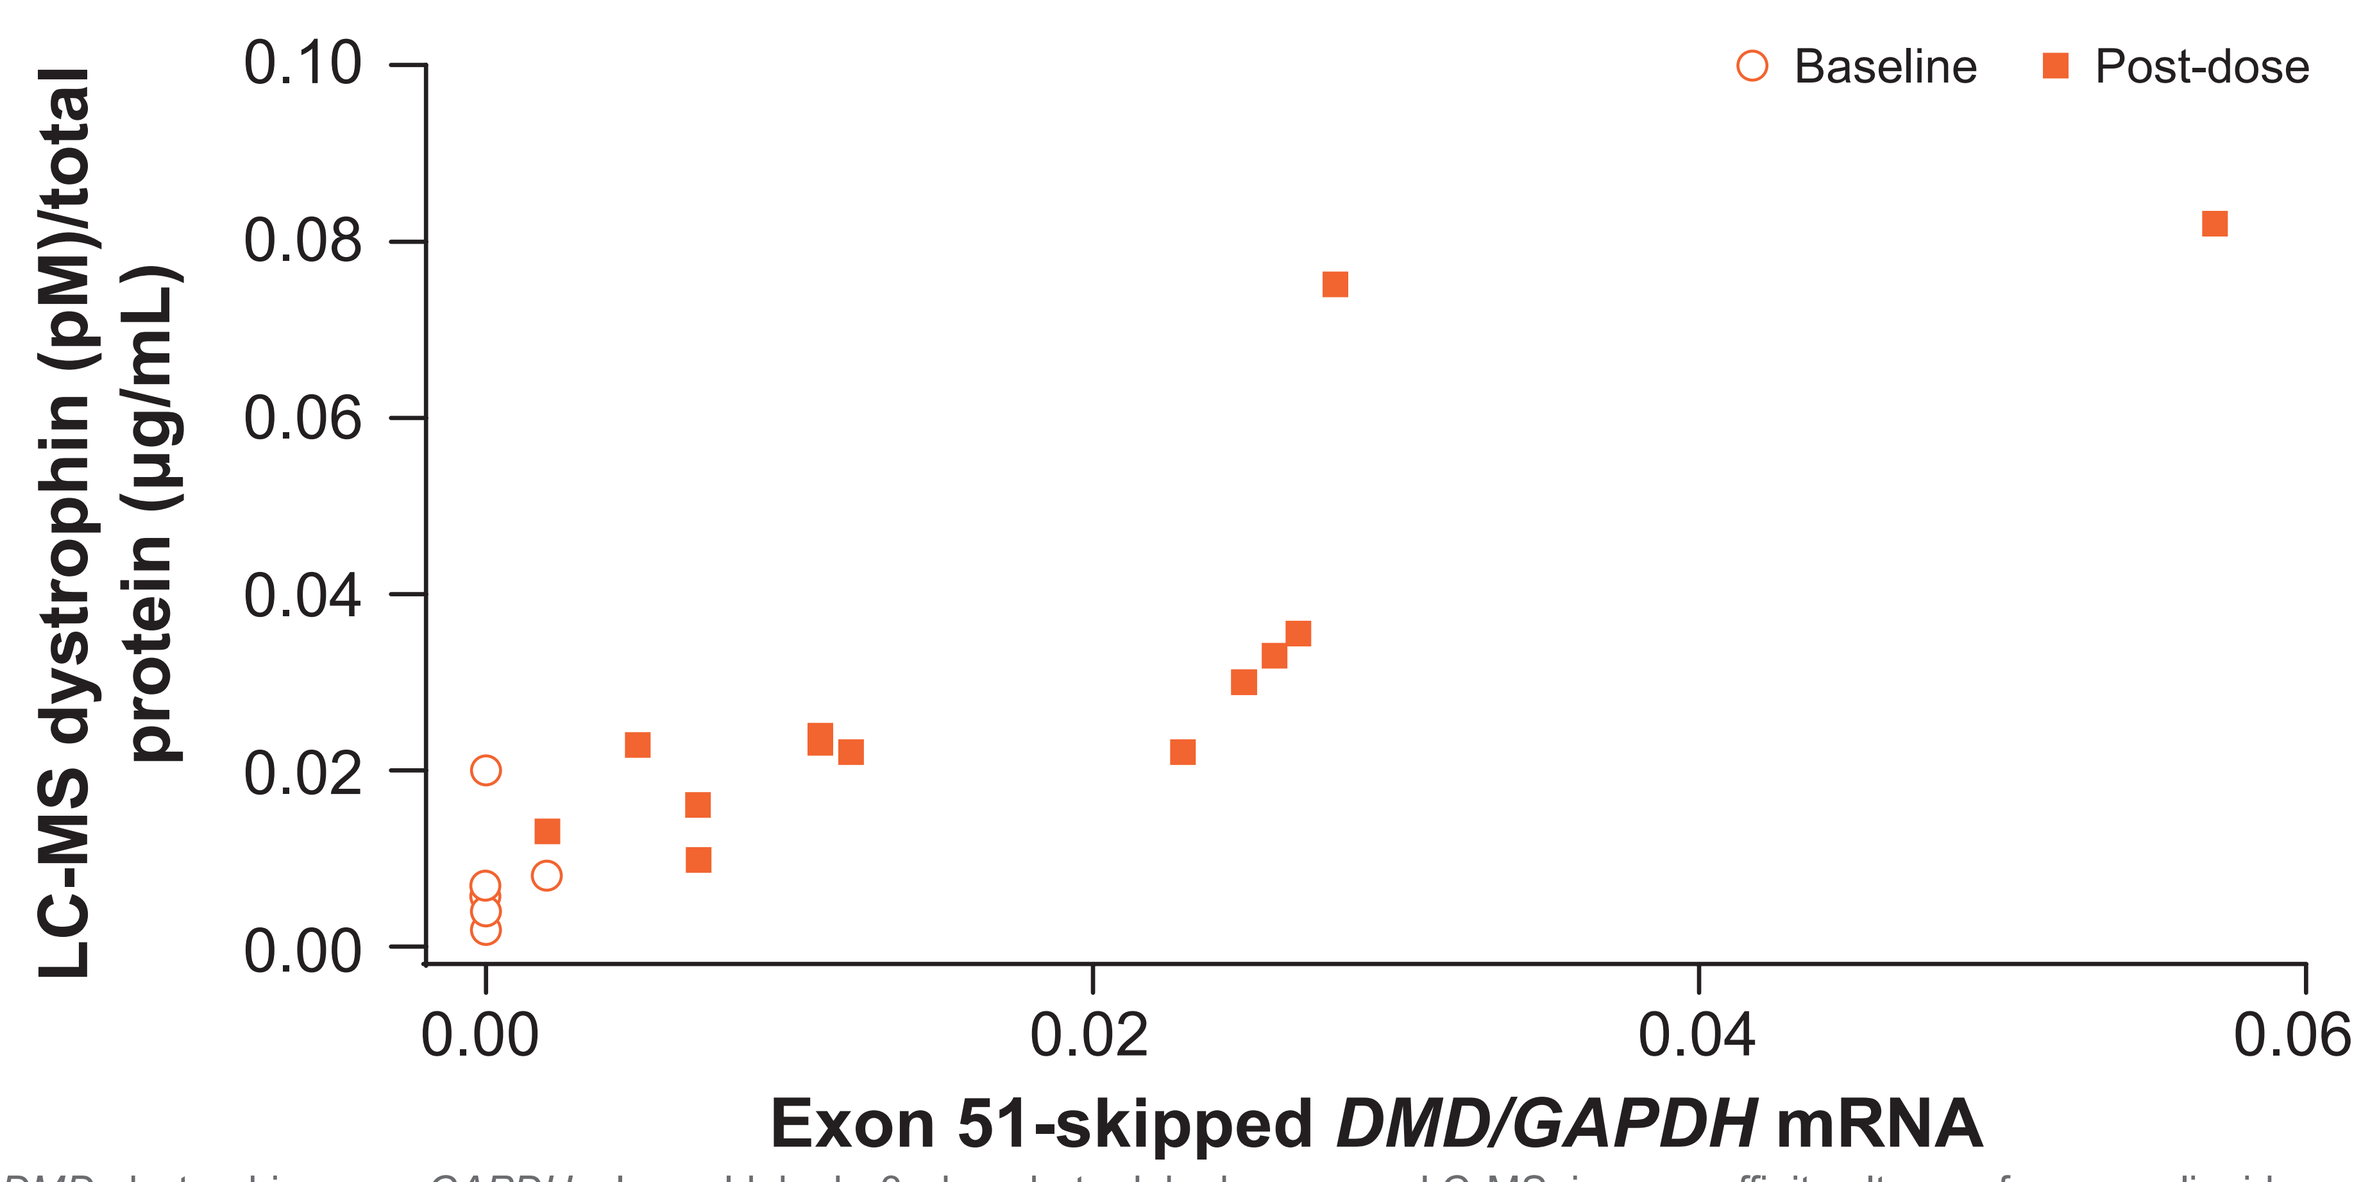
LC-MS dystrophin (pM)/total protein (µg/mL)
0.10
0.08
0.06
0.04
0.02
0.00
0.00
0.02
0.04
0.06
Exon 51-skipped DMD/GAPDH mRNA
Baseline
Post-dose

DMD, dystrophin gene; *GAPDH*, glyceraldehyde-3-phosphate dehydrogenase; LC-MS, immunoaffinity ultra-performance liquid chromatography coupled with tandem mass spectrometry.

Muscle exon-skipping and dystrophin expression data from study 351-201 are presented in poster #2.66P

351-201 clinical data

- Disease-associated gene expression in DMD mice involved muscle regeneration, extracellular matrix, and adaptive immune pathways (**Figure 7**)
- Muscle gene expression in DMD mice treated with nivudirsen for 13 weeks reverted to wild-type, suggesting broad impact on muscle biology

Figure 7. Disease-associated muscle gene expression and response to treatment in DMD mice

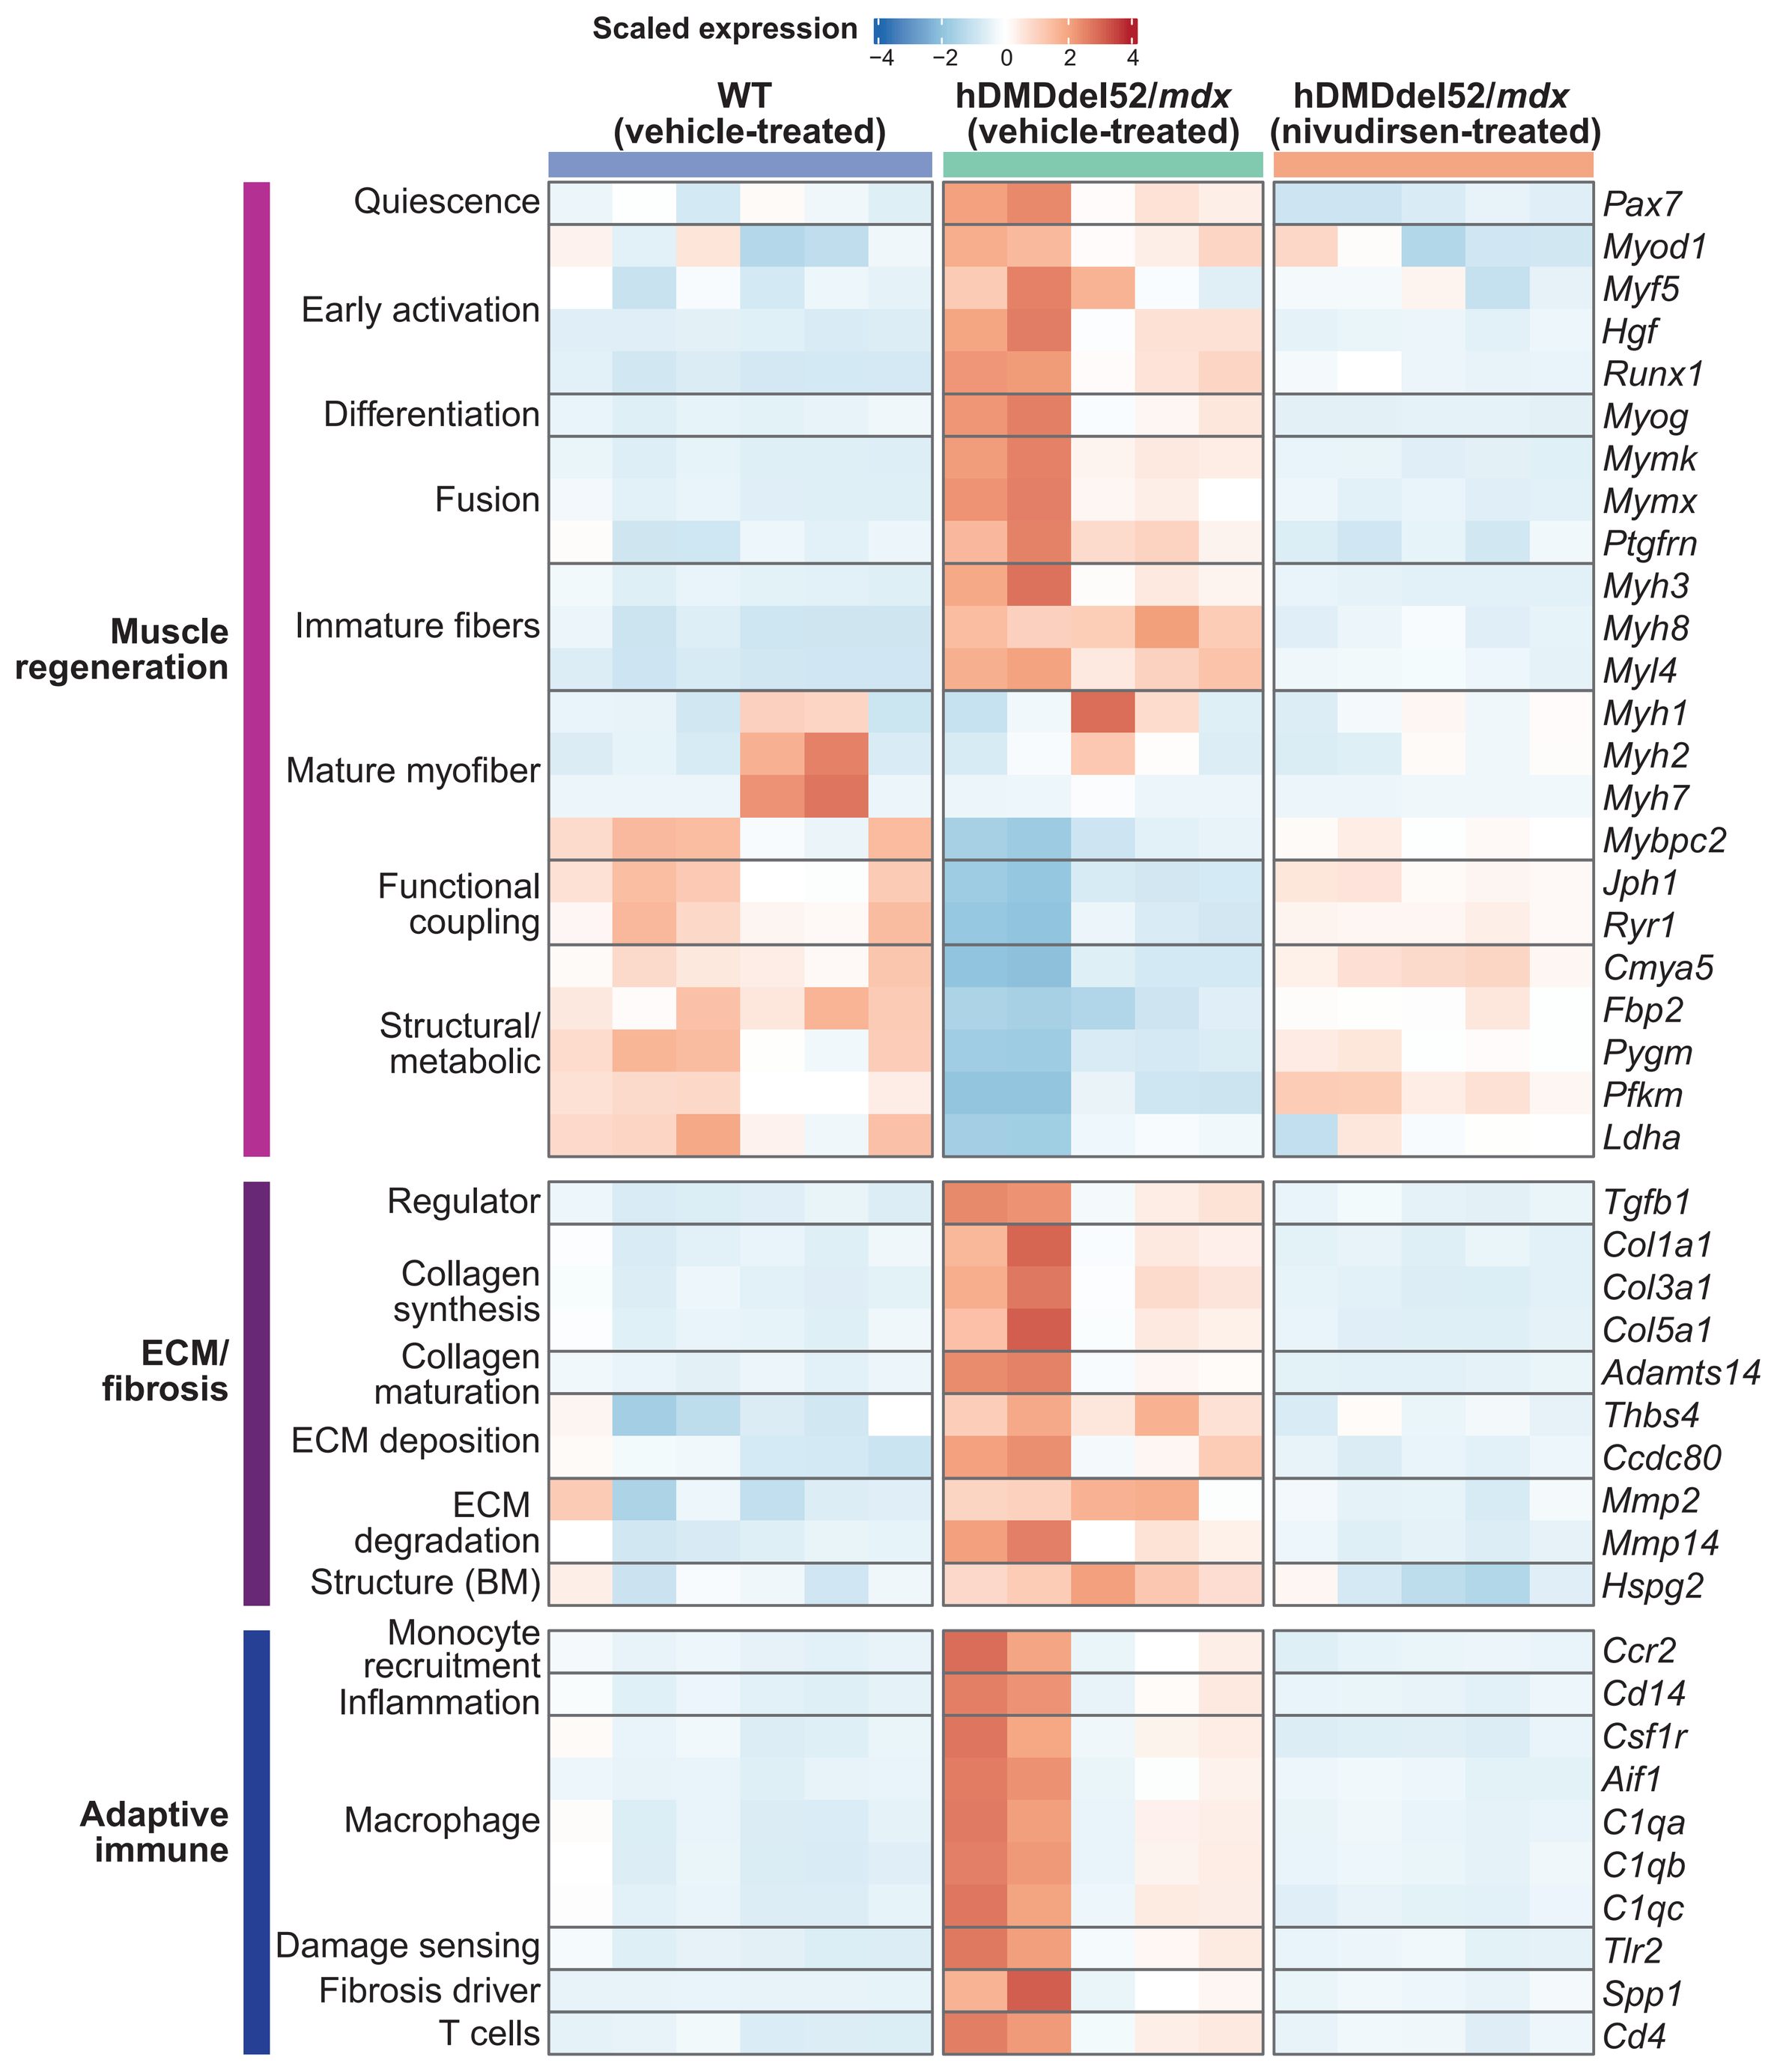
Scaled expression
−4 −2 0 2 4
WT (vehicle-treated)
hDMDdel52/mdx (vehicle-treated)
hDMDdel52/mdx (nivudirsen-treated)
Muscle regeneration
Quiescence
Early activation
Differentiation
Fusion
Immature fibers
Mature myofiber
Functional coupling
Structural/ metabolic
ECM/ fibrosis
Regulator
Collagen synthesis
Collagen maturation
ECM deposition
ECM degradation
Structure (BM)
Adaptive immune
Monocyte recruitment
Inflammation
Macrophage
Damage sensing
Fibrosis driver
T cells
Pax7
Myod1
Myf5
Hgf
Runx1
Myog
Mymk
Mymx
Ptgfrn
Myh3
Myh8
Myl4
Myh1
Myh2
Myh7
Mybpc2
Jph1
Ryr1
Cmya5
Fbp2
Pygm
Pfkm
Ldha
Tgfb1
Col1a1
Col3a1
Col5a1
Adamts14
Thbs4
Ccdc80
Mmp2
Mmp14
Hspg2
Ccr2
Cd14
Csf1r
Aif1
C1qa
C1qb
C1qc
Tlr2
Spp1
Cd4

Heatmaps in Figure 7 were generated using variance-stabilized expression values from an unsupervised DESeq2 model.
DMD, Duchenne muscular dystrophy; ECM, extracellular matrix; WT, wild-type.

- The muscle myoblast-to-myofiber gene expression ratio measured by quantitative ddPCR was elevated in DMD mice, reduced with nivudirsen treatment, and correlated with function measured by gait analysis (**Figure 8**)

Figure 8. Evaluating muscle myoblast-to-myofiber gene expression ratio as a disease biomarker: Reduction in treated DMD mice correlated with improved function

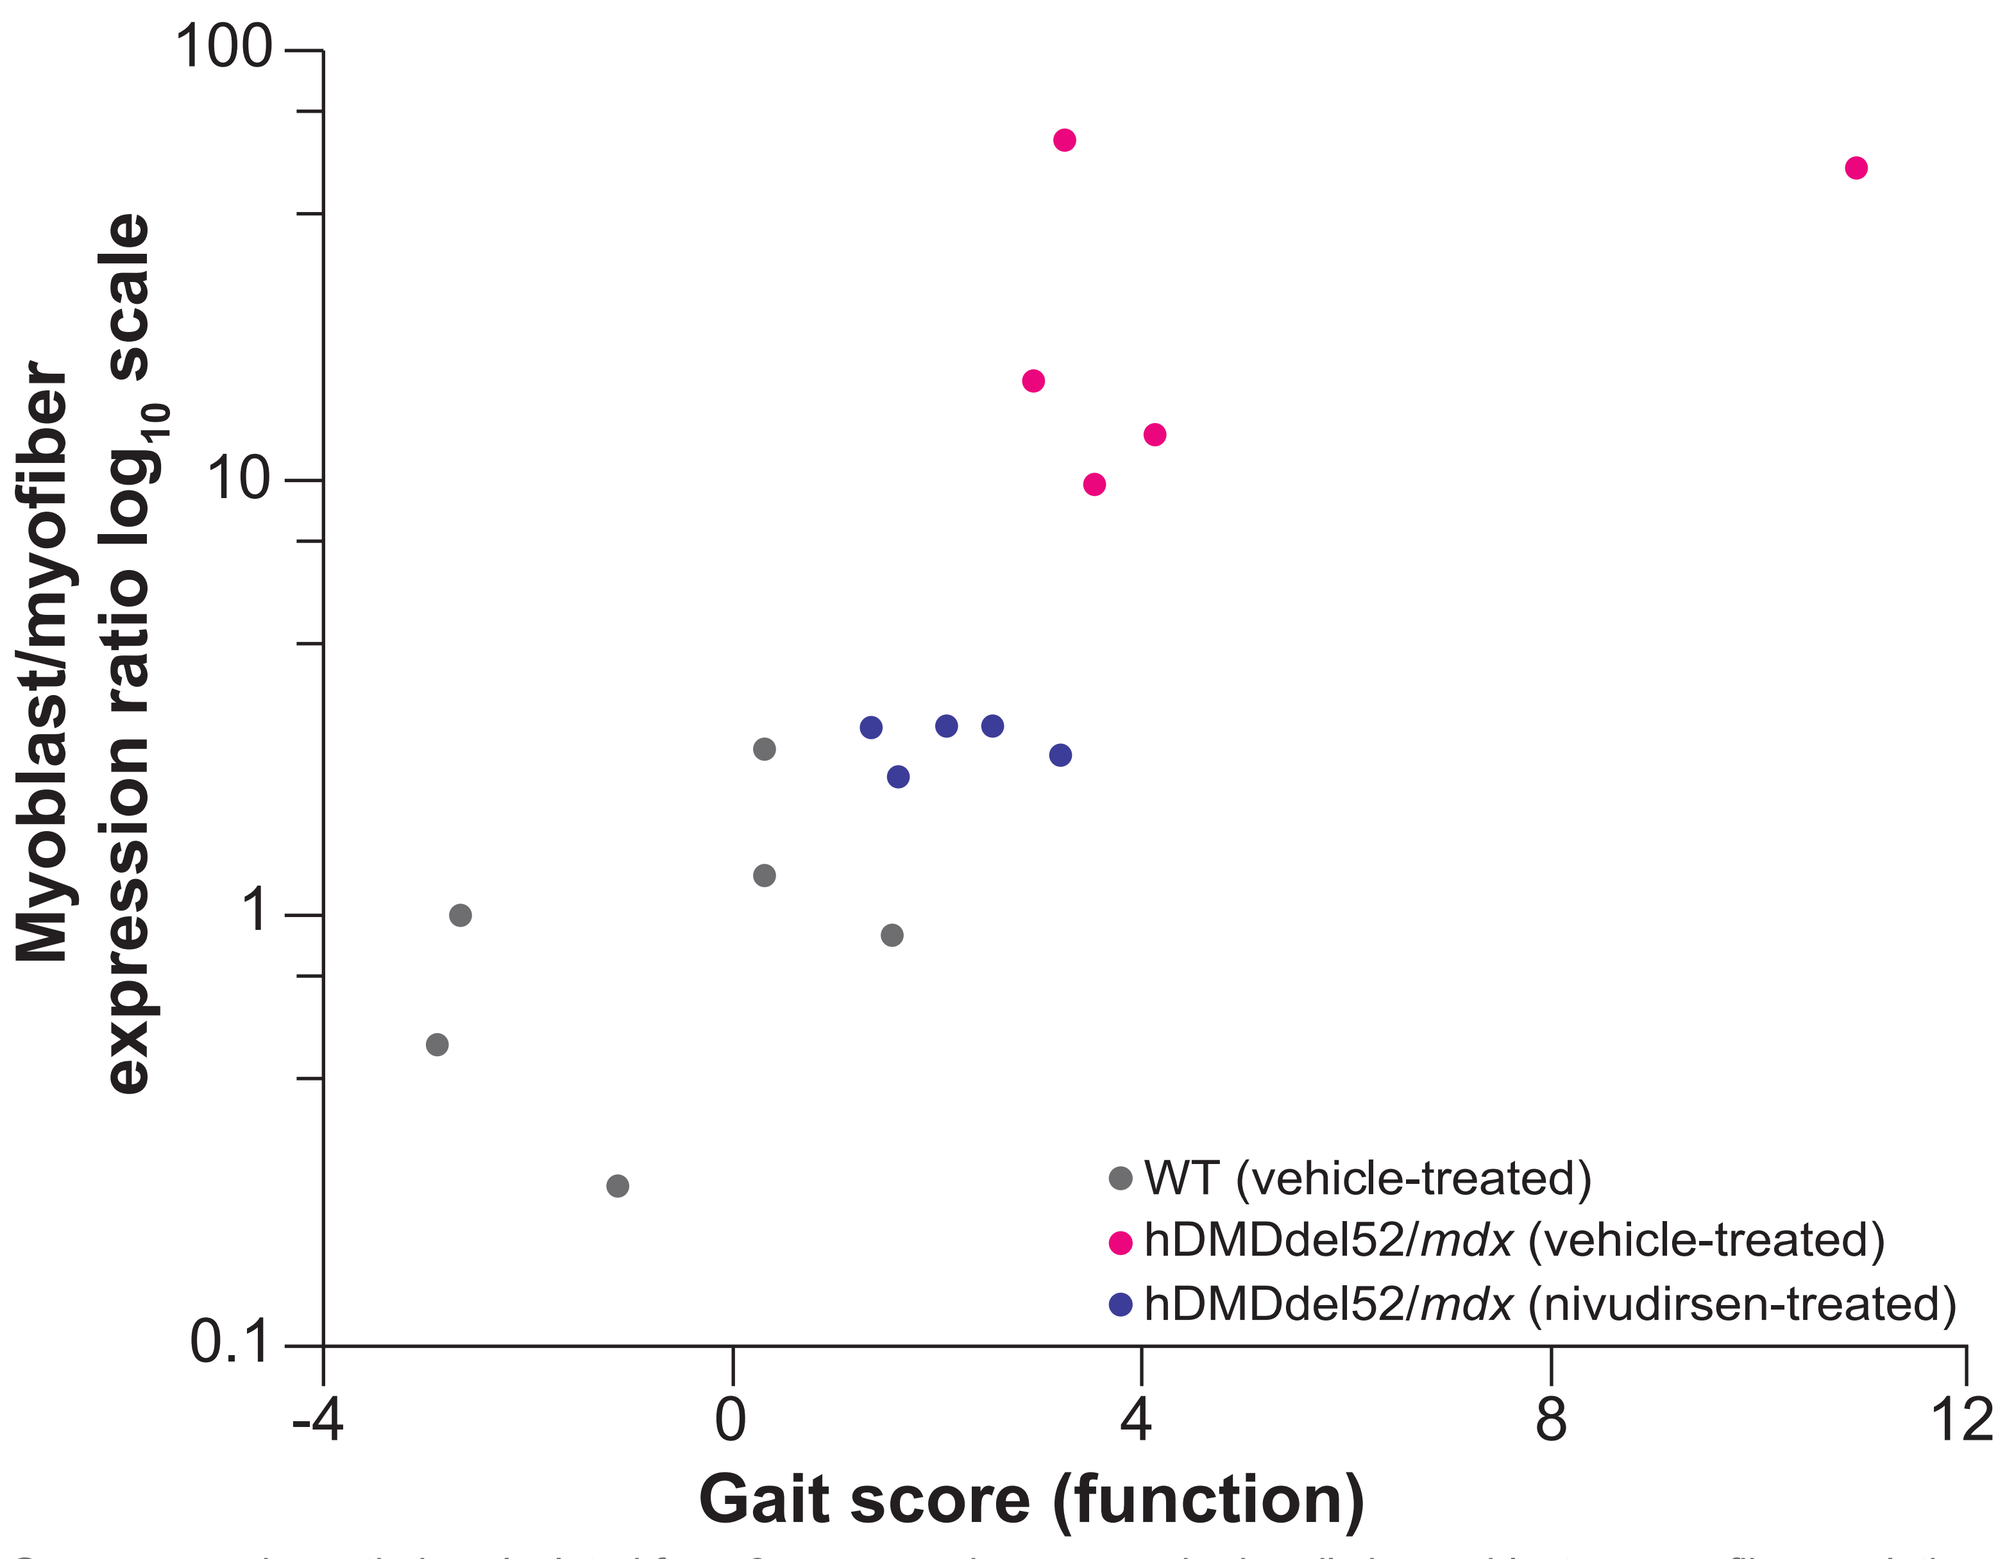
Myoblast/myofiber expression ratio log10 scale
100
10
1
0.1
-4
0
4
8
12
Gait score (function)
WT (vehicle-treated)
hDMDdel52/mdx (vehicle-treated)
hDMDdel52/mdx (nivudirsen-treated)

Gene expression ratio is calculated from 3 genes each expressed primarily in myoblasts or myofibers, relative to WT expression levels. Gait score was determined using fine motor kinematic testing; lower gait score indicates higher function.[3]
DMD, Duchenne muscular dystrophy; WT, wild-type.

- Serum LDH, serum CK, and urinary titin are elevated in untreated individuals with DMD compared with the expected range in a healthy pediatric population, reflecting muscle damage (**Figure 9**)

Figure 9. Pre-study characterization of systemic muscle injury biomarkers: Elevation in untreated individuals with DMD

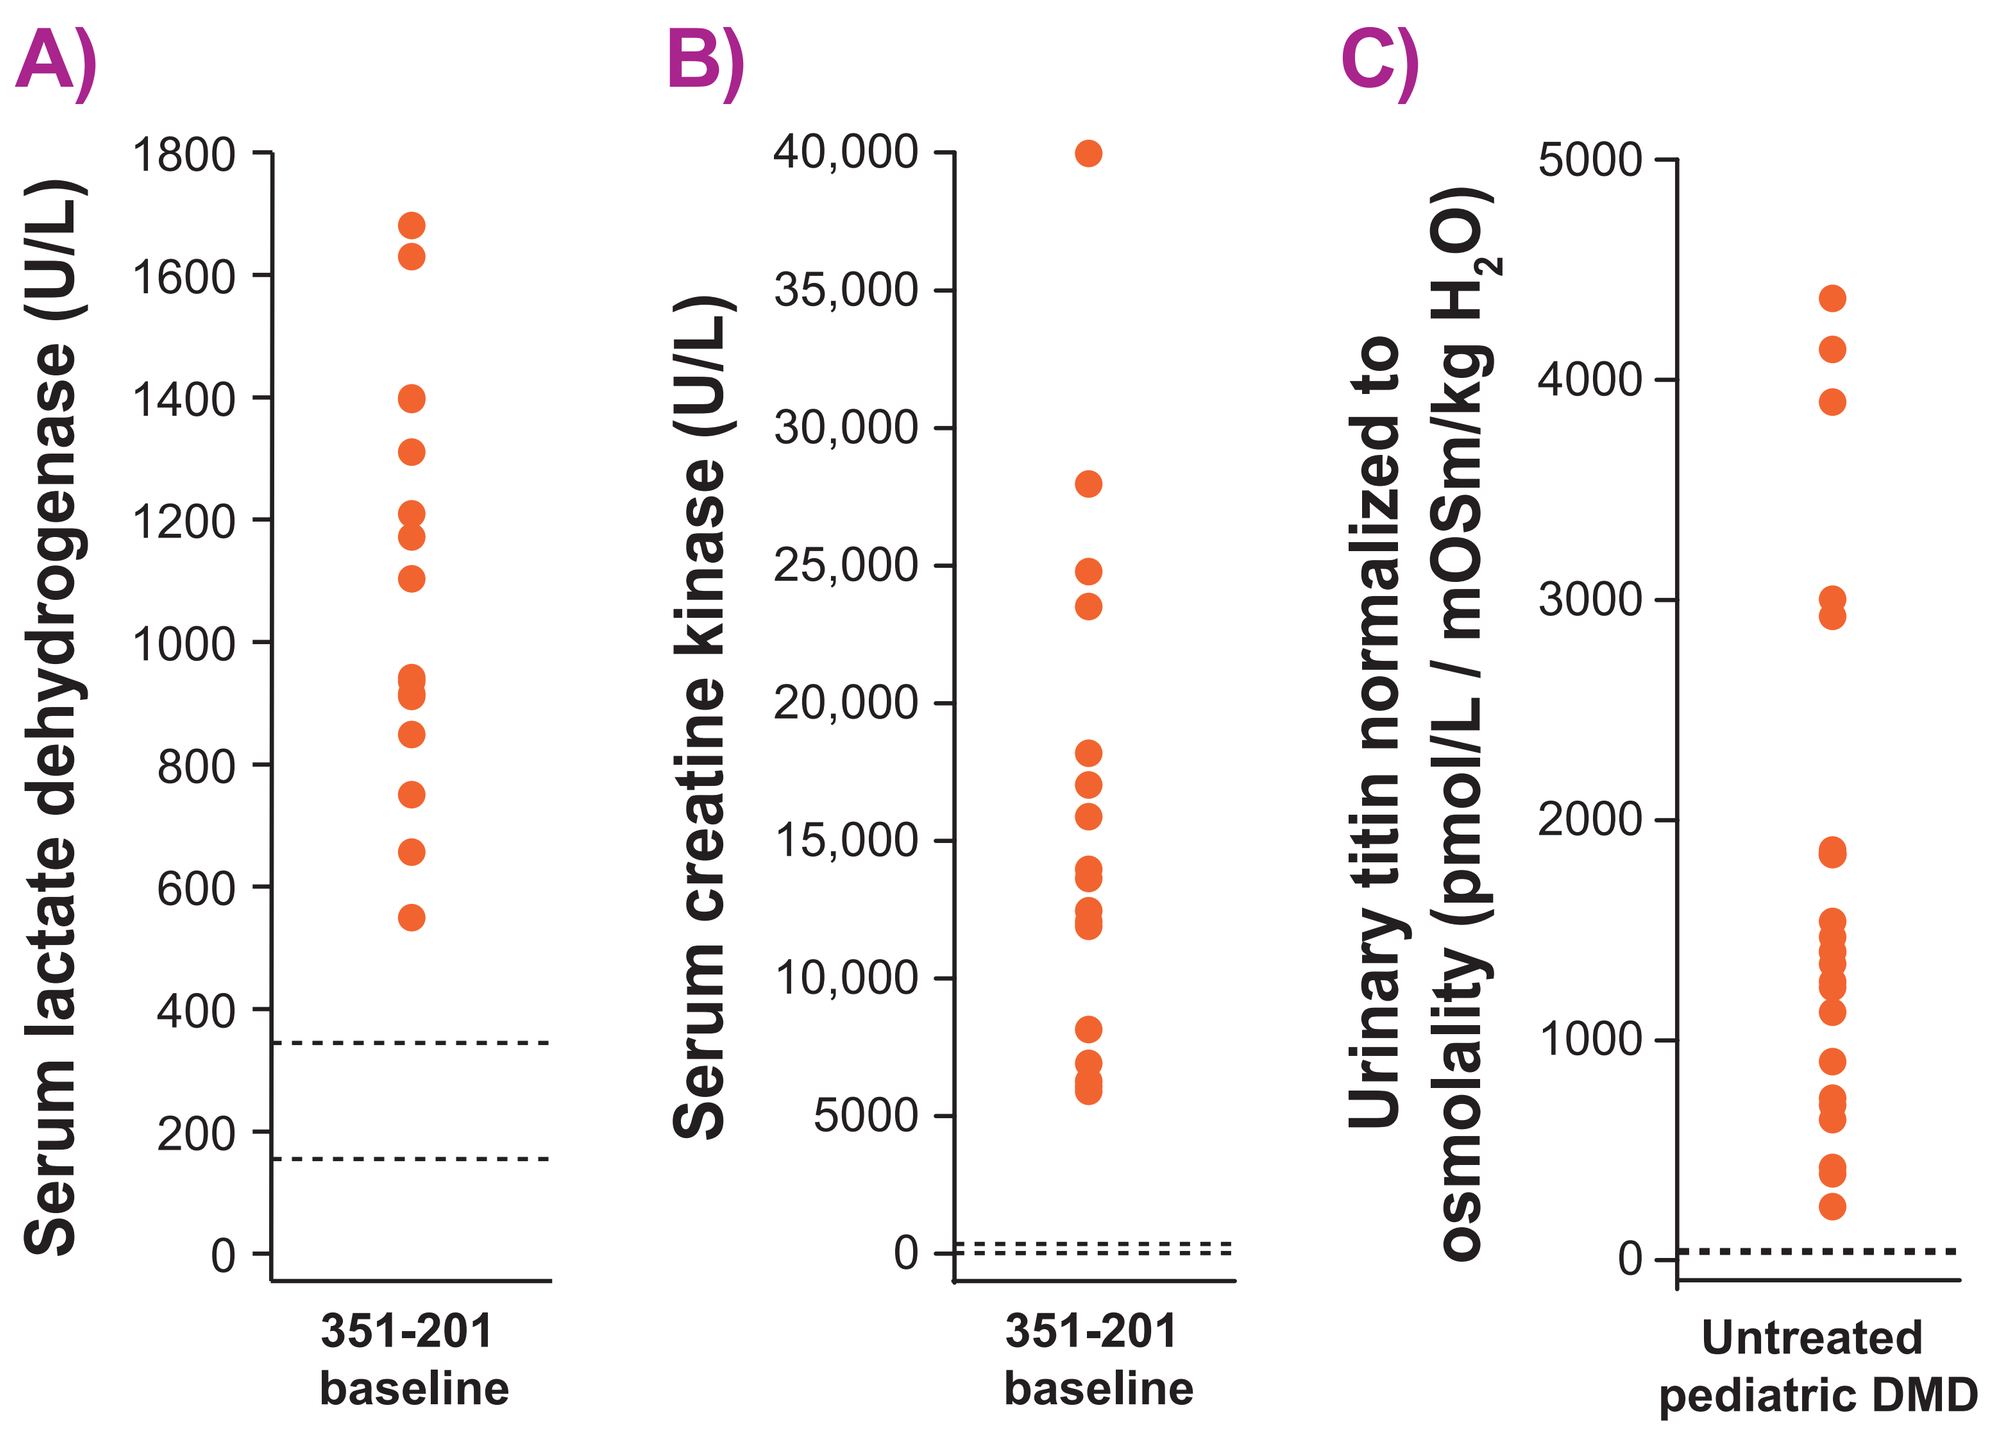
A)
Serum lactate dehydrogenase (U/L)
1800
1600
1400
1200
1000
800
600
400
200
0
351-201 baseline
B)
Serum creatine kinase (U/L)
40,000
35,000
30,000
25,000
20,000
15,000
10,000
5000
0
351-201 baseline
C)
Urinary titin normalized to osmolality (pmol/L / mOSm/kg H2O)
5000
4000
3000
2000
1000
0
Untreated pediatric DMD

Points represent baseline results, and dotted lines represent the range expected for a healthy pediatric population aged 4 to 9 years.
DMD, Duchenne muscular dystrophy

Muscle gene expression, myoblast-to-myofiber gene expression ratio, and systemic muscle injury biomarker results from study 351-201 are presented in poster #2.59eP

Biomarkers of muscle injury

Conclusions

- Together, the data from this suite of measures uniquely provide a deep and holistic understanding of the PK and PD of nivudirsen
- These assays have been leveraged in preclinical and clinical studies to understand the molecular and cellular changes resulting from nivudirsen treatment
- The preclinical and clinical data presented here, together with the subsequent ongoing phase 1/2 study, support the potential for benefit from nivudirsen treatment in individuals with DMD

References

1. Bladen CL, et al. *Hum Mutat.* 2015;36(4):395–402. **2.** Duan D, et al. *Nat Rev Dis Primers.* 2021;7(1):13. **3.** Oppeneer T, et al. *Nucleic Acid Ther.* 2025;35(2):68–80. **4.** Oppeneer T, et al. *Nucleic Acid Ther.* 2025;35(2):81–92.

Acknowledgments

We thank all trial participants, their families, study-site staff, and investigators. This research was funded by BioMarin Pharmaceutical Inc. Medical writing support was provided by Tony Sallese, PhD, of Red Nucleus, and funded by BioMarin Pharmaceutical Inc.

Disclosures

KL, **KOM**, **HZ**, **KH**, **EW**, **LW**, **MA**, **LS**, **AG**, **AY**, **KJ**, **PC**, **RP**, **PV**, and **AM** are employees and shareholders of BioMarin Pharmaceutical Inc.

Copies of the poster that are obtained through the QR code are for personal use only and may not be reproduced without permission from the authors.

PK/PD of nivudirsen | **Biomarkers of muscle injury** | **351-201 clinical data**

Presented at the 31st annual Congress of the World Muscle Society: September 29th – October 3rd; Hiroshima, Japan

©2026 BioMarin Pharmaceutical Inc. All rights reserved.